Population Pharmacokinetics of NKTR-102, a Topoisomerase I Inhibitor-Polymer Conjugate in Patients With Advanced Solid Tumors

Michael A. Eldon and Ute Hoch, Nektar Therapeutics, San Francisco, CA

Abstract

Background: NKTR-102 demonstrates significant anti-tumor activity with a tolerable toxicity profile in pts with platinum-resistant/refractory ovarian or metastatic breast cancers. NKTR-102 is a novel topoisomerase I inhibitor that uses Nektar's proprietary polymer conjugation to improve the PK of its active metabolite SN38 resulting in significantly reduced Cmax and sustained exposure throughout the dosing interval. **Methods**: NKTR-102 was administered as a 90-min IV inf. to 94 pts: 76 pts at 58-245 mg/m² wx3q4w, q14d or q21d, and 18 pts at 100 or 125 mg/m² q21d receiving concurrent cetuximab. Serial plasma samples were collected for the duration of treatment and assayed by LC-MS/MS for NKTR-102, irinotecan, SN38, SN38-Glucuronide (SN38-G), and APC. **Results**: NKTR-102 and its metabolites followed biphasic disposition kinetics with first-order appearance rates consistent with their metabolic progression (Tmax 1.5-24 hr) and first-order terminal $t_{1/2}$ between 21-61 days. PK of NKTR-102 and all metabolites were independent of dose and schedule. BSA was a significant covariate for CL. SN38 CL was lower (30%) in females and lower (30%) in pts who were homozygous for UGT1A1*28, but these reductions were not considered clinically important. Patient age, hepatic or renal functions were not correlated with SN38 CL. Coadministration of cetuximab had no effects on NKTR-102 PK and vice versa. PK in pts with ovarian and breast cancer were similar to those with non-ovarian or -breast tumors. The RP2D (145 mg/m²) results in approx. the same plasma SN38 AUC as 350 mg/m² of irinotecan, but exposure is continuous rather than intermittent and Cmax is 10-fold less. **Conclusions**: NKTR-102 results in sustained systemic exposure to SN38 between infrequent doses while preventing exposure to excessively high concentrations; these enhancements appear responsible for improved efficacy and tolerability profiles. The PK of NKTR-102 and metabolites are predictable and do not require complex dosing adjustments and support ongoing Phase 2 clinical development as single-agent in metastatic breast, platinum-resistant ovarian, and metastatic colorectal cancers.

Background

- NKTR-102 uses Nektar's proprietary polymer conjugation to improve the pharmacokinetics of irinotecan and its active metabolite SN38.
- NKTR-102 has superior activity compared to irinotecan in a wide range of mouse models of human xenograft tumors.
- NKTR-102 demonstrated high anti-tumor activity in a range of tumors in Phase 1 (11% confirmed PRs; ENA 2008, abs 595).
- NKTR-102 showed a 22% confirmed response rate per RECIST in heavily pre-treated women with platinum resistant/refractory ovarian cancer (ASCO 2010, abs 5013), and a 29% confirmed response rate in 2nd/3rd line patients with advanced breast cancer (SABCS 2010, abs S6-6)

Irinotecan is extensively metabolized to various metabolites. It is cleaved enzymatically to form SN38 which is conjugated to an inactive glucuronide, SN38-G. Other inactive irinotecan metabolites identified are the major plasma metabolite APC (7-ethyl-10-[4-N-(5-aminopentanoic acid)-1-piperidino]-carbonyloxycamptothecin), and NPC (7-ethyl-10-[4-amino-1-piperidino]-carbonyloxycamptothecin). In NKTR-102, irinotecan is conjugated to a multi-arm PEG and is cleaved in vivo.

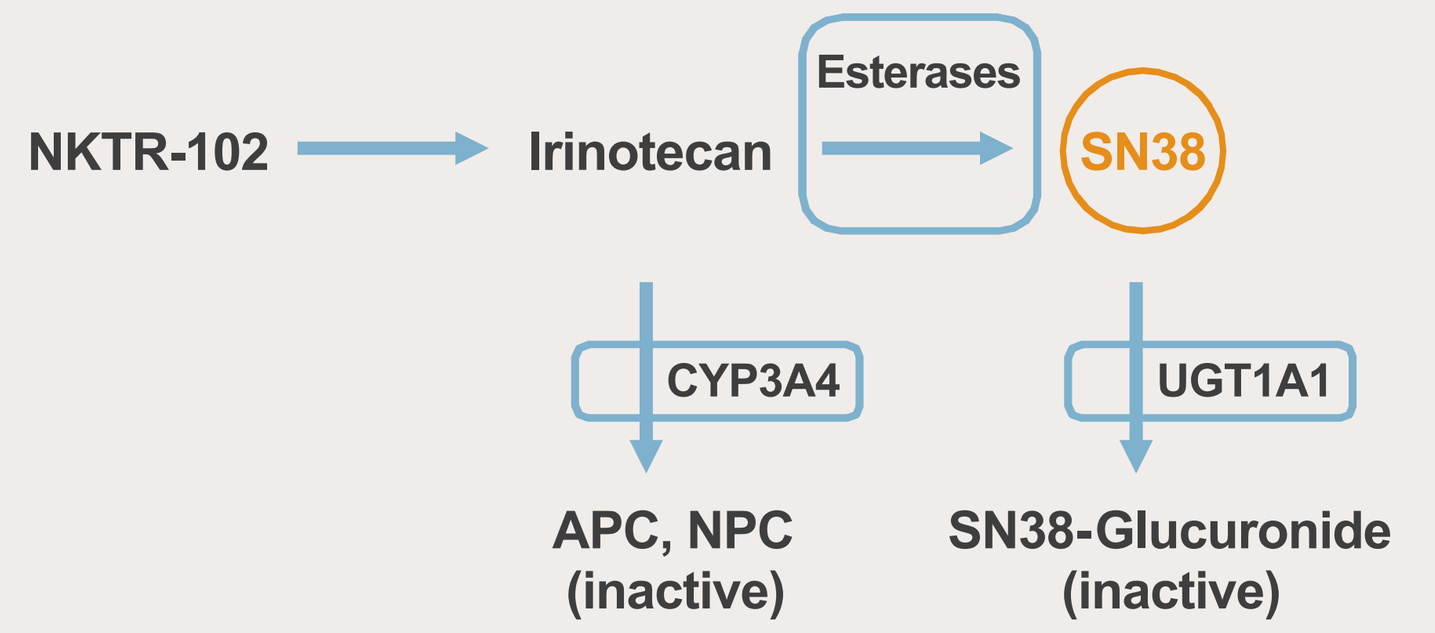
NKTR-102
Irinotecan
Esterases
SN38
CYP3A4
UGT1A1
APC, NPC (inactive)
SN38-Glucuronide (inactive)

Methods

NKTR-102 Pharmacokinetic Analysis: In Phase 1 Study 06-IN-IR001, NKTR-102 was administered as a 90-min IV infusion. Thirty-two patients received 58-230 mg/m² doses wx3q4wk, 19 pts received 145-230 mg/m² q14d, and 25 pts received 145-245 mg/m² doses q21d. Plasma samples were obtained throughout the treatment duration: rich PK sampling was performed during the first cycle and cycle 3 for the q14d and q21d treatment schedules; predose samples were obtained before each subsequent cycle. PK samples were analyzed using validated methods with LLOQ of 2000 ng/mL for NKTR-102, 1 ng/mL for irinotecan, SN38-G, and APC, and 0.2 ng/mL for SN38. Pharmacokinetics of all analytes were estimated using nonlinear mixed-effects modeling. Individual patient concentration-time profiles for all analytes were well represented by a 2-compartment pharmacokinetic model with either zero-order input for NKTR-102 or first-order input for all metabolites, and first-order output for all analytes. Data for each analyte were fitted with the appropriate PK model using Monolix 3.1 (INRIA Saclay, Orsay, France).

Covariate Analysis: The effect of patient demographics, renal or hepatic impairment as estimated from clinical laboratory results and UGT1A1 homozygosity on drug and metabolite CL or CL/F, V or V/F, and metabolite formation rate (K_{form}) were investigated using nonlinear mixed-effects modeling. Plasma concentration-time data for NKTR-102, irinotecan, SN38, SN38-G, or APC from 94 patients (76 receiving single agent NKTR-102; 18 receiving concurrent cetuximab; data were pooled because no drug-drug interaction was observed in the presence of cetuximab) with various solid tumors were combined for analysis. Individual patient concentration-time profiles for all analytes were well represented by a 2-compartment pharmacokinetic model with either zero-order input for NKTR-102 or first-order input for all metabolites, and first-order output for all analytes. Data for each analyte were fitted with the appropriate PK model and selected clinical covariates using Monolix 3.1. Inclusion of a covariate in the final model for each analyte was based on application of Wald's approximation of the likelihood ratio test as described by Kowalski and Hutmacher (Efficient screening of covariates in population models using Wald's approximation to the likelihood ratio test. J Pharmacokinet Pharmacodyn 2001 Jun;28(3):253-75).

NKTR-102 Pharmacokinetic Analysis

Observed and Model-Predicted Concentration-Time Profiles for Representative Patients at the Recommended Phase 2 Doses

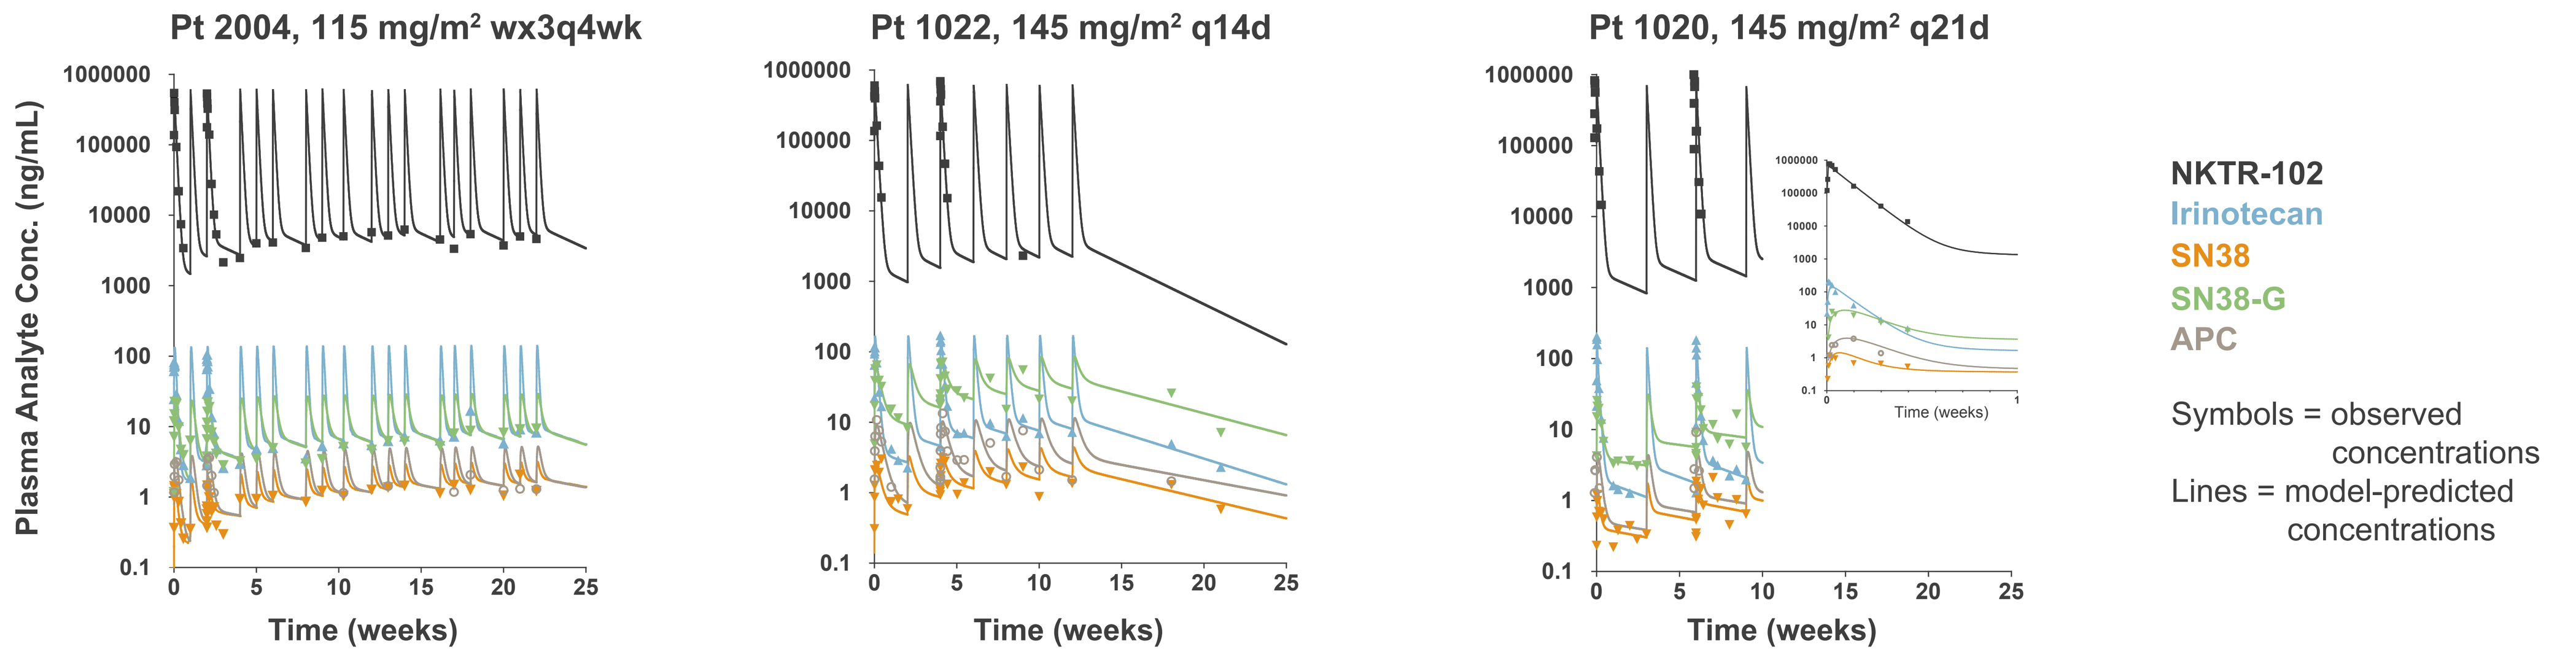
Pt 2004, 115 mg/m² wx3q4wk
Pt 1022, 145 mg/m² q14d
Pt 1020, 145 mg/m² q21d
Plasma Analyte Conc. (ng/mL)
Time (weeks)
NKTR-102
Irinotecan
SN38
SN38-G
APC
Symbols = observed concentrations
Lines = model-predicted concentrations

PK Parameters

Mean (%CV) plasma pharmacokinetic parameters for NKTR-102, irinotecan, SN38, SN38-G, and APC after a 90-min intravenous infusion of the first NKTR-102 dose between 58-245 mg/m²

Analyte	n	Tmax (hr)	Cmax (ng/mL)	AUC (ng*hr/mL)	$T_{1/2}$ (days)
NKTR-102	76	1.8 (31)	360000-1300000	5600000-29000000	21 (20)
Irinotecan	67*	4 (91)	50-600	3000-12000	27 (13)
SN38	76	13 (170)	2-6	400-2500	50 (28)
SN38-G	76	16 (69)	30-100	6500-67000	61 (37)
APC	76	20 (41)	4-30	800-3000	53 (8)

*Data from 9 pts were missing due to incorrect sample processing.

- Cmax of NKTR-102 observed shortly after the end of the 90-min infusion
- Cmax decreased in the order of NKTR-102>irinotecan≥SN38-G>APC>SN38
- Tmax for NKTR-102 metabolites increased, consistent with the proposed metabolic scheme
- NKTR-102 and metabolite PK were independent of dose and treatment schedule, and Cmax and AUC increased linearly with dose
- Interpatient variability in SN38 exposure after NKTR-102 was similar to that reported for SN38 after irinotecan dministration

Comparison of Irinotecan and SN38 Concentration-Time Profiles after NKTR-102 and Irinotecan Administration

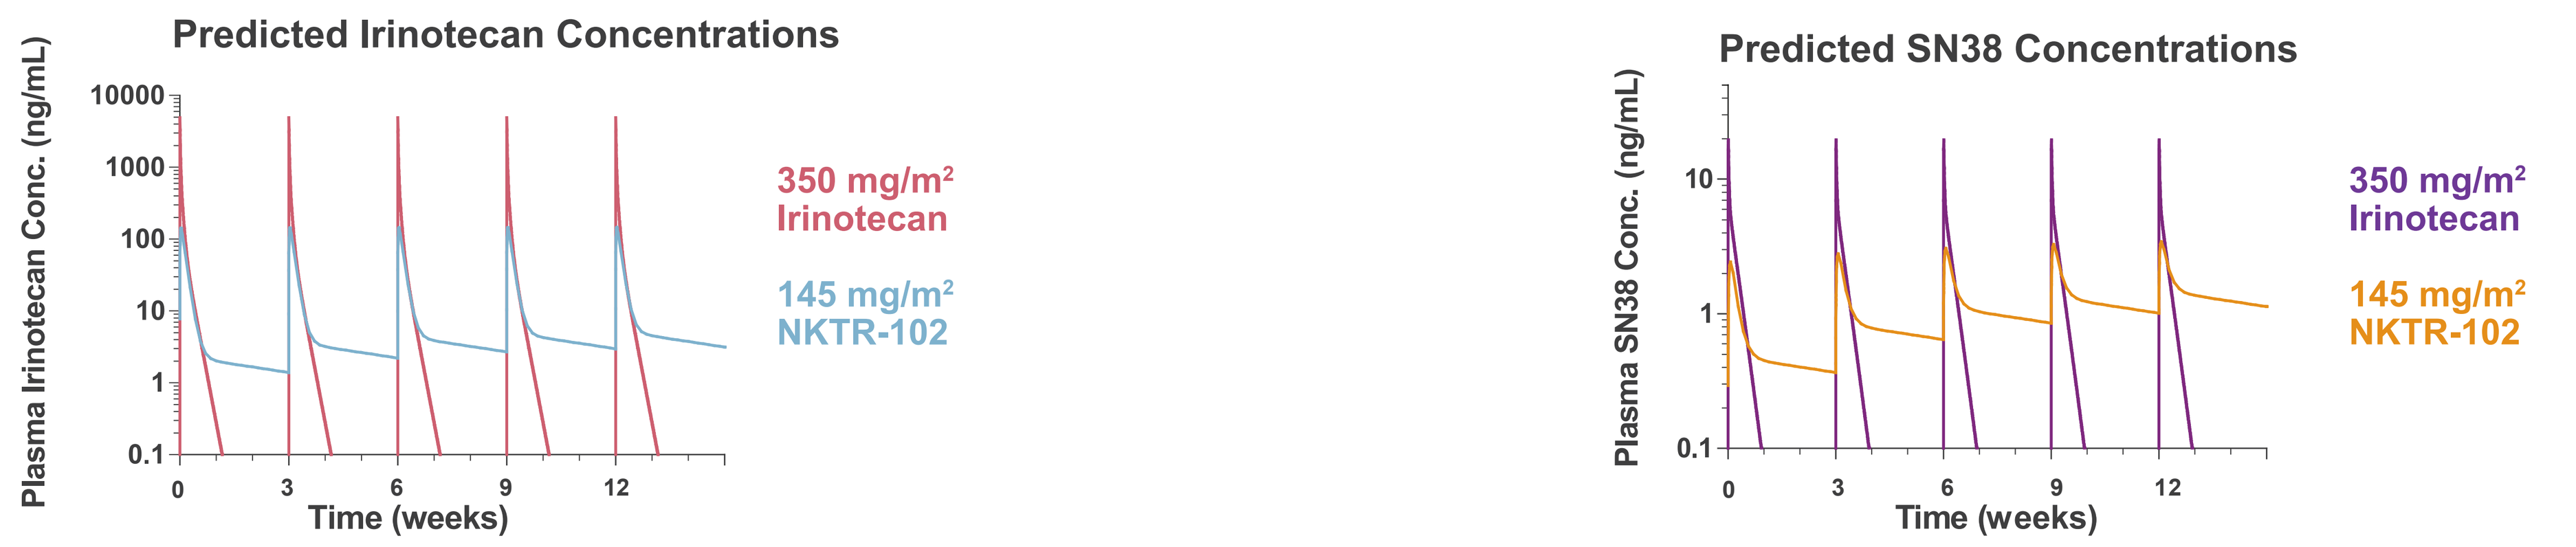
Predicted Irinotecan Concentrations
Plasma Irinotecan Conc. (ng/mL)
Time (weeks)
350 mg/m² Irinotecan
145 mg/m² NKTR-102
Predicted SN38 Concentrations
Plasma SN38 Conc. (ng/mL)
Time (weeks)
350 mg/m² Irinotecan
145 mg/m² NKTR-102

Irinotecan and SN38 concentrations after irinotecan administration are based on Xie et al. 2002, JCO 20:3293. Simulations of irinotecan and SN38 after NKTR-102 administration are based on the 06-IN-IR001 derived population parameters.

- Single agent approved dose of irinotecan is 350 mg/m² q21d
- Compared to irinotecan administration, irinotecan Cmax after administration of 145 mg/m² NKTR-102 is ~35-fold lower compared to administration of 350 mg/m² irinotecan
- Reduced Cmax is likely associated with lack of cholinergic reactions after administration of NKTR-102
- Compared to irinotecan administration, SN38 concentrations are sustained between dosing occasions after NKTR-102 administration: plasma SN38 remains >0.4 ng/mL after the first dose and > 1 ng/mL after ≥4 doses
- In contrast, plasma SN38 concentrations fall below 0.1 ng/mL 7 days post irinotecan dose, resulting in a drug holiday for 70% of the dosing cycle
- A single145 mg/m² dose of NKTR-102 results in approximately the same plasma exposure to SN38 as the 350 mg/m² dose of irinotecan, but exposure is continuous rather than intermittent and maximal concentrations are ~5- to 10-fold less

PK in Patients With Ovarian and Breast Cancers (Phase 3 planning underway for these tumor types)

Six of 94 patients had ovarian as the primary tumor. Patients with ovarian cancer tended to be smaller (mean BSA 1.72 vs 1.91 m²), older (mean age 65 vs 58 yr) and have more renal impairment (mean eGFR 53 vs 79 mL/min) than those with other cancers.

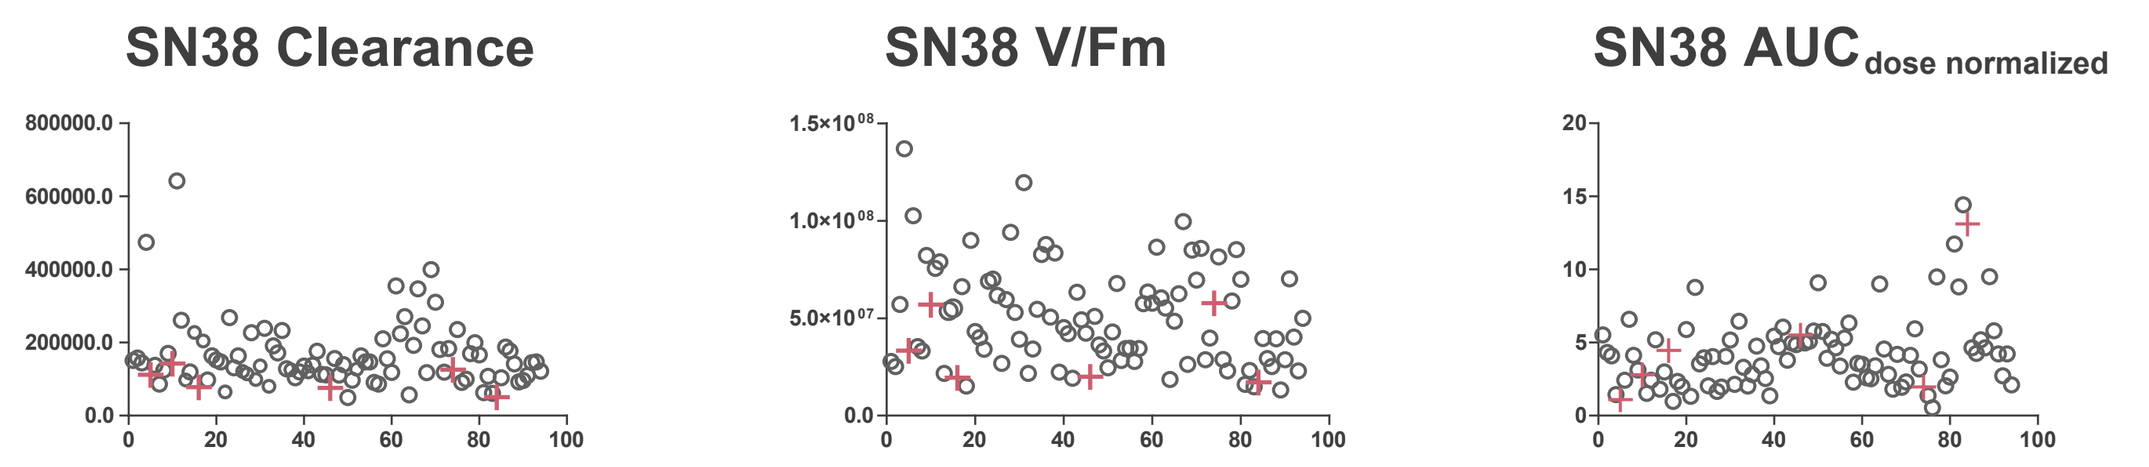
SN38 Clearance
SN38 V/Fm
SN38 AUC dose normalized

- PK in patients with ovarian cancer is similar to those with all other cancers

Five of 94 patients had breast as the primary tumor. Patients with breast cancer had similar characteristics to those with other cancers.

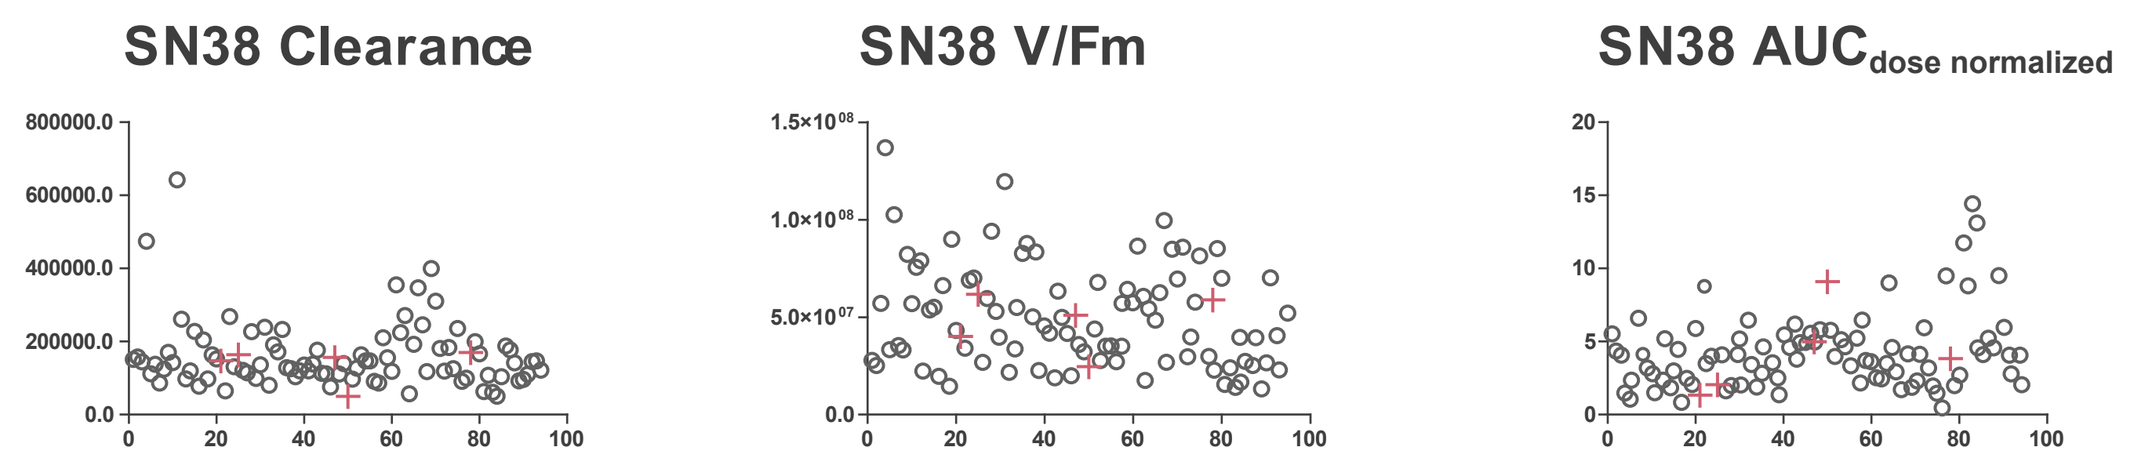
SN38 Clearance
SN38 V/Fm
SN38 AUC dose normalized

- PK in patients with breast cancer is similar to those with all other cancers

Covariate Analysis

Listing of Potential and Statistically Significant Covariates

	Demographics		Renal Impairment	Hepatic Impairment	Reduced UGT1A1 Activity
	Age 25-81 years	Gender 45 female, 49 male	eGFR# 30-134 mL/min	Bilirubin 0.2-1.6 mg/dL	UGT1A1*28 11 of 94 pts
NKTR-102*	CL, V1	CL, V1	CL, V1	CL, V1	not investigated
Irinotecan	CL, V/Fm	CL, V/Fm	CL, V/Fm	CL, V/Fm	not investigated
SN38	K_{form}, CL, V/Fm	K_{form}, CL, V/Fm	CL, V/Fm	CL, V/Fm	CL, V/Fm
SN38-G	K_{form}, CL, V/Fm	K_{form}, CL, V/Fm	CL, V/Fm	CL, V/Fm	K_{form}, CL, V/Fm
APC	K_{form}, CL, V/Fm	K_{form}, CL, V/Fm	CL, V/Fm	ka, CL, V/Fm	not investigated

#Estimated glomerular filtration rate based on corrected chronic kidney disease epidemiological models
*Black color indicates covariates investigated, red color indicates the covariates found statistically significant

Renal Impairment and NKTR-102 PK

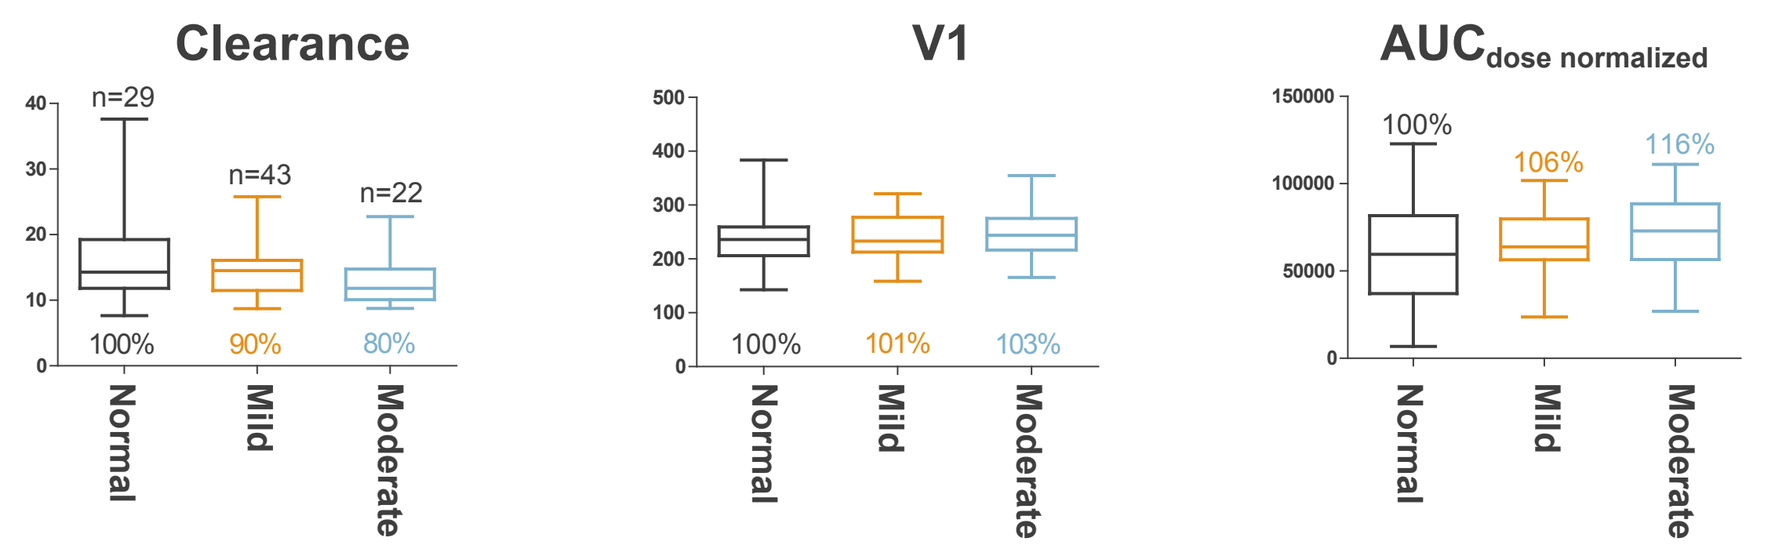
Clearance
V1
AUC dose normalized
Normal
Mild
Moderate

- Only NKTR-102 was observed to decrease with decreasing renal function
- Renal excretion of NKTR-102 constitutes only a minor pathway; predominant metabolic pathway is dependent on cleavage to irinotecan
- Due to the small changes in clearance, not expected that patients with moderate renal impairment (eGFR ≥30 mL/min) will require reduced NKTR-102 starting doses

Age and SN38-G PK or APC PK

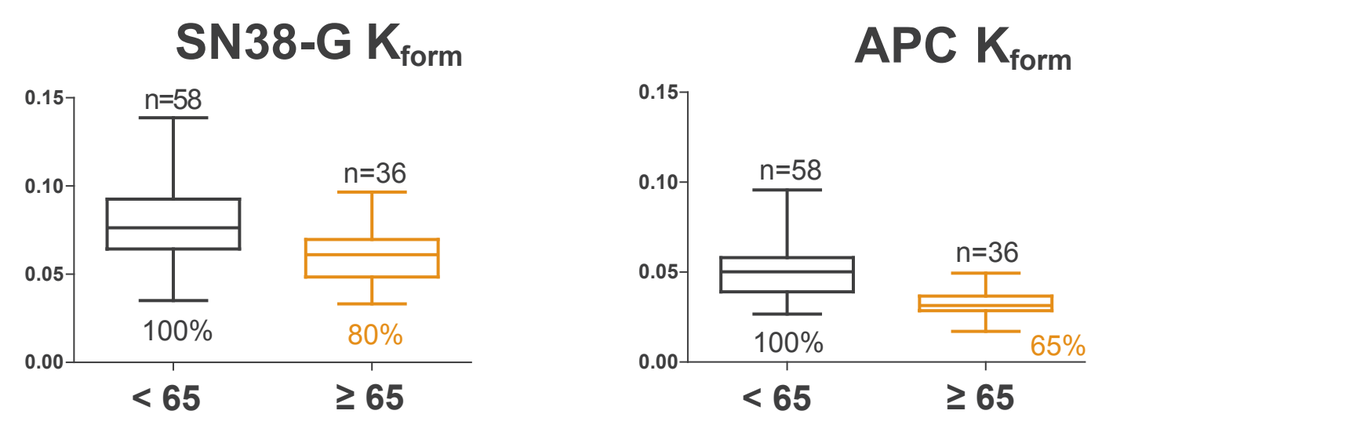
SN38-G Kform
APC Kform
< 65
≥ 65

- K_{form}, CL, and V/Fm for SN38-G and for APC all appear to decline with age, but only K_{form} was statistically significant
- Clinical significance of these trends, if any, is unknown

Age and Irinotecan PK

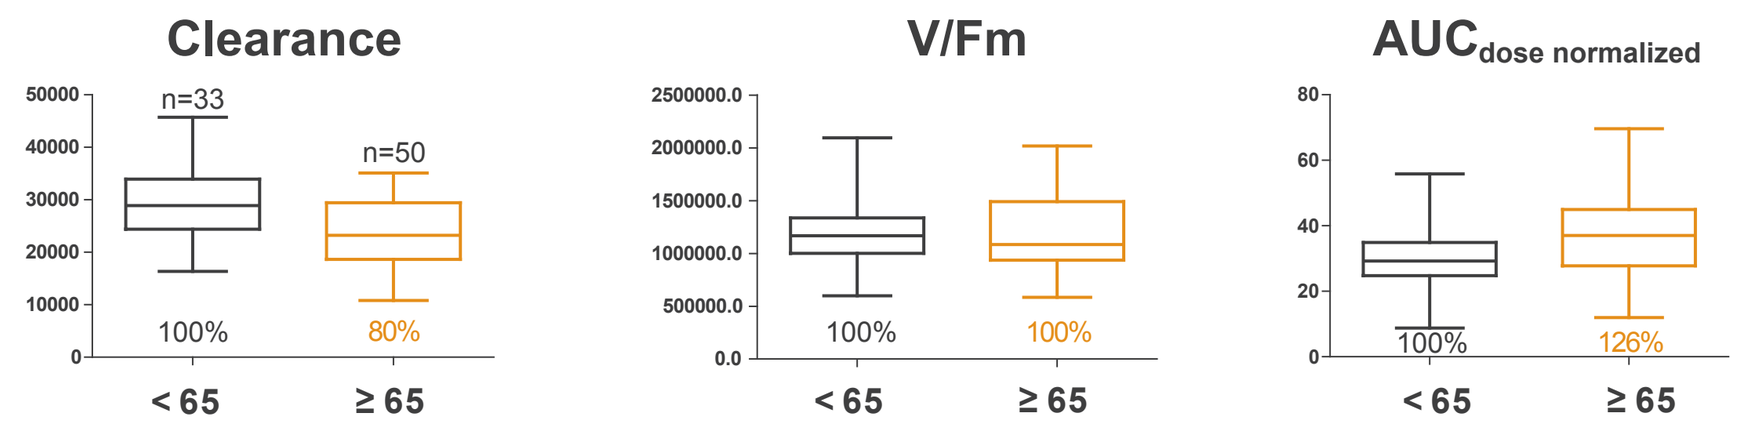
Clearance
V/Fm
AUC dose normalized
< 65
≥ 65

- Small but statistically significant reduction observed in irinotecan clearance between the age groups (ages ranged from 25-81 years)
- Small decline is not expected to be clinically important in the selection of NKTR-102 starting dose

UGT1A1 Status and SN38G PK

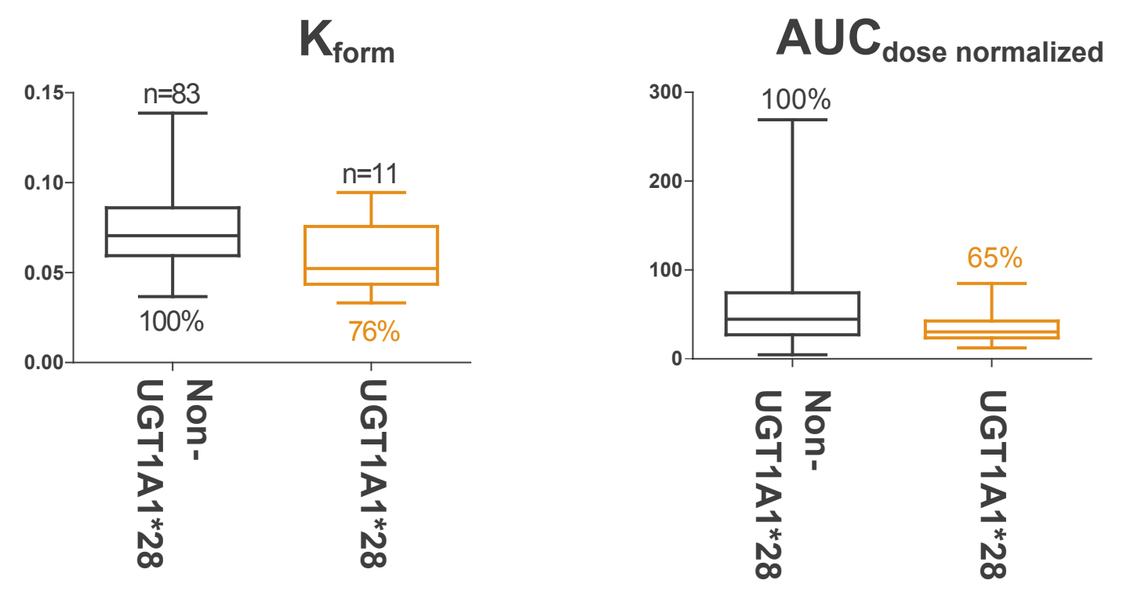
Kform
AUC dose normalized
Non-UGT1A1*28
UGT1A1*28

- Only SN38-G formation rate (K_{form}) found to be significantly different
- Consistent with role of UGT enzymes in SN38 to SN38-G metabolism
- This finding is not expected to be clinically important for the selection of NKTR-102 starting dose

Gender and SN38 PK

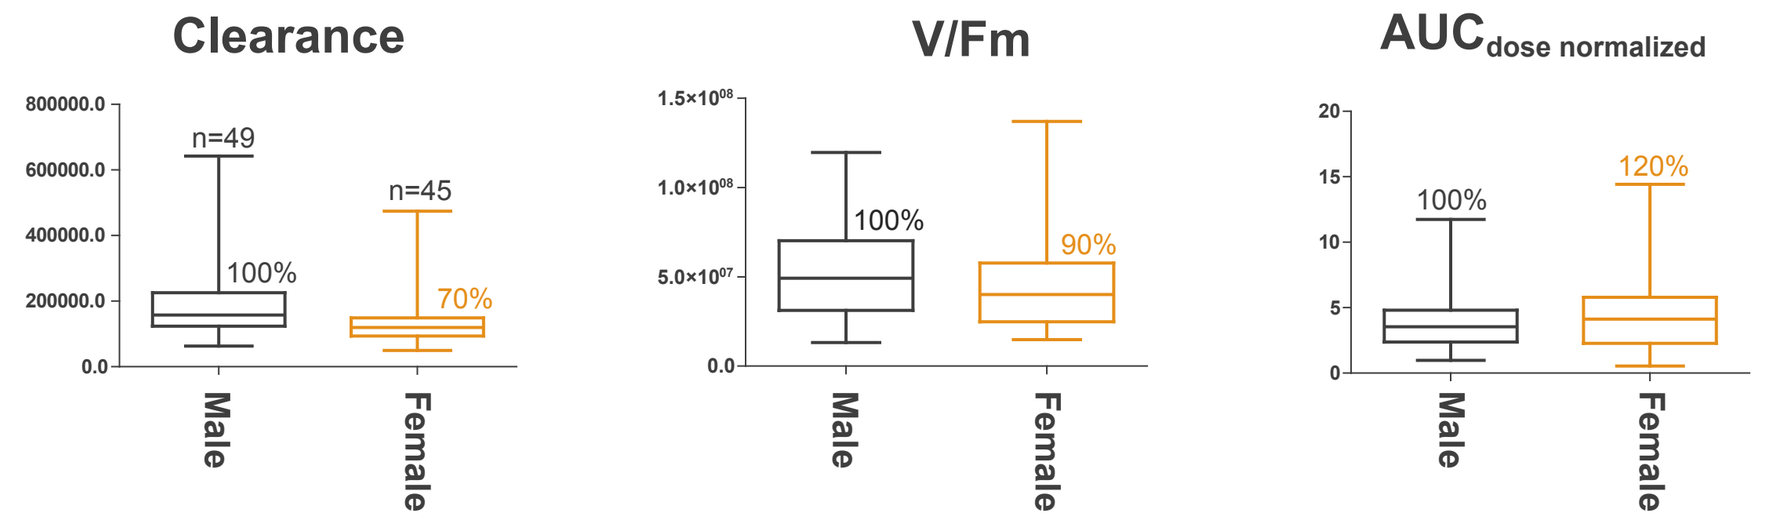
Clearance
V/Fm
AUC dose normalized
Male
Female

- SN38 CL/Fm is ~30% less in females
- No other parameter significantly influenced by patient gender

UGT1A1 Status and SN38 PK

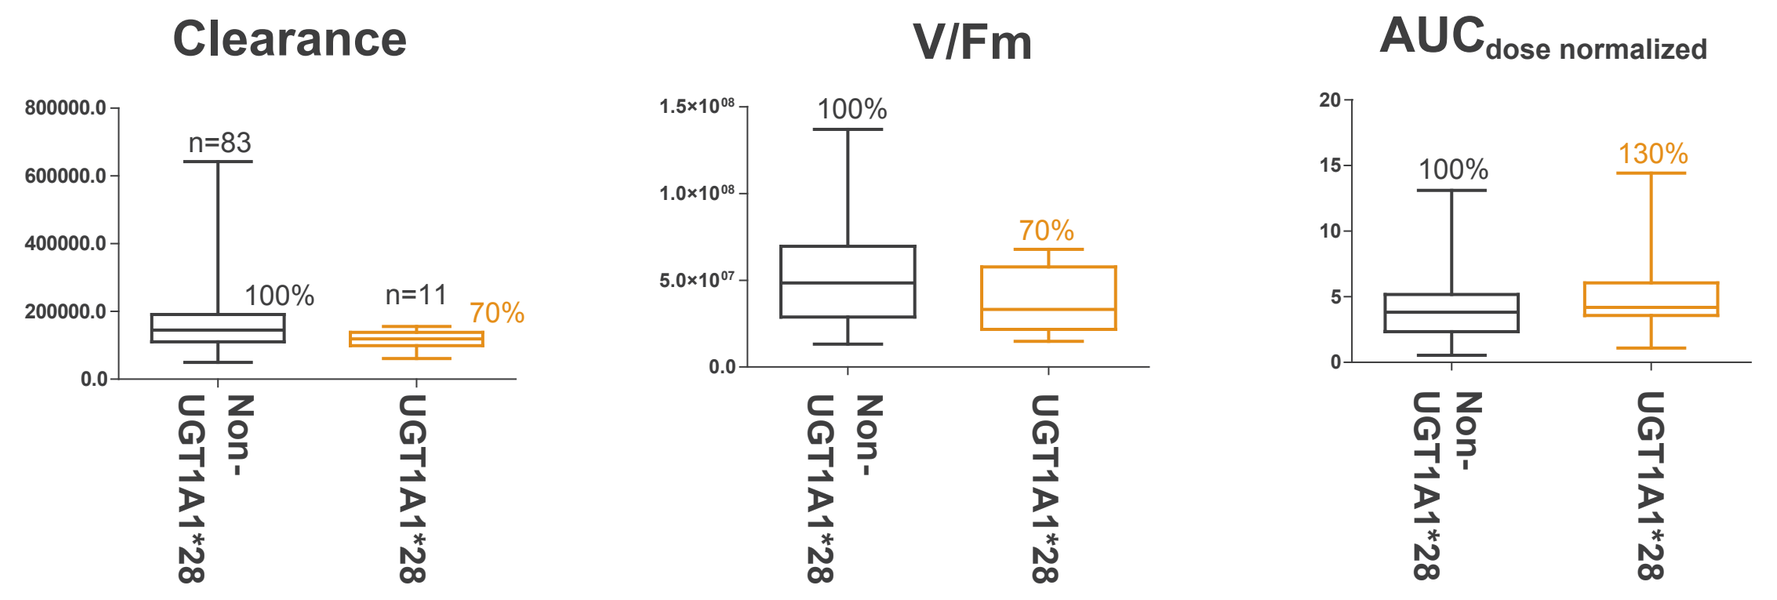
Clearance
V/Fm
AUC dose normalized
Non-UGT1A1*28
UGT1A1*28

- Reductions in clearance and volume in homozygous patients were not significant
- Reduced SN38 Cmax and overall lower SN38 plasma concentrations appear less likely to result in saturation of glucuronidation in homozygous patients
- Reduction in starting dose not expected to be required

Conclusions

- NKTR-102 and metabolites demonstrate dose proportional disposition kinetics with long disposition half-lives
- SN38 concentrations are sustained throughout a 21-day dosing interval; Cmax ~5-10-fold less for NKTR-102 vs. irinotecan
- Interpatient variability in PK parameters is comparable to irinotecan
- No unexpected clinical covariates of drug/metabolite exposure
- NKTR-102 appears to allow for a uniform starting dose across all patients
- Overall, the PK of NKTR-102 and its metabolites are predictable and do not require complex dosing adjustments
- NKTR-102 is currently being developed in ovarian, breast, and colorectal cancers and Phase 3 planning is underway for patients with ovarian and breast cancers

Abstract 2598, Poster 8E ASCO 2011, Developmental Therapeutics – Clinical Pharmacology and Immunotherapy, June 3-7, 2011
To download a copy of this poster, please visit http://www.nektar.com/