Significant Antitumor Activity in a Randomized Phase 2 Study Comparing Two Schedules of NKTR-102 in Patients With Pre-Treated Metastatic Breast Cancer

Authors: Ahmad Awada[1], Stephan Chan[2], Guy Jerusalem[3], Robert E Coleman[4], Manon Huizing[5], Aminder Mehdi[6], Susan M. O'Reilly[7], John T. Hamm[8], Peter J. Barrett-Lee[9], Veronique Cocquyt[10], Konstantine Sideras[11], David E. Young[12], Mollie Brown[13], Carol Zhao[13], Alison L. Hannah[13], Lorianne K. Masuoka[13], Agustin A. Garcia[14], Edith A. Perez[15], on behalf of the NKTR-102 Study Group
Institutions: [1]Institut Jules Bordet, Brussels, Belgium; [2]Nottingham City Hospital, Nottingham, UK; [3]Domaine Universitaire du Sart Tilman, Liege, Belgium; [4]Weston Park Hospital, Sheffield, UK; [5]UZ Antwerpen, Antwerp, Belgium [6]Stockton Hematology/Oncology, Stockton, CA; [7]Clatterbridge Centre for Oncology, Bebington, UK; [8]Louisville Oncology Research, Louisville, KY; [9]Velindre NHS Trust Cardiff, Cardiff, UK; [10]Gent University Hospital, Gent, Belgium; [11]Mayo Clinic, Rochester, MN; [12]Desert Hematology/Oncology, Rancho Mirage, CA; [13]Nektar Therapeutics, San Francisco, CA; [14]USC Norris Comprehensive Cancer Center, Los Angeles, CA; [15]Mayo Clinic, Jacksonville, FL; and the NKTR-102 Study Group.

Background

- NKTR-102 is a topoisomerase 1 inhibitor-polymer conjugate with reduced peak concentrations and a continuous exposure profile
- NKTR-102 has superior efficacy (measured both by tumor growth delay and regression rate) compared to irinotecan against a wide range of human xenograft tumors.[1]

MCF-7 Breast Tumor Model

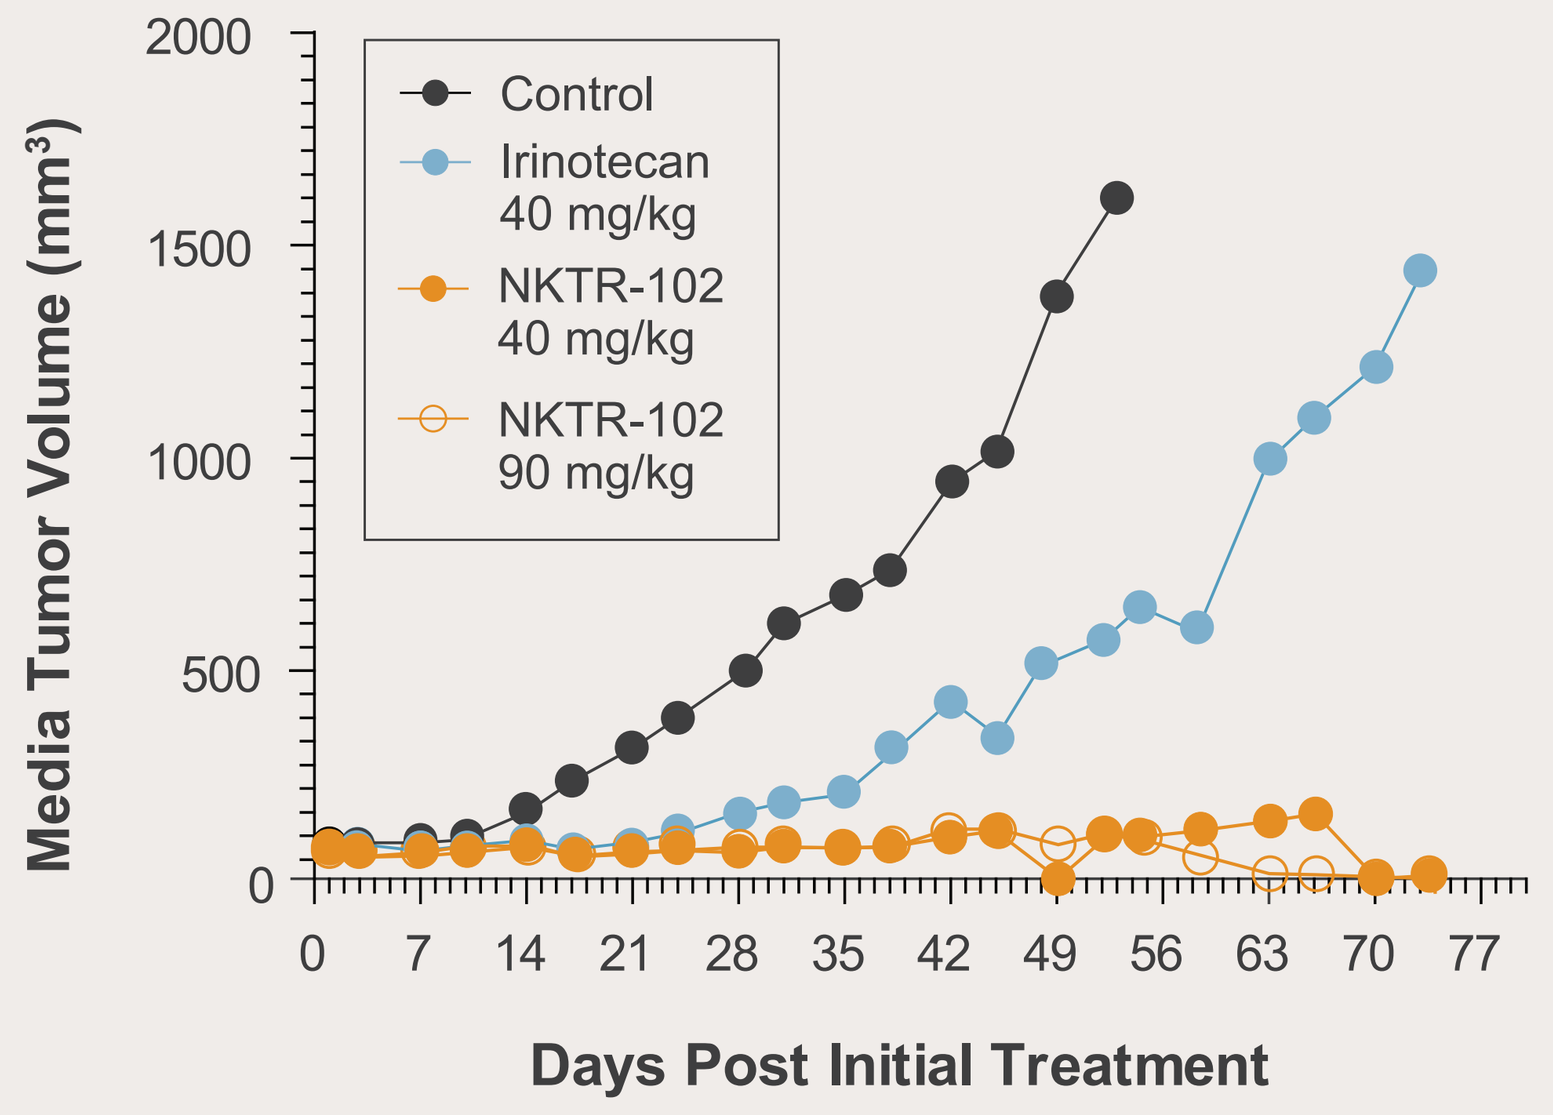
Control
Irinotecan 40 mg/kg
NKTR-102 40 mg/kg
NKTR-102 90 mg/kg
Media Tumor Volume (mm³)
2000
1500
1000
500
0
0 7 14 21 28 35 42 49 56 63 70 77
Days Post Initial Treatment

Polymer Conjugation Improves Pharmacokinetics of Irinotecan

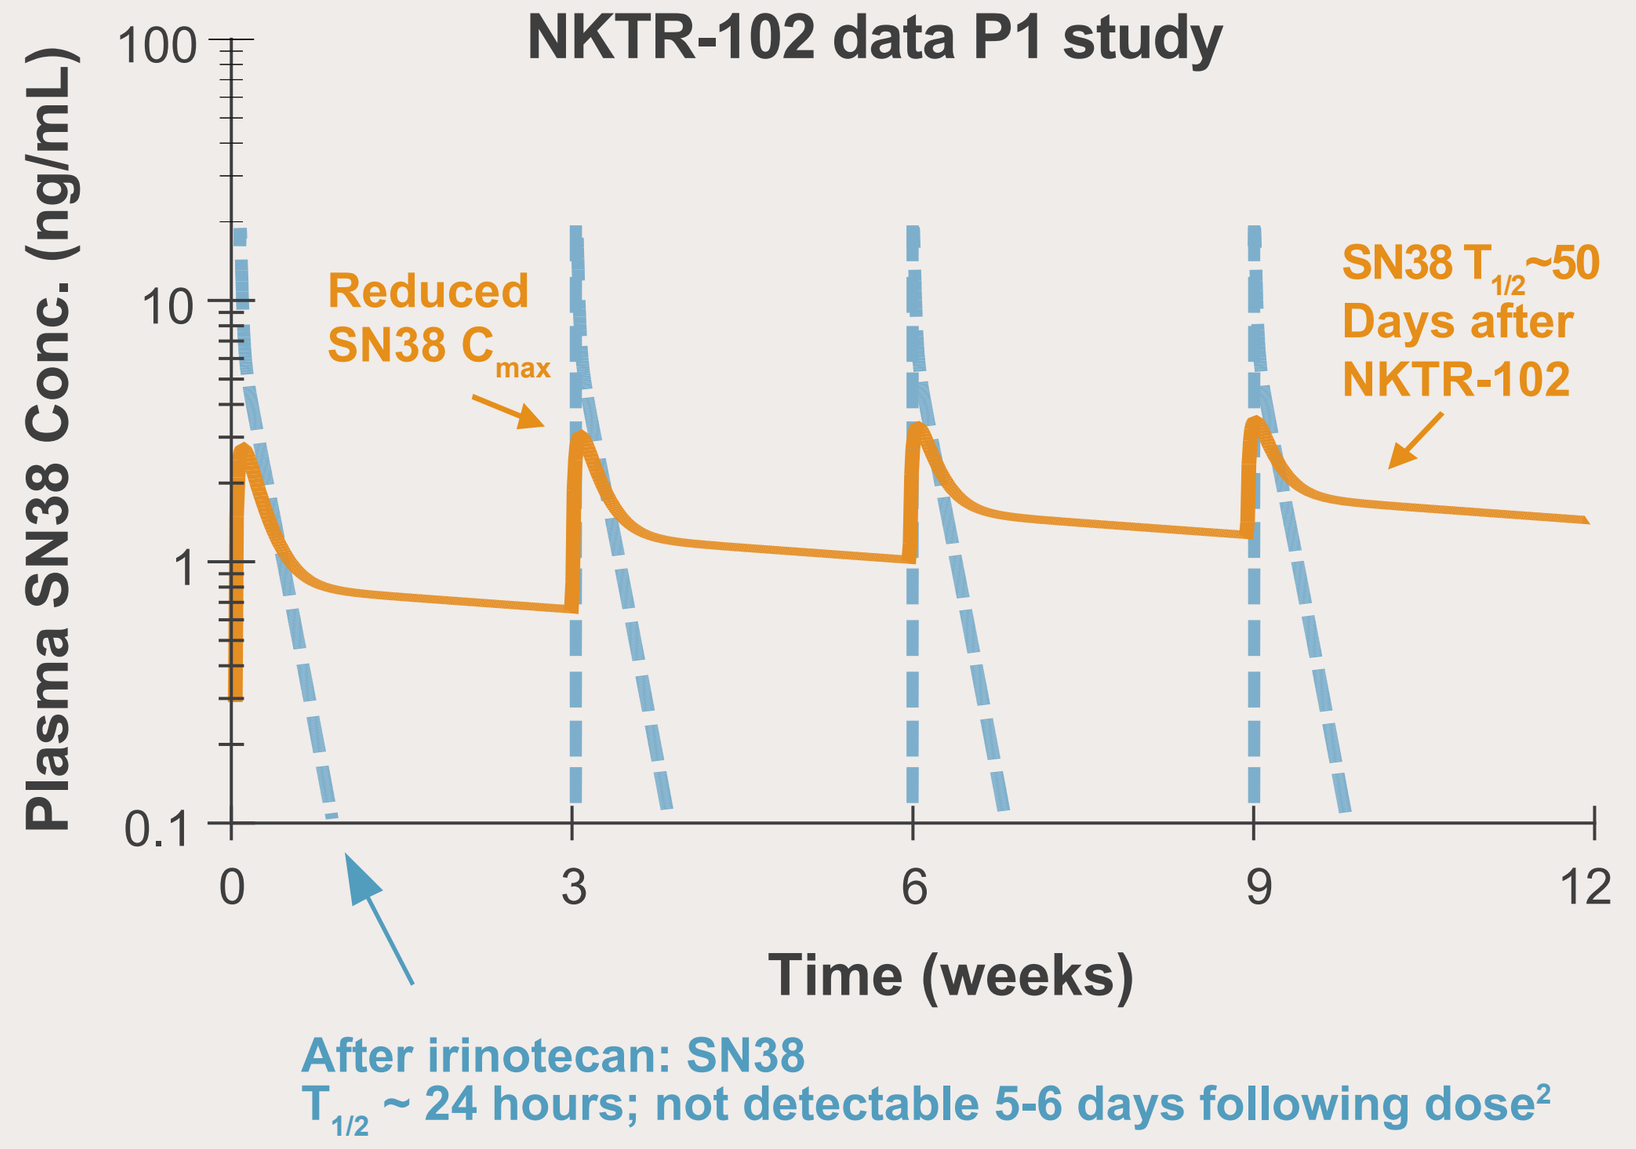
NKTR-102 data P1 study
Plasma SN38 Conc. (ng/mL)
100
10
1
0.1
Reduced SN38 Cmax
SN38 T1/2~50 Days after NKTR-102
0 3 6 9 12
Time (weeks)
After irinotecan: SN38
T1/2 ~ 24 hours; not detectable 5-6 days following dose2

- NKTR-102 demonstrated high anti-tumor activity in a range of tumors in phase 1 (11% confirmed PRs)[3]
 - Of interest, 3 patients in the Phase 1 study with triple-negative breast cancer (TNBC) showed significant response to single-agent NKTR-102[3]
- NKTR-102 showed a 22% confirmed response rate per RECIST in heavily pre-treated women with platinum resistant/refractory ovarian cancer (Vergote IB, et. al. Proc. Am. Soc. Clin. Oncol. 2010; 28: Abstract 5013)[4]

References:
1.Persson H., et. Al., NKTR-102, a novel polyethylene glycol conjugate of irinotecan, has improved anti-tumor activity in three mouse xenograft models. Poster presented at the 2007 AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics, Oct 22-26, 2007, San Francisco, CA USA. Poster no. C10

2. Xie R, Mathijssen RHJ, Sparreboom A, et al. Clinical pharmacokinetics of irinotecan and its metabolites: A population analysis. J Clin Oncol 20 (15): 3293-3301, 2002

3. Von Hoff DD, Jameson GS, Borad MJ et al. First Phase 1 trial of NKTR-102 (Peg-Irinotecan) reveals early evidence of broad anti-tumor activity in three different schedules. Presented at the 20th EORTC-NCI-AACR Symposium on "Molecular Targets and Cancer Therapeutics" Meeting, Oct 21-24, 2008, Geneva, Switzerland. Poster no. 595

4. Vergote IB, Micha JP, Pippitt Jr. CH, Rao GG, Spitz DL, Reed N, Dark GG, Garcia A, Maslyar DJ, and Rustin GJ. Phase II study of NKTR-102 in women with platinum-resistant/refractory ovarian cancer. J Clin Oncol 28:15s, 2010 suppl; abstr 5013.

Study Design & Objectives

Study Design: Randomized Simon Two-Stage

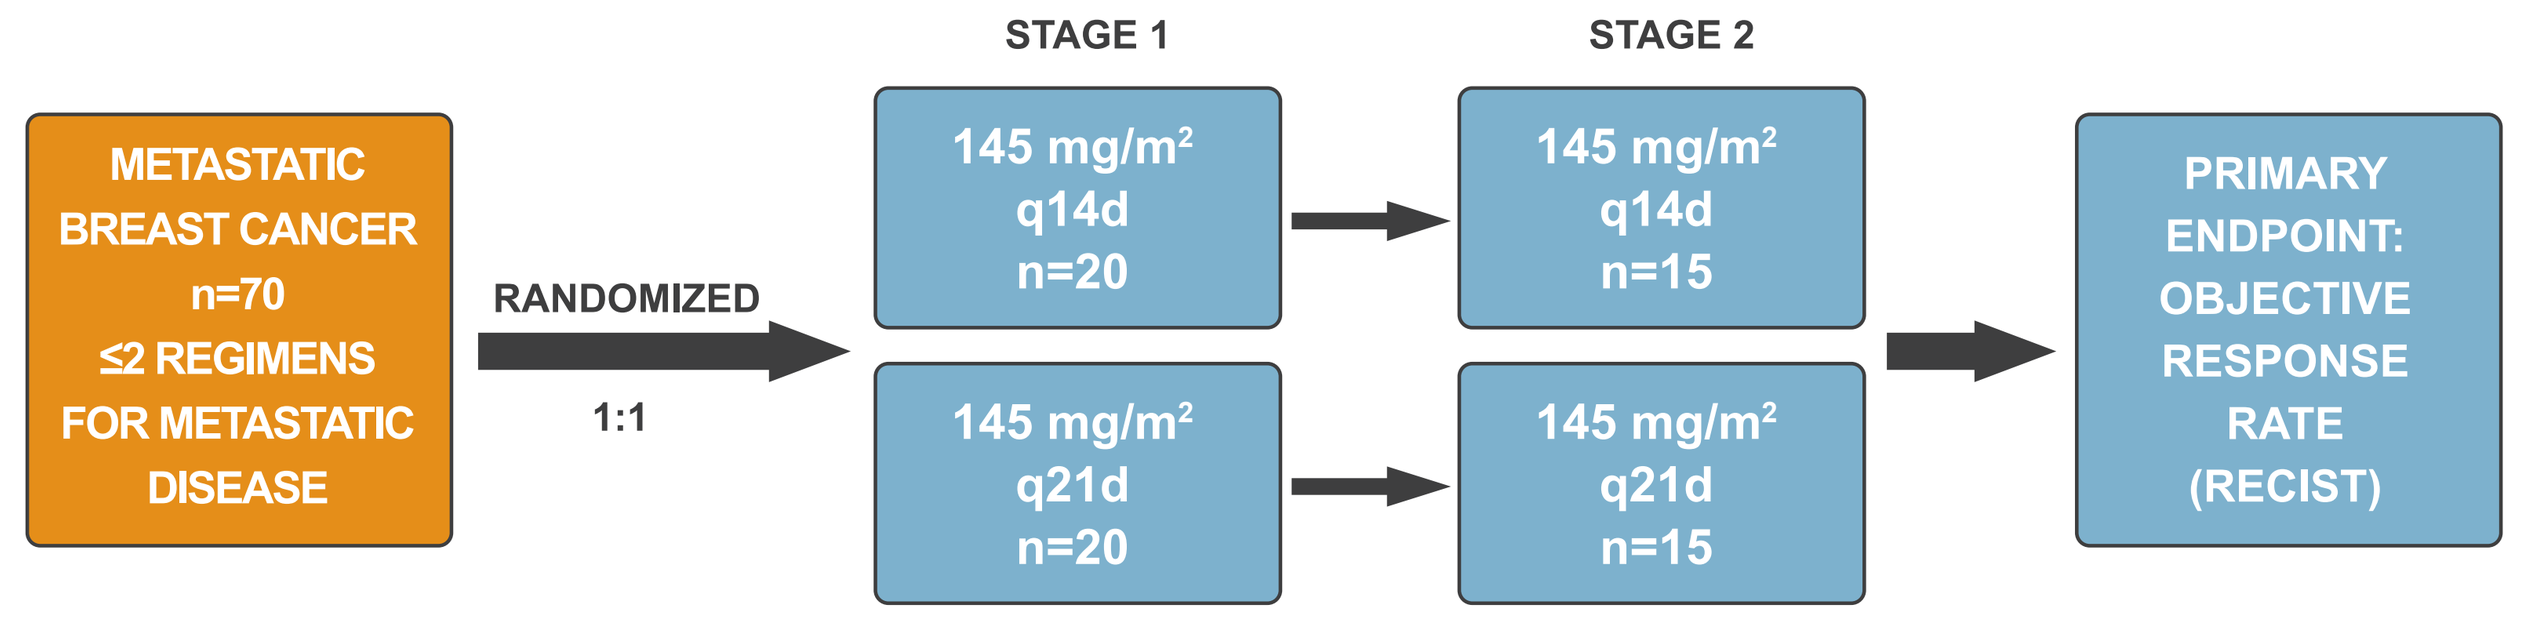
STAGE 1
STAGE 2
METASTATIC BREAST CANCER n=70 ≤2 REGIMENS FOR METASTATIC DISEASE
RANDOMIZED
1:1
145 mg/m² q14d n=20
145 mg/m² q21d n=20
145 mg/m² q14d n=15
145 mg/m² q21d n=15
PRIMARY ENDPOINT: OBJECTIVE RESPONSE RATE (RECIST)

Statistical Hypotheses: H_0 ORR (RECIST version 1.0) ≤ 5% and H_a ORR ≥ 20%. (Type 1 error = 0.029; type 2 error = 0.145)

Stage 1: If ≥ 1 patient responds, that treatment regimen proceeds to the next stage.
Stage 2: An additional 15 are patients enrolled

If > 4 patients respond out of 35 patients (Stage 1 + Stage 2 combined), the drug has met the efficacy threshold.

NKTR-102 Breast Cancer Study: Objectives

Primary Efficacy Objective:
- Determine the objective response rate (ORR) by RECIST v 1.0
- Determine the optimal schedule of NKTR-102 in breast cancer

Secondary Objectives:
- Estimate progression-free survival (PFS)
- Evaluate overall survival (OS) rates
- Characterize the safety profile

Key Eligibility Criteria

- Male or female patients with advanced breast cancer following taxane therapy (adjuvant or metastatic)
- Patients may also have received prior anthracycline or capecitabine
- No prior camptothecin therapy
- No more than two prior chemotherapy regimens given in the metastatic setting
- Measurable disease as defined by RECIST version 1.0
- ECOG PS: 0-1
- Adequate renal, hepatic and marrow function
- No known or suspected CNS metastases
- No significant pre-existing acute/chronic GI disorder

Study Demographics

- 70 patients (35 per schedule) randomized from February 2009 through May 2010 (median follow-up: 8 months)

		NKTR-102 145 mg/m² Q14 days N=35	NKTR-102 145 mg/m² Q21 days N=35
Age (years)	Median (Range)	53 (33-83)	56 (37-77)
ECOG PS	0 1	15 (43%) 20 (57%)	13 (37%) 22 (63%)
Receptor Status*	ER+ or PR+ ER-/PR-/HER-2 (triple negative) Her2+	22 (63%) 11 (31%) 5 (14%)	21 (60%) 10 (29%) 1 (3%)
Prior Systemic Treatments*	Neoadjuvant and/or Adjuvant therapy Taxane based regimen in metastatic setting Prior AT only (anthracycline/taxane) Prior AT only for metastatic disease Prior ATC (anthracycline/taxane/capecitabine) Median cytotoxic regimens (metastatic disease) Visceral (at least one lesion)	28 (80%) 32 (91%) 23 (66%) 7 (20%) 8 (23%) 2 28 (80%)	24 (69%) 33 (94%) 21 (60%) 9 (26%) 10 (29%) 2 32 (91%)
Time from primary diag. to metastatic	Median (years) (Range)	1.5 (0-7)	2 (0-12)
Time from Last Chemo to Entry	Median (months) (Range)	1.1 (0-22.6)	1.5 (0-84.7)

* Numbers may add up to more than 100% due to patients included in multiple rows
Source: Data as of May 9, 2011

Results

Objective Tumor Response Rate by RECIST (Investigator Assessment)

Response by RECIST v 1.0	NKTR-102 145 mg/m² Q14 days	NKTR-102 145 mg/m² Q21 days	TOTAL
N ORR (confirmed + unconfirmed) ORR (confirmed)	31* 11(35%) 10(32%)	35/35 11(31%) 9 (26%)	66 22 (33%) 19 (29%)
CR (confirmed) PR (confirmed) SD PD	2 (7%) 8 (26%) 12 (39%) 9 (29%)	0 9 (26%) 16 (46%) 10 (29%)	2 (3%) 17 (26%) 28 (42%) 19 (29%)
Duration of ORR (months)	8.3	4.4	5.8
Clinical benefit (CR+PR+SD≥6 months)	13 (42%)	17 (49%)	30 (46%)

*4 patients in the Q14 day arm with no post-baseline scans and no other evidence of progression were excluded from the evaluable population.
Source: Data as of May 9, 2011

Response Rate By Prior Therapy

Prior Therapy Subgroup	Response by RECIST v 1.0 n/N (%) Evaluable Patients		
	NKTR-102 145 mg/m² Q14 days	NKTR-102 145 mg/m² Q21 days	TOTAL
Prior A/T only ORR (confirmed)	7/22 (32%)	5/21 (24%)	12/43(28%)
Prior A/T in MBC ORR (confirmed)	3/7 (43%)	2/9 (22%)	5/16 (31%)
Prior A/T/C ORR (confirmed)	2/6 (33%)	3/10 (30%)	5/16 (31%)

Source: Data as of May 9, 2011

Response Rate By Tumor Characteristics

Disease Subgroup	Response by RECIST v 1.0 n/N (%) Evaluable Patients		
	NKTR-102 145 mg/m² Q14 days	NKTR-102 145 mg/m² Q21 days	TOTAL
ER+ and/or PR+ ORR (confirmed)	8/21 (38%)	4/21 (19%)	12/42 (29%)
TNBC ORR (confirmed)	2/8 (25%)	5/10 (50%)	7/18 (39%)
Visceral Disease ORR (confirmed)	8/25 (32%)	9/32 (28%)	17/57 (30%)

Source: Data as of May 9, 2011

Study Drug Administration and Discontinuation Due to AE

	NKTR-102 145 mg/m²		
	Q14 days (N=35)	Q21 days (N=35)	TOTAL (N=70)
Discontinuation from Study Drug Due to AE	7 (20.0%)	5 (14.3%)	12 (17.1%)
Total Exposure Duration (days) Median (Range)	85 (1-393)	113.5 (1-420)	85 (1-420)
Total No. of Cycles Received Median (Range)	6.0 (1-29)	6.0 (1-21)	6.0 (1-29)

Source: Data as of May 9, 2011

Maximum Decline in Tumor Measurements

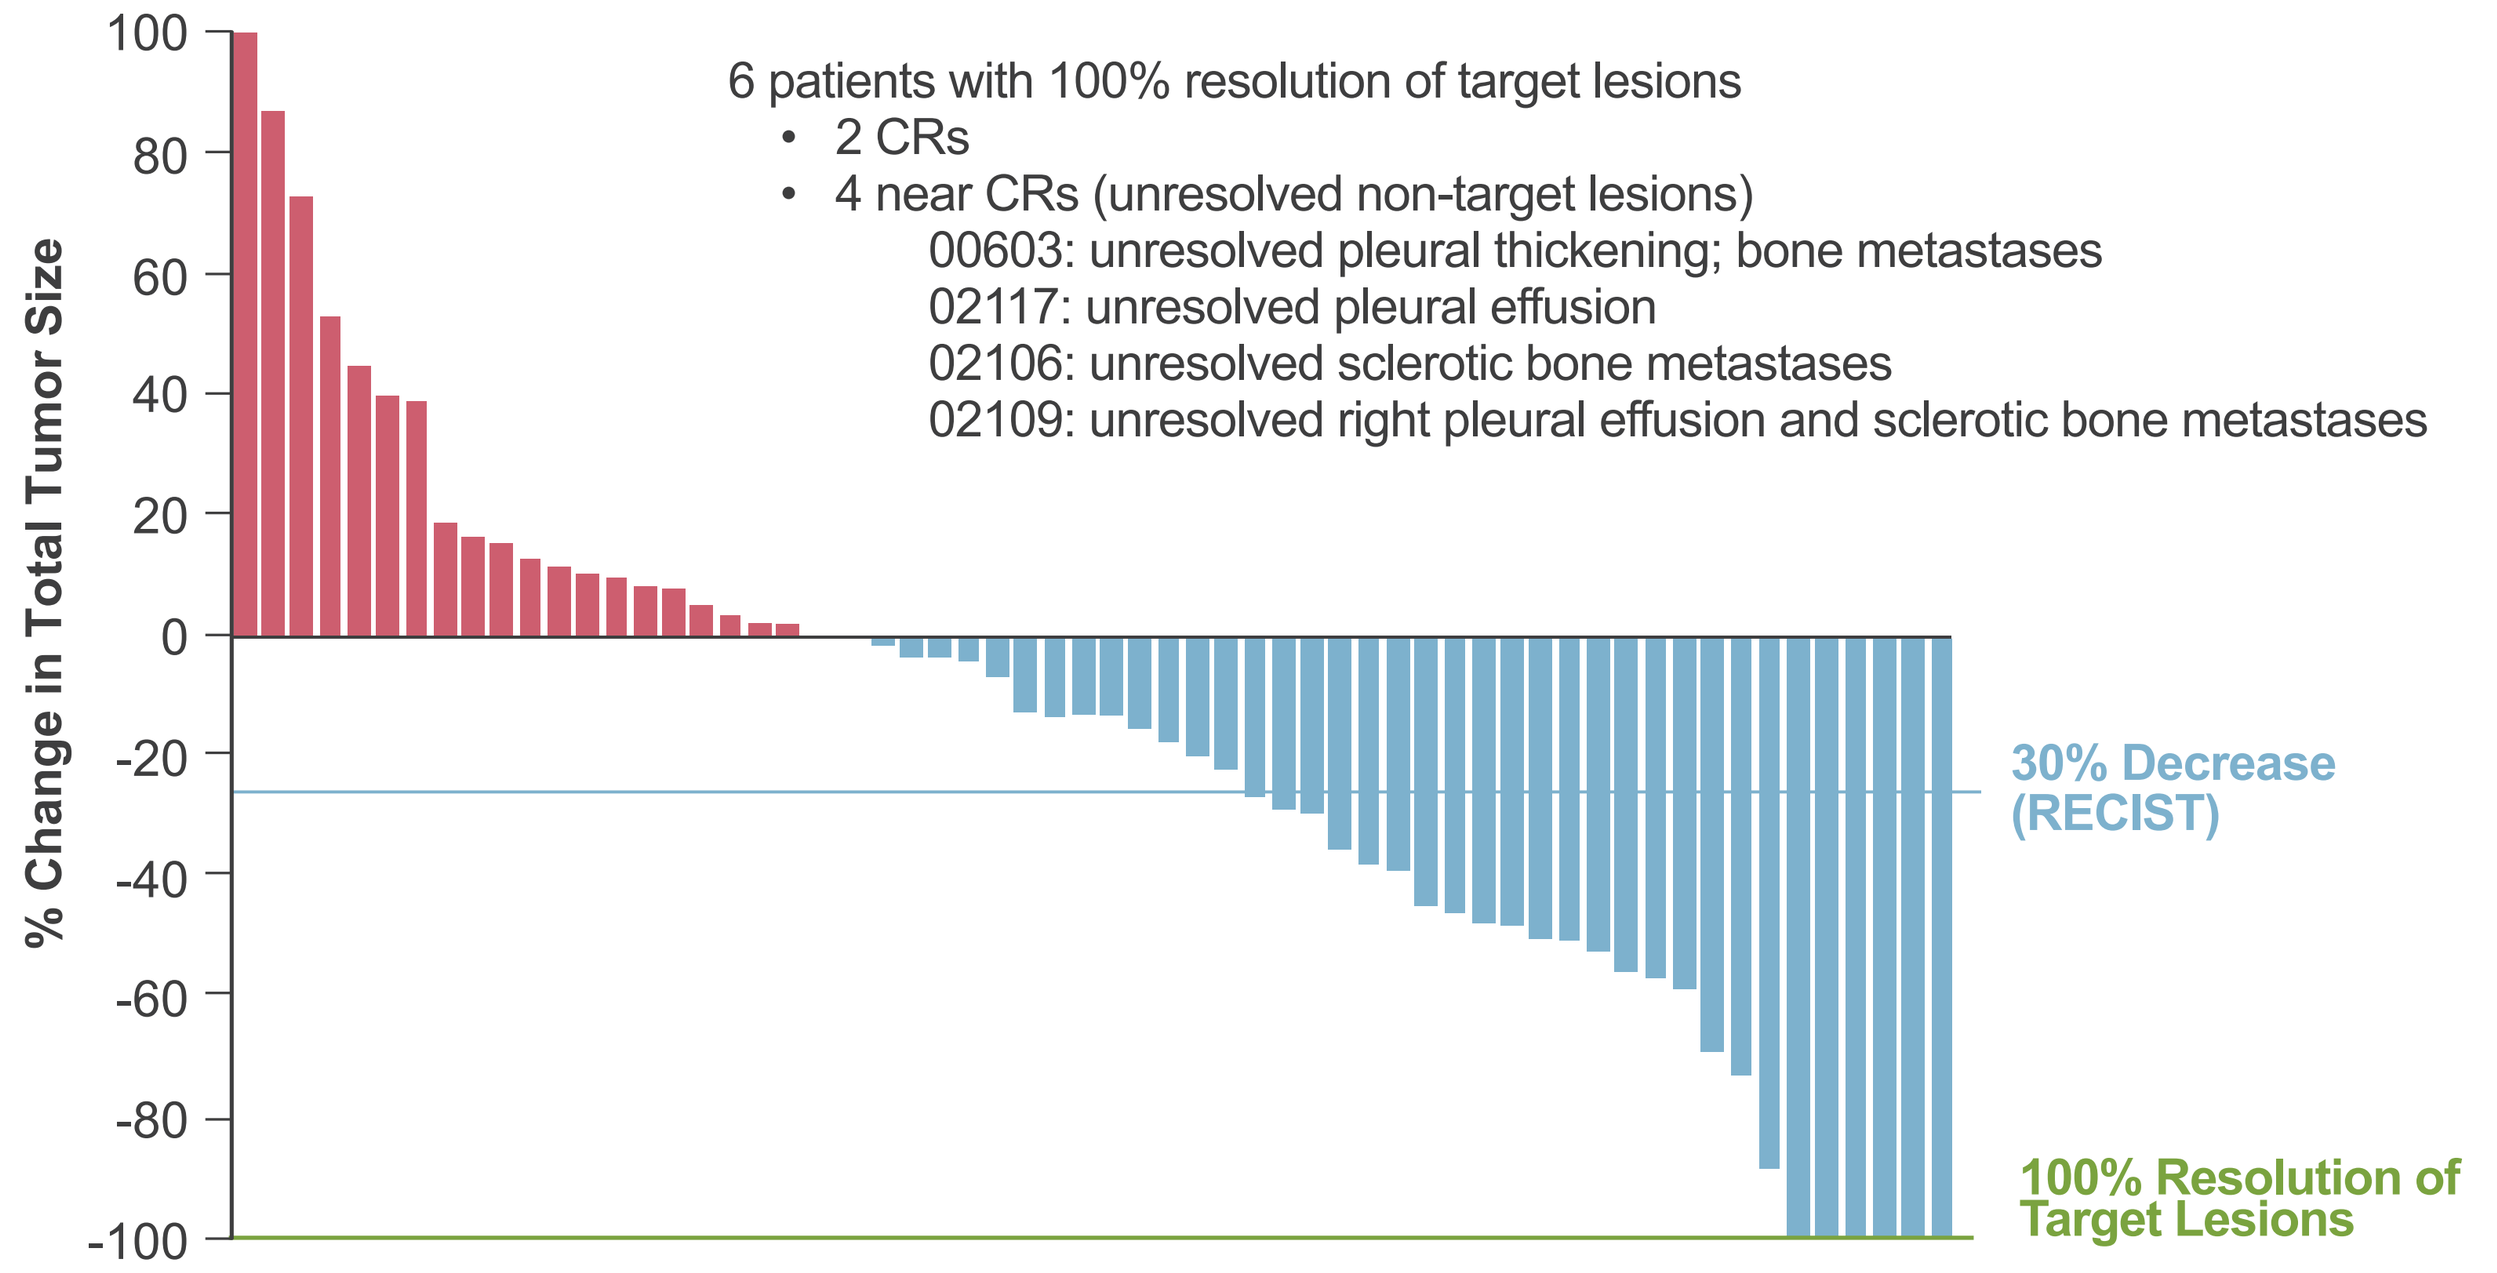
% Change in Total Tumor Size
100
80
60
40
20
0
-20
-40
-60
-80
-100
6 patients with 100% resolution of target lesions
• 2 CRs
• 4 near CRs (unresolved non-target lesions)
00603: unresolved pleural thickening; bone metastases
02117: unresolved pleural effusion
02106: unresolved sclerotic bone metastases
02109: unresolved right pleural effusion and sclerotic bone metastases
30% Decrease (RECIST)
100% Resolution of Target Lesions

Progression-Free Survival (All Patients)

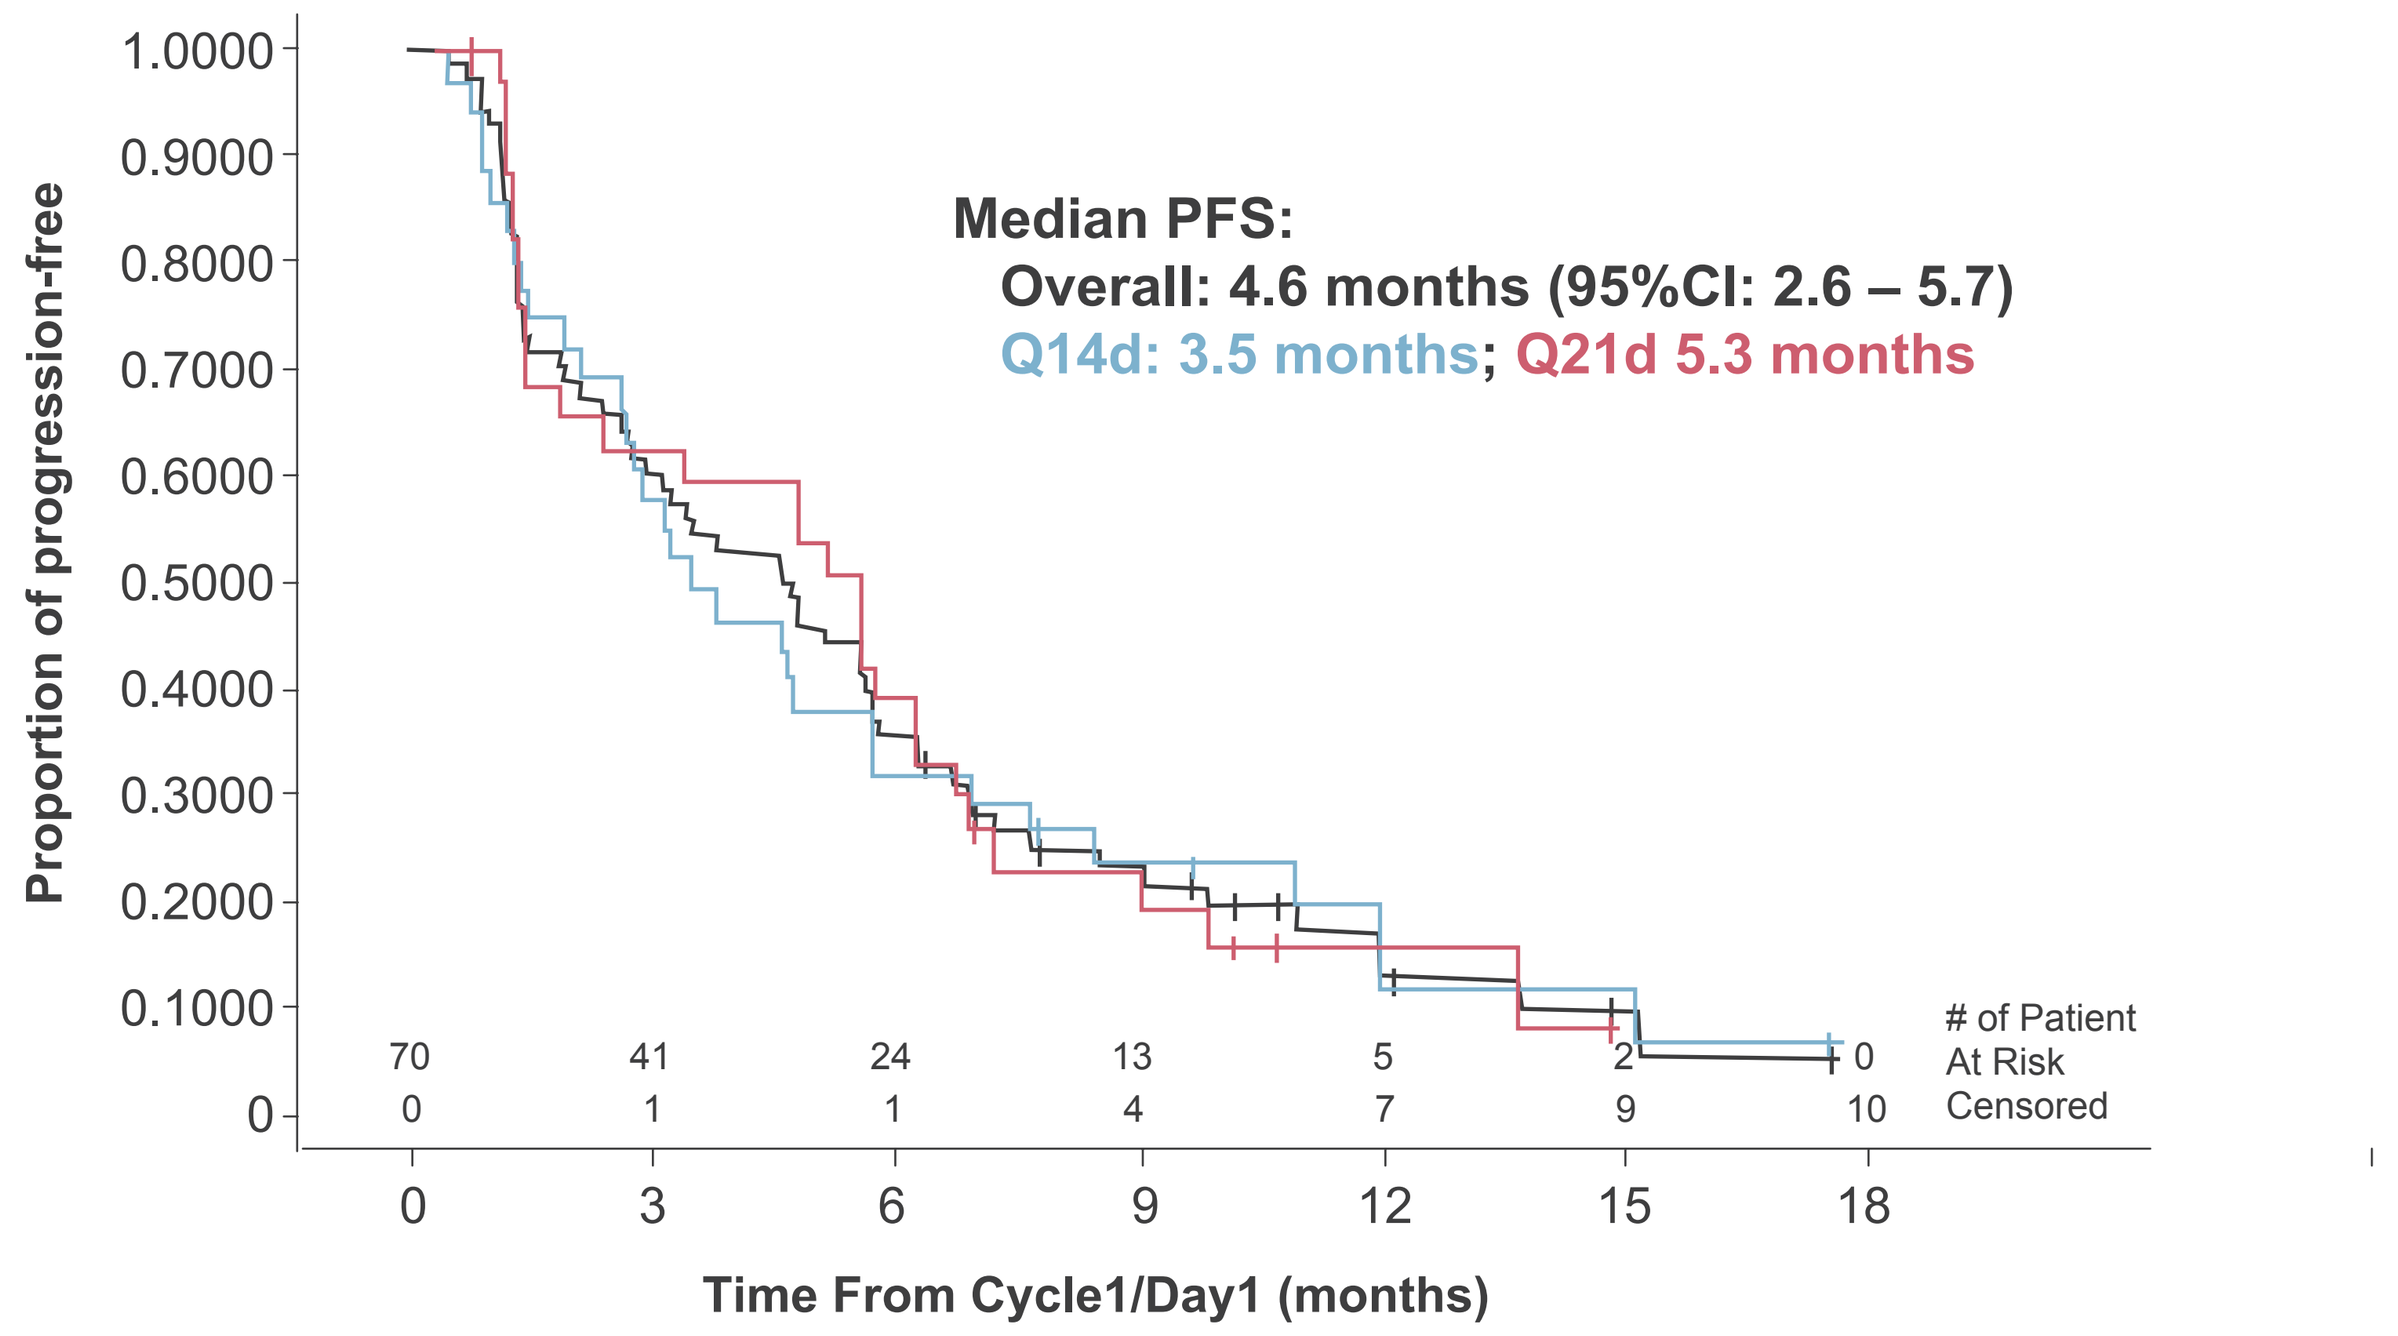
Proportion of progression-free
1.0000
0.9000
0.8000
0.7000
0.6000
0.5000
0.4000
0.3000
0.2000
0.1000
0
Median PFS:
Overall: 4.6 months (95%CI: 2.6 – 5.7)
Q14d: 3.5 months; Q21d 5.3 months
70 41 24 13 5 2 0
0 1 1 4 7 9 10
of Patient At Risk Censored
0 3 6 9 12 15 18
Time From Cycle1/Day1 (months)

Kaplan-Meier graph of progression-free survival
Analysis includes the intent-to-treat population. Tickmarks show censored data.
Source: Data as of May 9, 2011

Overall Survival (All Patients)

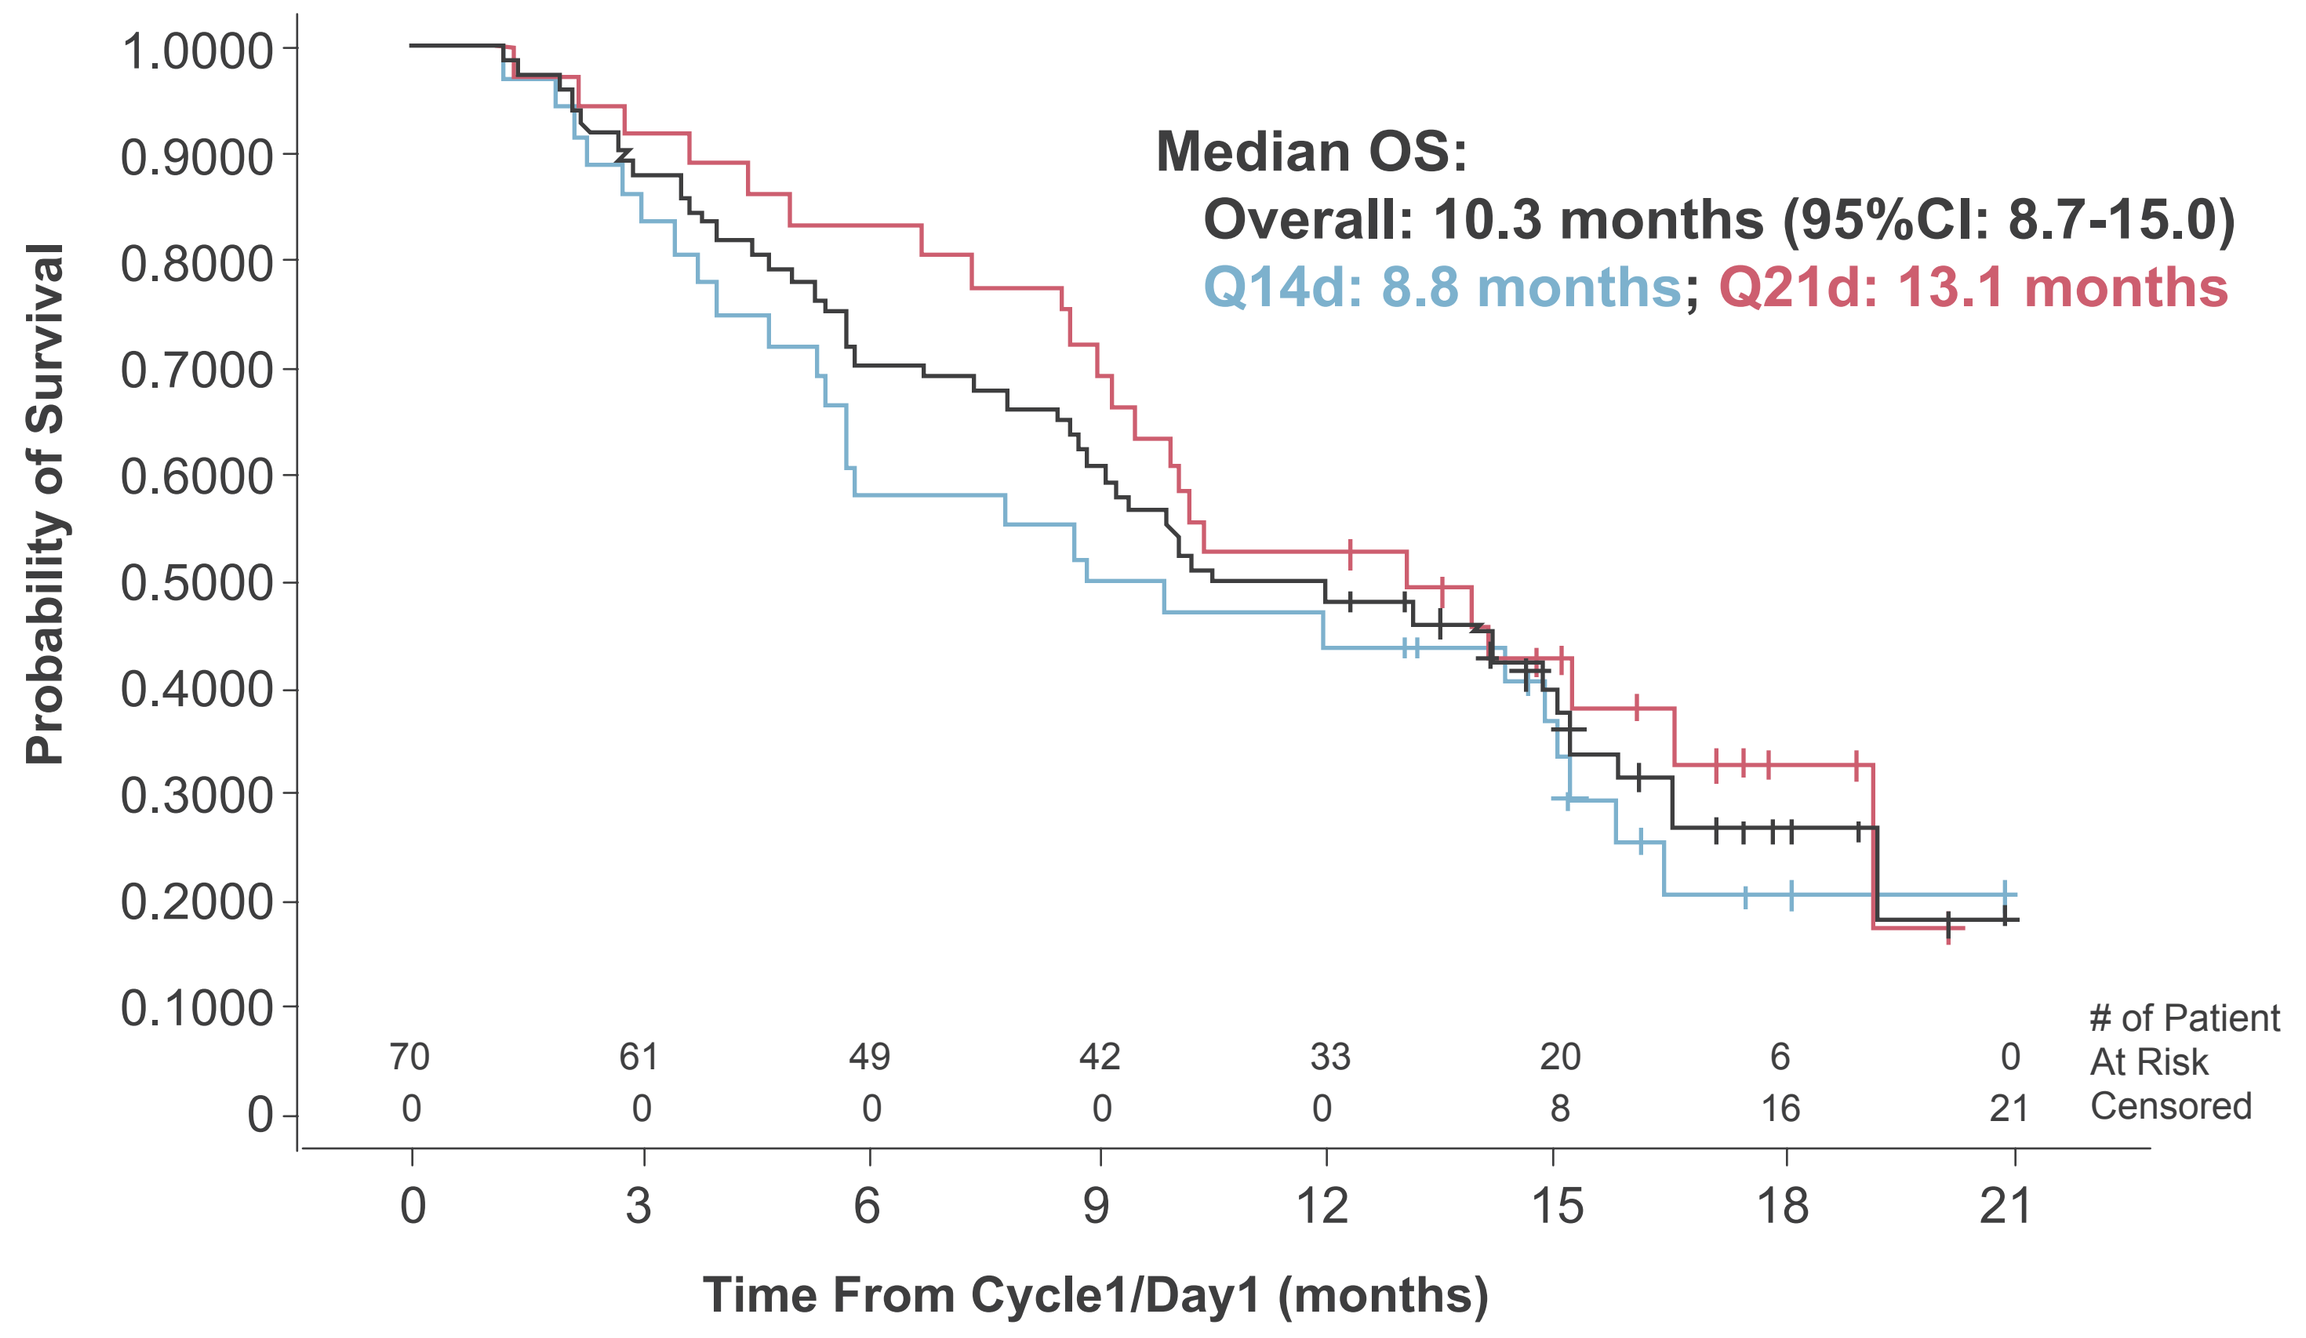
Probability of Survival
1.0000
0.9000
0.8000
0.7000
0.6000
0.5000
0.4000
0.3000
0.2000
0.1000
0
Median OS:
Overall: 10.3 months (95%CI: 8.7-15.0)
Q14d: 8.8 months; Q21d: 13.1 months
70 61 49 42 33 20 6 0
0 0 0 0 0 8 16 21
of Patient At Risk Censored
0 3 6 9 12 15 18 21
Time From Cycle1/Day1 (months)

Kaplan-Meier graph of overall survival
Analysis includes the intent-to-treat population. Tickmarks show censored data.
Source: Data as of May 9, 2011

Conclusions

- High confirmed objective response rate observed with single-agent NKTR-102 in patients with advanced breast cancer previously treated with taxane +/- capecitabine (89% with prior anthracycline):
 - 29% confirmed objective response rate
 - 32% ORR on 14-day schedule; 26% on -21-day schedule
 - PFS: 4.6 months in 2nd/3rd line
 - Preliminary estimate for median survival: 10.3 months
- Similar anti-tumor activity for both schedules; 21-day schedule appears better tolerated in a setting of improved survival.
- ORR is maintained in heavily pre-treated and poor prognosis subsets
 - A/T/C Pre-treated: 31%
 - Triple negative: 39%
 - Visceral disease: 30%
- Side effects generally manageable; most common Grade 3/4 toxicity was diarrhea (20-23%) typically occurring after 3 months of therapy for both chedules
- NKTR-102 is being evaluated in multiple cancer indications as a single and combination agent. Phase 3 planning is underway in ovarian and breast cancers.

Safety

Safety: Summary of Drug Related AEs

Most Common Drug-related Grade 3 and 4 Adverse Events > 5% or event of interest N (%)	145 mg/m² Q14 days* N=35		145 mg/m² Q21 days* N=35	
	Grade 3	Grade 4	Grade 3	Grade 4
Diarrhea	6 (17%)	1 (3%)	8 (23%)	0
Neutropenia	2 (6%)	2 (6%)	3 (9%)	1 (3%)
Dehydration	3 (9%)	0	4 (11%)	0
Fatigue	4 (11%)	0	3 (9%)	0
Vomiting	3 (9%)	0	0	0
Anaemia	1 (3%)	0	0	1 (3%)
Asthenia	2 (6%)	0	0	0
Lethargy	2 (6%)	0	0	0
Lymphopenia	1 (3%)	1 (3%)	0	0
Neutropenic sepsis	0	0	1 (3%)	0
Febrile neutropenia	0	0	1 (3%)	0
N (%)	Q14 days N=35		Q21 days N=35	
	Grade 1	Grade 2	Grade 1	Grade 2
Alopecia	7 (20%)	0	3 (9%)	1 (3%)

*2 possible treatment-related deaths: sepsis (Q21 days) and acute renal failure following diarrhea (Q14 days)
Source: Data as of May 9, 2011

Safety: Time Course of Diarrhea and Neutropenia

	NKTR-102 145 mg/m² q14d N=35	NKTR-102 145 mg/m² q21d N=35
Diarrhea (≥Grade 3) Cycle 1 and/or 2 Cycle 3 and/or 4 Cycle 4+ Onset Time, Median (Range) days [# cycle] Duration, Median (Range) days	 9% 0% 11% 88 (1-121) [6] 8.5 (1-16)	 3% (G3 only) 6% (G3 only) 14% (G3 only) 93 (8-107) [5] 14 (2-39)
Neutropenia (≥Grade 3) Cycle 1 and/or 2 Cycle 3 and/or 4 Cycle 4+ Onset Time, Median (Range) days [# cycle] Duration, Median (Range) days	 3% 0% 9% 98 (15-188) [6.5] 12 (6-15)	 3% 6% 3% 60 (28-203) [3] 8 (6-14)

*Anti-diarrheals given therapeutically; no prophylactic anti-diarrheals administered

Abstract 1034 – Poster #24 Poster presented at the 2011 American Society of Clinical Oncology Annual Meeting : Ovarian Cancer Poster Session, June 3-6, 2011
To download a copy of this poster, please visit http://www.nektar.com