NKTR-105, A NOVEL PEGYLATED-DOCETAXEL, DEMONSTRATES SUPERIOR ANTI-TUMOR ACTIVITY VERSUS DOCETAXEL IN HUMAN NON-SMALL CELL LUNG AND COLON MOUSE XENOGRAFT MODELS

Ron Wolff[1], Sheri Routt[2], Renee Hartsook[1], Jennifer Riggs[1], Wen Zhang[1], Heather Persson[1], Randall Johnson[3]

[1]Nektar Therapeutics, CA, USA [2]Piedmont Research Center, NC, USA [3]Independent Consultant, NM, USA

Poster #448

Background

- NKTR-105 is a novel PEGylated conjugate of docetaxel, an anti-neoplastic agent belonging to the taxoid family that acts by disrupting the microtubular network in cells. Docetaxel is a well-established chemotherapeutic agent used in the treatment of breast, prostate, non-small cell lung cancer, gastric, and head and neck cancers.
- Using Nektar's small molecule PEGylation technology, NKTR-105 was created to improve the time-concentration profile and anti-tumor activity of docetaxel. The molecule is currently in preclinical development.
- As with NKTR-102 (PEG-irinotecan), sustained exposure to active chemotherapeutic agents is associated with enhanced anti-tumor activity at equitoxic doses.

Objectives

- To evaluate the comparative anti-tumor activity of NKTR-105 and docetaxel in colorectal (LoVo and LS174T) and non-small cell lung (H460) mouse xenograft models.

Methods

- In separate studies, LS174T, LoVo, and H460 tumor fragments exhibiting varying degrees of docetaxel sensitivity were implanted subcutaneously into female, athymic (Nu:Nu) nude mice.
- Mice bearing subcutaneously implanted tumors were dosed as described in Table 1 and Table 2.

Group	Treatment	Dose
1	Control	–
2	NKTR-105	33.3 mg/kg
3		25 mg/kg
4		18.7 mg/kg
5		14 mg/kg
6	Docetaxel	25 mg/kg
7		18.7 mg/kg
8		14 mg/kg
9		10 mg/kg

Group	Treatment	Dose
1	Control	–
2	NKTR-105	40 mg/kg
3		30 mg/kg
4		22.5 mg/kg
5		16.9 mg/kg
6	Docetaxel	30 mg/kg
7		22.5 mg/kg
8		16.9 mg/kg
9		12.7 mg/kg

- NKTR-105 and docetaxel doses were selected to be equitoxic and to achieve the maximum tolerated dose (MTD).
- Each treatment group (n = 10) received a total of three intravenous doses administered once each week (qwk x3) on days 1, 8, 15. Control groups were untreated (n = 10).
- Dosing commenced when tumors reached minimum starting size (8-120 mm^3 for LS174T and H460; 100-150 mm^3 for LoVo).
- Tumor volumes were measured twice weekly after the first drug injection.
- Animals were weighed daily for the first five days and twice weekly thereafter.
- Tumor endpoint volume was 1500 mm^3 for LS174T, 1000 mm^3 for LoVo, and 2000 mm^3 for H460.
- Animals were euthanized when their tumor reached endpoint volume. Any remaining animals were sacrificed on Days 79, 77, or 57 for the LS174T, LoVo, or H460 studies, respectively.
- Anti-tumor activity was measured as changes in tumor volume, time to reach endpoint, and complete or partial tumor regression.
- Treatment outcome was evaluated by Tumor Growth Delay (TGD), which is defined as the increase in median time to endpoint in a treatment group compared to the control group. Here it is expressed as %TGD, as a percentage of the median time to endpoint of the control group.
- Partial regression was defined as a tumor whose volume was reduced to 50% or less for three consecutive measurements compared to its volume on Day 1. Complete regression was defined as volume equal to or less than 13.5 mm^3 for three consecutive measurements.
- Acceptable toxicity was defined as a group mean body weight loss of less than 20% during the study and not more than one treatment-related death among ten treated animals. Any dosing regimen that resulted in greater toxicity was considered above the MTD.

Poster #448
Presented at the:
20th EORTC-NCI-AACR Symposium on "Molecular Targets and Cancer Therapeutics"
21–24 October 2008, Geneva, Switzerland

Results

H460 Non-Small Cell Lung Cancer Xenograft

Figure 1. NKTR-105 Demonstrates Significant Dose-Related Increases in TGD Relative to Docetaxel at MTD in H460 Xenografts.

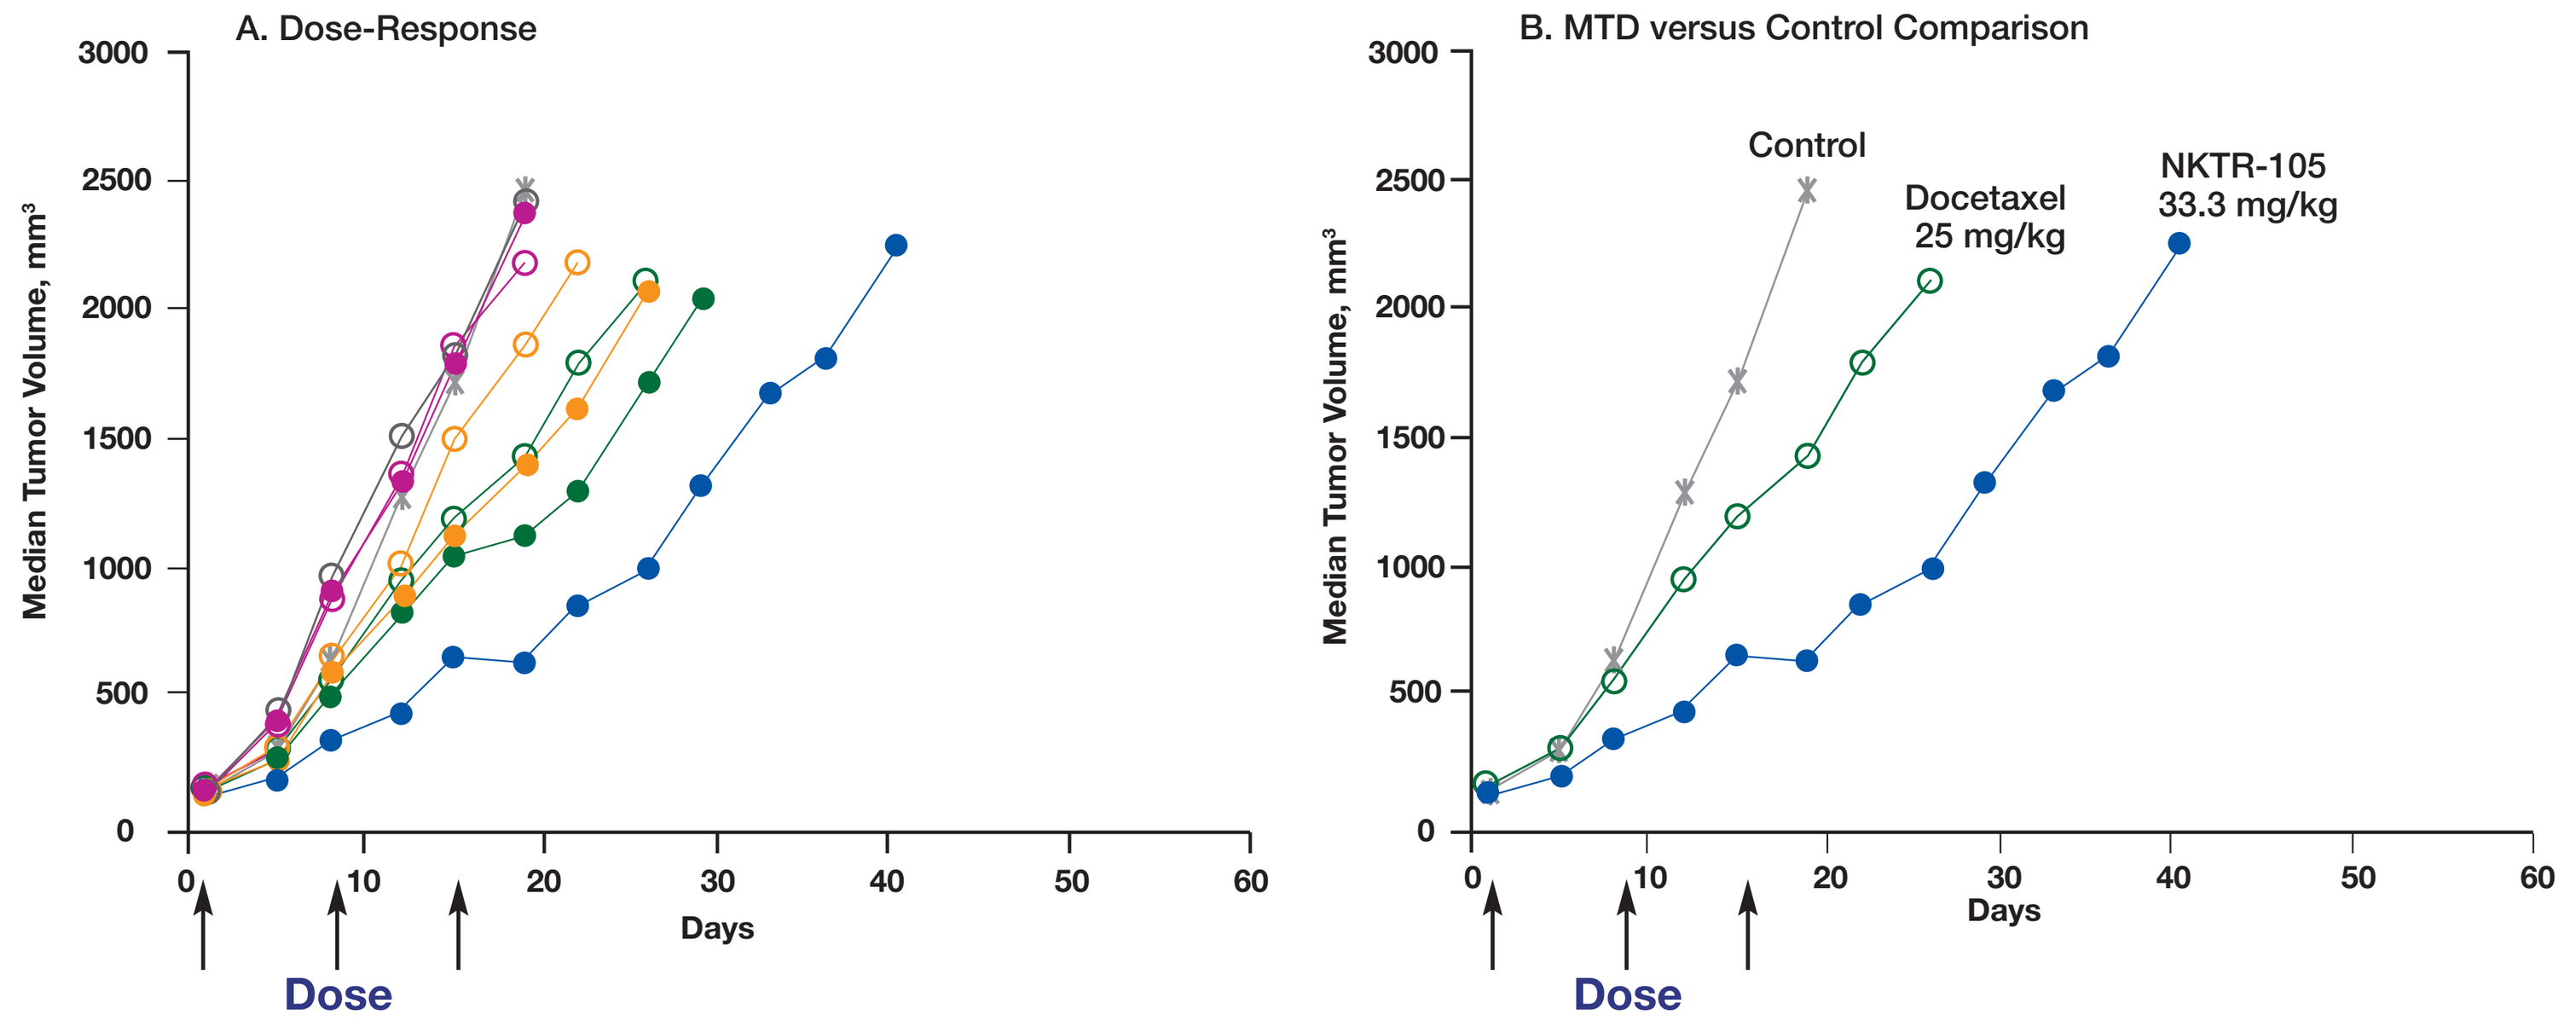

A. Dose-Response
B. MTD versus Control Comparison
Control
Docetaxel 25 mg/kg
NKTR-105 33.3 mg/kg
Median Tumor Volume, mm³
Days
Dose

	Symbol	Group	%TGD	Statistical Significance vs 1	vs 2	vs 3	vs 4	vs 5	PR	Mean Body Weight Nadir
1		Control	–	–	–	–	–	–	0	–
2		NKTR-105 33.3 mg/kg	122%	*p*<0.01	–	–	–	–	0	-14.2% Day 22
3		NKTR-105 25 mg/kg	72%	*p*<0.01	–	–	–	–	0	-10.4% Day 29
4		NKTR-105 18.7 mg/kg	56%	*p*<0.05	–	–	–	–	0	-6.2% Day 22
5		NKTR-105 14 mg/kg	-1%	ns	–	–	–	–	0	–
6		Docetaxel 25 mg/kg	48%	*p*<0.05	*p*<0.05	ns	–	–	0	-15.5% Day 26
7		Docetaxel 18.7 mg/kg	23%	ns	–	–	ns	–	0	-9.7% Day 22
8		Docetaxel 14 mg/kg	4%	ns	–	–	–	ns	0	-4.1% Day 19
9		Docetaxel 10 mg/kg	-2%	ns	–	–	–	–	0	–

%TGD = % Tumor Growth Delay as defined in Methods.
Statistical Significance by Logrank test: ns = not significant, — comparison not performed.
PR = Partial Regression as defined in Methods.
Mean Body Weight Nadir = lowest group mean body weight, as % change from Day 1;
— no decrease in mean body weight was observed.

- At MTD, 33.3 mg/kg of NKTR-105 resulted in a TGD of 122%, compared to a TGD of 48% for 25 mg/kg docetaxel (*p*<0.01).
- At the same dose of 25 mg/kg, NKTR-105 demonstrated a 72% TGD while docetaxel resulted in a 48% TGD (not statistically significant).
- Decreases in body weight were comparable between NKTR-105- and docetaxel-treated animals at MTD (14.2% and 15.5%, respectively).
- Leukopenia (decreased WBC, leukocytes, neutrophils) was first evident at Day 2 in docetaxel-treated animals, but not in NKTR-105–treated animals. This was transient and resolved by Day 4.

LoVo Colon Cancer Xenograft

Figure 2. NKTR-105 Demonstrates Partial Regressions at a Dose Above MTD and Increases in TGD Relative to Docetaxel in LoVo Xenografts.

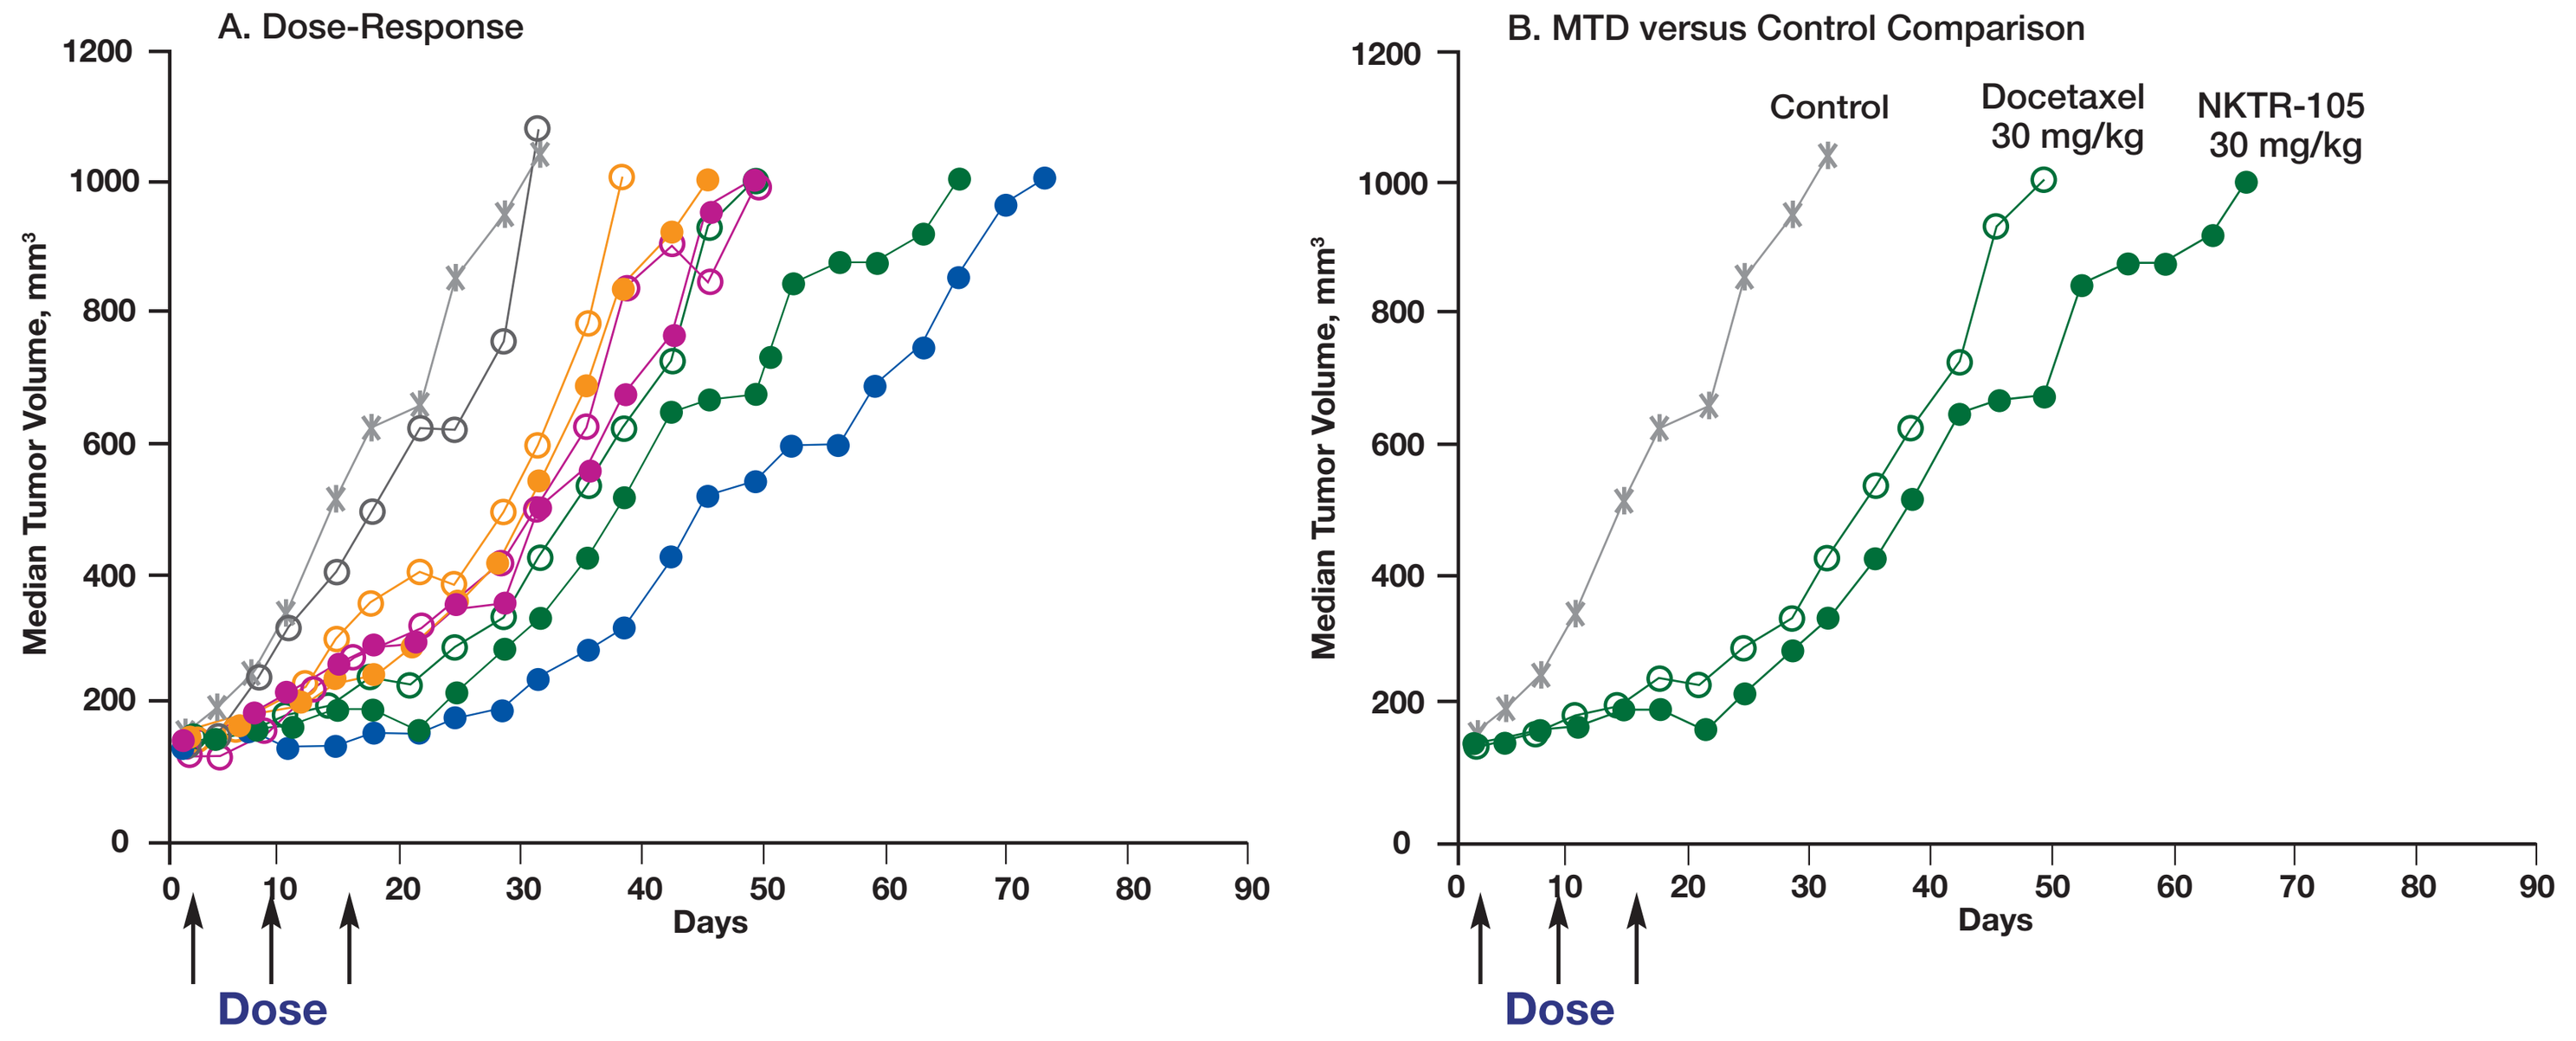

A. Dose-Response
B. MTD versus Control Comparison
Control
Docetaxel 30 mg/kg
NKTR-105 30 mg/kg
Median Tumor Volume, mm³
Days
Dose

	Symbol	Group	%TGD	Statistical Significance vs 1	vs 2	vs 3	vs 4	vs 5	PR	Mean Body Weight Nadir
1		Control	–	–	–	–	–	–	0	–
2		NKTR-105 40 mg/kg	148%	ne	–	–	–	–	2	-22.6% Day 24
3		NKTR-105 30 mg/kg	128%	*p*<0.001	–	–	–	–	0	-16.2% Day 24
4		NKTR-105 22.5 mg/kg	50%	*p*<0.001	–	–	–	–	0	-11% Day 24
5		NKTR-105 16.9 mg/kg	64%	*p*<0.001	–	–	–	–	0	-7% Day 21
6		Docetaxel 30 mg/kg	64%	*p*<0.001	ne	ns	–	–	0	-13.3% Day 21
7		Docetaxel 22.5 mg/kg	32%	*p*<0.01	–	–	ns	–	0	-14.6% Day 24
8		Docetaxel 16.9 mg/kg	70%	*p*<0.001	–	–	–	ns	0	-12.8% Day 21
9		Docetaxel 12.7 mg/kg	7%	ns	–	–	–	–	0	-5% Day 24

%TGD = % Tumor Growth Delay as defined in Methods.
Statistical Significance: ne = not evaluable, ns = not significant, — comparison not performed.
PR = Partial Regression as defined in Methods.
Mean Body Weight Nadir = lowest group mean body weight, as % change from Day 1;
— no decrease in mean body weight was observed.

- At MTD, 30 mg/kg of NKTR-105 resulted in a TGD of 128%, compared to a TGD of 64% for 30 mg/kg docetaxel. The difference, while pronounced, was not statistically significant.
- 40 mg/kg NKTR-105 was in excess of the MTD due to a body weight decrease of 22.6%. Two partial regressions were observed at this dose.
- Decreases in body weight were comparable between NKTR-105- and docetaxel-treated animals at MTD (16.2% and 13.3%, respectively).

LS174T Colon Cancer Xenograft

Figure 3. NKTR-105 Demonstrates Partial Regressions and a Significant Increase in TGD Relative to Docetaxel at MTD in LS174T Xenografts.

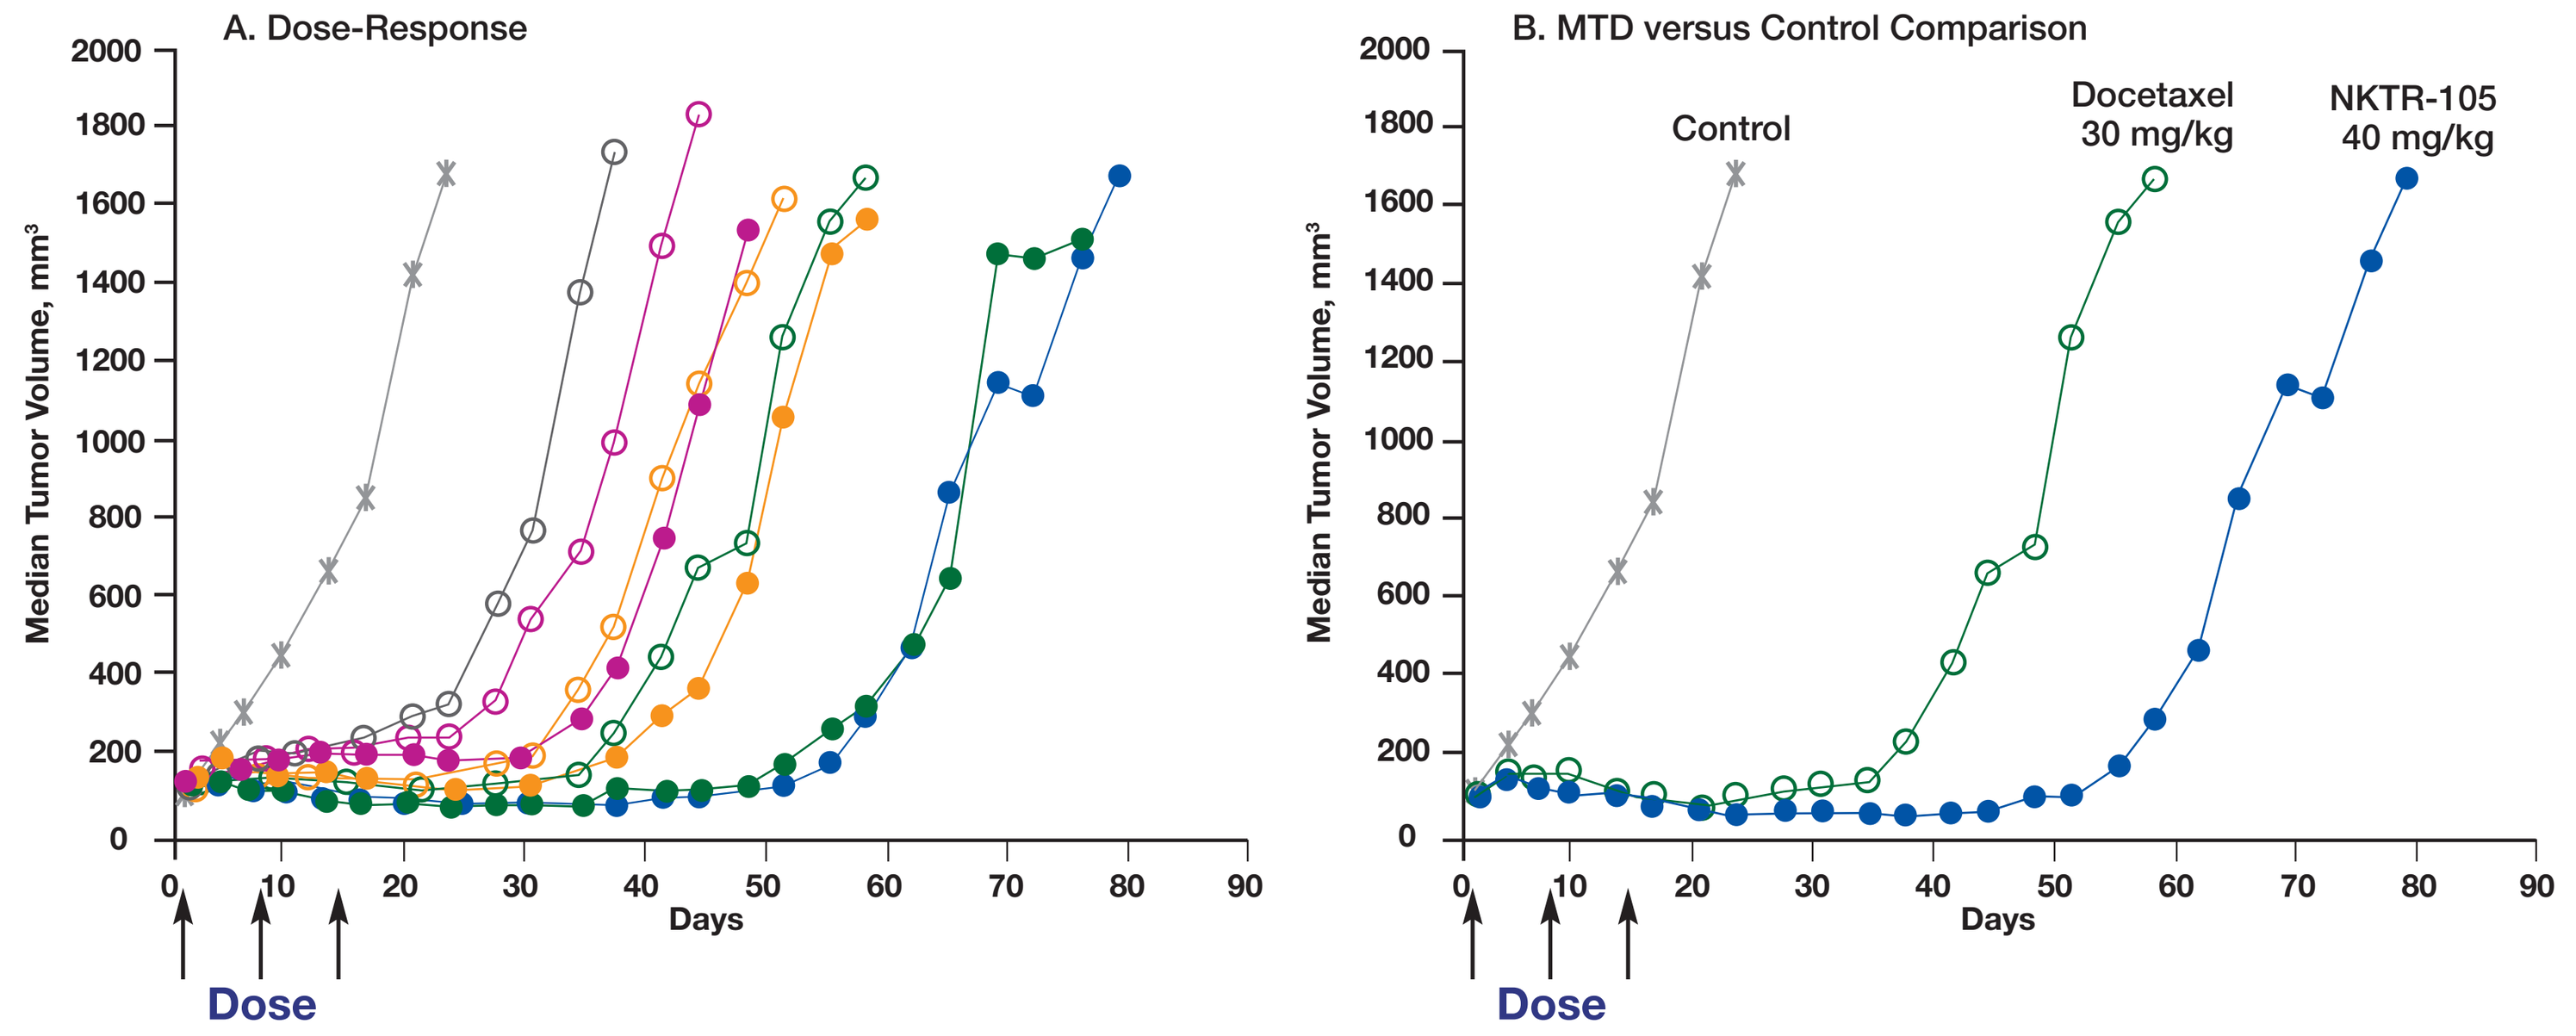

A. Dose-Response
B. MTD versus Control Comparison
Control
Docetaxel 30 mg/kg
NKTR-105 40 mg/kg
Median Tumor Volume, mm³
Days
Dose

	Symbol	Group	%TGD	Statistical Significance vs 1	vs 2	vs 3	vs 4	vs 5	PR	Mean Body Weight Nadir
1		Control	–	–	–	–	–	–	0	–
2		NKTR-105 40 mg/kg	266%	*p*<0.001	–	–	–	–	3	-18.8% Day 23
3		NKTR-105 30 mg/kg	250%	*p*<0.001	–	–	–	–	1	-17.9% Day 23
4		NKTR-105 22.5 mg/kg	169%	*p*<0.001	–	–	–	–	0	-11.1% Day 23
5		NKTR-105 16.9 mg/kg	126%	*p*<0.001	–	–	–	–	0	-12.4% Day 23
6		Docetaxel 30 mg/kg	166%	*p*<0.001	*p*<0.001	*p*<0.001	–	–	0	-16.6% Day 23
7		Docetaxel 22.5 mg/kg	135%	*p*<0.001	–	–	ns	–	0	-6.7% Day 23
8		Docetaxel 16.9 mg/kg	96%	*p*<0.001	–	–	–	*p*<0.05	0	-11.2% Day 23
9		Docetaxel 12.7 mg/kg	69%	*p*<0.001	–	–	–	–	0	-8.8% Day 23

%TGD = % Tumor Growth Delay as defined in Methods.
Statistical Significance: ns = not significant, — comparison not performed.
PR = Partial Regression as defined in Methods.
Mean Body Weight Nadir = lowest group mean body weight, as % change from Day 1;
— no decrease in mean body weight was observed.

- At MTD, NKTR-105 demonstrated three partial regressions.
- At MTD, 40 mg/kg of NKTR-105 resulted in a TGD of 266%, compared to a TGD of 166% for 30 mg/kg docetaxel (*p*<0.001).
- At the same dose of 30 mg/kg, NKTR-105 demonstrated a 250% TGD and one partial regression while docetaxel resulted in a 166% TGD and no regressions.
- Decreases in body weight were comparable between NKTR-105- and docetaxel-treated animals at MTD (18.8% and 16.6%, respectively).
- At Day 20, leukopenia was evident in all docetaxel-treated groups, but only at the two highest doses of NKTR-105 (40 and 30 mg/kg).

Conclusions

- NKTR-105 demonstrates significantly greater anti-tumor activity than docetaxel in H460 and LS174T mouse xenograft models. Partial regressions were observed in two of three cell lines for NKTR-105, while no regressions were observed for docetaxel.
- At MTD, the %TGD for NKTR-105 was 2.5-, 2.0-, and 1.6-fold greater than docetaxel in H460, LoVo, and LS174T xenograft models, respectively.
- NKTR-105 is a promising drug candidate that warrants study in clinical trials.

Nektar Therapeutics, 201 Industrial Road, San Carlos, CA 94070, USA

©2008 Nektar Therapeutics.