Pharmacokinetics and Pharmacodynamics of Oral NKTR-181, a Novel Opioid Analgesic: Results of a Single Ascending Dose Phase 1 Study

NEKTAR

[1]Lynn Webster, MD, [1]Matthew Iverson, MPH, [2]Robert Medve, MD, [2]Aleksandrs Odinecs, PhD and [2]Michael A Eldon, PhD, FCP
[1]Lifetree Clinical Research, Salt Lake City, UT; [2]Nektar Therapeutics, San Francisco, CA

Conclusions

- NKTR-181 produces a dose-dependent central analgesic response in healthy subjects, as measured by hand withdrawal latency in the cold pressor model.
- NKTR-181 produces a dose-dependent and time-delayed maximal central response, with E_{max} for miosis occurring at 4-6 hours, consistent with its reduced rate and extent of uptake into the CNS.
- The T_{max} of NKTR-181 is 2-4 hours and the compound exhibits an average half-life of approximately 12 hours following oral administration, which could support once or twice daily dosing.
- NKTR-181 demonstrates predictable dose-linear pharmacokinetics over a 50-fold range of single oral doses in healthy subjects.

Background

- The abuse properties of opioid drugs are believed to relate to their rapid entry into the CNS (1, 2). NKTR-181 is a novel mu-opioid analgesic molecule engineered using Nektar's small molecule polymer conjugate technology.
- NKTR-181 exhibited a reduced rate and extent of CNS uptake in brain uptake model in rats compared to commonly used opioids (3, 4).
- NKTR-181 has maximal analgesic activity in preclinical models comparable to that of oxycodone and morphine, but demonstrates significantly lower abuse liability (4).
- The properties of NKTR-181 are inherent in the molecular structure of NKTR-181 and no formulation technique has been incorporated into the molecule in preclinical or clinical studies.

Objectives

The objective of this Phase 1 clinical study was to determine the safety, tolerability, pharmacokinetic profile, and opioid pharmacodynamics of single oral ascending doses of NKTR-181 in healthy human subjects.

Methods

- A total of 105 subjects were evaluated in this Phase 1 study.
- Seven dose cohorts were evaluated with 15 subjects in each dose cohort (10, 20, 40, 80, 160, 320, and 500 mg). Subjects in each dose cohort received single doses of NKTR-181 solution given orally (n=12) or placebo (n=3) following an overnight fast. Dose escalation occurred only in the absence of dose-limiting adverse effects.
- Pharmacokinetics and pharmacodynamics was measured through serial blood samples and observations of cold pressor test (CPT) response, as well as pupillometry to determine opioid-induced miosis of drug effect and exposure over time.

References

1: Pharmacol Biochem Behav. 2007; 86:45-54. 2: J Pharmacol Exp Ther. 2001; 299 :1056-65. 3: Poster # HHH13, Society for Neuroscience 40th Annual Meeting: Neuroscience 2010, November 2010; 4: Anesthesiology 2010, A509 (Abstract #A509, American Society of Anesthesiologists (ASA) Annual Meeting: Anesthesiology 2010, 16-20 October 2010, San Diego, CA)

Results

NKTR-181 Shows Dose-linearity, with C_{max} at 2-4 hrs. Post-dose

Figure 1: Mean (±SEM) Plasma NKTR-181 Concentration-Time Profiles in Healthy Subjects (n=12 per dose level)

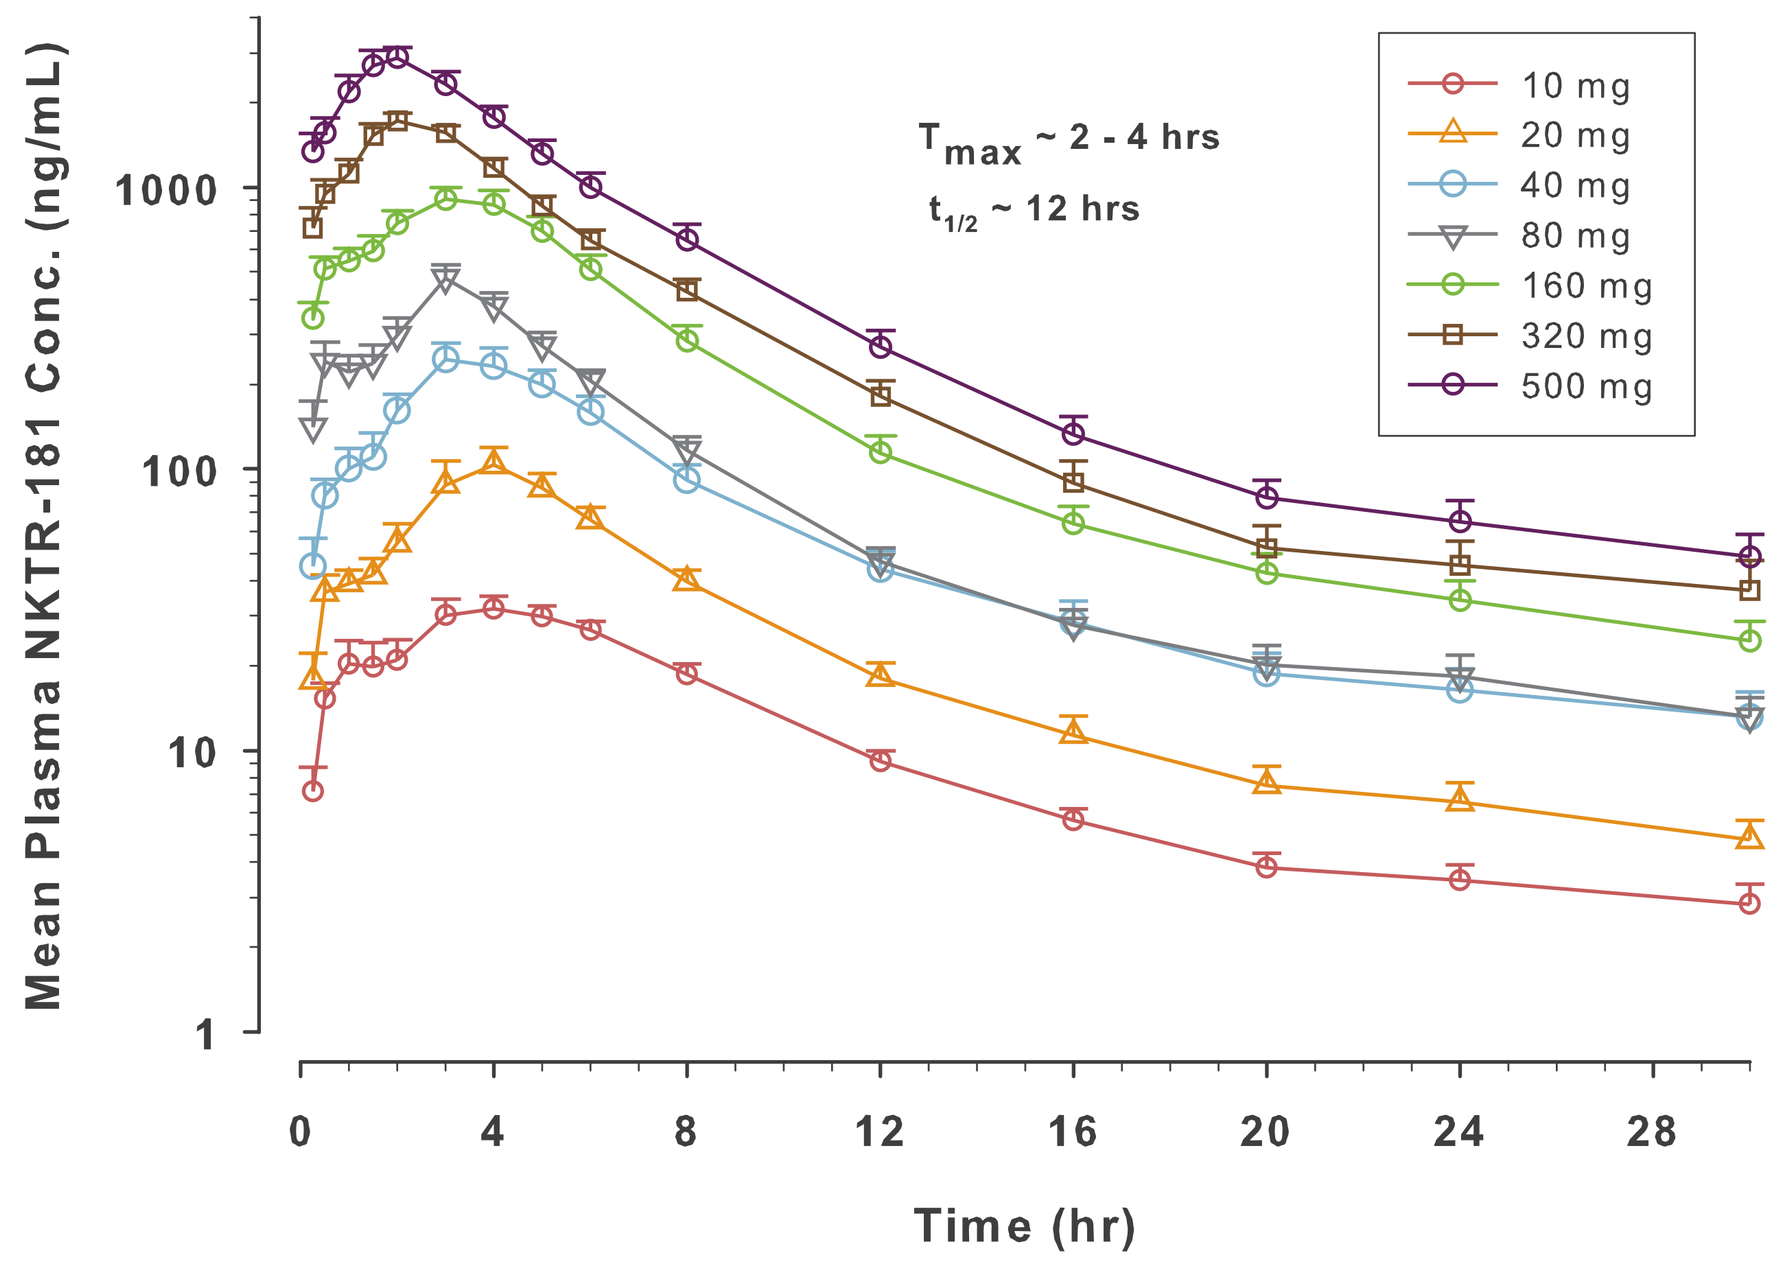
Mean Plasma NKTR-181 Conc. (ng/mL)
Time (hr)
T max ~ 2 - 4 hrs
t1/2 ~ 12 hrs
10 mg
20 mg
40 mg
80 mg
160 mg
320 mg
500 mg

Pharmacokinetics
NKTR-181 C_{max} values occurred ~2 to 4 hours postdose and C^{max} and $AUC_{0-\infty}$ values increased in proportion to dose over the studied range of 10 to 500 mg (Figure 1, Table 1), thus demonstrating dose-linear pharmacokinetics. Plasma half-life of was independent of dose and averaged approximately 12 hours.

Extent and Duration of Miosis Increases with Dose, with Maximum Miosis at 4-6 hrs.

Figure 2: Median (± 95% CI) Pupil Diameter vs. Time by NKTR-181 Dose Level

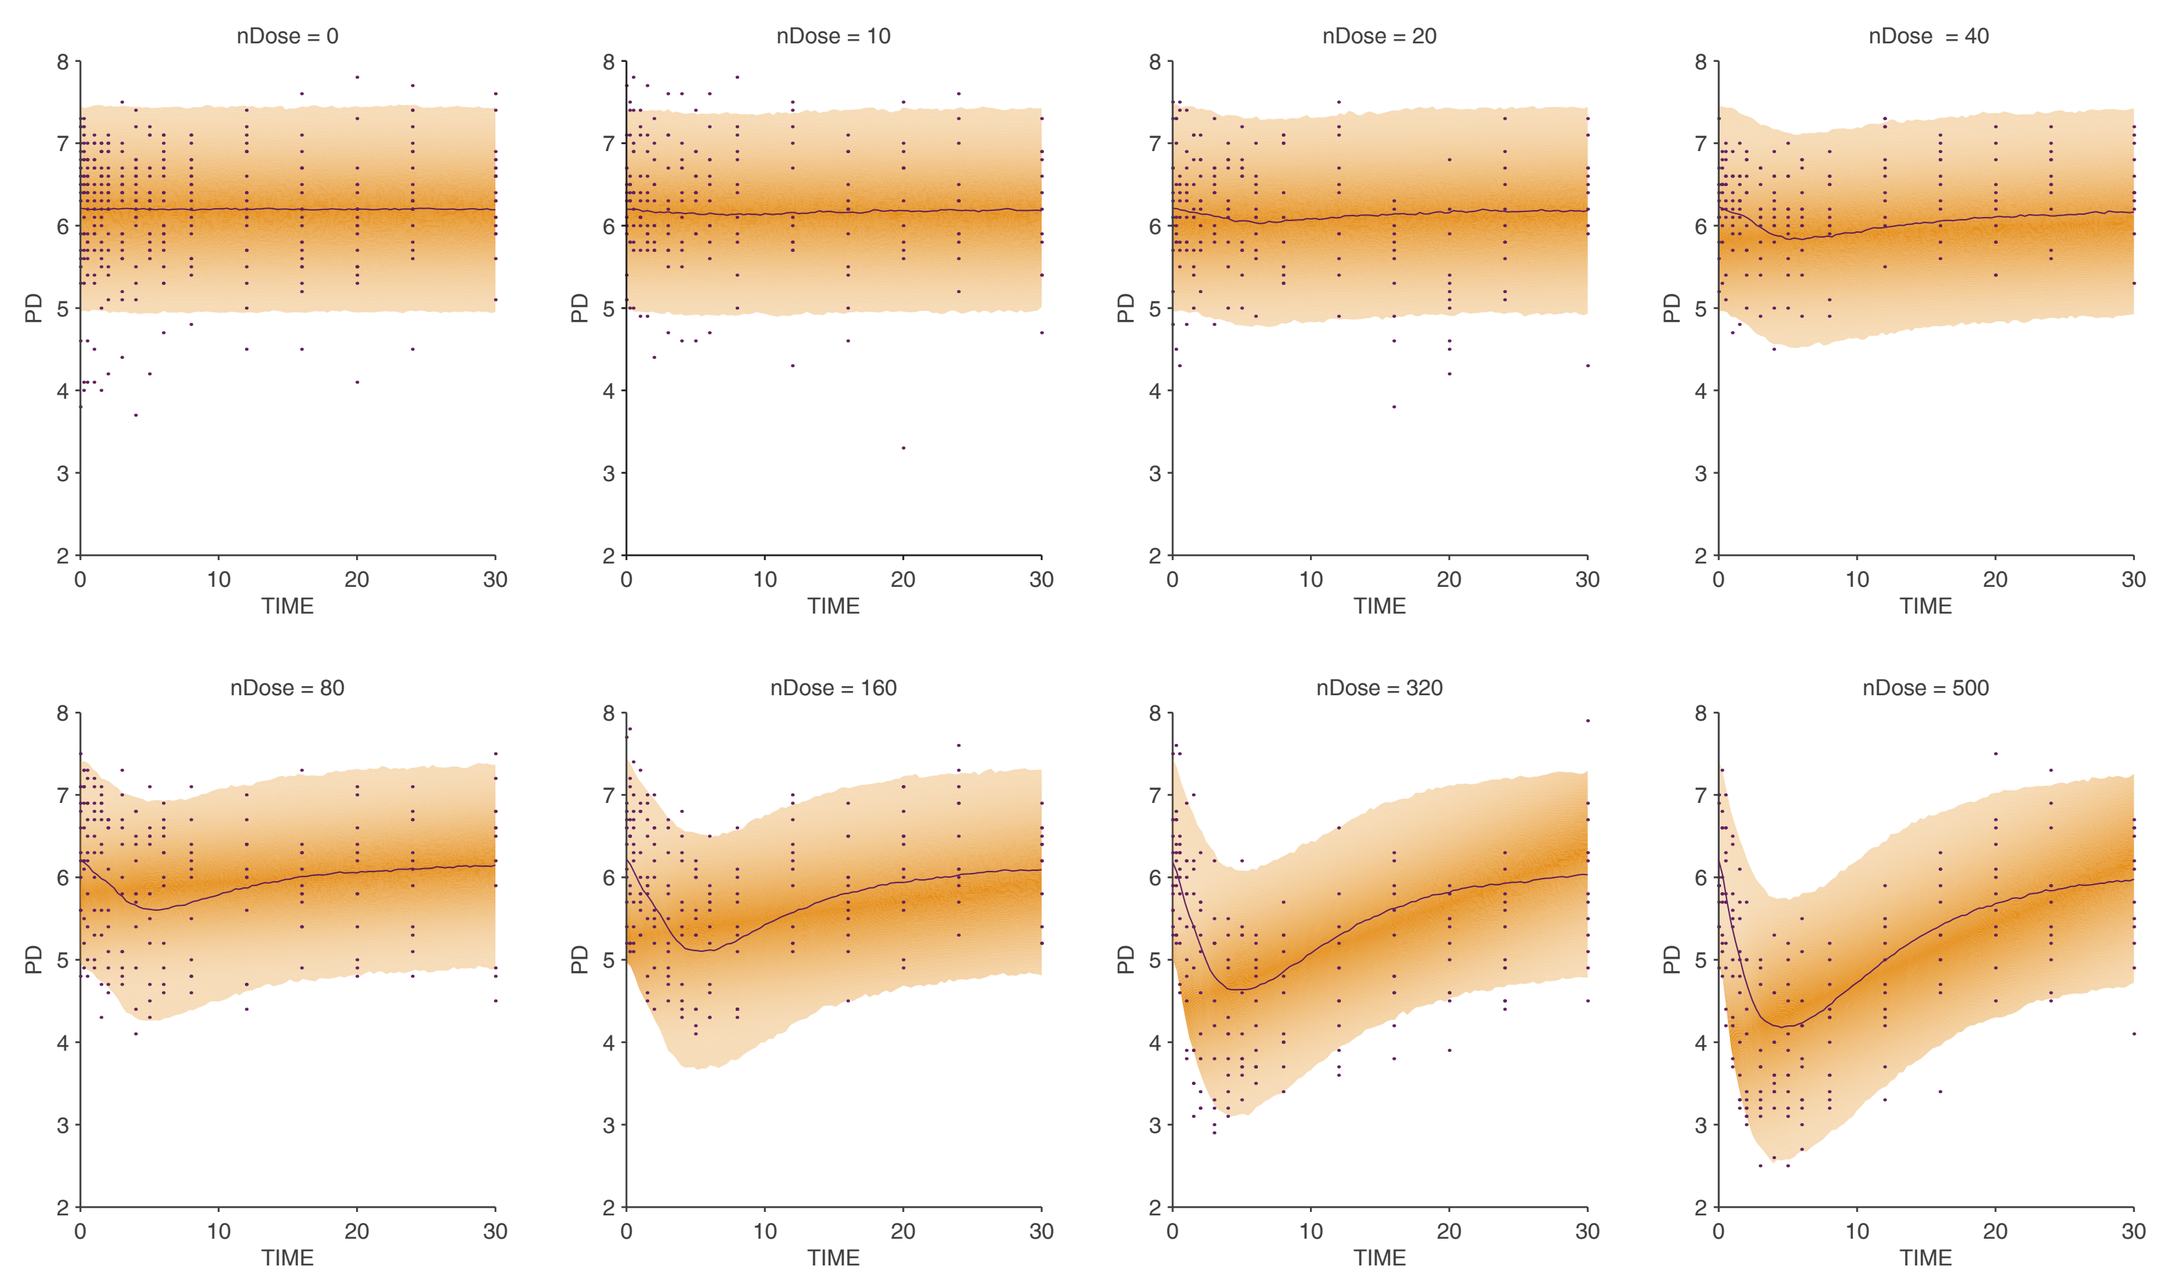
nDose = 0
nDose = 10
nDose = 20
nDose = 40
nDose = 80
nDose = 160
nDose = 320
nDose = 500
PD
TIME

Pharmacodynamics
The extent and duration of opioid effect, based on the time course of miosis, increased with administered dose, with the time of maximum miosis between 4 and 6 hours, and duration up to approximately 16 hours (Figure 2).

Reduction in Pupil Diameter is Proportional to Dose

Figure 3: Effect of NKTR-181 on Maximal Change of Pupil Diameter

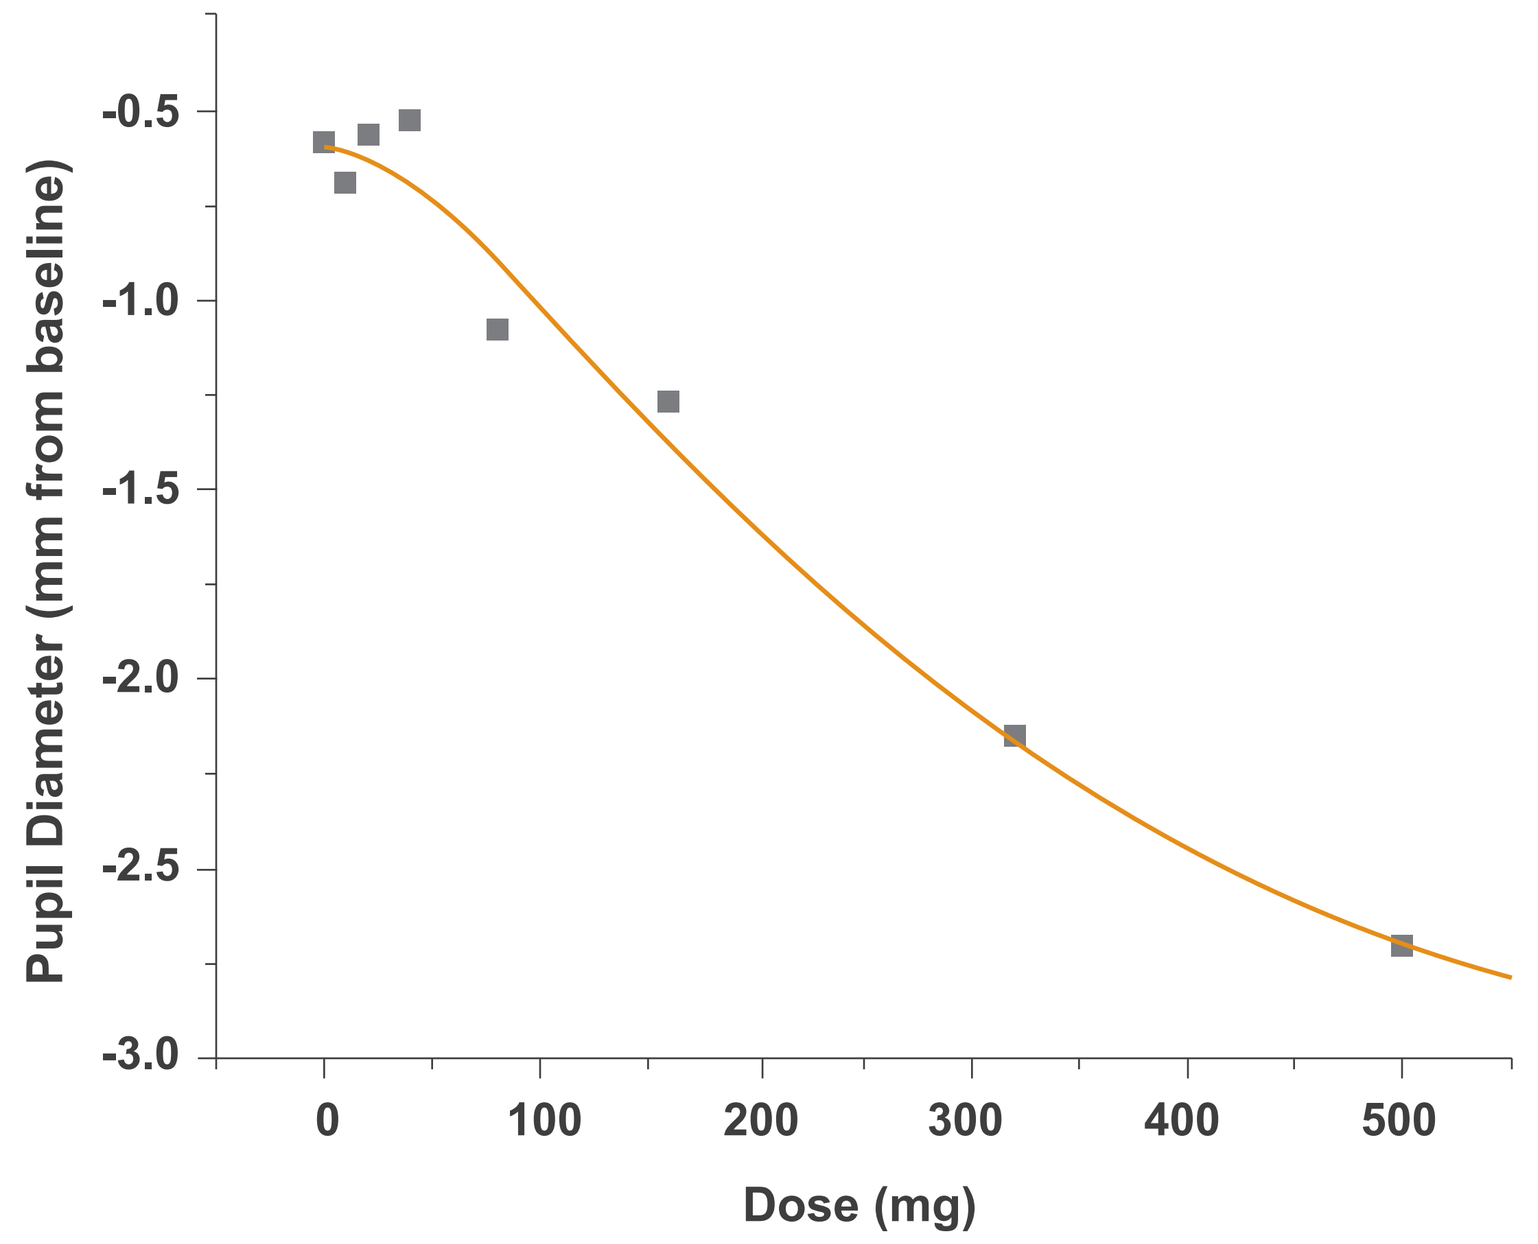
Pupil Diameter (mm from baseline)
Dose (mg)

Pharmacodynamics
The maximum reduction in pupil diameter decreased in a dose-dependent manner over the dose range of 80 to 500 mg (Figure 3).

Miosis Lags Appearance in Plasma, Consistent with Reduced Rate of CNS Uptake

Figure 4: Median Plasma NKTR-181 Concentration and Pupil Diameter (inverted scale) vs. Time

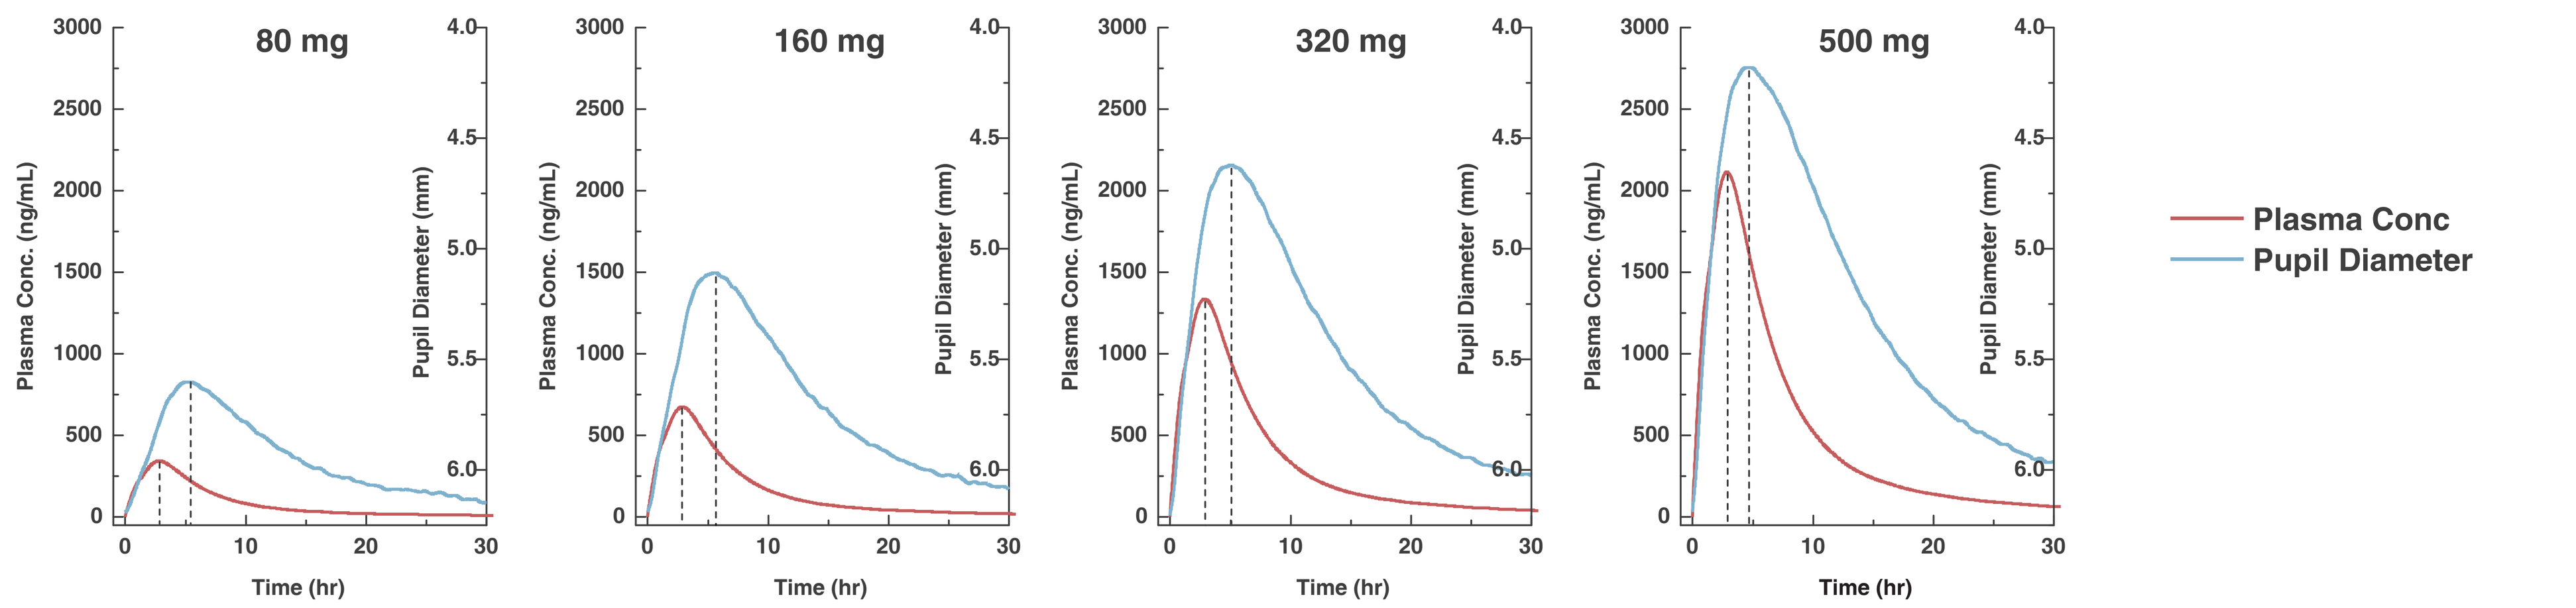
80 mg
160 mg
320 mg
500 mg
Plasma Conc. (ng/mL)
Pupil Diameter (mm)
Time (hr)
Plasma Conc
Pupil Diameter

Although NKTR-181 was rapidly absorbed and detected in plasma within 15 minutes (Figure 1), the time course of miosis lagged the time course of NKTR-181 in plasma, consistent with the reduced rate of NKTR-181 CNS uptake observed in preclinical studies. Figure 4 illustrates the controlled onset and sustained duration of miosis relative to the plasma NKTR-181 concentration-time profile for the doses resulting in maximal miotic effect.

Pain Tolerance Increases with Dose

Figure 5: Plasma concentration and CPT (mean latency to hand removal) vs. time profiles

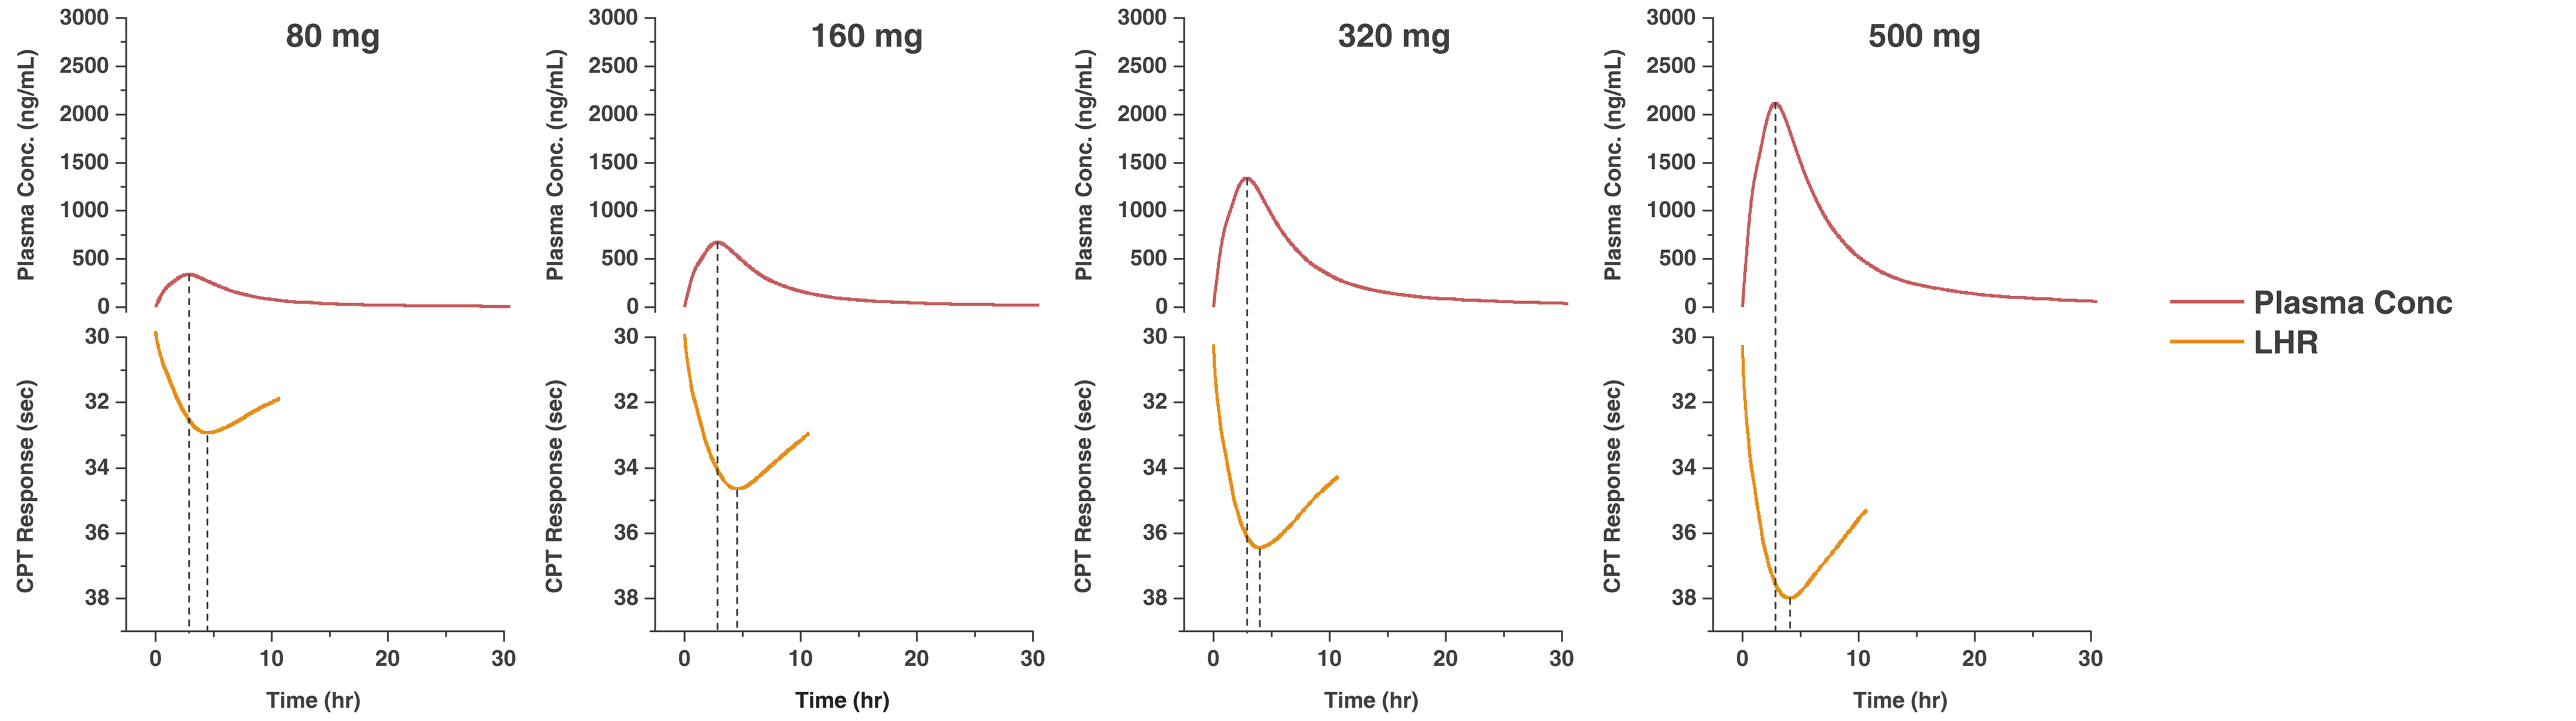
80 mg
160 mg
320 mg
500 mg
Plasma Conc. (ng/mL)
CPT Response (sec)
Time (hr)
Plasma Conc
LHR

The extent and duration of analgesic effect based on CPT parameters of Time to Pain Perception and Pain Tolerance also increased with administered dose. Figure 5 shows plasma NKTR-181 concentration and CPT response vs. time profiles for representative individuals who received single 80, 160, 320, or 500 mg doses of NKTR-181.

Safety and Tolerability

NKTR-181 was well-tolerated across a 50-fold range of doses. There were very few reports of AEs until the highest dose level tested. Adverse events (AEs) were transient and most were mild. Mild non-dose limiting AEs at the highest dose tested included AEs characteristic of an active opioid agonist, such as mild dizziness and nausea.

Presented at the American Academy of Pain Management 22nd Annual Meeting; September 20–23, 2011; Las Vegas, NV.