REVEAL: A phase 1/2, open-label, multicenter, dose escalation and dose expansion study of NKTR-262 [TLR 7/8 agonist] plus NKTR-214 [CD122-biased agonist] with or without nivolumab (nivo) in patients (pts) with locally advanced or metastatic solid tumor malignancies

NEKTAR®

Adi Diab[1], Mario Marcondes[2], Brian Kotzin[2], Mary Tagliaferri[2], Ute Hoch[2], Jenny Zhang[2], Werner Rubas[2], Saul Kivimäe[2], Jonathan Zalevsky[2], Anthony Paul Conley[1], Erkut Hasan Borazanci[3], Sandra P. D'Angelo[4]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]Nektar Therapeutics, San Francisco, CA; [3]HonorHealth/ TGen, Scottsdale, AZ; [4]Memorial Sloan Kettering Cancer Center & Weill Cornell Medical College, New York, NY

BACKGROUND

- Immune system activation with checkpoint inhibitors has proven to be an effective strategy for inhibiting tumor growth and prolonging survival.[1,2] However, anti-PD-1 therapies, such as nivolumab, depend on pre-existing T-cell infiltration within the tumors for optimal efficacy[3,4]
- Toll-like-receptor (TLR) stimulation can induce differentiation of functional antigen presenting cells in the tumor environment and reduce immune suppression in tumors, facilitating T cell priming[5]
- Stimulation of the interleukin-2 (IL-2) pathway is a potent means of expanding and activating immune cells, specifically lymphocytes including cytotoxic CD8$^+$ T cells[6]
- Abundance and functional quality of tumor infiltrating lymphocytes are positively linked with tumor response and improved survival with checkpoint inhibitors[1-4]
- Combination treatment with a novel intratumoral (IT) TLR7/8 targeting agent NKTR-262 and a systemic CD122-biased agonist NKTR-214 leads to synergistic activation of innate and adaptive anti-tumor immune response (**Figure 1**) resulting in highly efficacious growth inhibition of treated and abscopal lesions in multiple preclinical mouse tumor models

Figure 1: Mechanism of Action of NKTR-262 and NKTR-214 Combination Treatment

- NKTR-262 and NKTR-214 engage non-overlapping immune mechanisms enhancing antigen presentation and anti-tumor T cell response
 - Intratumorally delivered NKTR-262 provides local sustained release of TLR7/8 agonist with minimal systemic exposure[8]
 - Systemic sustained CD122-biased IL-2 pathway activation by NKTR-214 expands and maintains tumor infiltrating CD8$^+$ T cell clones[9-11]
 - NKTR-214 substantially increases CD8+ T cells that were newly proliferative (Ki67+) **(Figure 3)**[12]

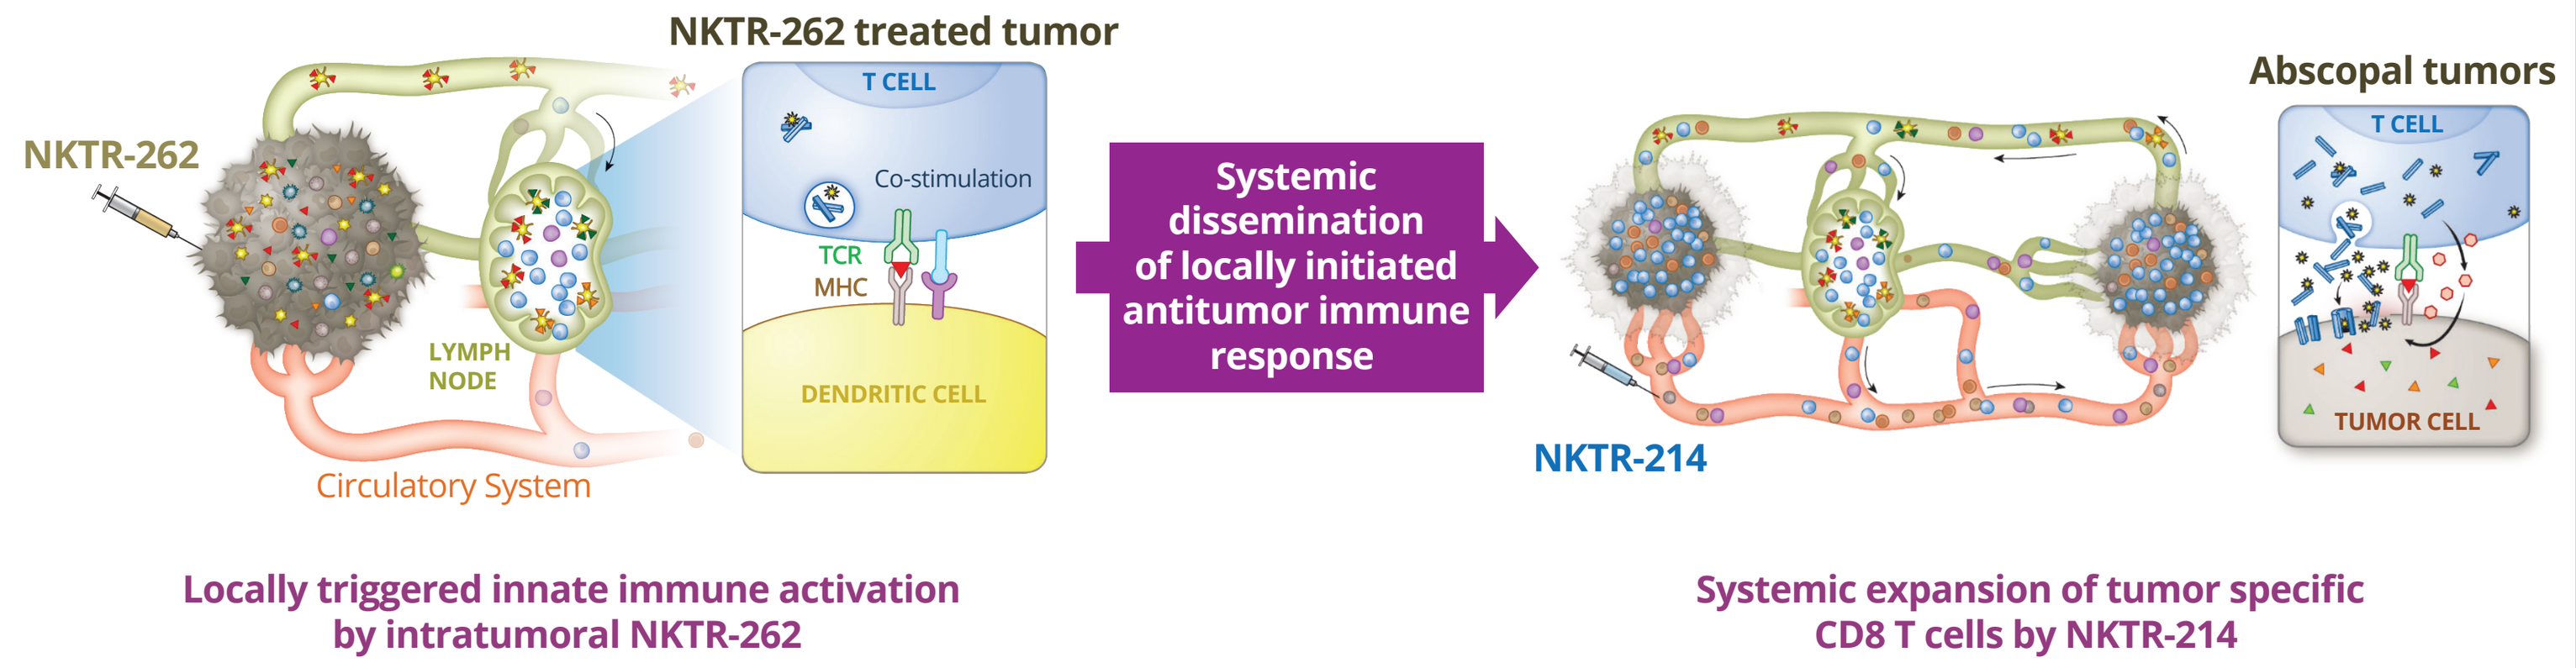
NKTR-262 treated tumor
NKTR-262
T CELL
Co-stimulation
TCR
MHC
LYMPH NODE
DENDRITIC CELL
Circulatory System
Systemic dissemination of locally initiated antitumor immune response
NKTR-214
Abscopal tumors
T CELL
TUMOR CELL
Locally triggered innate immune activation by intratumoral NKTR-262
Systemic expansion of tumor specific CD8 T cells by NKTR-214

Figure 2: Effective tumor growth inhibition by combining NKTR-262 with NKTR-214 in multiple syngeneic mouse tumor models

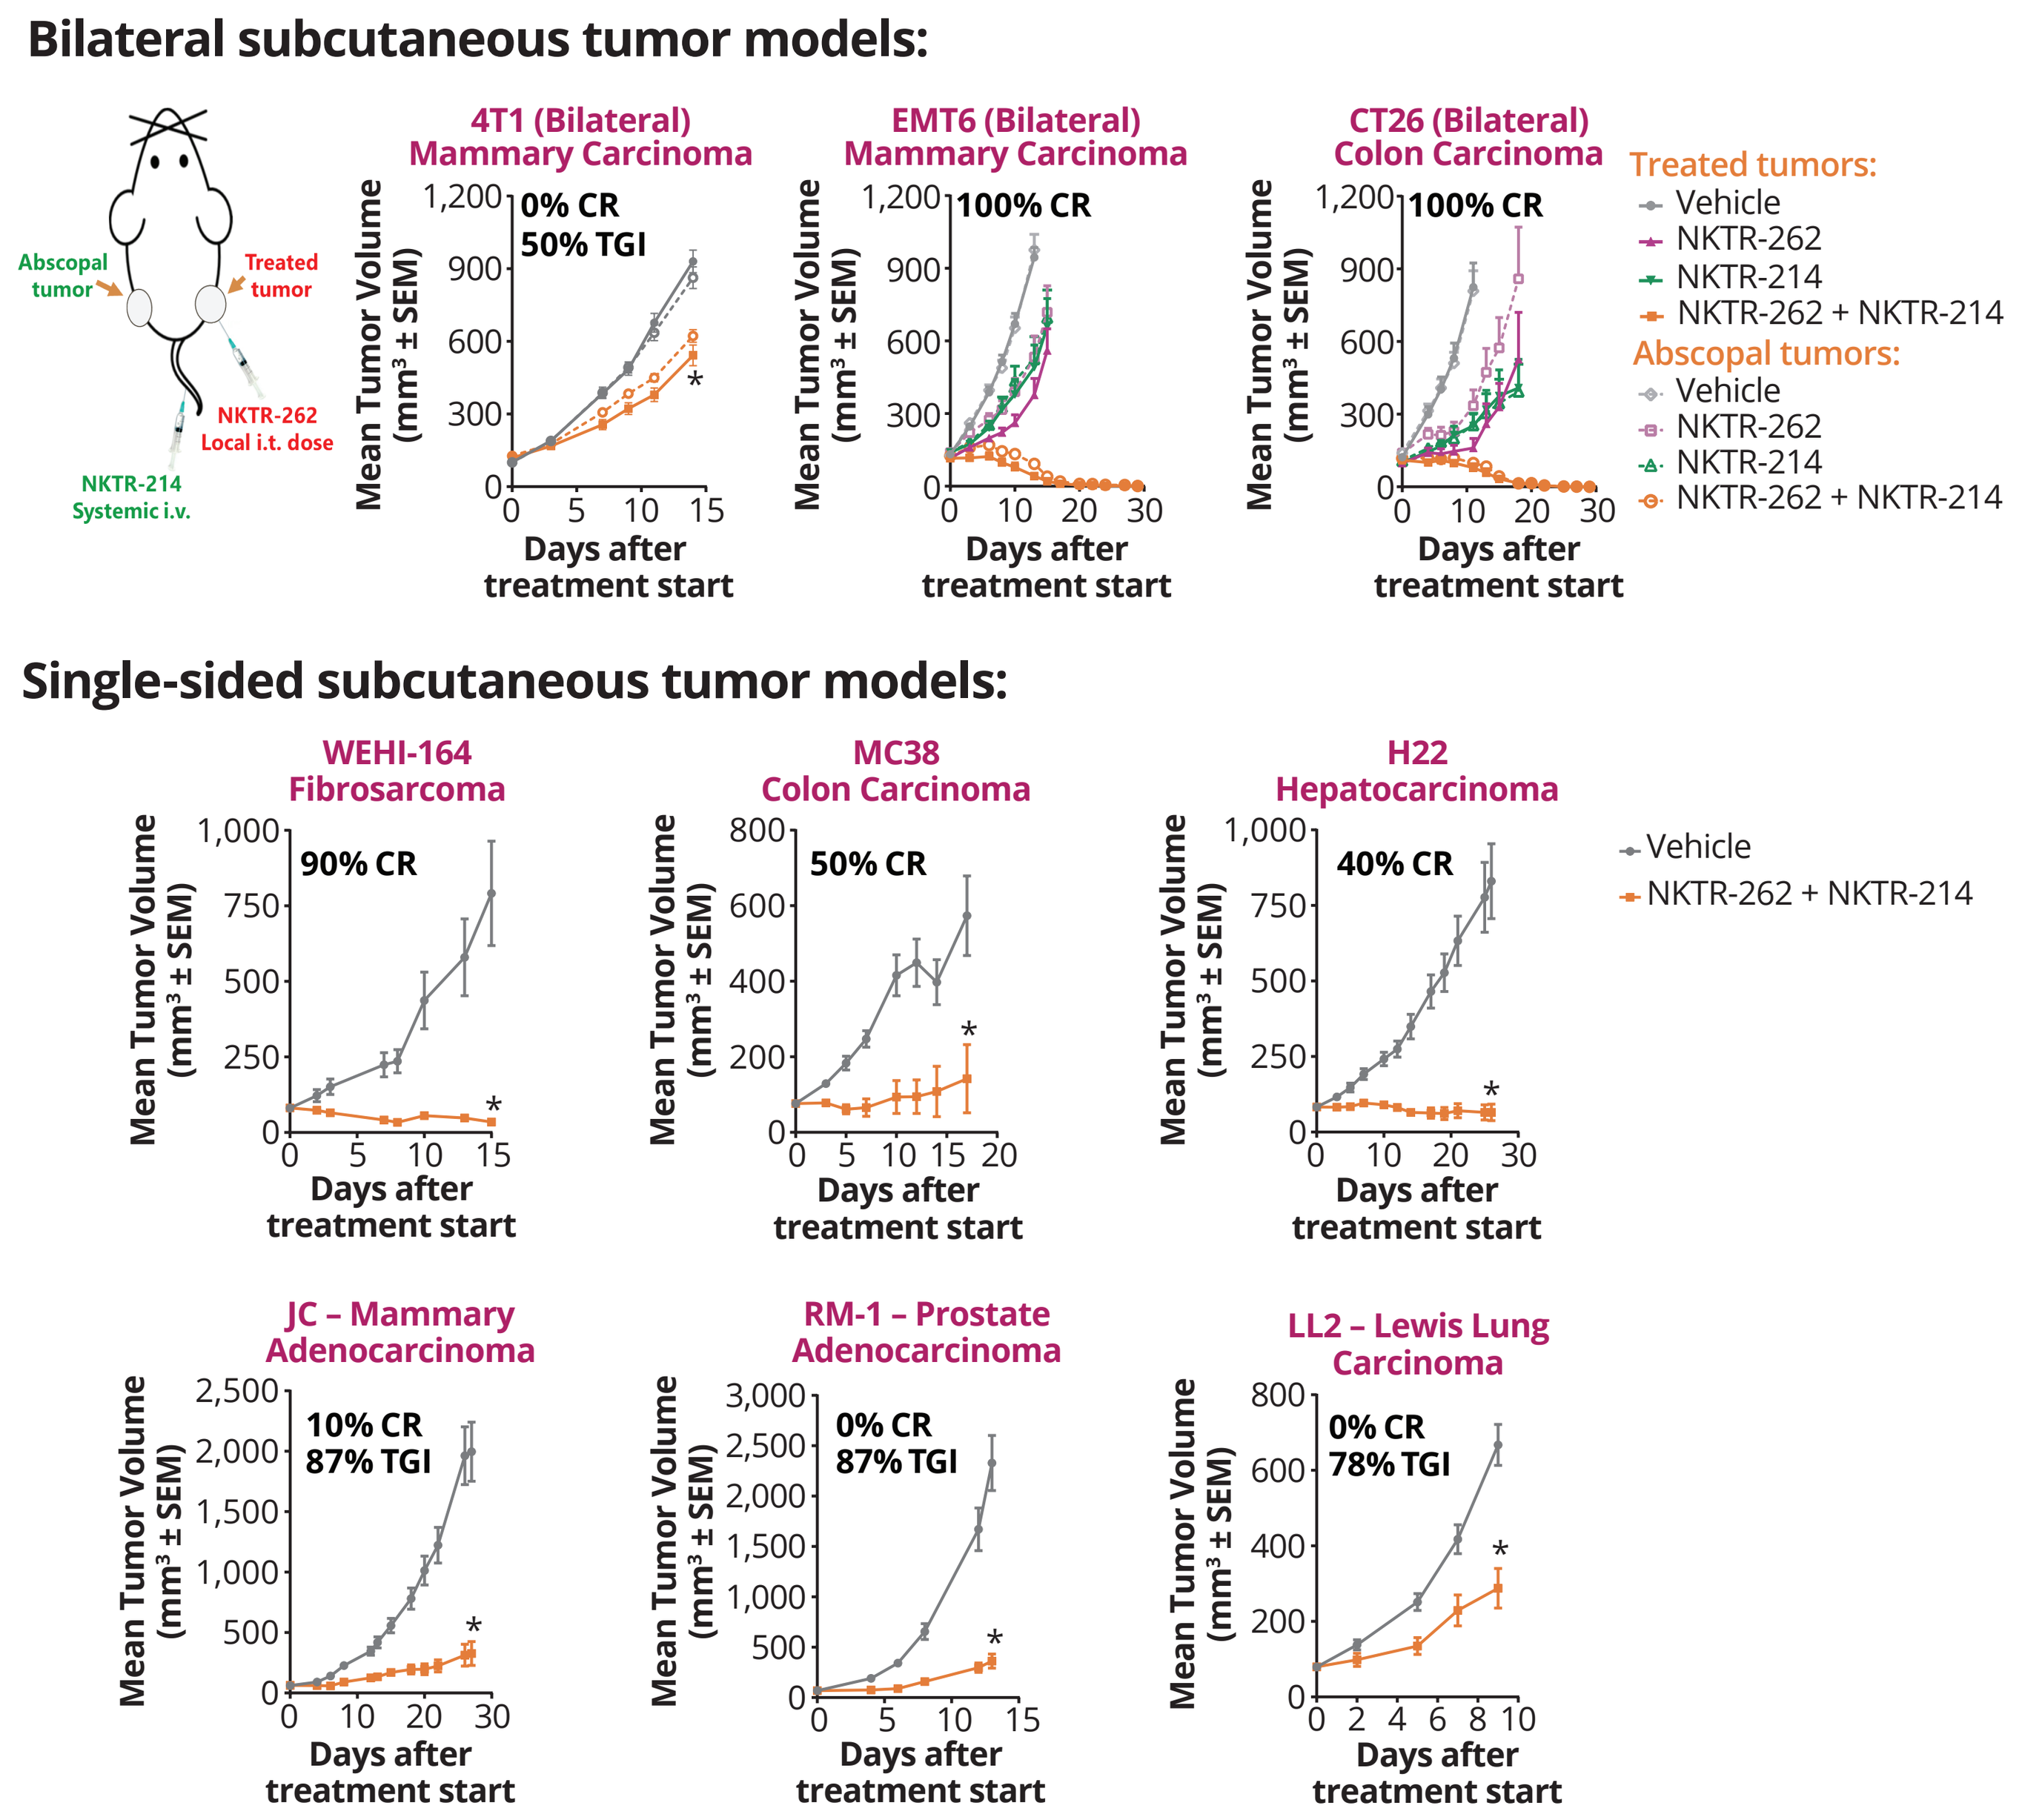
Bilateral subcutaneous tumor models:
Abscopal tumor
Treated tumor
NKTR-262 Local i.t. dose
NKTR-214 Systemic i.v.
4T1 (Bilateral) Mammary Carcinoma
0% CR
50% TGI
EMT6 (Bilateral) Mammary Carcinoma
100% CR
CT26 (Bilateral) Colon Carcinoma
100% CR
Mean Tumor Volume (mm³ ± SEM)
Days after treatment start
Treated tumors:
Vehicle
NKTR-262
NKTR-214
NKTR-262 + NKTR-214
Abscopal tumors:
Vehicle
NKTR-262
NKTR-214
NKTR-262 + NKTR-214
Single-sided subcutaneous tumor models:
WEHI-164 Fibrosarcoma
90% CR
MC38 Colon Carcinoma
50% CR
H22 Hepatocarcinoma
40% CR
Vehicle
NKTR-262 + NKTR-214
JC – Mammary Adenocarcinoma
10% CR
87% TGI
RM-1 – Prostate Adenocarcinoma
0% CR
87% TGI
LL2 – Lewis Lung Carcinoma
0% CR
78% TGI

- A single NKTR-262 dose (20 µg) combined with NKTR-214 (0.8 mg/kg, iv, q9d x 3 doses) inhibits tumor growth in syngeneic mouse tumor models
- In bilateral models, only the right flank tumor was treated with NKTR-262 to assess abscopal efficacy in the untreated left flank tumors (*, p<0.05 relative to vehicle)

Figure 3: NKTR-214 Promotes T Cell Proliferation and Selectively Increases T Cells in the Tumor

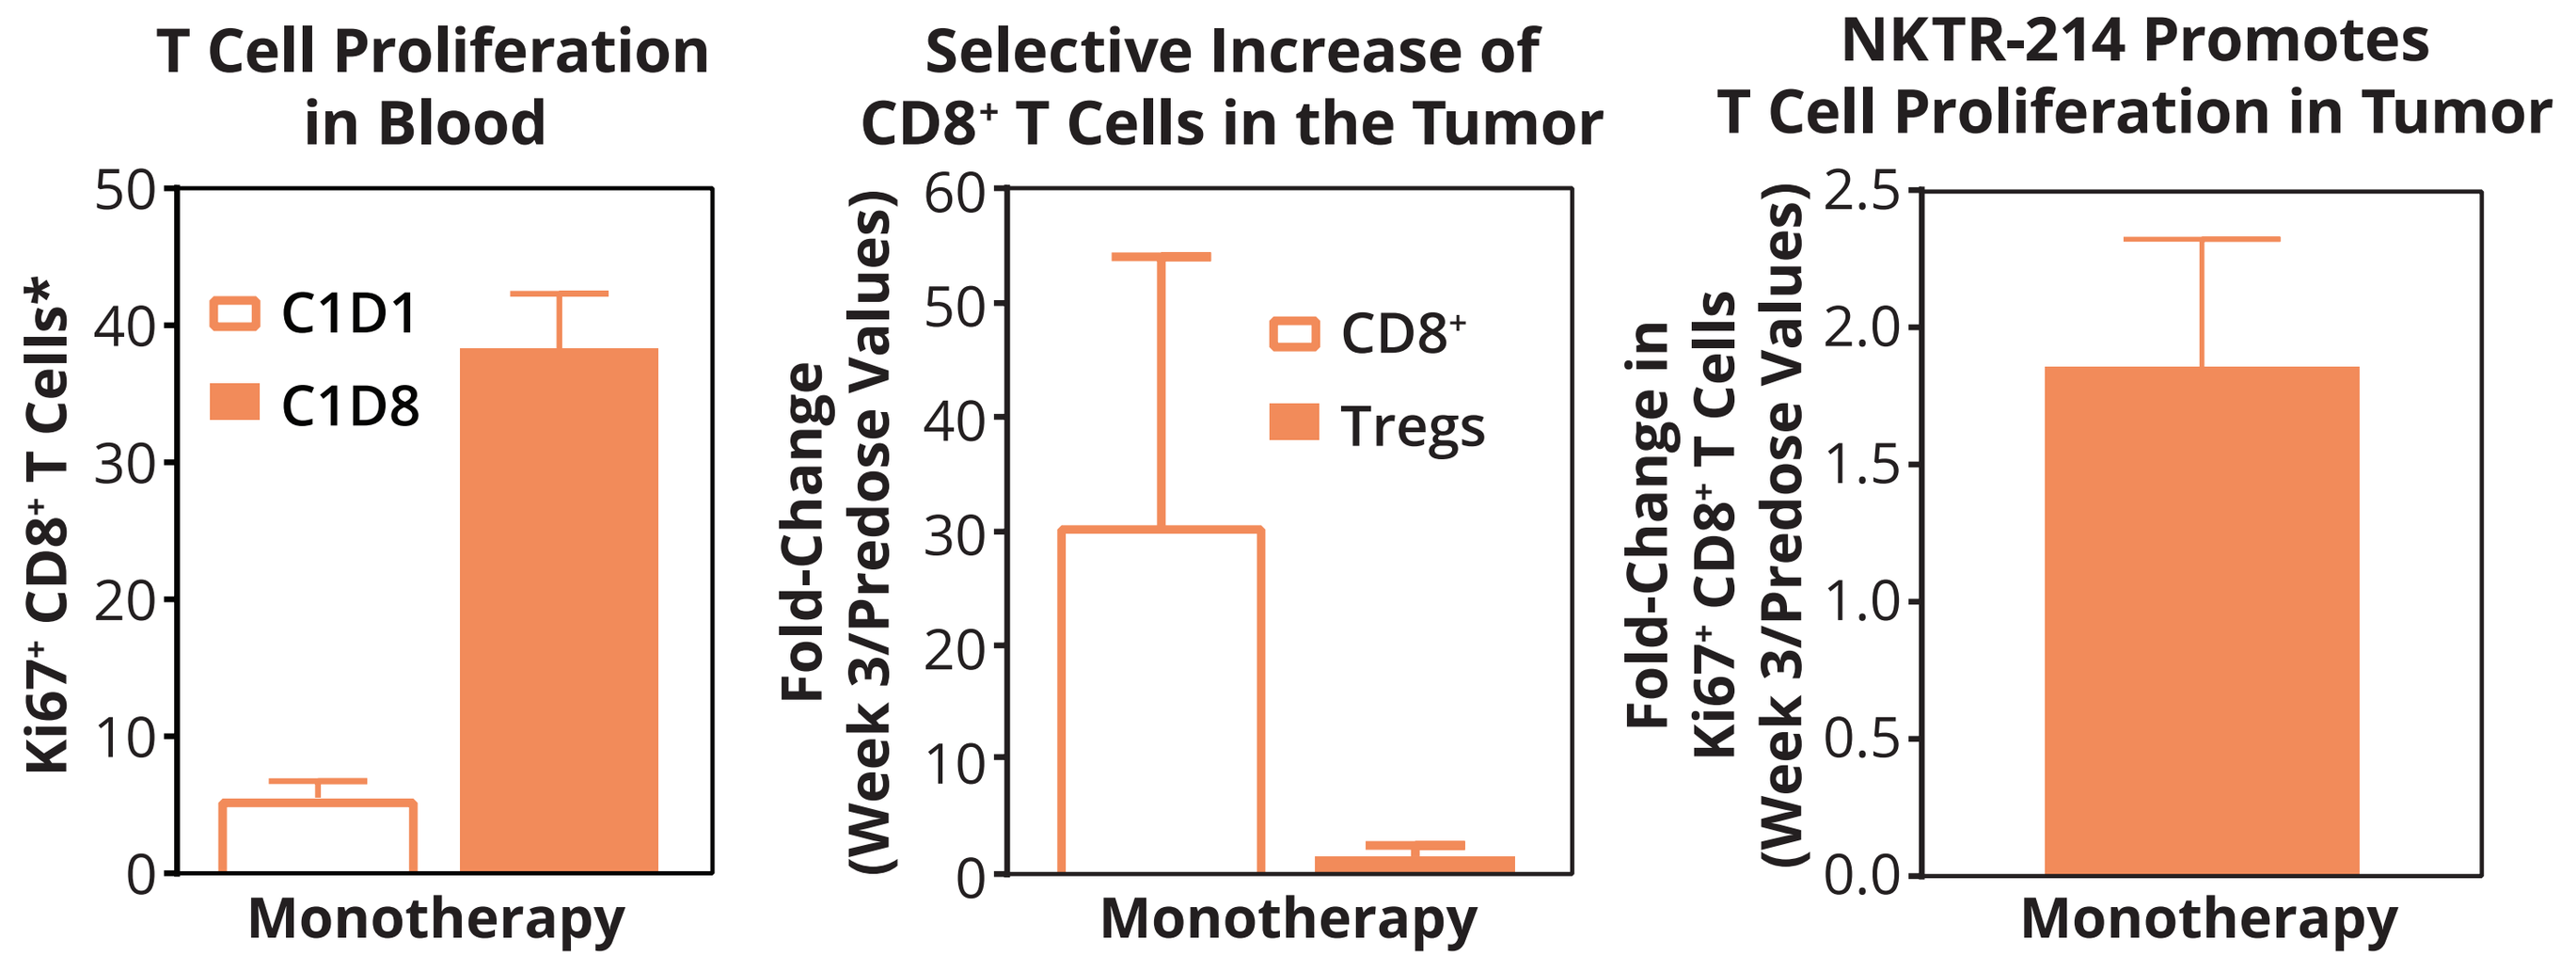
T Cell Proliferation in Blood
Ki67⁺ CD8⁺ T Cells*
C1D1
C1D8
Monotherapy
Selective Increase of CD8⁺ T Cells in the Tumor
Fold-Change (Week 3/Predose Values)
CD8⁺
Tregs
Monotherapy
NKTR-214 Promotes T Cell Proliferation in Tumor
Fold-Change in Ki67⁺ CD8⁺ T Cells (Week 3/Predose Values)
Monotherapy

*Data are expressed as %Ki67$^+$ CD8$^+$ T cells on Cycle 1 Day 1 (C1D1) and Cycle 1 Day 8 (C1D8) plotted as Mean ± Standard Error

REFERENCES

1) Reck M, Rodriguez-Abreu D, Robinson AG, et al. *N Engl J Med*. 2016;375(19):1823-1833.
2) Brahmer J, Reckamp KL, Baas P, et al. *N Engl J Med*. 2015;373(2):123-35
3) Daud AI, Wolchok JD, Robert C, et al. *J Clin Oncol.* 2016;34(34):4102-9
4) Daud AI, Loo K, Pauli ML, et al. *J Clin Invest.* 2016;126(9):3447–52
5) Huang, et al. *Cellular and Molecular Immunology*. 2018;15:428–437
6) Sim GC, Martin-Orozco N, Jin L, et al. *J Clin Invest*. 2014;124:99-110
7) Kivimäe S, Marlene H, Rhoneil P, et al. Presented at AACR 2018 Annual Meeting, April 14-18, 2018. Poster #3755
8) Lee M, Anand N, Haiying C, et al. Presented at AACR 2018 Annual Meeting, April 14-18, 2018. Poster #2755
9) Charych DH, Hoch U, Langowski JL, et al. Clin Cancer Res. 2016;22(3):680–90
10) Bernatchez C, Haymaker C, Tannir NM, et al. Presented at SITC 2016, National Harbor, MD. Poster #387
11) Bernatchez C, Bentebibel SE, Hurwitz ME, et al. Presented at ASCO 2017, Chicago, IL
12) Nektar Therapeutics Analyst & Investor Event at 2017 ASCO Annual Meeting (2017). [online] Available at: http://ir.nektar.com/events-and-presentations/events

Presented in Munich, Germany at ESMO, October 19 - 23, 2018
Study sponsored by Nektar Therapeutics

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster.

REVEAL STUDY

The combination of NKTR-262 and NKTR-214 with or without nivolumab could engage the entire immune activation cascade required for systemic tumor clearance from local tumor antigen production to a sustained systemic T cell response

STUDY DESIGN

Primary Objectives:

- Evaluate the safety and tolerability, and define the maximum tolerated dose (MTD) or recommended phase 2 dose (RP2D) of NKTR-262 in combination with NKTR-214 (doublet) and the safety and tolerability of NKTR-262 and NKTR-214 plus nivolumab (triplet)
- Evaluate the anti-tumor activity of the doublet and the triplet by assessing the objective response rate (ORR) per Response Evaluation Criteria In Solid Tumors version 1.1 (RECIST 1.1)

Secondary Objectives:

- Evaluate the anti-tumor activity of the combination of the doublet and the triplet by assessing progression-free survival (PFS) and overall survival (OS)
- Evaluate the proportion of patients with an abscopal response by RECIST 1.1 after the initiation of treatment of the doublet or the triplet

DESIGN

- The REVEAL study design is shown in **Figure 4**
- Enrollment into the dose expansion cohorts will commence once the RP2D is established
- Patients will be treated until unacceptable toxicity, death, or disease progression per RECIST 1.1, Investigator's decision to discontinue treatment, the patient withdraws consent, is lost to follow-up

Figure 4: Study design

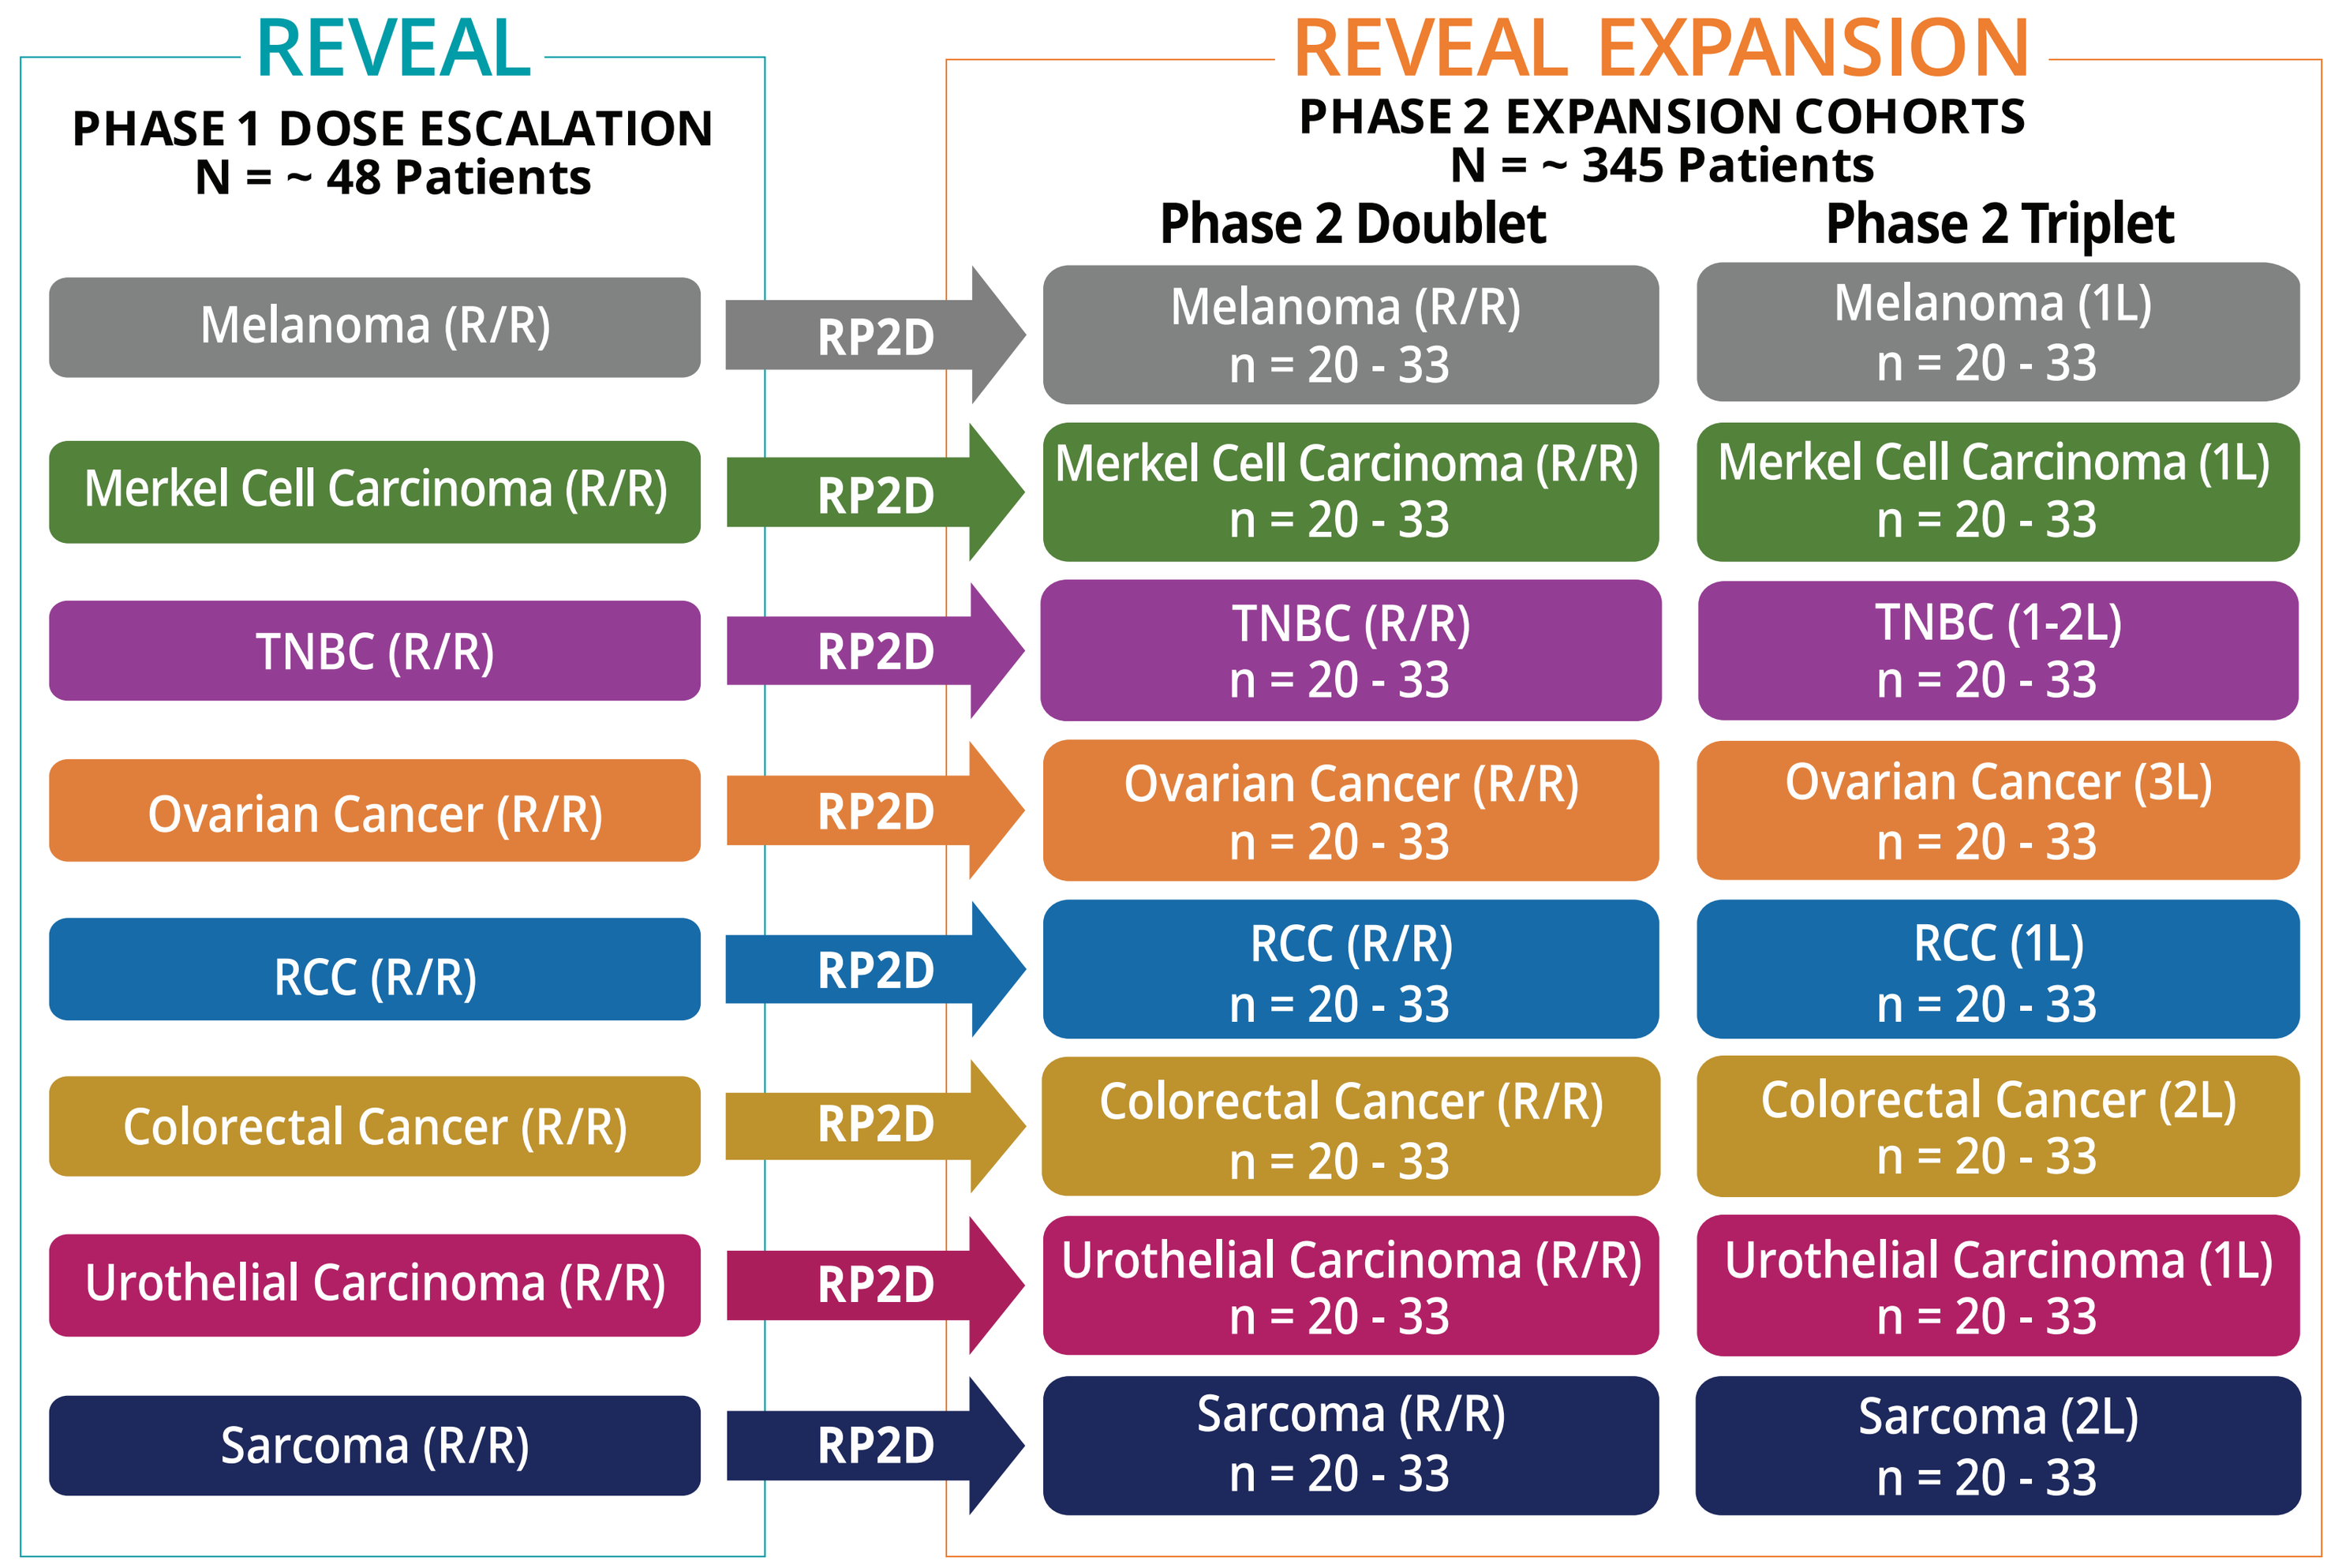
REVEAL
PHASE 1 DOSE ESCALATION
N = ~ 48 Patients
Melanoma (R/R)
Merkel Cell Carcinoma (R/R)
TNBC (R/R)
Ovarian Cancer (R/R)
RCC (R/R)
Colorectal Cancer (R/R)
Urothelial Carcinoma (R/R)
Sarcoma (R/R)
RP2D
REVEAL EXPANSION
PHASE 2 EXPANSION COHORTS
N = ~ 345 Patients
Phase 2 Doublet
Melanoma (R/R) n = 20 - 33
Merkel Cell Carcinoma (R/R) n = 20 - 33
TNBC (R/R) n = 20 - 33
Ovarian Cancer (R/R) n = 20 - 33
RCC (R/R) n = 20 - 33
Colorectal Cancer (R/R) n = 20 - 33
Urothelial Carcinoma (R/R) n = 20 - 33
Sarcoma (R/R) n = 20 - 33
Phase 2 Triplet
Melanoma (1L) n = 20 - 33
Merkel Cell Carcinoma (1L) n = 20 - 33
TNBC (1-2L) n = 20 - 33
Ovarian Cancer (3L) n = 20 - 33
RCC (1L) n = 20 - 33
Colorectal Cancer (2L) n = 20 - 33
Urothelial Carcinoma (1L) n = 20 - 33
Sarcoma (2L) n = 20 - 33

1L, 1st line; TNBC, triple negative breast cancer; RCC; Renal cell carcinoma; R/R, Relapsed refractory

Sample Dose Escalation Scheme: Simultaneous 3+3 Design (Phase 1)*

	NKTR-262	NKTR-214	Nivolumab
Cohort 1	0.03 mg	0.006 mg/kg q3w	
Cohort 2	0.06 mg	0.006 mg/kg q3w	
Cohort 3**	0.06 mg	0.006 mg/kg q3w	
Cohort 4	0.12 mg	0.006 mg/kg q3w	
Cohort 5	0.24 mg	0.006 mg/kg q3w	
Cohort A***	0.24 mg	0.006 mg/kg q3w	
Cohort B***	0.24 mg	0.006 mg/kg q3w	360 mg q3w

- Dose modifications of NKTR-262 (either dose escalation or de-escalation) may be adjusted based on clinical observations as per the sample dose escalation scheme

q3w: every 3 weeks
* With the exception of the Cohort 1 dose, dose levels for subsequent cohorts may be adjusted based on clinical observations. Dose escalation will not exceed double the prior dose of NKTR-262
** Starting at Cycle 2, Cohort 3-5 will explore same-day administration of NKTR-262 and NKTR-214 at the dose level of Cohort 2
*** Cohorts A and B (with up to 12 patients per cohort) will explore same-day administration of NKTR-262 and NKTR-214, or NKTR-262, NKTR-214, and nivolumab, respectively, starting at Cycle 1

Phase 2 (Dose Expansion)

DOUBLET (NKTR-262 Recommended Phase 2 Dose)		TRIPLET (NKTR-262 Recommended Phase 2 Dose)	
Cycles 1-3 (same day administration)	Cycles 4+	Cycles 1-3 (same day administration)	Cycles 4+
NKTR-262 + NKTR-214	NKTR-214	NKTR-262 + NKTR-214 + Nivolumab	NKTR-214 + Nivolumab

- Phase 2 dose expansion initiation for both the doublet or triplet combination cohorts will be based on safety and tolerability findings and anti-tumor effect using RECIST 1.1
- NKTR-262 will be administered in 3-week cycles
- Patients may receive an additional 3 cycles of NKTR-262 IT injection after assessment of anti-tumor effect at each designated disease staging in consultation with Medical Monitor
- Patients treated with doublet therapy will have NKTR-262 IT injection administered first, followed by NKTR-214 infusion
- Patients treated with triplet therapy will have NKTR-262 IT injection administered first, followed by NKTR-214 infusion, followed by nivolumab infusion

ELIGIBILITY

Key INCLUSION Criteria

- Histologically confirmed diagnosis of a locally advanced (not amenable to curative therapy such as surgical resection) metastatic cancer of the following histologies: melanoma, Merkel cell carcinoma, triple-negative breast cancer (TNBC), ovarian carcinoma, renal cell carcinoma (RCC), colorectal cancer, urothelial carcinoma, or sarcoma
- Life expectancy > 12 weeks as determined by the Investigator
- Eastern Cooperative Oncology Group (ECOG) performance status 0 to 1
- Measurable disease per RECIST 1.1
- Injected lesions (up to two) must be between 20 mm and 90 mm in diameter for IT injection; lesions must be accessible for baseline and on-treatment biopsies. Tumors encasing major vascular structures (ie, carotid artery or tumors close to other vital organs) are not considered appropriate for intratumoral injection

STATUS

- The Phase 1 dose escalation portion of REVEAL is open and recruiting. REVEAL Phase 2 is scheduled to open in the first half of 2019 (NCT03435640)