Etirinotecan Pegol (NKTR-102) in Patients With Platinum-Resistant Ovarian Cancer: Modeling CA-125 Response and its Correlation with Tumor Response

Yen Lin Chia, Ute Hoch, Abraham C. F. Leung, Michael A. Eldon, Nektar Therapeutics, San Francisco, CA

Introduction

- Etirinotecan pegol (NKTR-102) is a unique topoisomerase I inhibitor designed for prolonged tumor cell exposure.
- After q21d dosing of etirinotecan pegol to patients with cancer, plasma SN38 exposure is increased and greatly prolonged compared to irinotecan (elimination half-life 50 days compared to 2 days), yet maximal concentrations are 5- to 10-times less.

Figure 1. Model-Predicted Plasma SN38 Concentration-Time Profiles After Dosing with 145 mg/m² Etirinotecan Pegol (orange) and 350 mg/m² Irinotecan (blue) q21d.

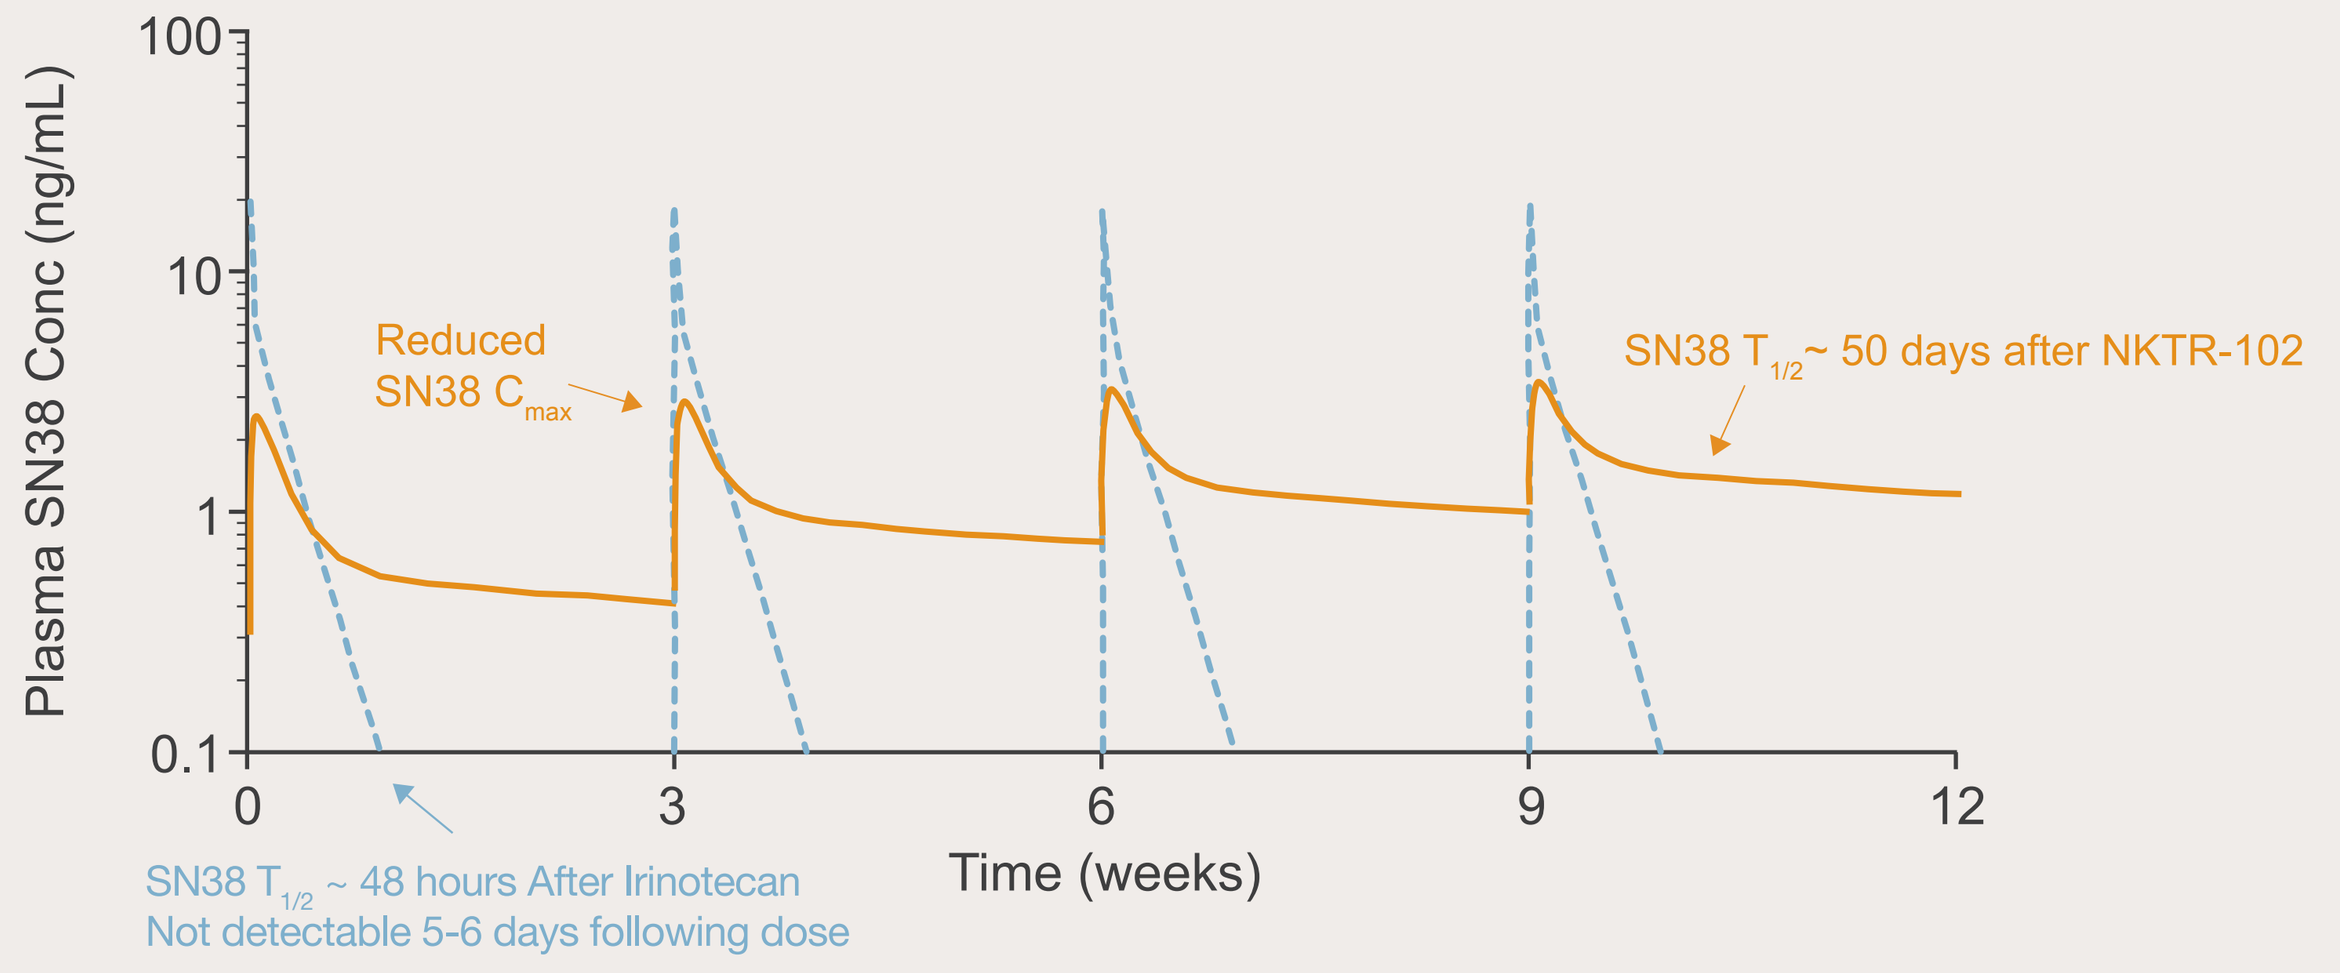
Plasma SN38 Conc (ng/mL)
100
10
1
0.1
Reduced SN38 C_{max}
SN38 $T_{1/2}$~ 50 days after NKTR-102
0
3
6
9
12
Time (weeks)
SN38 $T_{1/2}$ ~ 48 hours After Irinotecan
Not detectable 5-6 days following dose

Background

- In a phase 2 trial in heavily pretreated patients with platinum resistant/refractory ovarian cancer, etirinotecan pegol administered q14d or q21d demonstrated confirmed objective RECIST and GCIG response rates (ORR) of 20% and 33%, respectively.

Table 1. Efficacy Results: Objective Response Rate by RECIST, GCIG, CA-125 in MITT Population.

	145 mg/m² IV q14d N=35	145 mg/m² IV q21d N=31	Total N=66
RECIST			
N (evaluable)	35	31	66
Confirmed	7 (20%)	6 (19%)	13 (20%)
Duration of confirmed response (months)*	4.1 (2.8-8.0)	4.0 (2.6-12.6)	4.1 (2.8-8.0)
GCIG			
N (evaluable)	35	31	66
Confirmed	11 (31%)	11 (36%)	22 (33%)
CA-125			
N (evaluable)	30	25	55
Confirmed	12 (40%)	10 (40%)	22 (40%)

*Estimated using Kaplan-Meier method
The MITT Population includes platinum-resistant patients who have undergone radiographic tumor measurement evaluation at baseline with measurable tumors and received at least one dose of study drug.

- The strong CA-125 response observed in these heavily pretreated patients who responded to the drug (Figure 2) was encouraging and stimulated our focus on this established biomarker.[1]

Figure 2. % Change in Serum CA-125 Profiles after 145 mg/m² Etirinotecan Pegol q14d or q21d.

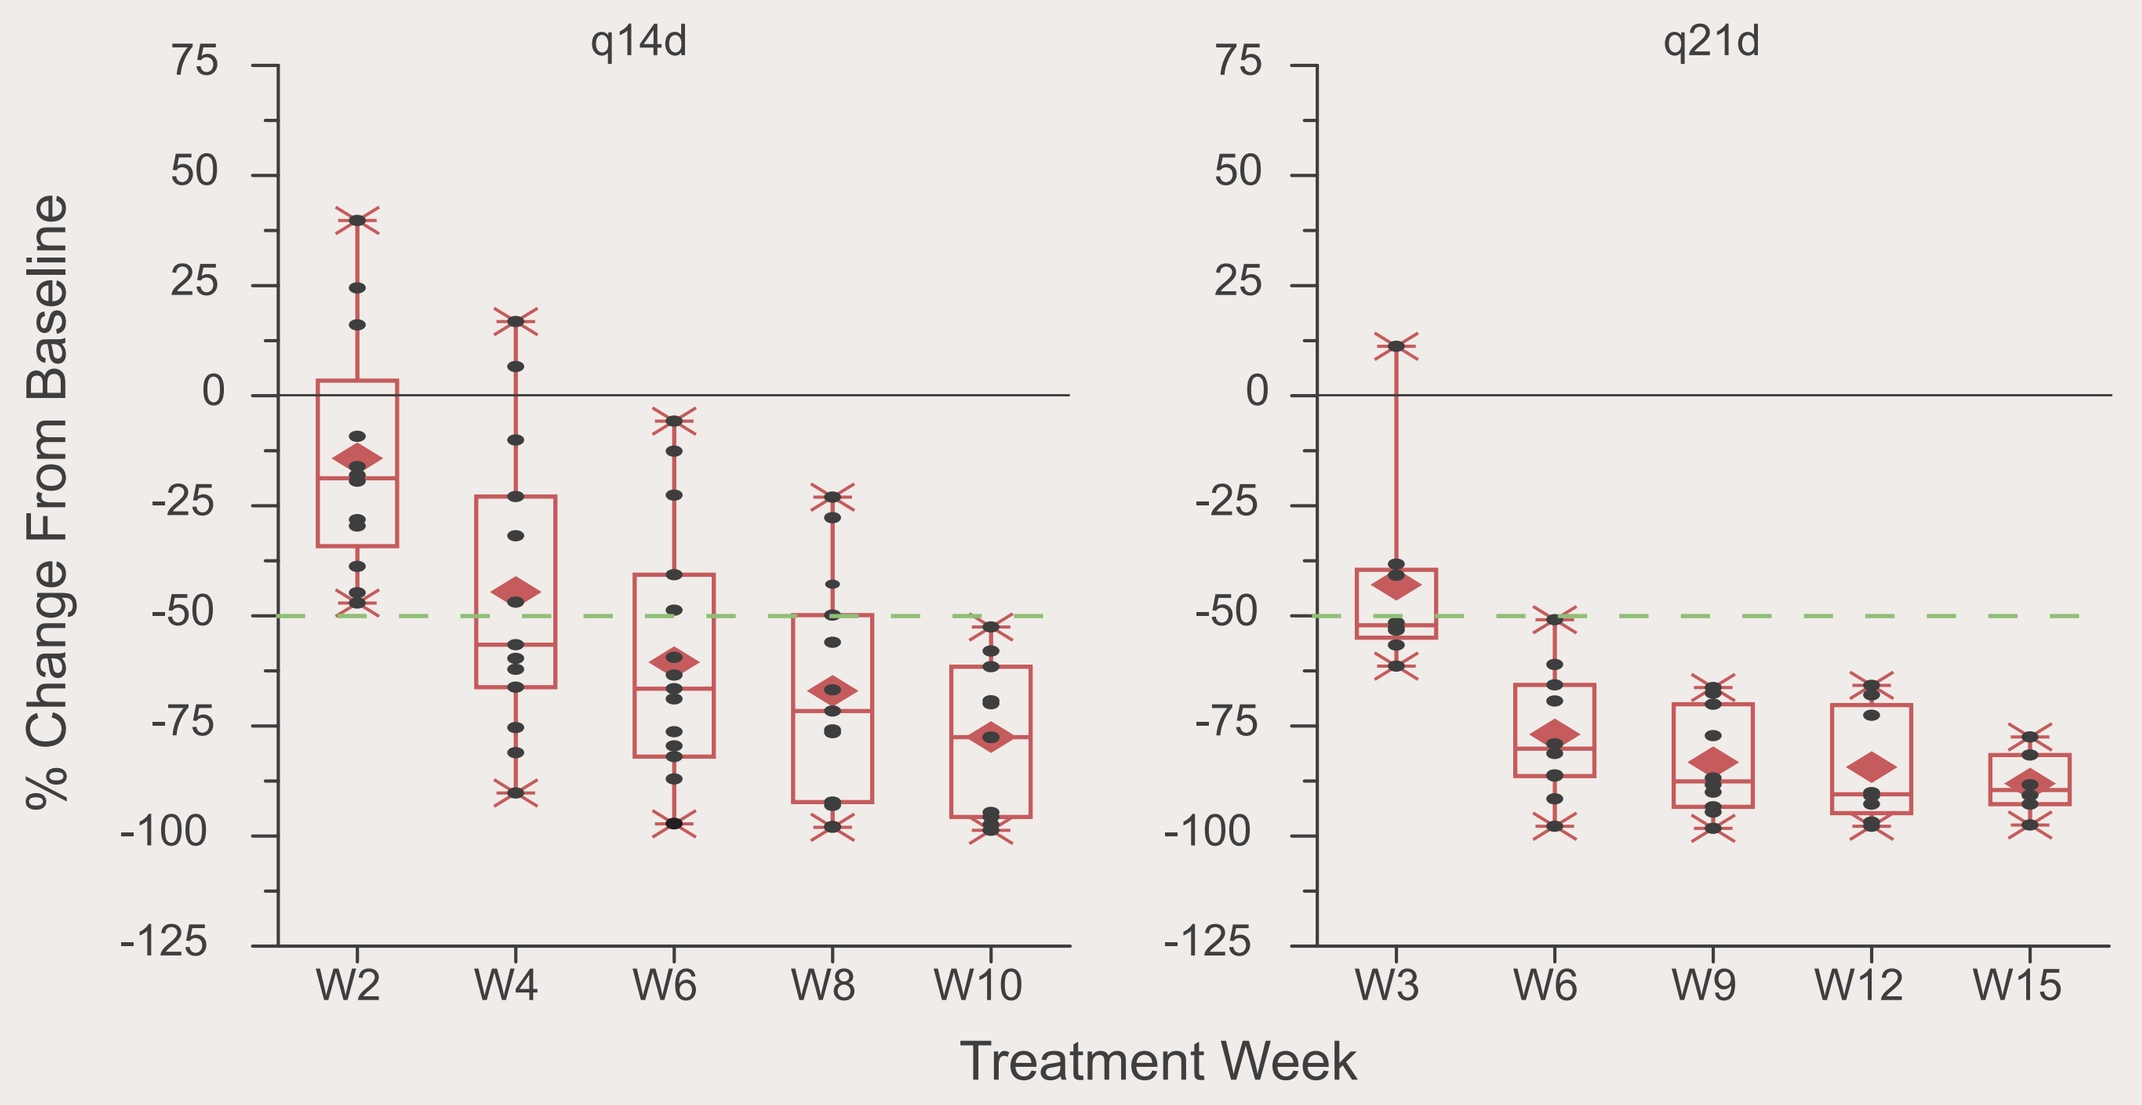
q14d
q21d
% Change From Baseline
75
50
25
0
-25
-50
-75
-100
-125
W2
W4
W6
W8
W10
W3
W6
W9
W12
W15
Treatment Week

- A recent study suggested that mathematical modeling may help describe CA-125 kinetic profiles that are predictive of relapse in patients with platinum-resistant ovarian cancer.[2]
- We developed a mathematical model linking the kinetics of CA-125 to SN38 exposure, tumor size, and RECIST and GCIG response criteria with the intent of predicting response in various clinical scenarios, such as changes in dose and/or schedule, or in the occurrence of dosing delays.

Data/Methods

- In the phase 2 study, patients with platinum-resistant ovarian cancer were randomized to etirinotecan pegol at a dose of 145 mg/m² q14d or q21d.
- Our analysis dataset consisted of the 55 patients whose baseline CA-125 was > 2 X upper normal limit and who had at least 1 post-baseline CA-125 measurement:

Table 2. Baseline CA-125 Measurements (U/mL).

	Min	25% quartile	Median	75% quartile	Max
q14d (N = 27)	118.5	244	522	1409	20805
q21d (N = 28)	71.3	250	507	1137	11421
Total	71.3	245.5	514.5	1341	20805

Model Building Steps

- Predict the plasma SN38 concentration-time profile for each patient using our previously developed population PK model.[3]
- Investigate the relationship of predicted plasma SN38 concentration with serum CA-125 concentration.

 Hypothesis: SN38 (indirectly) inhibits the production of CA-125.
- Evaluate the impact of dosing schedules on CA-125 response.
- Model:

$$\xrightarrow{k_{in}} \boxed{\text{CA-125}} \xrightarrow{k_{out}}$$

Exponential growth of CA-125 production rate

$$\frac{d[\mathrm{CA125}]}{dt} = k_{in} \cdot e^{\beta t} \cdot \underbrace{\left(1 - \frac{[C_{SN38}]}{IC50 + [C_{SN38}]}\right)}_{\text{Inhibitory effect}} - k_{out}[\mathrm{CA125}]$$

- k_{in}: production rate of serum CA-125
- k_{out}: elimination rate of serum CA-125
- [CA125]: Serum CA-125 (U/mL)
- [CSN38]: SN38 concentration (ng/mL)

Results

- CA-125 concentration-time profiles were well described by the model independent of response pattern.

Figure 3. Examples of Typical CA-125 Profiles: Observed (black) Versus Individual Predicted CA-125 (pink).

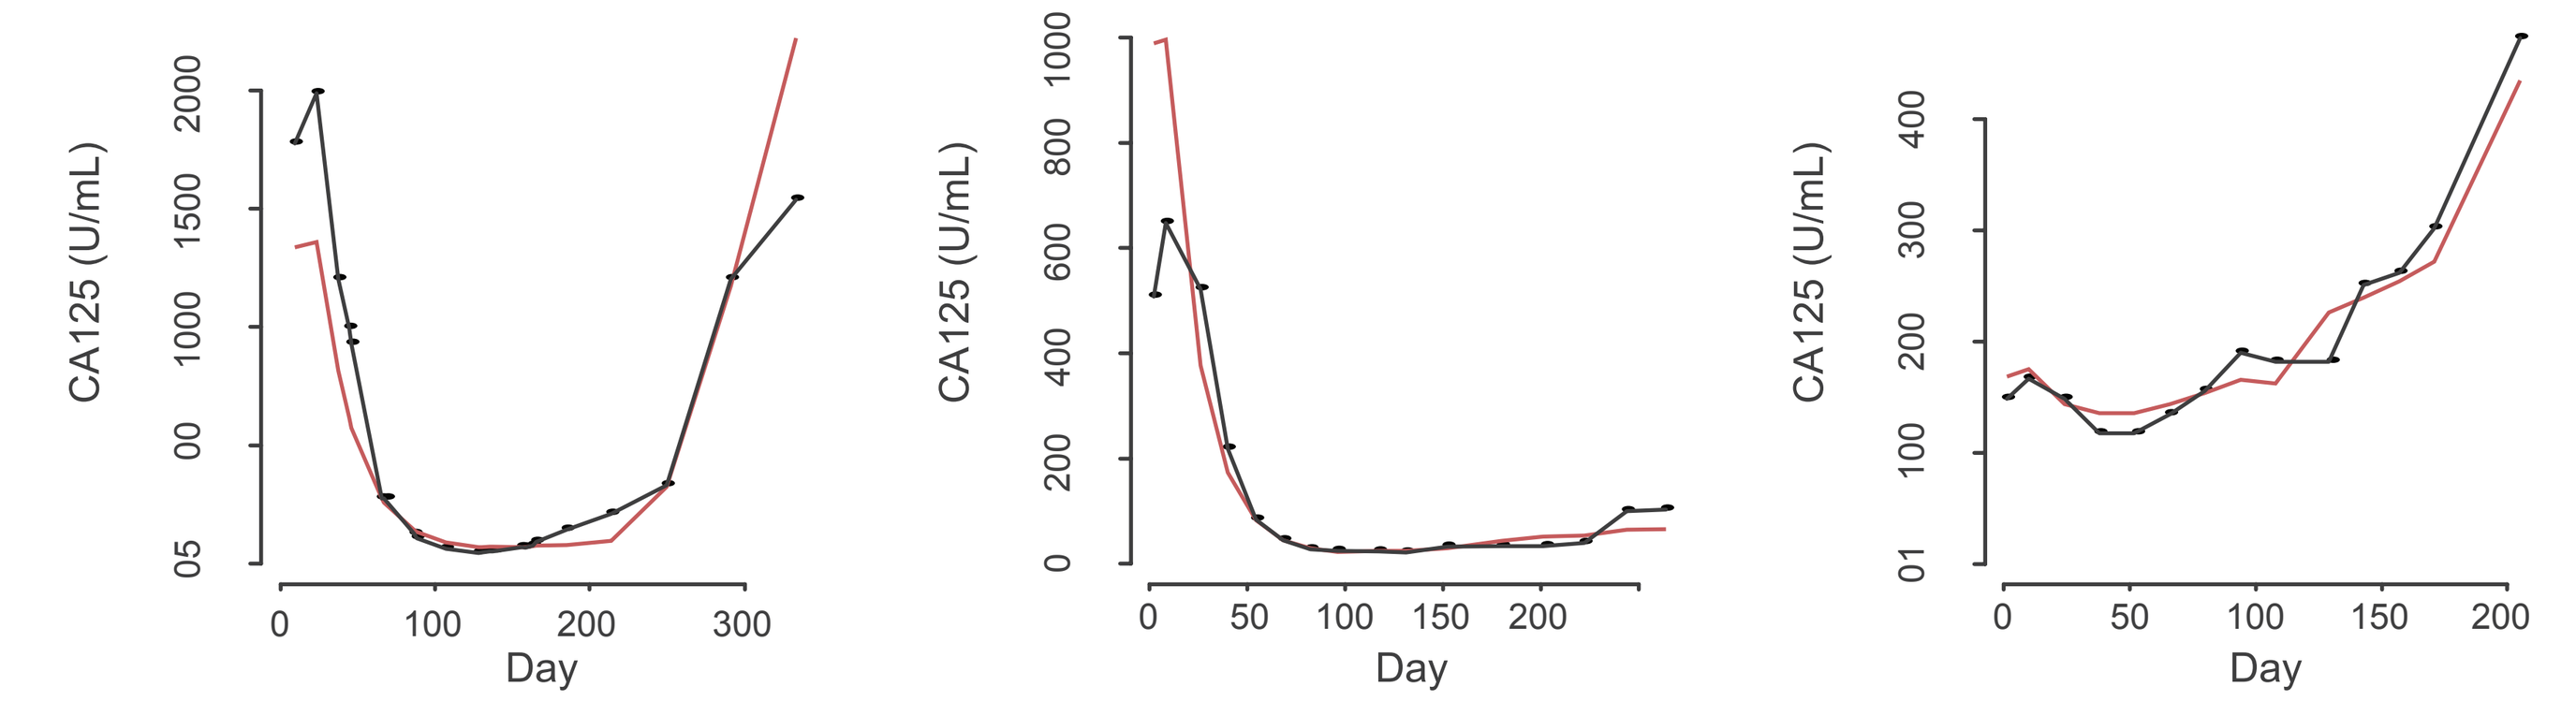
CA125 (U/mL)
Day

- The half-life of CA-125 decline was 6.4 days, consistent with previous reports.[4]

- The modeled population mean IC50 of SN38 was 1.1 ng/mL.
- The minimum plasma SN38 concentration (Cmin) at steady-state for the q14d regimen exceeded the IC50 in the absence of dose interruption as illustrated in Figure 4.

Figure 4. Simulated SN38 Profiles for 145 mg/m2 Etirinotecan Pegol q14d or q21d.

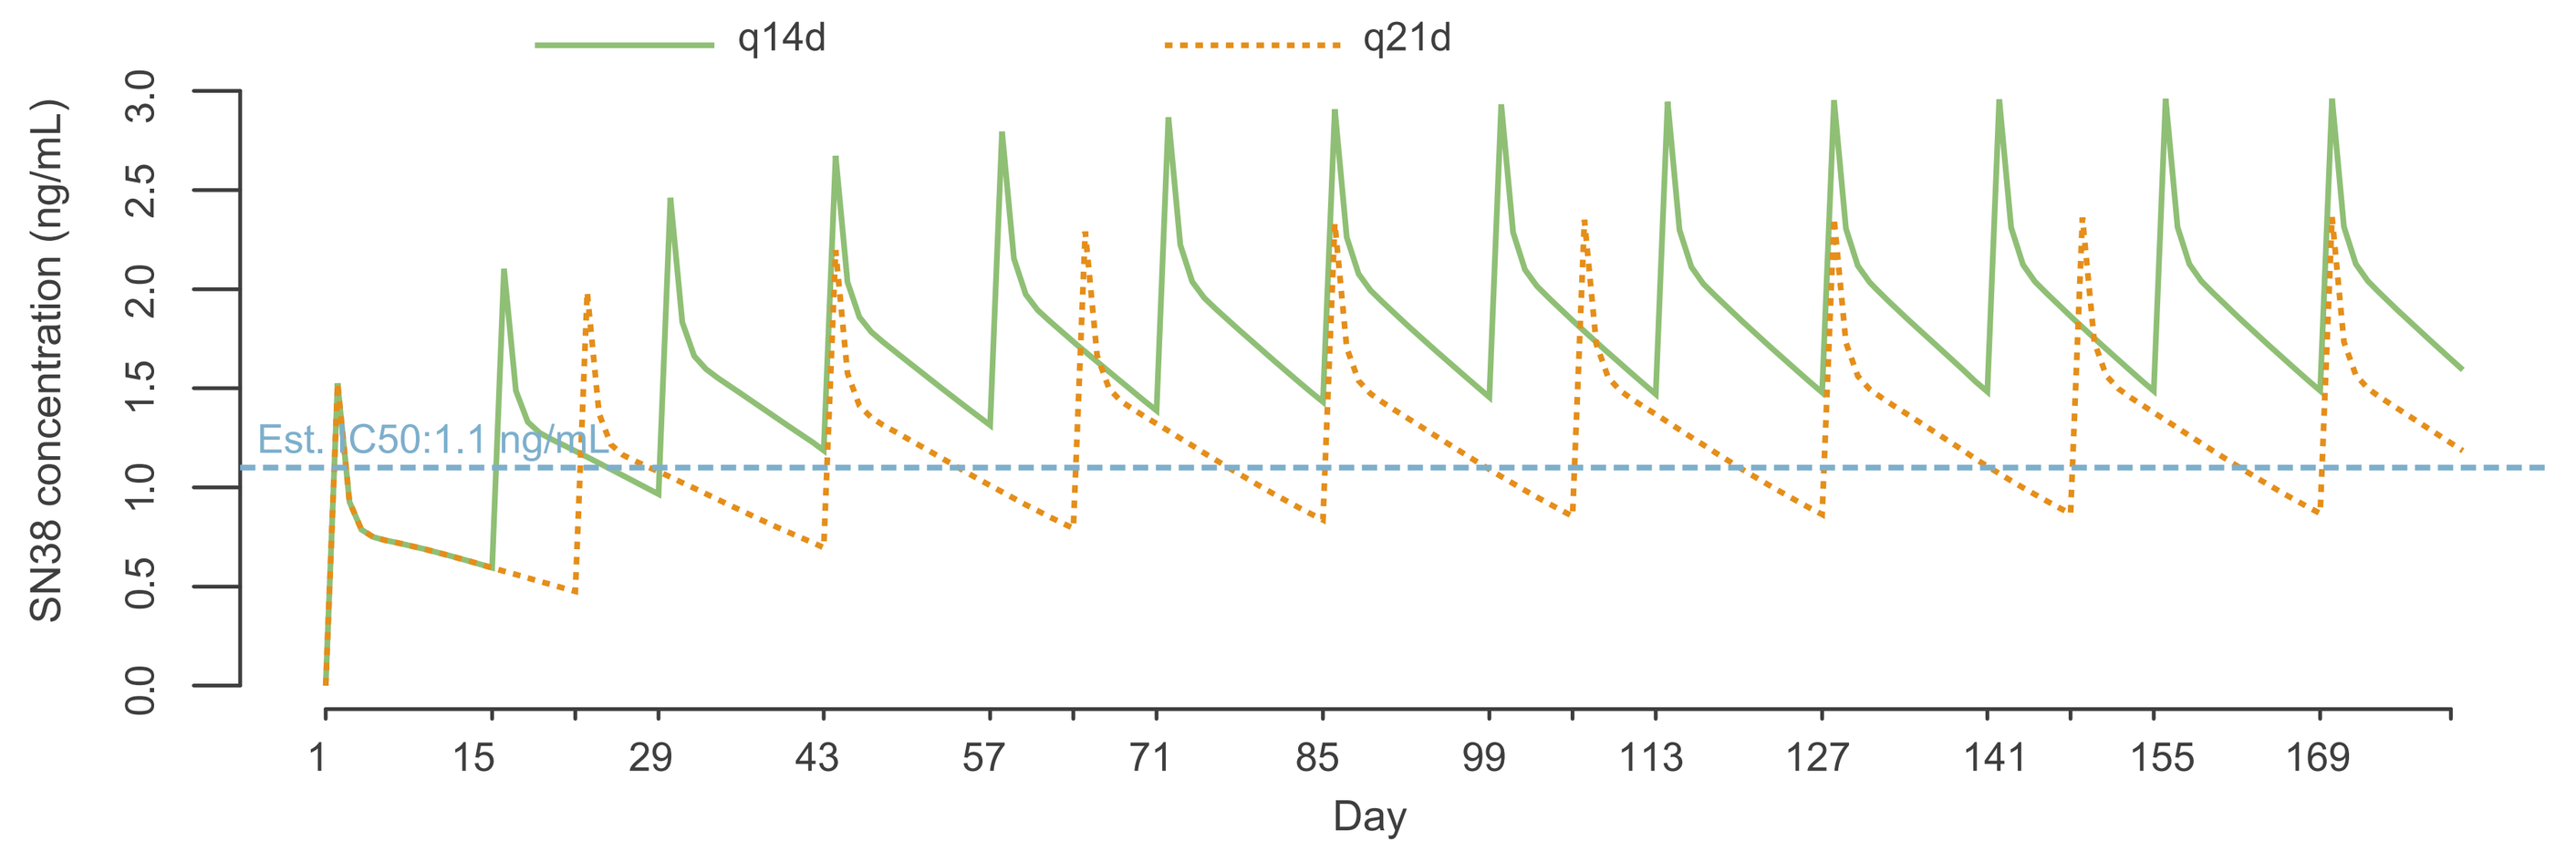
q14d
q21d
SN38 concentration (ng/mL)
Est. IC50:1.1 ng/mL
1
15
29
43
57
71
85
99
113
127
141
155
169
Day

- Model-predicted CA-125 profiles after q14d or q21d were very similar, with overlapping 95% prediction intervals. (Figure 5)
 - Predicted median CA-125 concentrations at steady state were 242 and 301 U/mL for the q14d and q21d schedules, respectively.

Figure 5. Simulated CA-125 Profiles for 145 mg/m2 Etirinotecan Pegol q14d or q21d.

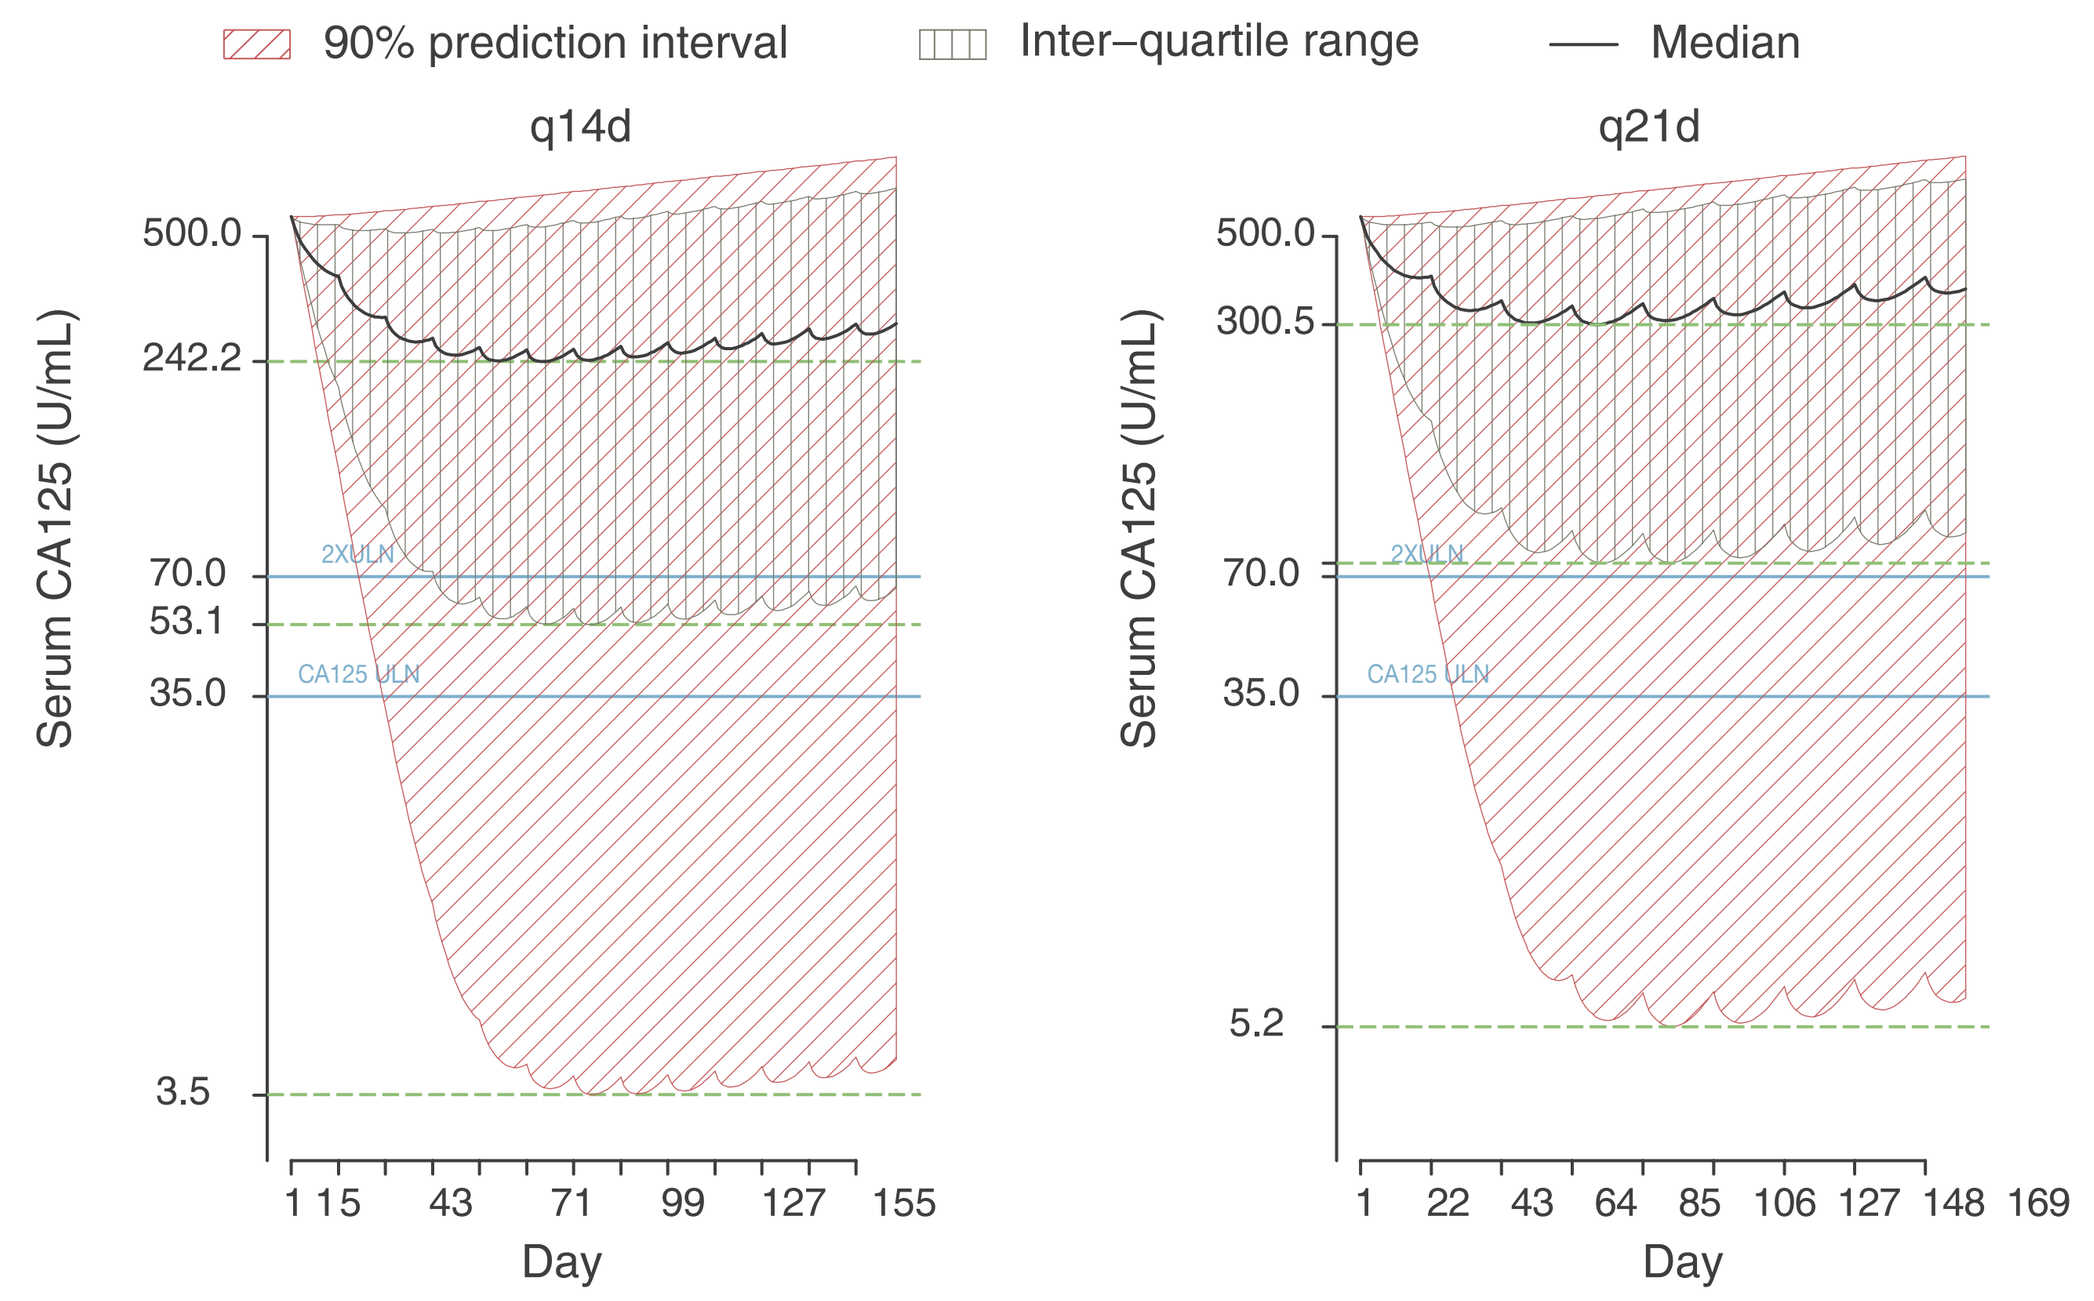
90% prediction interval
Inter–quartile range
Median
q14d
q21d
Serum CA125 (U/mL)
500.0
242.2
70.0
53.1
35.0
3.5
300.5
5.2
2XULN
CA125 ULN
Day

- To allow comparison of GCIG response between schedules, CA-125 profiles were simulated for 1000 pts receiving 145 mg/m² etirinotecan pegol q14d or q21d.
 - Projected GCIG response patterns for q14d and q21d over 180 days were very similar.
 - The % of patients achieving GCIG responses of CR, PR, SD, or PD was comparable between schedules, supporting use of the better-tolerated q21d regimen (Figure 6).

Figure 6. Projected GCIC Response for q14d and q21d.

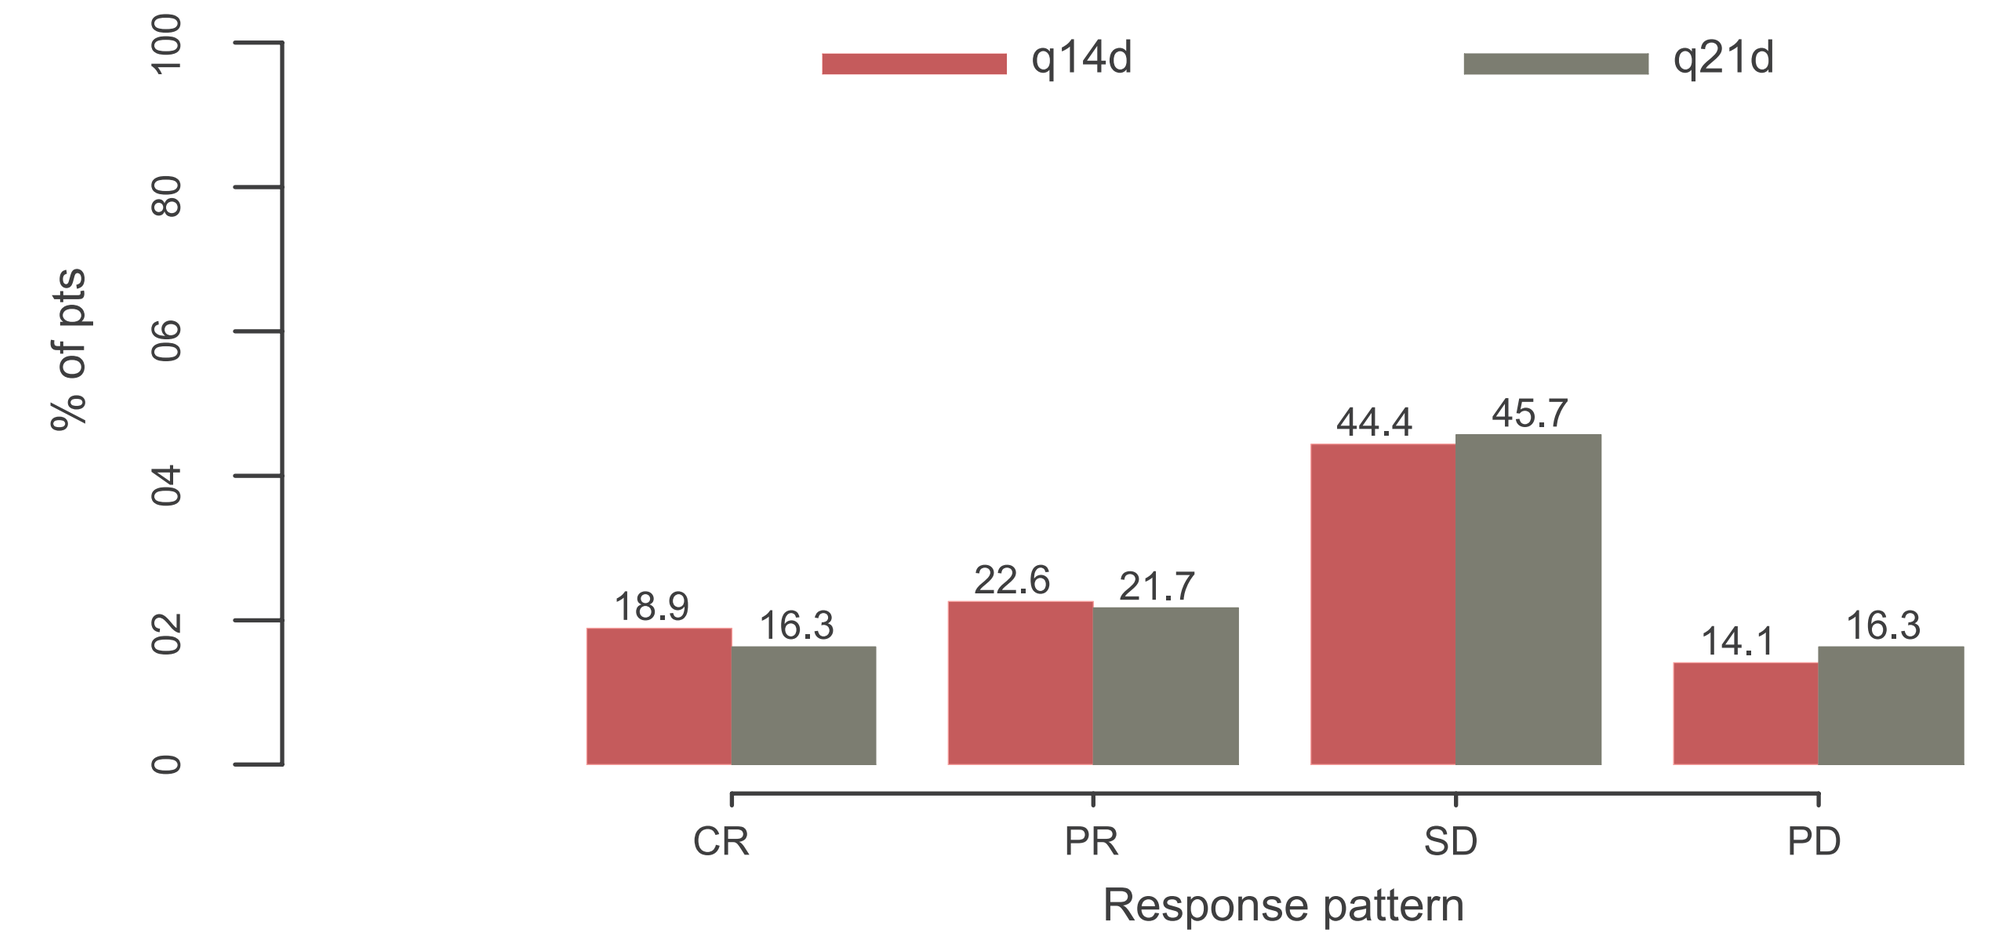
q14d
q21d
% of pts
18.9
16.3
22.6
21.7
44.4
45.7
14.1
16.3
CR
PR
SD
PD
Response pattern

CA-125 Response as Predictor of RECIST Response

Figure 7. Observed CA-125 Overall Best Response Rates showed Good Correlation with RECIST Responses (Pink: CA-125, Dark grey:Tumor size).

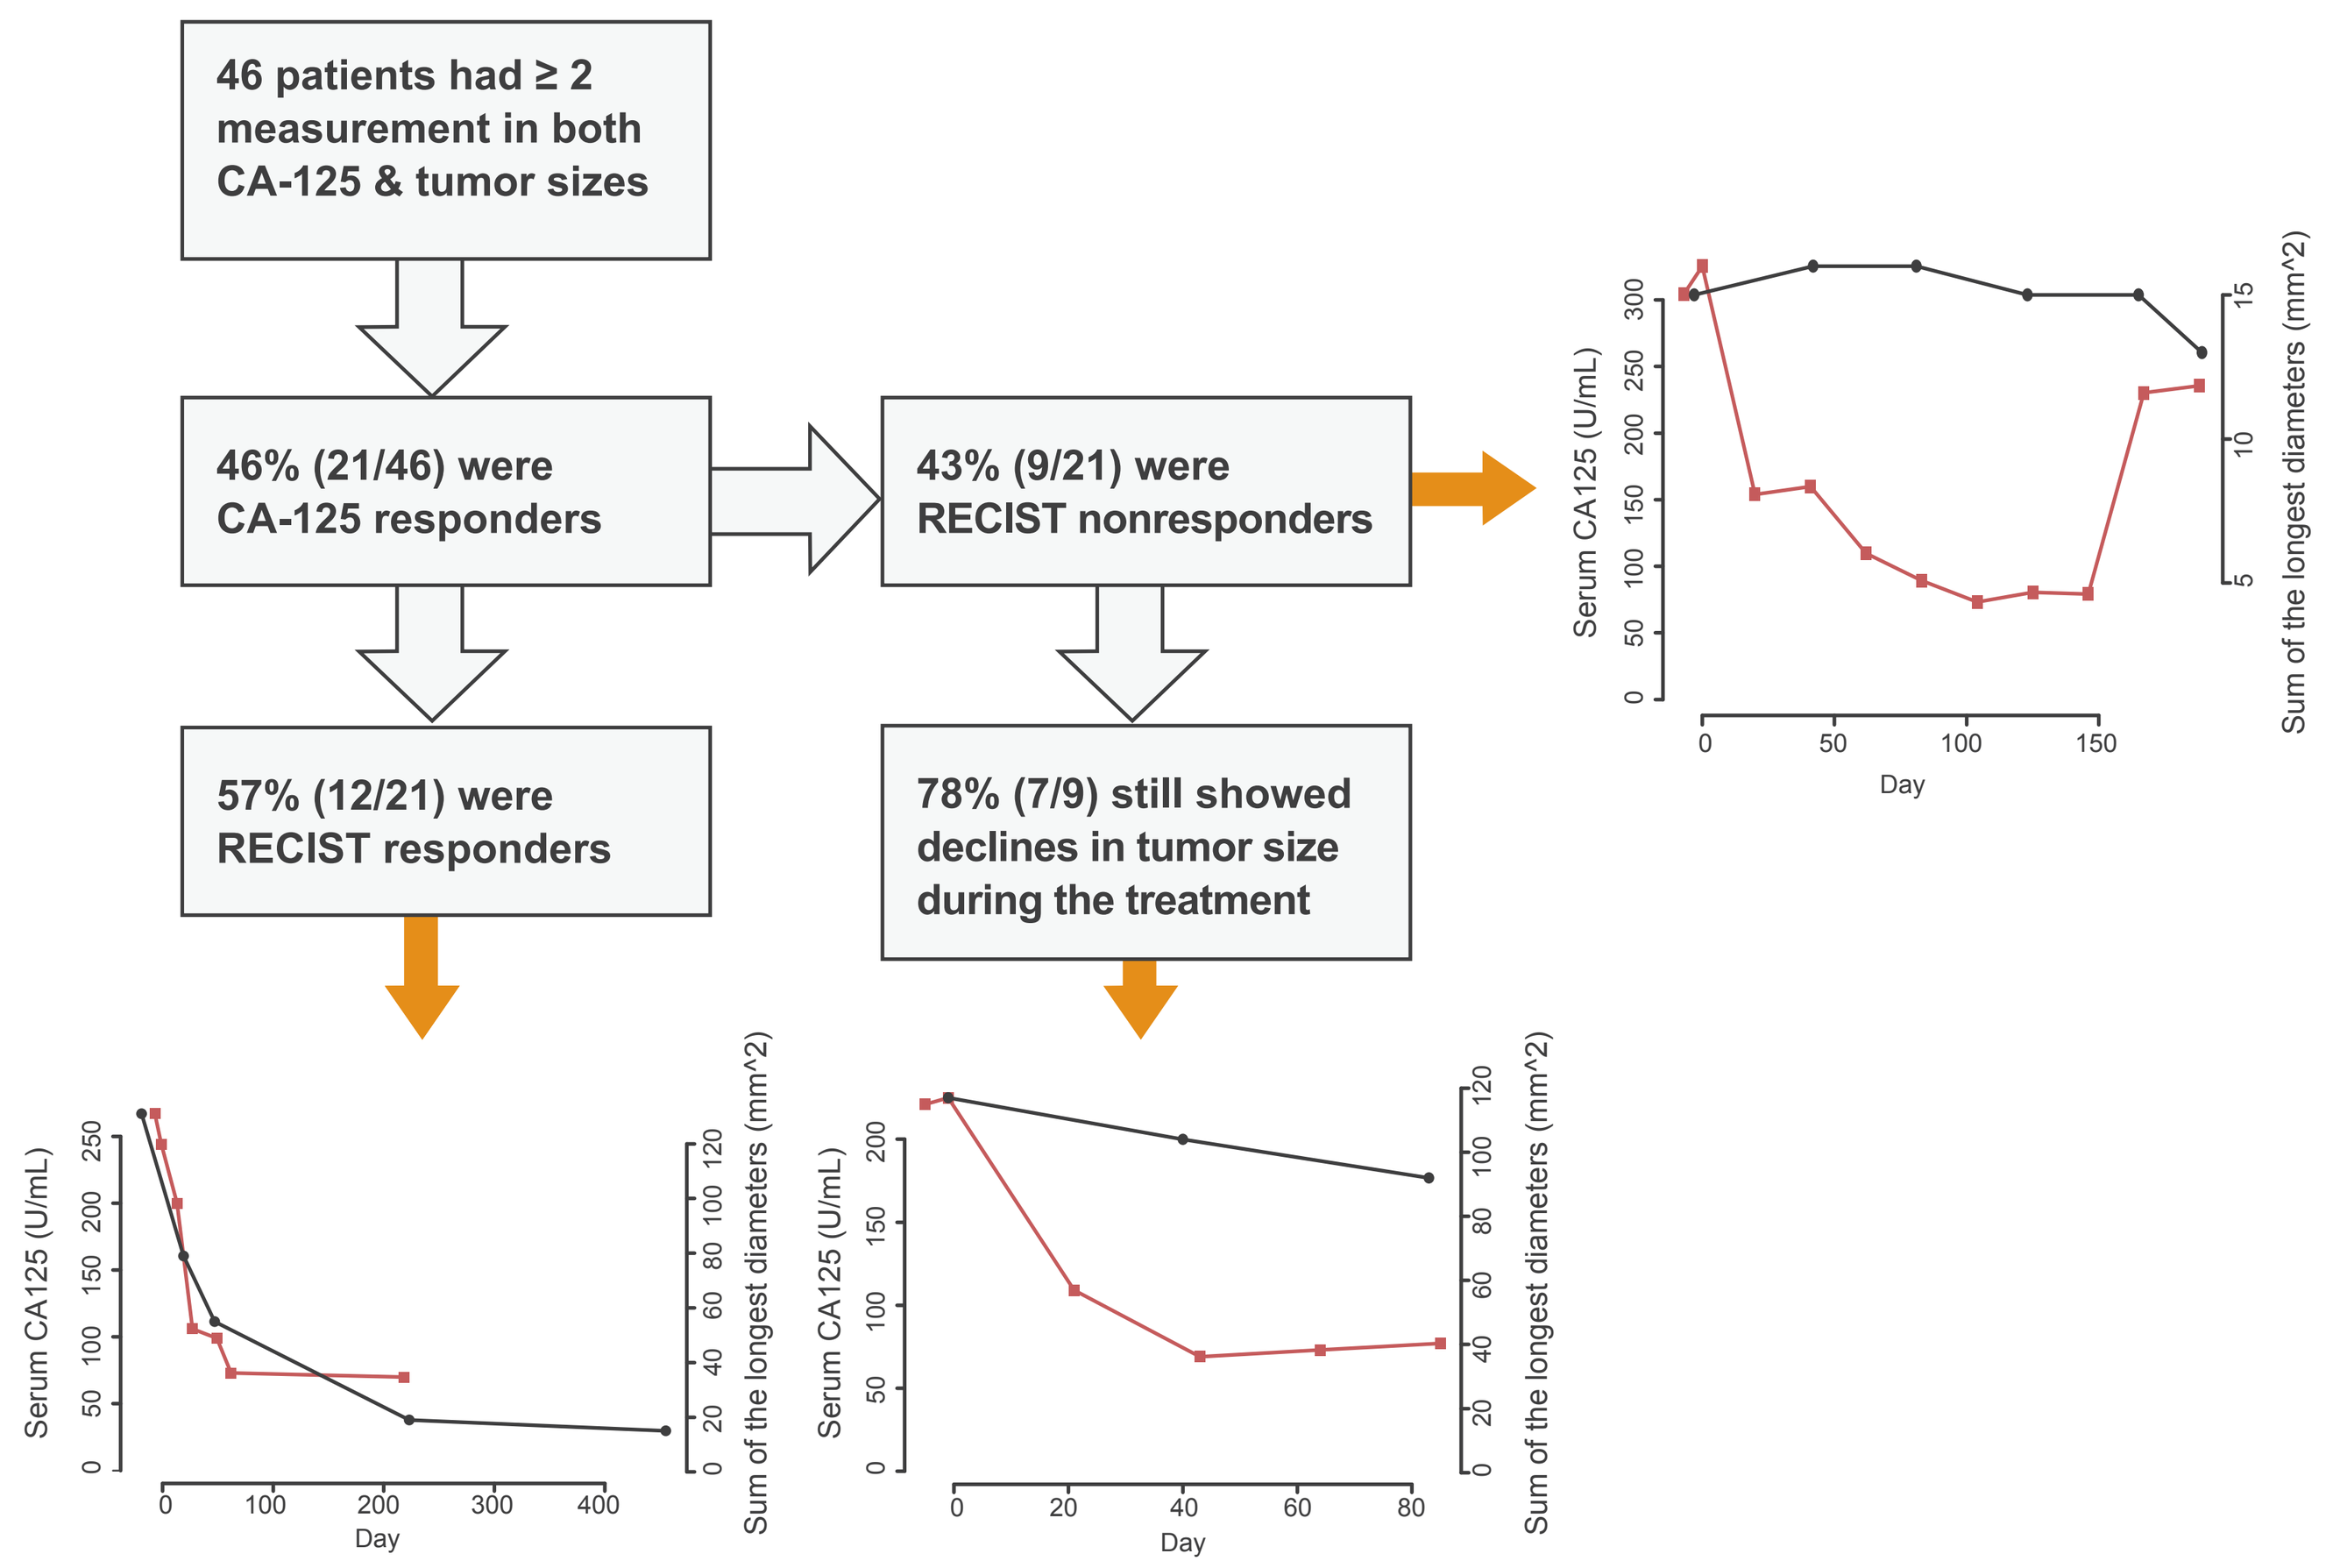
46 patients had ≥ 2 measurement in both CA-125 & tumor sizes
46% (21/46) were CA-125 responders
43% (9/21) were RECIST nonresponders
57% (12/21) were RECIST responders
78% (7/9) still showed declines in tumor size during the treatment
Serum CA125 (U/mL)
Sum of the longest diameters (mm^2)
Day

- Overall best response by RECIST and CA-125 correlated in 57% of platinum-resistant ovarian cancer patients.
- 43% of patients were classified as RECIST nonresponders.
 - Of these nonresponders, 78% still showed decline in tumor size and CA-125 during etirinotecan pegol treatment.
- Given that the simulated CA-125 response patterns for q14d and q21d were comparable, this strong correlation of CA-125 and RECIST response also supports use of the better-tolerated q21d regimen.

Conclusions

- The PK/PD model described CA-125 profiles accurately, providing a tool to predict drug response from SN38 PK data.
- The close correlation of CA-125 with tumor size and RECIST ORR in this dataset is consistent with historical use of CA-125 as a surrogate marker of tumor response.
- Model-predicted CA-125 profiles and GCIG responses for etirinotecan pegol q14d or q21d were similar, suggesting that the better-tolerated q21d schedule will produce clinical results consistent with those from both schedules used in the phase 2 study.

References

1. A.E. Guppy, G.J. Rustin. CA-125 response: can it replace the traditional response criteria in ovarian cancer? Oncologist. 2002;7(5):437-43.
2. B. You, et al. Mathematical modeling of CA-125 kinetics in recurrent ovarian cancer (ROC) patients treated with chemotherapy and predictive value of early modeled kinetic parameters in CALYPSO trial: A GCIG study. J Clin Oncol 29: 2011 (suppl; abstr 5065)
3. M.A. Eldon, U. Hoch. Population Pharmacokinetics of NKTR-102, a topoisomerase I inhibitor-polymer conjugate in patients with advanced solid tumors. J Clin Oncol 29: 2011 (suppl; abstr 2598)
4. P.A. Canney et al. Ovarian cancer antigen CA-125: A prospective clinical assessment of its role as a tumor marker. Br. J. Cancer (1984), 50, 765-679.

Abstract ID: 5048, Poster Board #: 18H, S HALL A2, McCormick Place, 2012 ASCO Annual Meeting, Chicago, IL.
To download a copy of this poster, please visit http://www.nektar.com/product_pipeline/oncology_nktr-102.html

Acknowledgement:
The authors would like to thank all patients and investigators who took part in the study. The authors would also like to thank Carol Zhao, Emily Xu and Shibani Harite from Nektar Biometrics and Data Management group for providing the datasets and the results in the background. The work was supported by Nektar Therapeutics.