Abstract # C209: Strong Synergistic Activity of NKTR-102 – Pegylated Liposomal Doxorubicin (PLD) Combination Therapy in a Nonclinical Model of Platinum Resistant A2780 Human Ovarian Cancer

Ute Hoch and Michael A. Eldon, Nektar Therapeutics, San Francisco, CA

NEKTAR

Conclusions

- Single agent NKTR-102 shows superior activity over single agent PLD in the A2780 ovarian tumor model, with a 100% complete response rate at both doses tested
- NKTR-102 and PLD in combination showed marked synergism demonstrated by complete response in all animals without tumor regrowth in 93% of animals
- NKTR-102 can be combined with PLD without additive toxicity
- These preclinical results support further study of NKTR-102 in combination with PLD in ovarian cancer

Abstract

Purpose: NKTR-102, a next generation topoisomerase I inhibitor, has been demonstrated to be superior to cisplatin, carboplatin and irinotecan in a mouse model of platinum-resistant A2780 Human Ovarian Cancer (Abstract 8015, ECCO 15-ESMO 34, 22 SEP 09). NKTR-102 as a single agent in women with platinum resistant/refractory ovarian cancer who have failed pegylated liposomal doxorubicin (PLD) therapy has demonstrated a confirmed ORR of 20% (RECIST, n = 30) to 36% (CGIC, n = 33) (JCO Vol 29, No 15_suppl, 2011: 5047). As a single agent, NKTR-102 appears highly active in the defined population, however, we hypothesize that the combination of NKTR-102 and PLD could result in synergistic activity through concurrent, long lasting inhibition of Topo I by NKTR-102 and Topo II by PLD, resulting in greater efficacy than observed as sequential single agents. This report is a presentation of the results of a study of NKTR-102 as a single agent, PLD as a single agent, and a combination of NKTR-102 and PLD in mice bearing A2780 Human Ovarian tumors that are minimally responsive to cisplatin. **Methods:** The MTD of PLD in mice was determined to be 10 mg/kg q7dx2 in a pilot study. NKTR-102 doses were limited to 150 mg/kg due to viscosity and dose volume limitations rather than tolerability. Mice bearing A2780 ovarian tumors (~ 100 mm3) received either vehicle, PLD, NKTR-102, or a combination of NKTR-102 and PLD as follows: 5 or 10 mg/kg PLD on days 1 and 8; 100 or 150 mg/kg NKTR-102 on day 1; or 5/100, 10/100, 5/150, or 10/150 mg/kg PLD on days 1 and 8/NKTR-102 on day 1 (n = 10/treatment). Anti-tumor efficacy was evaluated based on tumor growth delay (TGD) and response rate. Results: Control tumors grew rapidly and uniformly to the 2000 mm3 endpoint in a median of 12 days. PLD administered at 5 and 10 mg/kg q7dx2 resulted in TGD of 7 and 24 days, respectively, with one partial response at the highest dose. Tumor growth was slower following PLD, but was continuous in all animals. NKTR-102 at single doses of 100 and 150 mg/kg resulted in TGDs of 32 and 38 days, respectively, with 100% (10 of 10) complete responses in each group. Following complete tumor regression in all animals, growth resumed after approximately 25 and 35 days in the 100 and 150 mg/kg treatment groups, respectively. TGD was 57 days (maximum possible value; 474% TGD) following any combination of NKTR-102 and PLD and all mice achieved complete response. There was no tumor regrowth within the 69-day observation period in 37 of 40 animals (93%). There was no accentuated weight loss in any dose group, indicating that the combination treatments were tolerated as well as single agent PLD. **Conclusions:** NKTR-102 as a single agent shows superior activity compared to PLD as a single agent in the A2780 ovarian tumor model, with a 100% complete response rate at all doses. NKTR-102 and PLD in combination showed marked synergism demonstrated by complete response in all animals with no tumor regrowth in 93%. Nektar intends to conduct a Phase I study of NKTR-102 with PLD in patients with advanced/metastatic refractory solid tumors, including those with platinum-resistant ovarian cancer.

Background

- Nektar's proprietary polymer conjugation technology to improve the pharmacokinetics of irinotecan and its active metabolite SN38.

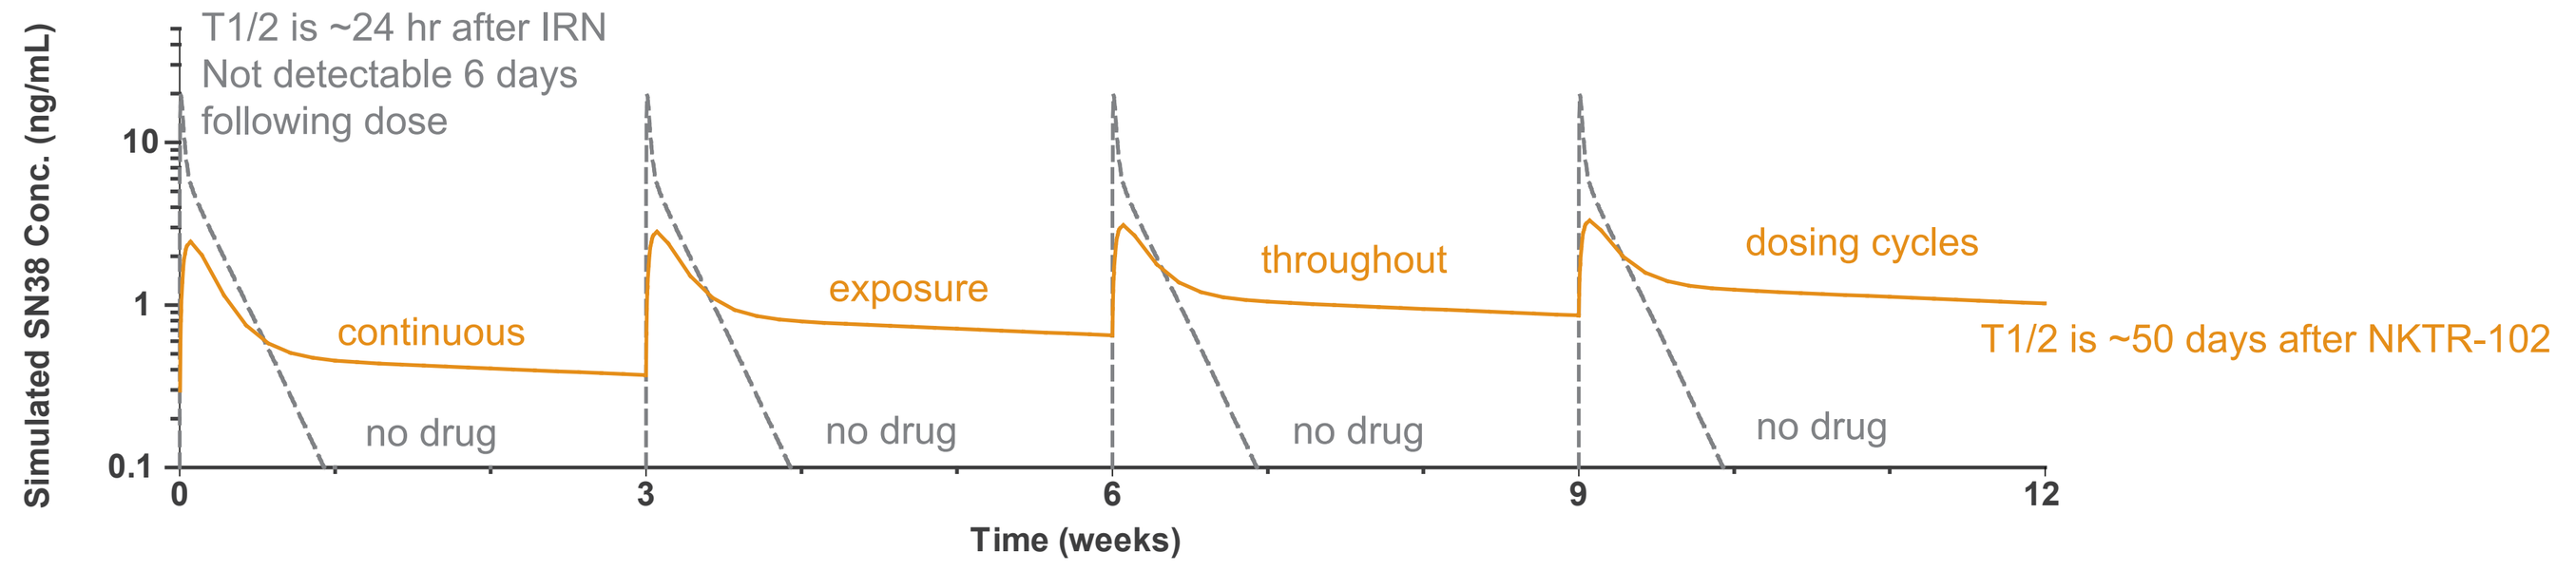
T1/2 is ~24 hr after IRN
Not detectable 6 days following dose
Simulated SN38 Conc. (ng/mL)
continuous
exposure
throughout
dosing cycles
T1/2 is ~50 days after NKTR-102
no drug
Time (weeks)

- NKTR-102 has superior activity compared to irinotecan in a wide range of mouse models of human xenograft tumors.

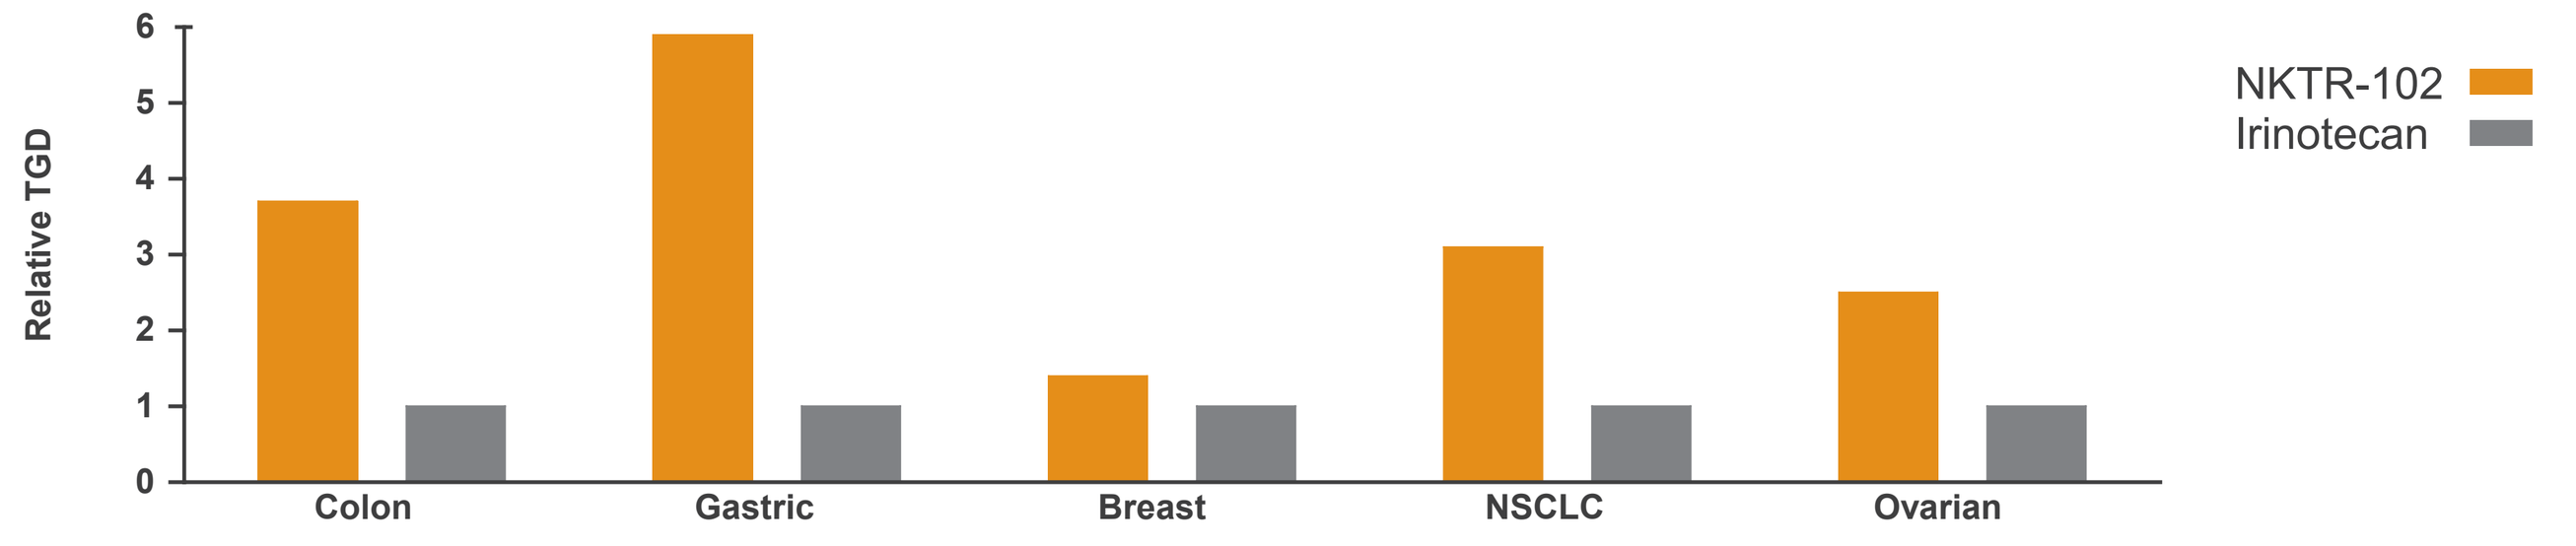
Relative TGD
NKTR-102
Irinotecan
Colon
Gastric
Breast
NSCLC
Ovarian

- NKTR-102 demonstrated high anti-tumor activity in a range of tumors in Phase I (11% confirmed PRs; ENA 2008, abs 595).
- NKTR-102 showed a 22% confirmed response rate per RECIST in heavily pre-treated women with platinum resistant/refractory ovarian cancer (ASCO 2010, abs 5013), and a 29% confirmed response rate in 2nd/3rd line patients with advanced breast cancer (SABCS 2010, abs S6-6).

Objective

To investigate the nonclinical anti-tumor activity of NKTR-102 (topoisomerase I inhibitor) in combination with PLD (topoisomerase II inhibitor) in the platinum resistant A2780 ovarian tumor model.

Study Design

Tumor cells were cultured and implanted subcutaneously in 9-10 week old female athymic nude mice. Animals were randomized into treatment groups (n=10/group) when their tumors reached 75-144 mm³ and subsequently received either vehicle, NKTR-102, PLD, or combinations of NKTR-102 with PLD as shown in the Figure below. Animals were weighed and monitored twice weekly, and tumor volumes were measured until an endpoint (2000 mm³ or Day 76) was met. Efficacy was measured by tumor growth delay (TGD) and regression response rate. To qualify as partial regression (PR), the tumor volume had to be 50% or less of its Day 1 volume for three consecutive measurements. To qualify as complete regression (CR), the tumor volume had to be less than 13.5 mm³ for three consecutive measurements. An animal with a CR at the termination of the study was classified as a tumor-free survivor (TFS).

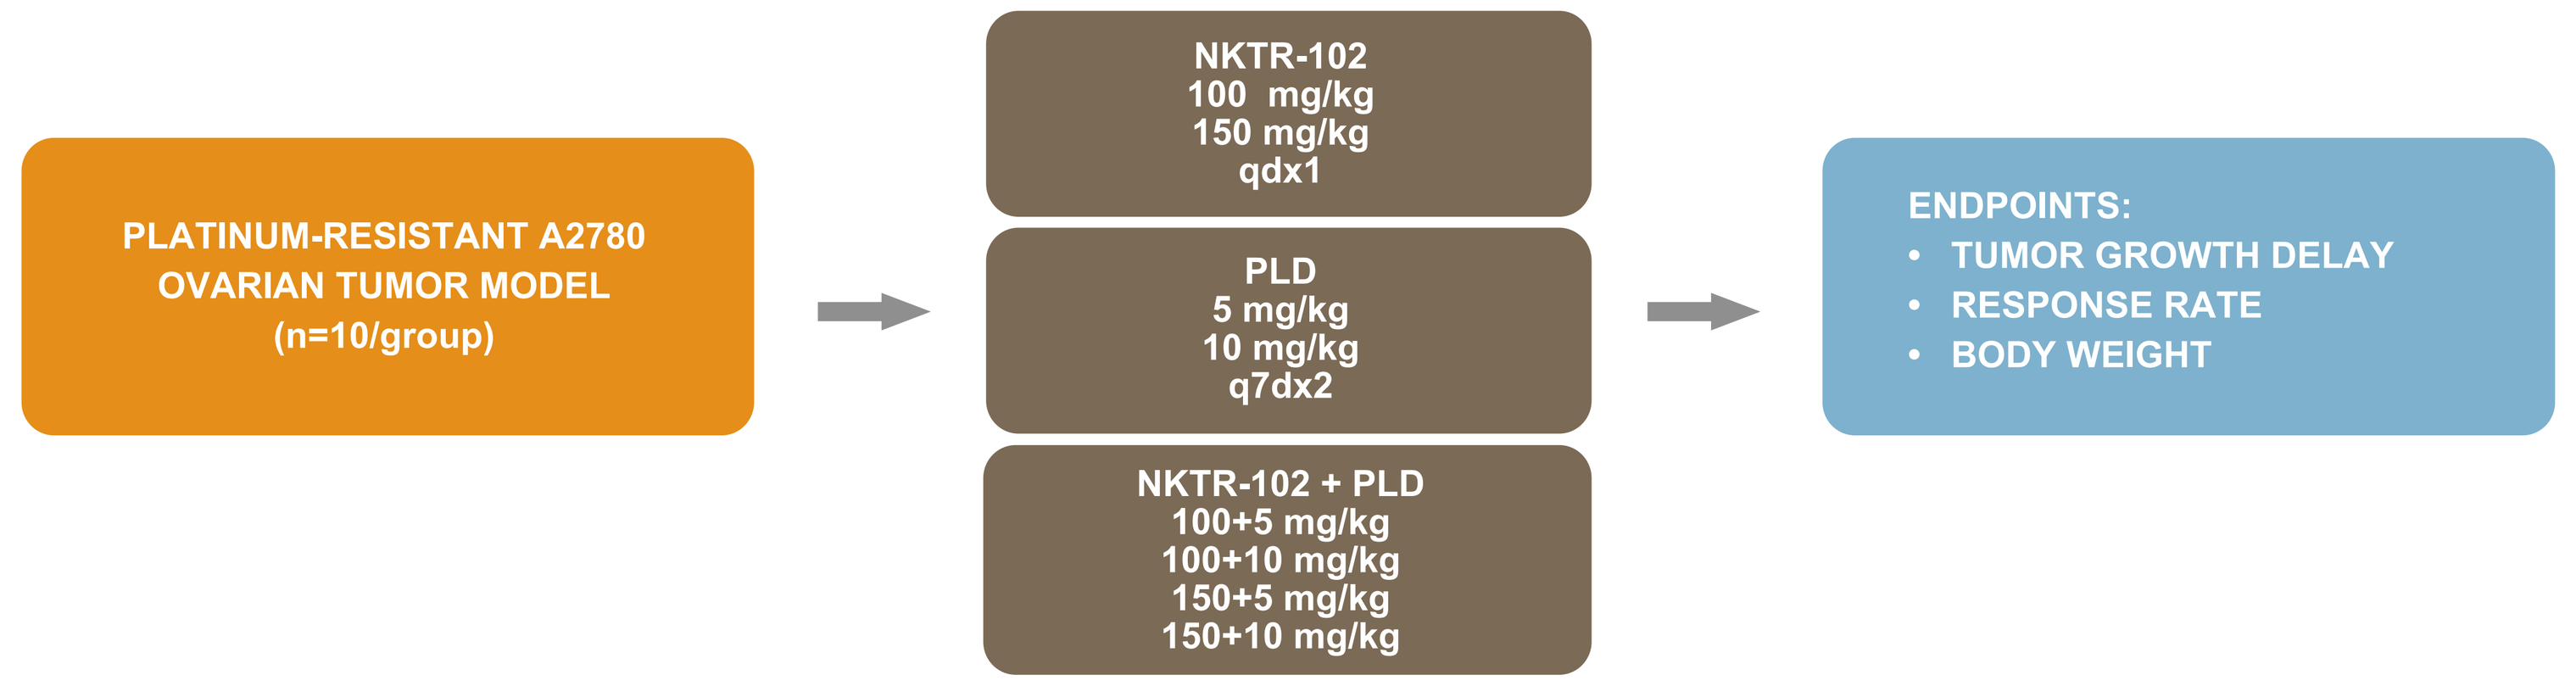
PLATINUM-RESISTANT A2780 OVARIAN TUMOR MODEL (n=10/group)
NKTR-102 100 mg/kg 150 mg/kg qdx1
PLD 5 mg/kg 10 mg/kg q7dx2
NKTR-102 + PLD 100+5 mg/kg 100+10 mg/kg 150+5 mg/kg 150+10 mg/kg
ENDPOINTS:
• TUMOR GROWTH DELAY
• RESPONSE RATE
• BODY WEIGHT

Results

NKTR-102 with PLD Shows Superior Activity Over Either Single Agent in A2780 Ovarian Tumor Model

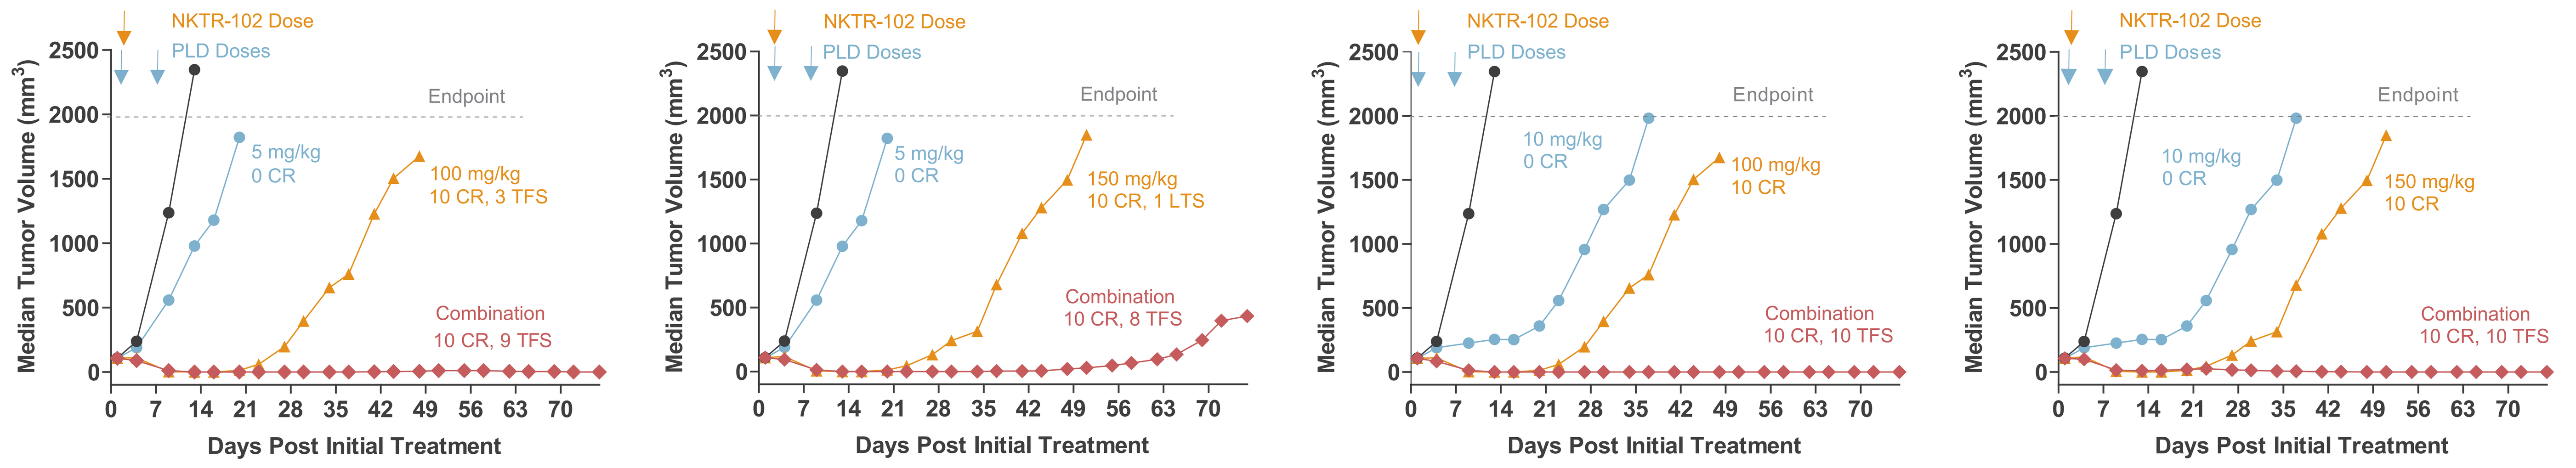
NKTR-102 Dose
PLD Doses
Endpoint
Median Tumor Volume (mm³)
Days Post Initial Treatment
5 mg/kg 0 CR
100 mg/kg 10 CR, 3 TFS
Combination 10 CR, 9 TFS
5 mg/kg 0 CR
150 mg/kg 10 CR, 1 LTS
Combination 10 CR, 8 TFS
10 mg/kg 0 CR
100 mg/kg 10 CR
Combination 10 CR, 10 TFS
10 mg/kg 0 CR
150 mg/kg 10 CR
Combination 10 CR, 10 TFS

Tolerability/Toxicity

- All treatments were acceptably tolerated
- Maximum mean body weight losses were minimal or zero for single agent NKTR-102
- Maximum mean body weight losses were less than 10% for single agent PLD
- Combination of NKTR-102 and PLD did not increase the body weight losses over single agents
- No remarkable clinical observations

Individual Times Until An Endpoint (Tumor Volume of 2000 mm³ or Day 76) Was Met

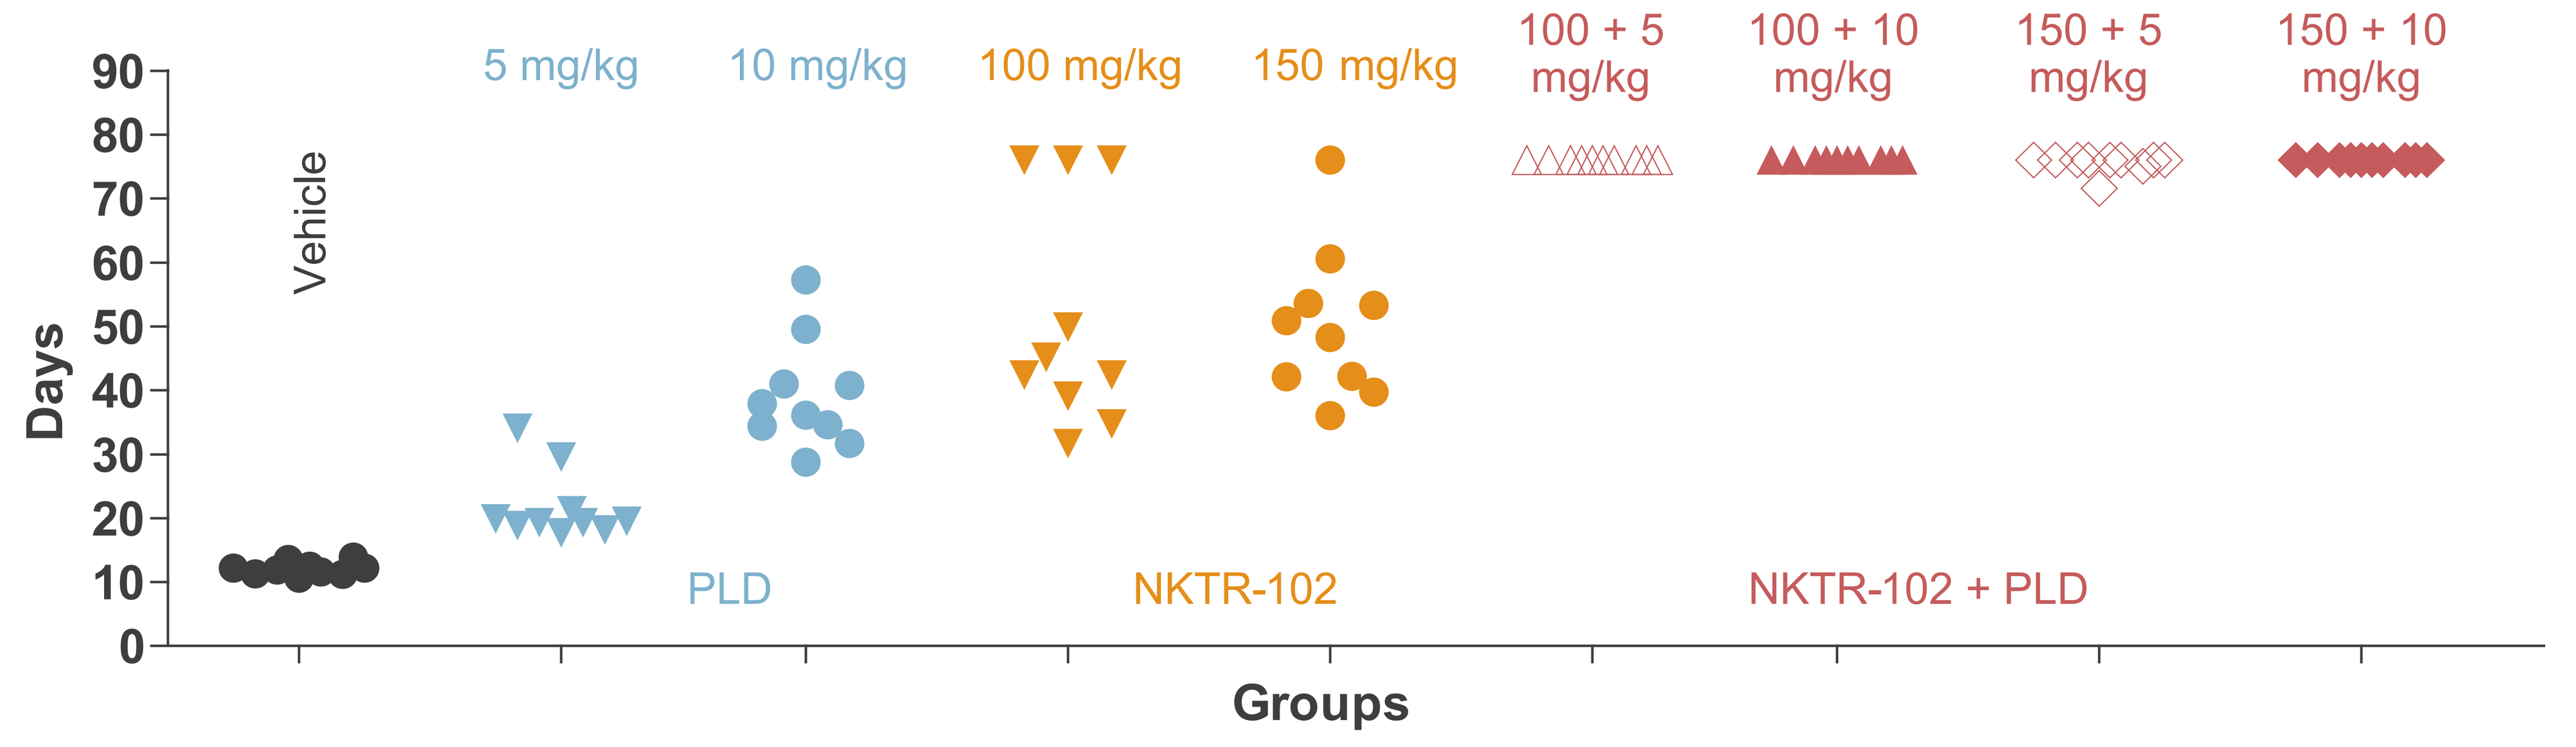
Days
Vehicle
5 mg/kg
10 mg/kg
100 mg/kg
150 mg/kg
100 + 5 mg/kg
100 + 10 mg/kg
150 + 5 mg/kg
150 + 10 mg/kg
PLD
NKTR-102
NKTR-102 + PLD
Groups

Response Summary

Treatment Regimen: Agent	mg/kg	Median TTE*	T-C (days)	TGD (%)	Regressions: PR	CR	TFS	Mean BW Nadir	Statistical Results For Groups Tested
vehicle	---	12	---	---	0	0	0	---	
PLD	5	19	7.4	62	0	0	0	-1%, Day 9	Significant against vehicle (p<0.001)
PLD	10	36	24	201	1	0	0	-9%, Day76	Significant against vehicle (p<0.001)
102	100	44	32	265	0	10	3	0.5%, Day2	Significant against vehicle (p<0.001)
102	150	50	38	313	0	10	1	0.1%, Day 4	Significant against vehicle (p<0.001)
102 + PLD	100+5	76	64	533	0	10	9	-5%, Day 3	Significant against vehicle (p<0.001), 100 mg/kg NKTR-102 (p<0.01), 5 mg/kg PLD (p<0.001)
102 + PLD	100+10	76	64	533	0	10	10	-10%, Day 76	Significant against vehicle (p<0.001), 100 mg/kg NKTR-102 (p<0.01), 10 mg/kg PLD (p<0.001)
102 + PLD	150+5	76	64	533	0	10	8	-4%, Day 9	Significant against vehicle (p<0.001), 150 mg/kg NKTR-102 (p<0.001), 5 mg/kg PLD (p<0.001)
102 + PLD	150+10	76	64	533	0	10	10	-7%, Day 9	Significant against vehicle (p<0.001), 150 mg/kg NKTR-102 (p<0.001), 10 mg/kg PLD (p<0.001)

*Abbreviations: TTE: time to endpoint; T-C: treatment vs control group (vehicle); TGD: tumor growth delay; PR: partial regression; CR: complete regression; TFS: tumor free survivor; BW: body weight

Poster C209, 2011 AACR-EORTC-NCI Molecular Targets and Cancer Therapeutics Conference in San Francisco
To download a copy of this poster, please visit http://www.nektar.com/product_pipeline/oncology_nktr-102.html

© 2011 Nektar Therapeutics, all rights reserved.

Acknowledgment:
We would like to thank Piedmont Research Center for conducting the xenograft study.