NKTR-181: An Orally Available Mu-Opioid Agonist with Slow Rate of Uptake into the CNS, Exhibits Comparable Analgesic Efficacy with Reduced Abuse Liability and CNS Mediated Side Effects Compared to Oxycodone

Kathleen Gogas[1], PhD, Juergen Pfeiffer[1], MS, Irene Choi[1], PhD, Juli Evans[1], RVT, Dan McWeeney[1], BA, Phi Quach[1], BS, Patricia Trinchero[1], David V Gauvin*, PhD, C. Simone Fishburn[1], PhD, Stephen D Harrison[1], PhD, Timothy A Riley[2], PhD, Stephen K Doberstein[1], PhD, Jennifer Riggs-Sauthier[2], PhD
Nektar Therapeutics, [1]San Francisco, CA; [2]Huntsville, AL *MPI Research Inc, Mattawan, Michigan

NEKTAR

Introduction

- Opioid analgesics are highly effective for the treatment of pain.[1]
- Opioids are associated with serious CNS related side effects, including respiratory depression, sedation and abuse liability.[1]
- The abuse properties of opioid analgesics are believed to relate to their rapid entry into the CNS.[2,3]
- NKTR-181 is a mu-opioid analgesic molecule designed to provide clinically relavent analgesia while reducing CNS mediated side effects.[4]
- NKTR-181 binds to the mu-opioid receptor and acts as a full agonist in adenylate cyclase inhibition assays.[4]
- NKTR-181 exhibits a slower rate of CNS uptake in rats compared to commonly used opioids.[5,6]
- NKTR-181 displays markedly lower abuse liability than commonly used opioids when evaluated in self-administration studies in non-human primates and rats.[6,7]

Background

NKTR-181 exhibits pharmacological behavior characteristic of opioids and has been shown to have maximal analgesic activity in preclinical models of pain following oral administration in mice.

NKTR-181 Shows Analgesic Activity in a Preclinical Model of Pain[4]

NKTR-181 shows comparable efficacy to oxycodone in the acetic acid writhing model in mice

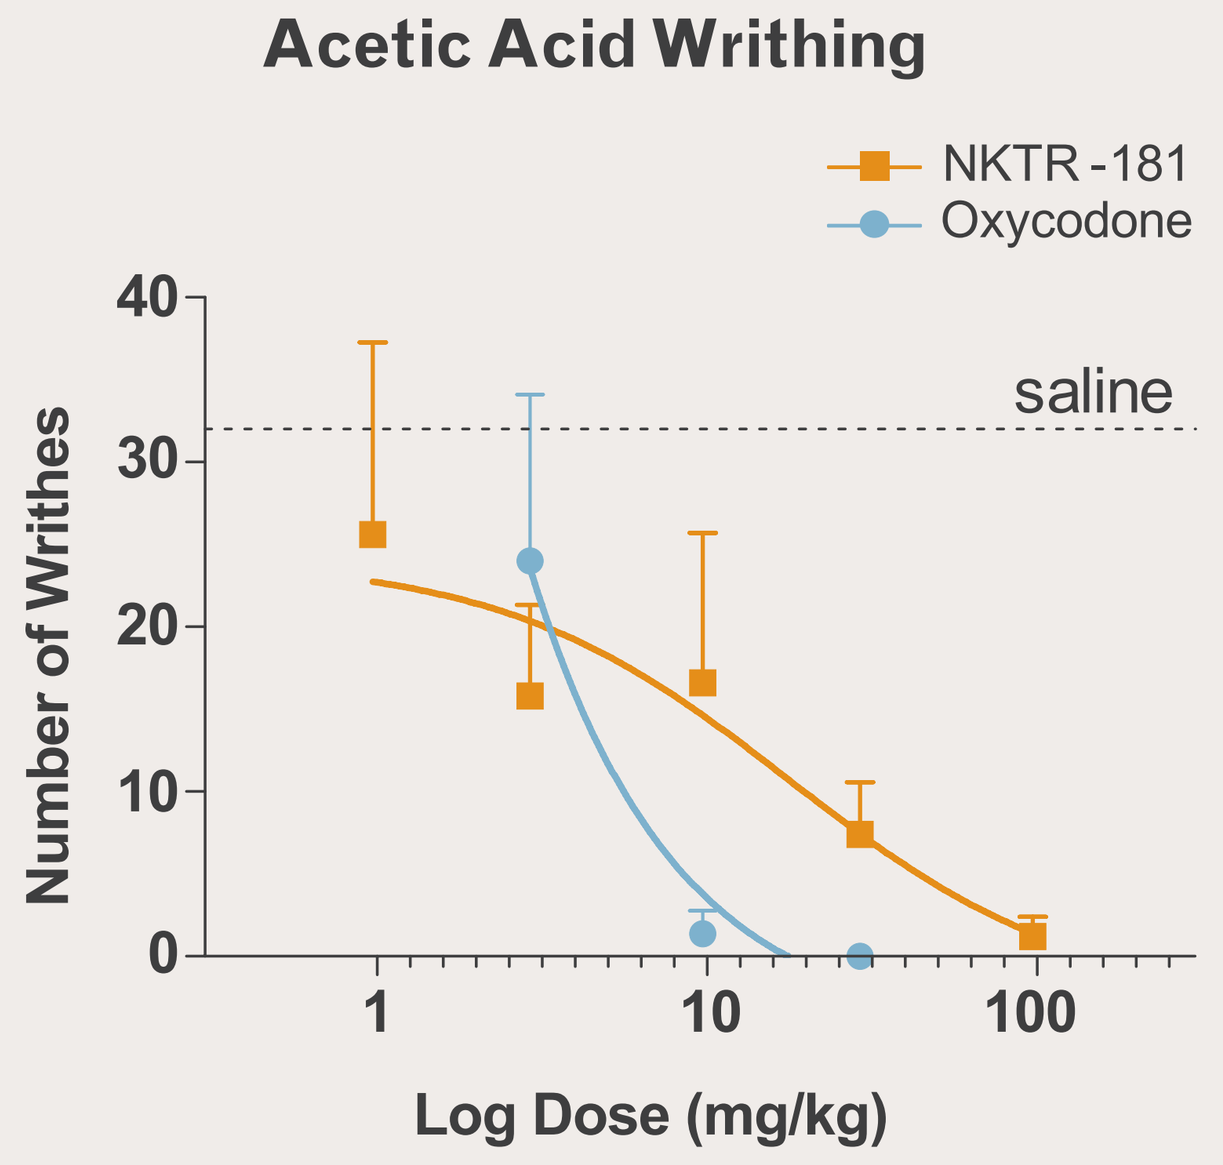
Acetic Acid Writhing
NKTR-181
Oxycodone
saline
Number of Writhes
40
30
20
10
0
1
10
100
Log Dose (mg/kg)

- NKTR-181 produces full suppression of acetic acid writhing
- Maximal efficacy achieved at 100 mg/kg p.o.
- Potency is maximal 1-2 hr after oral dosing
- Activity sustained for up to 8 hours after oral dosing

For acetic acid writhing studies, CD-1 mice were treated orally with test article, and 30 minutes later were injected intraperitoneally with 0.5% acetic acid (0.1 mL/10 g body weight). Writhes were counted over a 20 minute period. Data shown represent mean + SEM values (n=5) demonstrate the response 30 minutes post-dose, and is representative for NKTR-181 at all time points measured.

Results

NKTR-181 Displays Slowed Entry into the Brain Compared to Oxycodone

NKTR-181 displays a dramatically different distribution between brain and plasma following oral delivery in mice

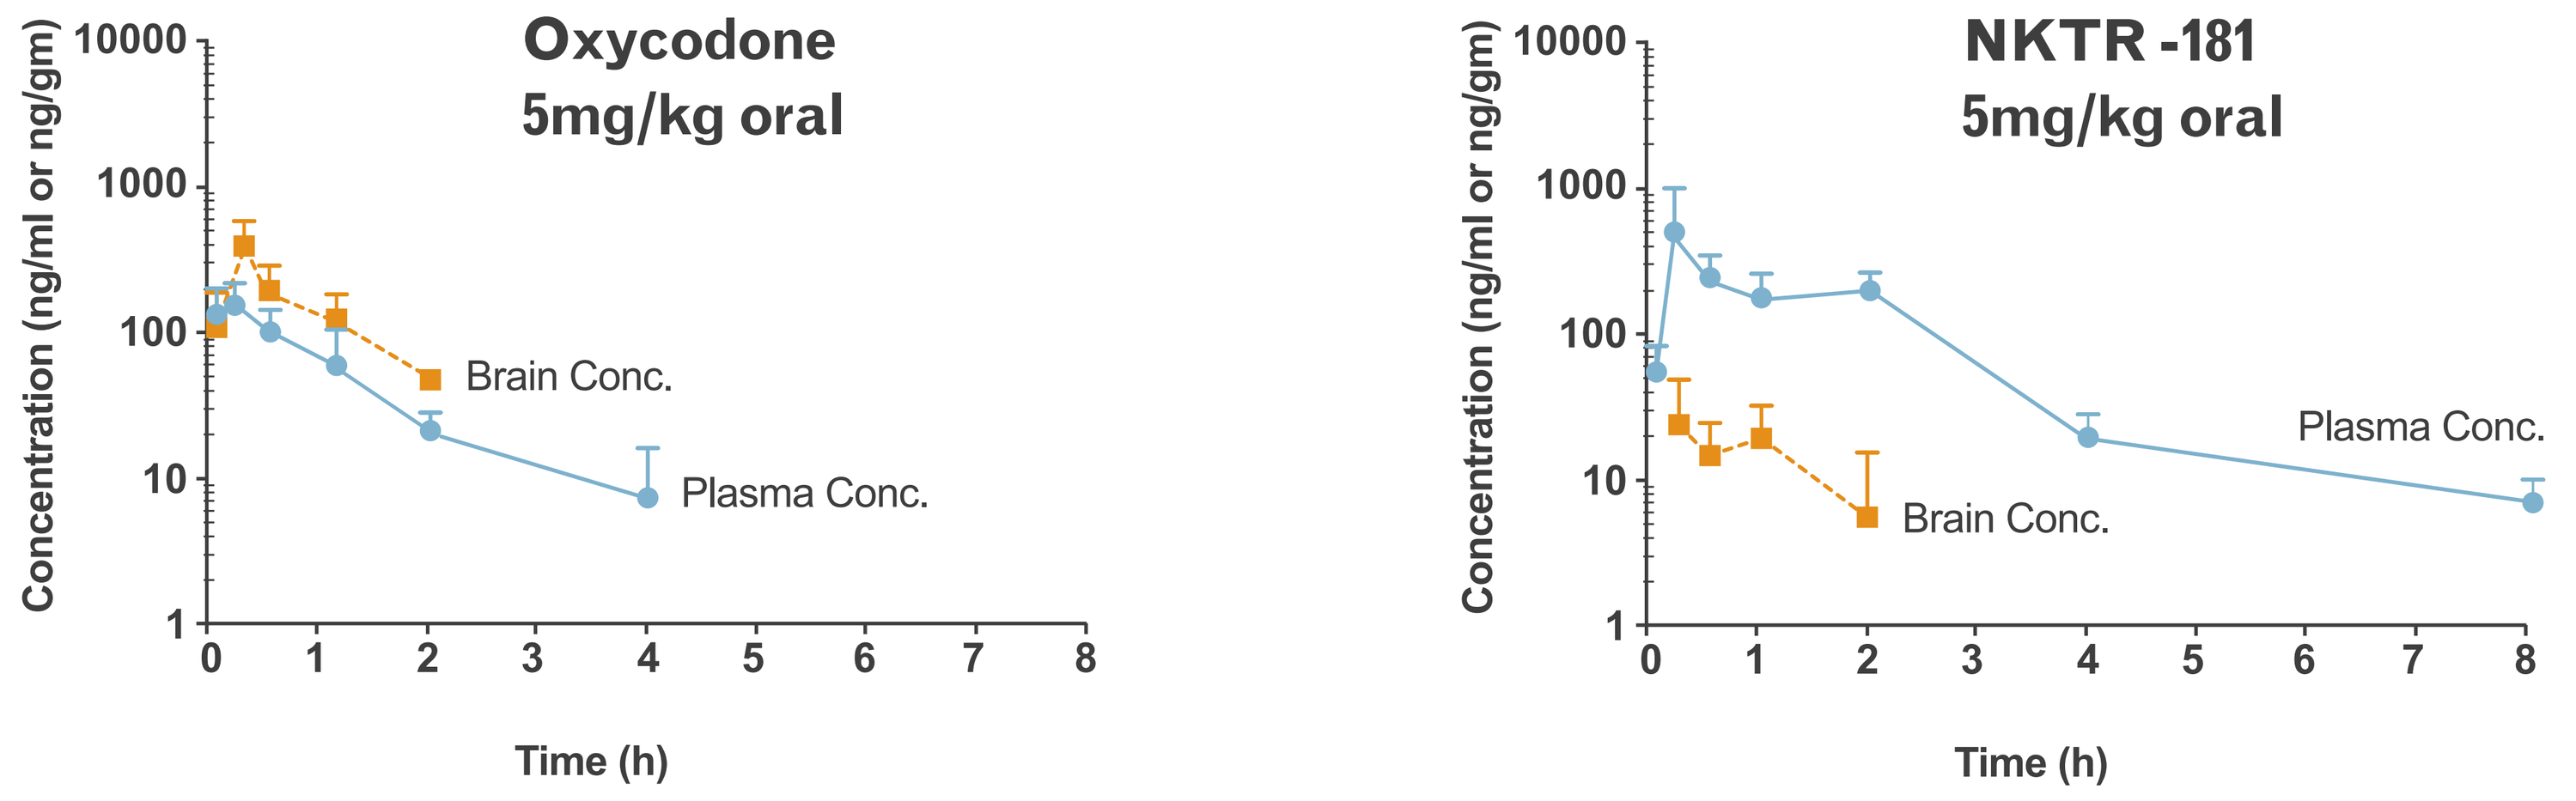
Oxycodone
5mg/kg oral
Brain Conc.
Plasma Conc.
NKTR-181
5mg/kg oral
Plasma Conc.
Brain Conc.
Concentration (ng/ml or ng/gm)
10000
1000
100
10
1
0 1 2 3 4 5 6 7 8
Time (h)

Mice were treated orally with NKTR-181 or oxycodone at 5 mg/kg, and were then sacrificed at the time points indicated. Plasma samples were taken, and, following exsanguination, brains were excised and homogenized. Oxycodone and NKTR-181 were quantitated using LC-MS/MS. Data represent the mean + SD (n=3). Rate of entry to the brain was calculated as the ratio of Cmax: Tmax for the brain concentrations of the test articles.

	Rate of Brain Entry (C_{max}/T_{max}: ug/g/hr)
Oxycodone	1.21
NKTR-181	0.09

NKTR-181 Demonstrates Less Sedative Potential Than Oxycodone at Equianalgesic Doses

NKTR-181 exhibits less impact on CNS coordination compared to oxycodone

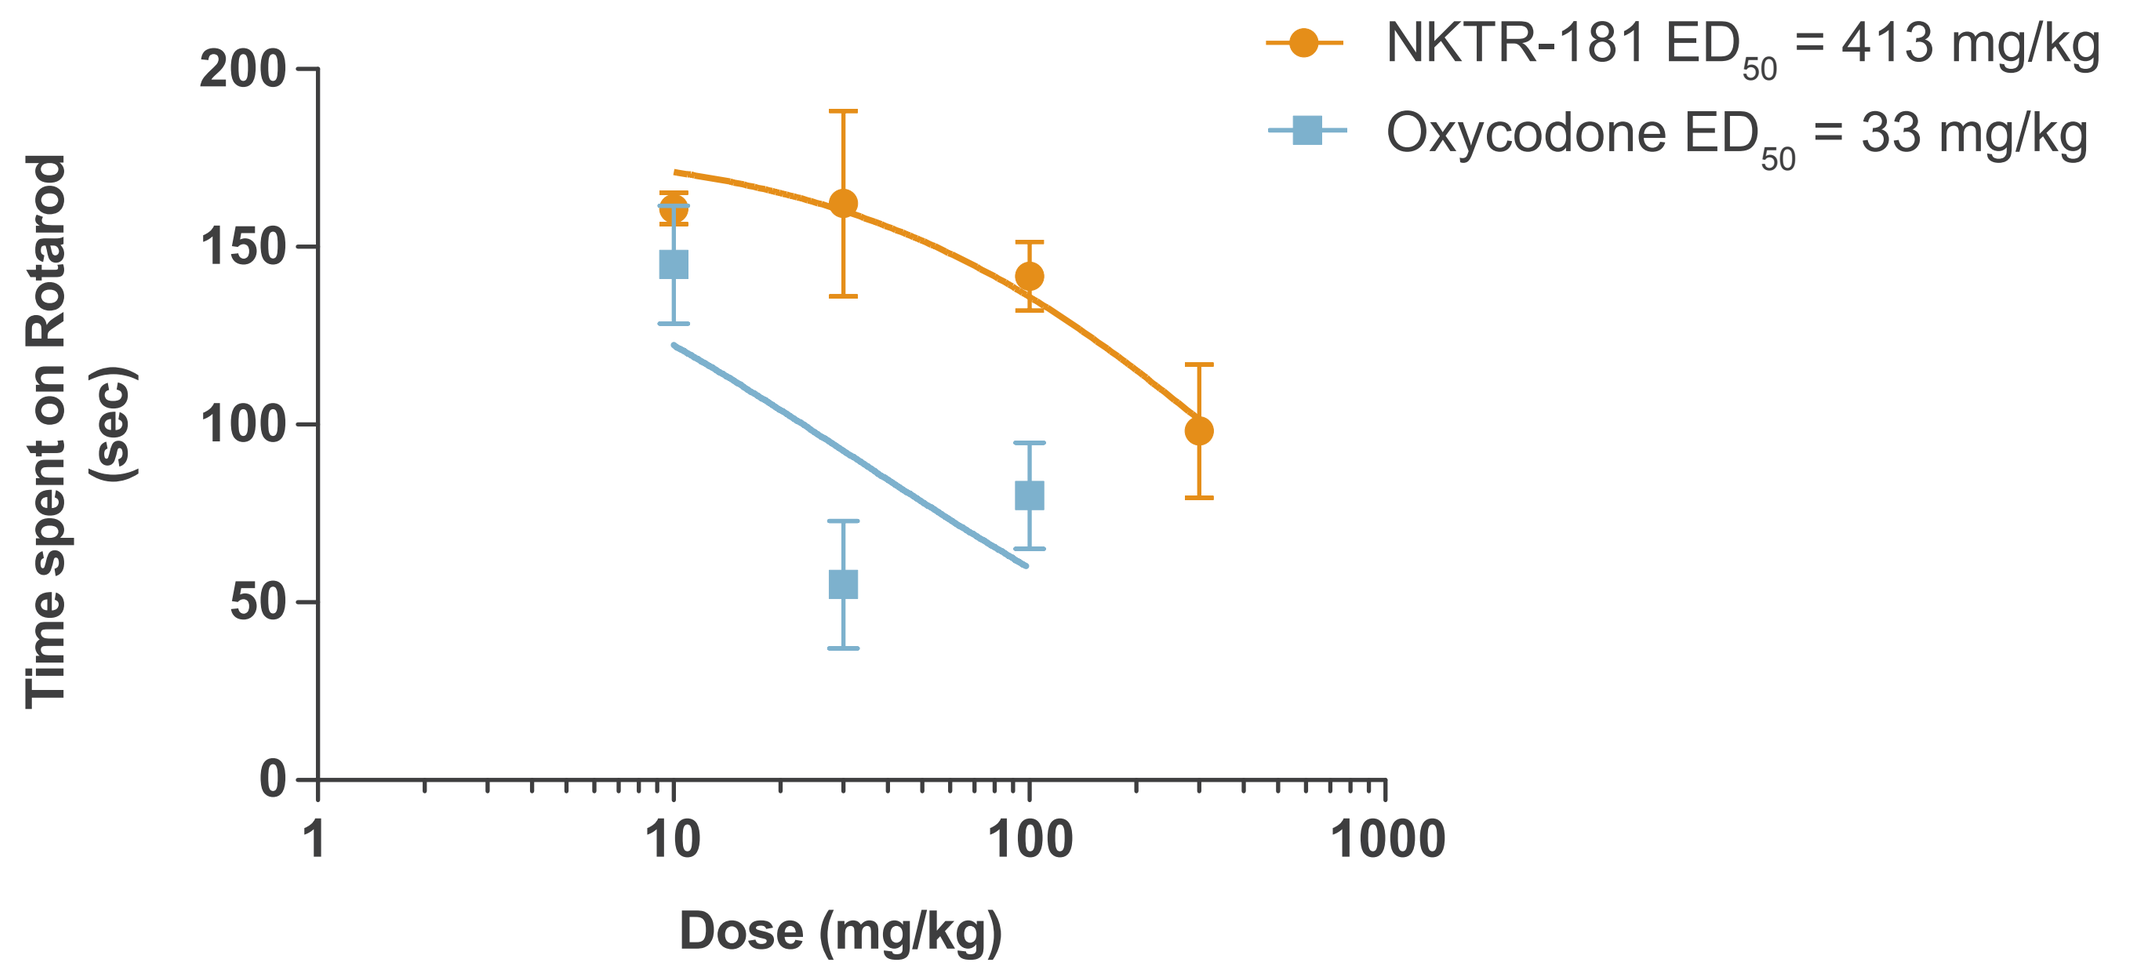
NKTR-181 ED50 = 413 mg/kg
Oxycodone ED50 = 33 mg/kg
Time spent on Rotarod (sec)
200
150
100
50
0
1
10
100
1000
Dose (mg/kg)

Animals were trained to run on the treadmill at a constant speed of 10 RPM one day prior to the day of study. Those rats that were able to stay on the rod for 300 seconds were considered trained. On the day of study, animals were dosed with either NKTR-181 [10-300 mg/kg, p.o.], oxycodone [10-100 mg/kg, p.o.] or vehicle and were placed on the rotarod treadmill at 0 and 60 minutes post dose. The treadmill was set at a constant speed of 4 RPM for 15 seconds, at this point the timer started (T=0). After this 15 second period, the rotarod was set to accelerate from 4-40 RPM over a five minute period (using the built in program of the rotarod). The time (in seconds) that each animal stopped running and tripped the plate was recorded as the animal's run time. Animals that ran for 300 seconds were taken off the treadmill and 300 seconds was recorded as the run time. Data shown represent mean + SEM values (n=5). Curve fits were done using Nonlinear Regression in GraphPad Prism.

NKTR-181 Displays Low Abuse Liability in Drug Discrimination Studies in Rats

Significant decrease in abuse liability for NKTR-181 compared to oxycodone following oral administration

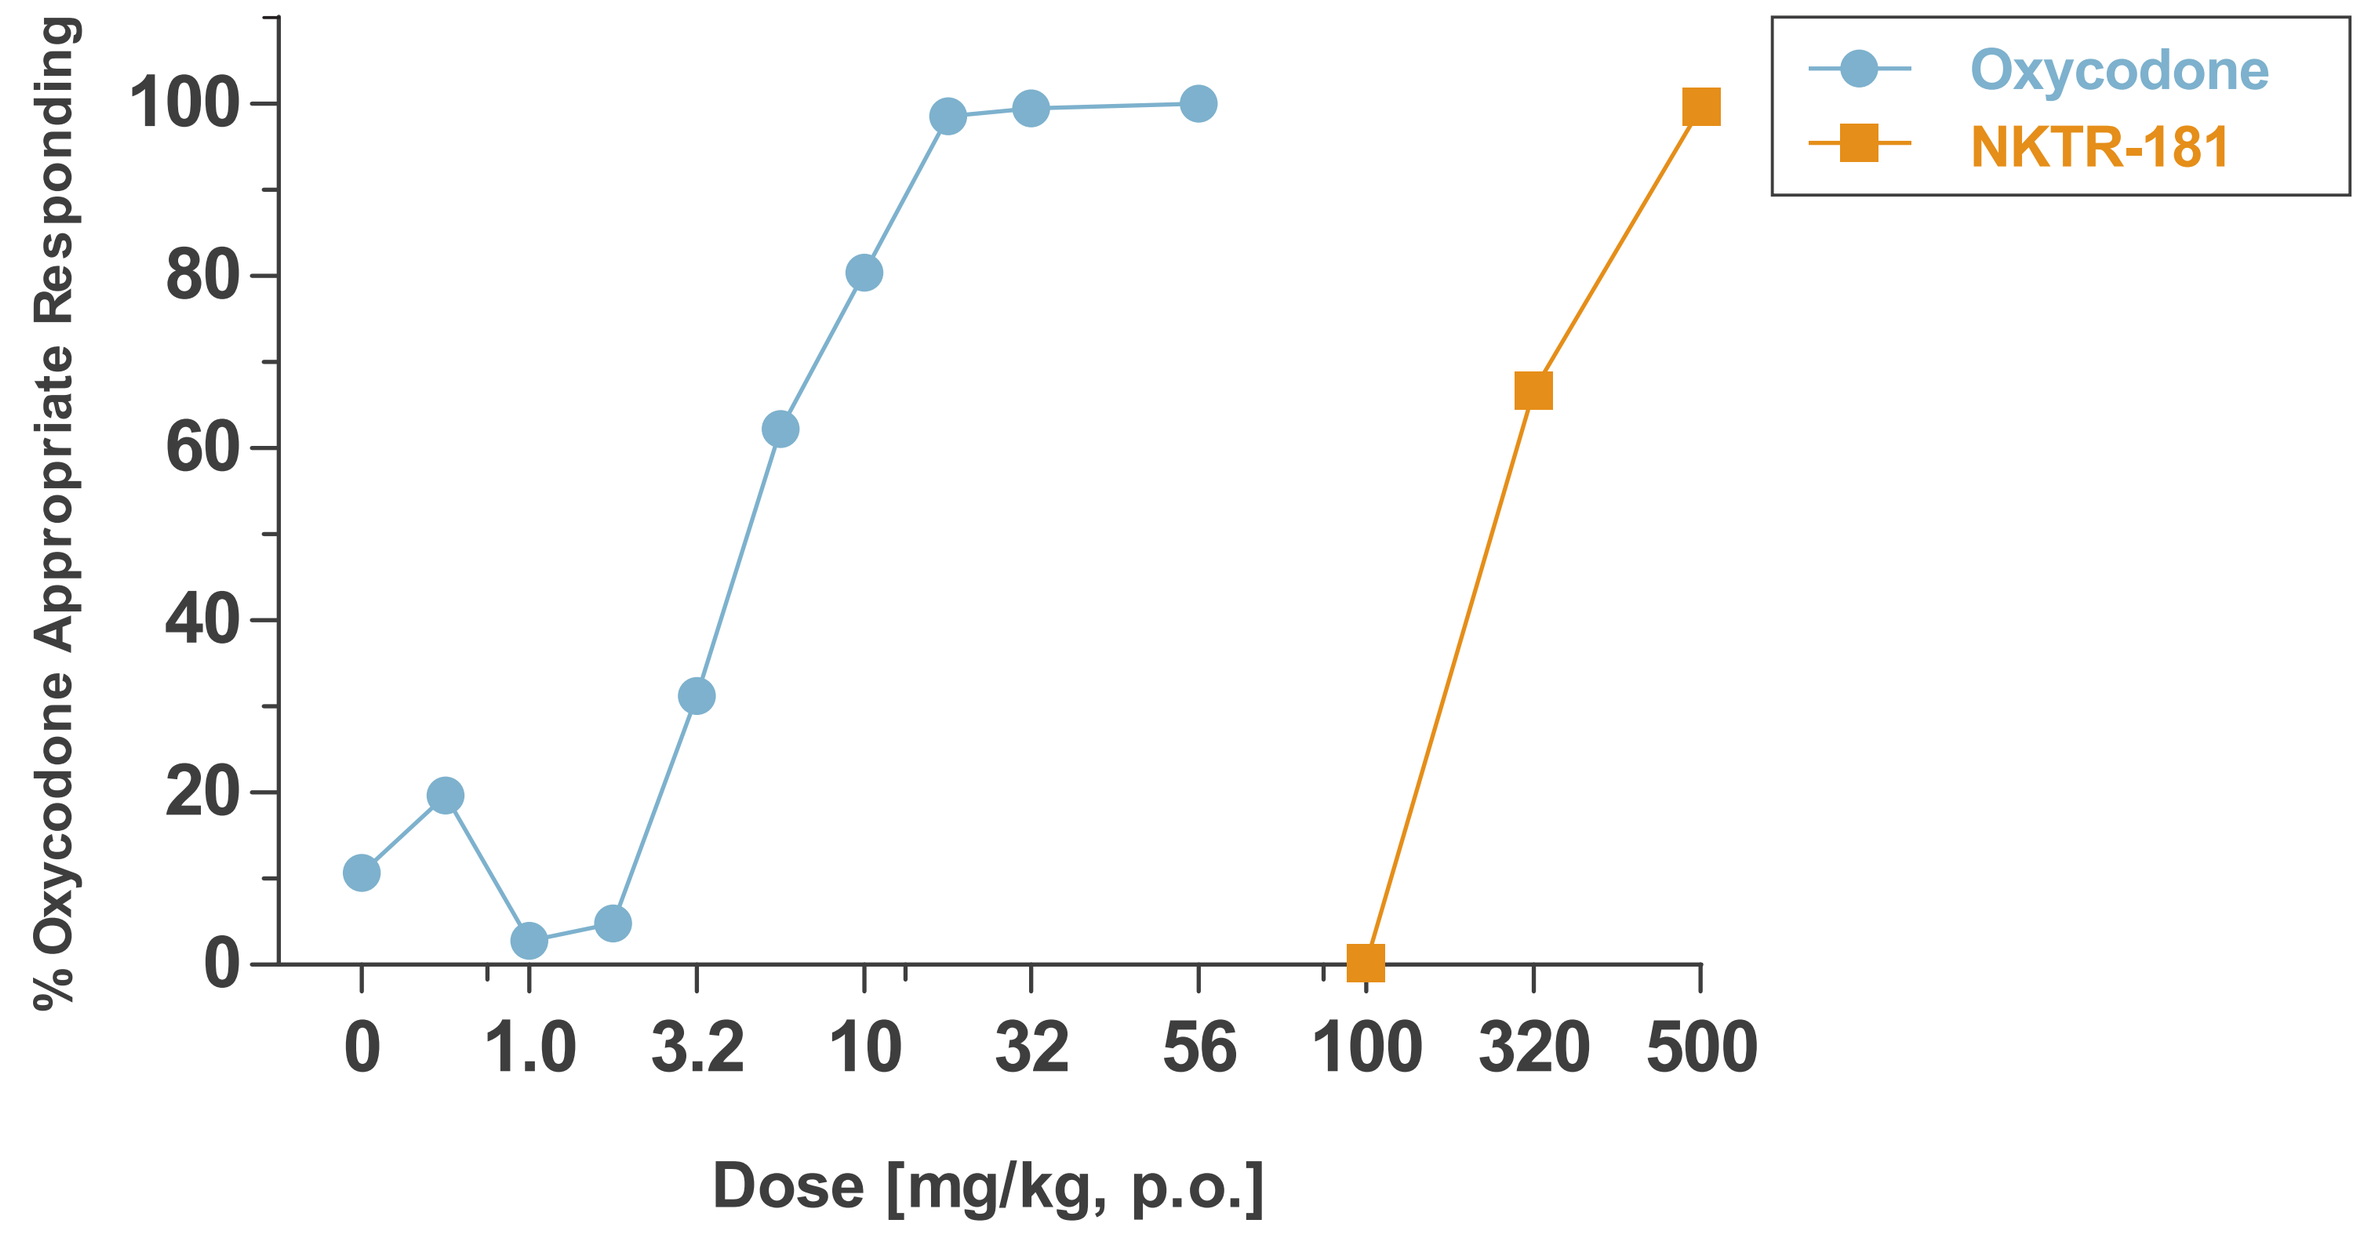
Oxycodone
NKTR-181
% Oxycodone Appropriate Responding
100
80
60
40
20
0
0 1.0 3.2 10 32 56 100 320 500
Dose [mg/kg, p.o.]

Drug discrimination studies were performed in rats trained to discriminate between oxycodone and vehicle. Animals then received oxycodone or NKTR-181 [N = 6] to determine if either produced a response similar to oxycodone or saline at the indicated unit doses. The "minimum discriminable dose" [MDD; the dose that produced >80% oxycodone appropriate responding] was determined for both oxycodone and NKTR-181 following oral dosing. The MDD for orally administered NKTR-181 compared to oral oxycodone showed a 30-fold shift in the dose response function [MDD oxycodone = 17.8 versus MDD NKTR-181 = 500 mg/kg].

References

1. Melnikova, I, Pain Market, Nature Reviews Drug Discovery, Volume 9, 589-90 (August 2010).
2. Pharmacol Biochem Behav. 2007; 86:45-54.
3. J Pharmacol Exp Ther. 2001; 299 :1056-65.
4. Fishburn, C.S. et al., NKTR-181: A Novel Opioid With Slowed Brain Entry Shows Low Abuse Liability and Reduced CNS Side Effects. Frontiers in Research Symposium, 2010.
5. Poster # HHH13, Society for Neuroscience 40th Annual Meeting: Neuroscience 2010, November 2010.
6. Anesthesiology 2010, A509 (Abstract #A509, American Society of Anesthesiologists (ASA) Annual Meeting: Anesthesiology 2010, 16-20 October 2010, San Diego, CA).
7. Harrison, S. et al., NKTR-181: A Novel Opioid Analgesic That Exhibits Reduced Abuse Potential and Maintains Full Analgesic Activity Following Repeat Dosing in Preclinical Rodent Models. American Academy of Pain Management 22nd Annual Clinical Meeting, September 2011.

NKTR-181 Exhibits a Wider Therapeutic Window Compared to Oxycodone

NKTR-181 shows a more favorable separation of analgesia from side effects compared with oxycodone

	Efficacy (acetic acid writhing)	CNS Side Effects (rotarod in rates)	Abuse Liability (drug discrimination in rats)
	ED_{50} mg/kg	ED_{50} mg/kg (ratio compared to efficacy)	Minimum discernable dose in mg/kg (ratio compared to efficacy)
NKTR-181	8.9	413 (46)	500(56)
Oxycodone	2.7	33 (12)	17.8 (~7)

Conclusions

- NKTR-181, a new chemical entity, is a mu-opioid agonist that exhibits a slower rate of brain uptake following oral administration compared with oxycodone.
- NKTR-181 exhibits full efficacy in the acetic acid writhing model in mice.
- NKTR-181 has reduced CNS mediated sedation.
- NKTR-181 shows low abuse liability in multiple animal models.
- NKTR-181 displays a wide therapeutic window through more favorable separation of analgesia from side effects compared with oxycodone.
- Subsequent Phase 1 clinical data validate preclinical observations and NKTR-181 is currently being prepared for Phase 2 development in chronic pain patients in 2012.

Abstract #224 / Poster #258: Presented at the American Academy of Pain Medicine 28th Annual Meeting: Pain Medicine, February 23-26, 2012, Palm Springs, CA