Multiple Dose Pharmacokinetics and Pharmacodynamics of the New Oral Opioid Analgesic NKTR-181

NEKTAR

Lynn Webster[1], MD, PhD, Aleksandrs Odinecs[2], PhD, Susan Herzog[2], RN, Michael A Eldon[2], PhD, Robert Medve[2], MD
[1] Lifetree Clinical Research, Salt Lake City, UT; [2]Nektar Therapeutics, San Francisco, CA

Introduction

- NKTR-181 is a new mu-opioid analgesic molecule designed to provide clinically relevant analgesia while reducing CNS mediated side effects.[1]
 - NKTR-181 produced a dose-dependent central analgesic response in healthy subjects, with onset apparent within one hour of dosing.[2]
- A completed Phase 1 single ascending dose study of NKTR-181 demonstrated that NKTR-181 has a predictable dose-linear PK over a 50-fold range of doses.[2]
- The abuse properties of opioid analgesics are believed to relate to their rapid entry into the CNS.[3,4]
 - NKTR-181 exhibited a slower rate of CNS uptake in rats compared to commonly used opioids.[5,6]
 - NKTR-181 displayed markedly lower abuse liability than commonly used opioids in self-administration studies in rats and non-human primates.[2,6]

Objectives

The study presented here is a phase 1, double-blind, randomized, placebo-controlled, ascending multiple dose study of NKTR-181 administered orally in healthy male volunteers. The study was conducted in the U.S. at Lifetree Clinical Research (Salt Lake City, UT).

The primary objective was to evaluate the safety and tolerability of ascending oral doses of NKTR-181 administered every 12 hours over 8 days; the secondary objectives were to determine the pharmacokinetic (PK) profile and pharmacodynamic (PD) activity.

Methods

This ascending multiple dose study evaluated four dose levels: 100, 200, 300 and 400 mg. Each of four dose cohorts enrolled 15 subjects, enrolling a total of 60 healthy subjects over an eight-day treatment period. Subjects in each cohort received oral doses of NKTR-181 (n=12) in aqueous solution or placebo (n=3) following an overnight fast. Pharmacokinetics were determined through serial blood samples. Serial opioid pharmacodynamic tests included the cold pressor test (CPT) and a UVB injury model to assess analgesic effects and pupillometry as an indicator of the onset of opioid effect. Statistical analyses are based on a repeated measures mixed model analysis of variance.

References

1. Fishburn, C.S. et al., NKTR-181: A Novel Opioid With Slowed Brain Entry Shows Low Abuse Liability and Reduced CNS Side Effects. Frontiers in Research Symposium, 2010.
2. American Academy of Pain Management Annual Meeting, September 2011.
3. Pharmacol Biochem Behav. 2007; 86:45-54.
4. J Pharmacol Exp Ther. 2001; 299 :1056-65.
5. Society for Neuroscience 40th Annual Meeting, November 2010.
6. American Society of Anesthesiologists Annual Meeting October 2010.
7. Lalovic et al., Clinical Pharmacology and Therapeutics, 2006. (Table IV - $t_{1/2eO}$, effect site equilibration half-life)

Results

NKTR-181 Shows Dose-linear Pharmacokinetics with ~12 Hour Half-life

Figure 1. Mean (+SEM) Plasma NKTR-181 Concentration-Time Profiles in Healthy Subjects on Day 1

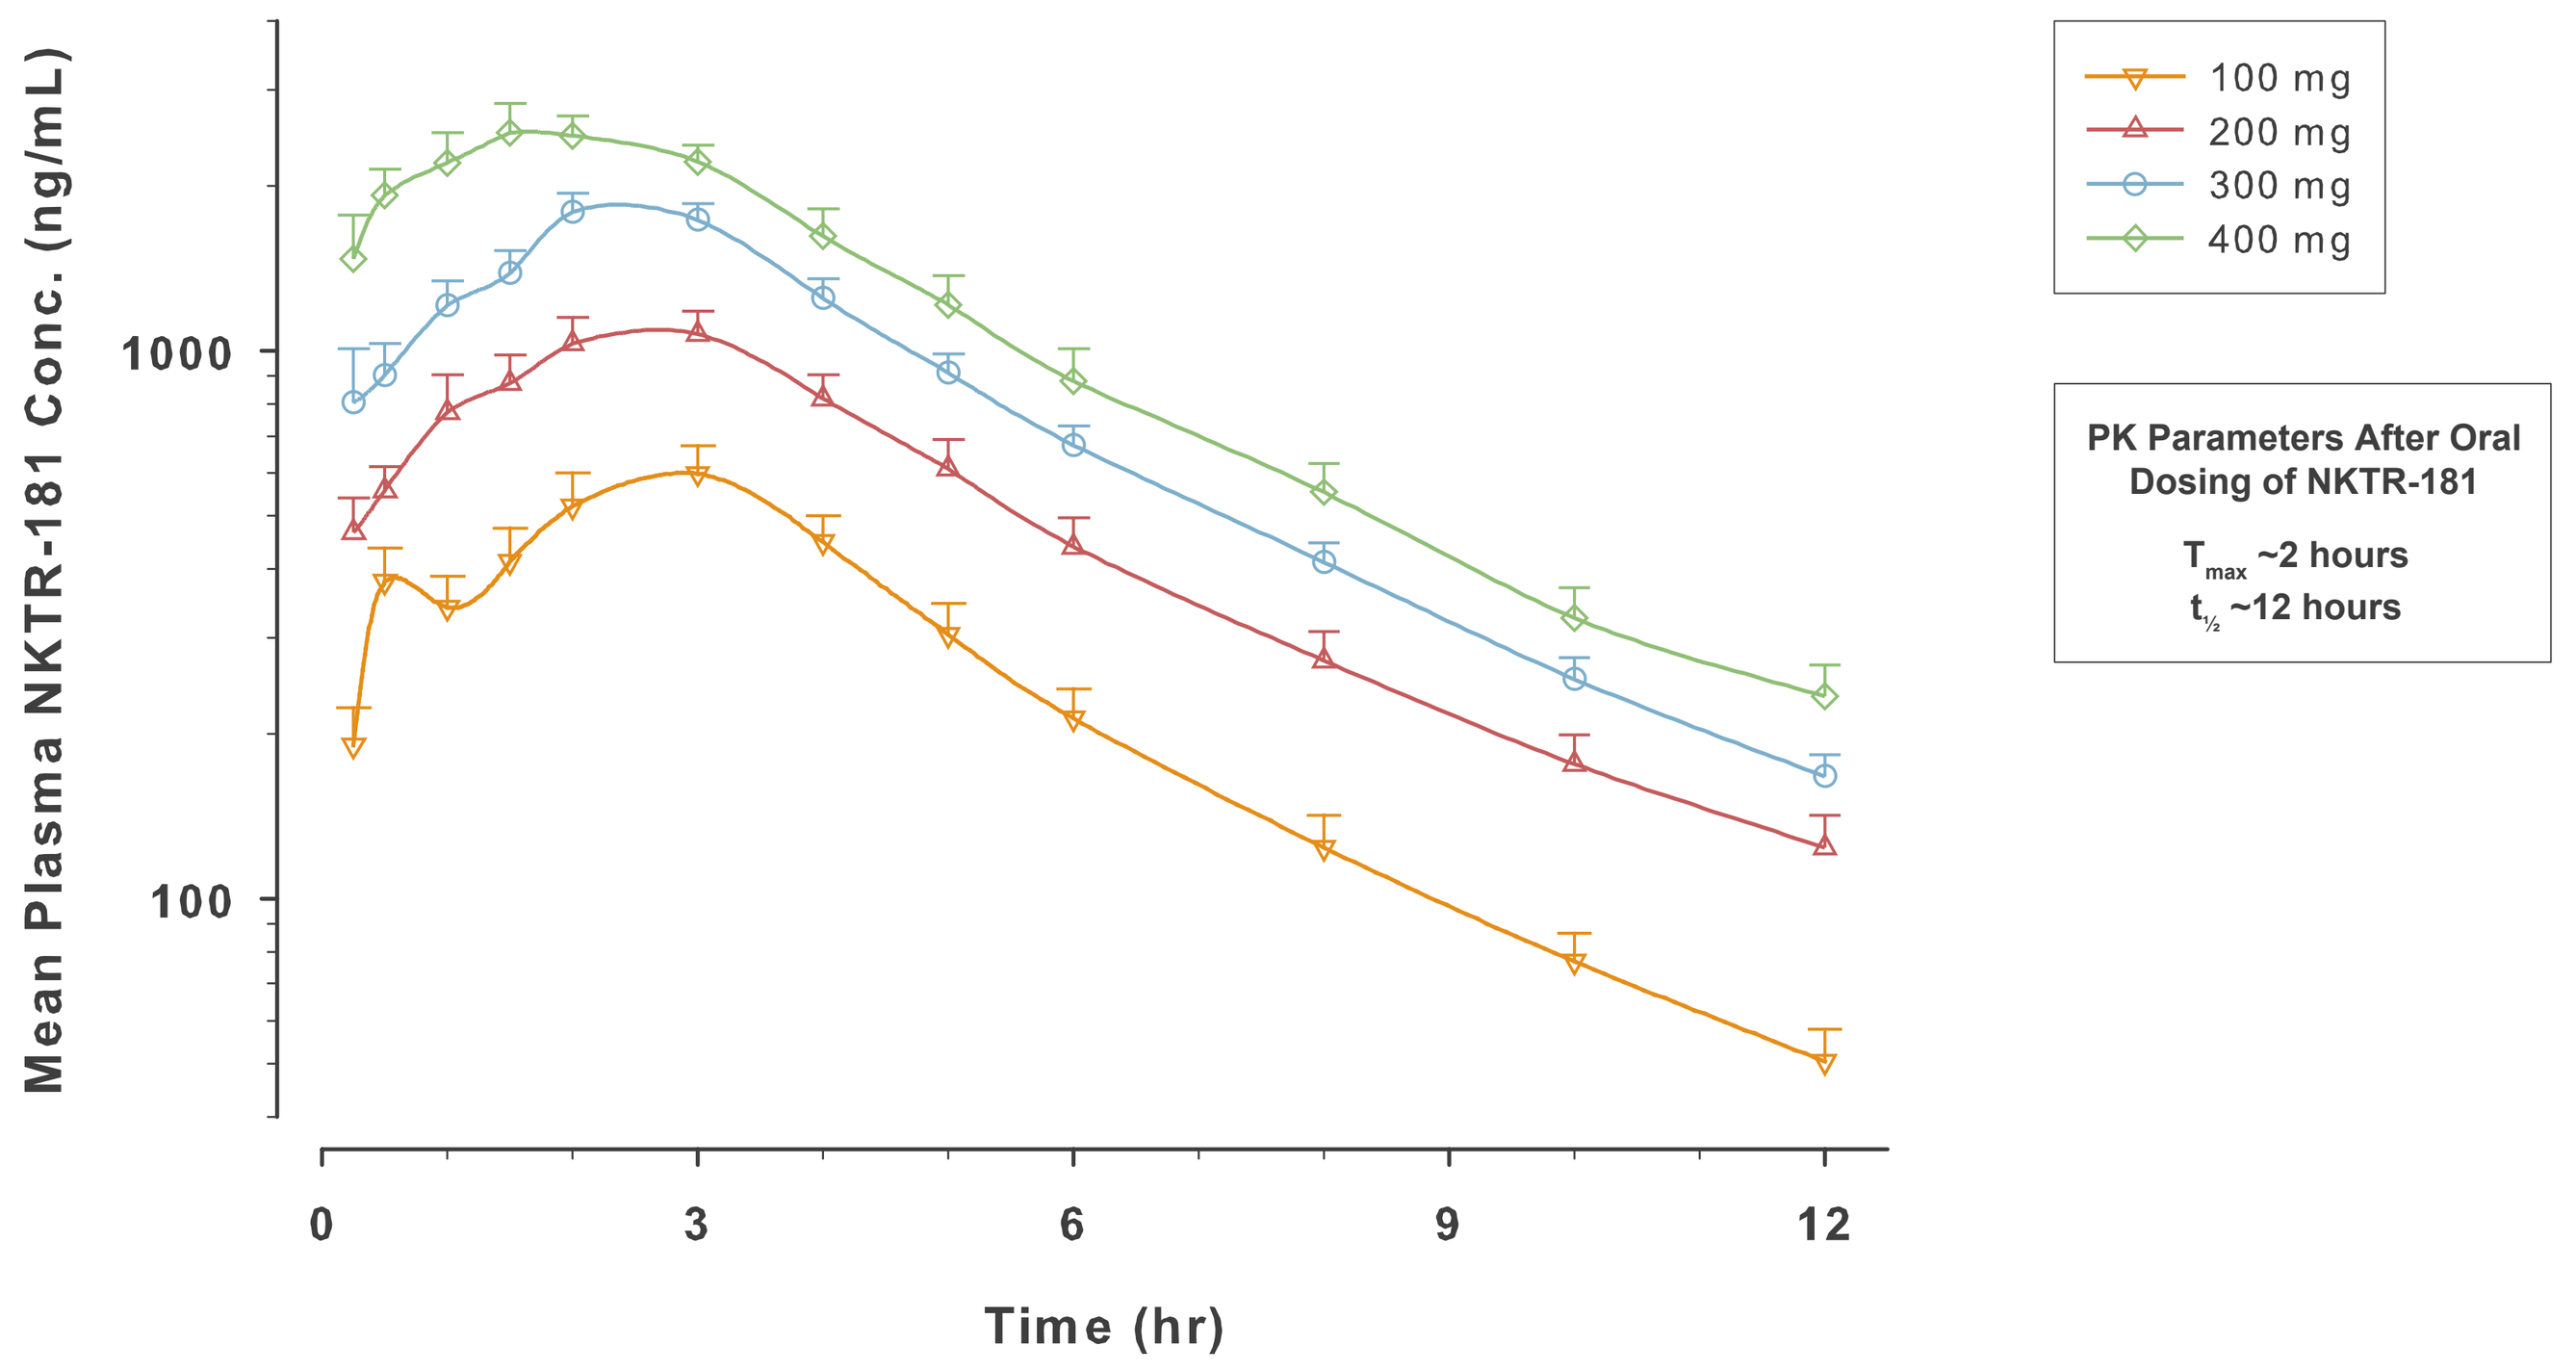
Mean Plasma NKTR-181 Conc. (ng/mL)
1000
100
0
3
6
9
12
Time (hr)
100 mg
200 mg
300 mg
400 mg
PK Parameters After Oral Dosing of NKTR-181
T_{max} ~2 hours
$t_{½}$ ~12 hours

Pharmacokinetics
NKTR-181 exhibited dose-linear pharmacokinetics across all dose levels on days 1 and 8. Pharmacokinetic steady state was achieved after day 3, with mean accumulation indexes of ~1.5 for C_{max} and ~1.8 for AUC_{0-12}. Maximum plasma NKTR-181 concentrations were achieved ~2 hours after dosing on both day 1 and day 8.

NKTR-181 Demonstrates Significant Central Effect Over 12 Hours

Figure 2. Peak central effect occurs ~4 hours following oral dosing

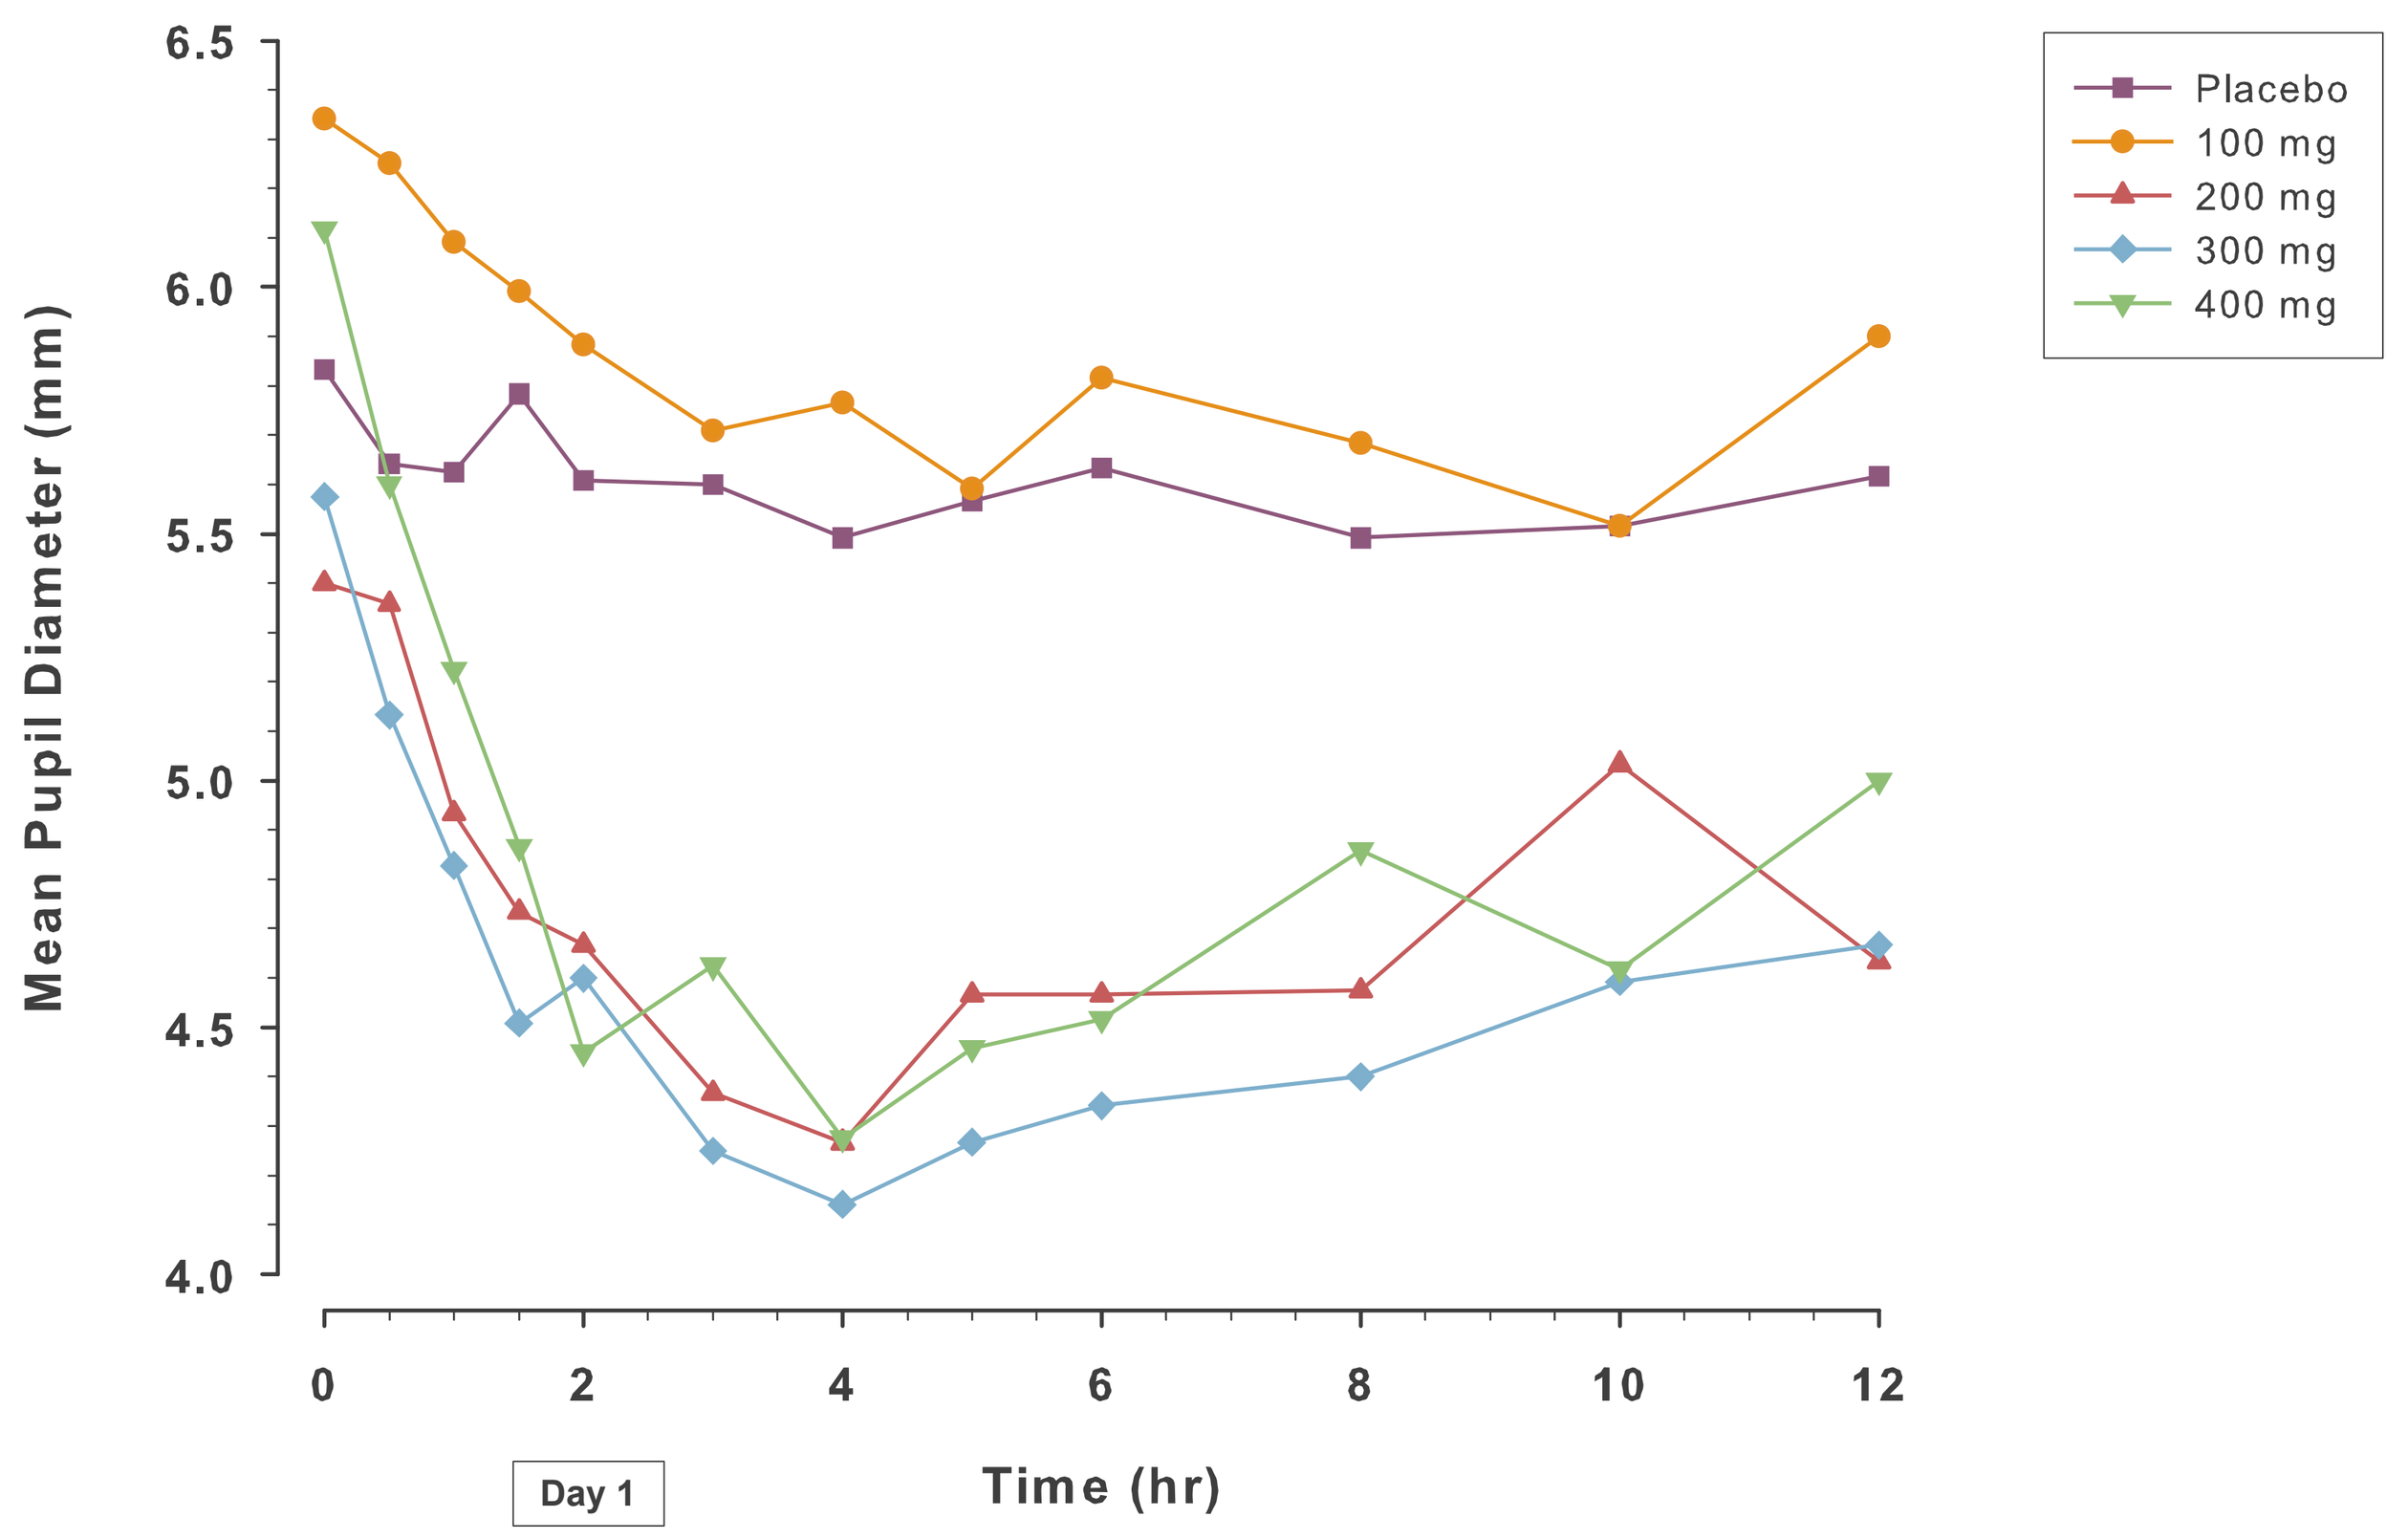
Mean Pupil Diameter (mm)
6.5
6.0
5.5
5.0
4.5
4.0
0
2
4
6
8
10
12
Day 1
Time (hr)
Placebo
100 mg
200 mg
300 mg
400 mg

Figure 2 presents the pupil diameter vs. time profiles that demonstrate a central effect of NKTR-181 that is statistically meaningful (P<0.001) following twice daily oral doses of 200-400 mg in this study. The peak central effect was consistently observed at ~ 4 hours after administration of NKTR-181, representing a significant time lag from the peak plasma level.

NKTR-181 Displays a 10-Fold Slower CNS Uptake As Compared to Oxycodone

Figure 3. Delay in onset of maximum central effect of NKTR-181 reflects slowed rate of entry to the CNS

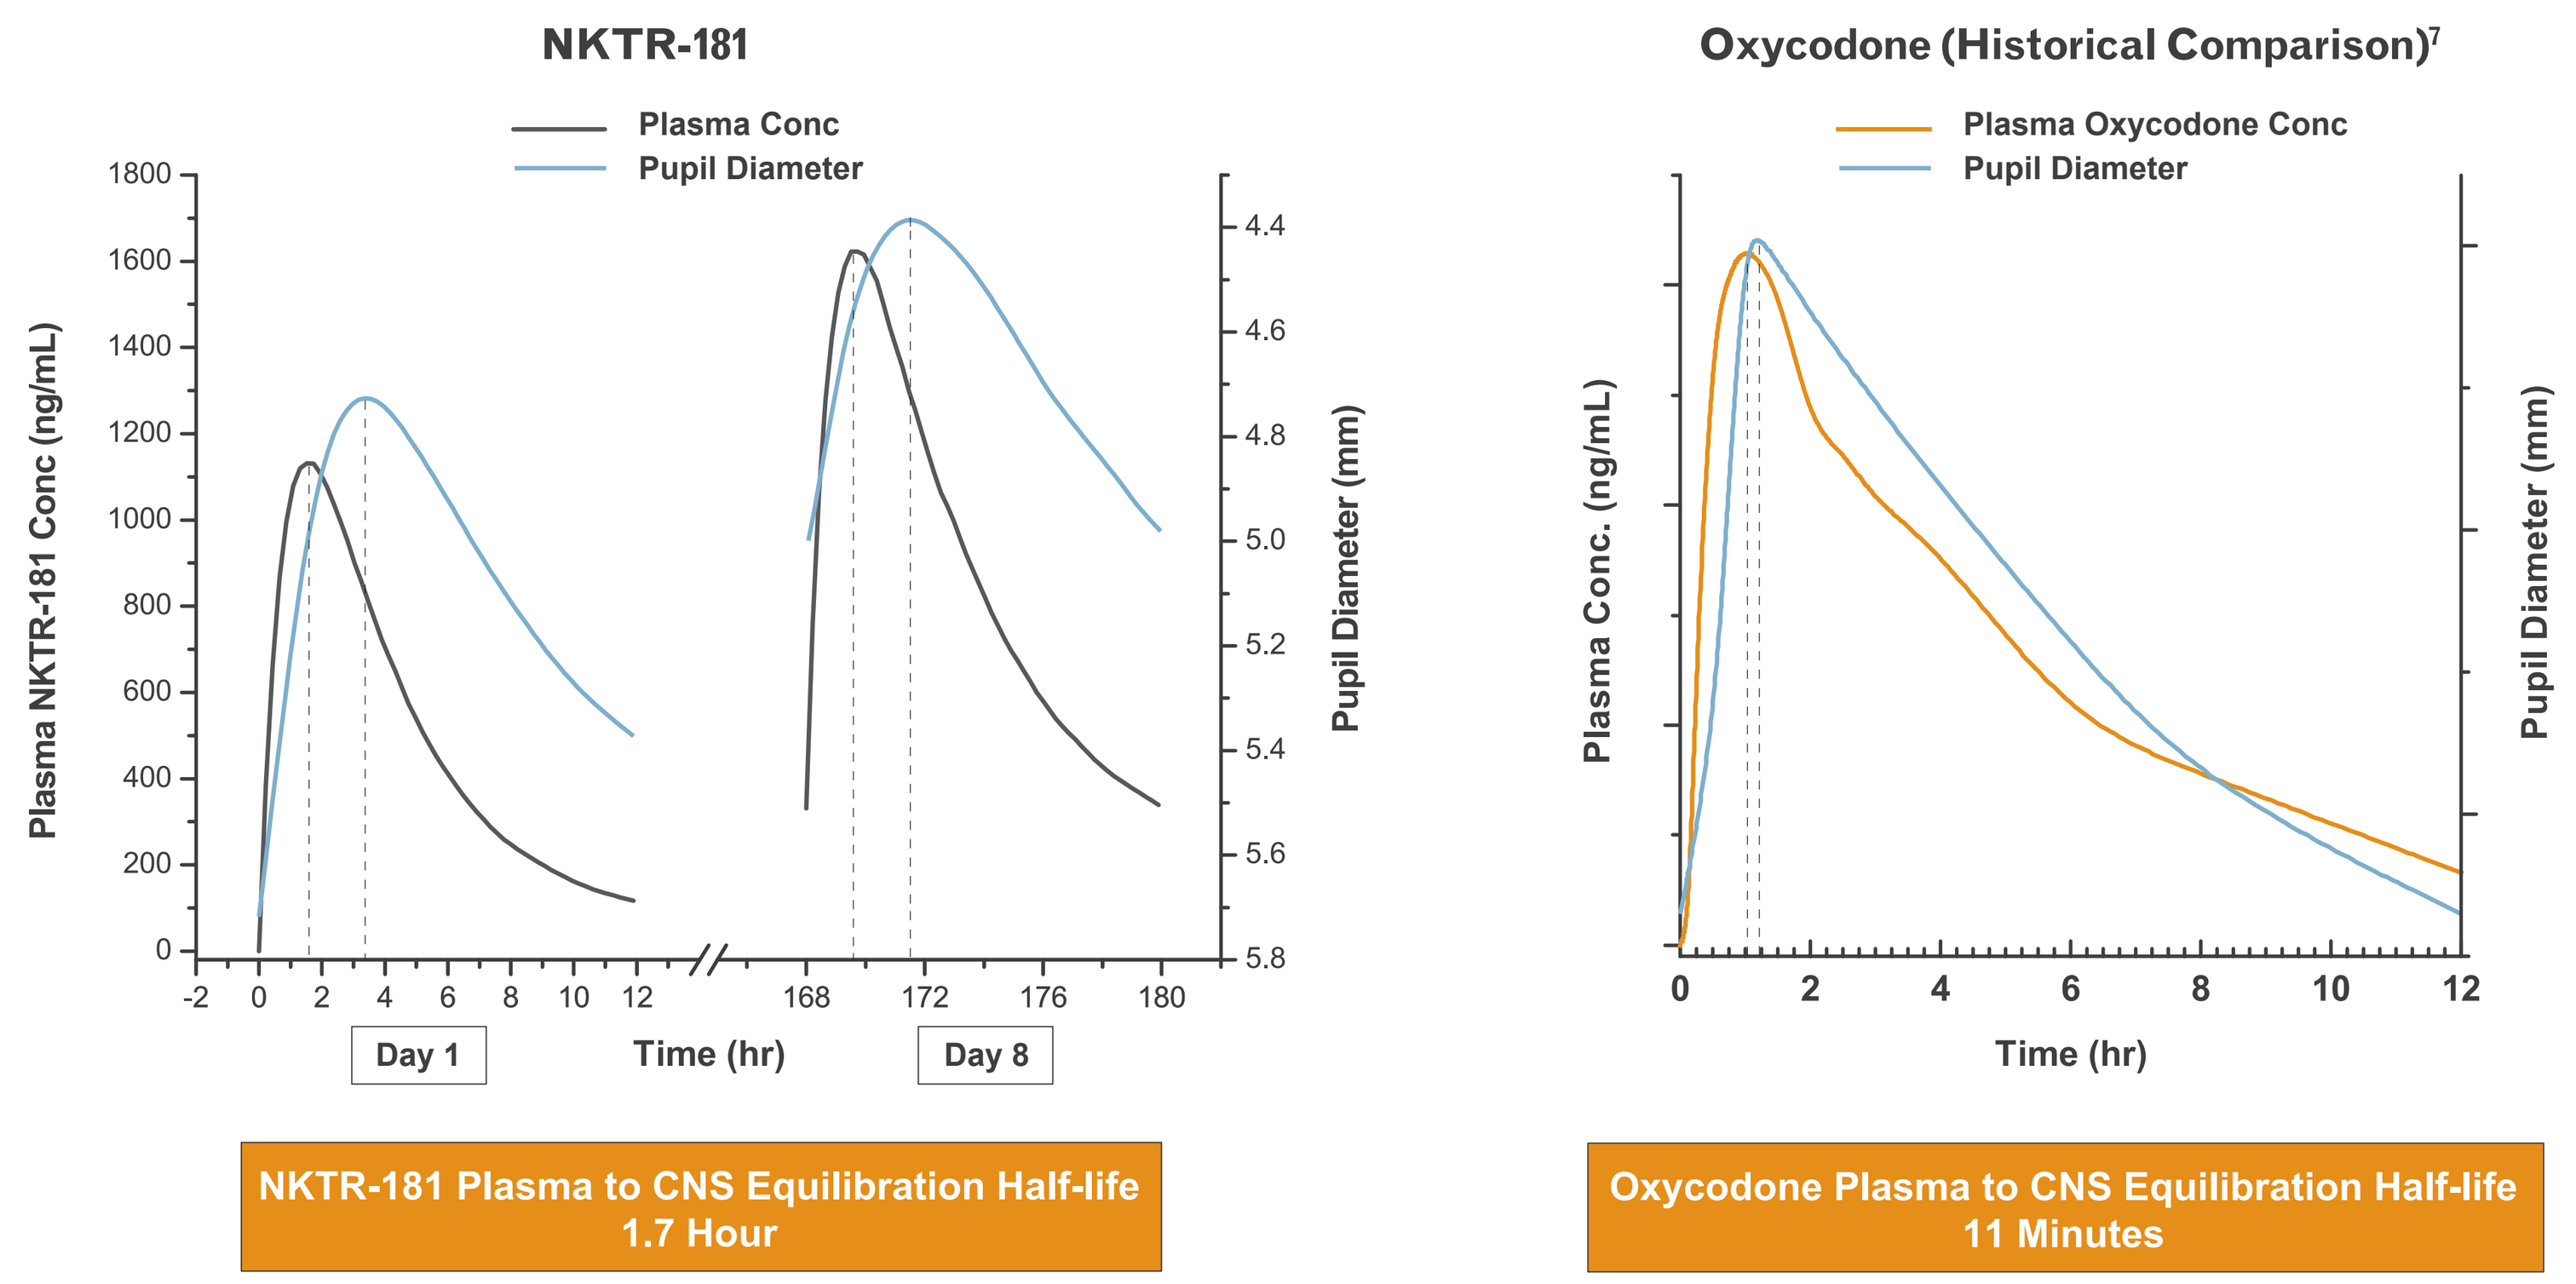
NKTR-181
Plasma Conc
Pupil Diameter
Plasma NKTR-181 Conc (ng/mL)
Pupil Diameter (mm)
Day 1
Time (hr)
Day 8
NKTR-181 Plasma to CNS Equilibration Half-life
1.7 Hour
Oxycodone (Historical Comparison)[7]
Plasma Oxycodone Conc
Pupil Diameter
Plasma Conc. (ng/mL)
Pupil Diameter (mm)
Time (hr)
Oxycodone Plasma to CNS Equilibration Half-life
11 Minutes

Figure 3 displays the onset and duration of miosis relative to the NKTR-181 concentration-time profile as illustrated by the 200 mg oral dose of NKTR-181. The time course of central effect (miosis) significantly lags the time course of NKTR-181 in plasma. The ~2 hour lag in peak central effects reflects the slowed entry of NKTR-181 to the CNS from the plasma. This slowed CNS entry is seen at all doses having central effect and is a property of the NKTR-181 molecule.

NKTR-181 Displays Sustained Dose-Dependent Central Effect With Twice Daily Dosing

Figure 4. Central effect (miosis) of NKTR-181 maintained with repeated dosing over multiple days

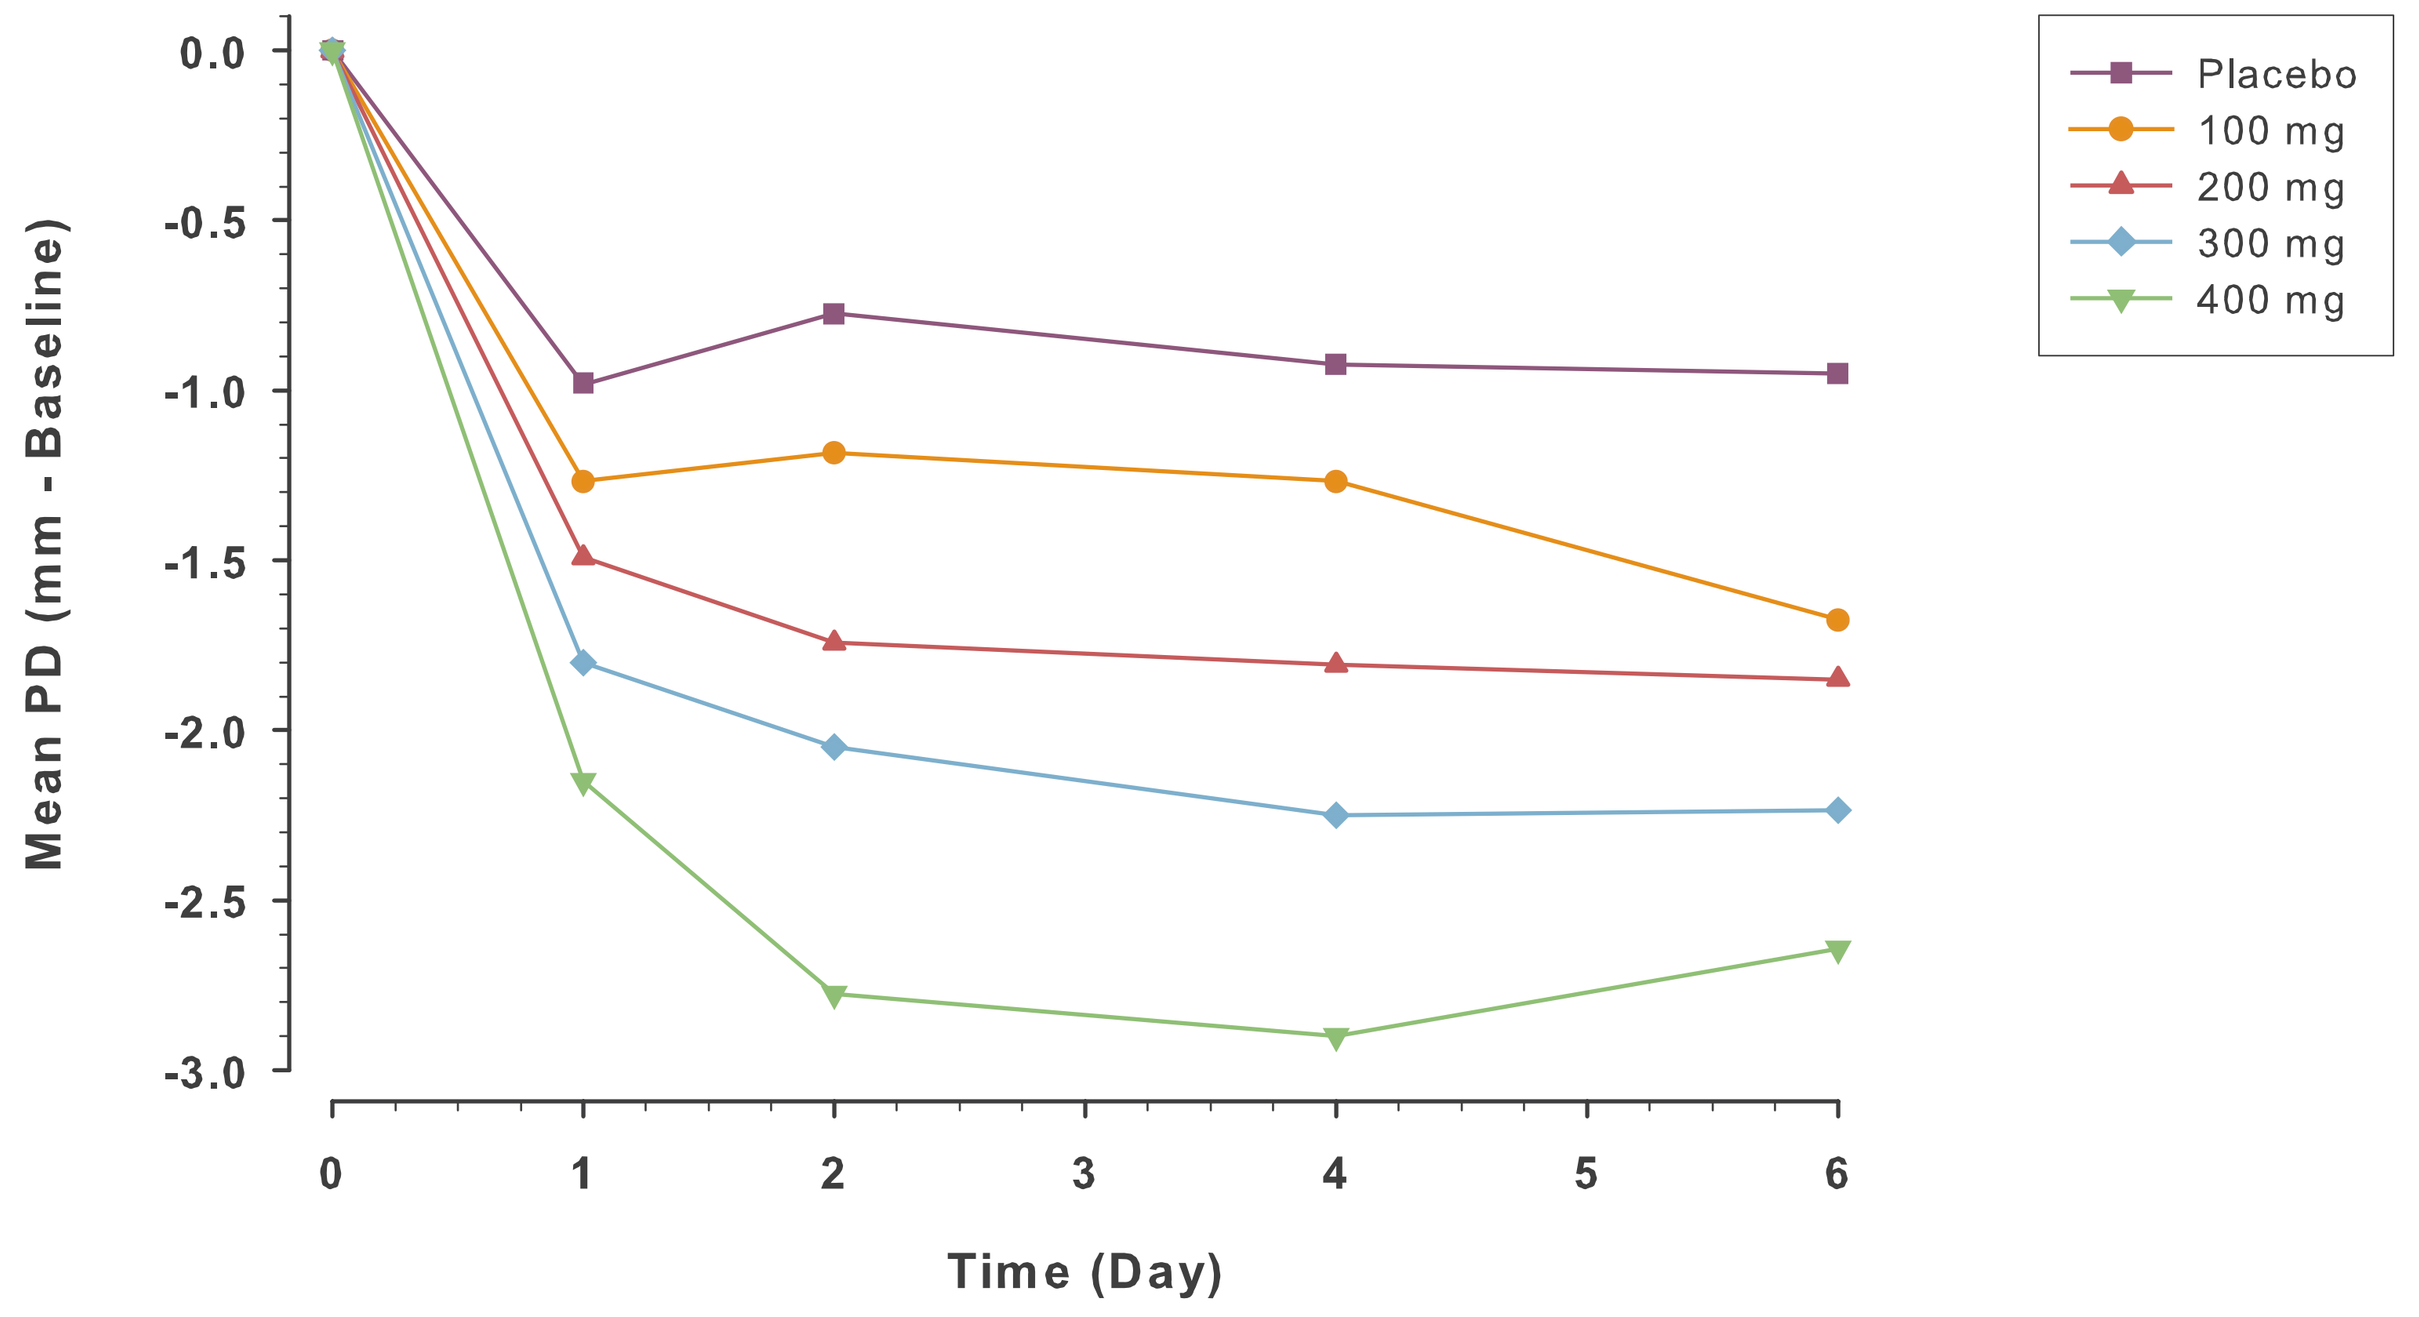
Mean PD (mm - Baseline)
0.0
-0.5
-1.0
-1.5
-2.0
-2.5
-3.0
0
1
2
3
4
5
6
Time (Day)
Placebo
100 mg
200 mg
300 mg
400 mg

Figure 4 presents the mean change in pupil diameter from baseline on days 1, 2, 4, and 6. Results show that there is a dose proportional NKTR-181 effect on the CNS and that this CNS-mediated effect persists with multiple dosing.

NKTR-181 is Safe and Well Tolerated at All Doses Over Entire Dosing Period

NKTR-181 was well tolerated over the entire 8-day dosing period in the study at all doses evaluated; most adverse events were mild and no serious adverse events were reported. Most frequent adverse events observed at the highest doses tested were consistent with AEs characteristic of an opioid agonist, such as constipation, headache and nausea.

NKTR-181 Displays Analgesic Activity in Multiple Pain Models

Figure 5. Central Analgesic Model: NKTR-181 shows sustained effect in Cold-Pressor Test (CPT)

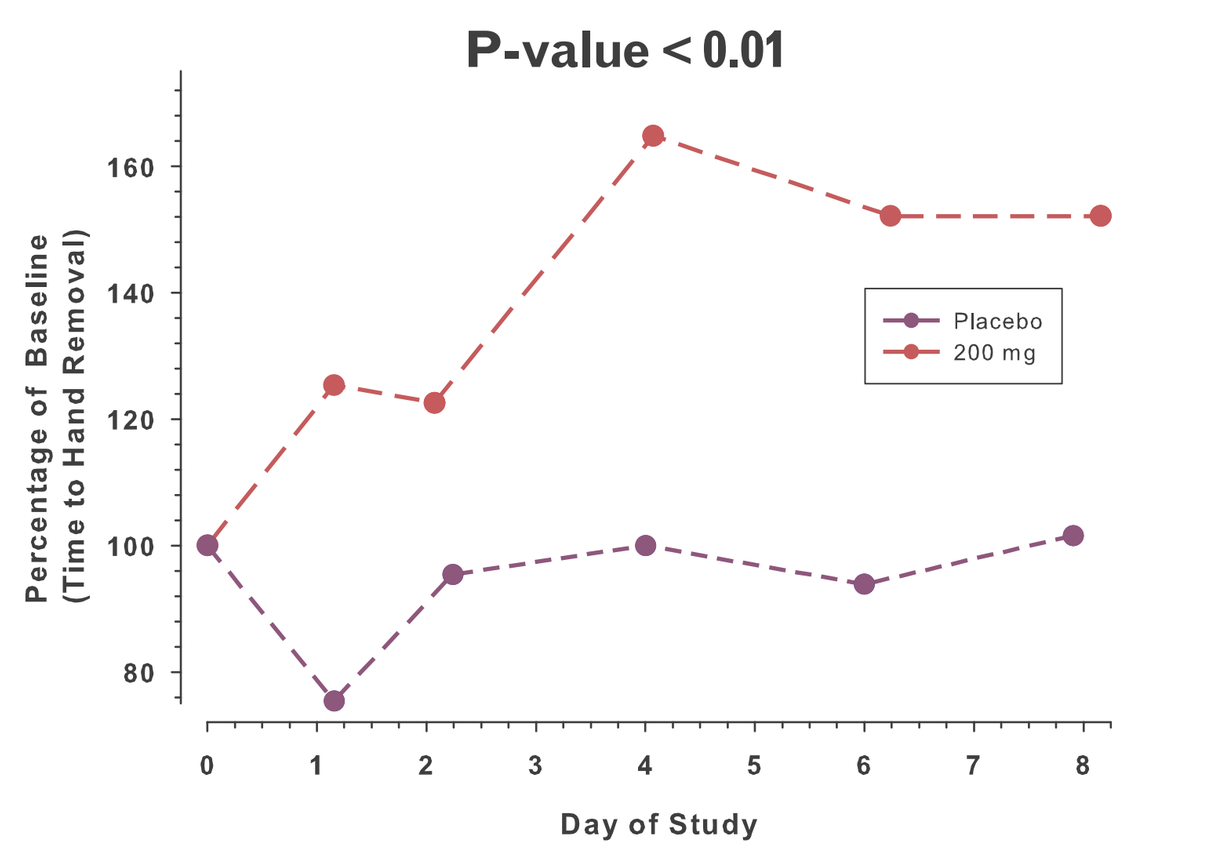
P-value < 0.01
Percentage of Baseline (Time to Hand Removal)
Day of Study
Placebo
200 mg

Results describing effect of 200 mg NKTR-181 on the latency-to-hand-removal (LHR) over 8-day dosing period are presented in Figure 5. The extent and duration of analgesic effect is assessed using increasing Latency to Hand Removal and an indicator of increased pain tolerance. The median of the individual subject responses increased from day 1 to day 8, reflecting increased analgesic effect once NKTR-181 reaches steady state.

Figure 6. Peripheral Analgesic Model: NKTR-181 demonstrates analgesia and anti-hyperalgesic activity following UVB injury

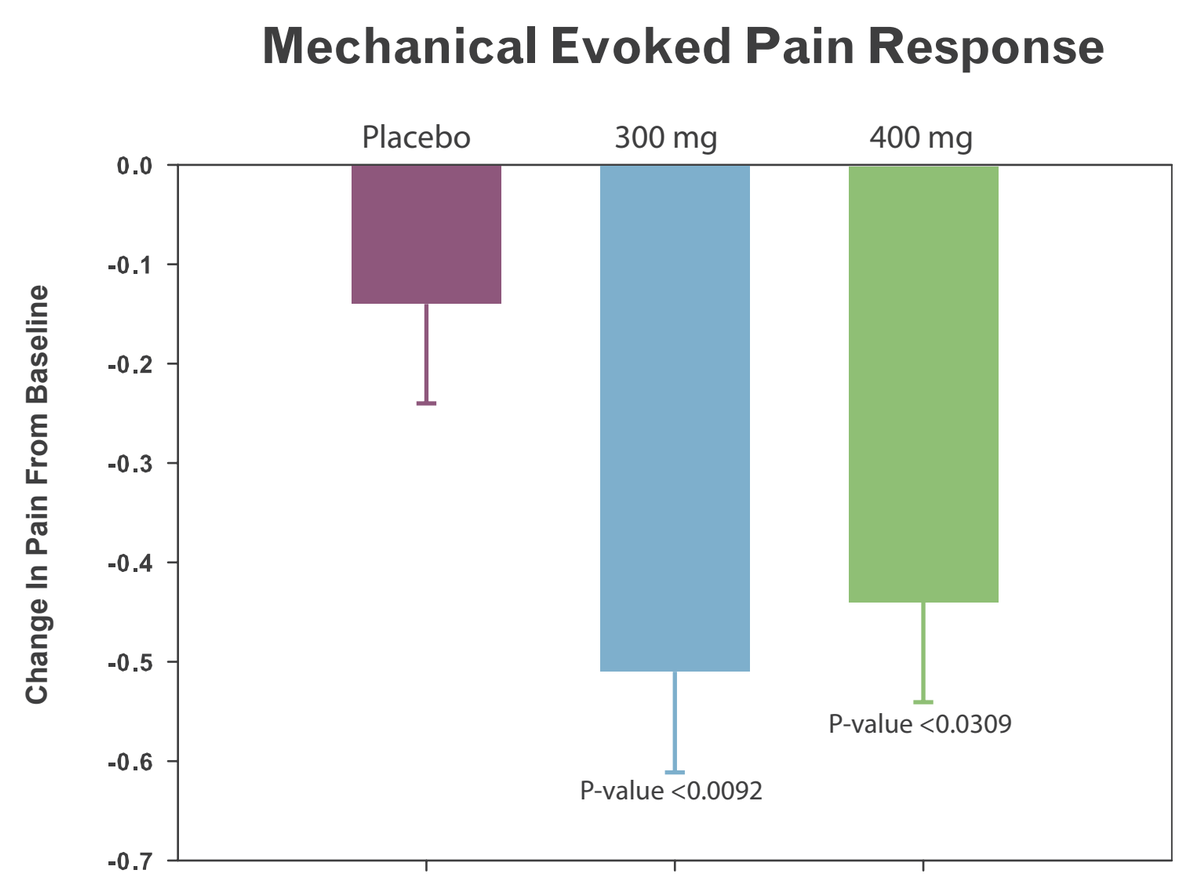
Mechanical Evoked Pain Response
Placebo
300 mg
400 mg
Change In Pain From Baseline
P-value <0.0092
P-value <0.0309

In an experimental model of induced UVB injury, NKTR-181 had significant anti-hyperalgesic effect following mechanical stimulation of irradiated skin. A consistent effect was observed in both mechanical and thermal stimulation in this model and maintained over the multiple dosing study period. These results indicate that NKTR-181 is producing effects on multiple pain modalities through both central and peripheral pathways.

Conclusions

- NKTR-181 is an orally-available new mu-opioid agonist molecule designed to have a slower rate of brain uptake compared with standard opioid therapies.
 - Human data confirms 10-fold slower CNS entry from plasma as compared to oxycodone.
- NKTR-181 produces significant peripheral and central analgesic responses.
 - NKTR-181 demonstrates a central analgesic response sustained over the 8-day dosing period in the cold-pressor test.
 - Response in the UVB model indicates that NKTR-181 provides relief of pain and hyperalgesia through both central and peripheral mechanisms.
- The PK profile of NKTR-181 is well-suited for the treatment of chronic pain.
 - Peak plasma level ~2 hours following dosing with an oral liquid.
 - Plasma half-life ~12 hours.
 - Plasma levels reach steady state after Day 3.
- Responses in the pharmacodynamic models and the overall safety profile of NKTR-181 over 8 days of twice-daily dosing to healthy volunteers suggest that NKTR-181 may have a wide therapeutic window: this hypothesis will be confirmed in Phase 2 testing.
 - Phase 2 studies of NKTR-181 in patients with chronic pain are currently being planned.

Poster #282B: Presented at the American Academy of Pain Medicine 28th Annual Meeting: Pain Medicine, February 23-26, 2012, Palm Springs, CA