Novel Regulatory T-cell enhancing Biologic Rezpegaldesleukin: Phase 2b Efficacy, Safety, and Baseline Severity–Dependent Treatment Response in Moderate-to-Severe Atopic Dermatitis

Jonathan I. Silverberg[1], Thomas Bieber[2], **Raj Chovatiya[3,4]**, Richard B. Warren[5], Melinda Gooderham[6], Spyridon Gkalpakiotis[7], Sohail Chaudhry[8], Yi Liu[8], Mary Tagliaferri [8], Jonathan Zalevsky[8], David Rosmarin[9]

[1]Department Of Dermatology, George Washington University School Of Medicine, Washington, DC, United States; [2]Department of Dermatology and Allergy, Ludwig-Maximilians University Hospital, Munich, Germany; [3]Rosalind Franklin University of Medicine and Science Chicago Medical School, North Chicago, IL, United States; [4]Center for Medical Dermatology + Immunology Research, Chicago, IL, United States; [5]Dermatology Centre, Northern Care Alliance NHS Foundation Trust & Division of Musculoskeletal and Dermatological Sciences, Manchester NIHR Biomedical Research Centre, Manchester Academic Health Science Centre, University of Manchester, United Kingdom; [6]Department of Medicine, Queen's University, Kingston, ON, Canada; [7]Department of Dermatovenereology, Third Faculty of Medicine, Charles University and University Hospital Kralovske Vinohrady, Prague, Czech Republic; [8]Nektar Therapeutics, San Francisco, CA, United States; [9]Indiana University School of Medicine, Indianapolis, IN, United States

Rezpegaldesleukin is a novel, first-in-class biologic targeting regulatory T cells (Tregs)

- Administered as an active drug, with extended half-life and preferential binding to IL-2R on Tregs compared to recombinant IL-2[1]
- *In vivo* selectively expands and enhances the function of Tregs[1,2]
- Demonstrated clinical activity in atopic dermatitis (AD), alopecia areata, psoriasis, systemic lupus erythematous[2-5]

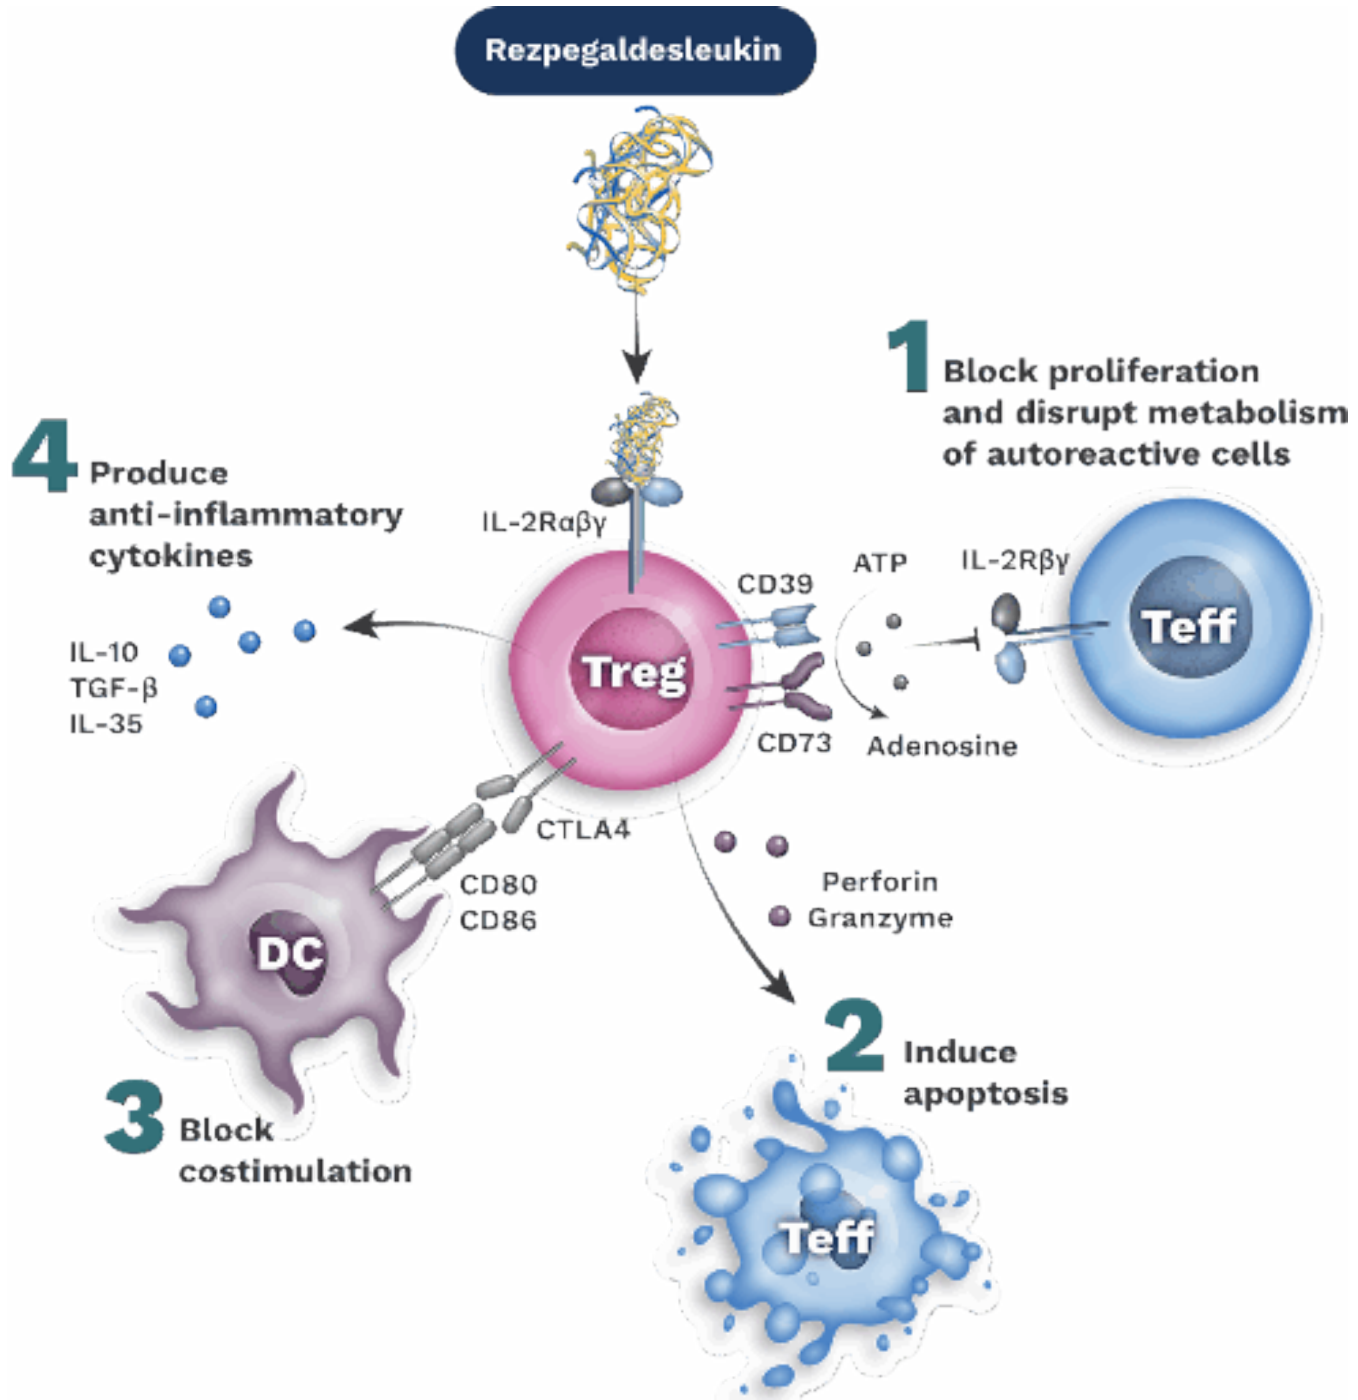
Rezpegaldesleukin
1 Block proliferation and disrupt metabolism of autoreactive cells
4 Produce anti-inflammatory cytokines
IL-2Rαβγ
CD39
ATP
IL-2Rβγ
Teff
Treg
IL-10
TGF-β
IL-35
CD73
Adenosine
CTLA4
CD80
CD86
DC
Perforin
Granzyme
2 Induce apoptosis
3 Block costimulation
Teff

Rezolve-AD: Study Schema Overview

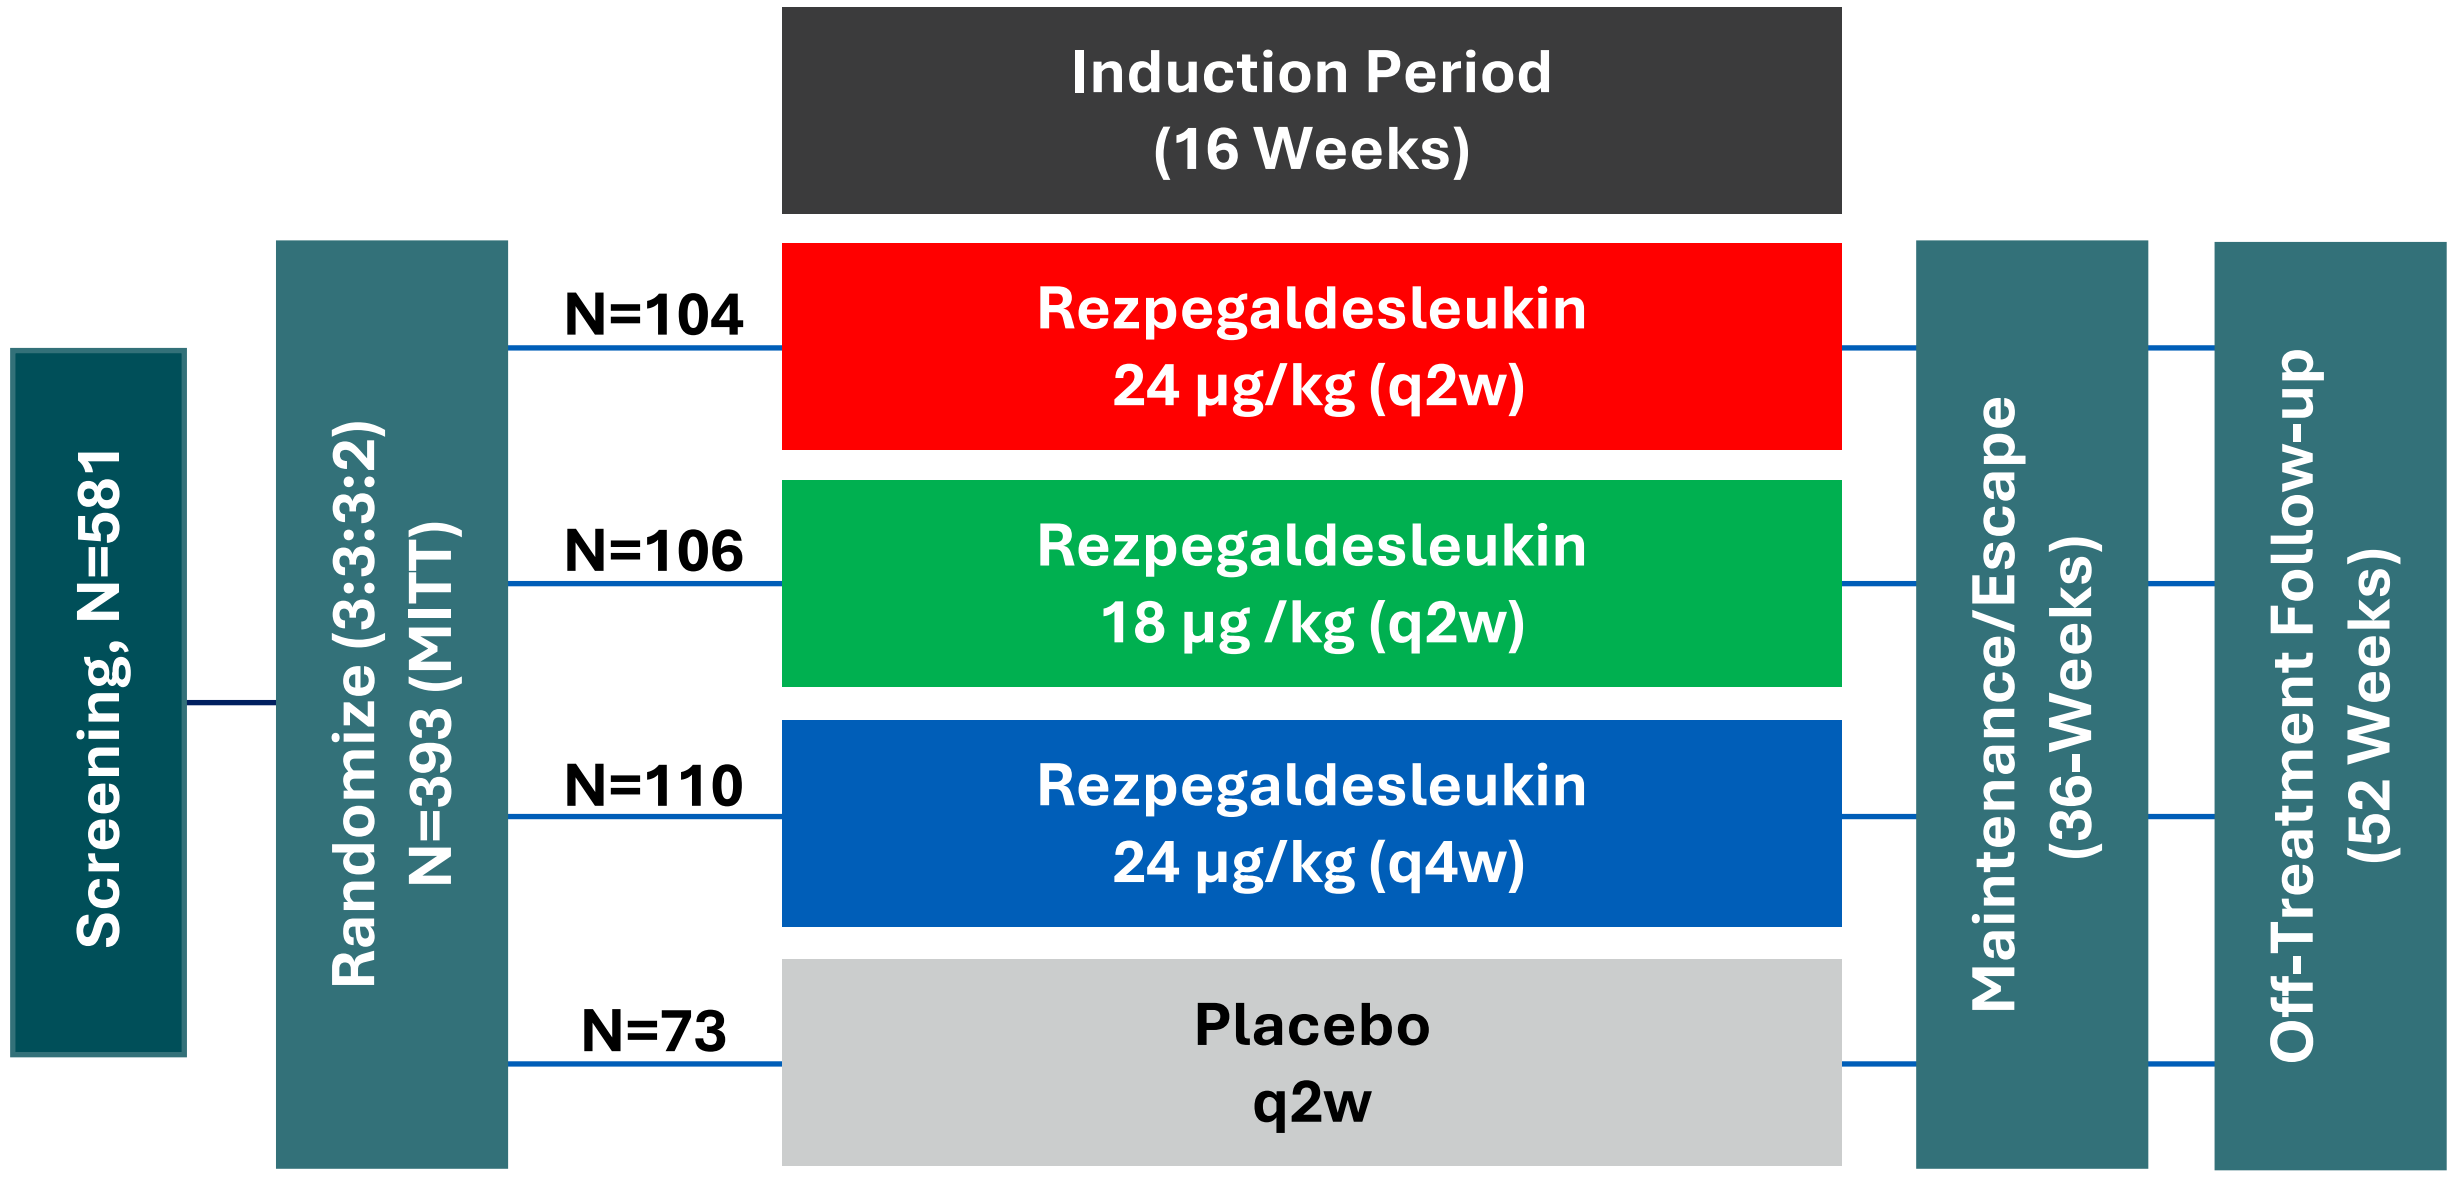
Induction Period (16 Weeks)
Screening, N=581
Randomize (3:3:3:2) N=393 (MITT)
N=104
Rezpegaldesleukin 24 μg/kg (q2w)
N=106
Rezpegaldesleukin 18 μg /kg (q2w)
N=110
Rezpegaldesleukin 24 μg/kg (q4w)
N=73
Placebo q2w
Maintenance/Escape (36-Weeks)
Off-Treatment Follow-up (52 Weeks)

1. *Dixit, N., et al. J Trans Autoimmunity* (2021) 4, 100103; 2. Silverberg, J. I. et al. *Nature Comm* (2024) 15(1), 9230; 3. Silverberg, J. I. et al. European Academy of Dermatology and Venereology Annual Meeting 2025, Sep. 17-20, 2025, Paris, LBA-108; 4. Rosmarin, D. et al. American Academy of Dermatology Annual Meeting 2026, March 27-31, 2026, Oral Late Breaker Abstract 79863; 5. Fanton, C. et al. *J. Transl. Autoimmun.*, 2022, 5, 100152.

1

Consistent Improvement in EASI % Change Across Disease Severity

Percent EASI Reduction in LS Mean from Baseline at Week 16 (Primary Estimand)

Overall

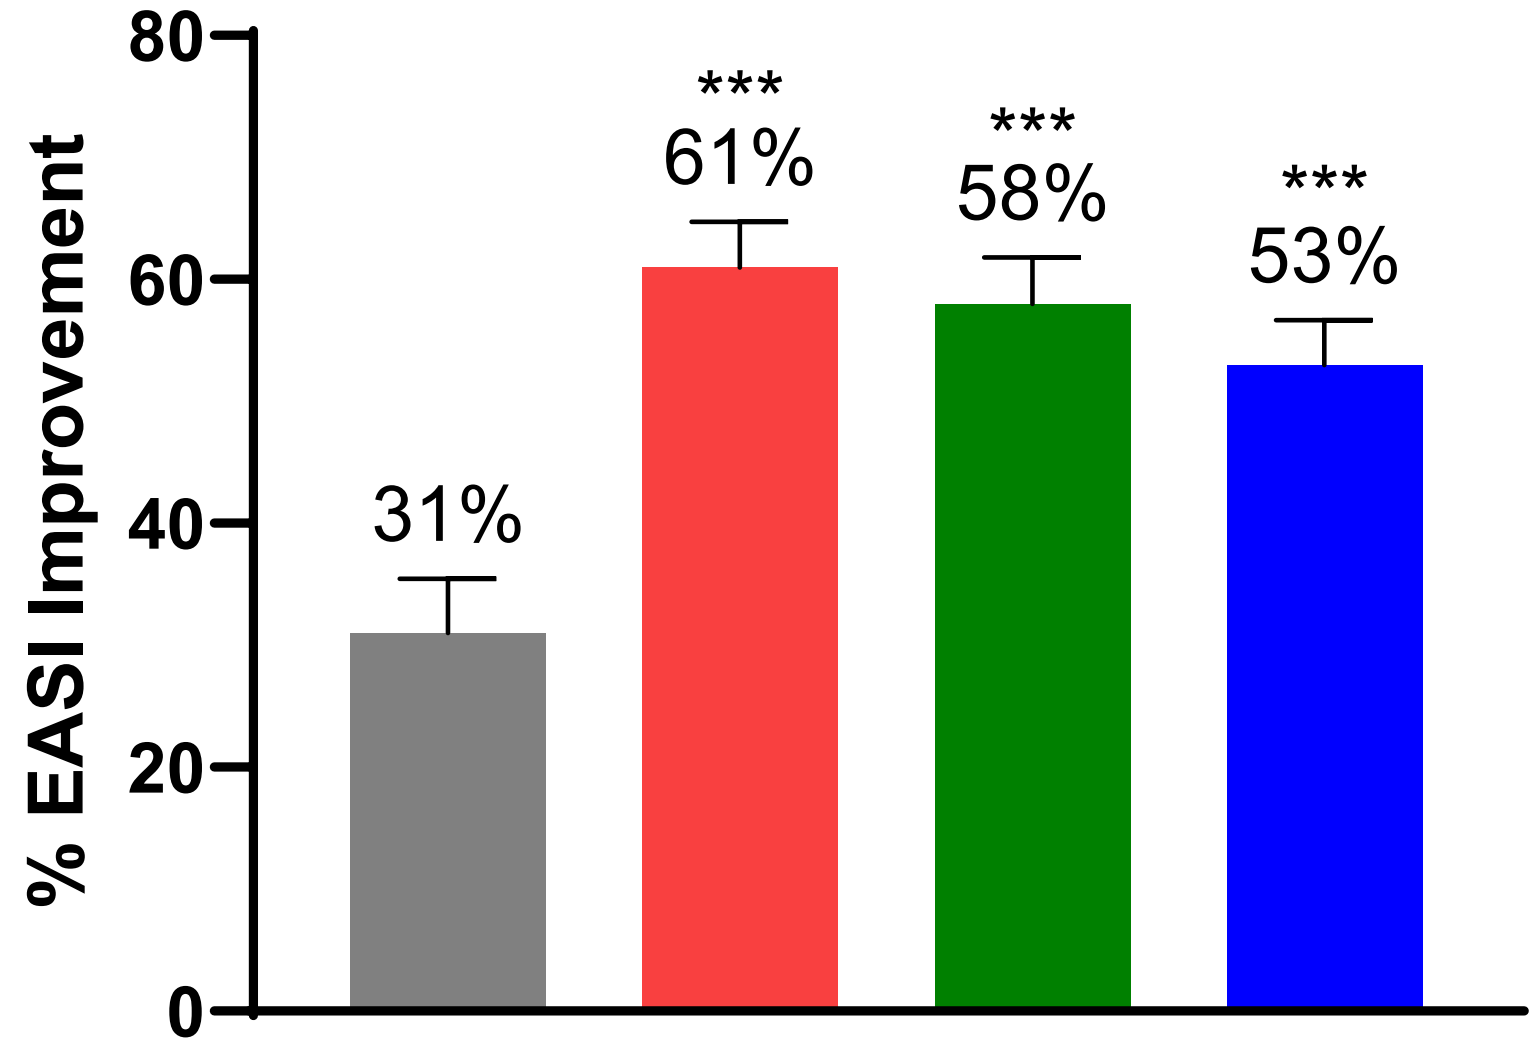

% EASI Improvement
80
60
40
20
0
31%

61%

58%

53%

Moderate (vIGA=3)

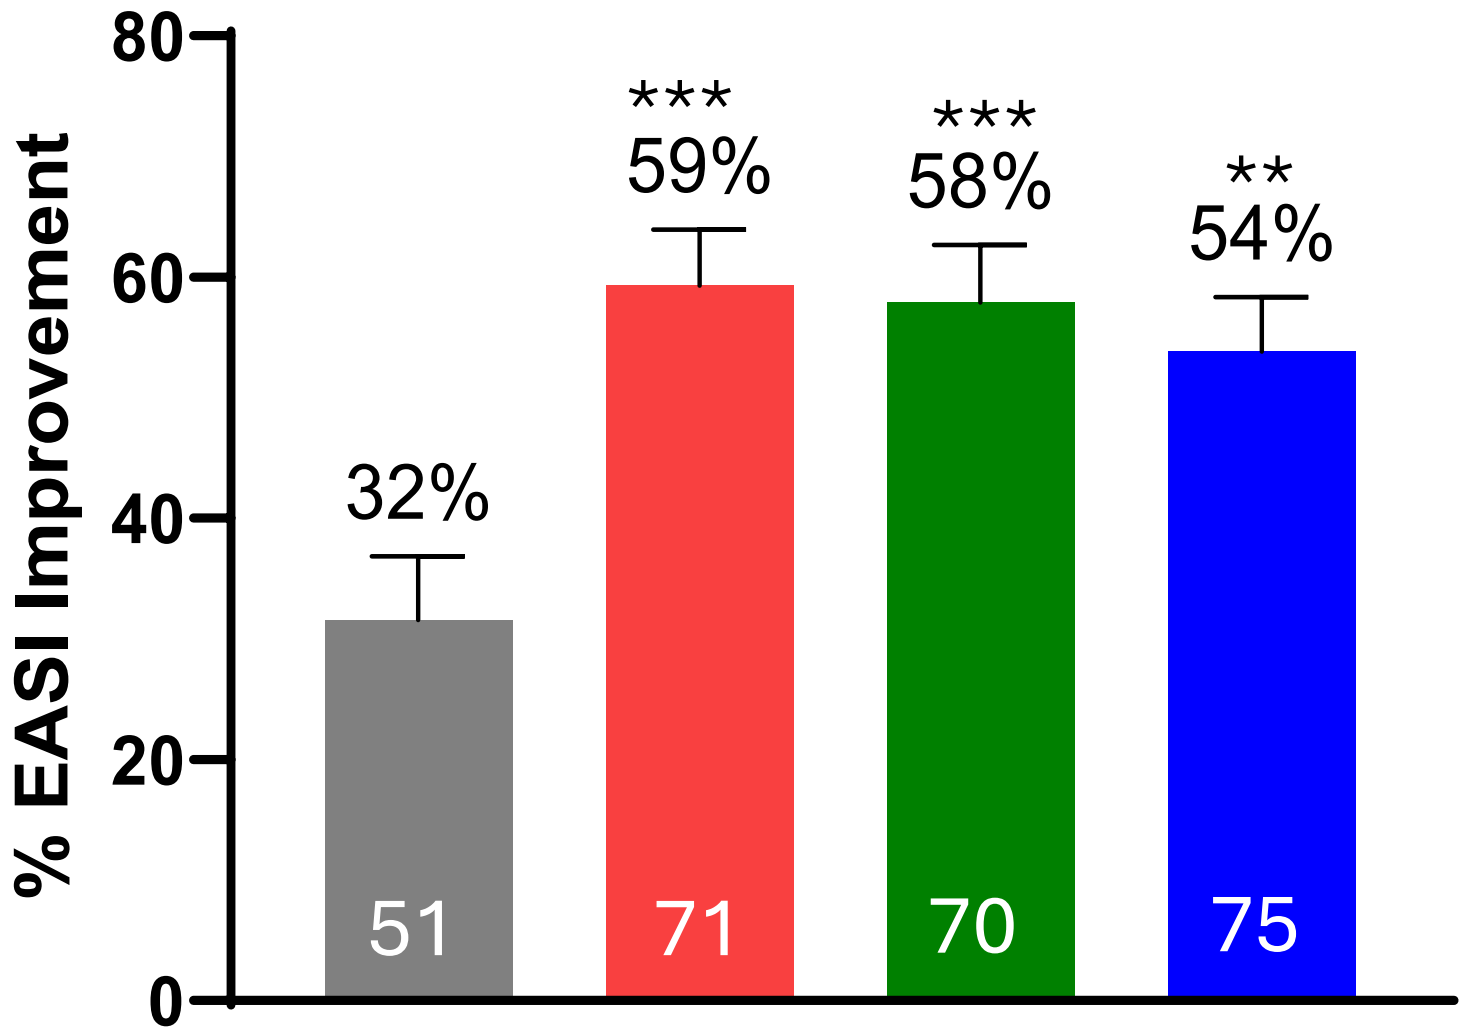

% EASI Improvement
80
60
40
20
0
32%

59%

58%
**
54%
51
71
70
75

Severe (vIGA=4)

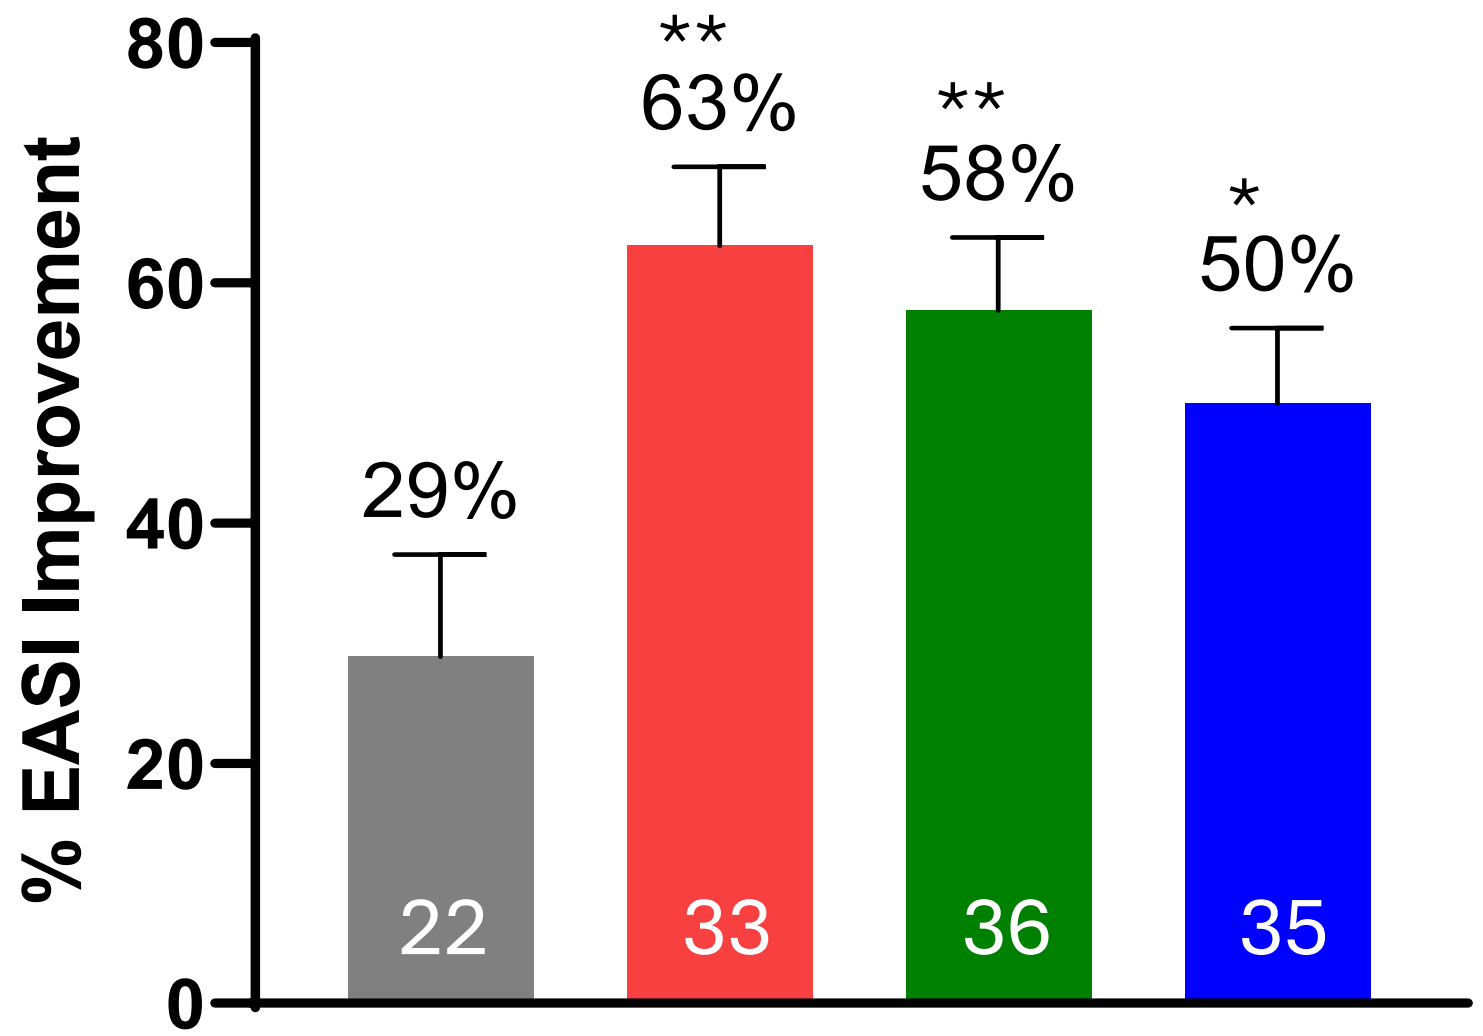

% EASI Improvement
80
60
40
20
0
29%
**
63%
**
58%
*
50%
22
33
36
35

- Placebo
- REZPEG 24 µg/kg, q2w
- REZPEG 18 µg/kg, q2w
- REZPEG 24 µg/kg, q4w

*p-value<0.05, **p-value<0.01, ***p-value<0.001 based on logistic regression

2

Consistent Improvement in EASI-75 and EASI-90 Across Disease Severity

Overall | Moderate | Severe

EASI-75

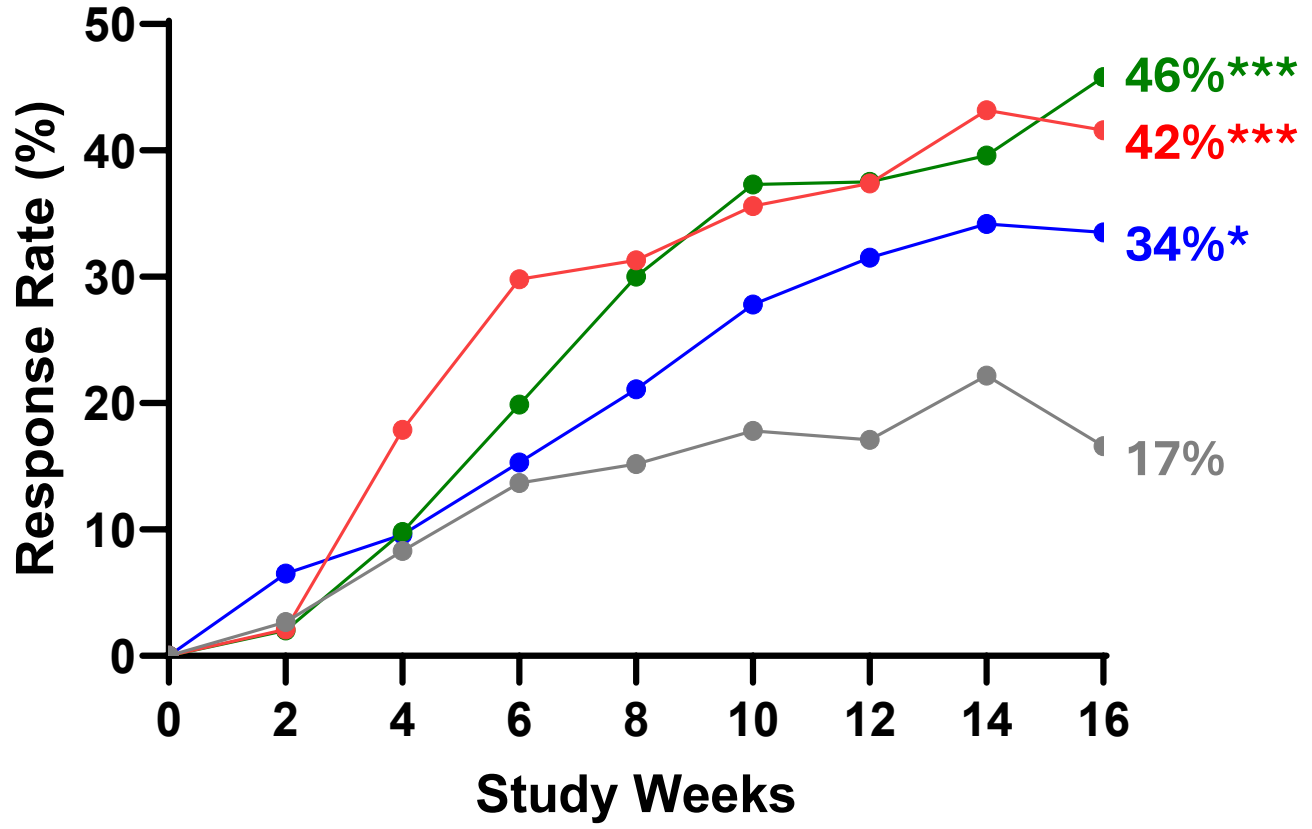
Response Rate (%)
0
10
20
30
40
50
0 2 4 6 8 10 12 14 16
Study Weeks
46%***
42%***
34%*
17%

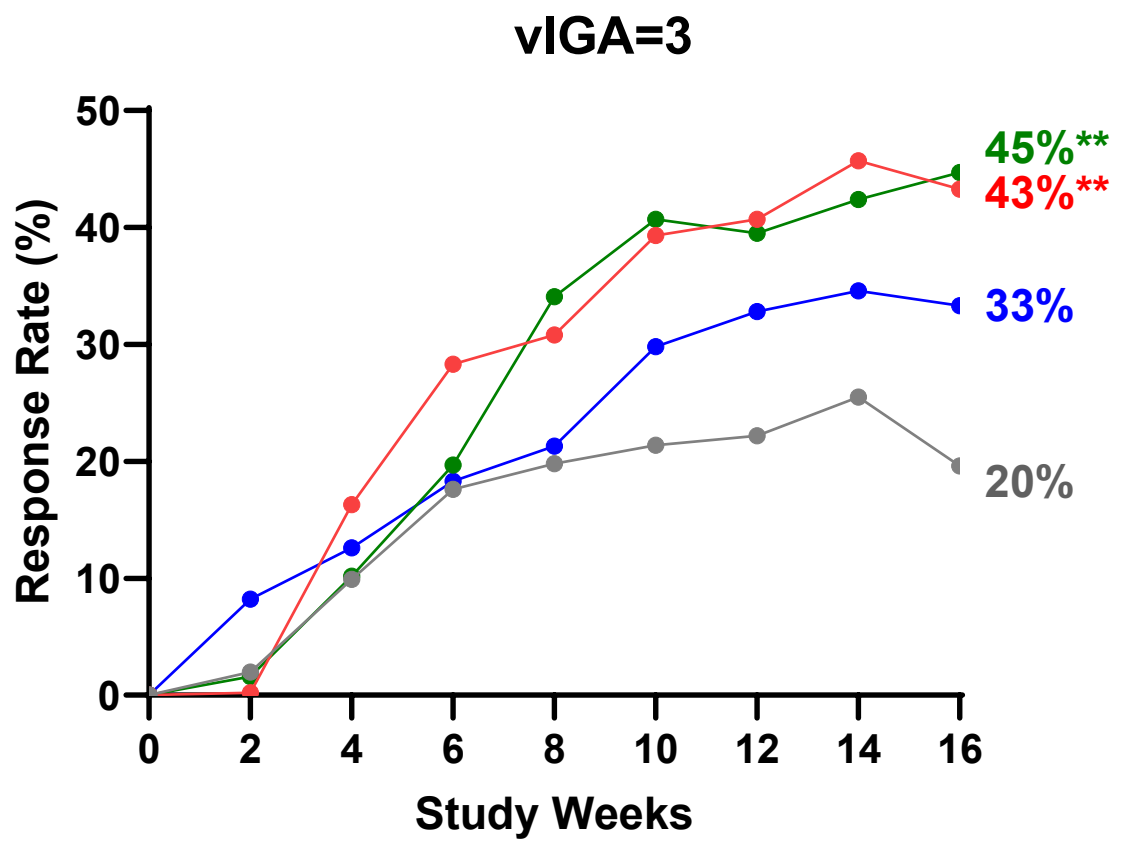
vIGA=3
Response Rate (%)
0
10
20
30
40
50
0 2 4 6 8 10 12 14 16
Study Weeks
45%**
43%**
33%
20%

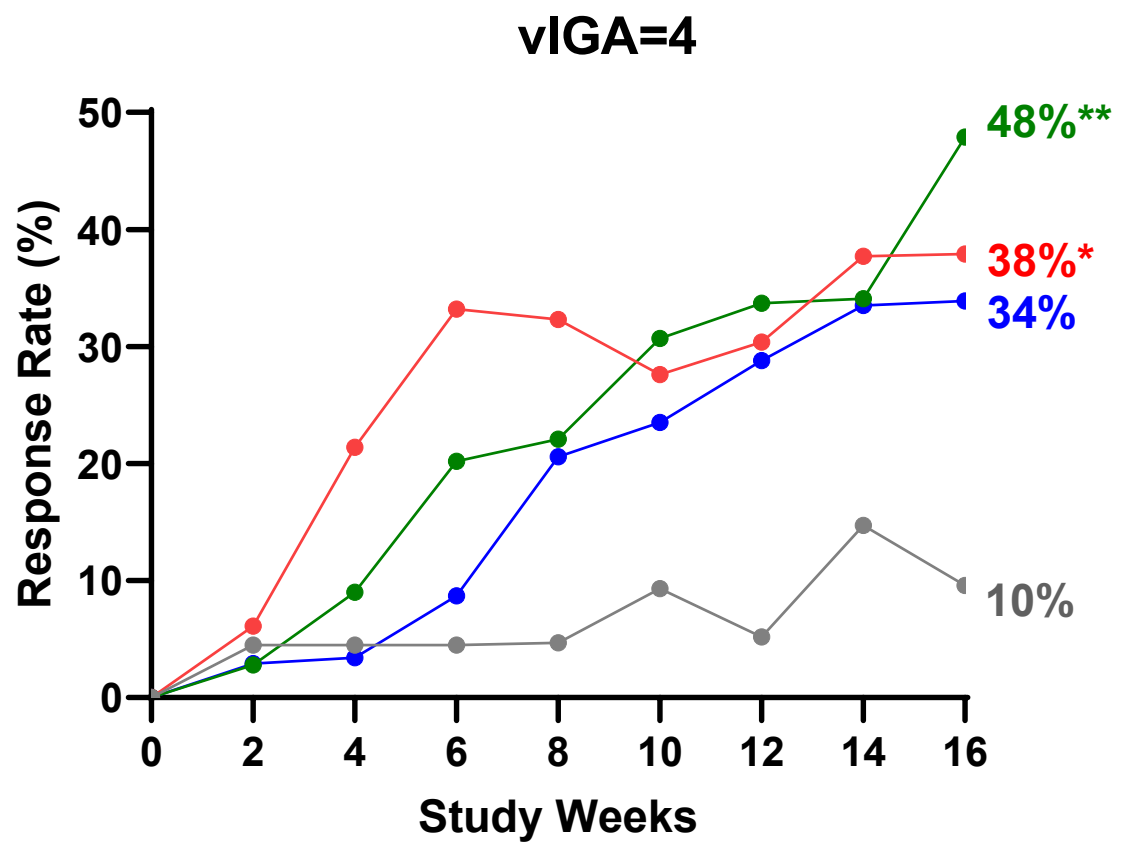
vIGA=4
Response Rate (%)
0
10
20
30
40
50
0 2 4 6 8 10 12 14 16
Study Weeks
48%**
38%*
34%
10%

EASI-90

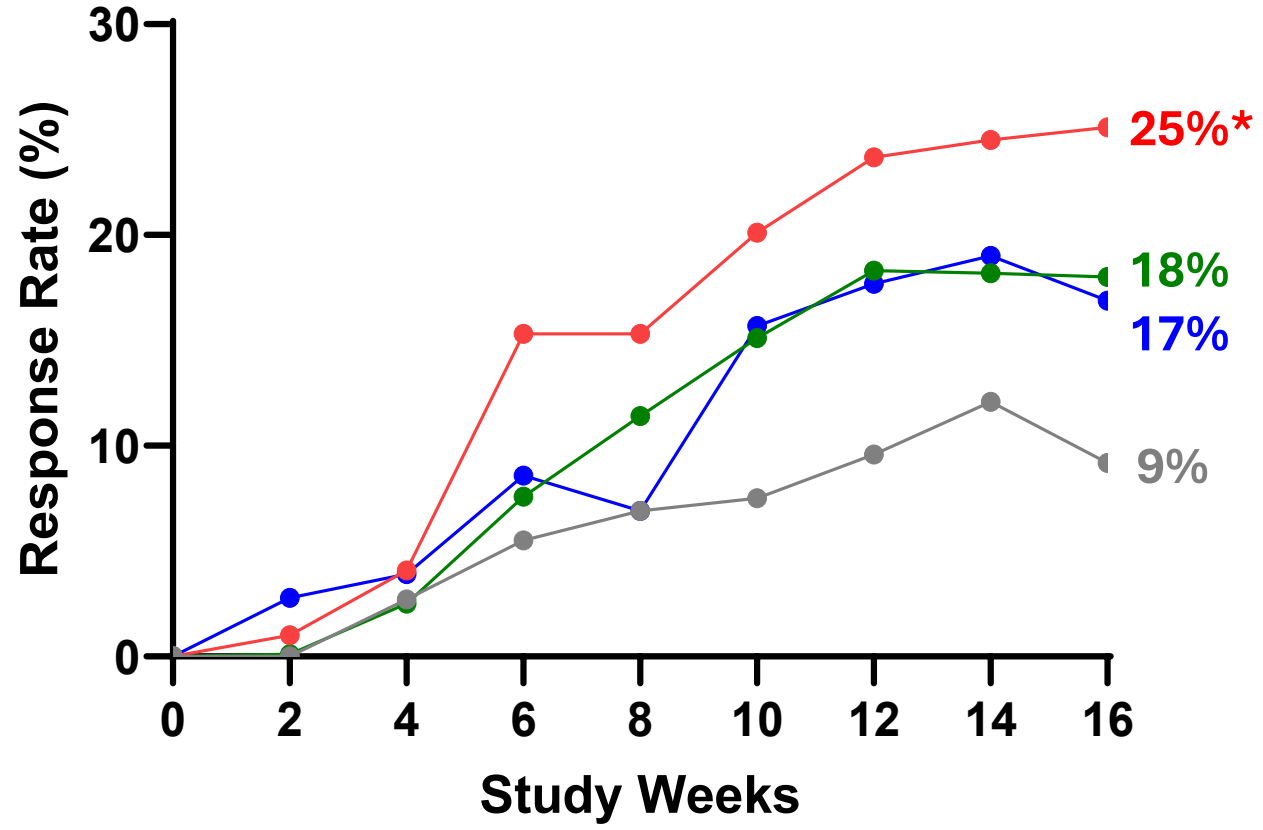
Response Rate (%)
0
10
20
30
0 2 4 6 8 10 12 14 16
Study Weeks
25%*
18%
17%
9%

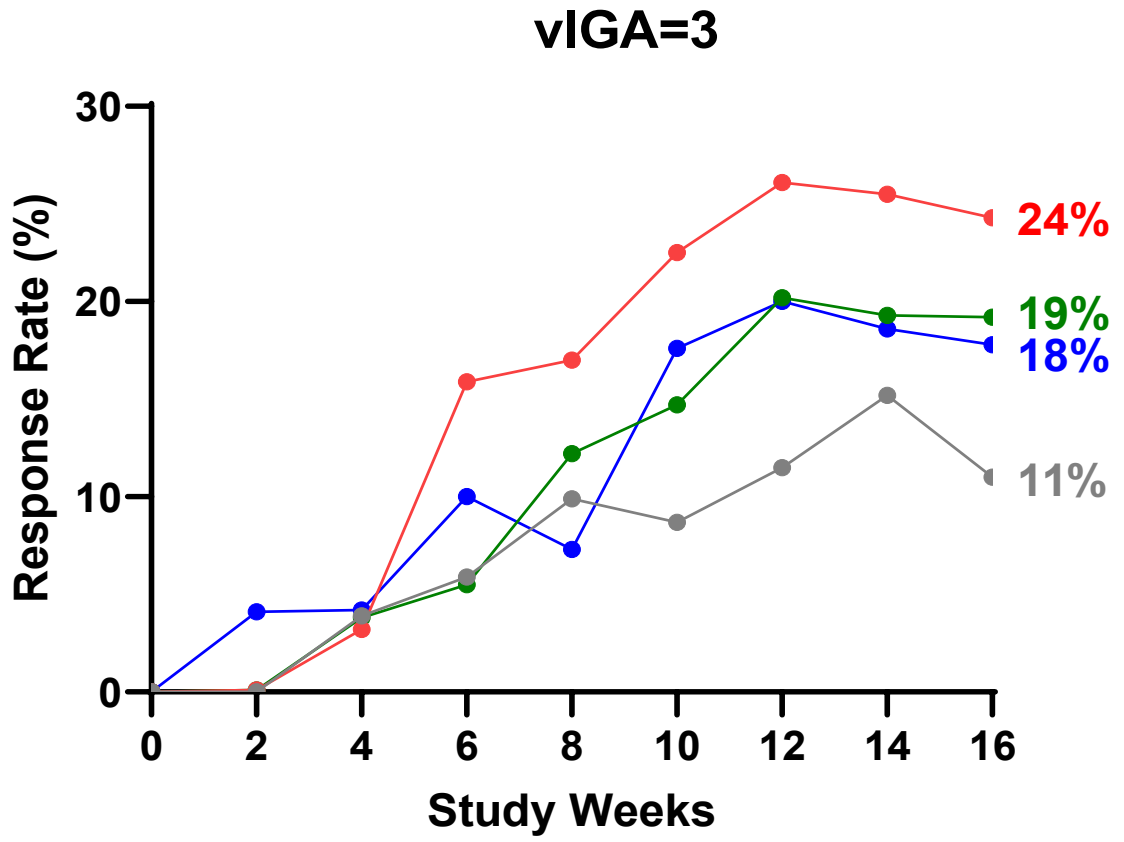
vIGA=3
Response Rate (%)
0
10
20
30
0 2 4 6 8 10 12 14 16
Study Weeks
24%
19%
18%
11%

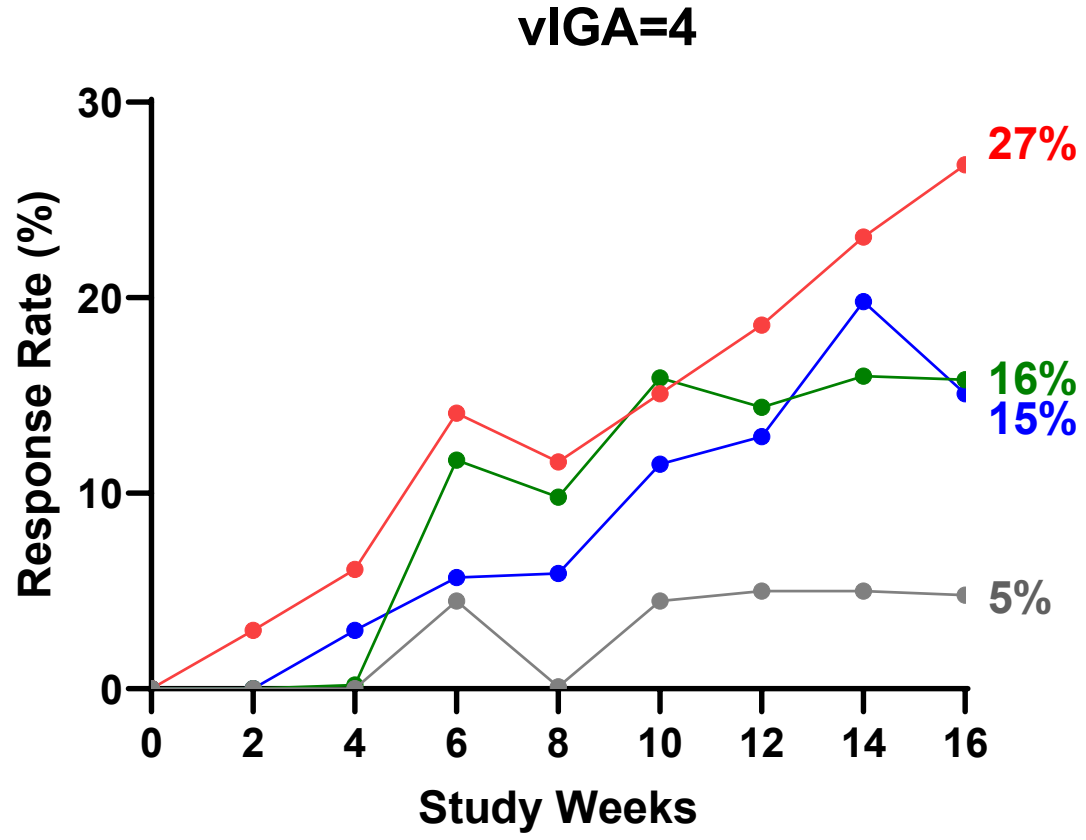
vIGA=4
Response Rate (%)
0
10
20
30
0 2 4 6 8 10 12 14 16
Study Weeks
27%
16%
15%
5%

- Placebo
- REZPEG 24 µg/kg, q2w
- REZPEG 18 µg/kg, q2w
- REZPEG 24 µg/kg, q4w

Overall:	IGA = 3	IGA = 4
N=73	N=51	N=22
N=104	N=71	N=33
N=108	N=70	N=36
N=110	N=75	N=35

*p-value<0.05, **p-value<0.01, ***p-value<0.001 based on logistic regression

3

EASI-75 Response for Rezpegaldesleukin 24 µg/kg q2w vs. Placebo

Consistent Treatment Effect Observed at Week 16 Across Baseline Severity of Disease, Geographical Region and Comorbidity of Asthma

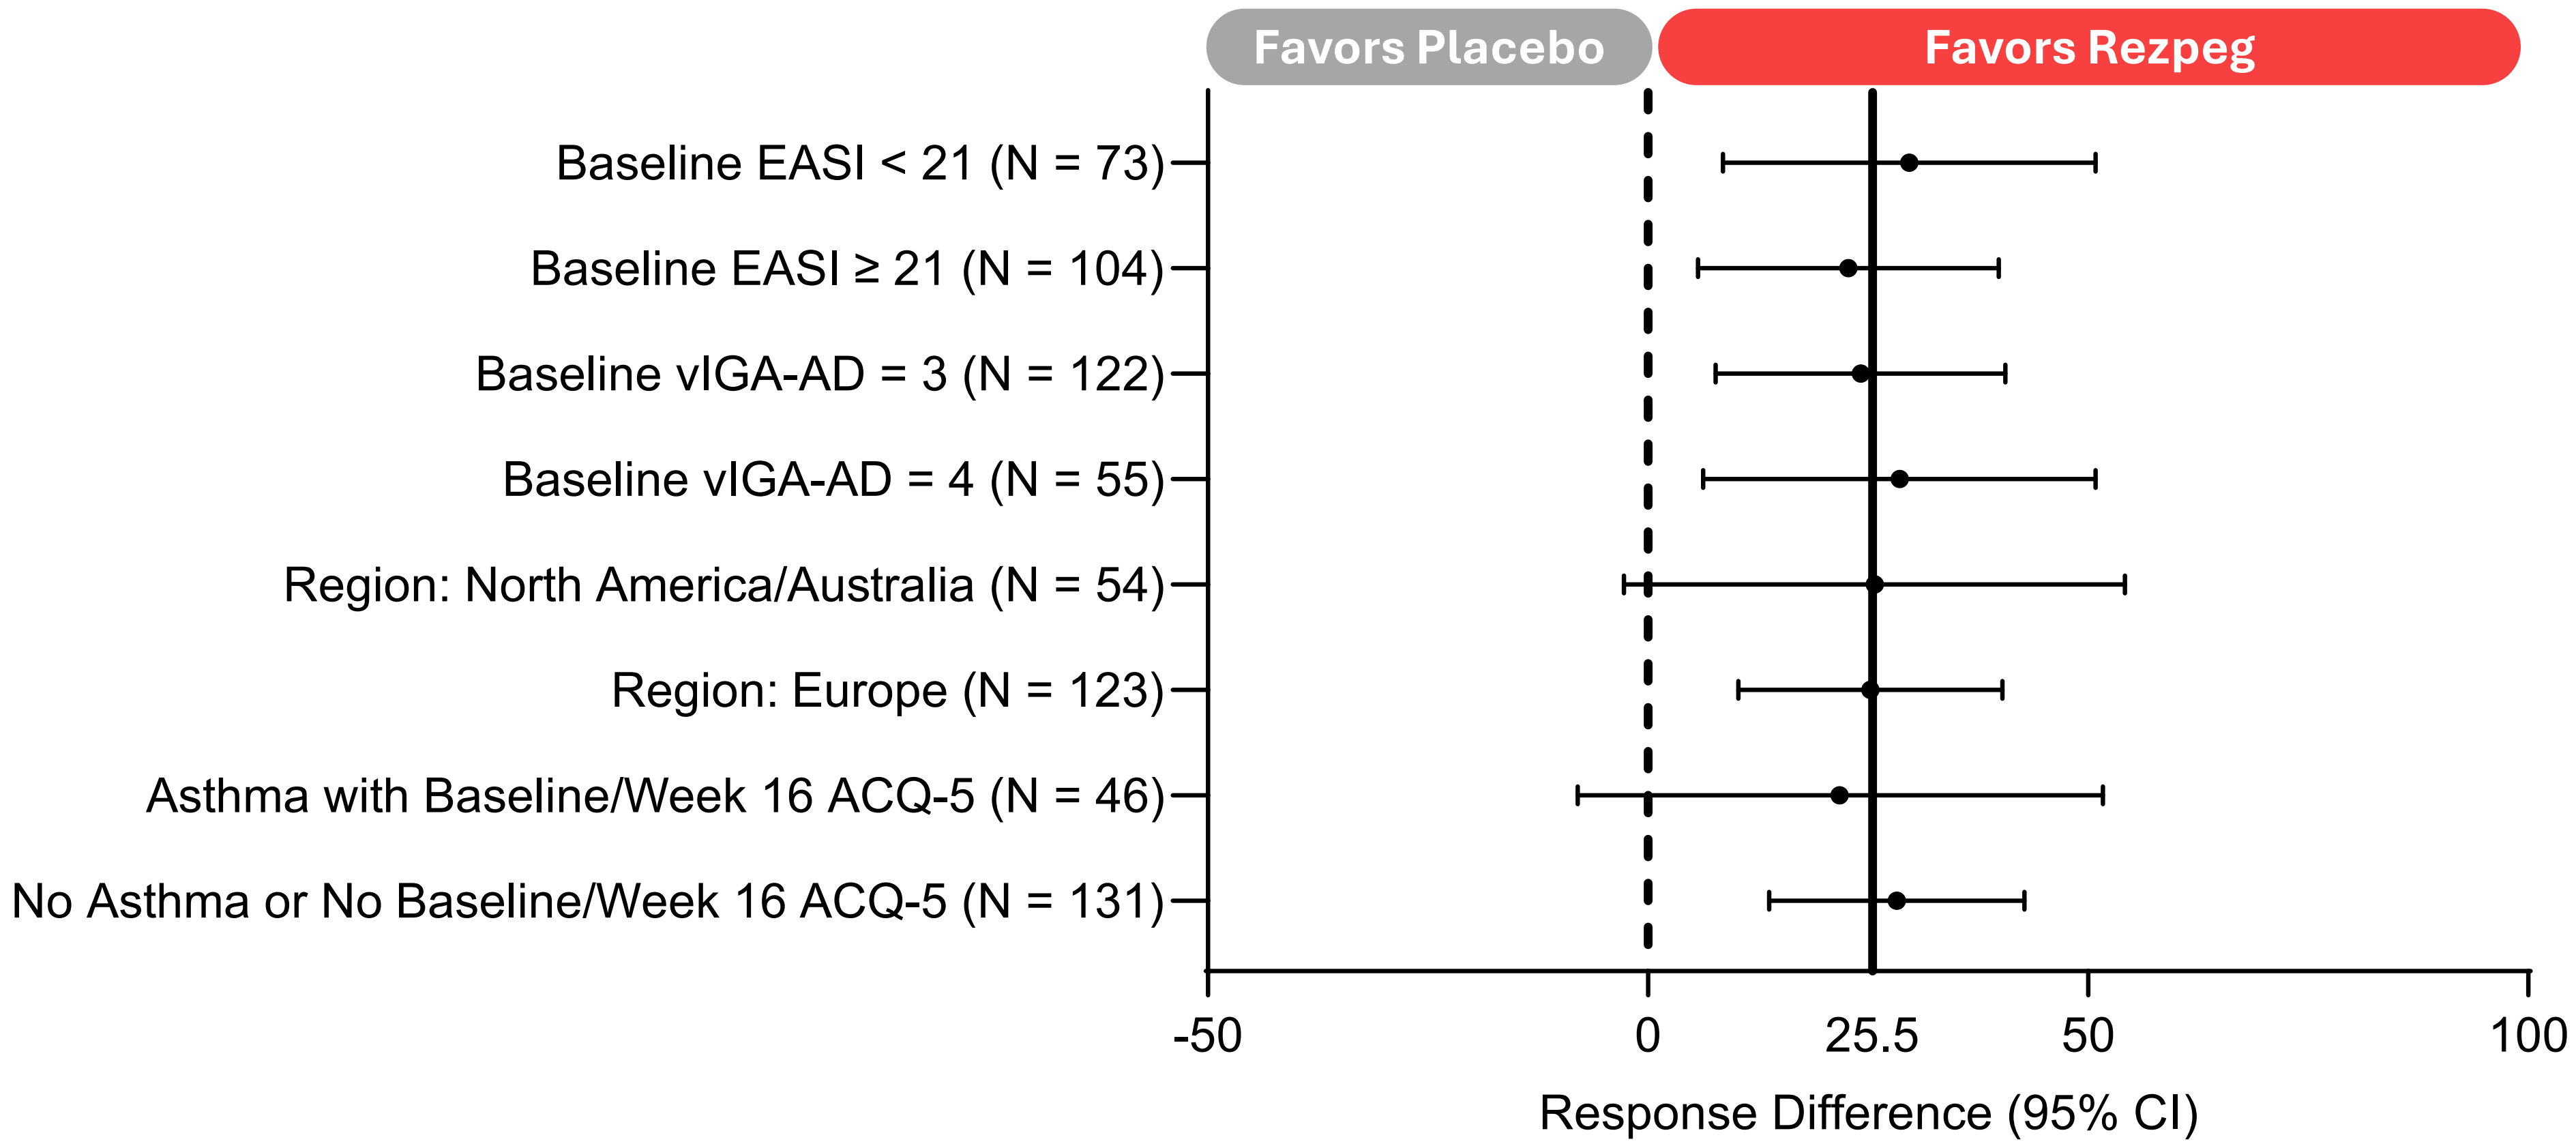

Favors Placebo
Favors Rezpeg
Baseline EASI < 21 (N = 73)
Baseline EASI ≥ 21 (N = 104)
Baseline vIGA-AD = 3 (N = 122)
Baseline vIGA-AD = 4 (N = 55)
Region: North America/Australia (N = 54)
Region: Europe (N = 123)
Asthma with Baseline/Week 16 ACQ-5 (N = 46)
No Asthma or No Baseline/Week 16 ACQ-5 (N = 131)
-50
0
25.5
50
100
Response Difference (95% CI)

The Primary Estimand analysis for binary endpoints use logistic regressions. Data after use of rescue therapy outside protocol specifications or discontinued treatment due to lack of efficacy were imputed as non-responders; data after patients who discontinued due to other reasons were set to missing and all missing data are imputed using the multiple imputation method.

4

Key Safety and Efficacy Summary

RESOLVE-AD: 16-Week Induction Period

Safety:

- Local injection site reactions were observed in 69.7% of all rezpegaldesleukin treated patients vs. 4.1% for placebo, nearly all being mild-to-moderate in severity (99.6%).
- Other TEAEs more commonly observed (≥5%) in the rezpegaldesleukin treated patients (n=320) versus placebo (n=73) included eosinophilia (7.8% vs. 2.7%), pyrexia (6.3% vs 2.7%), headache (6.3% vs. 4.1%), upper respiratory tract infections (5.9% vs. 5.5%) and arthralgia (5.0% vs 1.4%).
- No increased risk of conjunctivitis, oral ulcers, malignancy, or infections, including oral herpes, were identified.

Efficacy:

- Significant improvement over placebo was observed with rezpegaldesleukin 24 μg/kg q2w in mean percent change in EASI ($p<0.001$), EASI-75 ($p<0.001$), EASI-90 ($p<0.05$), vIGA-AD 0/1 ($p<0.05$), NRS-Itch ($p<0.01$)
- Other rezpegaldesleukin dose arms exhibited dose-dependent improvement in AD disease assessments throughout the induction period
- Consistent efficacy was observed irrespective of baseline AD severity for those with moderate or severe by EASI or vIGA-AD

The ZENITH-AD Phase 3 program of rezpegaldesleukin in moderate-to-severe atopic dermatitis is in final planning and will start in Q2 2026.

Disclosures:
RC has served as a speaker, advisory board member, consultant, and/or investigator for AbbVie, Acelyrin, Alumis, Amgen, AnaptysBio, Apogee Therapeutics, Arcutis Biotherapeutics Inc., Argenx, Astria Therapeutics Inc., Avalere Health, Beiersdorf, Boehringer Ingelheim, Bristol Myers Squibb, Cara Therapeutics, Castle Biosciences, Celldex CLn Skin Care, Dermavant, Eli Lilly and Company, EMD Serono, Formation Bio, Forte Biosciences, Galderma, Genentech, GSK, Incyte, Inmagene, Johnson & Johnson, Kenvue, LEO Pharma, L'Oréal, Nektar Therapeutics, Nia Health, Novartis, Opsidio, Organon, Oruka, Pfizer Inc., RAPT, Regeneron, Sanofi, Sitryx, Takeda, TRex Bio, UCB, Zai Lab. JS has served as a speaker, advisory board member, consultant and/or received institutional grants from AbbVie Inc, Alamar Biosciences, Inc., Aldena Therapeutics Inc., Amgen Inc., Abiomed, Inc., Arcutis Biotherapeutics, Inc., Arena Pharmaceuticals, Inc., Asana BioSciences LLC, Aslan Pharmaceuticals Pte Ltd., BioMX Inc., Biosion, Inc., Bodewell, Boehringer Ingelheim International GmbH, Bristol-Myers Squibb Company, Cara Therapeutics, Castle Biosciences, Inc., Celgene Corporation, Connect Biopharma Holdings Limited, Corevitas, LLC, Dermavant Sciences, Inc., Dermtech, LLC, Eli Lilly and Company, Galderma S.A., GlaxoSmithKline, Incyte Corporation, Kiniksa Pharmaceuticals, Ltd, Leo Pharma Inc., Nektar Therapeutics, Novartis AG, Optum, Inc., Pfizer Inc., RAPT Therapeutics, Inc., Recludix Pharma, Inc., Regeneron Pharmaceuticals, Inc., Sanofi, Shaperon Inc., Target PharmaSolutions, Inc., UNION therapeutics A/S, UpToDate, Inc.TB was speaker and/or consultant and/or Investigator for AbbVie, Affibody, Almirall, Amagma/Triveni, AnaptysBio, AOBiom, Anergis, Apogee, Arena, Aristea, Artax, Asana Biosciences, ASLAN pharma, Astria, Attovia, BambusTx, Bayer Health, Belenos, BioVerSys, Böhringer-Ingelheim, Bristol-Myers Squibb, BYOME Labs, Cantargia, CellDex, Connect Pharma, Daichi-Sanyko, Dermavant, DICE Therapeutics, DirigentBio, Domain Therapeutics, DS Pharma, EQRx, EMD Serono, Galderma, Galapagos, Gilead, Glenmark, GSK, Incyte, Innovaderm, Janssen, Kirin, Kymab, LEO, LG Chem, Lilly, Mabylon, MSD, Medac, Micreos, Nektar, Nextech, Novartis, Numab, OM-Pharma, Ornavi, Overtone, Pfizer, Pierre Fabre, Protagonist Tx, Q32bio, RAPT, Samsung Bioepis, Sanofi/Regeneron, Scienta Lab, TIRmed, UCB, Union Therapeutics, UPStream Bio, YUHAN. He is founder and chairman of the board of the non-profit biotech "Davos Biosciences AG" within the international Kühne-Foundation and founder of the consulting firm "Bieber Dermatology Consulting". RW has served as a speaker, advisory board member, and/or consultant, for AbbVie, Almirall, Amgen, Arena, Astellas, Avillion, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, DiCE, Galderma, GSK, Janssen, Eli Lilly and Company, Leo, Meiji Pharma, Novartis, Pfizer, RAPT pharmaceuticals, Sanofi, Sun Pharma, TAKEDA, UCB & UNION. Additionally, he has received research grants from AbbVie, Almirall, Amgen, Celgene, Janssen, Lilly, Leo, Novartis, Pfizer & UCB. MG has served as a speaker, advisory board member, consultant, investigator, and/or received institutional grants from AbbVie, Acelyrin, Amgen, AnaptysBio, Arcutis Biotherapeutics, Aristea Therapeutics, ASLAN Pharmaceuticals, Bausch Health, Boehringer Ingelheim, Bristol-Myers Squibb, Cara Therapeutics, Celgene Corporation, Dermavant Sciences, Dermira, Eli Lilly and Company, Galderma Laboratories, LP, Inmagene Biopharmaceuticals, Janssen Pharmaceuticals, Inc, Leo Pharma Inc, Leo Pharma Inc., Medimmune, Meiji Seika Pharma Co., Ltd, Moonlake, Nektar, Nimbus Therapeutics, Novartis Pharmaceuticals Corp., Pfizer Inc., Regeneron, Reistone Biopharma, Sanofi Genzyme, Sanofi/Regeneron, Sun Pharmaceutical Industries Ltd., Takeda Pharmaceuticals USA Inc, Tarsus, UCB, Ventyx Biosciences. SG has served as advisor and/or speaker for Almirall, Abbvie, LEO Pharma, UCB, Janssen Cilaq, Pfizer, Nektar, Novartis, Eli-Lilly, Sanofi and an investigator for Almirall, Alumis, Abbvie, LEO Pharma, GSK, UCB, Janssen Cilaq, Pfizer, Nektar, Novartis, Eli-Lilly, Sanofi and Takeda. SC, YL, MT, JZ are Employees and stockholders at Nektar Therapeutics DR has served as a speaker, advisory board member, consultant and/or investigator for AbbVie Inc, Abcuro, Inc., AltruBio Inc., Amgen Inc., Arena Pharmaceuticals, Inc., Boehringer Ingelheim International GmbH, Bristol-Myers Squibb Company, Celgene Corporation, Concert Pharmaceuticals, Inc., CSL Behring LLC, Dermavant Sciences, Inc., Dermira, Inc., Eli Lilly and Company, Galderma S.A., Incyte Corporation, Janssen Pharmaceuticals, Kyowa Kirin Co., Ltd., , Merck & Co., Inc., Novartis AG, Pfizer Inc., Recludix Pharma, Inc., Regeneron Pharmaceuticals, Inc., Revolo Biotherapeutics Limited, Sanofi, Sun Pharmaceuticals Industry Limited, UCB S.A., Viela Bio, Inc.

Scan QR code to download copy of presentation:

Study sponsored by Nektar Therapeutics