Bempegaldesleukin* (NKTR-214) + nivolumab in first-line advanced/metastatic urothelial carcinoma: Updated results from PIVOT-02

Arlene Siefker-Radtke[1], Mayer Fishman[2], Arjun V. Balar[3], Giovanni Grignani[4], Adi Diab[1], Jianjun Gao[1], Mary Tagliaferri[5], Alison Hannah[5], Erin Karski[5], Jonathan Zalevsky[5], Ute Hoch[5], Ahsan Rizwan[5], EJ Liao[5], Mehmet A. Bilen[6]

[1]University of Texas MD Anderson Cancer Center, Houston, TX; [2]H. Lee Moffit Cancer Center & Research Institute, Tampa, FL; [3]Perlmutter Cancer Center at NYU Langone Health, New York, NY; [4]Candiolo Cancer Institue - FPO, IRCCS, Candiolo, Italy; [5]Nektar Therapeutics, San Francisco, CA; [6]Winship Cancer Institute of Emory University, Atlanta, GA

*rINN

NEKTAR

BACKGROUND

- Two anti-PD1/PD-L1 agents have approval in the 1L cisplatin-ineligible setting. Regulatory authorities recently revised the labels to restrict the usage to patients whose tumors have PD-L1 expression, CPS ≥ 10 or PD-L1 IC ≥ 5%[1,2,6]
- Approximately 70% of cis-ineligible patients have tumors with low PD-L1 expression, which leaves a high unmet need for new therapy options for these patients[1,2]
- Bempegaldesleukin (NKTR-214) is a CD122-preferential IL-2 pathway agonist that has been shown to increase tumor-infiltrating lymphocytes, T cell clonality and increase PD-1 expression (**Figure 1**)[7-9]
- Bempegaldesleukin combined with the checkpoint inhibitor nivolumab has been shown to convert tumors from PD-L1 non-expressers to expressers (PD-L1 negative <1% to PD-L1 ≥1%)[8]
- PIVOT-02 is a multicenter, Phase 1/2 study evaluating bempegaldesleukin plus nivolumab and includes a cohort of patients with locally advanced or metastatic UC who are cisplatin-ineligible or cisplatin eligible who have refused standard of care (SOC)

Figure 1. NKTR-214 Delivers a Controlled, Sustained, and Biased Signal Through the IL-2 Receptor Pathway

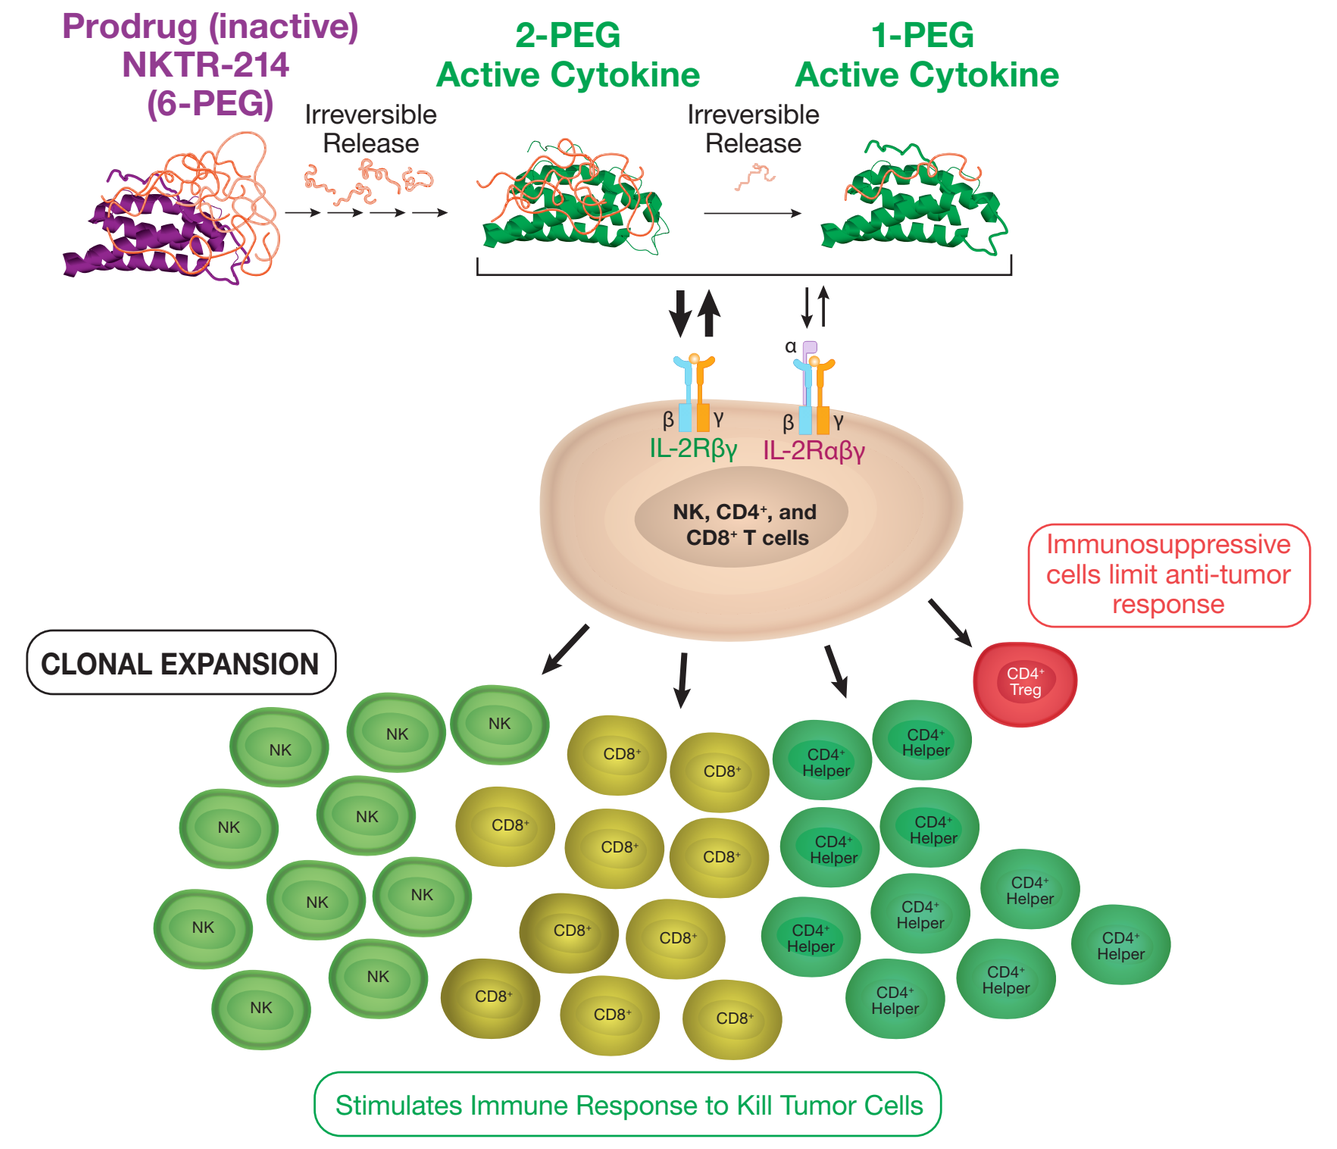
Prodrug (inactive) NKTR-214 (6-PEG)
Irreversible Release
2-PEG Active Cytokine
Irreversible Release
1-PEG Active Cytokine
IL-2Rβγ
IL-2Rαβγ
NK, CD4+, and CD8+ T cells
Immunosuppressive cells limit anti-tumor response
CLONAL EXPANSION
Stimulates Immune Response to Kill Tumor Cells

PIVOT-02 STUDY DESIGN AND ENDPOINTS

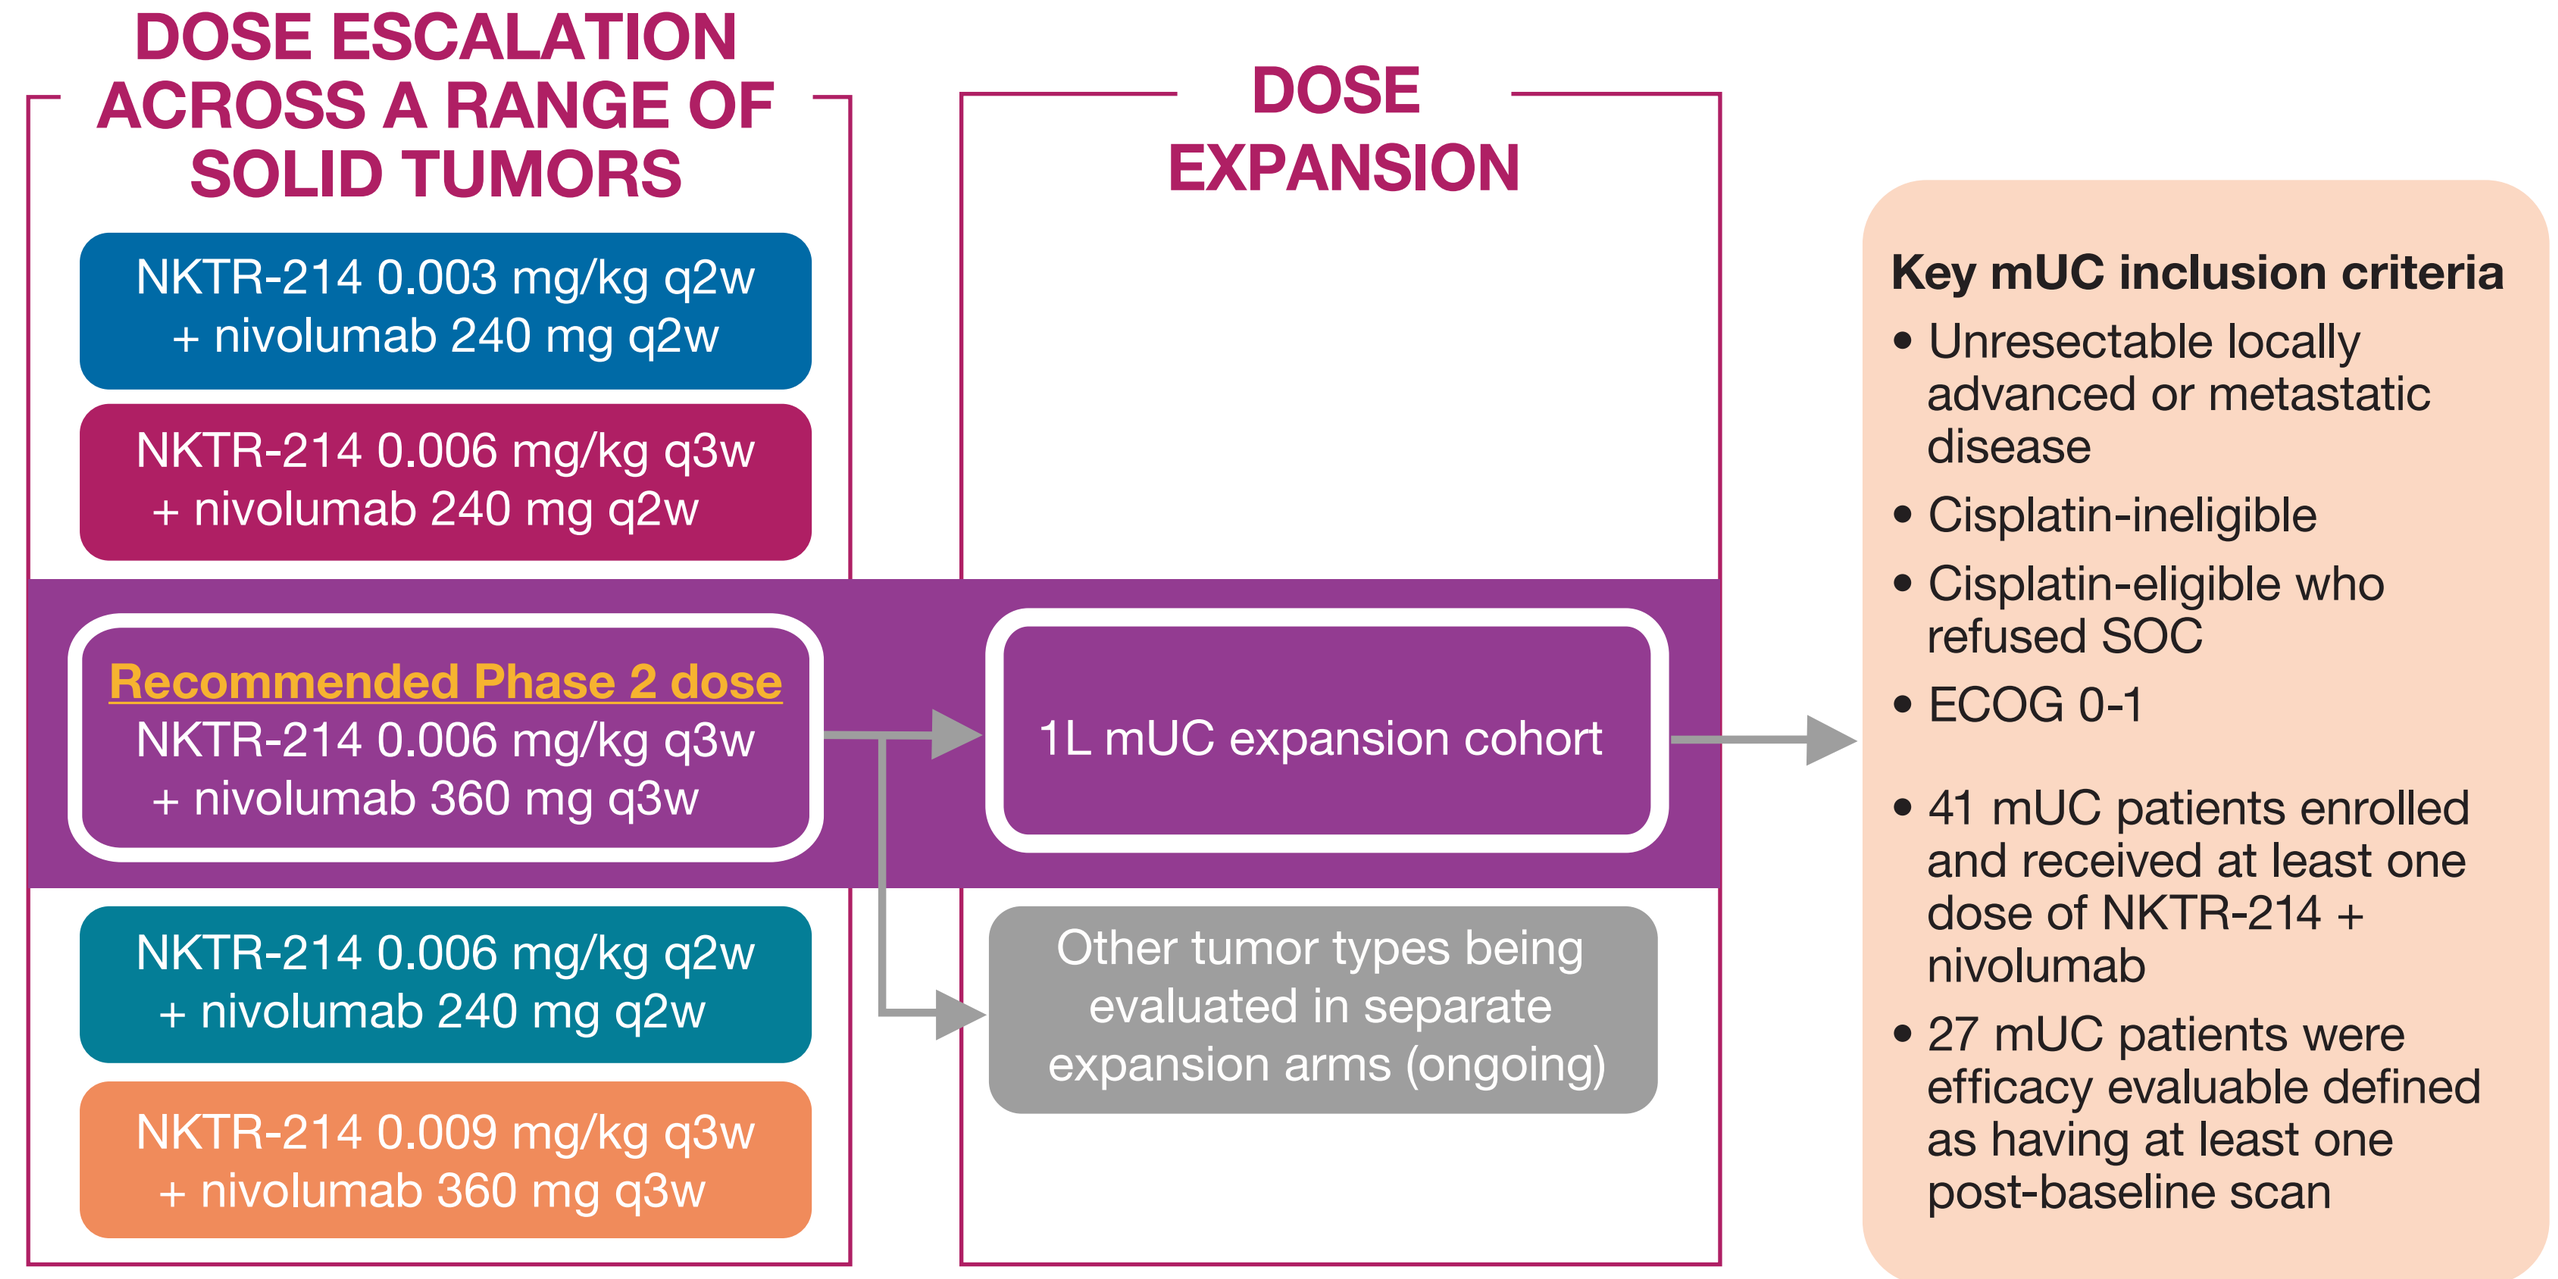
DOSE ESCALATION ACROSS A RANGE OF SOLID TUMORS
NKTR-214 0.003 mg/kg q2w + nivolumab 240 mg q2w
NKTR-214 0.006 mg/kg q3w + nivolumab 240 mg q2w
Recommended Phase 2 dose
NKTR-214 0.006 mg/kg q3w + nivolumab 360 mg q3w
NKTR-214 0.006 mg/kg q2w + nivolumab 240 mg q2w
NKTR-214 0.009 mg/kg q3w + nivolumab 360 mg q3w
DOSE EXPANSION
1L mUC expansion cohort
Other tumor types being evaluated in separate expansion arms (ongoing)
Key mUC inclusion criteria
• Unresectable locally advanced or metastatic disease
• Cisplatin-ineligible
• Cisplatin-eligible who refused SOC
• ECOG 0-1
• 41 mUC patients enrolled and received at least one dose of NKTR-214 + nivolumab
• 27 mUC patients were efficacy evaluable defined as having at least one post-baseline scan

Primary endpoints:
- Safety and tolerability per CTCAE v4.03
- ORR per RECIST v1.1 assessed every 8 (±1) weeks
- Per protocol, efficacy evaluable is defined as patients with ≥1 post-baseline scan

Secondary and exploratory endpoints:
- Duration of response, OS, PFS, clinical benefit rate, PK
- ORR by immune related RECIST (irRECIST)

Biomarker endpoints (subset of patients in each cohort):**
- Absolute lymphocyte count and blood immuno-phenotyping
- Baseline and on-treatment biopsies (3 weeks) were collected in patients, when clinically feasible

Preliminary data presented have a cut-off of Dec 3, 2018

ECOG PS: Eastern Cooperative Oncology Group performance score; mUC: locally advanced or metastatic urothelial carcinoma; ORR: overall response rate; OS: overall survival; PFS: progression free survival; PK: pharmacokinetics; RECIST: response evaluation criteria in solid tumors; RP2D: recommended phase 2 dose; SOC: standard of care

**Patients submitted tissue during screening and underwent tumor biopsy during week 3 of treatment

RESULTS

Table 1. Baseline Patient Characteristics (n, %)

	All Patients (n=41)	Efficacy Evaluable (n=27)*
Age, Median (Range) in Years	70 (41, 91)	70 (41, 83)
Population		
Cisplatin Ineligible	27 (66%)	16 (59%)
Renal Impairment	19 (70%)	12 (75%)
Hearing Loss	4 (15%)	2 (13%)
Peripheral Neuropathy, Grade ≥2	4 (15%)	1 (6%)
Missing	1 (4%)	1 (6%)
Cisplatin Eligible (Refused Standard Of Care)	14 (34%)	11 (41%)
Sex		
Male	29 (71%)	20 (74%)
Female	12 (29%)	7 (26%)
PD-L1 Status[a]		
Positive [≥1% TC]	13 (32%)	12 (44%)
Negative [<1% TC]	13 (32%)	11 (41%)
Not Evaluable[b]	2 (5%)	-
Not Available[b]	13 (32%)	4 (15%)
Locally Advanced Disease	1 (2%)	1 (4%)
Metastatic Disease (Stage IV)	40 (98%)	26 (96%)
Lymph Node Only[c]	16 (39%)	11 (41%)
Visceral (Non-Nodal Metastases)[d]	24 (59%)	15 (56%)
ECOG Performance Score		
0	18 (44%)	13 (48%)
1	22 (54%)	14 (52%)
Not Available	1 (2%)	-
Prior Systemic Neoadjuvant Therapy	5 (12%)	4 (15%)
Prior Systemic Adjuvant Therapy	4 (10%)	4 (15%)
Previous Cystectomy	5 (12%)	2 (7%)

All patients (N=41) have received at least one dose of NKTR-214 and nivolumab
[a] PD-L1 status evaluated using the 28-8 PharmDx assay; negative defined as <1% of tumor cells with PD-L1 expression on IHC; positive defined as ≥1% of tumor cells with PD-L1 expression on IHC; TC: tumor cells
[b] Patients with PD-L1 assessment not evaluable; biopsy sample collected but tissue not evaluable; Patients with PD-L1 assessment not available: 4 patients no available biopsy samples for analysis due to physician waivers; 9 patients pending biopsy sample analysis
[c] Defined as disease metastasized to lymph node (LN) only (includes only pts with LN disease or LN + primary site of disease)
[d] Defined as disease metastasized outside of lymph nodes (excludes pts with LN disease or LN + primary site of disease)
* Efficacy evaluable defined per protocol as patients with at least one post-baseline scan. As of 12/3/2018, 1 patient was excluded for non-eligibility (no target lesion), and 3 patients discontinued prior to first scan [1 due to patient decision; 1 due to clinical progression; 1 due to death from disease]; 10 patients pending first scan in database

Table 2. Treatment-Related Adverse Events (TRAE)

Adverse Event	N=41 (n, %)
Patients Experiencing at Least One TRAE	36 (88%)
Most Common Grade 1 or 2 TRAEs Occurring in >15% of the Population[a]	
Flu-like Symptoms[b]	29 (71%)
Fatigue	23 (56%)
Rash[c]	19 (46%)
Pruritus	13 (32%)
Decreased Appetite	11 (27%)
Nausea	9 (22%)
Patients Experiencing at Least One Grade 3 TRAE	6 (15%)
Flu-like Symptoms[b,e]	2 (5%)
Hypotension[e]	1 (2%)
Drug Reaction With Eosinophilia and Systemic Symptoms[d]	1 (2%)
Encephalopathy[d,f]	1 (2%)
Hypereosinophilic Syndrome[f]	1 (2%)
Myasthenic Syndrome[d]	1 (2%)
Complete Atrioventricular Block[d,g]	1 (2%)
Myocarditis[g]	1 (2%)
Myositis[g]	1 (2%)
Patients Experiencing at Least One Grade 4 or 5 TRAE	0
Patients Who Discontinued Due to TRAE	4 (10%)

[a] All AE's Grade 1 or 2 except for two events of Grade 3 flu-like symptoms
[b] Includes the following preferred terms: chills, influenza like illness, pyrexia, influenza
[c] Includes the following preferred terms: Erythema, Rash, Rash erythematous, Rash generalized, Rash macular, Rash maculo-papular, Rash maculovesicular, Rash papular, Rash pruritic, Rash pustular, Rash vesicular, Exfoliative rash.
[d] Adverse events leading to treatment discontinuation
[e] One event of flu-like symptoms and hypotension occurred in the same patient
[f] Encephalopathy and hypereosinophilic syndrome occurred in the same patient
[g] Complete atrioventricular block, myocarditis, and myositis occurred in the same patient

Table 3. RECIST v1.1 Objective Response Rate

	Total Efficacy Evaluable[±]	PD-L1 <1%	PD-L1 ≥1%	PD-L1 Unknown
Efficacy Evaluable Patients, n	27	11	12	4
ORR* (CR+PR)	**13 (48%)**	**5 (45%)**	**6 (50%)**	**2 (50%)**
CR	5 (19%)	2 (18%)	3 (25%)	0
PR	8 (30%)	3 (27%)	3 (25%)	2 (50%)
DCR (CR+PR+SD)	**19 (70%)**	**8 (73%)**	**9 (75%)**	**2 (50%)**
SD	6 (22%)	3 (27%)	3 (25%)	0
PD	8 (30%)	3 (27%)	3 (25%)	2 (50%)

ORR for efficacy evaluable cis-ineligible population (n=16) is 44%
CR: complete response; DCR: disease control rate; PR: partial response; SD: stable disease
*As of data cut-off date of 12/3/2018, ORR by primary investigator assessment included 4 unconfirmed responses: 2 patients with uPR and 1 patient with uCR pending confirmatory scan and one patient with uPR discontinued for AE after first scan with no confirmatory scan. Since 12/3/2018, 3 of 4 patients have had scans confirming responses (including CR)
[±]Efficacy evaluable defined per protocol as patients with at least one post-baseline scan. As of 12/3/2018, 1 patient was excluded for non-eligibility (no target lesion), and 3 patients discontinued prior to first scan [1 due to patient decision; 1 due to clinical progression; 1 due to death from disease]; 10 patients pending first scan in database

Figure 2. Best Percentage Change from Baseline in Target Lesions

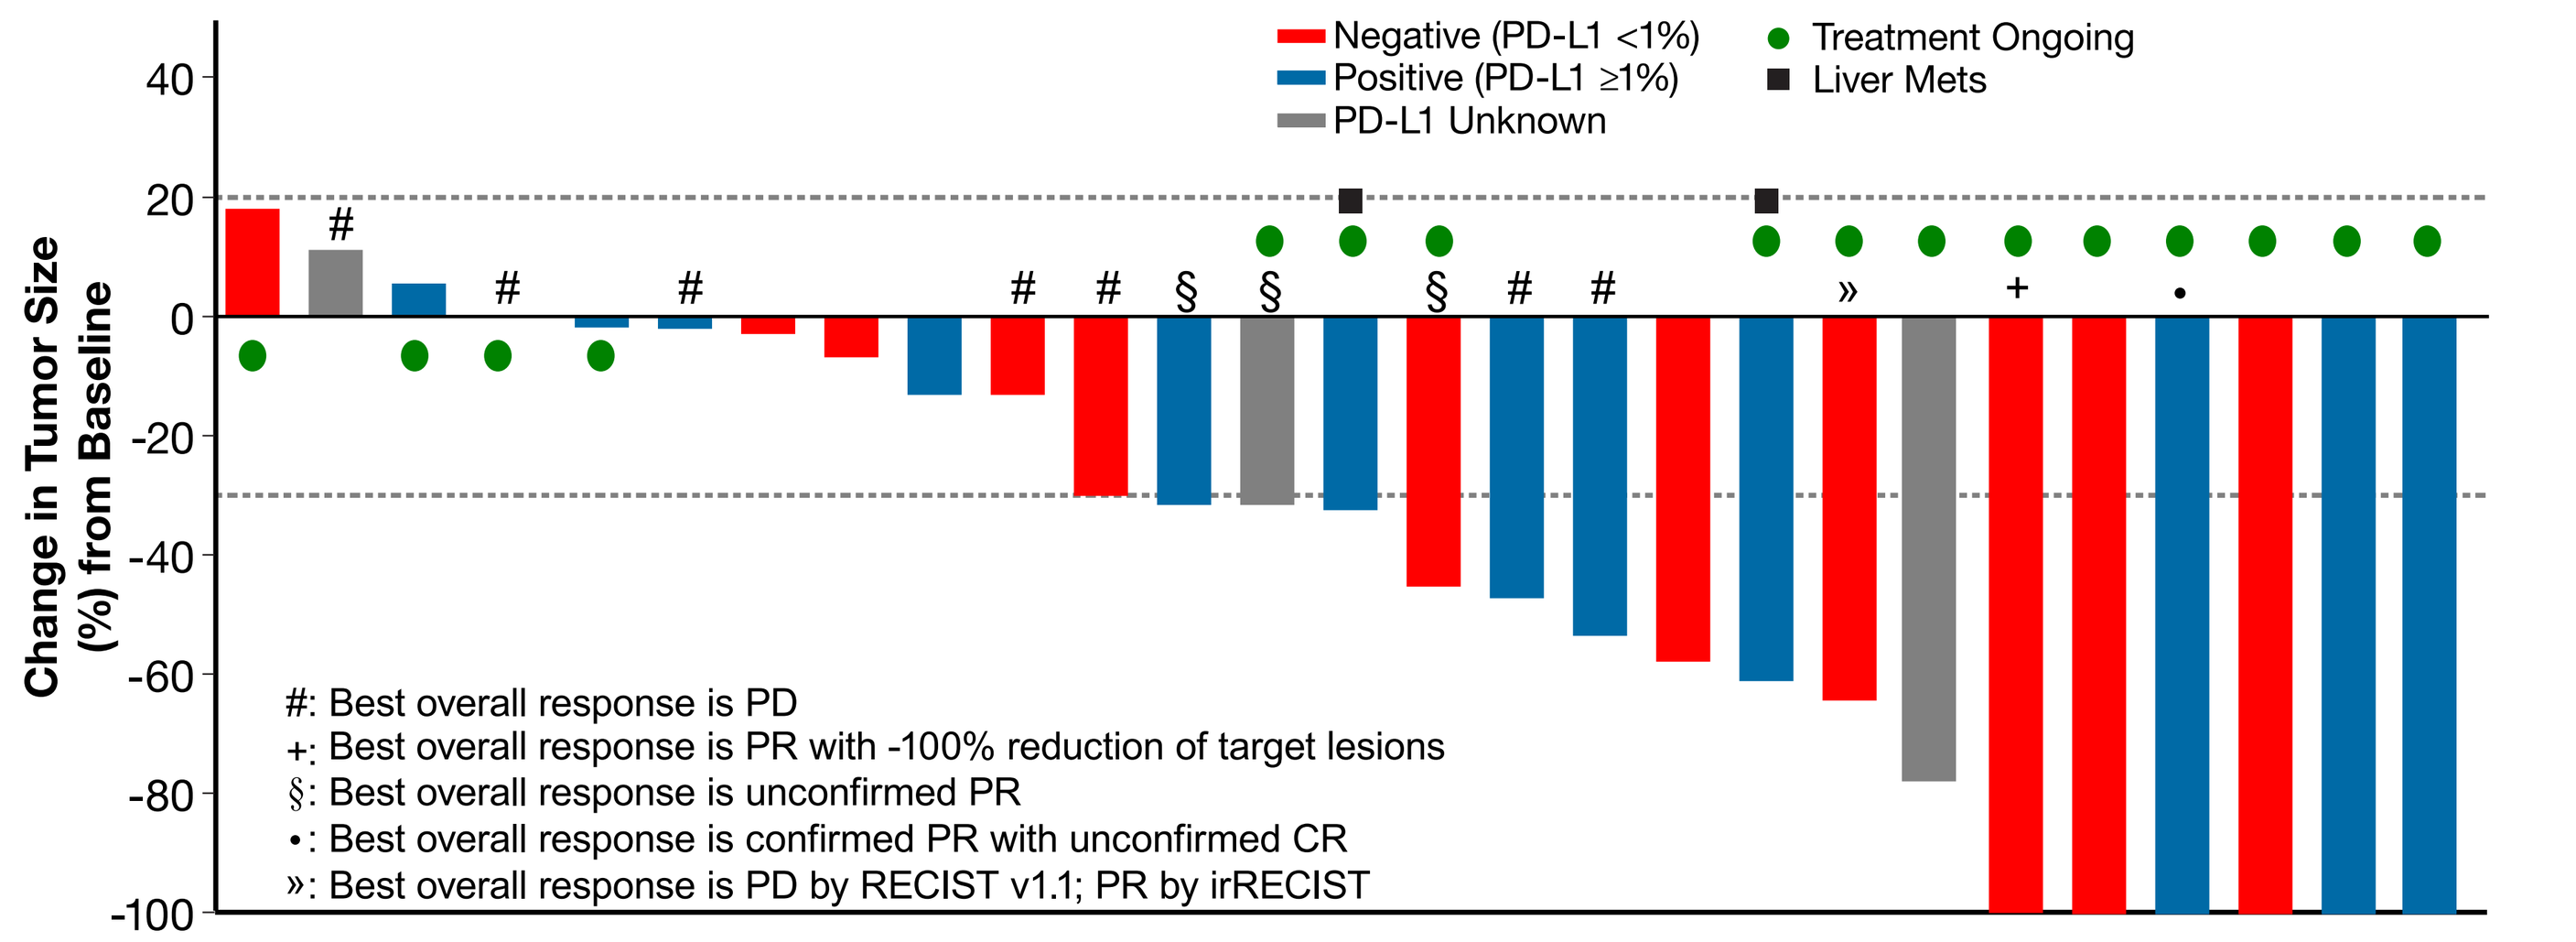
Negative (PD-L1 <1%)
Positive (PD-L1 ≥1%)
PD-L1 Unknown
Treatment Ongoing
Liver Mets
Change in Tumor Size (%) from Baseline
#: Best overall response is PD
+: Best overall response is PR with -100% reduction of target lesions
§: Best overall response is unconfirmed PR
•: Best overall response is confirmed PR with unconfirmed CR
»: Best overall response is PD by RECIST v1.1; PR by irRECIST

Efficacy Evaluable n=27	ORR
ORR by RECIST	13 (48%)
ORR by irRECIST	14 (52%)
Responses noted across all disease locations	
Visceral non-nodal metastases (n=15)	8 (53%)
Nodal metastases (n=11)	5 (46%)

Figure 3. Percent Change in Tumor Size by Week

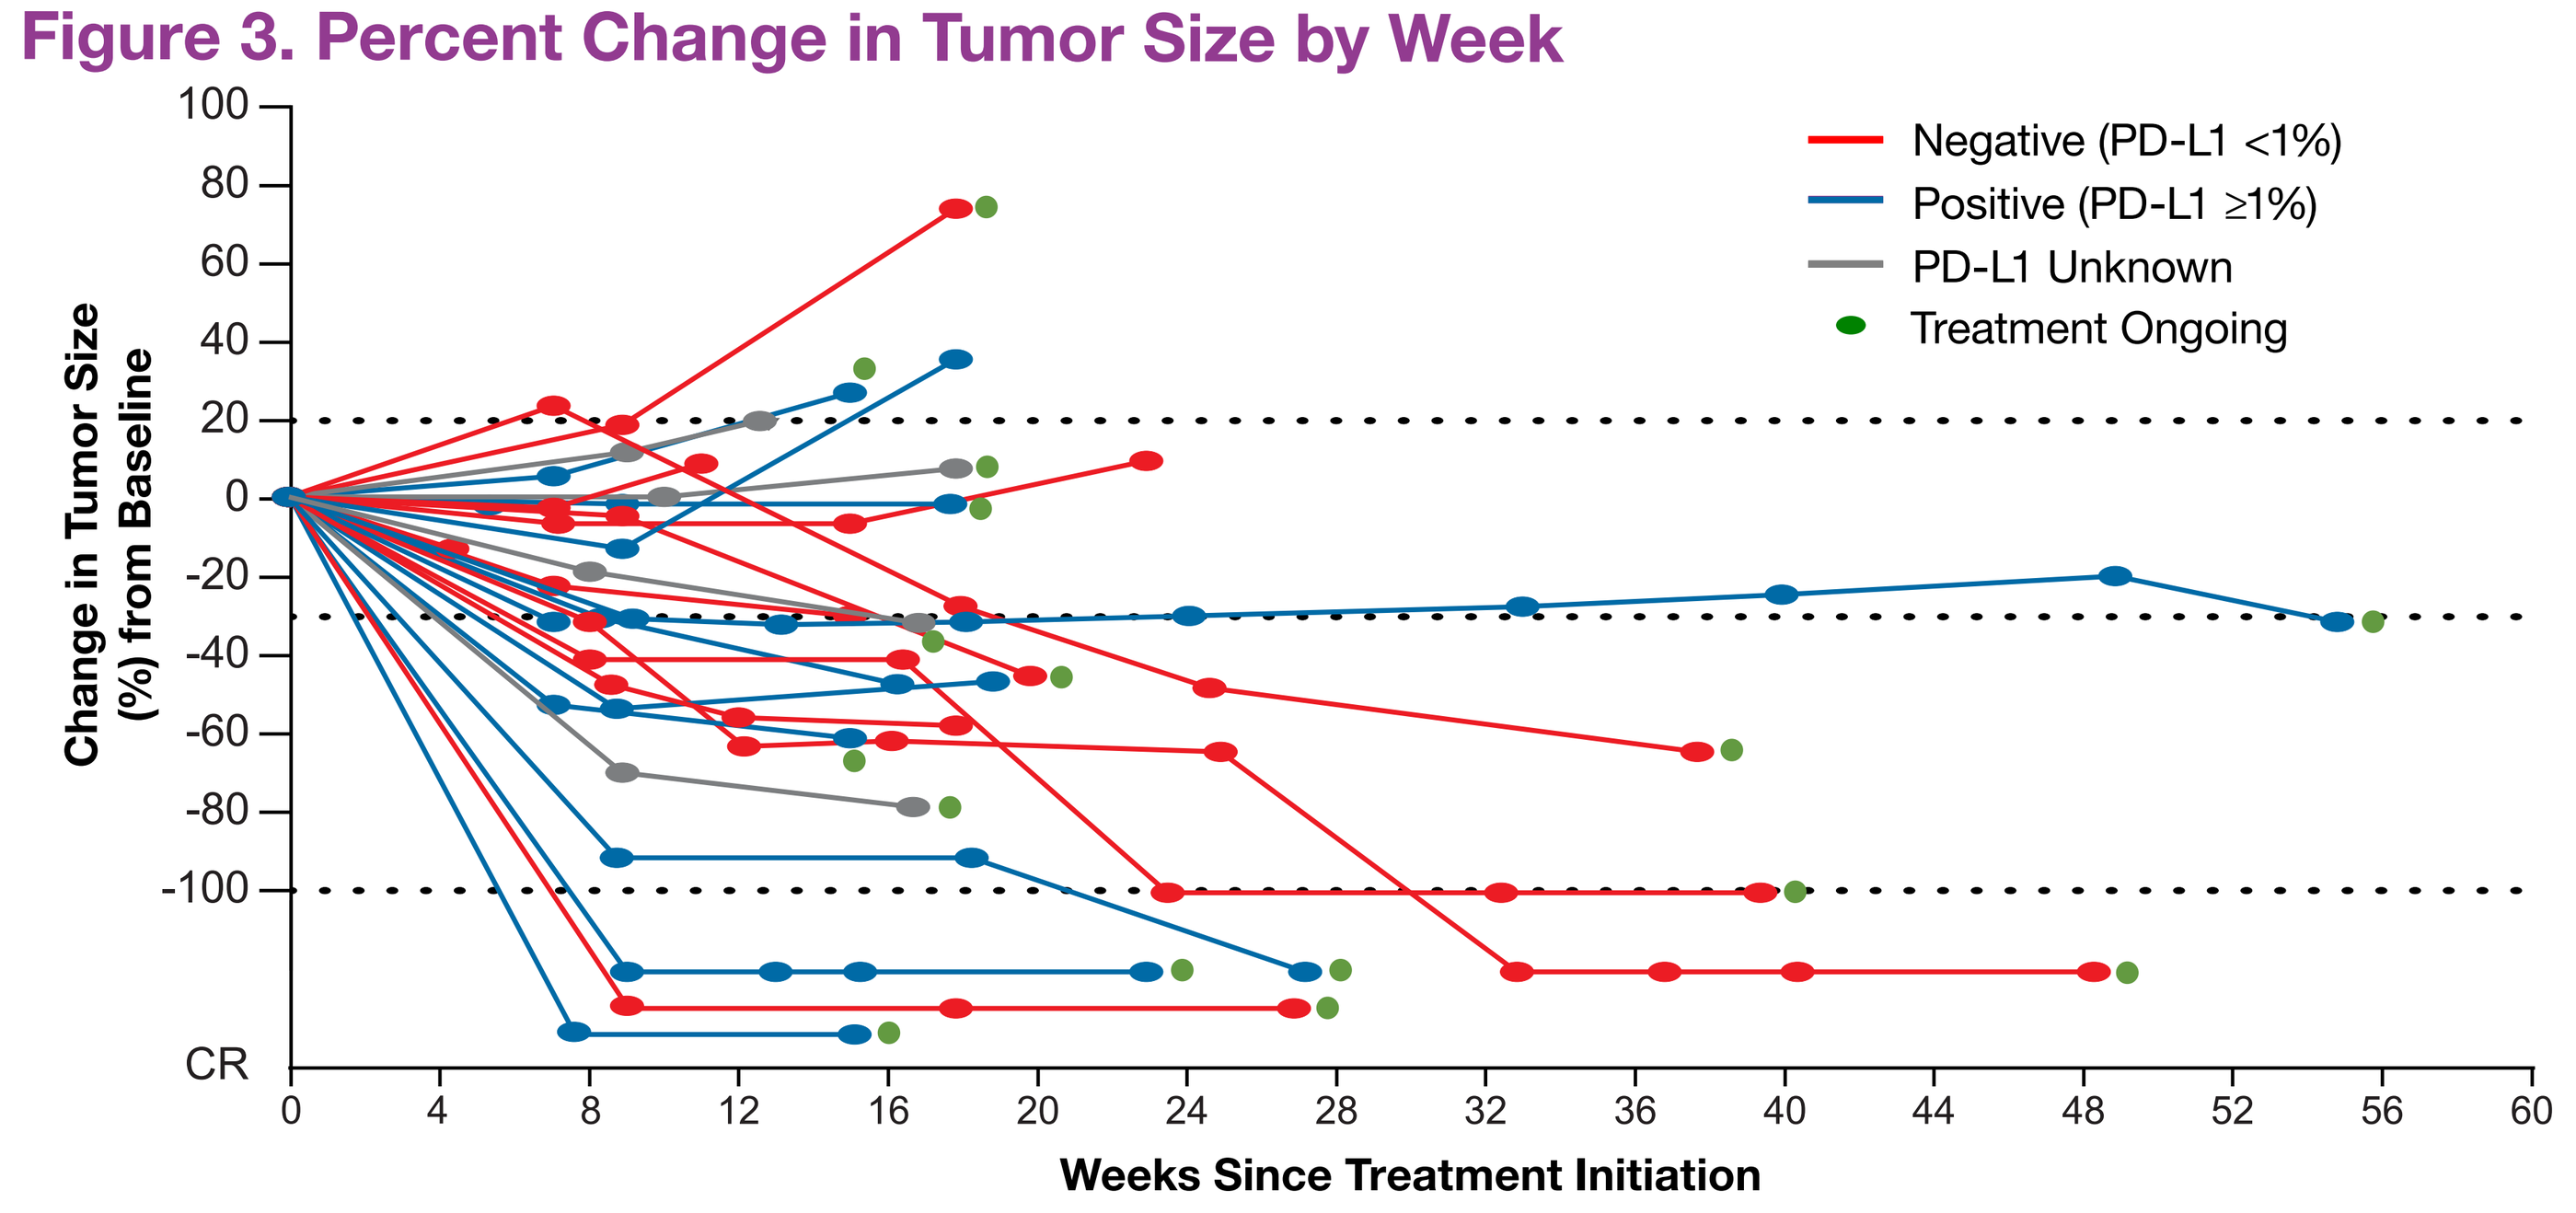
Negative (PD-L1 <1%)
Positive (PD-L1 ≥1%)
PD-L1 Unknown
Treatment Ongoing
Change in Tumor Size (%) from Baseline
CR
Weeks Since Treatment Initiation

1L mUC (n=27 Efficacy Evaluable)	
Median duration of follow-up (months)	5.1
Median time to response (months)	2
Patients with ongoing responses	11/13 (85%)
Median % reduction from baseline in responders as of 3DEC2018 (ongoing)	78%
Median % reduction from baseline, all efficacy evaluable patients	32%

In patients with RECIST response, no patients discontinued due to relapse. Two patients discontinued for TRAE.

Patient Case: Pseudoprogression

- One documented case of pseudoprogression was observed in a 70-year-old male with disease that included left external iliac lymph nodes (target lesion) and bilateral pulmonary nodules (non-target lesions) at baseline
- Initial tumor assessment (week 9) revealed 23% increase in target lesions; biopsy of progressing lesions revealed lymphocytic infiltrate on IHC staining
- Patient continued on treatment; the following scan (week 20) revealed a 28% decrease in target lesions from baseline. (**Figure 4**)
- The week 24 scan revealed a 48% reduction in target lesions from baseline and patient met criteria for PR by irRECIST. Further improvement was noted on subsequent scans. (**Figure 4**)
- Patient continues on study treatment; week 40 scan showed 64% decrease from baseline. (**Figure 4**)

Figure 4. Serial CT Scans Consistent With Pseudoprogression

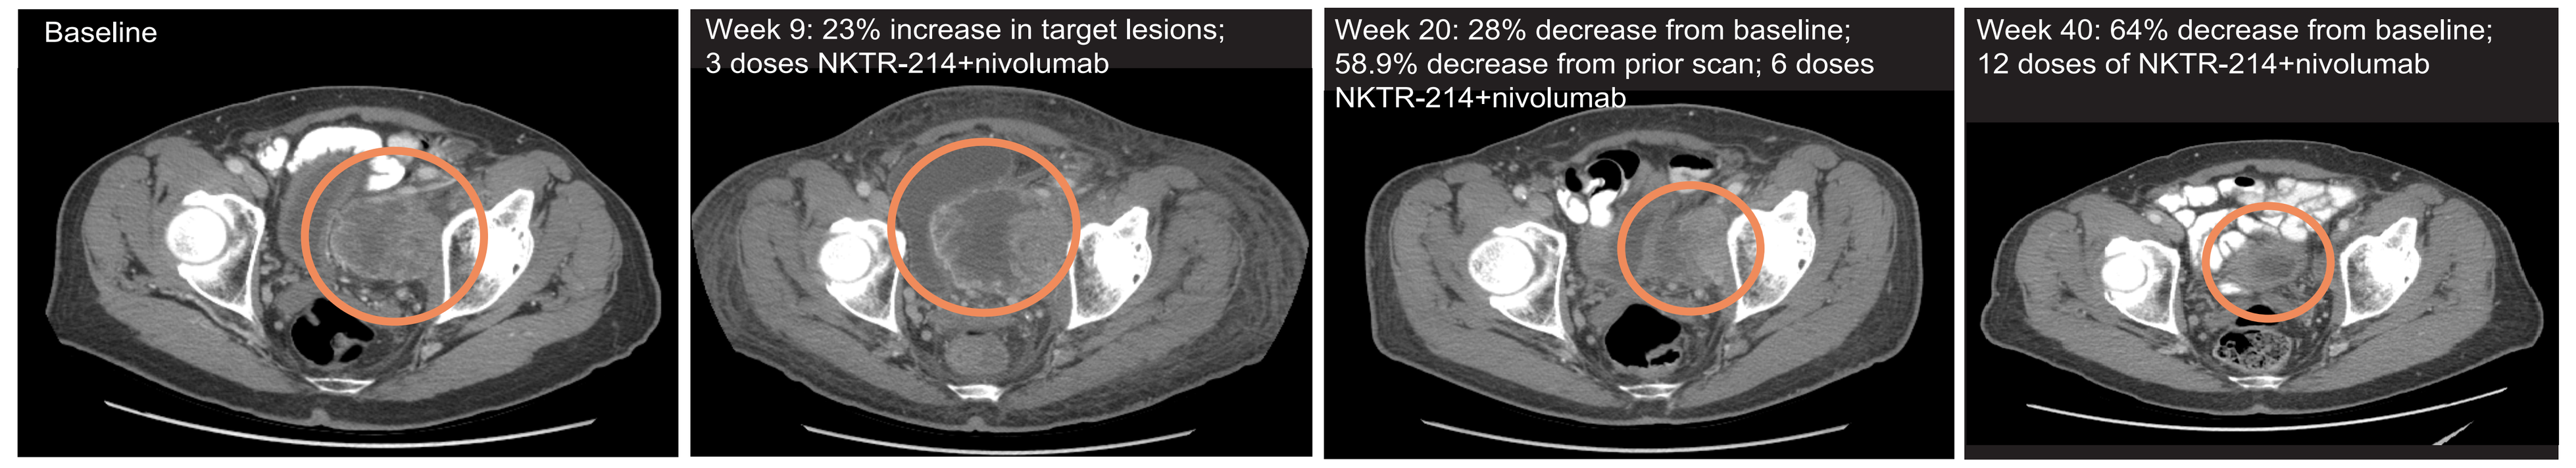
Baseline
Week 9: 23% increase in target lesions; 3 doses NKTR-214+nivolumab
Week 20: 28% decrease from baseline; 58.9% decrease from prior scan; 6 doses NKTR-214+nivolumab
Week 40: 64% decrease from baseline; 12 doses of NKTR-214+nivolumab

Biomarker Evaluation

- 13 paired tissue samples were evaluated for changes in PD-L1 expression (28-8 IHC PharmDx Assay; **Figure 5**)
- 7 of 10 (70%) PD-L1 negative samples at baseline converted to PD-L1 positive by week 3
- 3 of 3 patients who were PD-L1 positive at baseline remained PD-L1 positive

Figure 5. On-Treatment PD-L1 Conversion

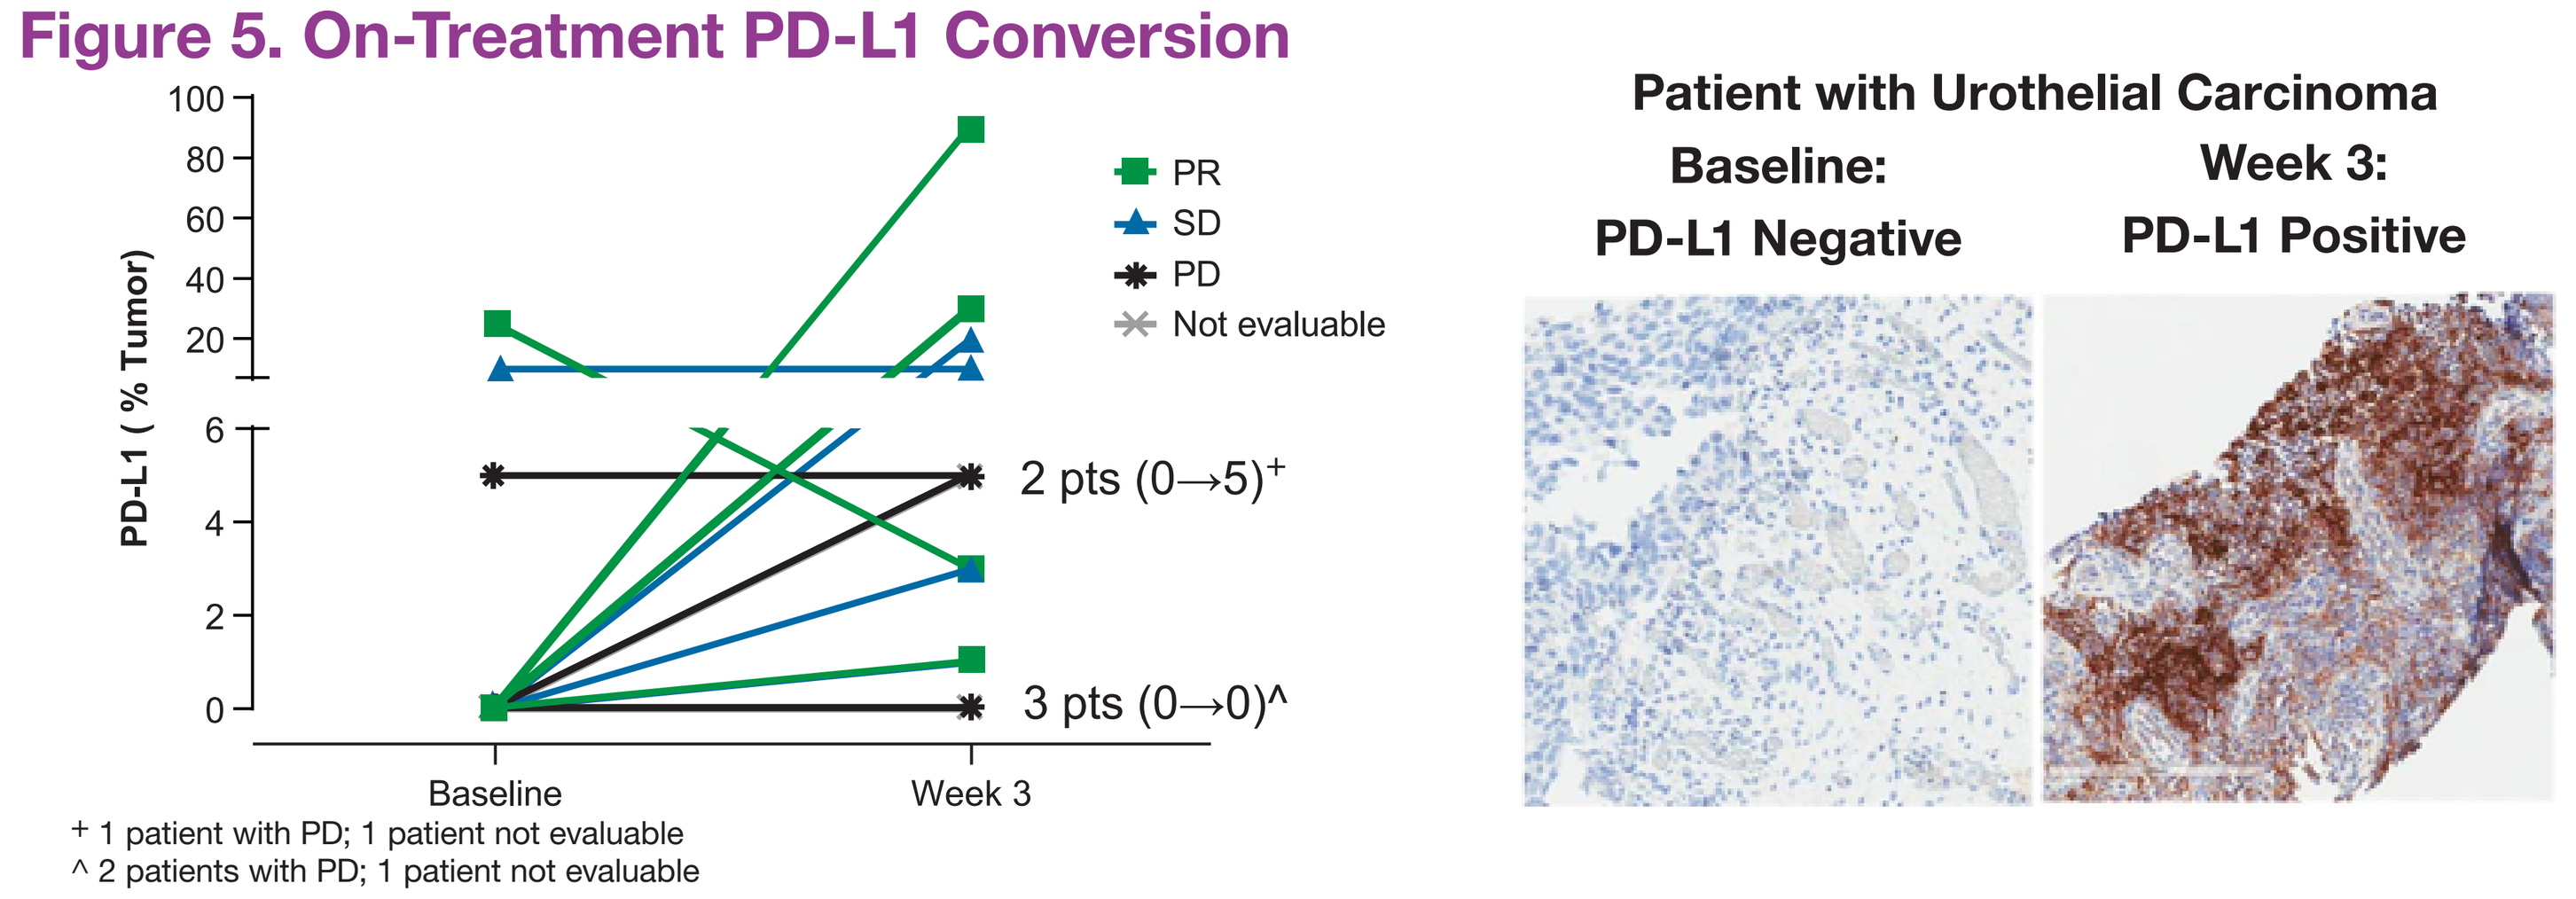
PD-L1 (% Tumor)
PR
SD
PD
Not evaluable
2 pts (0→5)+
3 pts (0→0)^
Baseline
Week 3
Patient with Urothelial Carcinoma
Baseline:
PD-L1 Negative
Week 3:
PD-L1 Positive

+ 1 patient with PD; 1 patient not evaluable
^ 2 patients with PD; 1 patient not evaluable

Figure 6. RECIST Responses Observed Independent of PD-L1 Status and CD8+ Infiltrate

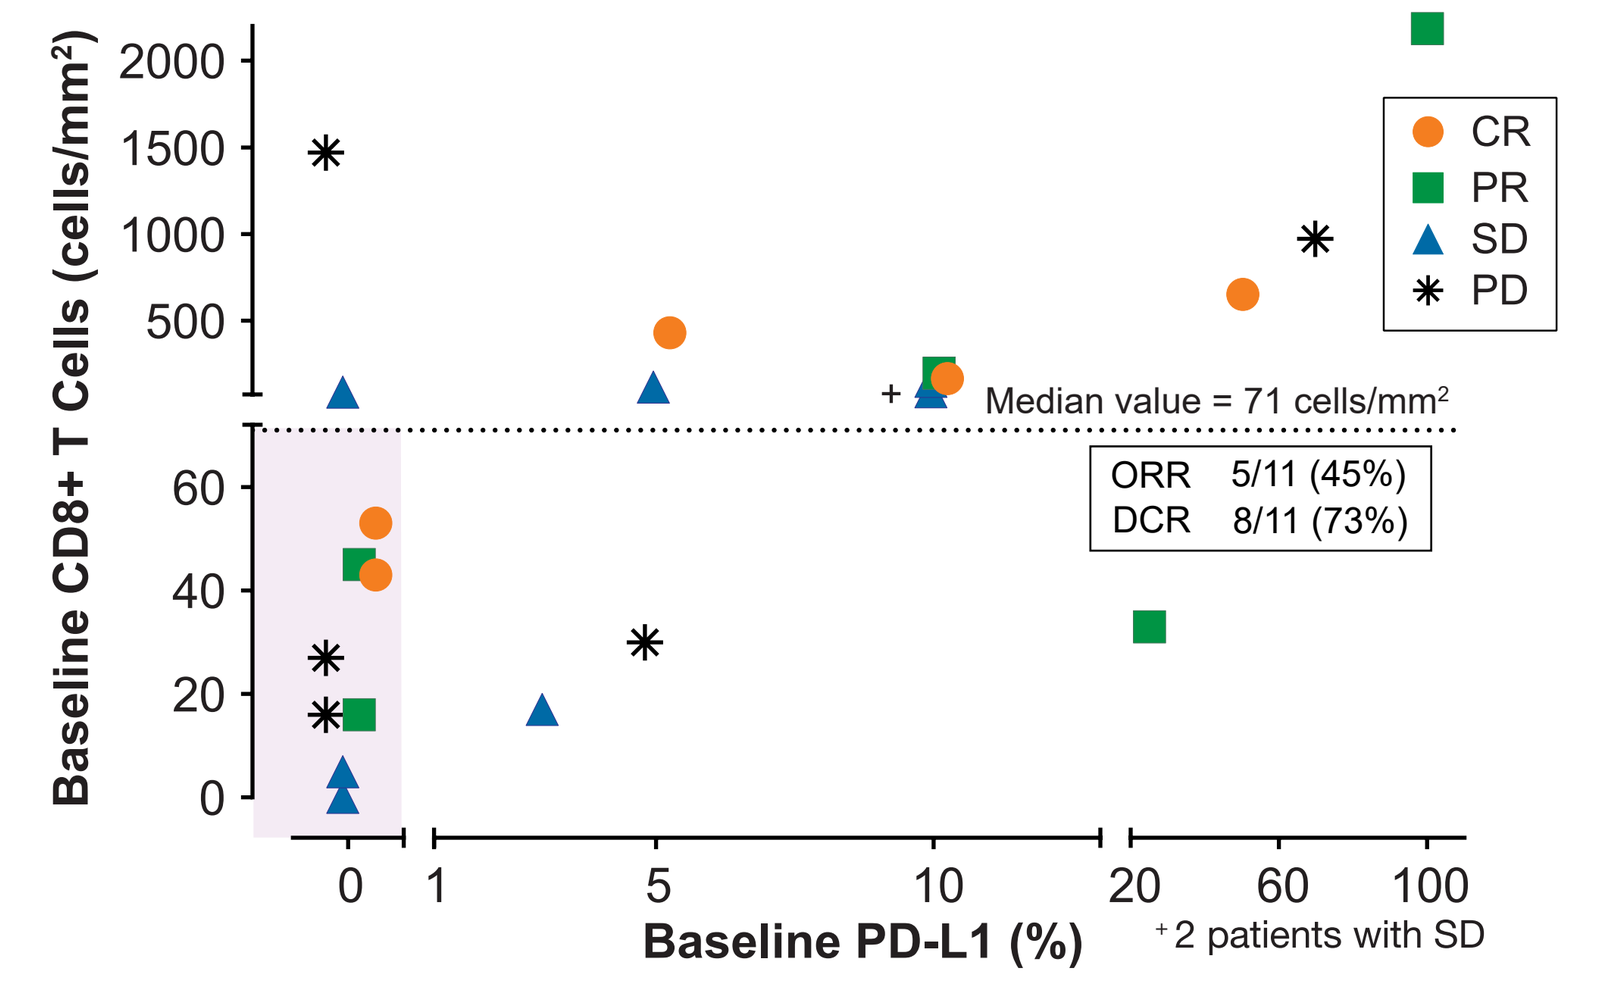
Baseline CD8+ T Cells (cells/mm²)
CR
PR
SD
PD
Median value = 71 cells/mm²
ORR 5/11 (45%)
DCR 8/11 (73%)
Baseline PD-L1 (%)
+2 patients with SD

- 22 baseline tissue samples* were evaluated for PD-L1 expression (28-8 IHC PharmDx Assay) and CD8 (Agilent CD8/144B antibody) positive cells and correlation with response
- Similar responses observed across patients regardless of baseline CD8+ TIL and PD-L1 expression
- 4/8 patients with both low CD8+ TIL and no PD-L1 expression achieved responses (noted by shading in figure)

*All patients with available baseline PD-L1 status and CD8+ TIL (n=22) were included in the analysis. Five patients from the 27 efficacy evaluable patients did not have sufficient biomarkers to be included in the analysis (4 were not available for PD-L1 and 1 was not evaluable for CD8+ TIL)

CONCLUSIONS

- Bempegaldesleukin (NKTR-214) plus nivolumab in 1L advanced/metastatic urothelial carcinoma was well tolerated and demonstrated promising clinical benefit in patients who were either cisplatin ineligible or cisplatin eligible who refused SOC
 - ORR in cisplatin-ineligible was 44%; ORR in refused SOC was 55%
 - Therapy demonstrated deep responses with CR rate of 19% (median 78% tumor shrinkage among responders)
 - No relapses observed among responders
- Responses were observed regardless of baseline PD-L1 expression
 - ORR in PD-L1 positive patients was 50% and ORR in PD-L1 negative was 45%
- Bempegaldesleukin plus nivolumab demonstrated conversion of PD-L1 status from negative at baseline to positive on treatment
 - 70% (7/10) of matched biopsies converted
- These data support the potential benefit of this combination in patients with urothelial cancer
 - Phase 2 study of bempegaldesleukin and nivolumab in progress to further evaluate efficacy and safety in the 1L cisplatin-ineligible population of patients whose tumors have low expression of PD-L1 (PIVOT-10, NCT03785925)

ACKNOWLEDGMENTS

The investigators would like to acknowledge the patients and clinical teams for their participation in this study; Dako for collaborative development of the PD-L1 IHC 28-8 pharmDx assay; Bristol-Myers Squibb (Princeton, NJ) and ONO Pharmaceutical Company Ltd. (Osaka, Japan). This study was sponsored by Nektar Therapeutics. All authors contributed to and approved the presentation. Writing and editorial assistance was provided by Mark Phillips, PharmD, MBA of Phillips Gilmore Oncology Communications and funded by Nektar Therapeutics.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster

REFERENCES

1. Tecentriq (atezolizumab) [package insert]. South San Francisco, CA: Genentech, Inc.; 2018.
2. Keytruda (pembrolizumab) [package insert]. Whitehouse Station, NJ: Merck & Co., Inc.; 2014-2018.
3. Opdivo (nivolumab) [package insert]. Princeton, NJ: Bristol-Myers Squib Company; 2018.
4. Imfinzi (durvalumab) [package insert]. Wilmington, DE: AstraZeneca Pharmaceuticals LP; 2018.
5. Bavencio (avelumab) [package insert]. New York, NY: EMD Serono, Inc. and Pfizer Inc.; 2018.
6. National Cancer Institute. FDA approves immunotherapy drugs for patients with bladder cancer. Cancer Current Blog. May 30, 2017.Available at https://www.cancer.gov/news-events/cancer-currents-blog/2017/approvals-fda-checkpoint-bladder.
7. Bentebibel S. et al. The Novel IL-2 Cytokine Immune Agonist NKTR-214 Harnesses the Adaptive and Innate Immune System for the Treatment of Solid Cancers. Presented as a part of SITC 2017; November 10, 2017; National Harbor, MD.
8. Diab A. et al. NKTR-214 (CD-122-biased agonist) plus nivolumab in patients with advanced solid tumors: Preliminary phase 1/2 results of PIVOT. Presented as a part of ASCO 2018; June 2, 2018; Chicago, IL.
9. Diab A. et al. Nektar Therapeutics Investor & Analyst Call with Melanoma Specialists. Presented as a part of SITC 2018; November 10, 2018; Washington DC.

Presented at ASCO GU, February 13-16, 2019, San Francisco, CA, USA