Interspecies Comparison of Pharmacokinetics of the Novel Opioid Analgesic NKTR-181

Aleksandrs Odinecs, Michael A Eldon, Yuan Song, Juergen Pfeiffer, Susan Herzog, and Robert Medve
Nektar Therapeutics, San Francisco, CA

NEKTAR®

Background

- NKTR-181 is a new molecular entity opioid specifically designed to have a reduced rate and extent of CNS uptake.
- NKTR-181 is a mu opioid agonist, with preclinical antinociceptive activity comparable to oxycodone and morphine, but with reduced CNS side effects and a lower abuse liability (1).
- The chemical structure of NKTR-181 prevents conversion into an abuseable opioid, independent of formulation.
- NKTR-181 is currently in Phase 2 clinical studies in osteoarthritis patients with chronic pain of the knee.

Objectives

The objectives of this work were:

- to characterize NKTR-181 pharmacokinetics in mice, rats, and dogs.
- to predict NKTR-181 exposure in the First In Human (FIH) study, and then to compare predictions with results of the FIH study.

Methods

NKTR-181 IV and PO pharmacokinetics were determined in male CD-1 mice (1 mg/kg IV; 5 and 30 mg/kg PO, 4 mice/timepoint), male Sprague-Dawley rats (1 mg/kg IV; 1, 5,15 mg/kg PO, 4 rats per group), and male Beagle dogs (1 mg/kg IV; 1, 5,15 mg/kg PO, 3 dogs, crossover design). In humans seven groups of 15 subjects received single 10, 20, 40, 80, 160, 320, and 500 mg oral doses of NKTR-181 solution (n=12) or placebo (n=3) after an overnight fast. NKTR-181 in plasma was assayed using validated LC-MS/MS methods. The limits of quantitation (LOQ) for NKTR-181 in plasma were 0.2 ng/mL in mouse and 0.05 ng/mL in rat, dog and human. The in vitro protein binding of NKTR-181 in mouse, rat, dog and human plasma was determined using equilibrium dialysis.

PK parameters were calculated using compartmental methods (WinNonlin Pro version 5.3). For interspecies allometric scaling, PK parameters obtained for each species with 2-compartmental analysis were plotted against corresponding body weights in log-log scale. Linear least-squares regression was applied to fit relationship to the equation:

$$Y = aW^b$$

where Y - PK parameters, a – allometric coefficient, W – body weight , and b – allometric exponent. The unbound fraction corrected intercept method (FCIM) was used for prediction of human CL/F and AUC values (2):

$$\text{Predicted human drug CL} = 33.35^*(a/Rf_u)^{0.77}$$

where the term "a" is the intercept obtained from the log–log plot of CL vs. body weight and Rf_u is the ratio of unbound fraction in plasma between rats and humans.

Interspecies population PK parameters were estimated also by mixed-effects modeling of plasma NKTR-181 concentration-time data from mice, rats and dogs receiving IV NKTR-181. Unbound NKTR-181 concentration-time data were simultaneously fit with a two-compartment PK model parameterized in clearances and volumes with allometric coefficients and exponents for each term (3) using Monolix 3.1.

References

1: Anesthesiology 2010; A509.
2: Drug Metab Dispos. 2005; 33:1297–1303.
3: J Pharmacokin Biopharm. 1997; 25:149-167.

Results

In Vitro Plasma Protein Binding

NKTR-181 protein binding values for mouse, rat, dog and human plasma, determined using equilibrium dialysis, are presented in Table 1. Binding was low to moderate in all species, with the greatest extent of binding ranging from 33 to 45% in human plasma. There was a trend for binding to decrease with increasing concentration which appeared to be greater in rodent plasma than in dog and human plasma.

Table 1: Mean (±SD) Plasma NKTR-181 Protein Binding in Mice, Rats, Dogs, and Humans (n=3)

Species	0.03 µg/mL	0.3 µg/mL	3 µg/mL
Human	45 ± 8.1	40 ± 6.0	33 ± 4.3
Dog	23 ± 9.0	11 ± 4.6	16 ± 1.0
Rat	32 ± 6.0	22[a]	5.3[a]
Mouse	27 ± 15	8.5 ± 5.5	5.6 ± 3.3

[a] – n=2

IV Pharmacokinetics

Mean plasma NKTR-181 concentration vs. time profiles by specie are presented in Figure 1.

Figure 1: Mean (+SD) Plasma NKTR-181 Concentration-Time Profiles after Administration of Single 1 mg/kg IV Doses to Mice, Rats and Dogs

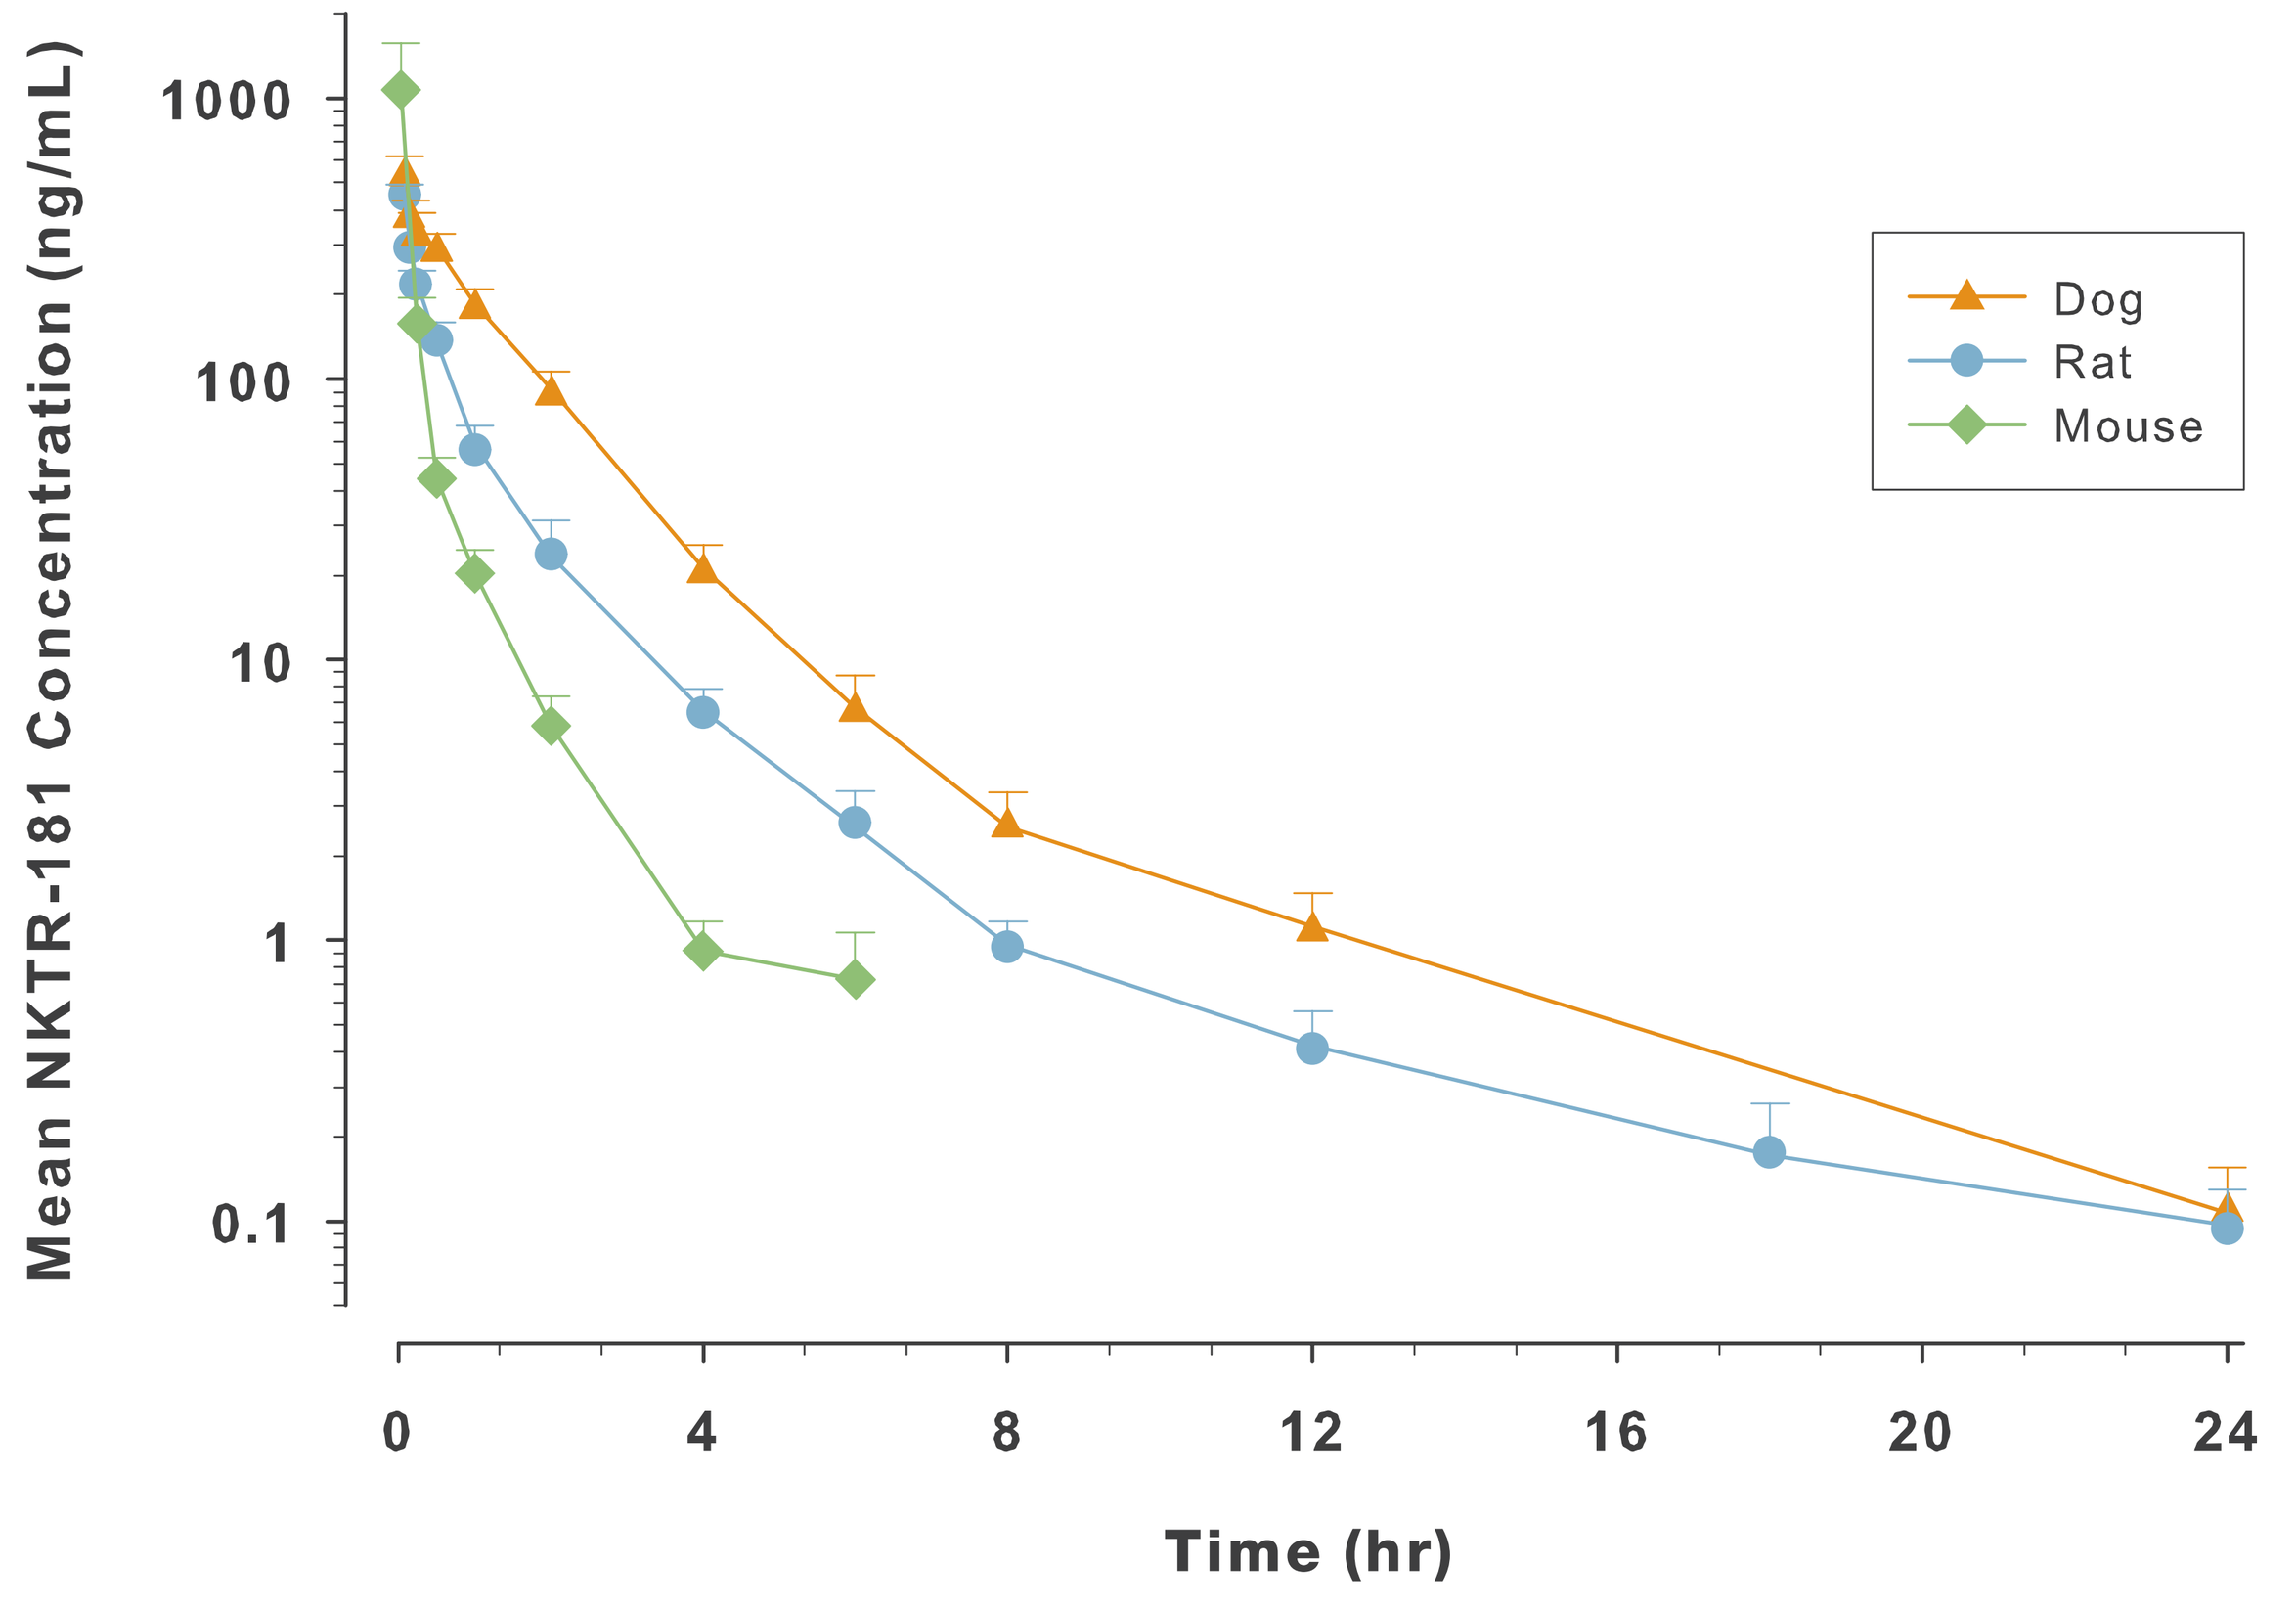
Mean NKTR-181 Concentration (ng/mL)
1000
100
10
1
0.1
0
4
8
12
16
20
24
Time (hr)
Dog
Rat
Mouse

Observed and predicted plasma NKTR-181 unbound concentrations vs. time from the simultaneous fit of data from all animal species using Mixed-Effects Allometric Scaling are presented in Figure 2.

Figure 2: Simultaneous Fit of Plasma NKTR-181 Unbound Concentration-Time Data After IV Administration to Mice, Rats, and Dogs with a Mixed-Effects Allometric Scaling Model

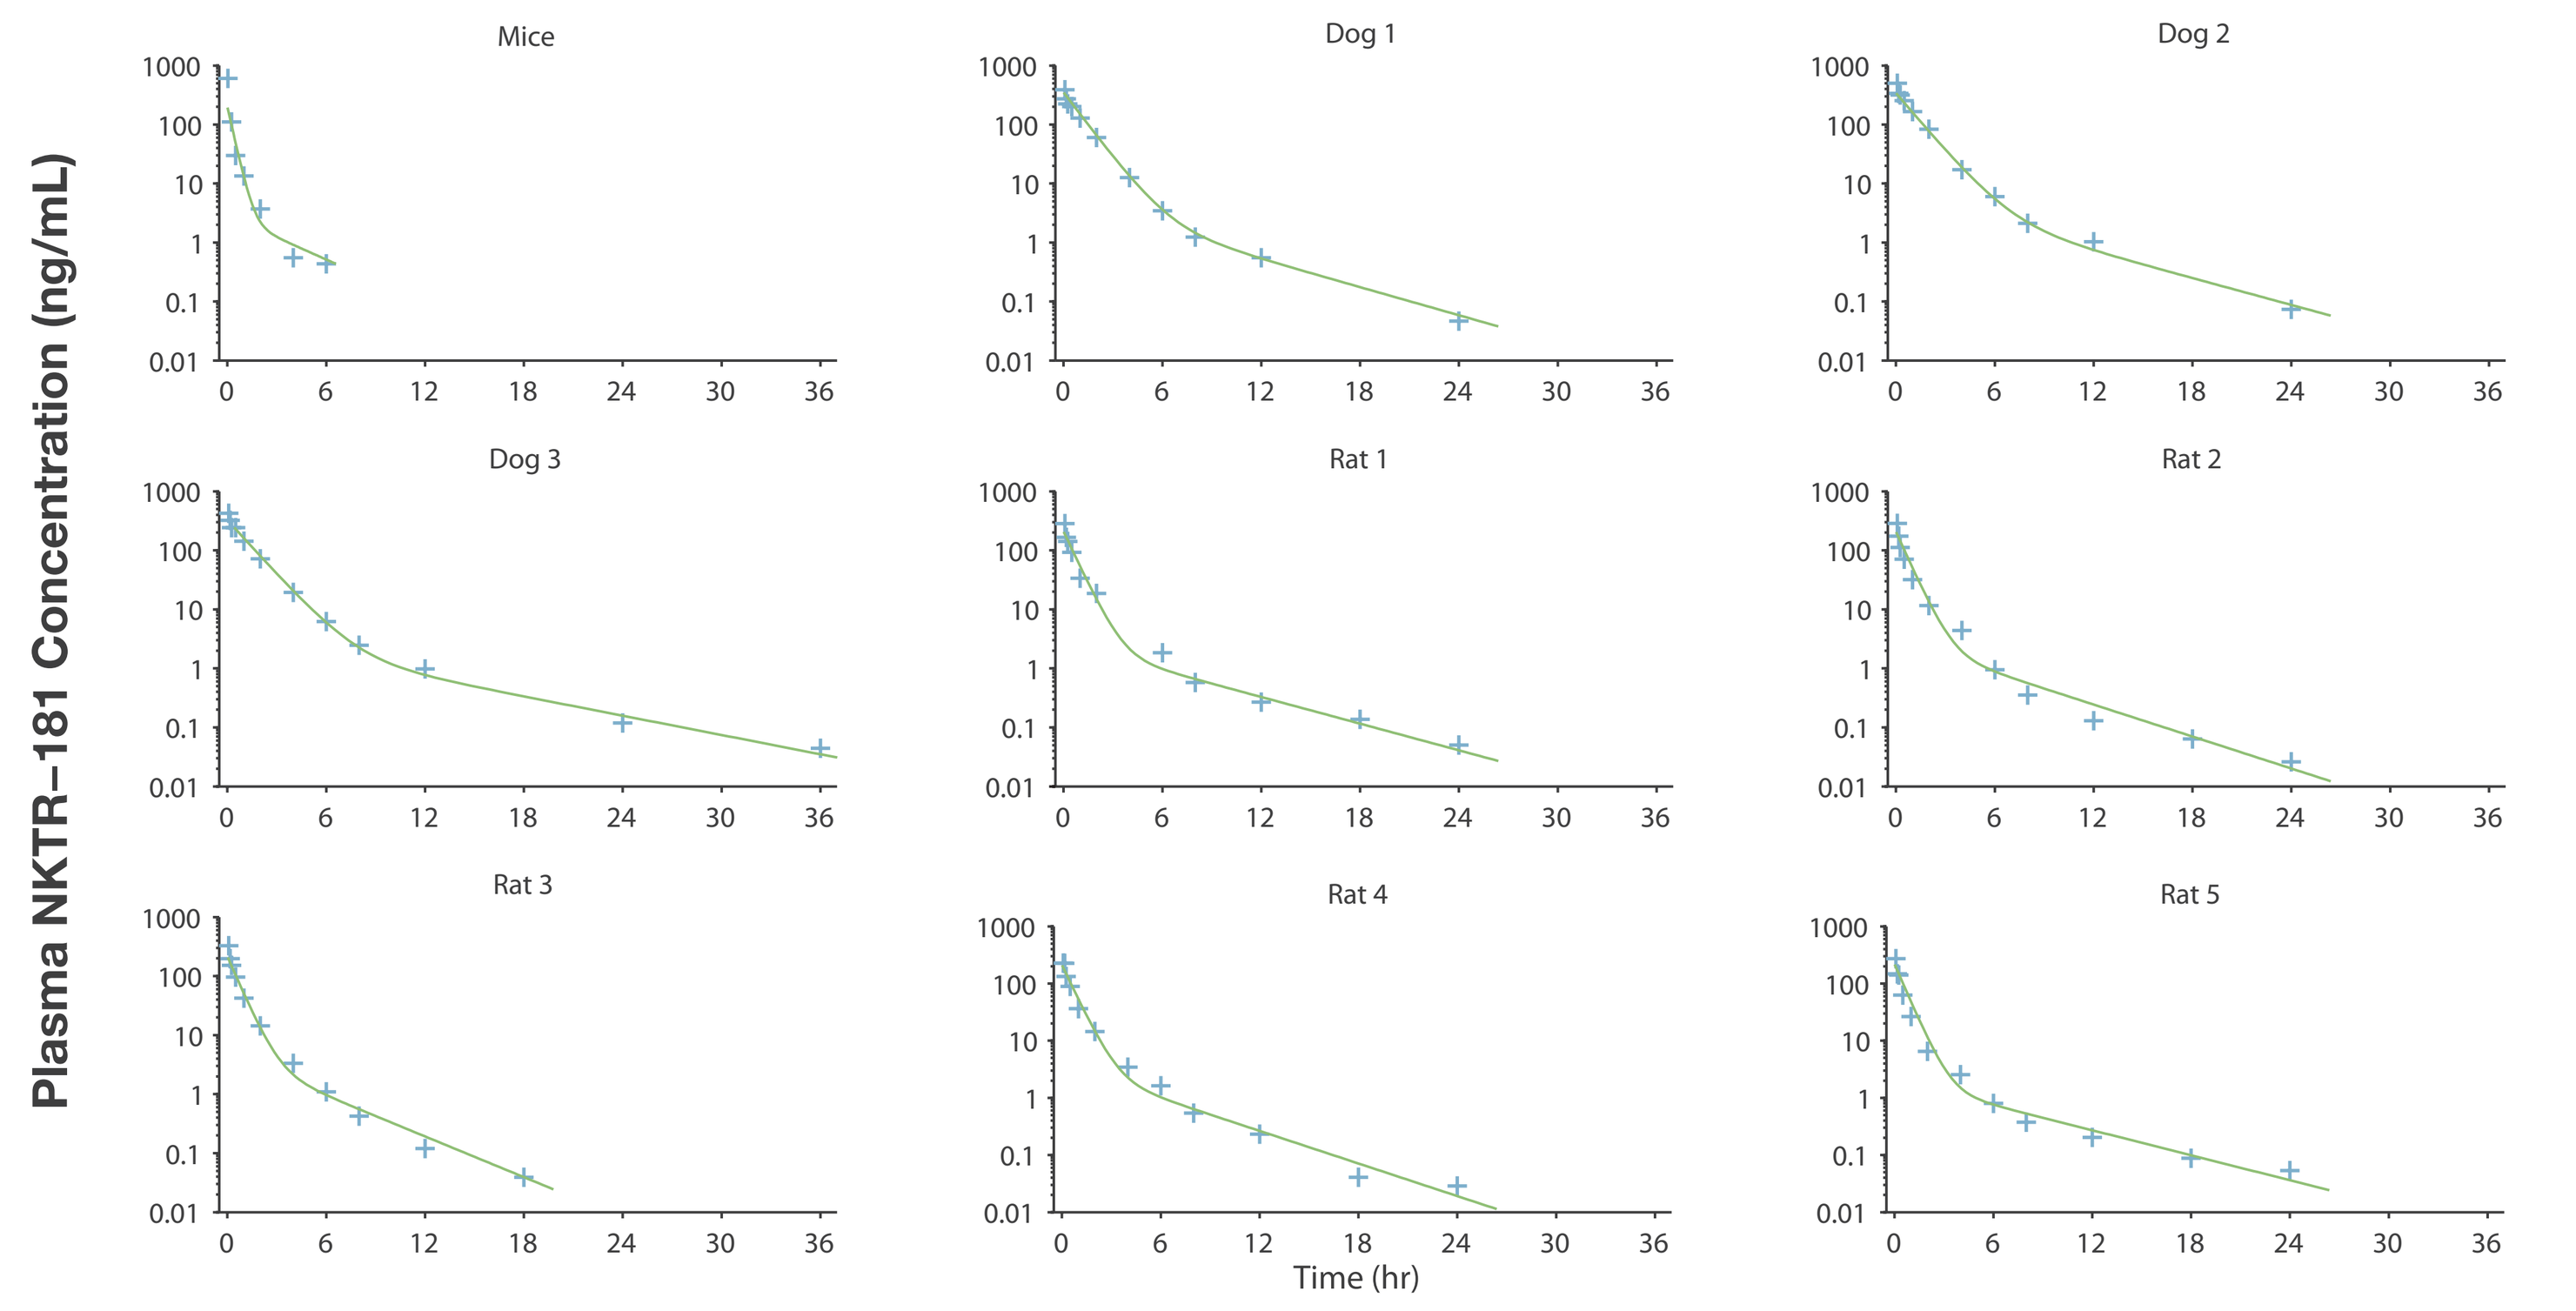
Plasma NKTR-181 Concentration (ng/mL)
Mice
Dog 1
Dog 2
Dog 3
Rat 1
Rat 2
Rat 3
Rat 4
Rat 5
Time (hr)

PO Pharmacokinetics

Mean dose-normalized plasma NKTR-181 concentration vs. time profiles after oral administration of 5 mg/kg NKTR-181 to mice, rats, and dogs, and 4.6 mg/kg to humans (320 mg fixed dose) are presented in Figure 3. Assessment of dose proportionality in mice, rats, dogs, and humans is presented in Figure 4.

Figure 3: Mean (+SD) Dose-Normalized Plasma NKTR-181 Concentration-Time Profiles after Administration of Single Oral ~5 mg/kg Doses to Mice, Rats, Dogs, and Humans

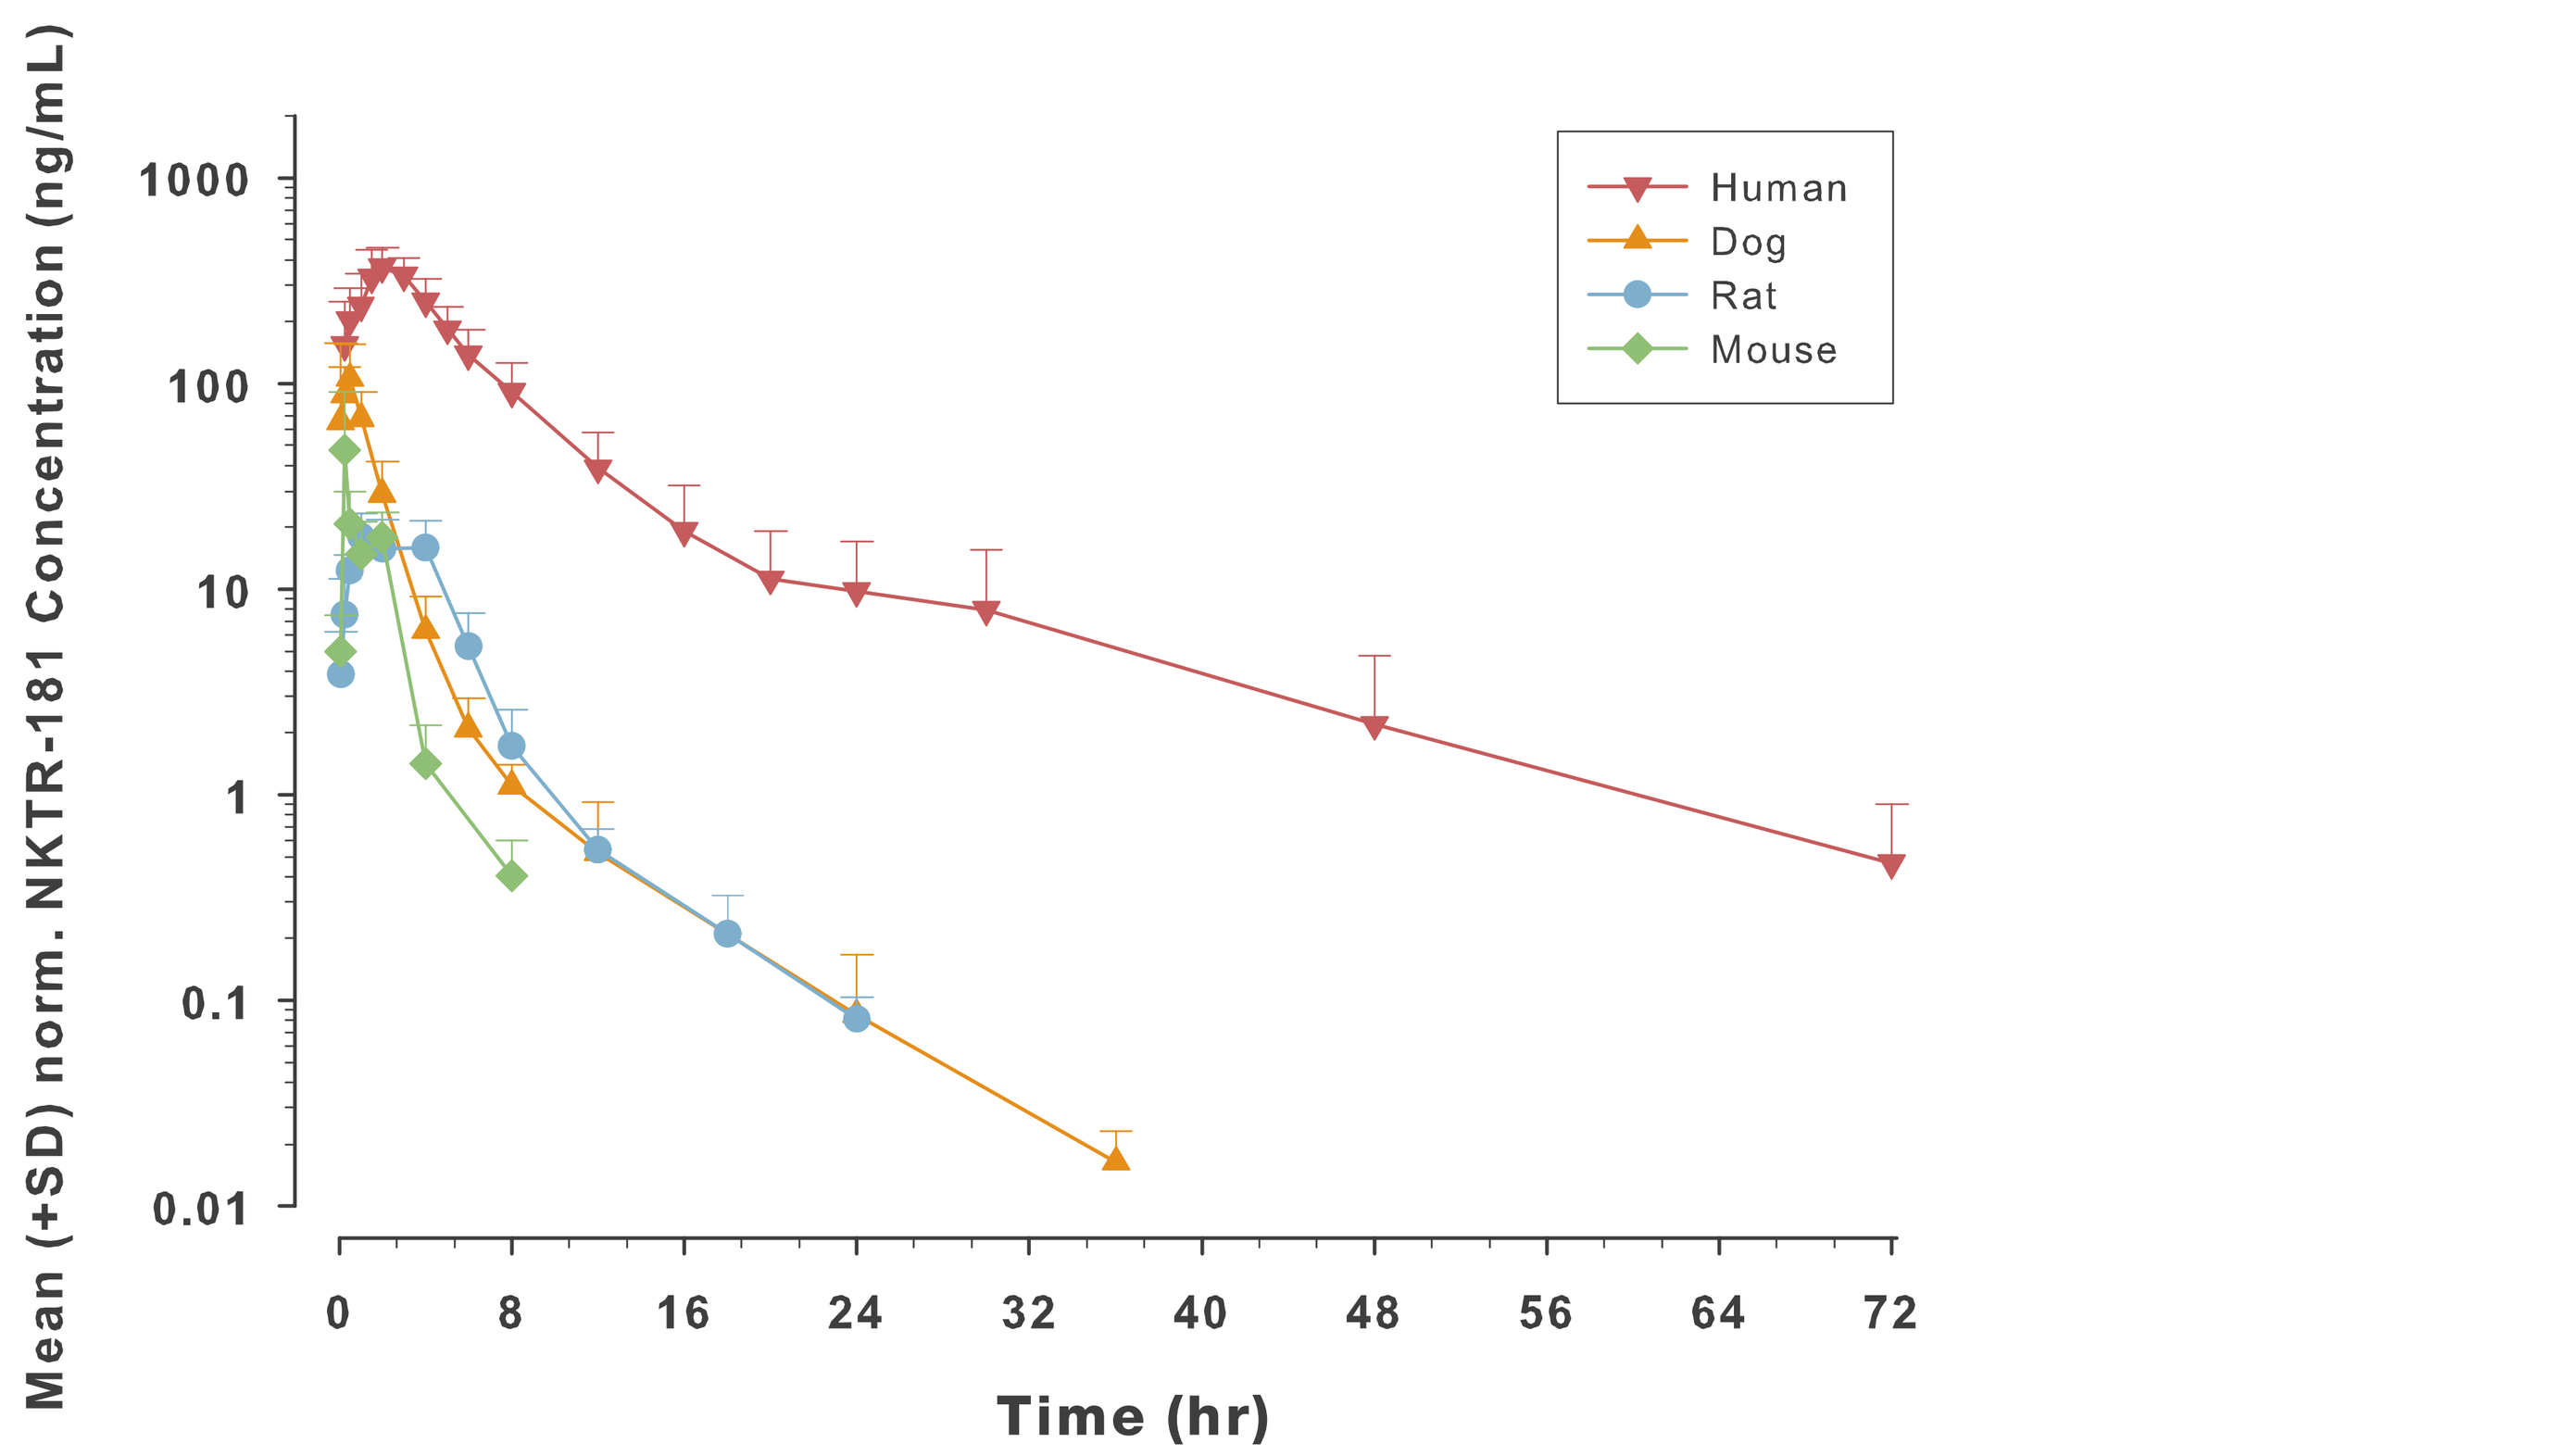
Mean (+SD) norm. NKTR-181 Concentration (ng/mL)
1000
100
10
1
0.1
0.01
0
8
16
24
32
40
48
56
64
72
Time (hr)
Human
Dog
Rat
Mouse

NKTR-181 was rapidly absorbed with absolute oral bioavailability in mice, rats, and dogs of about 30% at pharmacologic doses, and eliminated by mechanisms displaying first-order kinetics (Figure 3).

Figure 4: Mean (±SD) Dose Normalized Plasma NKTR-181 AUC_{inf} Values after Oral Administration of NKTR-181 to Mice, Rats, Dogs, and Humans

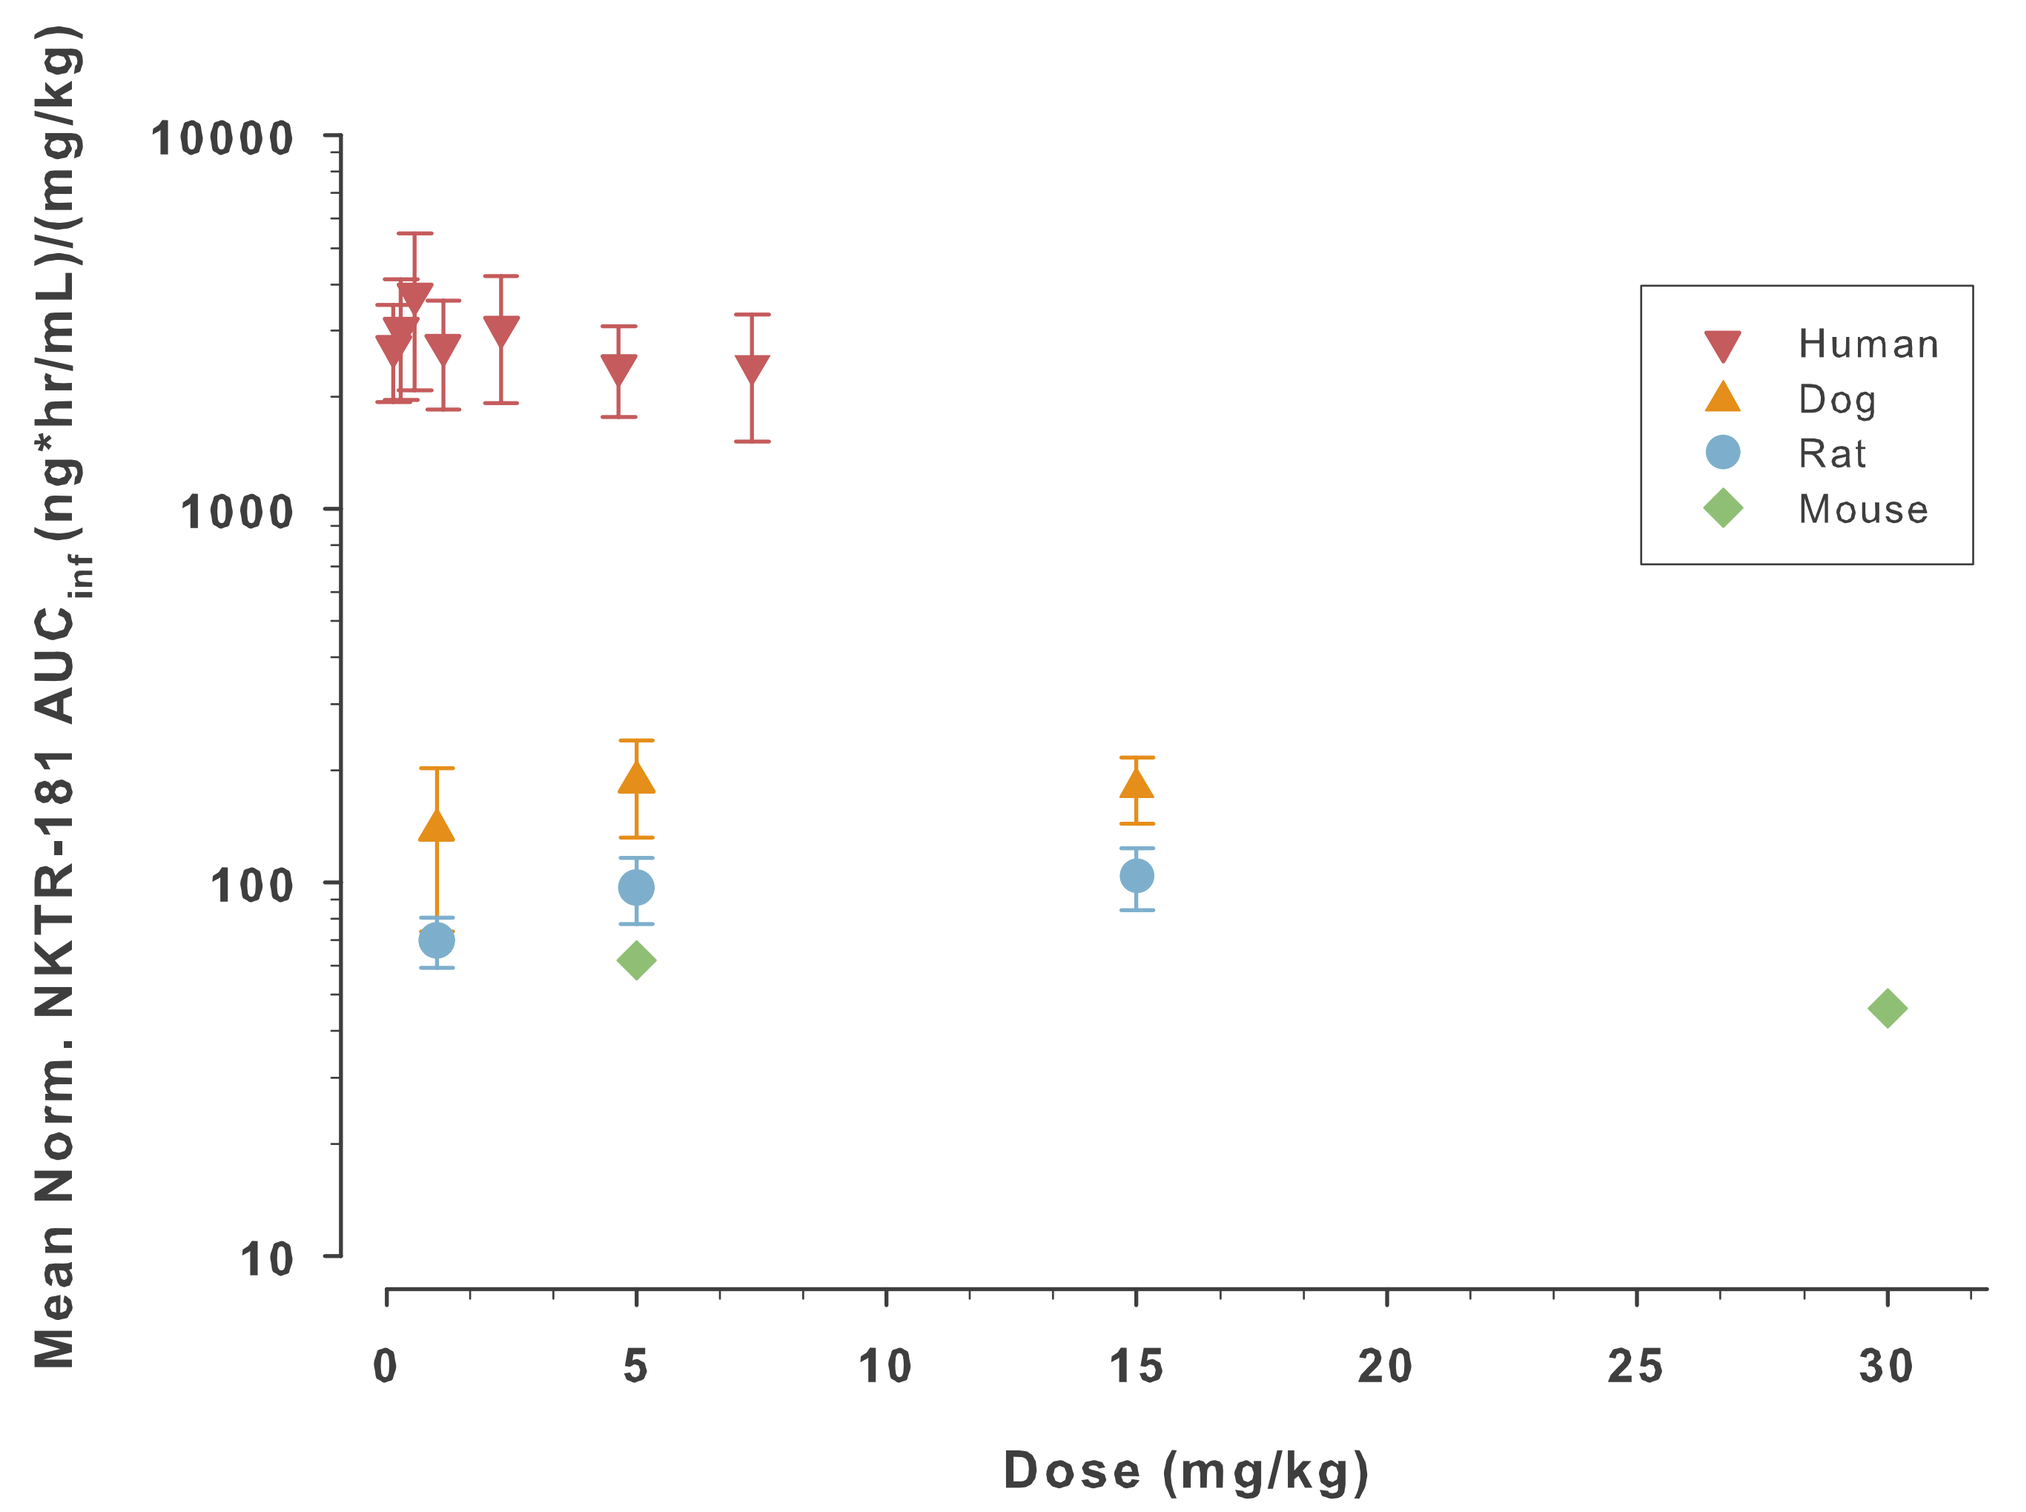
Mean Norm. NKTR-181 AUCinf (ng*hr/mL)/(mg/kg)
10000
1000
100
10
0
5
10
15
20
25
30
Dose (mg/kg)
Human
Dog
Rat
Mouse

Systemic exposure increased proportionally with dose at doses in the range associated with antinociception (Figure 4). In the single ascending dose FIH study, NKTR-181 T_{max} was 2-4 hours with an average half-life of approximately 12 hours. Dose proportional exposure was obtained over a wide dose range (10 to 500 mg), similar to that obtained in preclinical species.

Figure 5. Allometric Scaling Plots for Clearance (CL) and Volume at Central Compartment (V1) (left) and Predicting of Human CL/F (right)

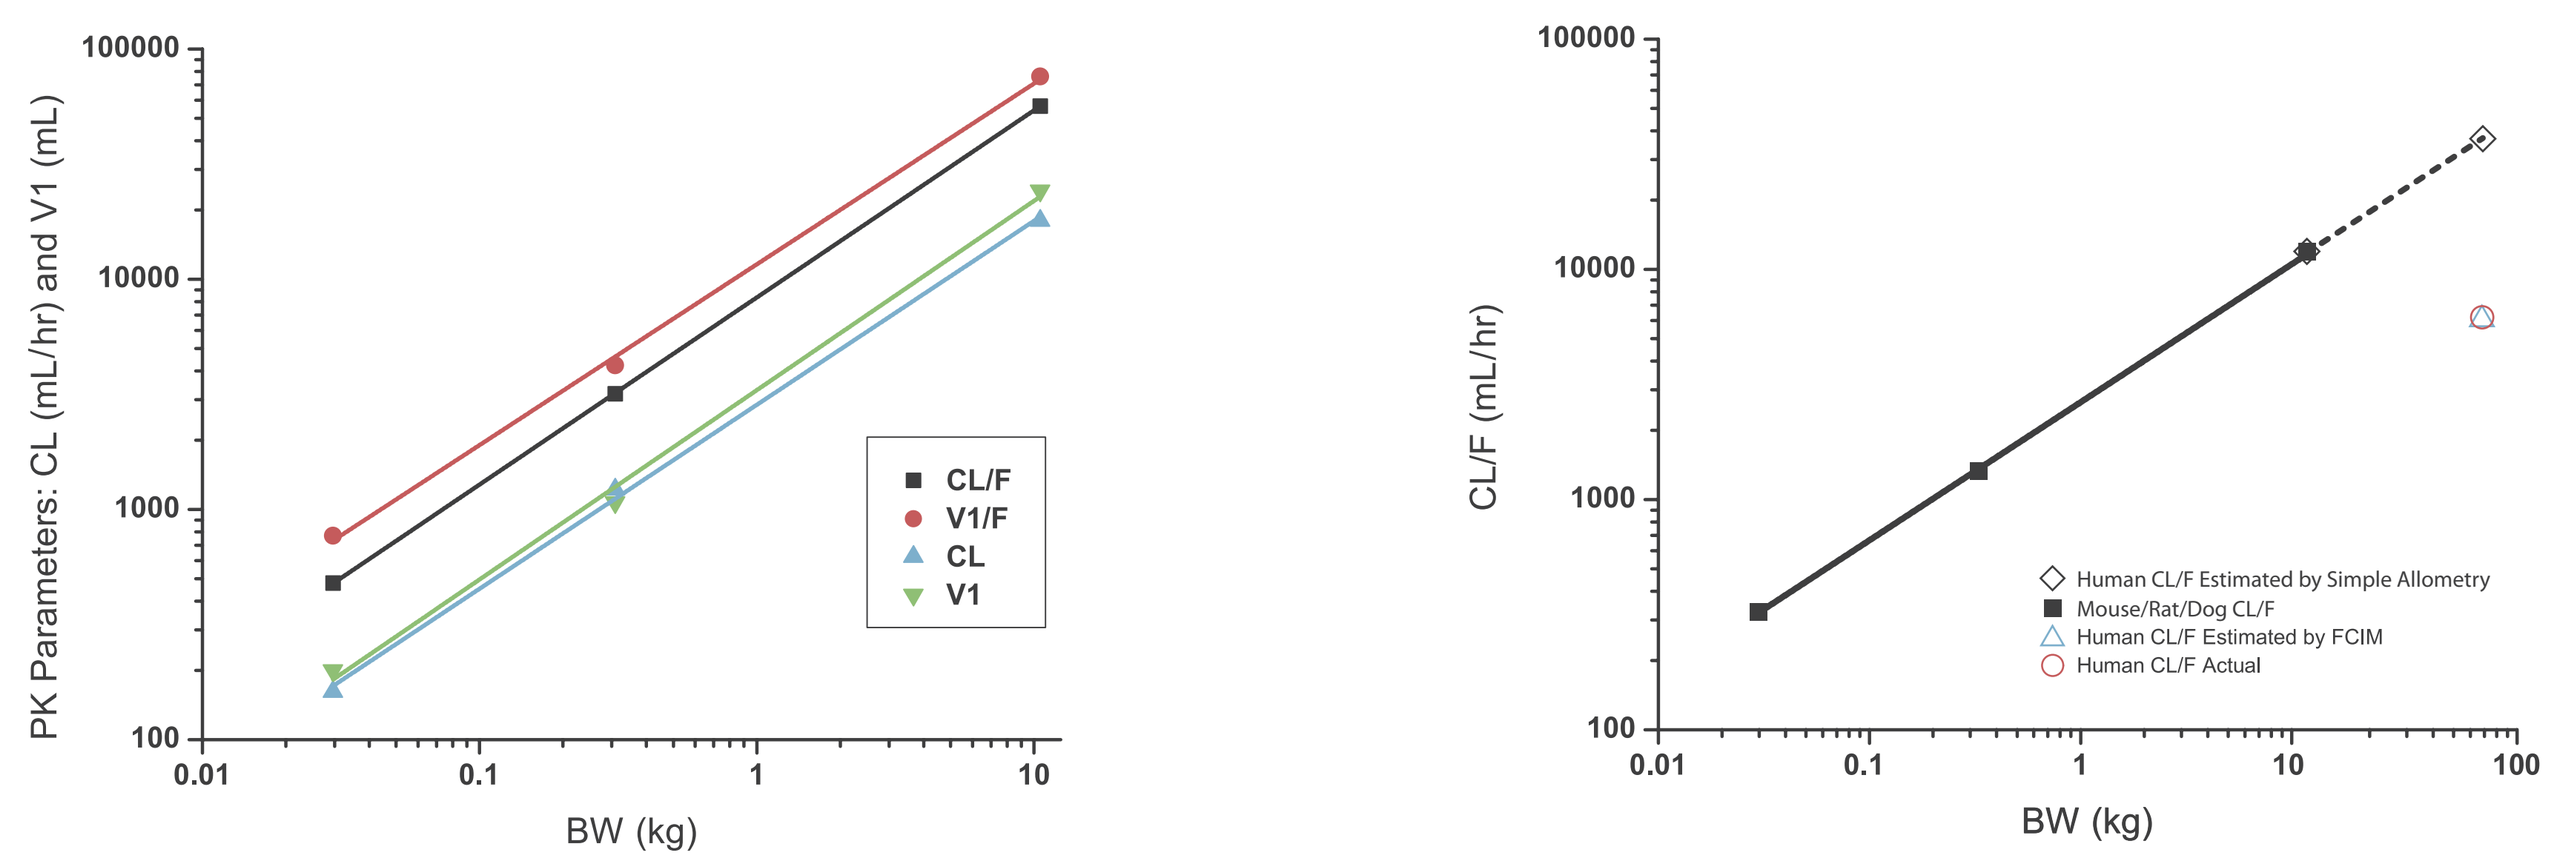
PK Parameters: CL (mL/hr) and V1 (mL)
BW (kg)
CL/F
V1/F
CL
V1
CL/F (mL/hr)
Human CL/F Estimated by Simple Allometry
Mouse/Rat/Dog CL/F
Human CL/F Estimated by FCIM
Human CL/F Actual

Simple allometric scaling of NKTR-181 CL and V1 values after IV administration as well as CL/F and V1/F after PO administration across mice, rats and dogs showed excellent correlation, but actual CL/F values in humans were much lower than values predicted from simple allometry (Figure 5). However, three-species scaling using the FCIM provided accurate prediction of CL/F and dose normalized AUC values in humans: predicted and observed (mean across all dose range) CL/F and dose normalized AUC values were 24.1 vs. 24.7 (range 18.6 to 28.5) L/hr and 41.5 vs. 41.3 (range 35.1 to 53.9) ng*hr/mL/mg, respectively.

Conclusions

- NKTR-181 exhibits dose-linear pharmacokinetics and good oral bioavailability in mice, rats, dogs, and humans.
- Results of the FIH study confirmed allometrically predicted CL/F and AUC.
- The favorable pharmacokinetic characteristics of NKTR-181 support its progression into Phase 2 clinical trials.

Presented at the American Association of Pharmaceutical Scientists Annual Meeting; October 14–18, 2012; Chicago, IL.