38 Measuring Withdrawal in a Phase 3 Study of a New Analgesic, NKTR-181, in Subjects With Moderate to Severe Chronic Low-Back Pain

Jack E. Henningfield,[1] John Markman,[2] Jeffrey Gudin,[3] Richard Rauck,[4] Joseph Gimbel,[5] Ivan Gergel,[6] Mary Tagliaferri,[6] Lin Lu,[6] Nathaniel P. Katz,[7] Suresh Siddhanti,[6] Sid Schnoll[1]

[1]Pinney Associates, Bethesda, MD and The Johns Hopkins University School of Medicine, Baltimore, MD; [2]University of Rochester School of Medicine and Dentistry, Rochester, NY; [3]Englewood Hospital and Medical Center, Englewood, NJ; [4]Carolinas Pain Institute and The Center for Clinical Research, Winston-Salem, NC; [5]Arizona Research Center, Phoenix, AZ; [6]Nektar Therapeutics, San Francisco, CA; [7]Tufts University School of Medicine, Boston, MA

NEKTAR®

Purpose

- Conventional opioid analgesics can produce withdrawal symptoms after cessation of chronic administration, which can lead to drug craving and opioid misuse. Certain extended-release formulations may produce intermittent symptoms of withdrawal even while patients are on therapy as a result of pharmacokinetic variability and patient metabolism. NKTR-181 is a new chemical entity, orally administered full mu-opioid receptor agonist designed to have an intrinsically slow rate of entry into the central nervous system, compared with conventional opioids.[1] It has a longer duration of action than many opioids with a half-life of approximately 12 hours, thus clinical dosing is twice daily. In a human abuse potential study of recreational opioid users, drug-high and drug-liking scores for NKTR-181 administered across the therapeutic dose range of 100 to 400 mg were significantly lower than those for oxycodone and closely resembled the scores for placebo.[2] At the supratherapeutic dose of 1200 mg, drug-liking scores exceeded placebo but were significantly lower than those produced by 60 mg oral oxycodone, and peaked more than an hour later with a more gradual increase and decline than oral oxycodone (60 and 40 mg) consistent with its slower onsetting and offsetting effects In an enriched-enrollment, randomized-withdrawal study of opioid-naïve subjects with chronic low-back pain (SUMMIT-07), NKTR-181 administered at 100 to 400 mg twice daily was associated with a significantly greater analgesic effect compared with placebo.
- In the study, opioid withdrawal was evaluated using the Clinical Opiate Withdrawal Scale (COWS) and the Subjective Opiate Withdrawal Scale (SOWS). Here we present measurements of withdrawal for NKTR-181 and placebo patients during the randomized treatment and safety follow-up period in the SUMMIT-07 study.

Methods

- This enriched-enrollment, double-blind, randomized-withdrawal study included a screening period, an open-label titration period, a 12-week double-blind treatment period, a 1-week study-drug taper, and a 2-week safety follow-up period. All study participants were opioid-naïve adults with moderate to severe chronic, non-neuropathic low-back pain of ≥6-month duration, for which non-opioid analgesia therapies had been inadequate. Opioid naïve is defined as taking no more than 10 mg morphine sulfate equivalent per day of short-acting opioids for 14 days prior to the study. Subjects with a COWS score >12 at the start of the study, indicating at least moderate withdrawal, were excluded prior to open-label titration. During open-label titration, NKTR-181 was initiated at 100 mg twice daily and adjusted to an effective and stable analgesic dose (maximum of 400 mg twice daily). Subjects achieving adequate efficacy were randomized to double-blind treatment with NKTR-181 or placebo. Those who were randomized to placebo had their NKTR-181 dose stopped abruptly without taper. For 2 weeks following randomization, subjects were allowed 5 mg hydrocodone/300 mg acetaminophen for breakthrough pain (1 tablet every 6 hours as needed). After 2 weeks, up to 2 tablets of acetaminophen per day was permitted. After 12 weeks of randomized treatment, subjects began a 1-week taper of NKTR-181 or placebo.
- Opioid withdrawal associated with NKTR-181 or placebo was assessed by COWS and SOWS throughout the trial. These scales were originally validated in patients with active substance abuse with opioids.[3,4] The COWS is an 11-item investigator-administered scale to assess clinical symptoms related to opiate withdrawal (**Table 1A**). The SOWS is a self-administered scale to assess 16 symptoms of opioid withdrawal (**Table 1B**). Greater scores correspond to increased perception of any of the 16 symptoms.
- Withdrawal, in addition to potential abuse, was also assessed by the Misuse, Abuse, and Diversion Drug Event Reporting System (MADDERS), which is a rigorous approach in which adverse events and drug accountability discrepancies that signify potentially abuse-related events are systematically captured, then reviewed and adjudicated by experts in assessing abuse potential, physical dependence and withdrawal.[5,6] Critically, potential withdrawal symptoms were assessed during three different phases of the study: 1) early termination of NKTR-181 during titration; 2) randomization from NKTR-181 to placebo during double-blind, randomized treatment; and 3) tapering from active drug at the end randomized treatment.
- COWS and SOWS scores were obtained at pre-specified time points or following early discontinuation (**Figure 1**).
- During randomized treatment, COWS scores were obtained at days 1, 8, 15, and 85. For subjects who completed randomized treatment, COWS scores were also obtained at the end of study-drug tapering and twice during the safety follow-up period. However, COWS scores were obtained at the end of the safety follow-up period only for subjects who did not enter a long-term NKTR-181 safety study. During randomized treatment, SOWS scores were obtained at day 1, twice daily for the next 3 days, and then daily through day 15. For subjects who completed randomized treatment, SOWS scores were also obtained twice daily for the first 3 days of study-drug tapering and daily during the safety follow-up period. COWS and SOWS findings are summarized descriptively.

Table 1. COWS and SOWS Items and Scoring

A.

COWS: 11-Item Scale Scored Variably from 0–5 Total Score Ranges from 0–48
• Resting pulse rate (0–4)
• Sweating (0–4)
• Restlessness (0–5)
• Pupil size (0–5)
• Bone or joint aches (0–4)
• Runny nose or tearing (0–4)
• GI upset (0–5)
• Tremor (0–4)
• Yawning (0–4)
• Anxiety or irritability (0–4)
• Gooseflesh skin (0–5)

COWS Score	Withdrawal
5–12	Mild
13–24	Moderate
25–36	Moderately Severe
>36	Severe Withdrawal

B.

SOWS: 16-Item Scale Scored Variably from 0–4 Total Score Ranges from 0–64
• I feel anxious
• I feel like yawning
• I am perspiring
• My eyes are teary
• My nose is running
• I have goosebumps
• I am shaking
• I have hot flushes
• I have cold flushes
• My bones and muscles ache
• I feel restless
• I feel nauseous
• I feel like vomiting
• My muscles twitch
• I have stomach cramps
• I feel like using now

COWS, Clinical Opiate Withdrawal Scale; SOWS, Subjective Opiate Withdrawal Scale.

Figure 1. COWS and SOWS Observation Time Points During Each Study Period

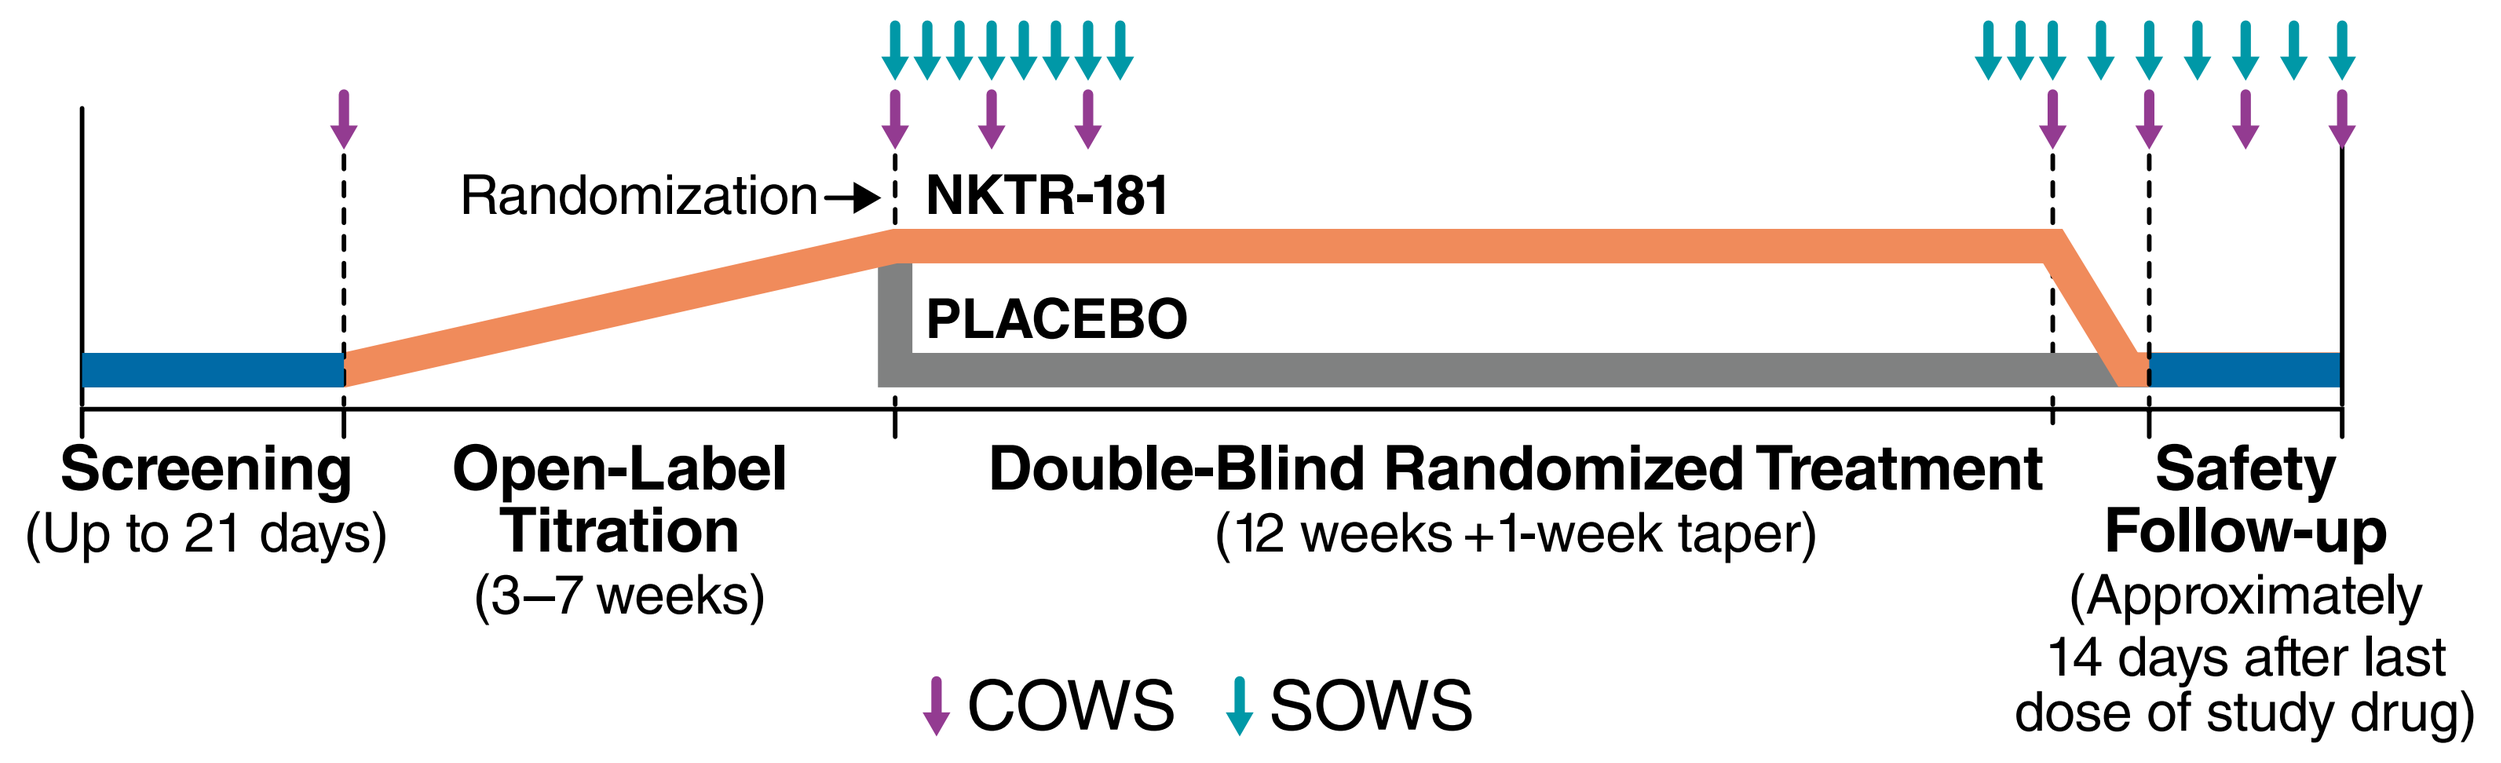

Randomization
NKTR-181
PLACEBO
Screening
(Up to 21 days)
Open-Label Titration
(3–7 weeks)
Double-Blind Randomized Treatment
(12 weeks +1-week taper)
Safety Follow-up
(Approximately 14 days after last dose of study drug)
COWS
SOWS

COWS, Clinical Opiate Withdrawal Scale; SOWS, Subjective Opiate Withdrawal Scale

Results

- A total of 1,189 subjects were exposed to NKTR-181 during open-label titration, and 610 were randomized, 309 to NKTR-181 and 301 to placebo. Characteristics of the randomized patients (the study's intention-to-treat population) are summarized by treatment group in **Table 2**.

Table 2. Subjects' Characteristics By Randomized Treatment Group (ITT Population)

Characteristic		NKTR-181 Group (N=309)	Placebo Group (N=301)
Age (years)	Mean (SD)	52.0 (12.7)	50.7 (12.5)
	Range	20–74	20–75
Sex, n (%)	Female	187 (60.5%)	170 (56.5%)
	Male	122 (39.5%)	131 (43.5%)
Race, n (%)	White	205 (66.3%)	196 (65.1%)
	Black	95 (30.7%)	93 (30.9%)
	Other	9 (2.9%)	12 (4.0%)
Time since LBP onset (years), mean (SD)		13.3 (10.0)	13.0 (9.8)
Pain score,[a] mean (SD)	At screening	6.70 (0.98)	6.76 (0.91)
	At baseline[b]	2.29 (1.08)	2.35 (1.09)
COWS score at screening, Mean (SD)		0.5 (0.9)[c]	0.5 (0.9)[d]
Withdrawal classification, n (%)			
	None	305 (99.0%)[c]	298 (99.3%)[d]
	Mild	3 (1.0%)[c]	2 (0.7%)[d]
	Moderate	0[c]	0[d]
	Moderately severe	0	0
	Severe	0	0

[a]Seven-day average of daily scores on an 11-point rating scale ranging from 0 ("No pain") to 10 ("Pain as bad as you can imagine"); [b]The end of open-label NKTR-181 titration, immediately preceding randomization; [c]N=308; [d]N=300.
COWS, Clinical Opiate Withdrawal Scale; ITT, intention-to-treat; LBP, low-back pain; SD, standard deviation.

Table 3. COWS Scores At and After Randomization (ITT Population)

Time Point	Withdrawal Classification (COWS Score),[a] n (%)	NKTR-181 Group (N=309)	Placebo Group (N=301)
Randomization	None	300 (99.7%)	298 (100.0%)
	Mild	1 (0.3%)	0
	Moderate	0	0
	Moderately severe	0	0
	Severe	0	0
End of week 1	None	292 (99.0%)	284 (97.6%)
	Mild	3 (1.0%)	7 (2.4%)
	Moderate	0	0
	Moderately severe	0	0
	Severe	0	0
End of week 2	None	287 (98.6%)	272 (99.6%)
	Mild	4 (1.4%)	1 (0.4%)
	Moderate	0	0
	Moderately severe	0	0
	Severe	0	0
End of week 12	None	264 (99.6%)	263 (99.6%)
	Mild	1 (0.4%)	1 (0.4%)
	Moderate	0	0
	Moderately severe	0	0
	Severe	0	0
End of tapering	None	213 (97.7%)	219 (99.5%)
	Mild	5 (2.3%)	1 (0.5%)
	Moderate	0	0
	Moderately severe	0	0
	Severe	0	0

[a]Mild, 5–12; moderate, 13–24; moderately severe, 25–36; severe, >36.
COWS, Clinical Opiate Withdrawal Scale; ITT, intention-to-treat.

- COWS scores at and after randomization are presented in **Table 3**. During randomized treatment period, COWS scores indicating mild withdrawal were recorded in 7 subjects (2.4%) in the placebo group (all scores >8) and 3 subjects (1.0%) in the NKTR-181 group on day 8, and in 1 subject (0.4%) in the placebo group (all scores ≥8) and 4 subjects (1.4%) in the NKTR-181 group on day 15. At the end of the tapering period (ie, 1 week after the end of maintenance study-drug dosing) COWS scores indicating mild withdrawal were recorded in 1 subject (0.5%) in the placebo group and 5 subjects (2.3%) in the NKTR-181 group. The maximum COWS score was 8 during this period. In both treatment groups, there were no COWS scores that indicated moderate, moderately severe, or severe withdrawal at any point during randomized treatment and the ensuing follow-up period.
- Mean SOWS scores during the first two weeks of randomized treatment are presented in **Figure 2**. Mean SOWS scores in the placebo group achieved no higher than a score of 2.7 on the 64-point SOWS scale. Mean SOWS scores in subjects randomized to NKTR-181 remained ≤1.8 on the 64-point SOWS scale.
- During the titration period, MADDERS identified 24/1189 (2.0%) cases of withdrawal. During and after the randomization period, MADDERS identified 9/309 (2.9%) subjects assigned to NKTR-181 and 11/301(3.7%) subjects assigned to placebo as having events of withdrawal. Thus, the difference in withdrawal rates was approximately 1% higher in the NKTR-181 group vs. placebo whether measured by either MADDERS or the COWS.

Figure 2. Mean SOWS Scores During the First Two Weeks of Randomized Treatment

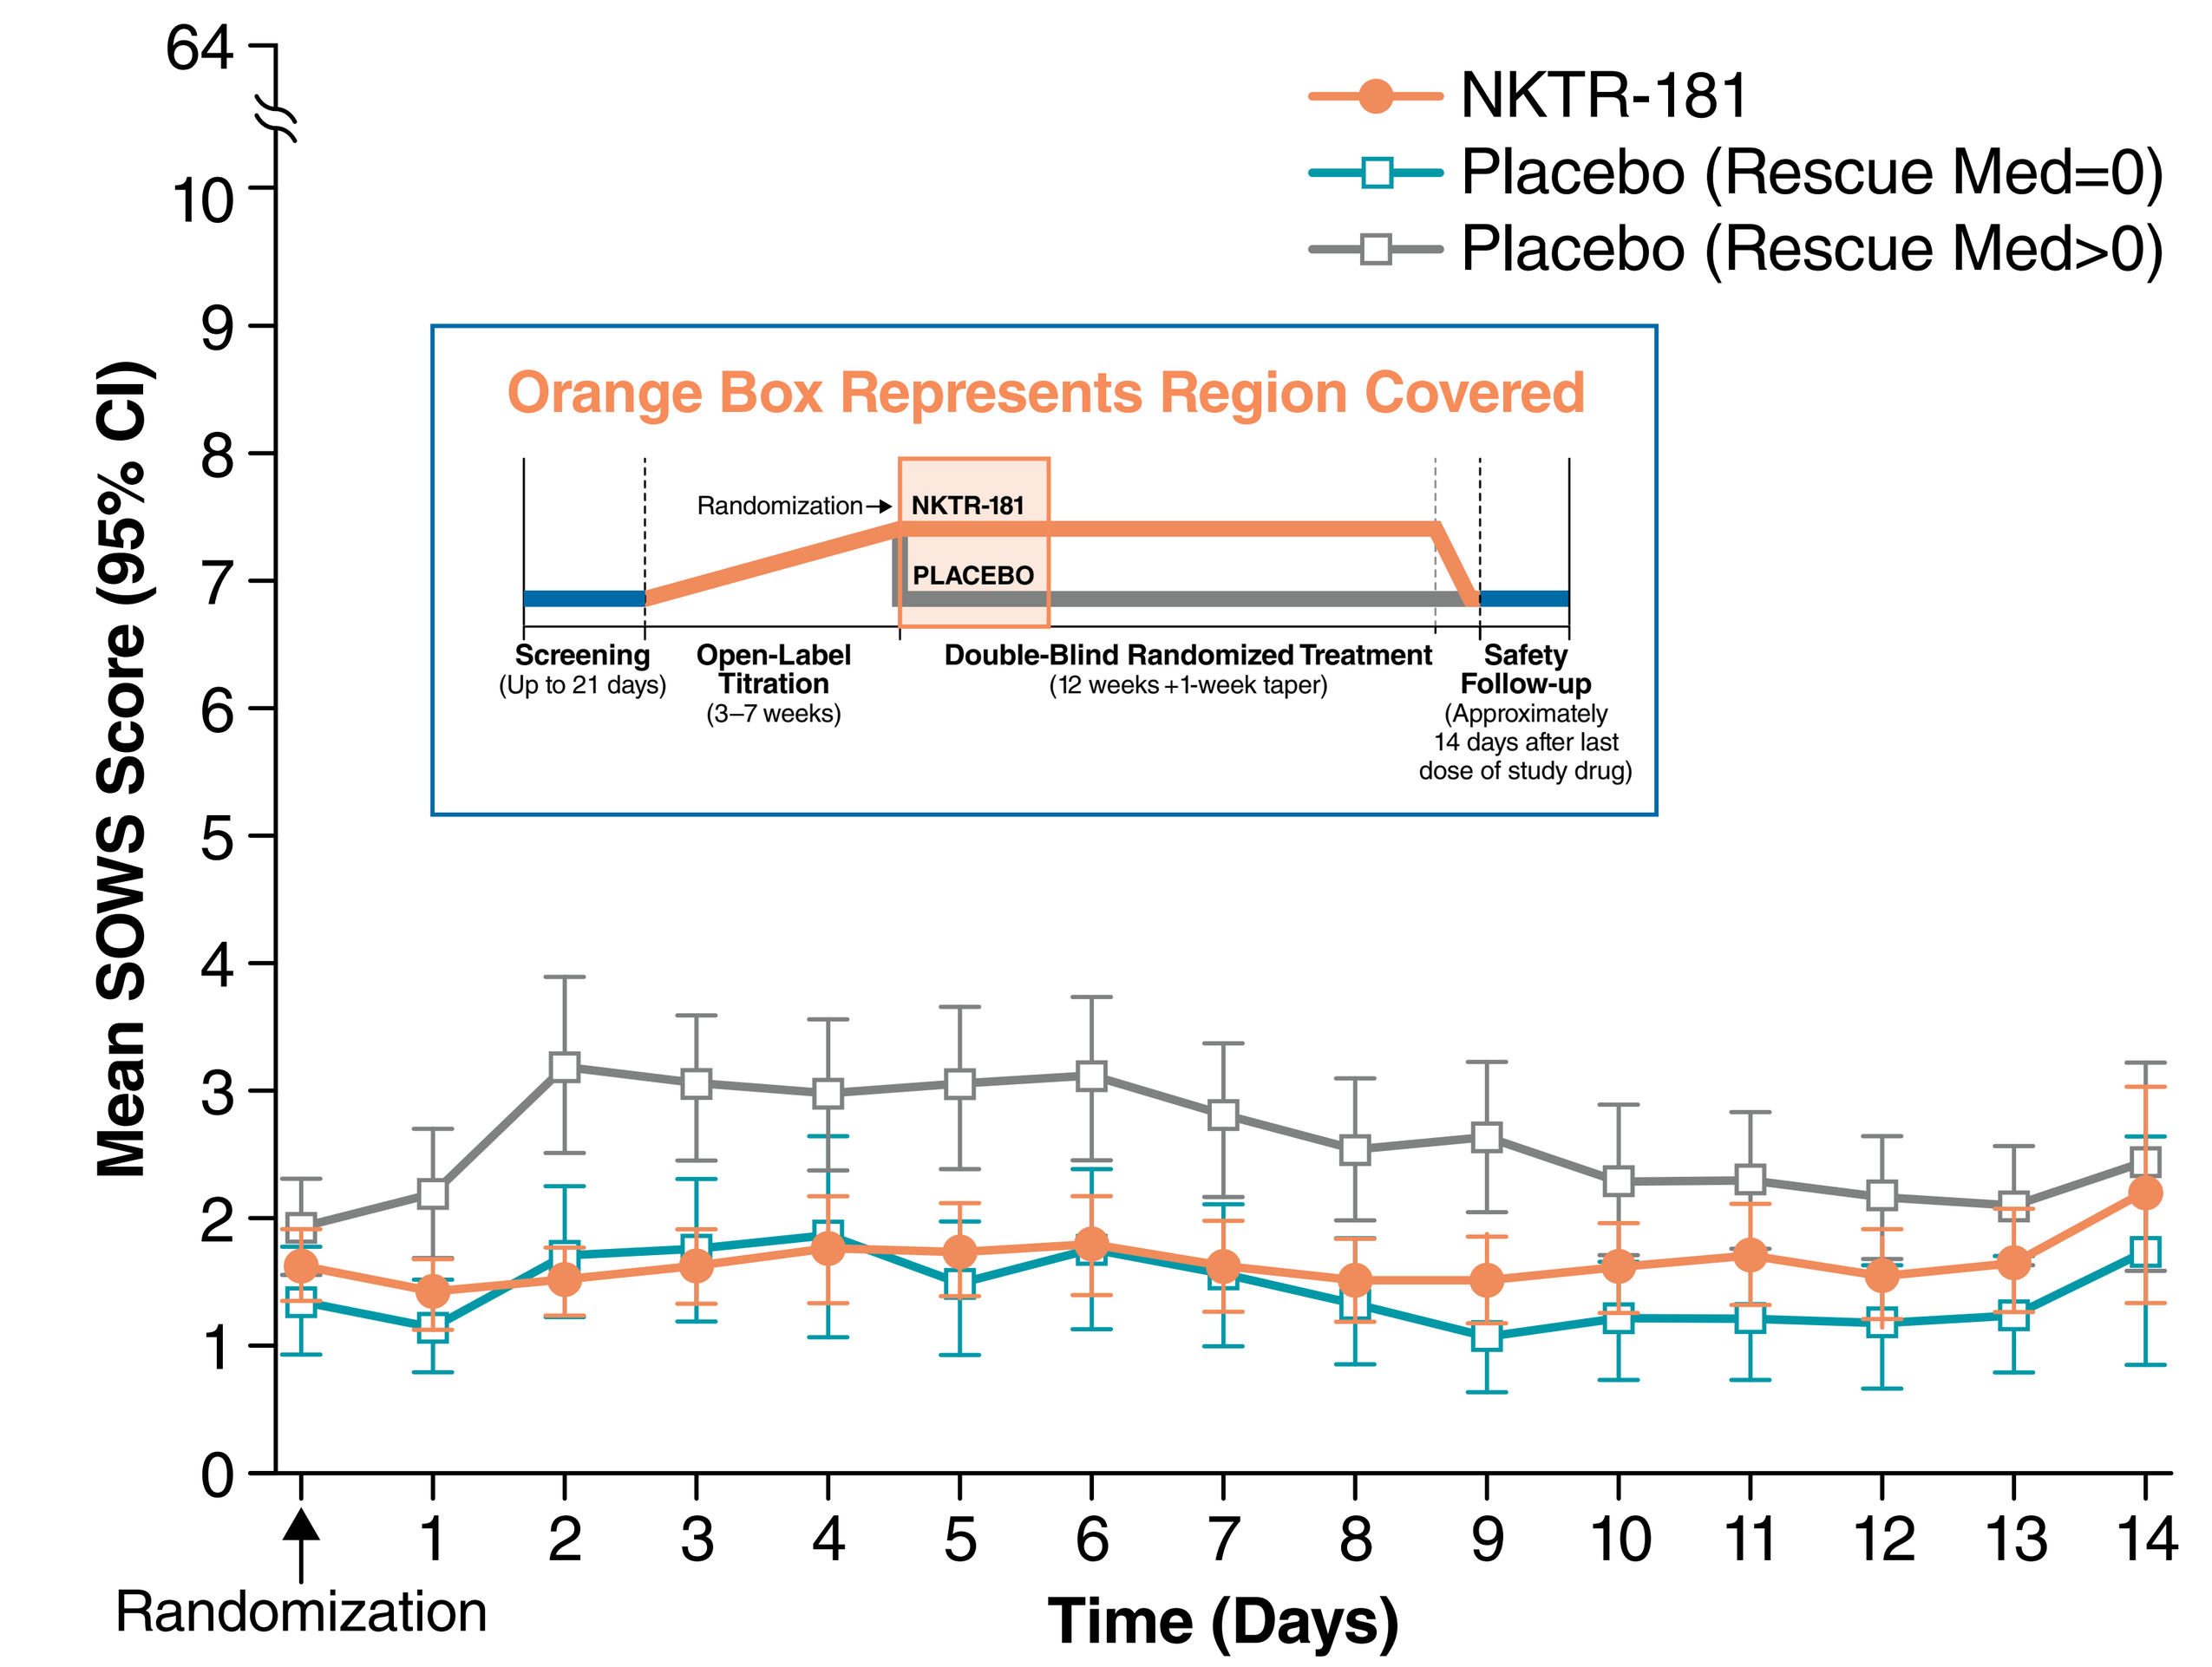

NKTR-181
Placebo (Rescue Med=0)
Placebo (Rescue Med>0)
Orange Box Represents Region Covered
Mean SOWS Score (95% CI)
Randomization
Time (Days)

	0	1	2	3	4	5	6	7	8	9	10	11	12	13	14
Placebo	111	111	110	111	107	107	110	108	108	108	109	111	103	85	40
Placebo	182	183	182	184	177	180	180	178	183	178	181	183	177	165	73
NKTR-181	299	295	298	300	296	296	293	288	294	291	298	289	279	250	107

CI, confidence interval; ITT, intention-to-treat; SOWS, Subjective Opiate Withdrawal Scale.

- Mean SOWS scores during and after the tapering period are presented in **Figure 3**. In both treatment groups, the mean score remained <2.7, with no discernible increase at any time point.

Figure 3. Mean SOWS Scores During and After the Tapering Period

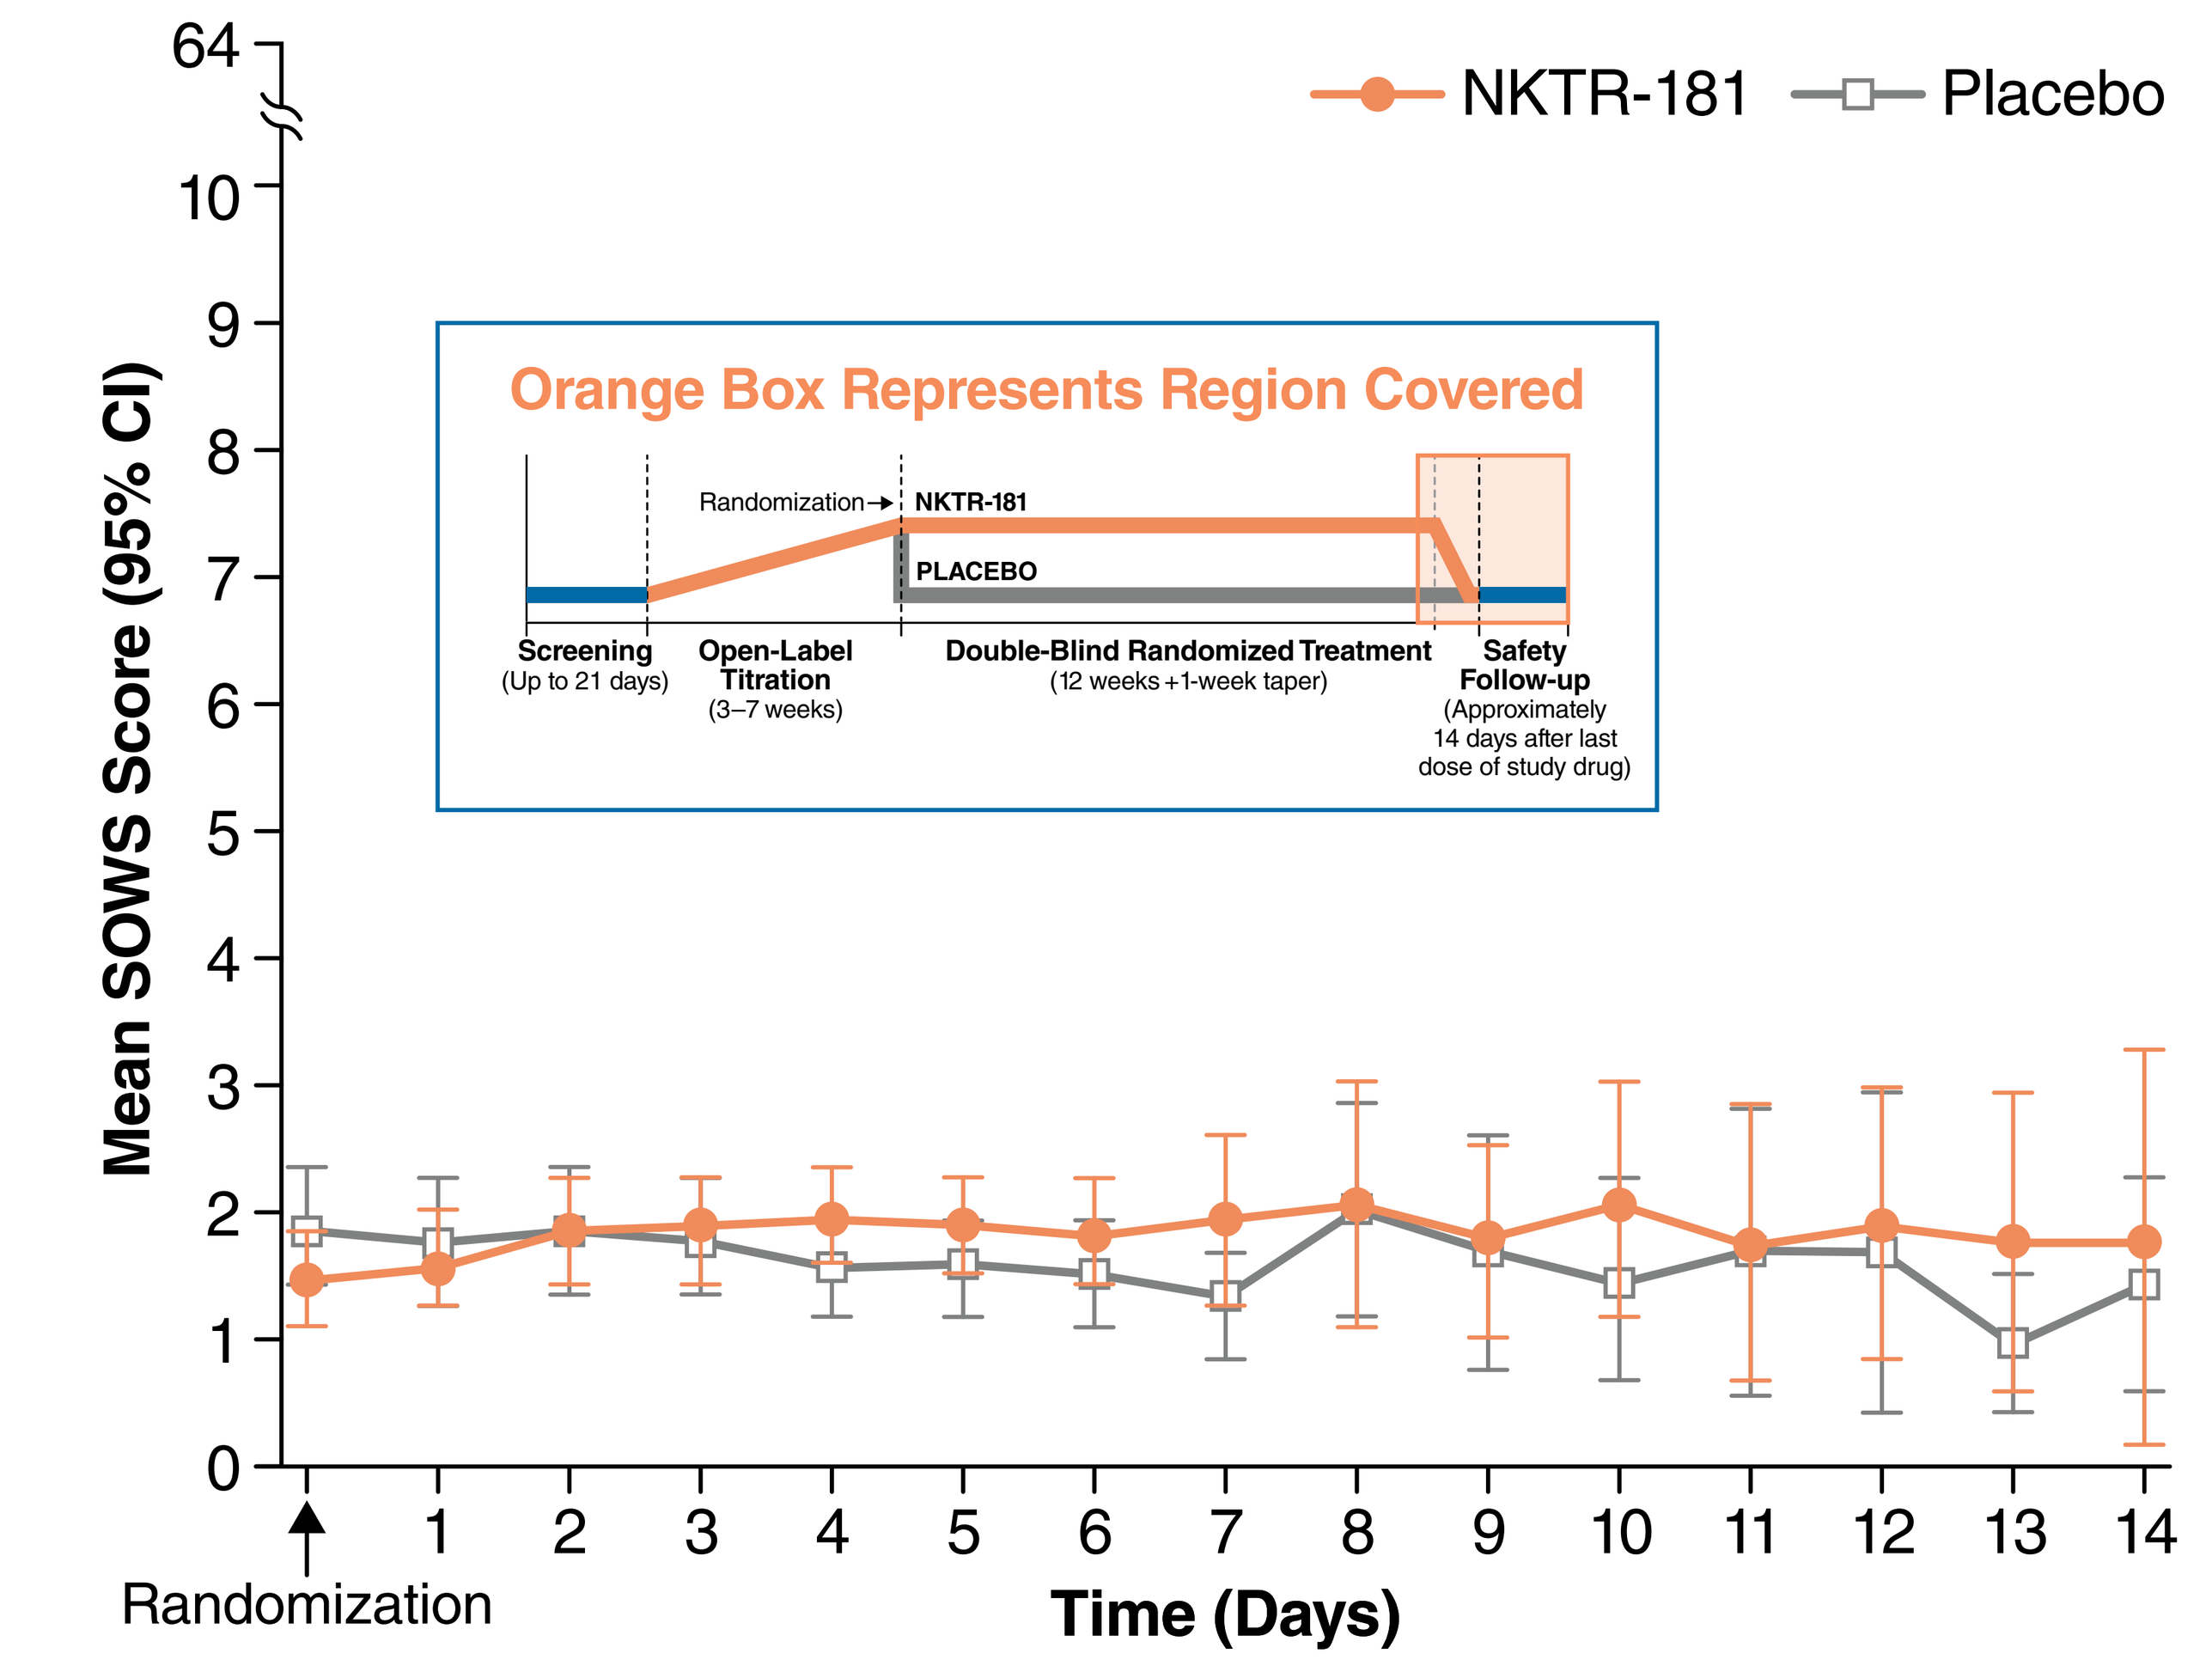

NKTR-181
Placebo
Orange Box Represents Region Covered
Mean SOWS Score (95% CI)
Randomization
Time (Days)

	0	1	2	3	4	5	6	7	8	9	10	11	12	13	14
Placebo	251	246	248	244	239	237	205	100	80	65	63	57	51	50	40
NKTR-181	259	267	265	269	262	252	218	114	86	77	72	67	62	60	43

CI, confidence interval; ITT, intention-to-treat; SOWS, Subjective Opiate Withdrawal Scale.

Conclusions

- In the SUMMIT-07 study, NKTR-181 administered at 100 to 400 mg twice daily to subjects with chronic low-back pain exhibited a low rate and intensity of opioid withdrawal, as rated by clinicians and subjects themselves. We observed no major difference in withdrawal symptoms between NKTR-181 and placebo following abrupt NKTR-181 discontinuation or during 1-week study-drug taper. During randomized treatment, clinical symptoms of withdrawal were rare, with no moderate or severe cases reported based on COWS and SOWS scores, even after abrupt NKTR-181 discontinuation in subjects randomized to placebo. These results show that reducing the rate of CNS entry does not predispose patients to end-of-dose withdrawal symptoms following cessation of treatment with NKTR-181. Opioids that are associated with minimal withdrawal symptoms when discontinued may be of clinical utility in treating patients with chronic pain, in that withdrawal-related adverse events may be less common, and withdrawal-induced drug craving and associated abuse may be a lesser concern.

Support This study was funded by Nektar Therapeutics.

Disclosures **JEH** provided consulting services, with financial support provided by Nektar Therapeutics. **JM** has consulted with Pfizer, Teva, Quark, Quartet, Biogen, Nektar, ENDO, Immune Pharma, Chromocell, Collegium, Purdue, Novartis, Depomed, Allergan, Egalet, Sanofi, Aptinyx, Diaachi Sankyo, Plasmasurgical, Grunenthal, Kempharm, and Inspirion. He has received research grant funding from Pfizer and Depomed and participated on clinical trial data safety monitoring committees for Novartis and Allergan. **JGudin** has served as a consultant and received consulting fees from Nektar Therapeutics; has received consulting and speaker fees from AstraZeneca Pharmaceuticals LP, Purdue Pharma LP, and Salix Pharmaceuticals, Inc. **RR** has received research funding from Endo Pharmaceuticals and BioDelivery Sciences International Inc; has servedas consultant for Endo Pharmaceuticals; has served as a consultant and received consulting fees from Nektar Therapeutics. **JGimbel** has received investigator payments from Endo Pharmaceuticals and served as a consultant for Endo; has served as a consultant and received consulting fees from Nektar Therapeutics. **IG**, **MT**, **LL**, and **SSiddhanti** are employed by Nektar Therapeutics. **NK** is an employee of Analgesic Solutions, which consults with many companies that develop and market opioid products, and provides the MADDERS system for evaluation of abuse-related events in clinical trials. **SSchnoll** is an employee of Pinney Associates, Inc., which consults with many pharmaceutical companies that develop and market opioid products. Editorial assistance was provided by The Curry Rockefeller Group, LLC. Nektar Therapeutics provided funding for editorial support.

References

1. Miyazaki T, Choi IY, Rubas W, et al. NKTR-181: a novel mu-opioid analgesic with inherently low abuse potential [published online August 4, 2017]. *J Pharmacol Exp Ther.* 2017. doi:10.1124/jpet.117.243030. **2.** Webster L, Henningfield J, Buchhalter AR,et al. Human abuse potential of the new opioid analgesic molecule NKTR-181 compared with oxycodone [published online March 10, 2017]. *Pain Med.* 2017. doi:10.1093/pm/pnw344. **3.** Gossop M. The development of a Short Opiate Withdrawal Scale (SOWS). *Addict Behav* 1990;15(5):487–490. **4.** Handelsman L, Cochrane KJ, Aronson MJ, Ness R, Rubinstein KJ, Kanof PD. Two new rating scales for opiate withdrawal. *Am J Drug Alcohol Abuse* 1987;13(3):293–308. **5.** Smith SM, Jones JK, Katz NP, Roland CL, Setnik B, Trudeau JJ, Wright S, Burke LB, Comer SD, Dart RC, Dionne R, Haddox JD, Jaffe JH, Kopecky EA, Martell BA, Montoya ID, Stanton M, Wasan AD, Turk DC, Dworkin RH. Identification of prescription medication misuse, abuse, and related events in clinical trials: ACTTION critique and recommended considerations. *Journal of Pain*, in press. **6.** Treister R, Trudeau JJ, Van Inwegen R, Jones JK, Katz NP. Development and feasibility of the misuse, abuse, and diversion drug event reporting system (MADDERS®). *The American Journal on Addictions* 2016; 25(8):641–651.

PRESENTED AT **PAINWEEK 2017** • SEPTEMBER 5–9, 2017 • LAS VEGAS, NEVADA