San Antonio Breast Cancer Symposium
December 4-8, 2012

Etirinotecan Pegol (NKTR-102) in Patients With Metastatic Breast Cancer (MBC): Modeling CA27.29 Response and its Correlation with Tumor Response

Yen Lin Chia, Ute Hoch, Alison Hannah, Michael A. Eldon, Nektar Therapeutics, San Francisco, CA

INTRODUCTION

- Etirinotecan pegol (NKTR-102) is a long acting next-generation topoisomerase I inhibitor with a unique pharmacokinetic (PK) profile that provides continuous active drug exposure with reduced peak concentration.[1]
- Etirinotecan pegol is in clinical trials for patients with solid tumors, including breast, ovarian and colorectal cancers.
- The BEACON study (BrEAst Cancer Outcomes with NKTR-102) is a phase 3 randomized, open-label, international study of etirinotecan pegol in patients with MBC that will evaluate single agent etirinotecan pegol in patients previously treated with an anthracycline, a taxane, and capecitabine (ATC) versus a comparator arm consisting of an active single agent Treatment of Physician's Choice (TPC).
- This phase 2 study[2] evaluated two treatment regimens of etirinotecan pegol (145 mg/m^2 q14d and q21d; n=70; 35 per treatment regimen), for patients with a median of 2 cytotoxic regimens for MBC (100% prior taxane, 89% prior anthracyline; 26% with prior ATC):
 - 29% ORR observed with single agent etirinotecan pegol (both schedules showed similar ORR).
 - ORR was also maintained in other heavily pre-treated and poor prognosis patient subsets:
 - ER+ and /or PR+: 29%
 - Triple-negative: 39%
 - Visceral disease: 30%
- Side effects were generally manageable; most common severe toxicity was diarrhea (G3 23% in q21d), typically occurring after 3 months of therapy.

Table 1. Phase 2 Efficacy Results in Patients with MBC

	Total
Overall Response Rate (ORR)[2]	29% (N=66) 31% in prior ATC subset (N=16)
Progression-Free Survival (PFS)[2]	4.6 months (5.3 months in q21d)
Overall Survival (OS)[2]	10.3 months (13.1 months in q21d)
Overall best CA27.29 response (50% or better decline in at least one observation from baseline)	36% (16/45)

BACKGROUND

- For many cancers, serum tumor markers play an important role for monitoring therapy in patients with advanced disease.
- The most widely used serum tumor markers in breast cancer are CA15-3, CA27.29, and carcinoembryoinic antigen (CEA).
- CA15-3 and CA27.29 are glycoproteins secreted by breast cancer cells, with well-characterized assays that allow detection of the circulating MUC-1 antigen in peripheral blood.
- ASCO guidelines[3] recommend that CA15-3 and CA27.29 can be used in conjunction with diagnostic imaging, history, and physical examination for monitoring patients with metastatic disease during active therapy.
- The majority of patients with MBC have elevated levels of serum CA27.29.
- CA27.29 response was encouraging in our Phase 2 study of etirinotecan pegol in patients with MBC whose disease had failed prior taxane-based treatment, and stimulated our focus on this biomarker.

Figure 2. Change (%) in Serum CA27.29 Profiles after 145 mg/m^2 Etirinotecan Pegol q14d or q21d (n = number of CA27.29 evaluable samples available at each cycle)

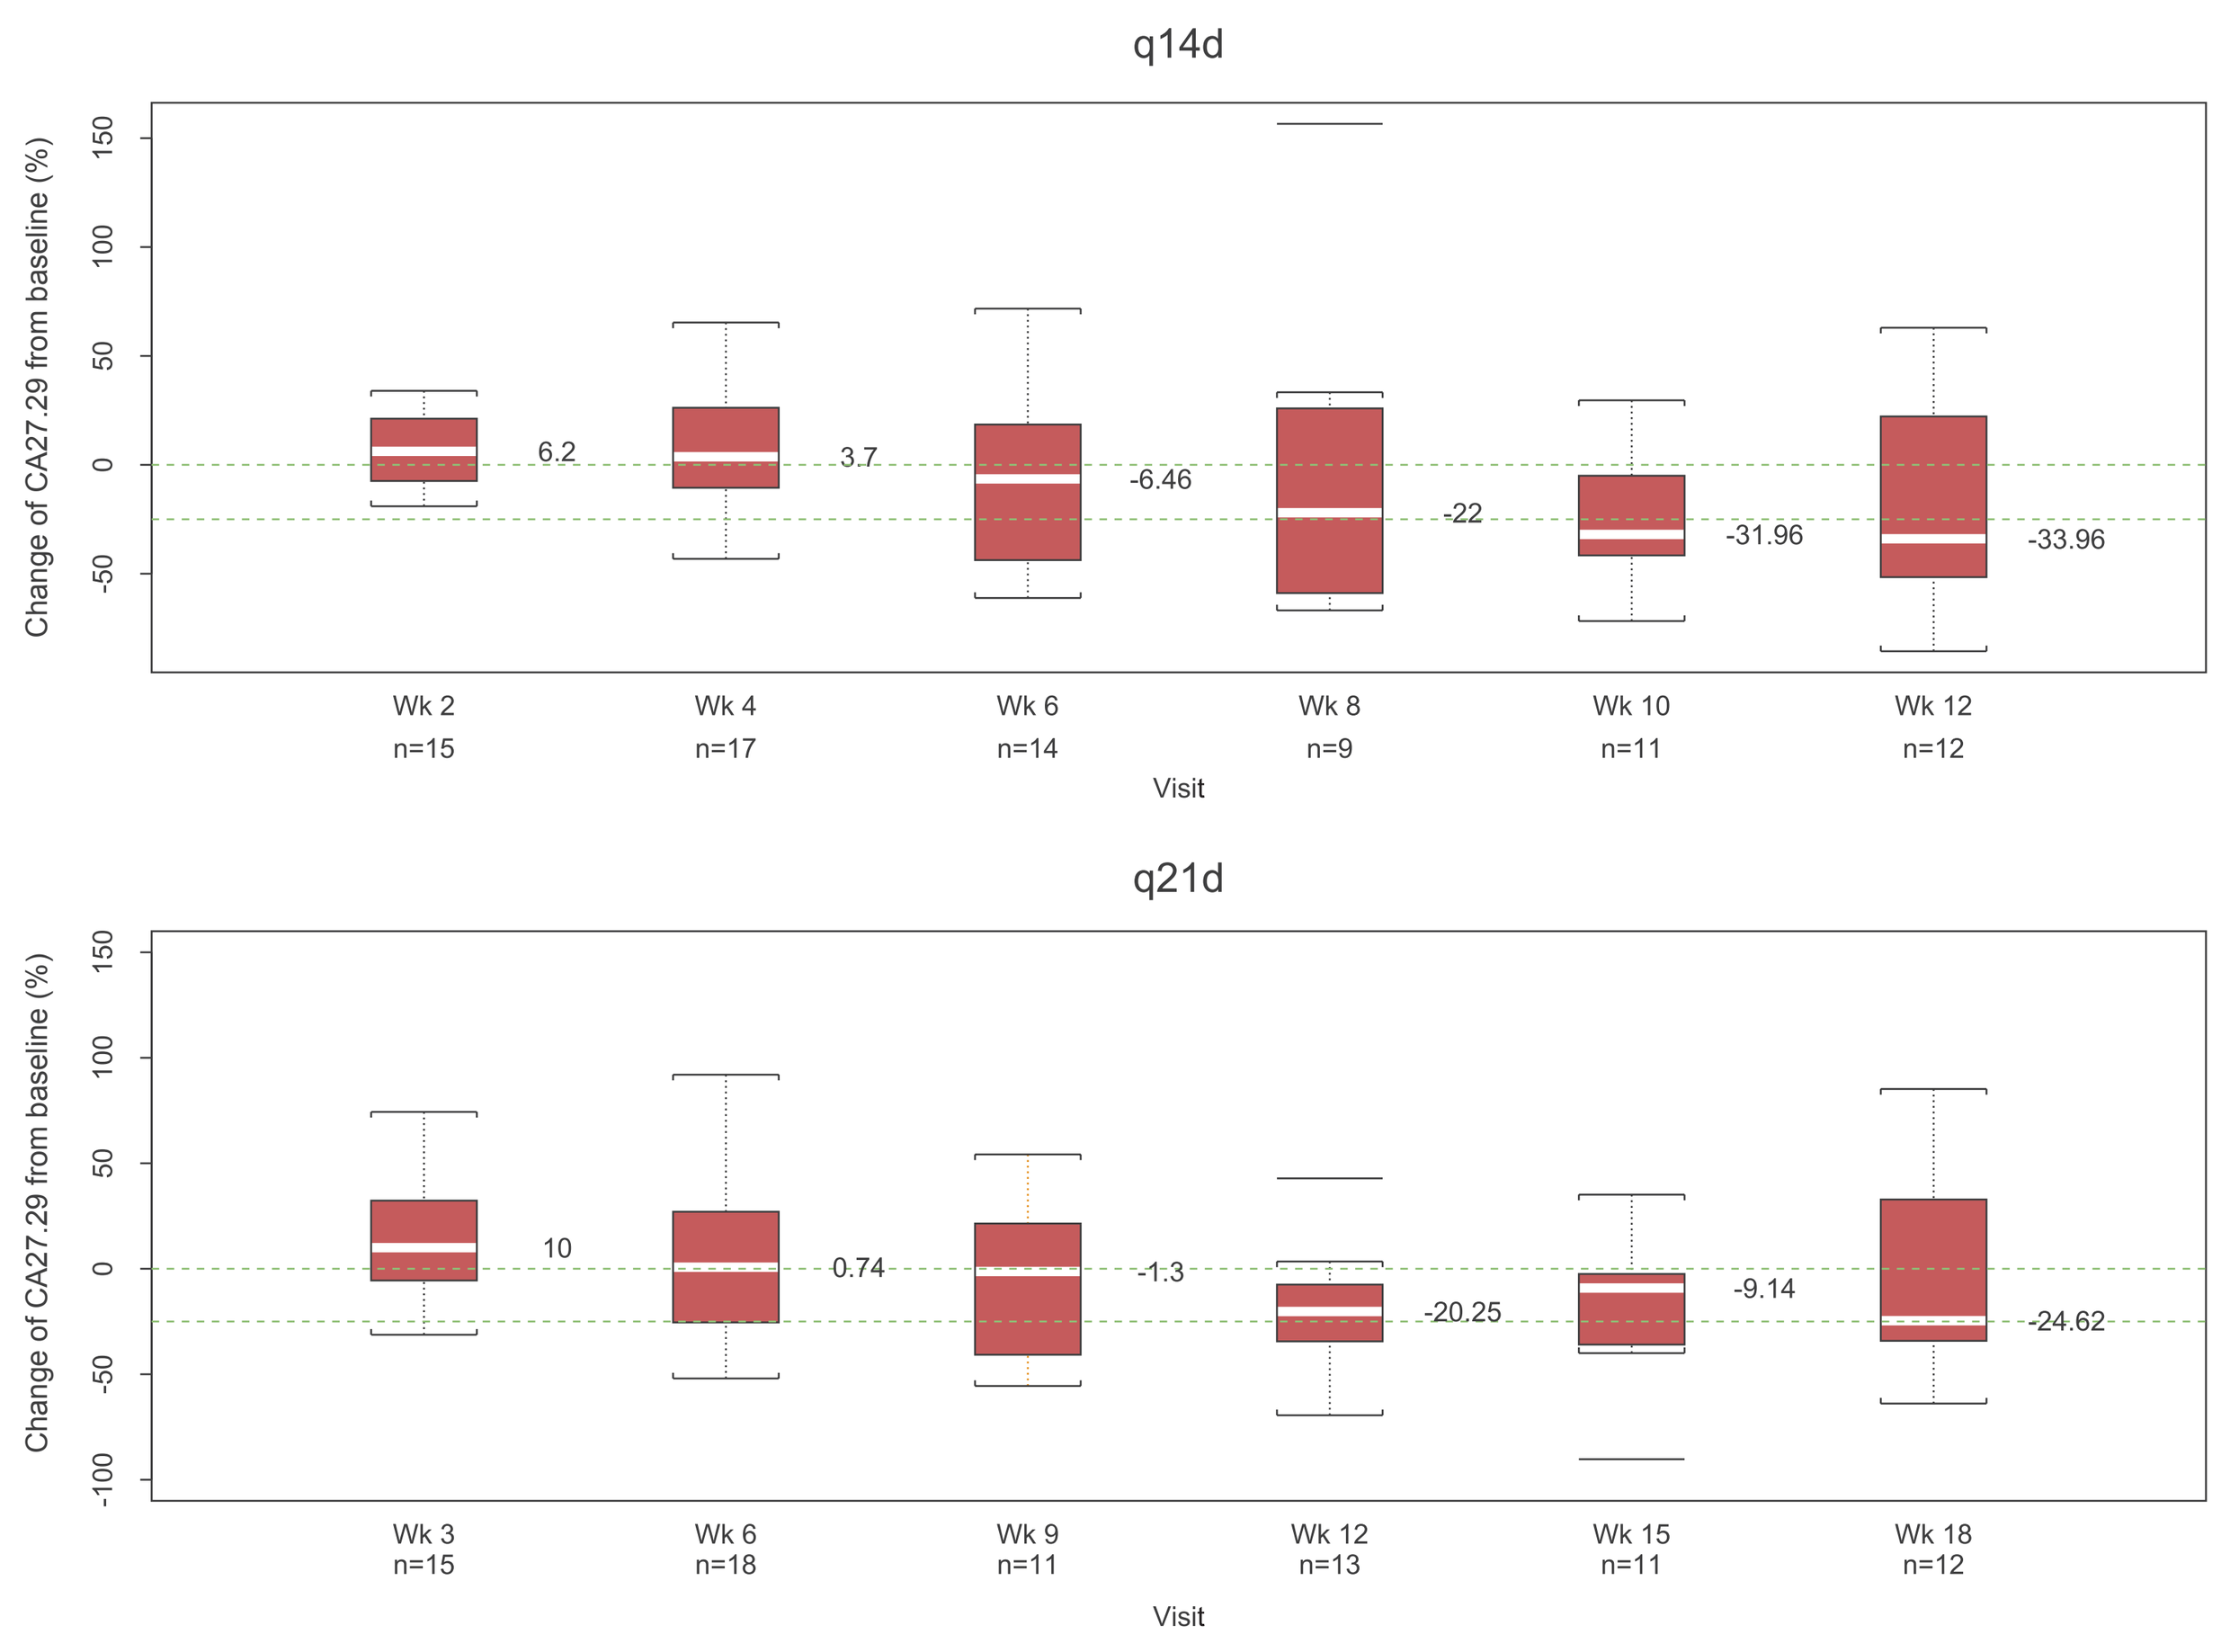
q14d
Change of CA27.29 from baseline (%)
6.2
3.7
-6.46
-22
-31.96
-33.96
Wk 2 n=15
Wk 4 n=17
Wk 6 n=14
Wk 8 n=9
Wk 10 n=11
Wk 12 n=12
Visit
q21d
10
0.74
-1.3
-20.25
-9.14
-24.62
Wk 3 n=15
Wk 6 n=18
Wk 9 n=11
Wk 12 n=13
Wk 15 n=11
Wk 18 n=12

One extreme observation of >700% change from baseline at Wk 3 for q21d was excluded.

- We developed a mathematical model linking the kinetics of CA27.29 to SN38 exposure, tumor size, and RECIST with the intent of predicting response in various clinical scenarios, such as changes in dose and/or schedule, or the occurrence of dosing delays.

DATA

- In the Phase 2 study, 70 patients with MBC were randomized to 145 mg/m^2 etirinotecan pegol q14d or q21d.
- Baseline CA27.29 measurements from 48 patients with available screening values as shown in Table 2.

Table 2. Baseline CA27.29 Measurements (U/mL) at Screening

	Min	25% quartile	Median	75% quartile	Max
q14d (N = 26)	13	38.8	89.0	149.5	3383
q21d (N = 22)	14	25.0	61.5	242.0	669
Total (N=48)	13	27.0	78.0	184.0	3383

- Our analysis dataset consisted of the 45 patients with at least two CA27.29 measurements (pre- and post-first dose).

MODEL BUILDING

- Predict plasma SN38 concentration-time profile for each patient based on dosing information using our previously developed population PK model.[4]
- Investigate the relationship of predicted plasma SN38 concentration and serum CA27.29 concentration to test the hypothesis.

 Hypothesis: SN38 (indirectly) inhibits the production of CA27.29.
- Model:

$$\xrightarrow{k_{in}} \boxed{CA27.29} \xrightarrow{k_{out}}$$

$$\frac{d[CA27.29]}{dt} = k_{in} \cdot \overbrace{e^{\beta t}}^{\text{Exponential growth of CA27.29 production rate}} \cdot \underbrace{\left(1 - \frac{[C_{SN38}]}{IC50 + [C_{SN38}]}\right)}_{\text{Inhibitory effect}} - k_{out}[CA27.29]$$

- k_{in}: production rate of serum CA27.29
- k_{out}: elimination rate of serum CA27.29
- [CA27.29]: serum CA27.29 (U/mL)
- [C_{SN38}]: plasma SN38 concentration (ng/mL)

RESULTS

Model Diagnostics and Projections

- CA27.29 concentration-time profiles were well described by the model independent of response pattern.

Figure 3. Examples of Typical CA27.29 Profiles: Observed (black circle) Versus Individual Predicted CA27.29 (red line)

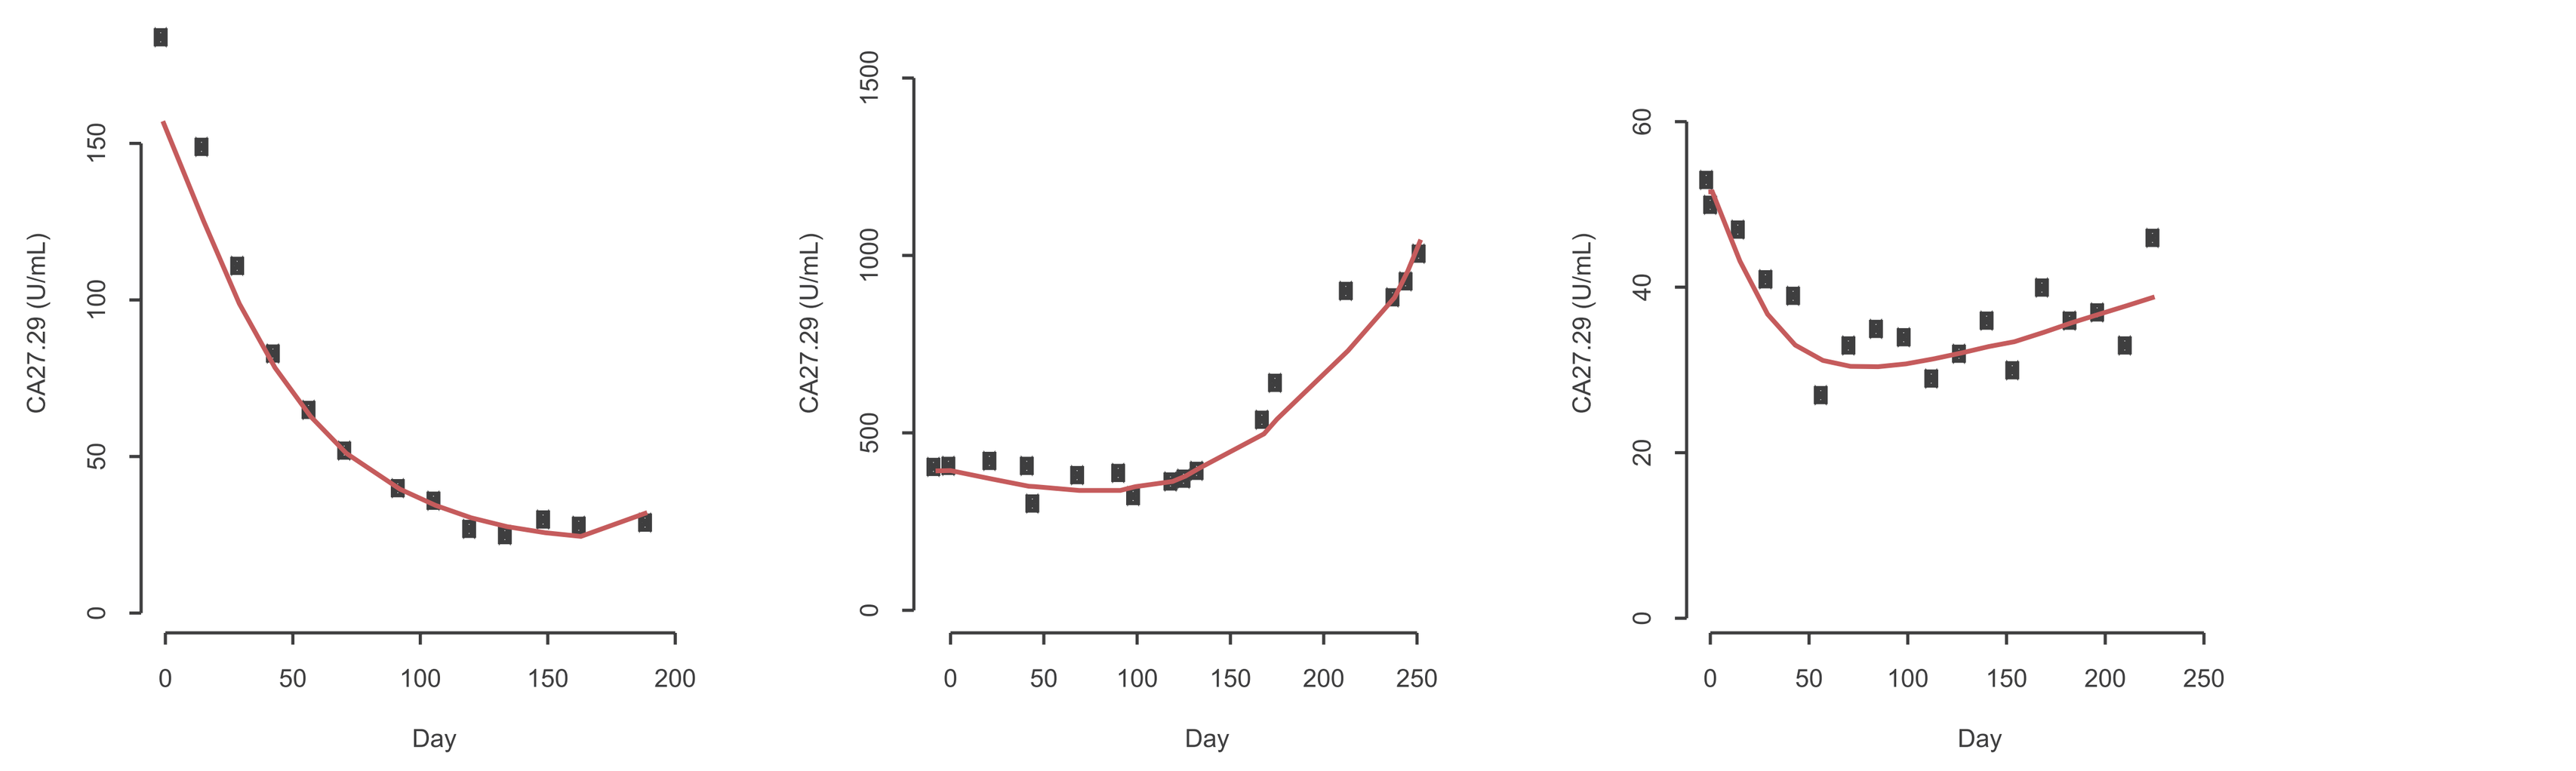
CA27.29 (U/mL)
Day

- The half-life of CA27.29 decline was estimated to be 15 days.
- The modeled population mean IC_{50} of SN38 was 1.6 ng/mL.
- In the absence of dose interruptions, the minimum plasma SN38 concentration (C_{min}) at steady state reached approximately 94% and 55% of IC_{50} for the q14d and q21d regimen, respectively.

Figure 4. Projections of SN38 Concentrations after 145 mg/m^2 q14d and q21d

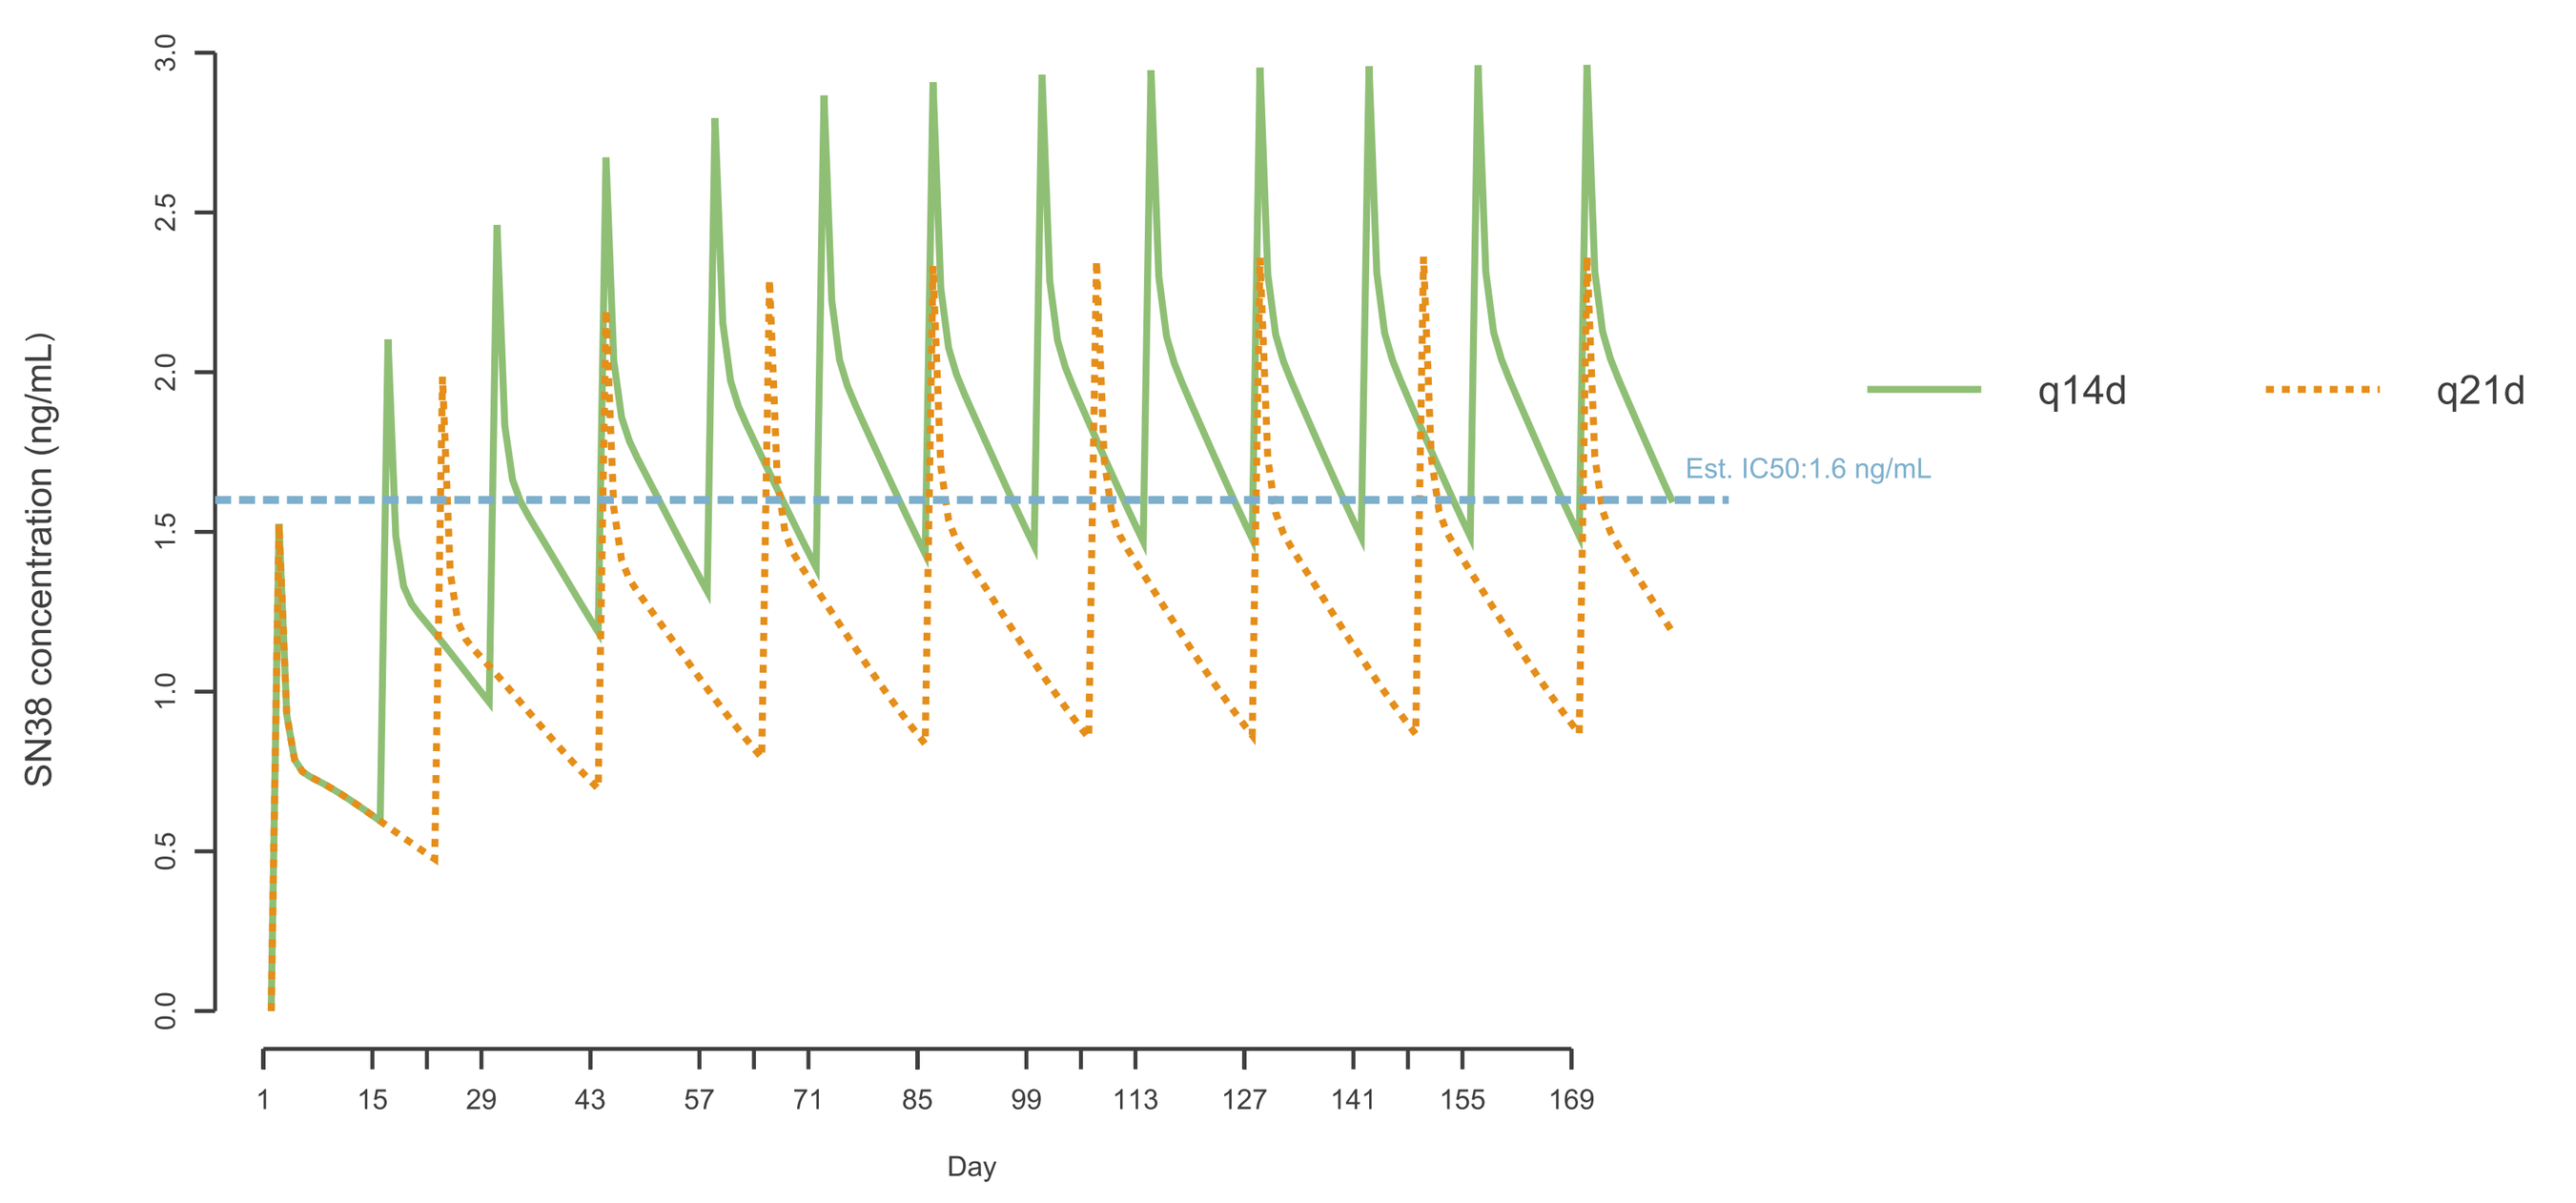
SN38 concentration (ng/mL)
Est. IC50:1.6 ng/mL
q14d
q21d
Day

Correlation of CA27.29 with RECIST Response

- Of the 45 patients, 43 patients had pre- and post-treatment tumor measurements for correlation of CA27.29 response with tumor size.
 - 93% (14/15) of the patients with RECIST CR or PR exhibited at least 10% declines in CA27.29 from baseline during the study.
 - 83% (5/6) of the patients with RECIST SD ≥ 6 months also manifested ≥10% declines, whereas only 45% (5/11) of the patients with SD < 6 months showed ≥10% decline in CA27.29.
 - 82% (9/11) of the patients with RECIST PD, showed CA27.29 elevation from baseline.

Figure 5. Observed Percent Change Of Serum CA27.29 Time Course Stratified by Response (Each Line Represents One Patient)

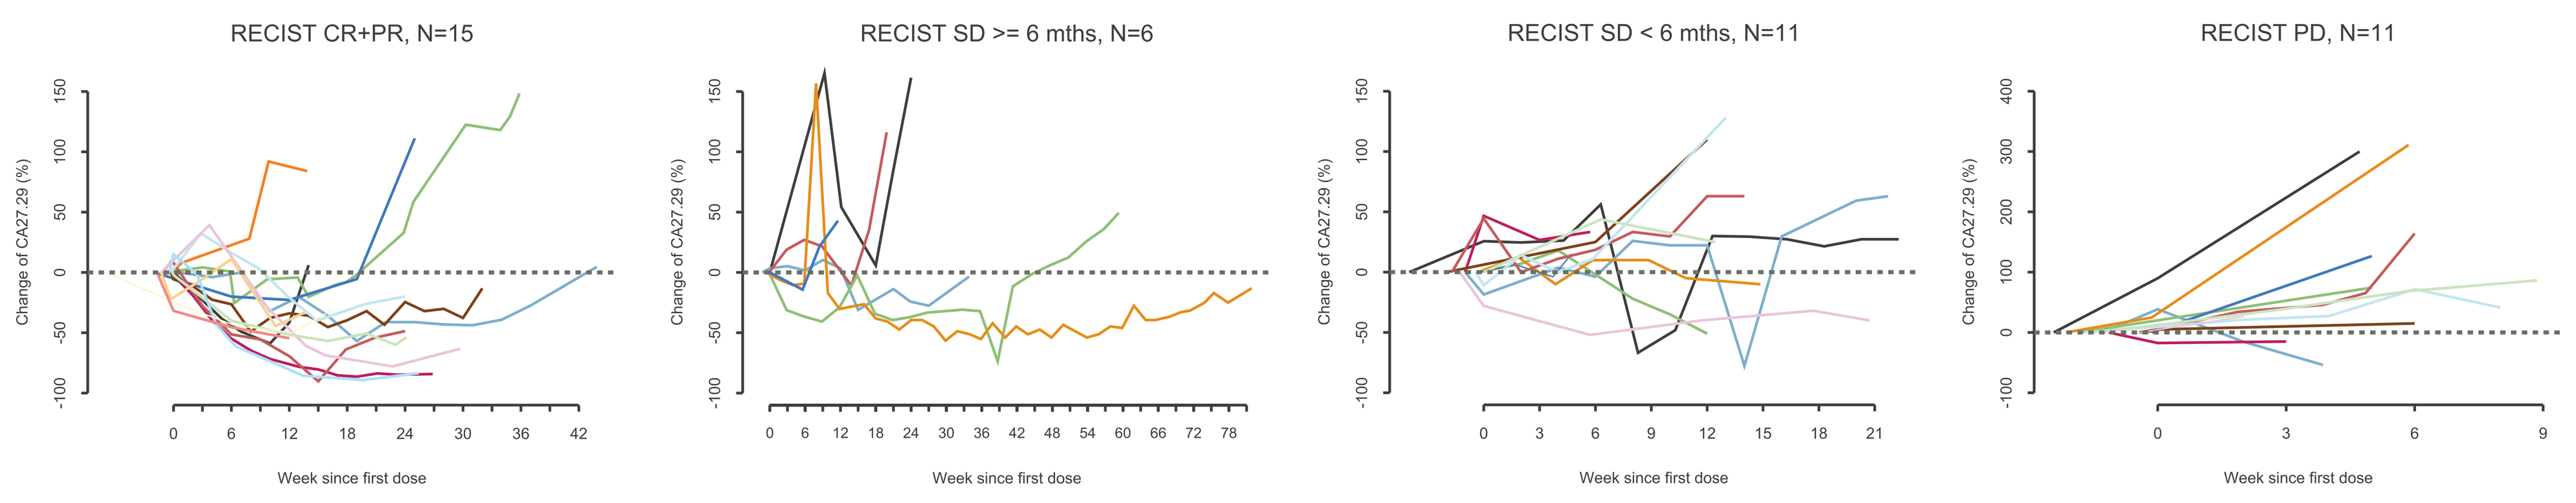
RECIST CR+PR, N=15
RECIST SD >= 6 mths, N=6
RECIST SD < 6 mths, N=11
RECIST PD, N=11
Change of CA27.29 (%)
Week since first dose

One extreme outlier observation of >700% from baseline, under RECIST SD >=6 months, was excluded from the plot.

Larger Declines in CA27.29 from Baseline were Associated with Better RECIST Response

Figure 6. Observed Maximum Changes in CA27.27 from baseline by RECIST Response

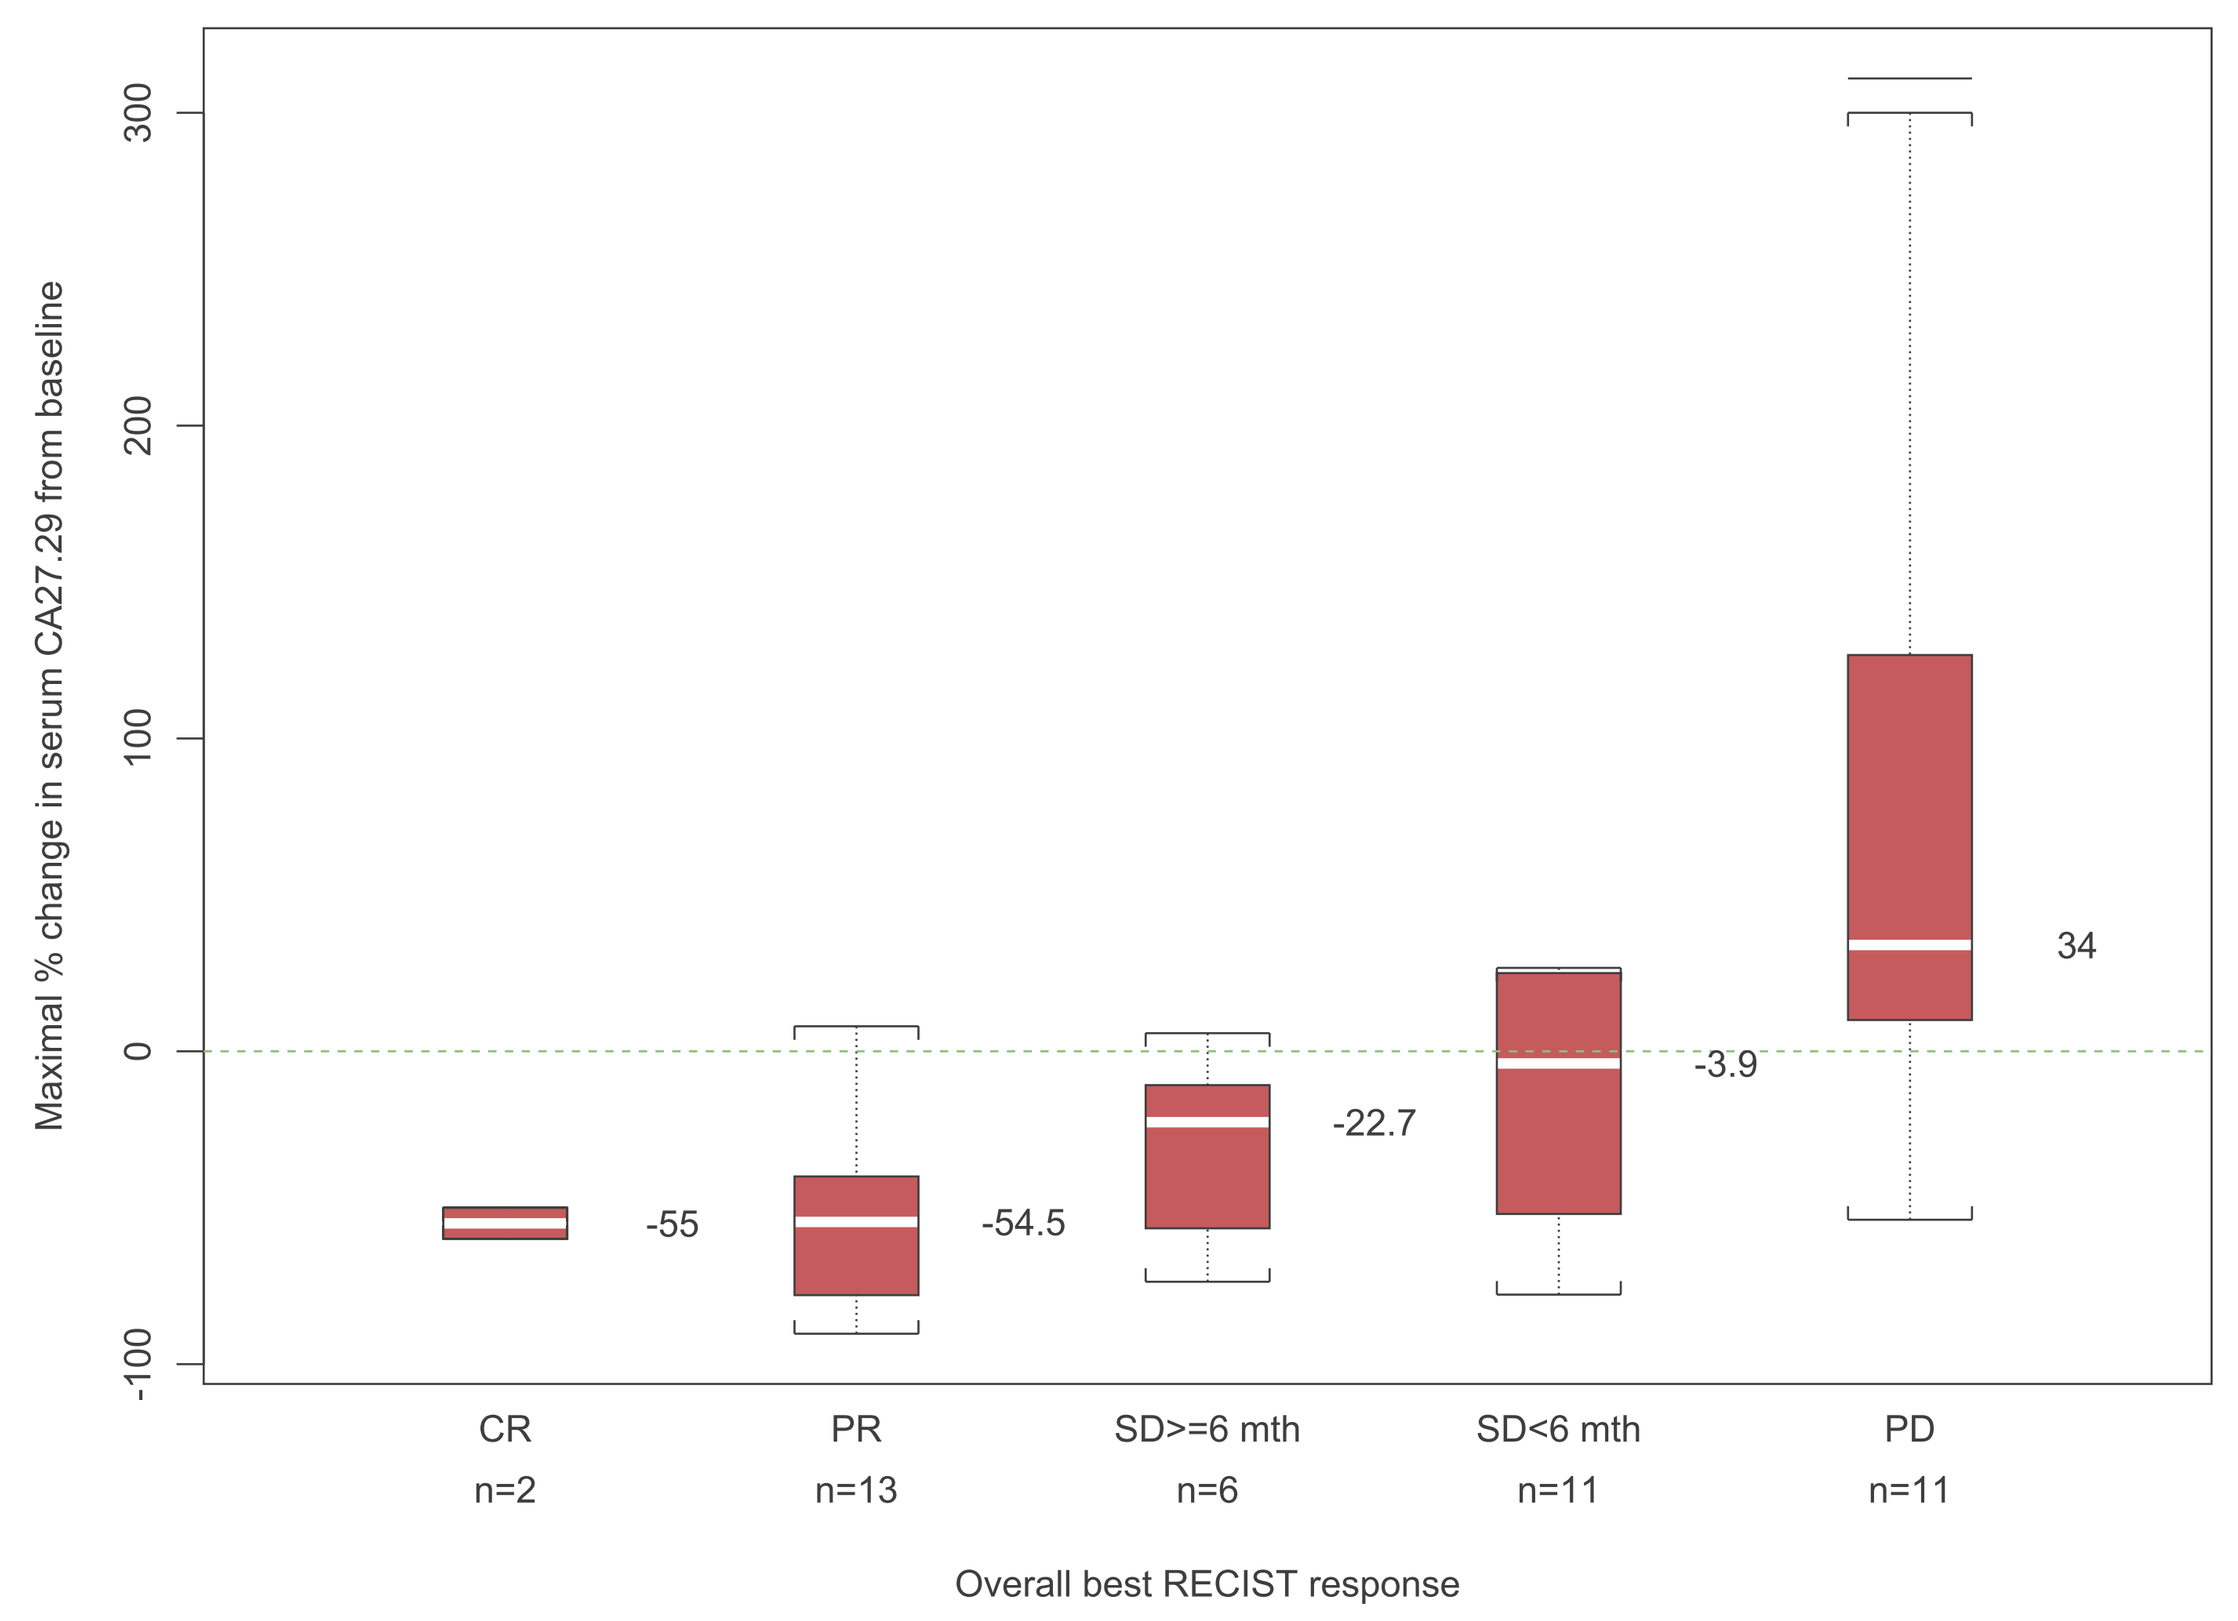
Maximal % change in serum CA27.29 from baseline
-55
-54.5
-22.7
-3.9
34
CR n=2
PR n=13
SD>=6 mth n=6
SD<6 mth n=11
PD n=11
Overall best RECIST response

- For PR and CR, the median of the maximum CA27.29 decline was larger than 50%.
- For SD ≥ 6 months, the median of the maximum decline in CA27.29 was 23%, whereas the median decline for SD < 6 months was negligible.
- For PD, CA27.29 increased in 8 of 10 patients; median minimum CA27.29 elevation from baseline during the study was 34%.

25% Reduction in CA27.29 at Week 6 as Predictor of Progression-free Survival (PFS)

- Based on ASCO recommendations, reduction in CA27.29 at week 6 was chosen to avoid spurious early rises that may occur during the first 4-6 weeks.
- Among the 26 patients with ≥ 25% reduction in CA27.29, 12 had not progressed or discontinued treatment by week 6:
 - The patients with ≥ 25% reduction by week 6 (n=9) had a median PFS of 12 months.
 - The patients with < 25% reduction (n=4) or elevations (n=13) in CA27.29 by week 6 had a median PFS of 6 months.
- Simulation of 1000 patients at population mean baseline CA27.29 receiving 145 mg/m^2 q21d projected that 46% achieve ≥25% reduction in CA27.29 at week 6 in the absence of dose reduction/interruption.

CONCLUSIONS

- The PK/PD model described CA27.29 profiles accurately, providing a tool to predict drug response from SN38 PK data.
- Change in CA27.29 from baseline may constitute an early marker for treatment response to etirinotecan pegol.
- Model-predicted CA27.29 profiles afer 145 mg/m^2 etirinotecan pegol q21d suggests that the better-tolerated q21d schedule will produce clinical results consistent with those from both schedules used in the phase 2 study.

REFERENCES

1. Jameson GS, et al. A Multicenter, Phase I, Dose-Escalation Study to Assess the Safety, Tolerability and Pharmacokinetics of Etirinotecan Pegol in Patients with Refractory Solid Tumors. Clinical Cancer Research: OnlineFirst Nov 7, 2012.
2. Awada A, et al. Antitumor activity in a randomized phase II study comparing two schedules of NKTR-102 in patients (Pts) with pretreated metastatic breast cancer (MBC). J Clin Oncol 29: 2011 (suppl; abstr 1034)
3. Harris L, Fritsche H, Mennel R, et al. American Society of Clinical Oncology 2007 update of recommendations for the use of tumor markers in breast cancer. J Clin Oncol 2007;25:5287-5312
4. Eldon MA, Hoch U. Hoch. Population Pharmacokinetics of NKTR-102, a topoisomerase I inhibitor-polymer conjugate in patients with advanced solid tumors. J Clin Oncol 29: 2011 (suppl; abstr 2598)

Scan to download poster

Program #P3-06-31 2012 CTRC-AACR San Antonio Breast Cancer Symposium.
This presentation is the intellectual property of the author/presenter. Contact ychia@nektar.com for permission to reprint and/or distribute.

Acknowledgment: This work was supported by Nektar Therapeutics.