Promising Activity of NKTR-102 in Women with Platinum Resistant/Refractory Ovarian Cancer and Failure on Pegylated Liposomal Doxorubicin (PLD)

A. A. Garcia, I. Vergote, J. P. Micha, C. H. Pippitt Jr., G. G. Rao, D. L. Spitz, N. Reed, G. G. Dark, E. Ibrahim, V. A. Armenio, L. Duska, C. Poole, C. Gennigens, L. Y. Dirix, J-S West, C. Zhao, A. C. Leung, L. Masuoka, G. J. Rustin

Background

Nektar's polymer conjugate technology, an established platform, is used to develop NKTR-102, a next-generation polymer releasable linker attached topoisomerase I inhibitor conjugate macromolecule. The main significant potential benefits of NKTR-102 are two-fold:

1) The potential for improved efficacy as the drug provides continuous exposure throughout dosing intervals due to marked increase in the half-life of the active metabolite (SN38, $T_{1/2}$=50 days from NKTR-102 versus ~2 days from irinotecan).[1] The polymer also concentrates in xenograft tumors, indicating the potential of NKTR-102 to concentrate at the site of tumors with prolonged mean residence time (enhanced permeation and retention effect)[2]:

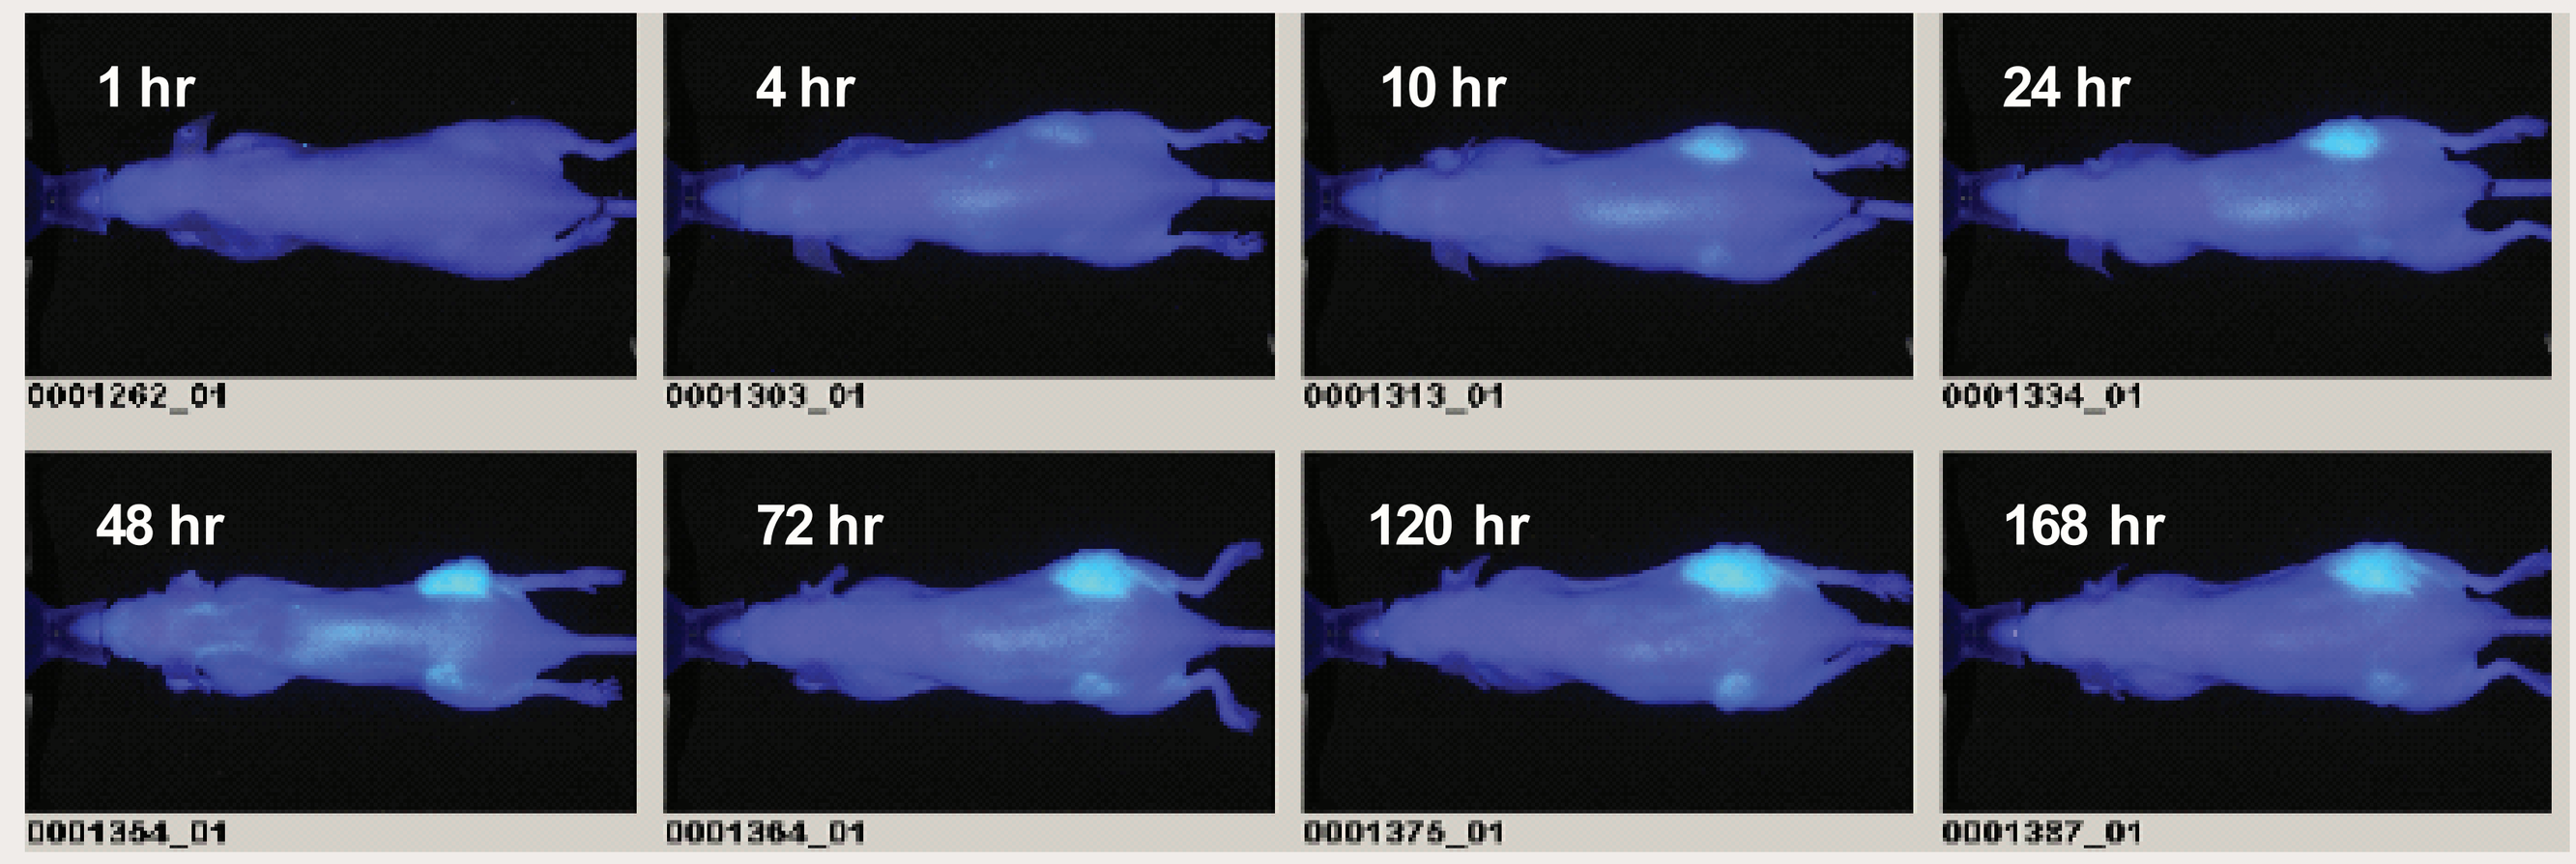
1 hr
4 hr
10 hr
24 hr
48 hr
72 hr
120 hr
168 hr
0001262_01
0001303_01
0001313_01
0001334_01
0001354_01
0001364_01
0001375_01
0001387_01

IR Dye signal remains elevated in the tumor tissue at the end of the study

2) The potential for significant reduction in certain adverse events associated with the parent compound, irinotecan, such as early, cholinergic diarrhea, likely due to the markedly reduced Cmax and slowed metabolic conversion of irinotecan to SN38, leading to a smaller SN38 pool. The metabolic inactivation of SN38 for drug elimination is less rate limiting for NKTR-102 due to this reduced SN38 pool which should minimize the need to alter starting dose for patients with metabolic enzyme mutations such as UGT1A1*28.[3]

Clinical results from the Phase 1 demonstrated potential NKTR-102 anti-tumor activity in a variety of tumors[1]:

Tumor	RECIST Response/ Tumor Reduction	Prior Agents
Breast (triple negative)	PR: ↓41%	Doxil, bevacizumab, docetaxel, carboplatin, tamoxifen, paclitaxel, cisplatin
Ovarian	uPR: ↓48%	Investigational agent, docetaxel, carboplatin, tamoxifen, paclitaxel, cisplatin
Cervical	PR: ↓53%	Cisplatin, gemcitabine, investigational agent
Colorectal	uPR: ↓51%	5-FU/leuvocovorin, oxaliplatin, bevacizumab, irinotecan, cetuximab, capecitabine
Colorectal	PR: ↓34%	5-FU/leuvocovorin, oxaliplatin, bevacizumab, panitumumab
Pancreatic (neuroendocrine)	PR: ↓36%	Doxil, gemcitabine, pemetrexed, etopisode, carboplatin, paclitaxel, capecitabine, investigational agent
Non-small cell lung	PR: ↓52%	Docetaxel, carboplatin, pemetrexed
Small cell lung	PR: ↓56%	Gemcitabine, topotecan, carboplatin, etopisode, cisplatin, investigational agent
Head and neck	PR: ↓45%	None
Bladder	PR: ↓53%	Gemcitabine, cisplatin, paclitaxel

NKTR-102 has been evaluated in a Phase 2 study in patients with platinum resistant/refractory ovarian cancer. The updated results for the subpopulation who had also received prior PLD is reported here.

Need for New/Novel Approaches to Platinum Resistant/Refractory Population

Current Treatment Options for platinum resistant/refractory disease can be summarized as follow:
- Challenging because of the marginal benefit obtained with current treatment options
- Having no evidence that any one of the available drugs being superior to another in this setting
- In randomized Phase 2 and 3 studies, objective response rates are low; for active drugs ORR typically less than 20%[4]
- For patients with highly platinum resistant disease (platinum-free interval less than 3 months) response rates are particularly low, typically less than 10%[5]

The lack of effective therapy for this highly chemoresistant subset of patients with ovarian cancer underscores the need for new approaches to this disease.

References:

1. Von Hoff DD, Jameson GS, Borad MJ et al. First Phase 1 trial of NKTR-102 (Peg-Irinotecan) reveals early evidence of broad anti-tumor activity in three different schedules. Presented at the 20th EORTC-NCI-AACR Symposium on "Molecular Targets and Cancer Therapeutics" Meeting, Oct 21-24, 2008, Geneva, Switzerland. Poster no. 595

2. Source: Nektar Therapeutics

3. Eldon M., et. Al., Population Pharmacokinetics of NKTR-102, a Topoisomerase I Inhibitor-Polymer Conjugate in Patients with Advanced Solid Tumors. 2011 American Society of Clinical Oncology Annual Meeting June 3-6, 2011 Abstract #2598

4. Vergote I, Finkler N, del Campo J, et al. Phase 3 Randomized Study of Canfosfamide (Telcyta®, TLK286) Versus Pegylated Liposomal Doxorubicin or opotecan as Third-Line Therapy in Patients with Platinum-Refractory or –Resistant Ovarian Cancer. European Journal of Cancer. 2009; 45: 2324-2332.

5. Pujade LE, Paraiso D, Joly F, et al. Is There a Role for Platinum in the Treatment of Patients with "Platinum-Resistant" Relapsed Advanced Ovarian Cancer (AOC) A GINECO study. Proc Am Soc Clin Oncol. 2003; 22: 451.

6. Vergote IB, Micha JP, Pippitt Jr. CH, Rao GG, Spitz DL, Reed N, Dark GG, Garcia A, Maslyar DJ, and Rustin GJ. Phase II study of NKTR-102 in women with platinum-resistant/refractory ovarian cancer. J Clin Oncol 28:15s, 2010 suppl; abstr 5013.

Study Design & Objectives

Original Study Design (33 previously treated with PLD, 37 PLD naïve)

NKTR-102 Phase 2 Study Design: Platinum Resistant Ovarian Cancer

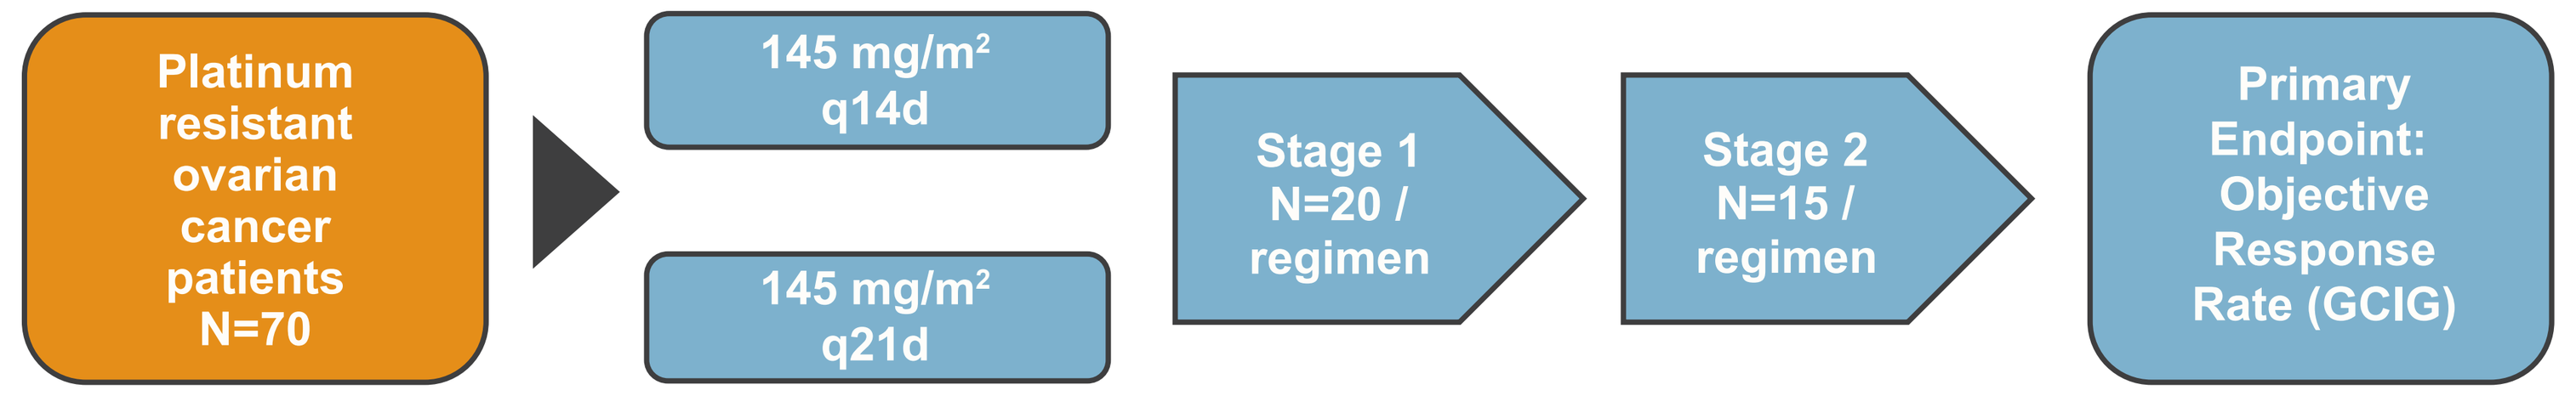
Platinum resistant ovarian cancer patients N=70
145 mg/m² q14d
145 mg/m² q21d
Stage 1 N=20 / regimen
Stage 2 N=15 / regimen
Primary Endpoint: Objective Response Rate (GCIG)

Statistical Hypotheses: 2-stage design (with power of 0.85 for a RR of 20% and with alpha of 0.03 for RR of 5%)

Stage 1: If ≥ 1 patient responds, that treatment regimen proceeds to the next stage
Stage 2: An additional 15 are patients enrolled

If ≥ 5 patients respond out of 35 patients (Stage 1 and Stage 2 combined), the drug has met the efficacy threshold.

NKTR-102 – Ovarian Cancer Study: Objectives

Primary Objective:

To determine the objective response rate (ORR) in patients with PROC who had also prior PLD treated with NKTR-102 given on one of two schedules: q14d and q21d

Secondary Objectives:

- To estimate progression-free survival (PFS) with NKTR-102
- To evaluate overall survival (OS) rates with NKTR-102
- To characterize the safety profile of NKTR-102

all results concern the group who were previously treated with PLD

Phase 2 Study Subpopulation Having Prior PLD

71 pts with platinum resistant/refractory ovarian cancer (PROC) were originally randomized to NKTR-102 IV at a dose of 145 mg/m² q14d vs. q21d [6]. Reported here are the results in the important subset of patients who are doubly chemoresistant – those who are platinum resistant and have progressed following treatment with PLD. This study subpopulation analysis includes 33 pts;16 pts in the q14d regimen and 17 pts in the q21d regimen. Six of these 33 pts were platinum-refractory (progression within 28 days of last platinum dose).

Key Eligibility Criteria

Key Inclusion Criteria:
Platinum-resistant ovarian cancer defined as:
– Progression during platinum-based therapy OR
– Progression within 6 months of receiving their last platinum-based therapy

Measurable disease as defined by RECIST version 1.0 in at least 1 lesion not previously irradiated or non-measurable disease

Key Exclusion Criteria:
Patients who have received any treatment with a camptothecin derivative (eg, irinotecan, topotecan, SN-38 investigational agents, etc.).

Study Demographics (Subpopulation Previously Treated with PLD)

	NKTR-102 q14d (N = 16)	NKTR-102 q21d (N = 17)	Total (N = 33)
Age			
Median (range)	58 (42, 78)	64 (48, 80)	59 (42, 80)
≥ 65	31%	47%	39%
ECOG PS			
0	44%	47%	46%
1	56%	53%	54%
Measurable disease (by RECIST)	100%	82%	91%
CA125 evaluable	94%	82%	88%
Histological Subtype			
Serous	94%	77%	85%
Malignant Mixed epithelial tumor	6%	6%	6%
Endometrioid	0%	12%	6%
Other	0%	6%	3%

	NKTR-102 q14d (N = 16)	NKTR-102 q21d (N = 17)	Total (N = 33)
Platinum-Free Interval			
≤ 6 weeks	38%	47%	42%
> 6 weeks	63%	53%	58%
Previous Platinum Regimens			
1	13%	35%	24%
2	56%	24%	39%
3	25%	18%	21%
4+	6%	24%	15%
No. Prior Therapy (median range)	4 (2 -8)	4 (1 -19)	4 (1 -19)
Prior taxane	100%	100%	100%
Prior gemcitabine	56%	59%	58%
Prior bevacizumab	6%	24%	15%

Efficacy Results: Subpopulation Previously Treated with PLD

Objective Response Rate by RECIST and GCIG

(Investigator Assessment)

	NKTR-102 q14d	NKTR-102 q21d	Total
RECIST			
N (evaluable)	16	14	30
Confirmed + Unconfirmed	4 (25%)	4(29%)	8 (27%)
Confirmed	3 (19%)	3 (21%)	6 (20%)
Duration for Confirmed Response (months, Range)*	4.2 (3-14)	4.4 (3-9)	4.3 (3-14)
GCIG			
N (evaluable)	16	17	33
Confirmed + Unconfirmed	8 (50%)	6 (35%)	14 (42%)
Confirmed	6 (38%)	6 (35%)	12 (36%)
CA-125			
N (evaluable)	15	14	29
Confirmed	6 (40%)	5 (36%)	11 (38%)

*Estimated using Kaplan-Meier method
Source: Data as of May 9, 2011

Progression-Free Survival

(All patients previously treated with PLD – dose groups pooled)

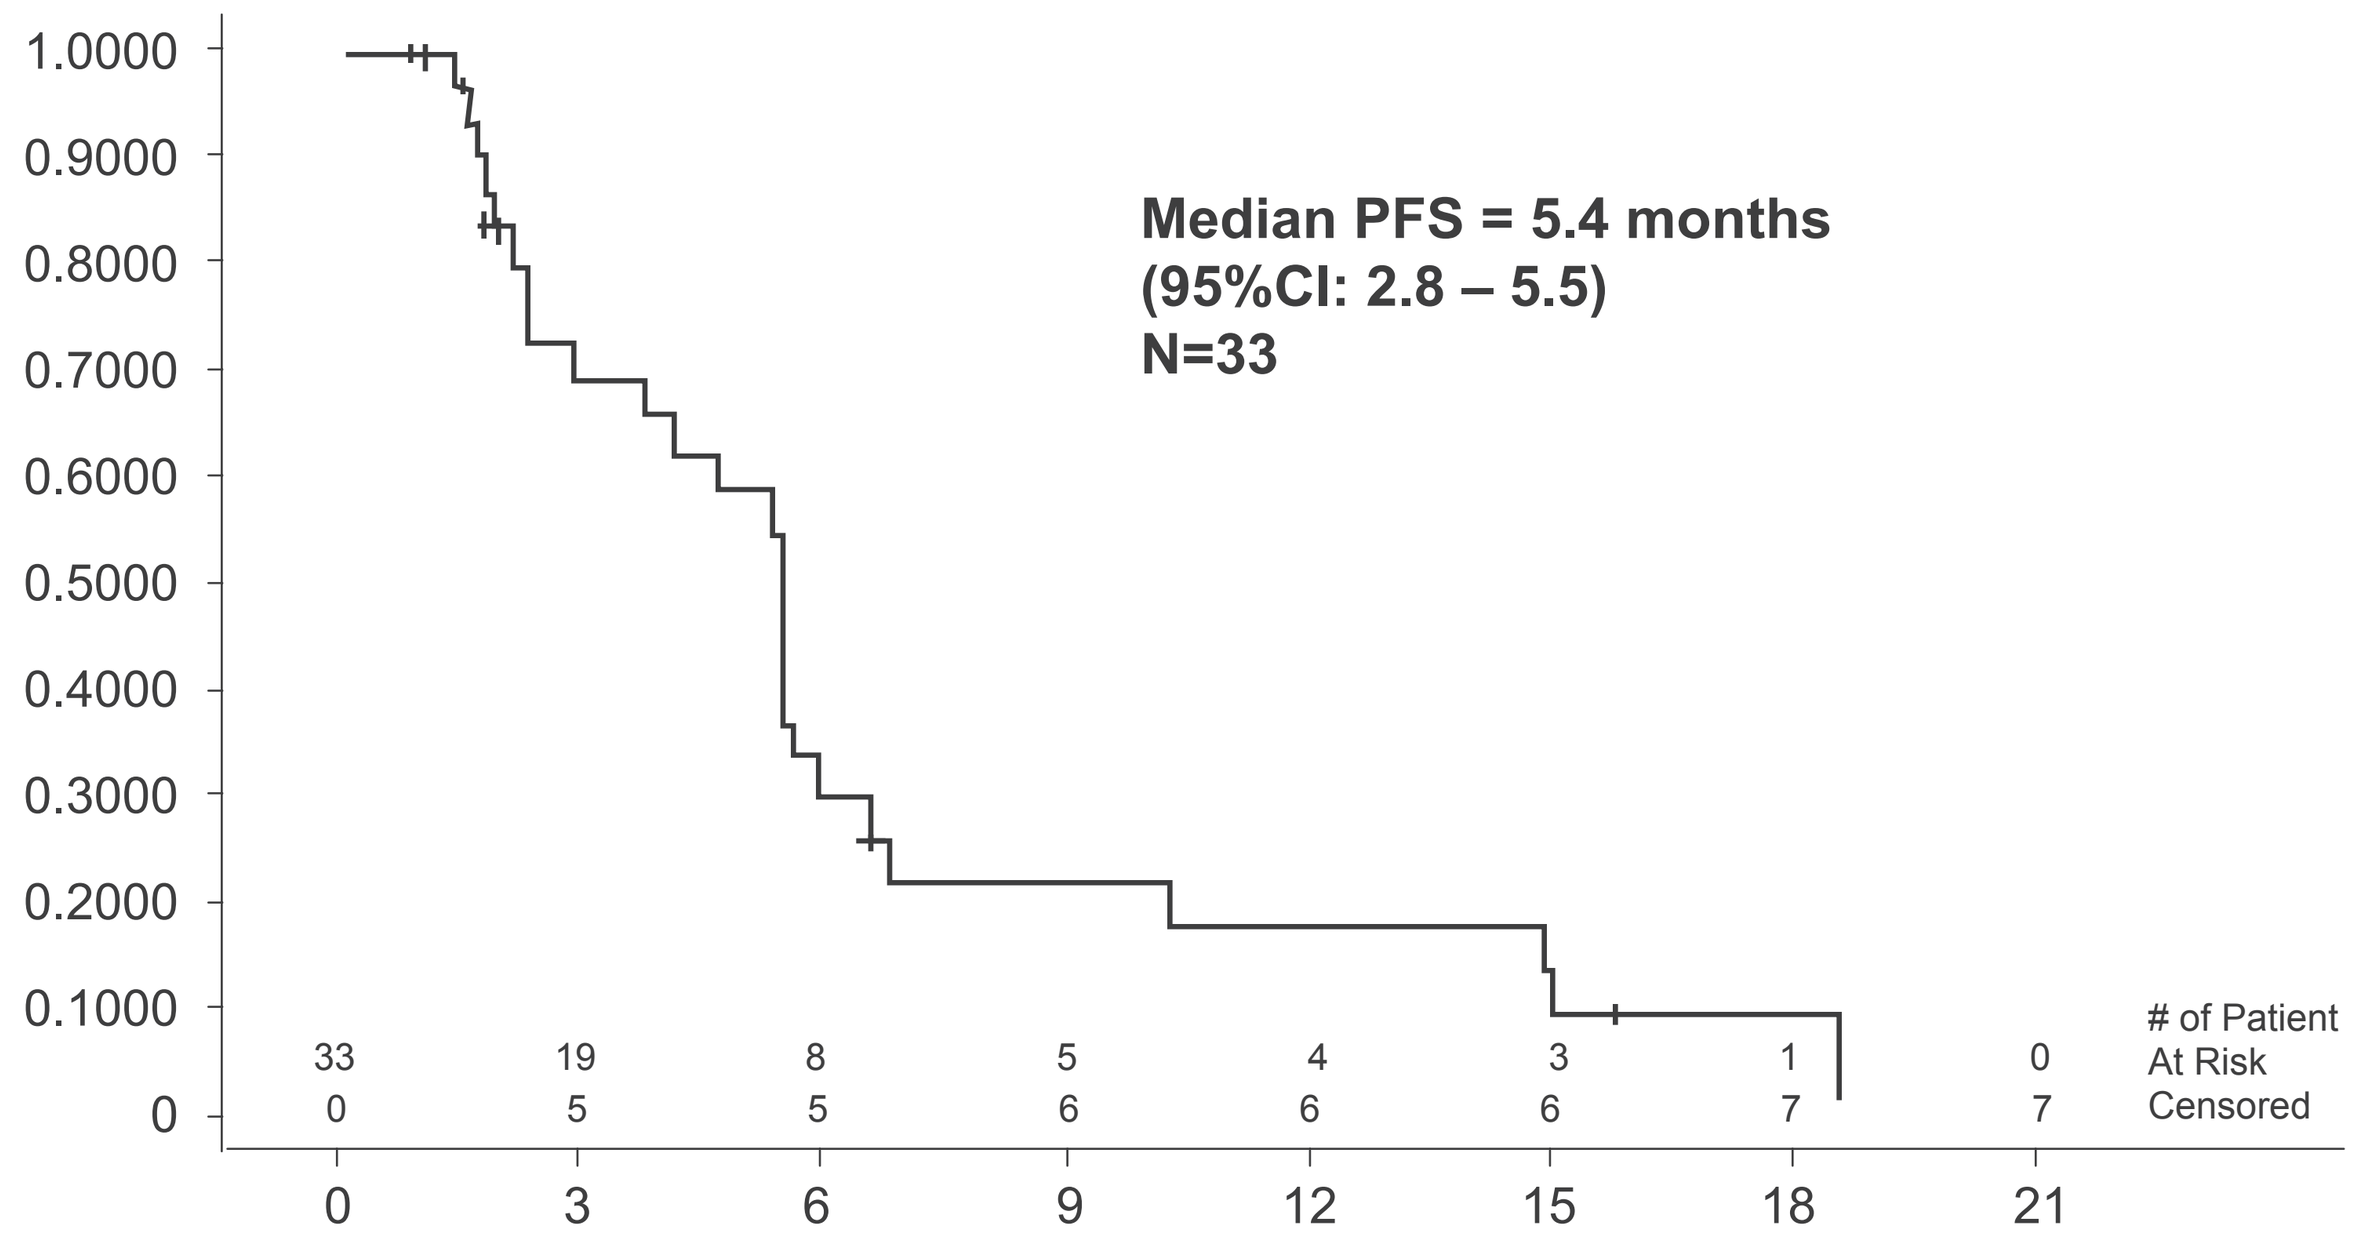
1.0000
0.9000
0.8000
0.7000
0.6000
0.5000
0.4000
0.3000
0.2000
0.1000
0
Median PFS = 5.4 months
(95%CI: 2.8 – 5.5)
N=33
33 19 8 5 4 3 1 0
0 5 5 6 6 6 7 7
of Patient
At Risk
Censored
0 3 6 9 12 15 18 21

Kaplan-Meier graph of progression-free survival
Analysis includes the intent-to-treat population. Tickmarks show censored data.
Source: Data as of May 9, 2011

Overall Survival

(All patients previously treated with PLD – dose groups pooled)

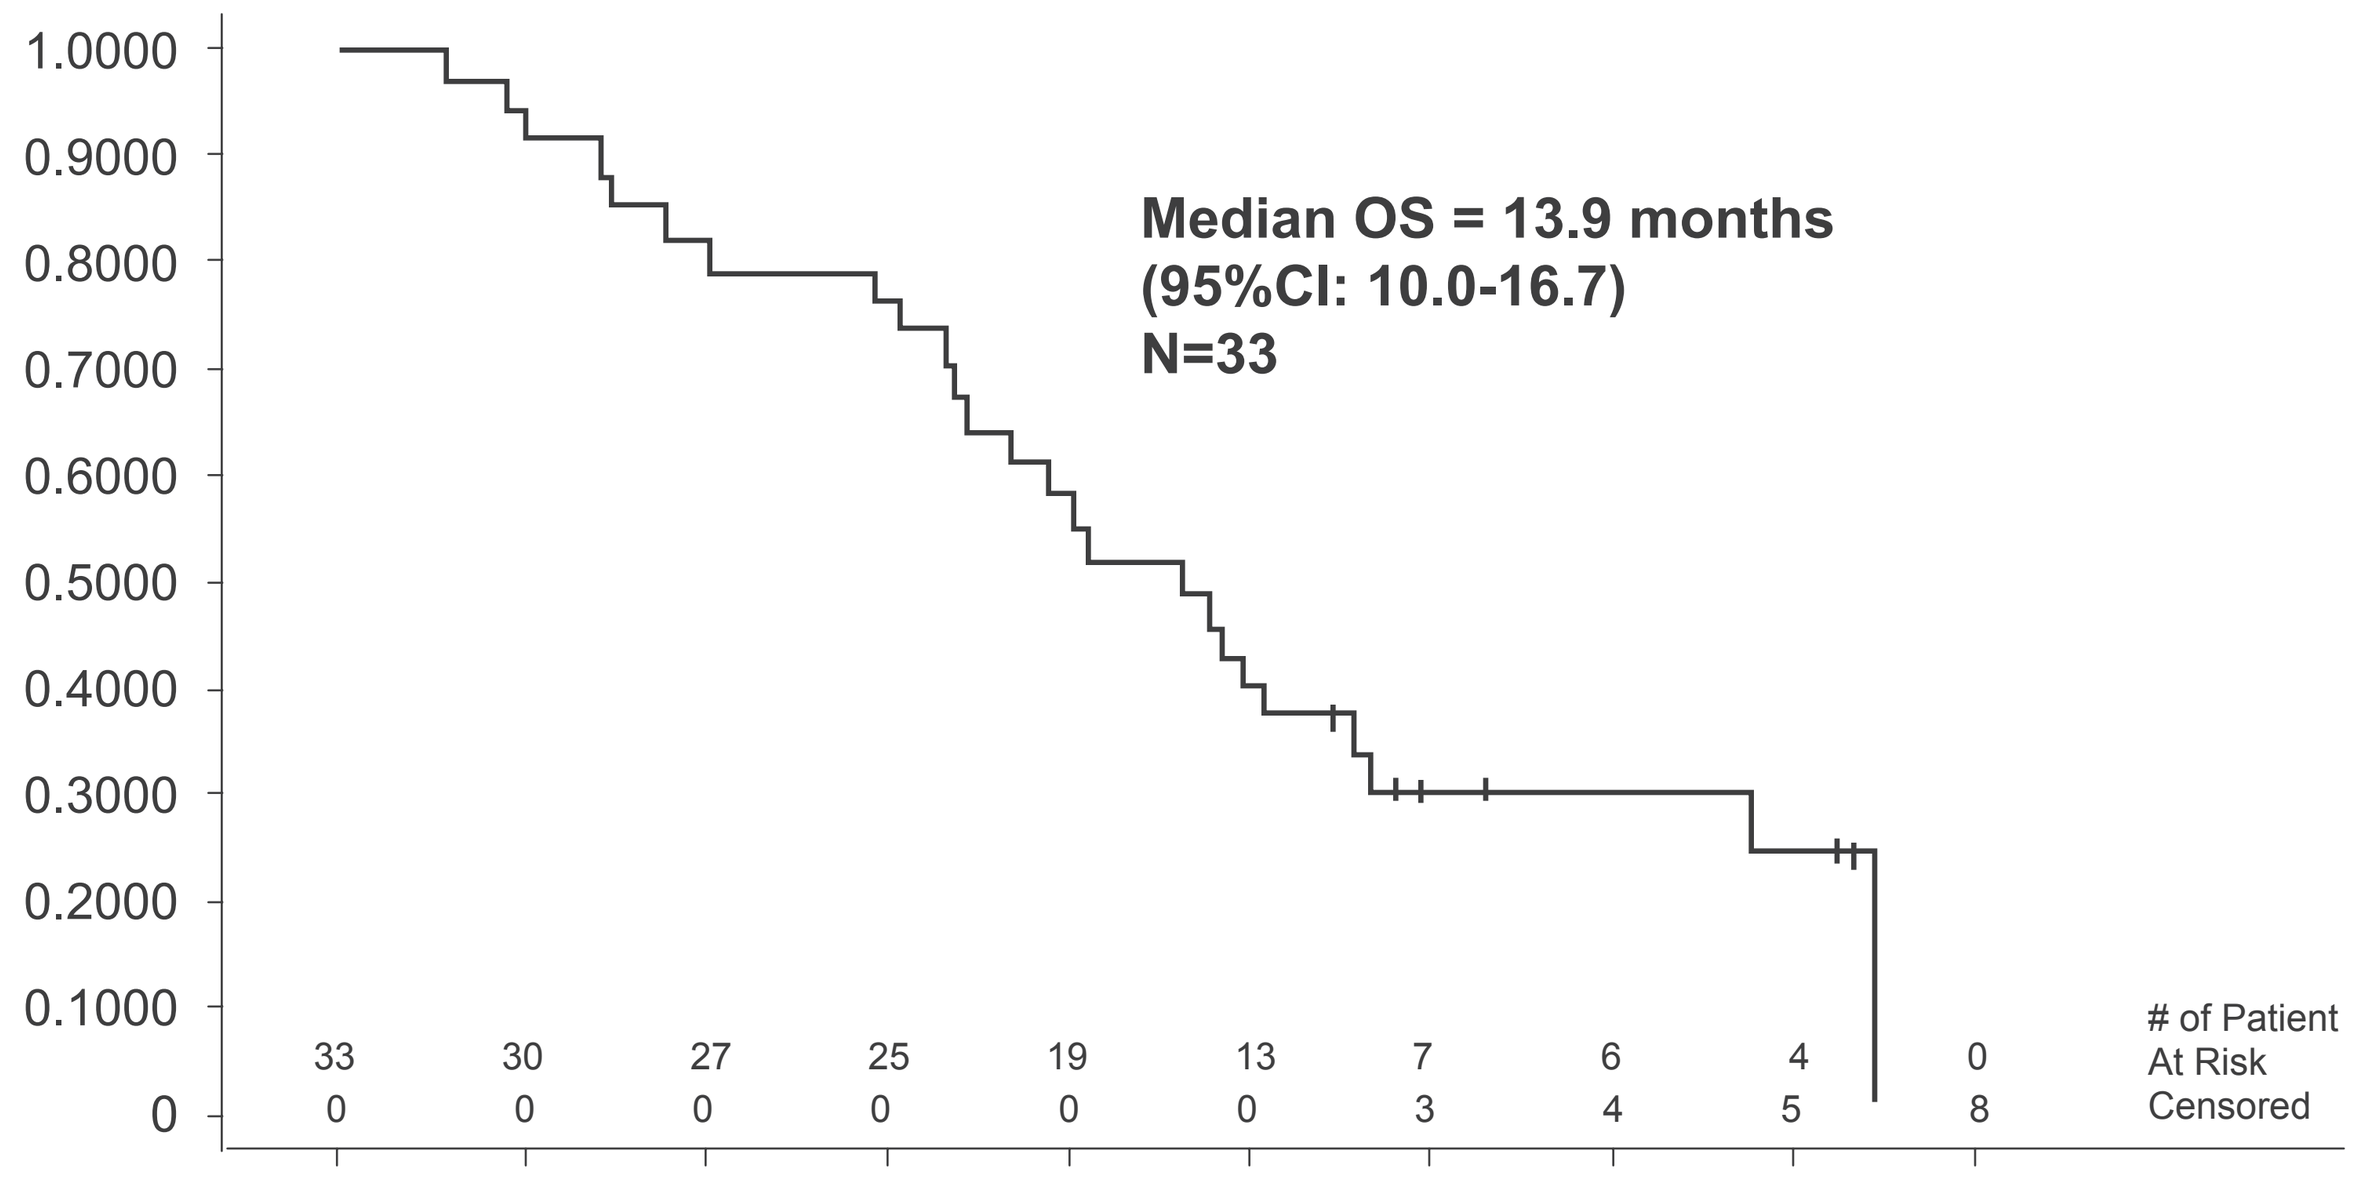
1.0000
0.9000
0.8000
0.7000
0.6000
0.5000
0.4000
0.3000
0.2000
0.1000
0
Median OS = 13.9 months
(95%CI: 10.0-16.7)
N=33
33 30 27 25 19 13 7 6 4 0
0 0 0 0 0 0 3 4 5 8
of Patient
At Risk
Censored
0 3 6 9 12 15 18 21 24 27

Kaplan-Meier graph of overall survival
Analysis includes the intent-to-treat population. Tickmarks show censored data.
Source: Data as of May 9, 2011

Conclusions

- NKTR-102 is new and novel approach to treating patients with platinum resistant/refractory ovarian cancer (PROC)
- NKTR-102 highly active in platinum resistant/refractory patients who previously received treatment with PLD
 - Confirmed response rate 19% for the q14d schedule, 21% for the q21d schedule
 - Duration of confirmed response: 4.2 months/q14d, 4.4 months/q21d
 - Median PFS (combined): 5.4 months
 - Median OS (combined): 13.9 months
- Q21d schedule identified as the optimal regimen for Phase 3 testing due to better safety profile than the q14d schedule. The most common treatment related Grade ≥ 3 toxicities were late diarrhea and fatigue. Grade 4 toxicity was uncommon, occurring in only 1 patient in the q21 day schedule.
- No febrile neutropenia in the q21d schedule or clinically significant neuropathy in either schedule
- This study is currently enrolling an additional 110 platinum-resistant/refractory ovarian cancer patients who have progressed after receiving PLD therapy to further evaluate single agent NKTR-102 in this population with a high degree of unmet medical need.
- NKTR-102 is being evaluated in multiple cancer indications as a single and combination agent. Phase 3 planning is underway in ovarian and breast cancers.

Safety

Safety: Summary of Drug-Related Adverse Events

Most Common Drug-related Grade 3/4 Adverse Events > 5% and Febrile Neutropenia N (%) ALL PATIENTS	145 mg/m² Q14 days N=36		145 mg/m² Q21 days N=35	
	Grade 3	Grade 4	Grade 3	Grade 4
Diarrhea	9 (25.0%)	1 (2.8%)	5 (14.3%)	0
Dehydration	9 (25.0%)	0	3 (8.6%)	0
Hypokalemia	7 (19.4%)	1 (2.8%)	3 (8.6%)	0
Nausea	7 (19.4%)	0	1 (2.9%)	0
Fatigue	2 (5.6%)	0	5 (14.3%)	0
Hyponatraemia	4 (11.1%)	1 (2.8%)	1 (2.9%)	0
Neutropenia	2 (5.6%)	0	2 (5.7%)	1 (2.9%)
Vomiting	4 (11.1%)	0	1 (2.9%)	0
Anaemia	4 (11.1%)	0	0	0
Febrile neutropenia	2 (5.6%)	1 (2.8%)	0	0

Most Common Drug-related Grade 3/4 Adverse Events > 5% and Febrile Neutropenia N (%) PTS PREVIOUSLY TREATED WITH PLD	145 mg/m² Q14 days N=16		145 mg/m² Q21 days N=17	
	Grade 3	Grade 4	Grade 3	Grade 4
Diarrhea	5 (31.3%)	0	3 (17.6%)	0
Hypokalemia	5 (31.3%)	1 (6.3%)	0	0
Nausea	5 (31.3%)	0	1 (5.9%)	0
Dehydration	3 (18.8%)	0	1 (5.9%)	0
Fatigue	1 (6.3%)	0	3 (17.6%)	0
Vomiting	4 (25.0%)	0	0	0
Neutropenia	1 (6.3%)	0	1 (5.9%)	1 (5.9%)
Anaemia	2 (12.5%)	0	0	0
Decreased appetite	1 (6.3%)	0	1 (5.9%)	0
Hyponatraemia	2 (12.5%)	0	0	0
Lymphopenia	1 (6.3%)	0	1 (5.9%)	0

One patient in the q14d schedule died due to renal failure from dehydration, possibly related to treatment in the prior PLD subset.
One patient not previously treated with PLD died due to neutropenic sepsis which was possibly related to study drug treatment.

Abstract 5047 – Poster #15C Poster presented at the 2011 American Society of Clinical Oncology Annual Meeting : Ovarian Cancer Poster Session, June 3-6, 2011
To download a copy of this poster, please visit http://www.nektar.com