NKTR-118, AN ORAL PERIPHERAL OPIOID ANTAGONIST, HAS LOW POTENTIAL FOR DRUG-DRUG INTERACTIONS

Aleksandrs Odinecs, PhD, Yuan Song, PhD, Shibani Harite, MS, Myong Gyong Lee, PhD, Alan R Kugler, PhD*, and Michael A Eldon, PhD, FCP
Nektar Therapeutics, San Carlos, CA

NEKTAR

Background

Nektar Therapeutics is developing NKTR-118, a PEGylated derivative of naloxone, for the treatment of opioid-induced constipation (OIC) and other manifestations of opioid-induced bowel dysfunction (OBD) in patients receiving opioid therapy for pain.

PEGylation of naloxone alters its distribution (reduced CNS penetration) and metabolism (reduced first pass effect) while its opioid antagonist properties are retained.

NKTR-118 was selected from a series of PEGylated naloxone candidates in part because of its greater metabolic stability observed in animal models.

This presentation combines a variety of *in vitro* and clinical experimental results to assess the potential of NKTR-118 to cause and be susceptible to drug-drug interactions.

Objectives

To evaluate the drug-drug interaction potential of NKTR-118 using *in vitro* methods.

To investigate whether concurrent administration of NKTR-118 and morphine affects the pharmacokinetics of morphine and its glucuronide metabolites.

In vitro Methods

Metabolic stability of NKTR-118 in the presence of human CYP isoforms (CYP1A2, CYP2C9, CYP2C19, CYP2D6, CYP3A4, and CYP2C8) expressed in Bactosomes™.

Metabolic stability and formation of NKTR-118-Glucuronide mediated by Uridine Diphosphate Glucuronyltransferase (UDPGT) in cryopreserved dog and human hepatocytes.

Inhibition by NKTR-118 of human P450 isoform-specific metabolism (CYP1A, CYP2C9, CYP2C19, CYP2D6, and CYP3A4) in human liver microsomes.

Induction by NKTR-118 of human P450 isoforms (CYP1A2 or CYP3A4) in fresh human hepatocytes from 3 individual donors.

Protein binding in human plasma (equilibrium dialysis with overnight incubation).

In vitro Results

Only a small portion of NKTR-118 was metabolized during a 2 hr incubation with human hepatocytes, indicating that it has relatively high metabolic stability. Naloxone under the same conditions was extensively metabolized during the first hour of incubation (Figure 1).

Incubation in human hepatocytes produced little glucuronide conjugates whereas dog hepatocytes produced significant amounts.

Figure 1. Stability of NKTR-118 and Naloxone in Cryopreserved Human Hepatocytes

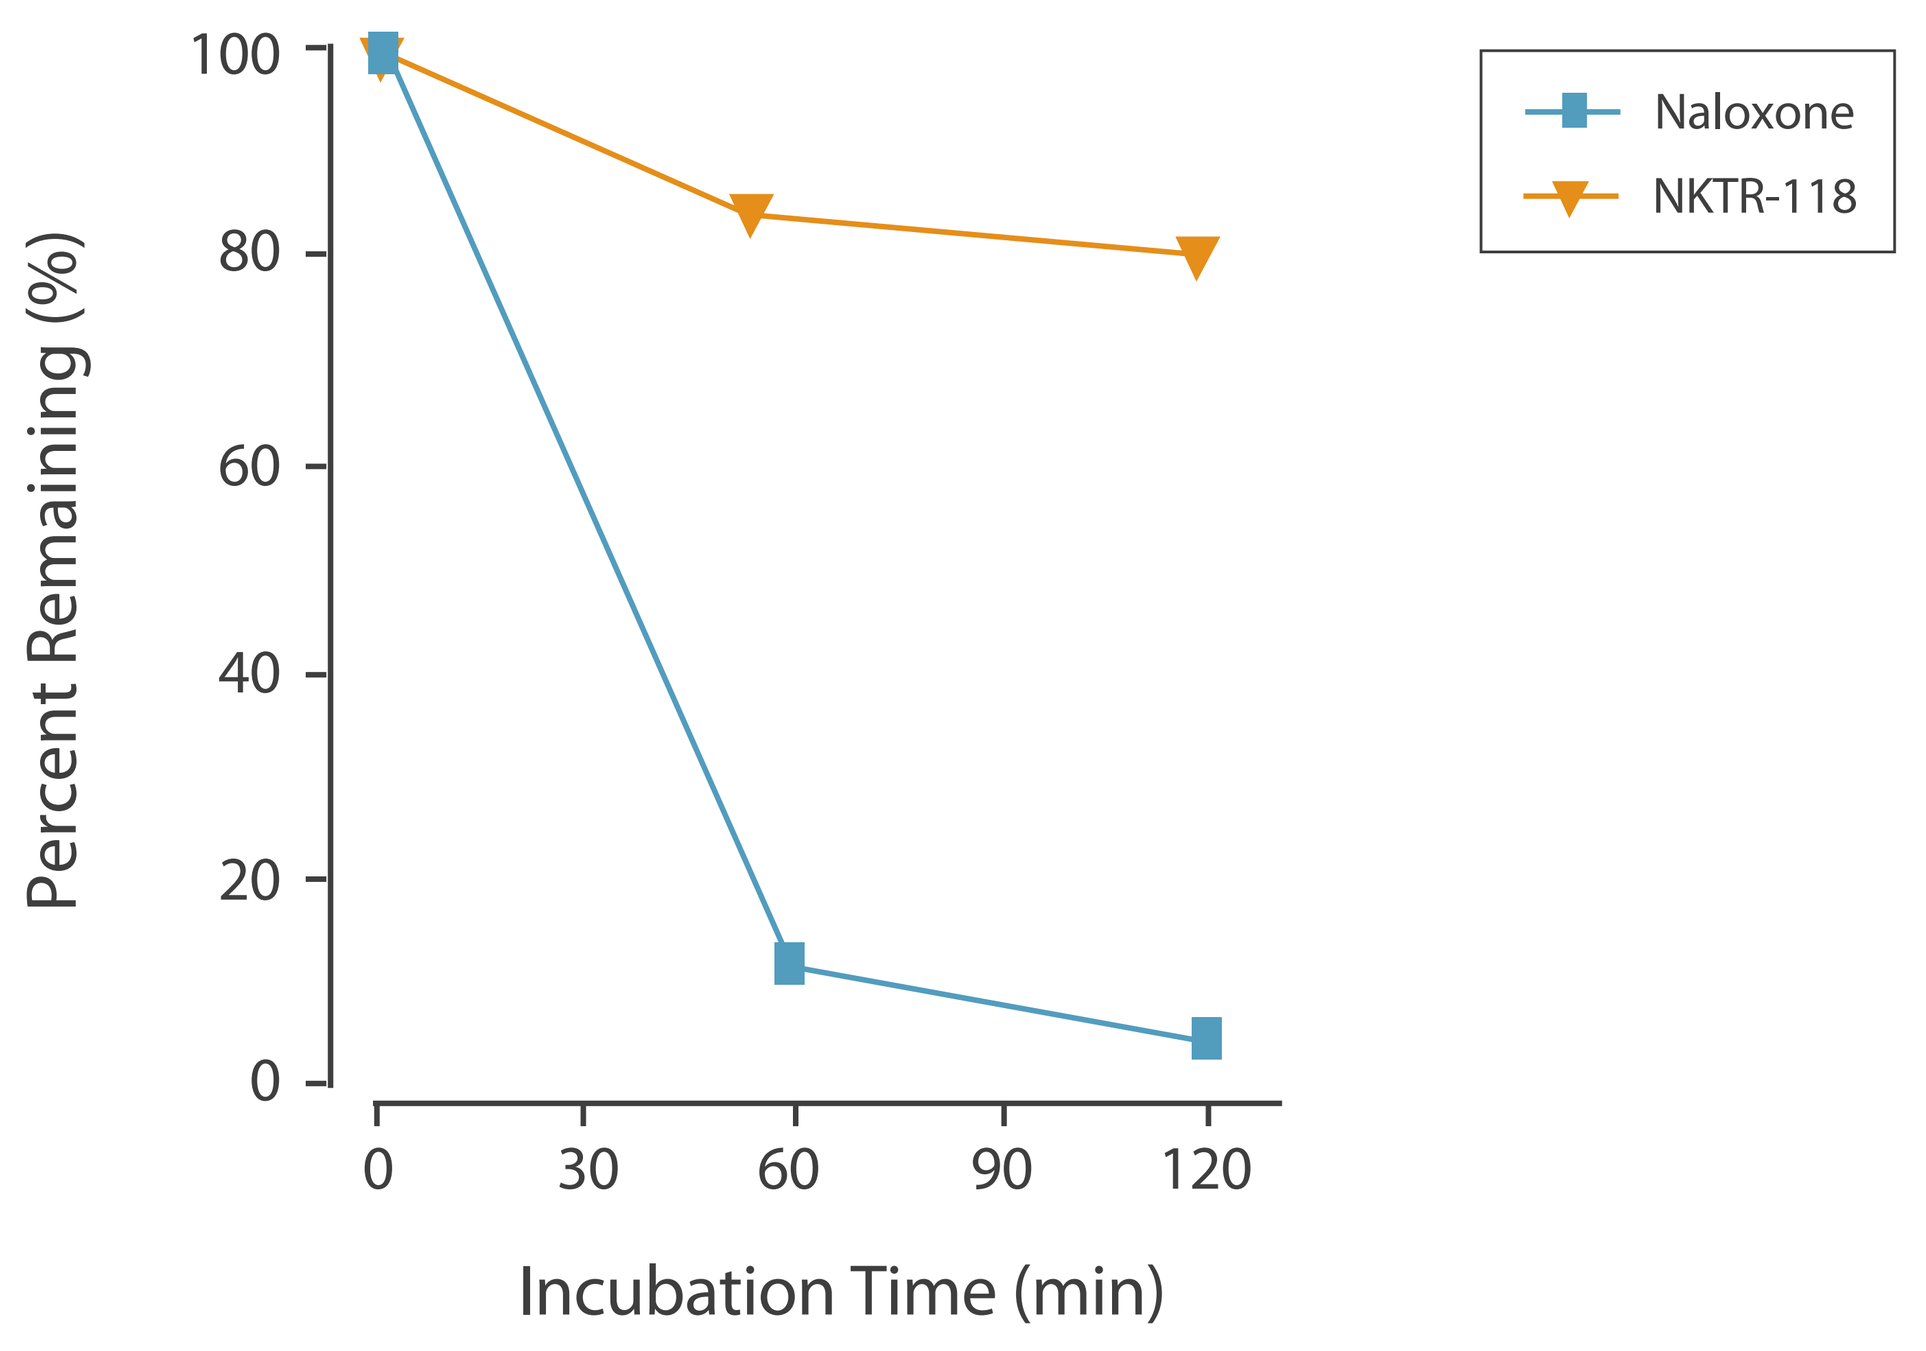
Naloxone
NKTR-118
Percent Remaining (%)
100
80
60
40
20
0
0
30
60
90
120
Incubation Time (min)

NKTR-118 is metabolically stable in the presence of CYP1A2, CYP2C19, CYP2C9, CYP2D6, and CYP2C8; it is metabolized *in vitro* only by CYP3A4.

NKTR-118 is a weak inhibitor of CYP2D6 (IC50 = 84.7 μM). No inhibition of CYP1A, CYP2C9, and CYP3A4 (testosterone) occurred and inhibition of CYP2C19 and CYP3A4 (midazolam) was 38.2 and 41.4%, respectively, in 100 μM NKTR-118. Since IC50 values for all tested P450 isoforms are much higher than therapeutically relevant plasma NKTR–118 concentrations, the results of this study suggest that NKTR-118 will not inhibit major human P450 isoforms (CYP1A, CYP2C9, CYP2C19, CYP2D6, and CYP3A4) *in vivo*.

No induction of CYP1A2 or CYP3A4 by NKTR-118 was observed in any of the three hepatocyte samples.

The unbound fraction of NKTR-118 in human plasma is 80–100%.

In vivo Methods

The effect of concurrent administration of NKTR-118 and morphine on the pharmacokinetics of morphine and its glucuronide metabolites was assessed in a randomized, double-blind, placebo-controlled, single-dose, dose escalation, crossover study in 46 healthy subjects who received IV morphine to investigate the potential for NKTR–118 to antagonize morphine-induced delay in oral-cecal transit time[1].

Subjects received 5 mg/70 kg IV morphine alone and with NKTR-118 at doses from 8 to 1000 mg (N = 6 in each of 8 dose levels) Serial blood samples were collected for pharmacokinetic comparison between treatments.

Plasma NKTR-118, NKTR-118-Glucuronide, morphine, morphine-3-glucuronide, and morphine-6-glucuronide concentrations were determined using validated LC-MS/MS methods.

In vivo Results

Plasma NKTR-118-Glucuronide concentrations were ~100x less than those of NKTR-118, consistent with *in vitro* results.

Mean plasma morphine and its metabolites concentration-time profiles pooled across dose cohorts were essentially superimposable, independent of treatment, as shown in Figure 2.

Ratios of Morphine+NKTR-118 to Morphine+Placebo Least-Square Mean Cmax and AUC values for morphine and both metabolites were between 90.8 and 101.5% (see table), indicating a high degree of similarity in morphine pharmacokinetics between treatments.

Ratio of (Morphine+NKTR-118) / (Morphine+Placebo) Least-Square Means (% of (Morphine+Placebo) Least-Square Mean)

Analyte	C_{max}	$AUC_{(0\text{-}last)}$	$AUC_{(0\text{-}inf)}$
Morphine	92.8	90.8	91.2
Morphine-3-Glucuronide	98.9	100.5	95.4
Morphine-6-Glucuronide	97.1	101.5	97.3

Overall, NKTR-118 at doses as high as 1000 mg did not affect the pharmacokinetics of morphine or its glucuronide metabolites.

Figure 2. Mean (±SD) Plasma Morphine and its Glucuronide Metabolites Concentration-Time Profiles Pooled Across Dose Cohorts

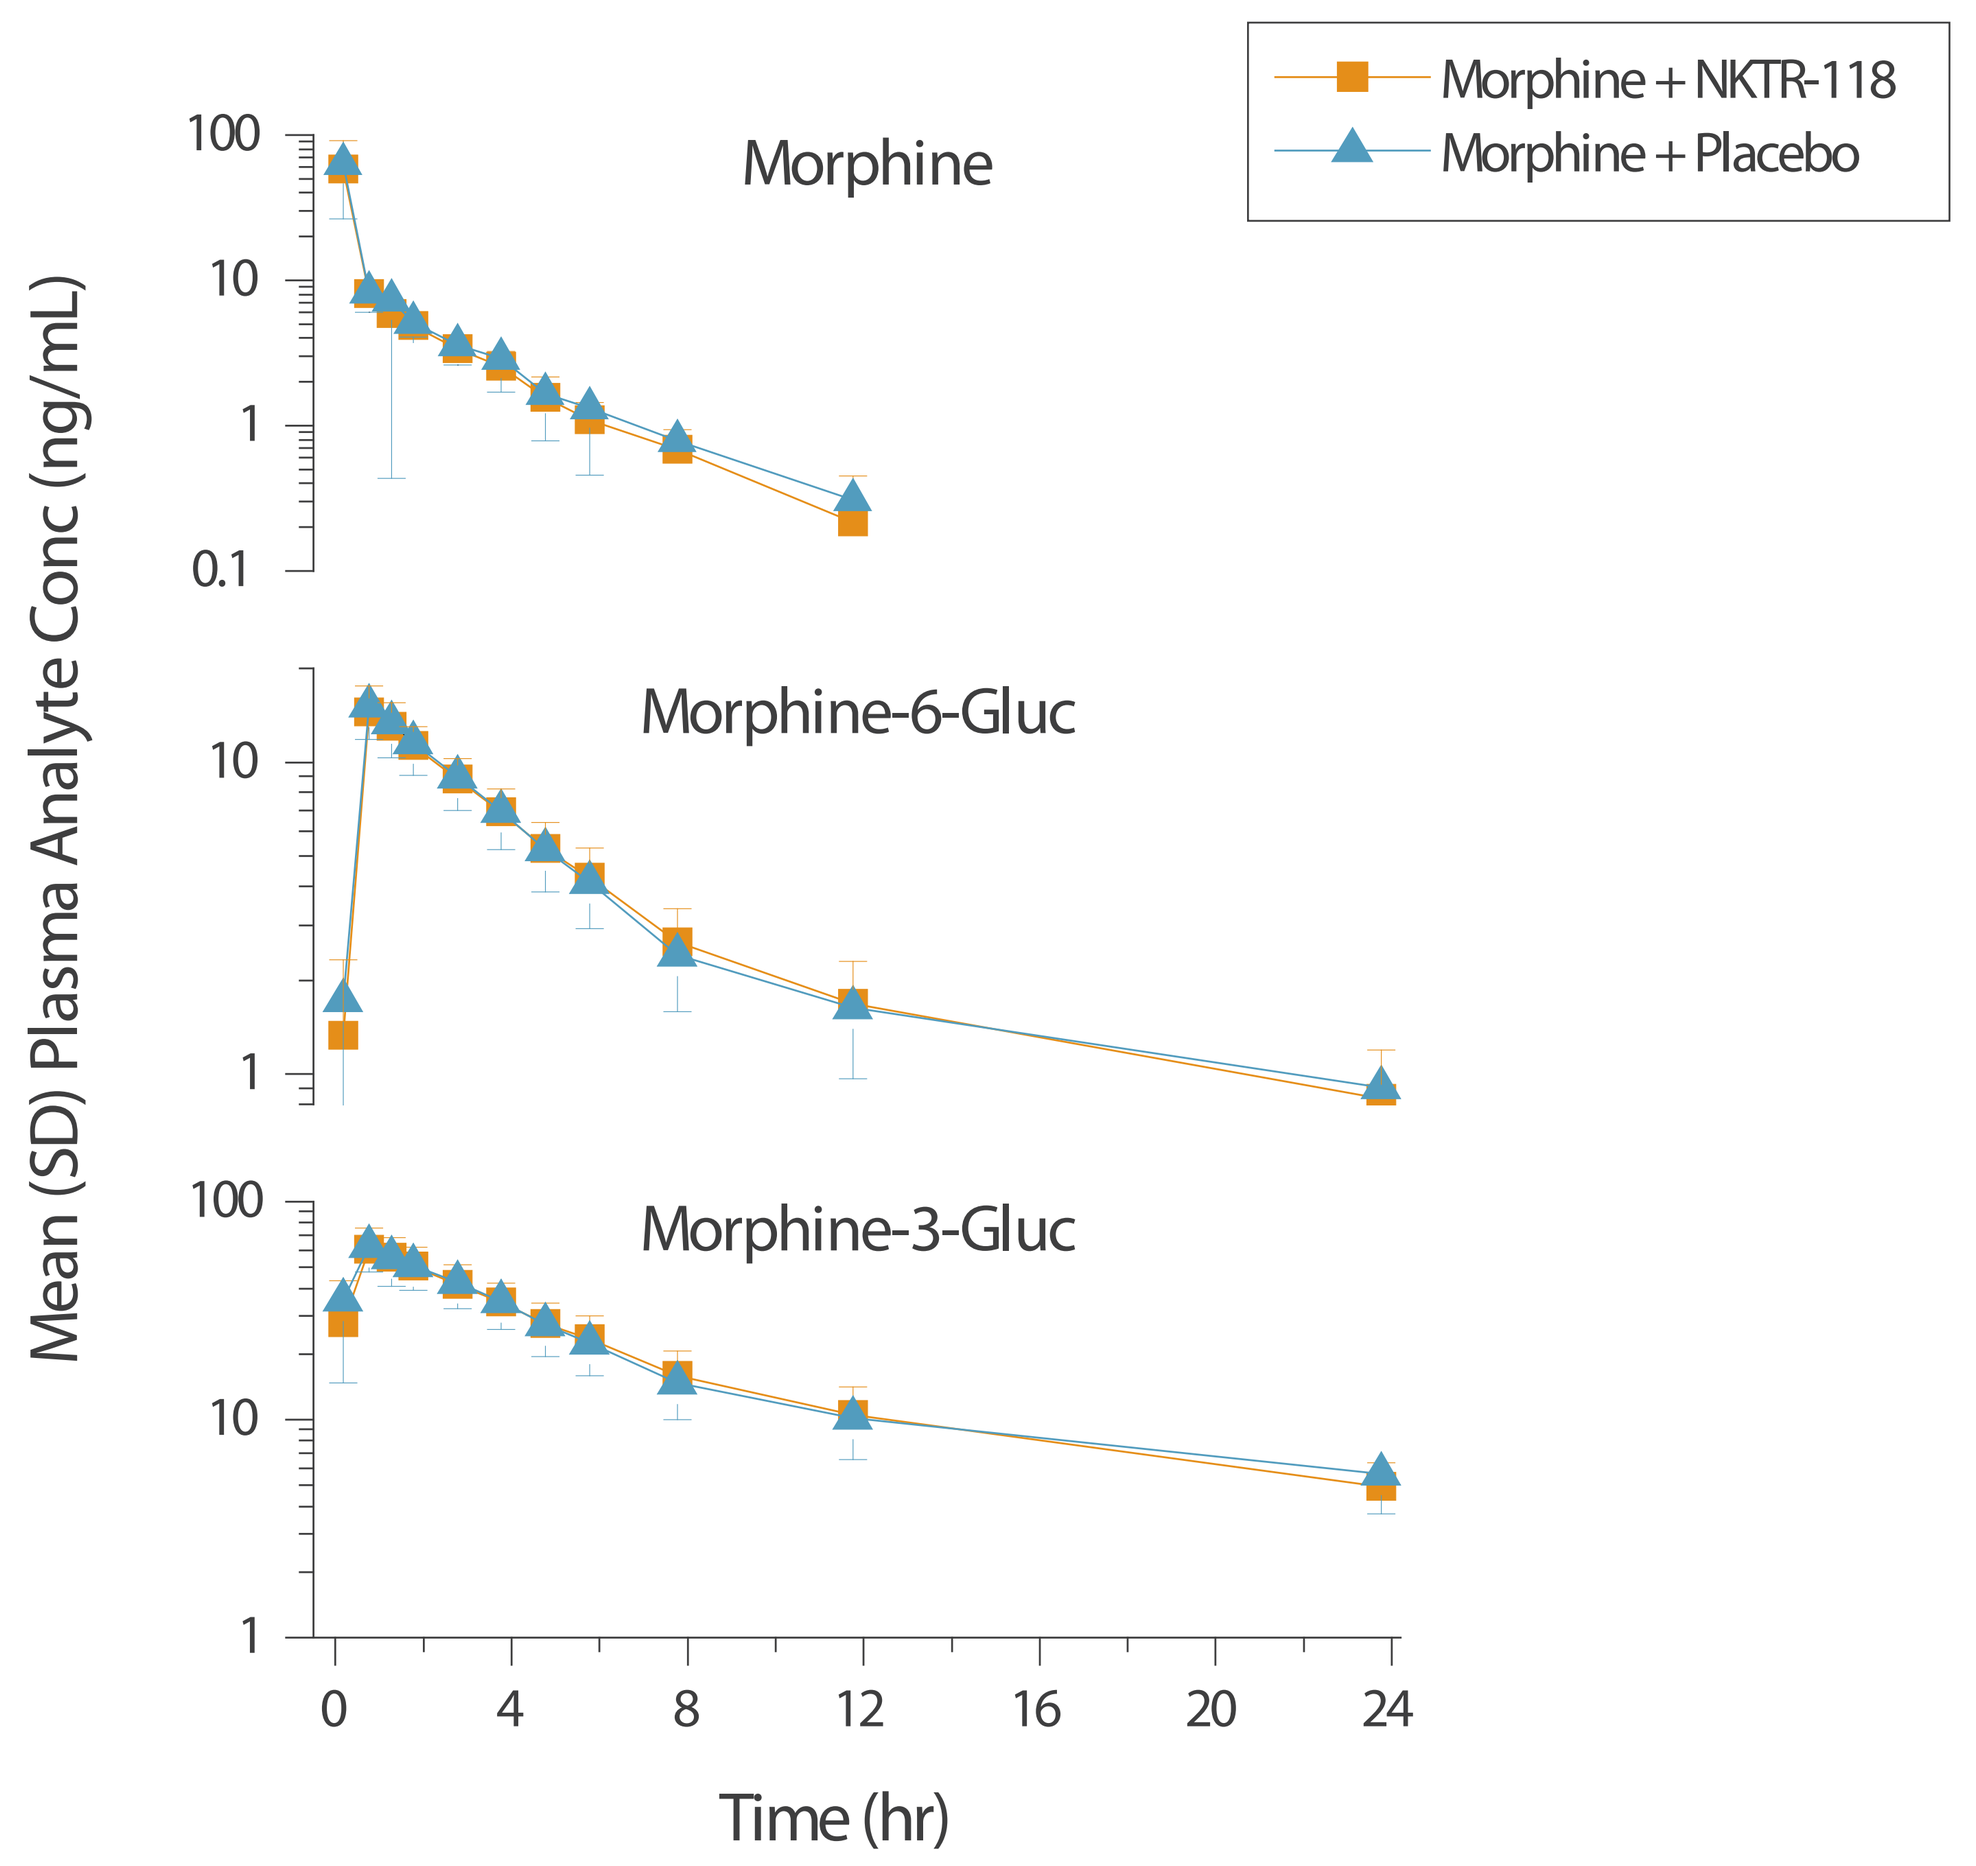
Morphine + NKTR-118
Morphine + Placebo
Morphine
Morphine-6-Gluc
Morphine-3-Gluc
Mean (SD) Plasma Analyte Conc (ng/mL)
100
10
1
0.1
0
4
8
12
16
20
24
Time (hr)

Conclusion

- Clinically important DDIs with NKTR-118 are not expected based on its *in vitro* profile of low or no inhibition and no induction of Cytochrome P450 isoforms, low affinity to human UDPGT, and low extent of plasma protein binding.
- The lack of effect of concomitant NKTR-118 on the pharmacokinetics of morphine and its glucuronide metabolites confirms the *in vitro* prediction based on low affinity to UDPGT.
- NKTR-118 carries low risk for mediating clinically significant drug-drug interactions.

Acknowledgments:
We would like to thank Cyprotex (Macclesfield, UK) and XenoTech (Lenexa, KS) for conducting in vitro studies and Kendle Clinical Pharmacology Unit (Utrecht, The Netherlands) for conducting the clinical study.

*Alan R Kugler presently with Coastal Pharma Group; Montara, CA

[1] Poster 36, ACCP 36th Annual Meeting; September 9, 2007; San Francisco, CA.

Presented at the American College of Clinical Pharmacology 38th Annual Meeting; September 13–15, 2009; San Antonio, TX (Poster 141).

© 2009 Nektar Therapeutics.