Abuse Potential Assessment of Novel Opioid Analgesic NKTR-181: Implications for Labeling and Scheduling

NEKTAR

L. Webster[1], S. Smith[2], J. Silowsky[2], K. Gogas[2], A. Odinecs[2], M. A. Eldon[2], N. Abrouk[2], R. Medve[2], J. E. Henningfield[3], A. R. Buchhalter[3], E. J. Cone[3], R. V. Fant[3], S. H. Schnoll[3]
[1]Lifetree Clinical Research, Salt Lake City, UT; [2]Nektar Therapeutics, San Francisco, CA; [3]PinneyAssociates, Bethesda, MD

INTRODUCTION

The FDA has acknowledged the need to positively and proactively address the problem of prescription opioid abuse and recently provided Draft Guidance (Guidance for Industry Abuse-Deterrent Opioids Evaluation and Labeling, January 2013) (1) that seeks to not only establish standards for the development of abuse-deterrent products, but to provide incentive to develop these products by offering differentiated labeling claims for products that are able to meet the standards described in the Guidance.

Abuse potential refers to the likelihood that a drug is attractive for nonmedical use repeatedly or even sporadically, for the positive psychoactive effects it produces. These drugs are characterized by central nervous system (CNS) activity, in particular psychoactive effects including sedation, euphoria, perceptual and other cognitive distortions, hallucinations, and mood changes. Drugs with abuse potential often (but not always) produce psychic or physical dependence and may lead to the disorder of addiction. (2)

Assessment of the abuse potential of a new drug is based on a composite analysis of chemistry, pharmacology, and clinical data, and the potential public health risk that the drug presents. Data from human abuse potential studies is an important factor in the development of product labeling and drug scheduling recommendations. (2)

Human abuse liability (HAL) assessment is typically conducted by comparing an investigational drug to a known drug of abuse and to placebo.

NKTR-181 is a new molecule opioid designed to have a reduced rate and extent of entry into the CNS, with the intent of preserving analgesic effectiveness while reducing CNS related effects, including psychoactive effects. If NKTR-181 provides relief of moderate to severe pain comparable to conventional opioid analgesics, but with reduced CNS liabilities, it would represent a significant improvement over existing opioids.

The study described here is a single-center, randomized, double-blind, active and placebo-controlled 5-period crossover study to assess the abuse potential of the investigational new molecule NKTR-181 in non-dependent, recreational opioid users.

REFERENCES

1) FDA Guidance for Industry Abuse-Deterent Opioids Evaluation and Labeling, January 2013

2) FDA Guidance for Industry Assessment of Abuse Potential of Drugs – January 2010

SLOW CNS ENTRY AND ANALGESIC RESPONSE: Pharmacodynamic Profile of NKTR-181 in Healthy Volunteers

NKTR-181 Demonstrates Slow Rate of CNS Entry

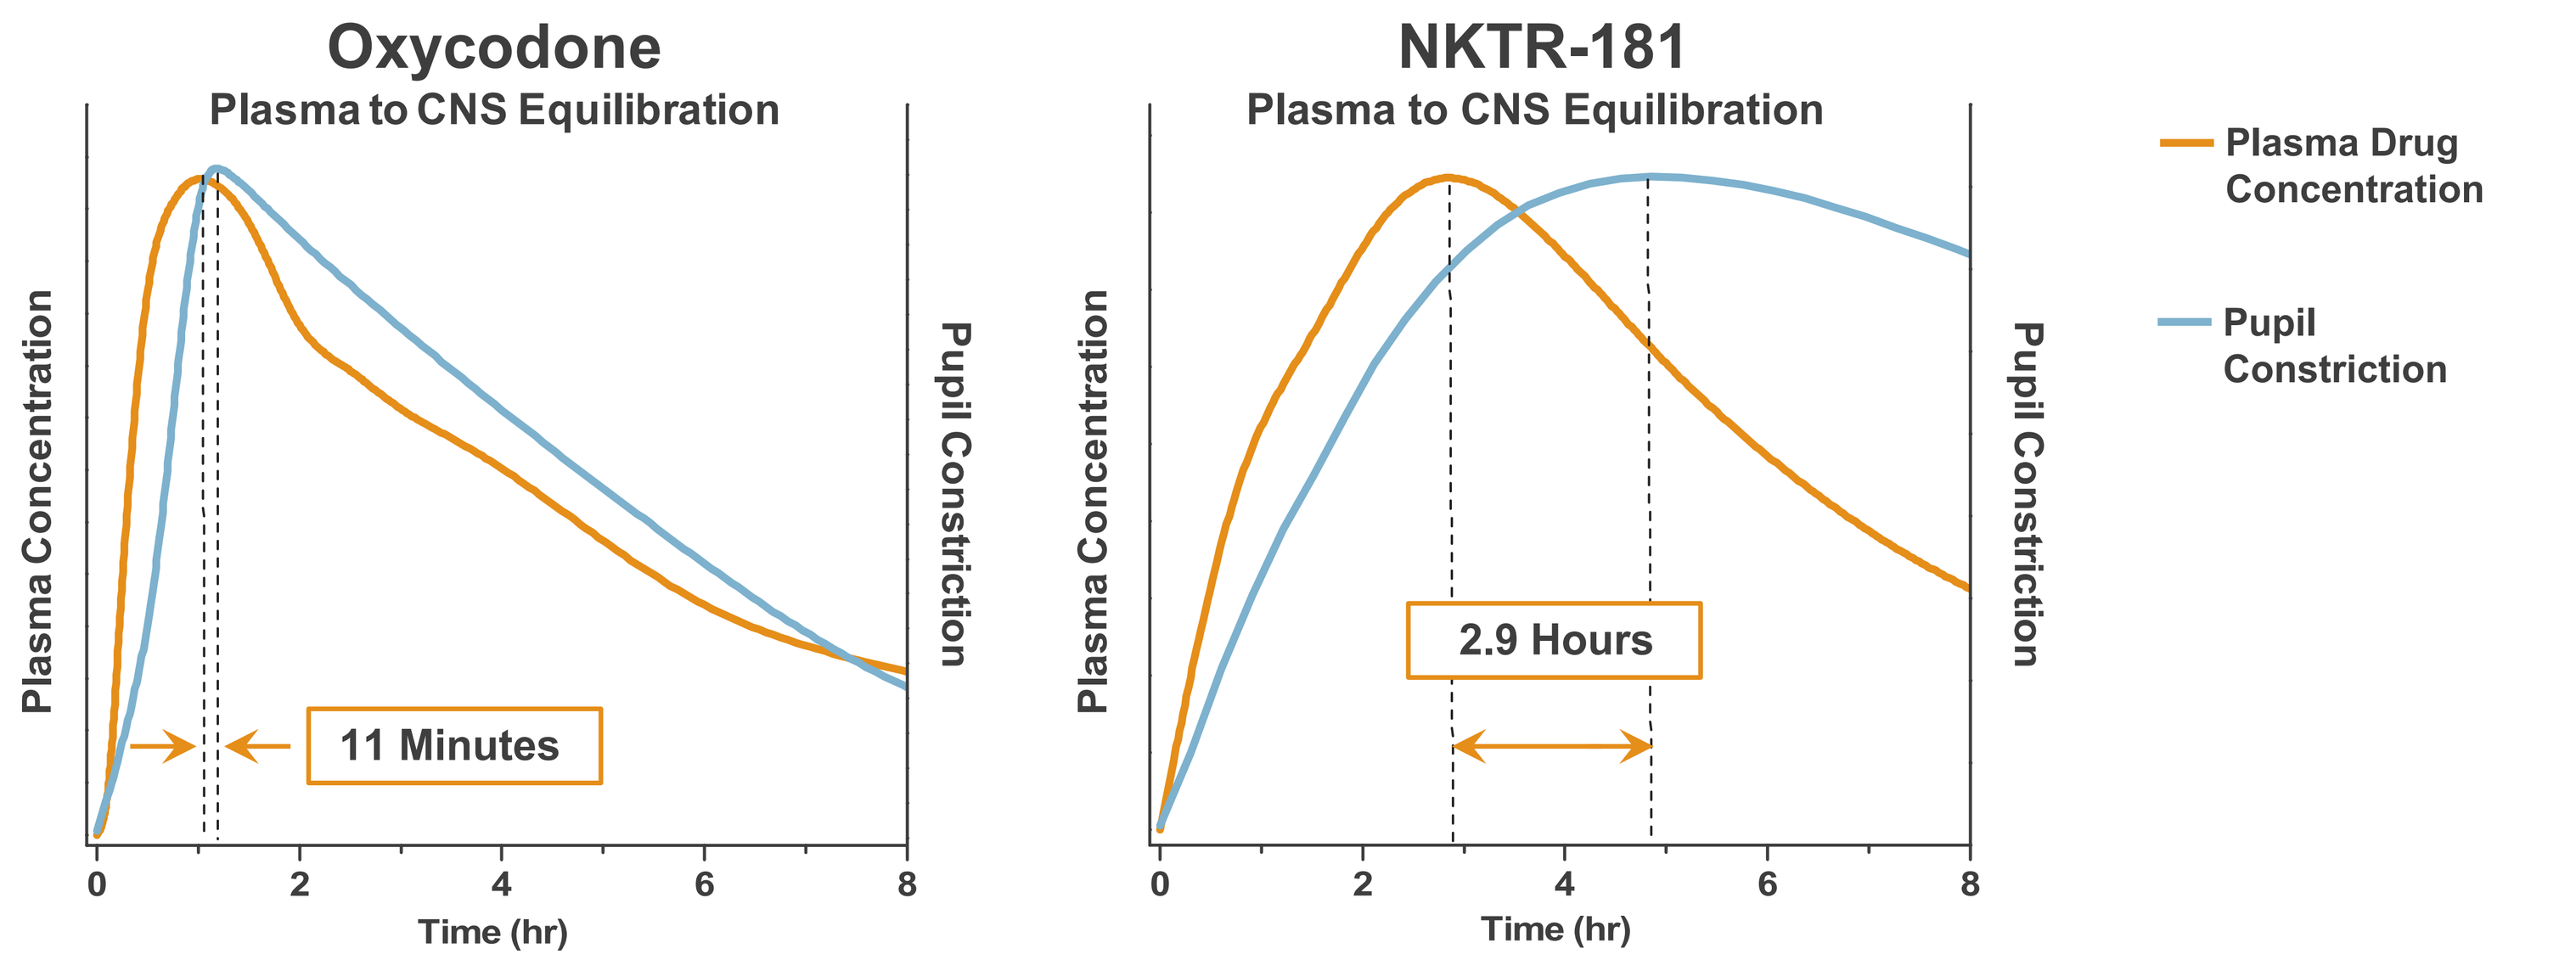
Oxycodone
Plasma to CNS Equilibration
Plasma Concentration
Pupil Constriction
11 Minutes
Time (hr)
NKTR-181
Plasma to CNS Equilibration
2.9 Hours
Plasma Drug Concentration
Pupil Constriction

Rate of CNS entry defined using pupillometry compared to plasma PK

NKTR-181 Demonstrates Analgesic Response

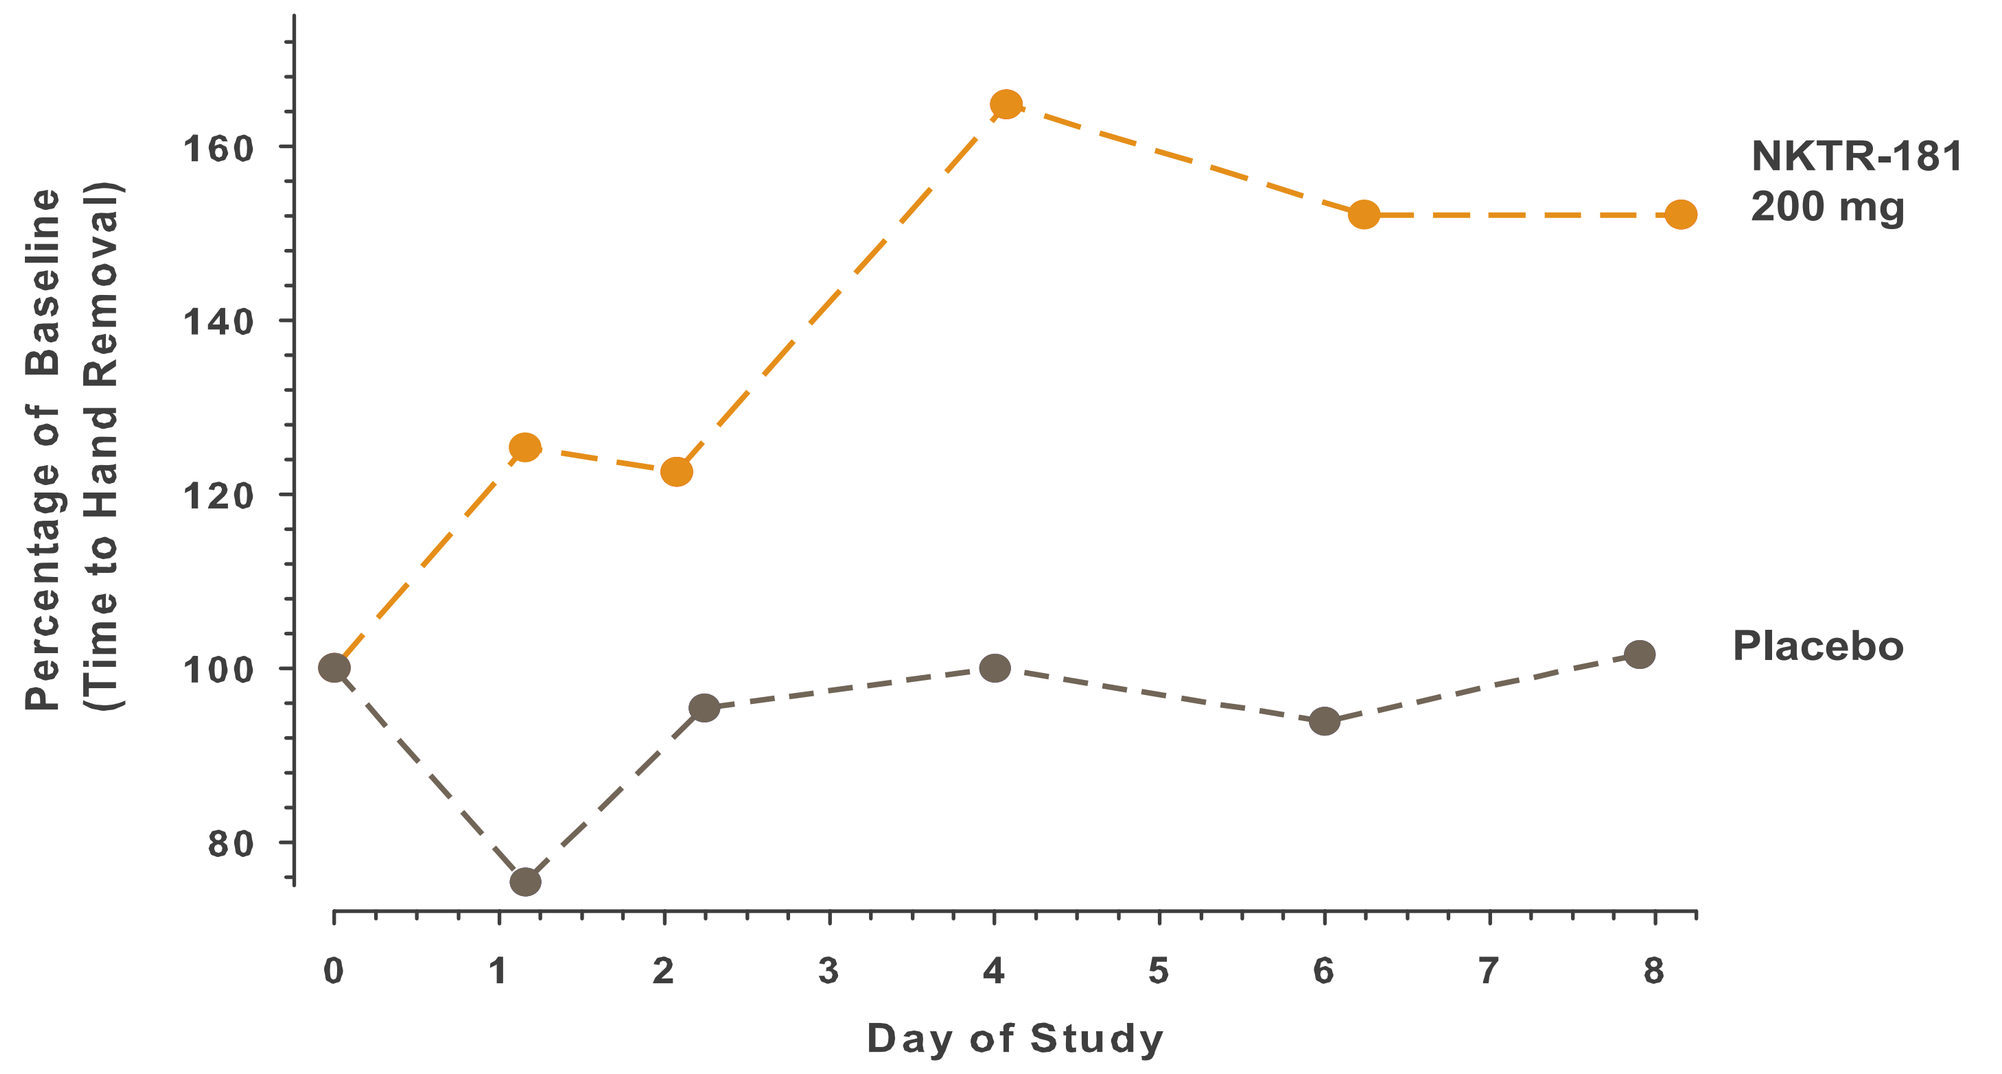
Percentage of Baseline (Time to Hand Removal)
Day of Study
NKTR-181 200 mg
Placebo

Increase in Pain Tolerance in Cold Pressor Test of Centrally-Mediated Pain

Results From NKTR-181 Human Abuse Liability (HAL) Study

Study Design

Single doses of NKTR 181 (100 mg, 200 mg and 400 mg administered as an oral solution) were compared with a 40 mg dose of oxycodone and with placebo. The study consisted of a Screening Stage (Visit 1); a Qualifying Stage comprised of a Naloxone Challenge Test and a Drug Discrimination Test (Visit 2); a randomized Treatment Stage (Visit 2); and a Follow-up Telephone Contact. The Treatment Stage consisted of 5 dosing days, each of which consisted of a single treatment of one of the study medications and/or placebo followed by a 72hr washout. Subjects received a single dose of each of the 5 study treatments in random order using a Williams Square design; study procedures were the same for each treatment arm.

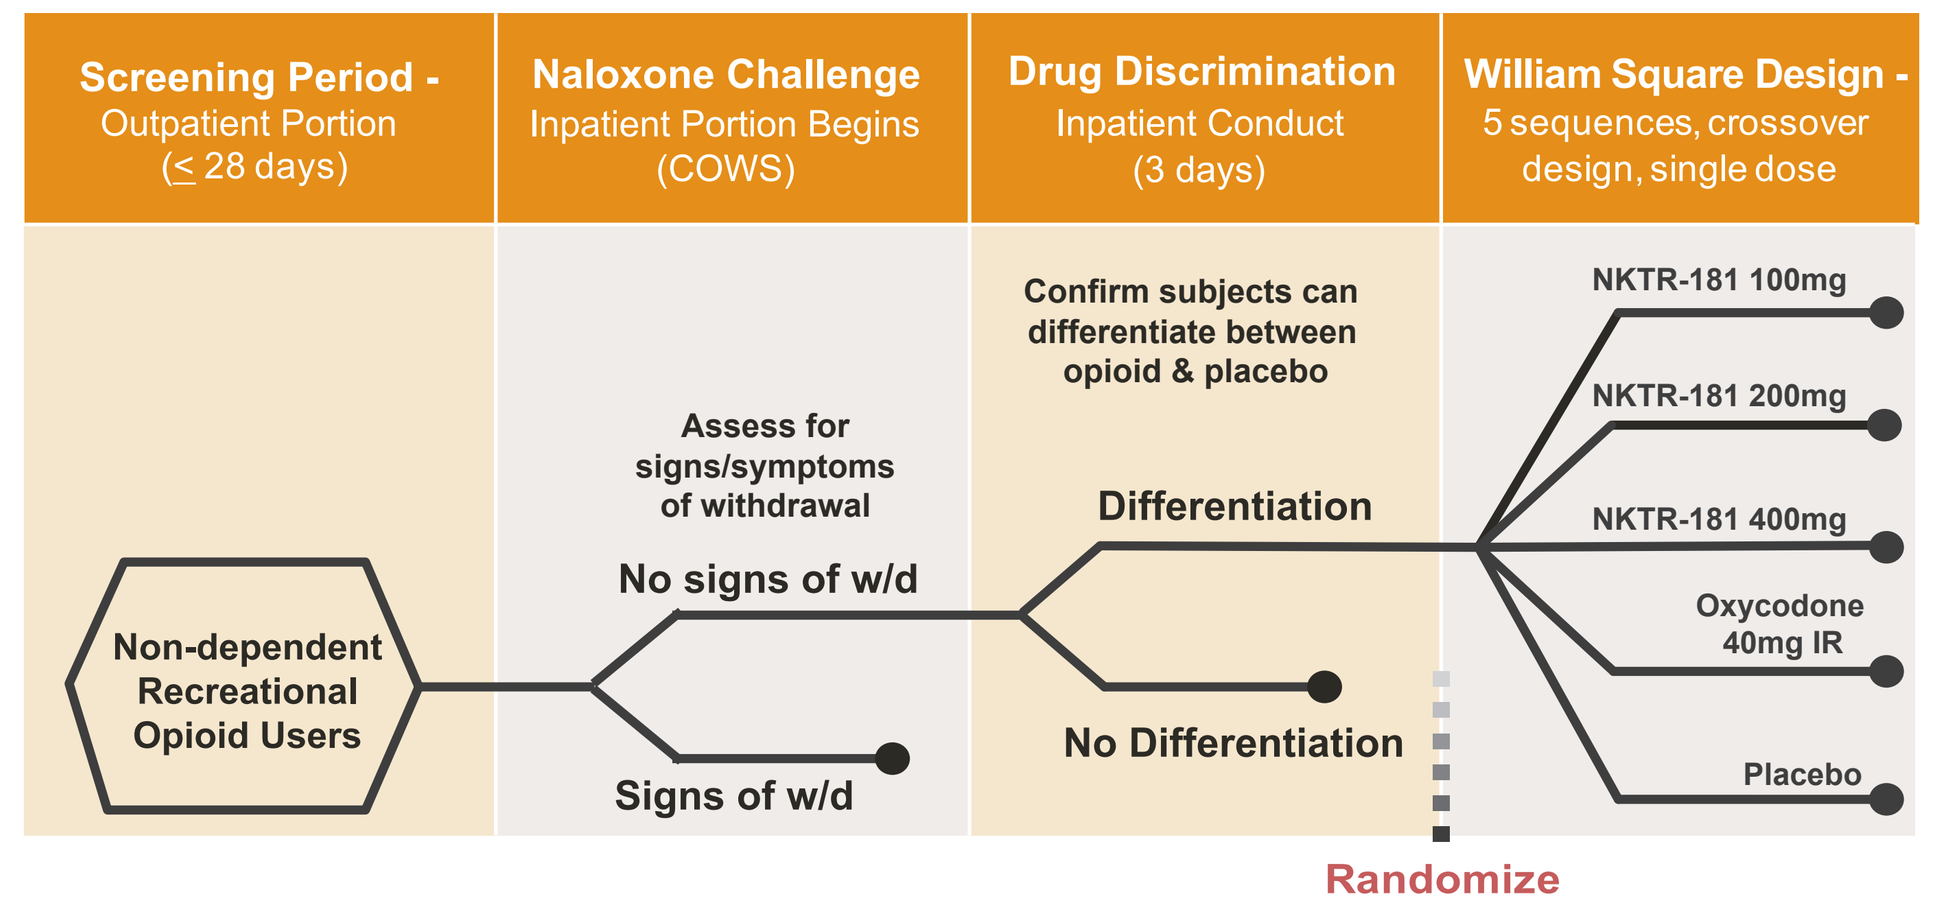
Screening Period - Outpatient Portion (≤ 28 days)
Naloxone Challenge Inpatient Portion Begins (COWS)
Drug Discrimination Inpatient Conduct (3 days)
William Square Design - 5 sequences, crossover design, single dose
Non-dependent Recreational Opioid Users
Assess for signs/symptoms of withdrawal
No signs of w/d
Signs of w/d
Confirm subjects can differentiate between opioid & placebo
Differentiation
No Differentiation
NKTR-181 100mg
NKTR-181 200mg
NKTR-181 400mg
Oxycodone 40mg IR
Placebo
Randomize

NKTR-181 Rated Similar to Placebo for "Drug Liking" and "High" VAS scores at All Doses

- The abuse potential of NKTR-181 was tested for three doses (100 mg, 200 mg, 400 mg administered as an oral solution) which were doses that exhibited analgesic response in Phase 1.
- Oxycodone performed as expected based upon published data: drug "liking" and "high" scores were significantly elevated compared to placebo [and NKTR-181] starting at 15 minutes, and remained significantly elevated compared to placebo [and NKTR-181] through 4 hours after dosing, at which time the oxycodone effects were decreasing.
- At the 100 mg and 200 mg doses, NKTR-181 demonstrated no difference from placebo in liking and high scores at any time point. At the 400 mg dose (the highest dose studied in the Phase 2 efficacy study), NKTR-181 produced a small increase 90-120 minutes post dose but remained significantly less p<0.0001 than oxycodone through 4 hours.

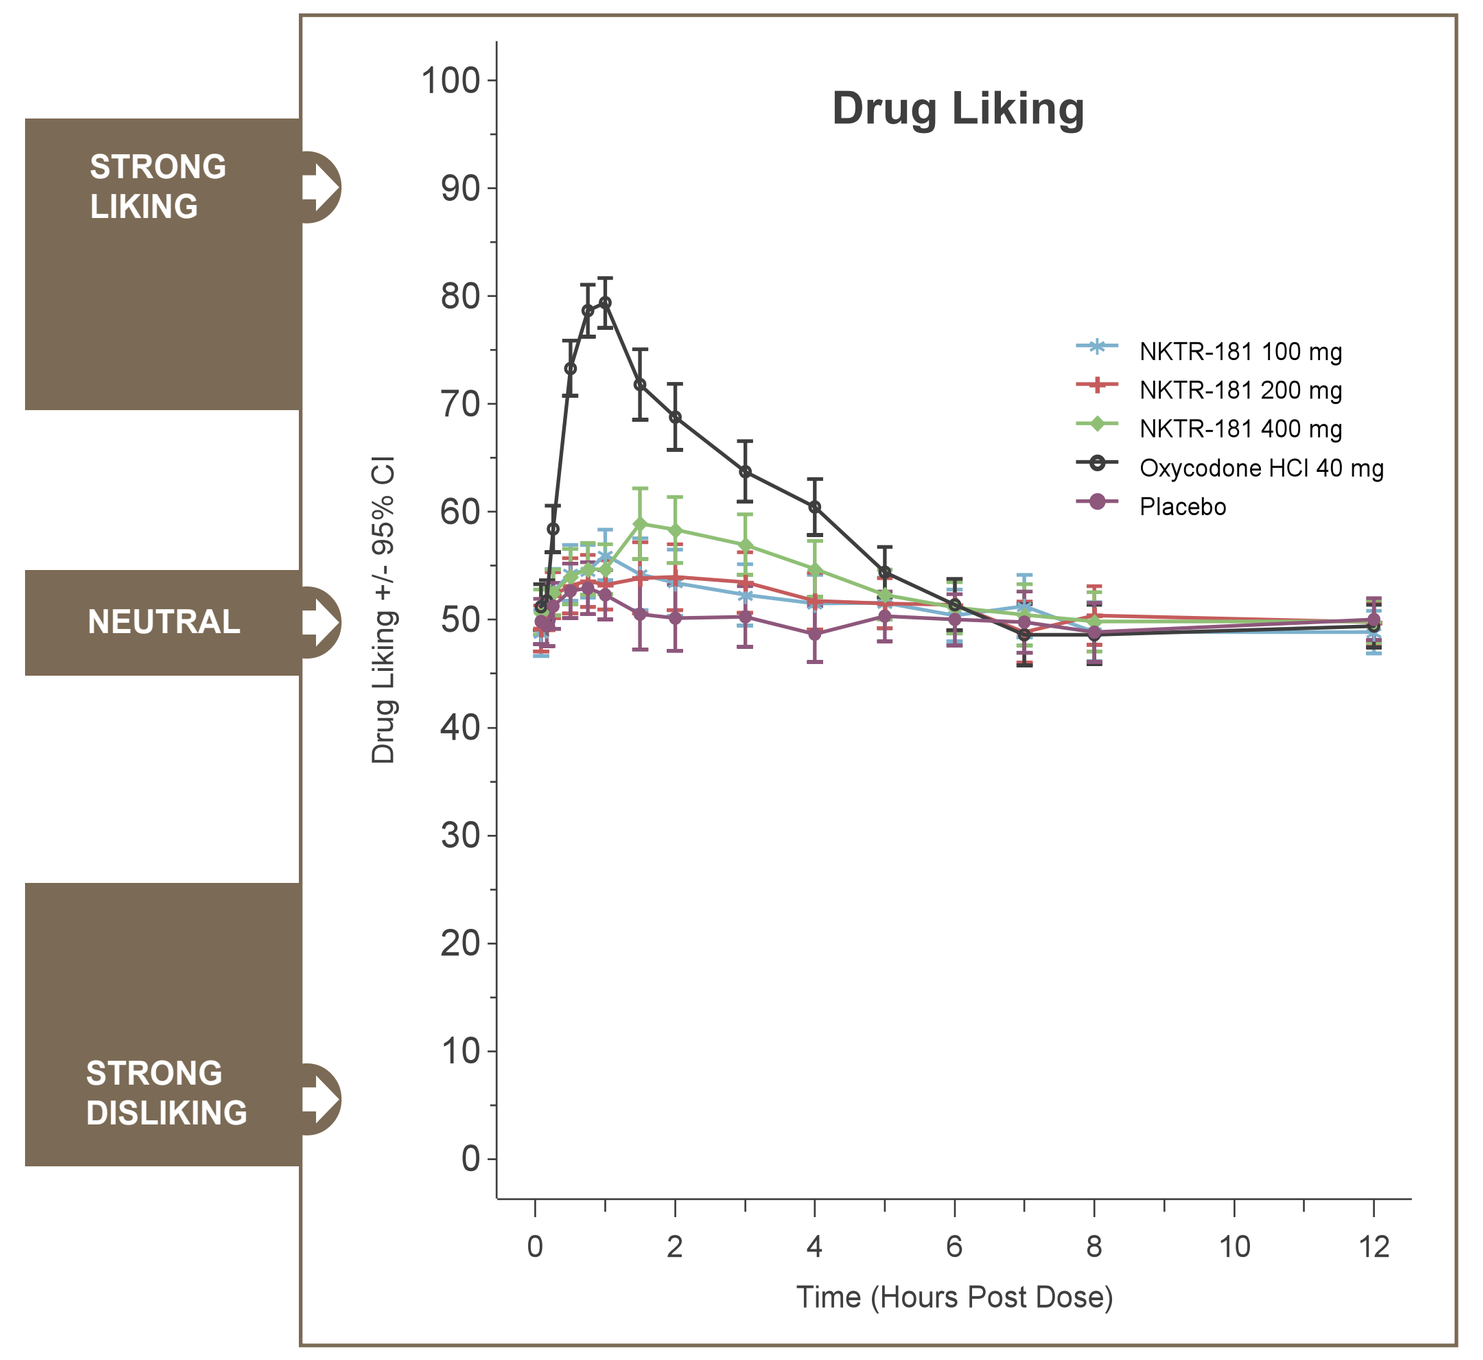
STRONG LIKING
NEUTRAL
STRONG DISLIKING
Drug Liking
Drug Liking +/- 95% CI
NKTR-181 100 mg
NKTR-181 200 mg
NKTR-181 400 mg
Oxycodone HCl 40 mg
Placebo
Time (Hours Post Dose)

NKTR-181 Primary Drug Liking Profile
On a scale of 0 to 100 with 50 being you don't feel positively or negatively about the drug, how would you rate the drug?

Drug Liking

TIME AFTER DOSING / NKTR-181 DOSE	15m	30m	45m	60m	90m	2h	3h	4h	Overall (0-24h)
100 mg	0.0002	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001
200 mg	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001
400 mg	0.0002	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	0.0009	<0.0001	<0.0001

Drug High

TIME AFTER DOSING / NKTR-181 DOSE	15m	30m	45m	60m	90m	2h	3h	4h	Overall (0-24h)
100 mg	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001
200 mg	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001
400 mg	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001

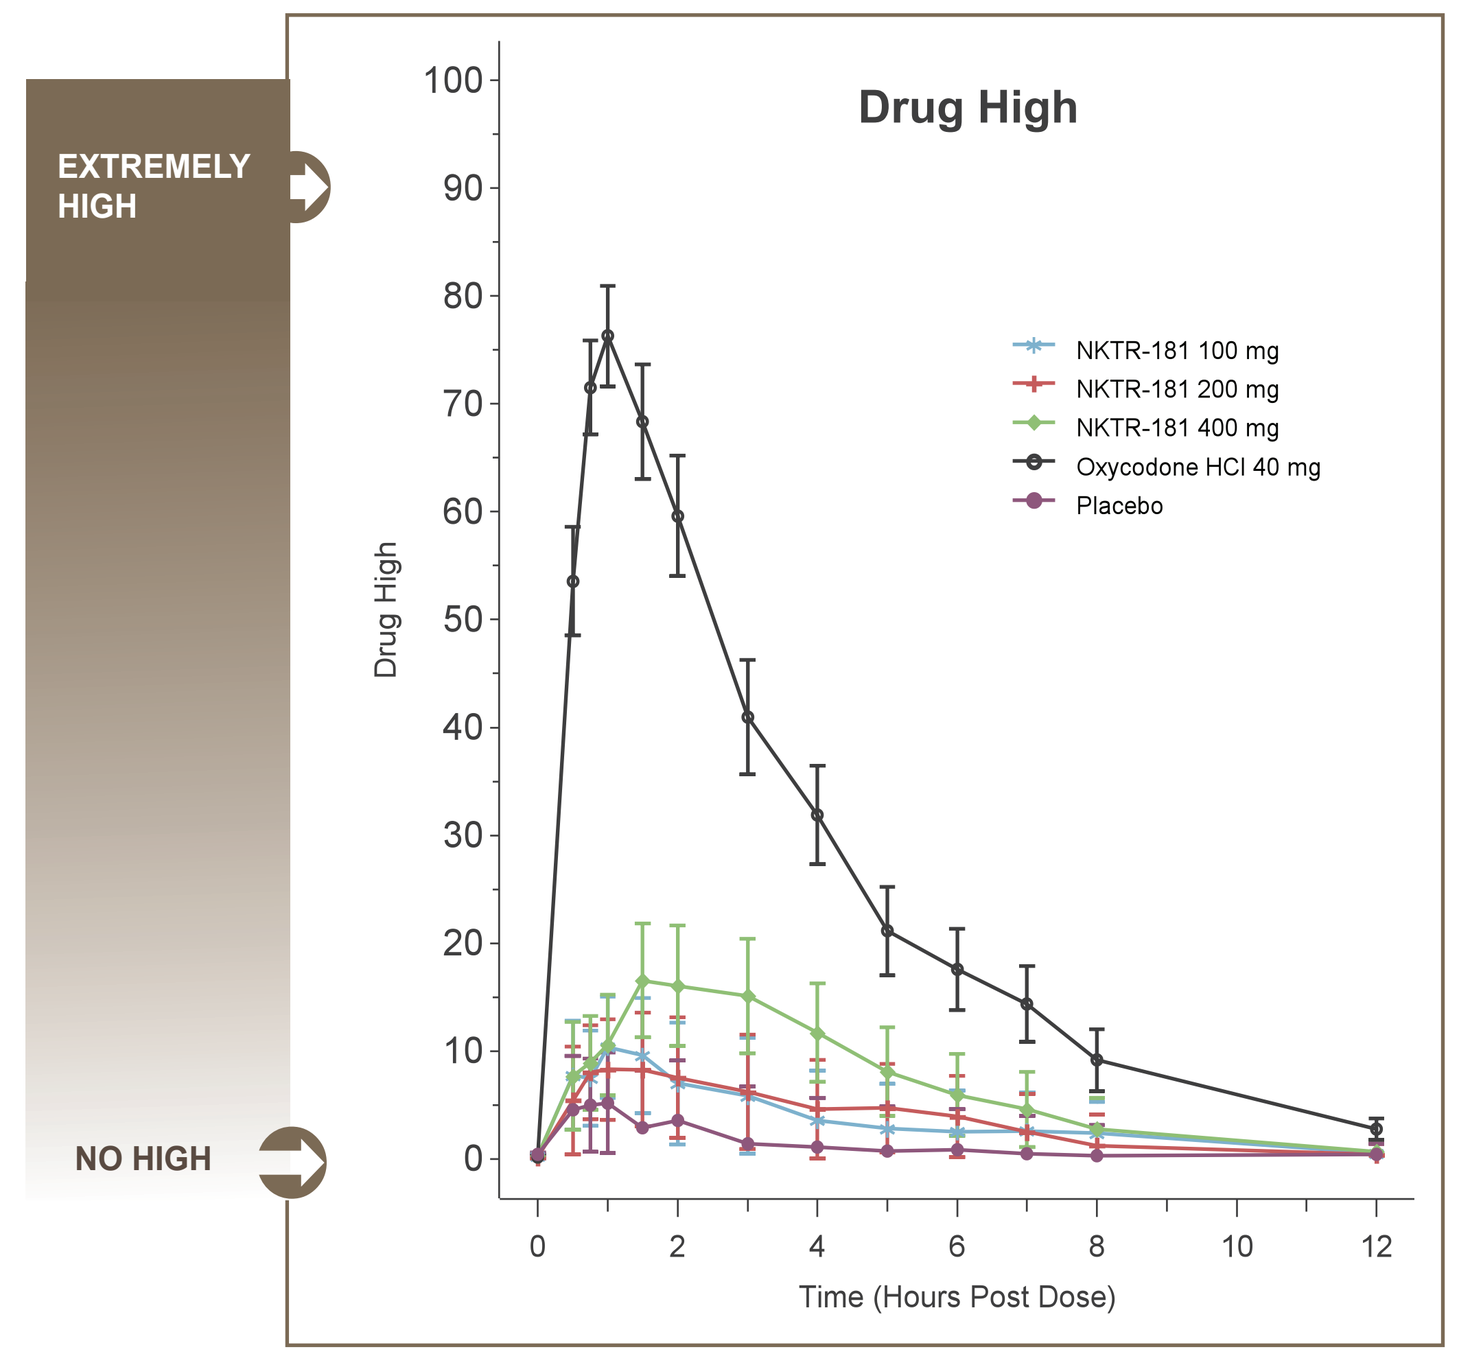
EXTREMELY HIGH
NO HIGH
Drug High
Drug High
NKTR-181 100 mg
NKTR-181 200 mg
NKTR-181 400 mg
Oxycodone HCl 40 mg
Placebo
Time (Hours Post Dose)

NKTR-181 Primary Drug Liking Profile
On a scale of 0 to 100, how high do you feel right now?

In Vitro Manipulation and Extraction Studies

- NKTR-181 is a unique new chemical entity (NCE) specifically designed to reduce the rate of CNS entry: the clinical profile, including liking scores similar to placebo, are intrinsic to the molecule and not dependent upon a formulation.
- Traditional tamper resistant strategies are commonly subject to conversion (e.g. extraction) into immediate-release forms of the current opioids. NKTR-181 is not subject to physical manipulation and was administered as an oral solution in this HAL study.
- NKTR-181 has been subjected to a wide variety of chemical (conventional and household agents) and temperature conditions in an attempt to convert the molecule to an abusable opioid. The results suggest that NKTR-181 is not readily manipulated into a more abusable substance.

Nektar NKTR-181: On the Path to Less Restrictive Scheduling and Tiered Labeling?

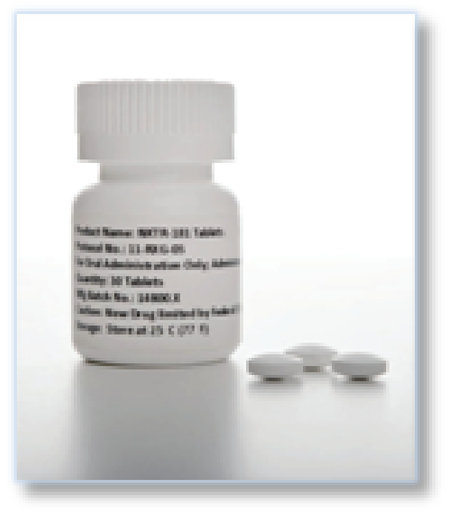

NKTR-181 Received Fast Track Status from FDA in 2012

NKTR-181: New Mu Opioid Analgesic Molecule for Chronic Pain Intended to Deter Abuse and Reduce CNS Side Effects by Reducing the Rate and Extent of Entry into the CNS

- NKTR-181 is a novel mu opioid molecule designed to treat chronic pain but have lower abuse potential than prototypic Schedule II opioids.
- As a mu-agonist opioid molecule, NKTR-181 is by default placed in Schedule II of the CSA during development.
- If the NKTR-181 profile observed in the clinic is validated by the ongoing clinical development program, less restrictive scheduling and/or other differentiating labeling may be warranted.
- The 2013 Guidance presents the possibility that the "abuse deterrent pharmacology" and slow CNS absorption of NKTR-181, independent of formulation, may merit one or more of the 4 Tiers of labeling differentiation.
- The regulatory and scientific path to less restrictive scheduling may be defined by NKTR-181 development.

Preliminary 8-Factor Analysis

Summary and Status of NKTR-181 as a Potential Candidate for Scheduling Less Restrictive than CII and for a Tiered Label

Factor	Key Findings
1. Known abuse potential (includes human and animal abuse potential assessment data)	Receptor binding studies suggest predominant activity of NKTR-181 at mu opioid receptors. NKTR-181 displayed very low abuse potential in preclinical studies in monkeys and in rats, based on results from standard self-administration and drug discrimination experiments. NKTR-181 rates similar to placebo on "drug liking" and "feeling high" scores at all doses studied and differentiates from oxycodone with high statistical significance on both measures (p<0.0001).
2. Pharmacology (overall pharmacologic profile)	NKTR-181's entry into the CNS is significantly slower than oxycodone based upon pupillometry (2.9 hours for NKTR-181 versus 11 minutes for oxycodone). NKTR-181's rate of crossing the blood brain barrier is substantially slower compared to prototypic CII opioids. These findings suggest that regardless of the speed of delivery to the systemic circulation (e.g., by swallowing, injecting or smoking) entry to the brain will be gradual, thus preventing euphoriant effects and deterring abuse.
3. Current state of science (including *in vitro* manipulation and extraction)	To date, there is no known chemical or physical method to alter NKTR-181 to generate an active opioid that would increase its rate of entry into the CNS.
4. History of abuse of substance & related substances	Opioids vary widely in abuse and other associated problems. NKTR-181 is not an approved drug and so there is no history of abuse, internet monitoring suggests interest from abusers is very low based upon slow CNS entry.
5. Scope and nature of problems	Findings to date suggest that NKTR-181 will be less of a target for abuse than prototypic CII drugs because it is designed to minimize euphoriant effects regardless of route of administration.
6. Public health implications (positive and negative)	The dosage form does not use an extended-release formulation. Therefore, unit doses will not be expected to carry a high risk of overdose if the patient or abuser crushes or chews the tablet before swallowing.
7. Psychic & physiological dependence	NKTR-181 has shown no evidence of physical tolerance or dependence in animal models. Clinical assessment is ongoing.
8. Chemistry (is it already scheduled or a prodrug of a scheduled drug?)	NKTR-181 is a new opioid analgesic molecule. It is not a pro-drug or a reformulation of a marketed opioid, but the first in a new class of small molecule mu- agonist opioids engineered to have slower entry into the CNS.

Outlook Concerning Scheduling and Tiered Labeling

- Preclinical and clinical results to date suggest that a scheduling recommendation less restrictive than CII is a plausible outcome for NKTR-181.
- Tiered labeling appears highly likely if the findings to date are sufficiently replicated and extended.
- The recent Draft FDA Guidance for Industry (Abuse-Deterrent Opioids Evaluation and Labeling) recognizes the need for safer opioids designed to deter abuse through their chemical structure. Current experience in the field has been with re-formulations of the drugs that are highly abused, hence the draft guidance does not provide specific consideration of new molecules with reduced abuse potential specifically engineered into the chemical structure. Since physical manipulation is not relevant in this setting, a new molecule with reduced abuse potential that is not subject to degradation or transformation of the chemical bonds to yield an abusable opioid may warrant an additional tier in the draft FDA guidance.

Abstract #683 / Poster Board #21: Presented at the 2013 College on Problems of Drug Dependence (CPDD) Annual Meeting, San Diego, CA