NKTR-255 + Cetuximab in Patients with Solid Tumors: Interim Safety and Efficacy Results from the Phase 1b Dose-escalation Study

957

Copies of this poster obtained through this Quick Response (QR) Code are for personal use only and may not be reproduced without written permission from SITC and the authors.

Mehmet Altan[1], Amita Patnaik[2], Minal A. Barve[3], Lara A. Dunn[4], Patrick W. Cobb[5], Ari Rosenberg[6], Sunil Sharma[7], Ammar Sukari[8], Manish R. Patel[9], Xiaoli Wang[10], Haijun Ma[11], Neha Dixit[12], Wildaliz Nieves[12], Christie Fanton[12], Sue L. Currie[13], Zachary Lee[13], Mario Q. Marcondes[13], Jonathan Zalevsky[14], Mary A. Tagliaferri[13], Assuntina G. Sacco[15]

[1]Thoracic/Head and Neck Medical Oncology, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Clinical Research, South Texas Accelerated Research Therapeutics (START), San Antonio, TX, USA; [3]Medical Oncology/Hematology, Mary Crowley Cancer Research Center, Dallas, TX, USA; [4]Medical Oncology/Hematology, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [5]Medical Oncology/Hematology, St. Vincent Frontier Cancer Center, Billings, MT, USA; [6]Medical Oncology/Hematology, University of Chicago, Chicago, IL, USA; [7]Medical Oncology/Hematology, HonorHealth Research Institute, Scottsdale, AZ, USA; [8]Medical Oncology, Karmanos Cancer Center, Detroit, MI, USA; [9]Hematology, Oncology, and Transplantation, University of Minnesota Medical Center, Minneapolis, MN, USA; [10]Clinical Pharmacology, Nektar Therapeutics, San Francisco, CA, USA; [11]Biostatistics, Nektar Therapeutics, San Francisco, CA, USA; [12]Research Biology, Nektar Therapeutics, San Francisco, CA, USA; [13]Clinical Development, Nektar Therapeutics, San Francisco, CA, USA; [14]Research and Development, Nektar Therapeutics, San Francisco, CA, USA; [15]Medical Oncology, University of California San Diego, La Jolla, CA, USA

BACKGROUND

- There is an unmet need for new cancer immunotherapies that can boost the number and function of NK cells to improve clinical outcomes
- NKTR-255 is a polymer-conjugated rhIL-15 agonist that engages the full IL-15 receptor pathway providing sustained PD responses without the need for daily dosing[1]
- In preclinical models, NKTR-255 induced the proliferation and activation of NK cells and promoted the survival and expansion of CD8+ T cells.[1] NKTR-255 also enhanced the antitumor activity of tumor-targeted antibodies with an ADCC mechanism[1,2]
- This phase 1b/2 (NCT04616196)[3] dose-escalation study with expansion cohorts evaluates the safety and antitumor activity of NKTR-255 plus cetuximab in patients with R/R HNSCC and CRC
- **Here we report preliminary data on safety, PK/PD, and efficacy from the dose-escalation portion**

NKTR-255 Engages the IL-15Rα/IL-2Rβγ Complex to Boost NK Cell and CD8+ T-cell Expansion, Proliferation, Activation, Function, and Survival[2]

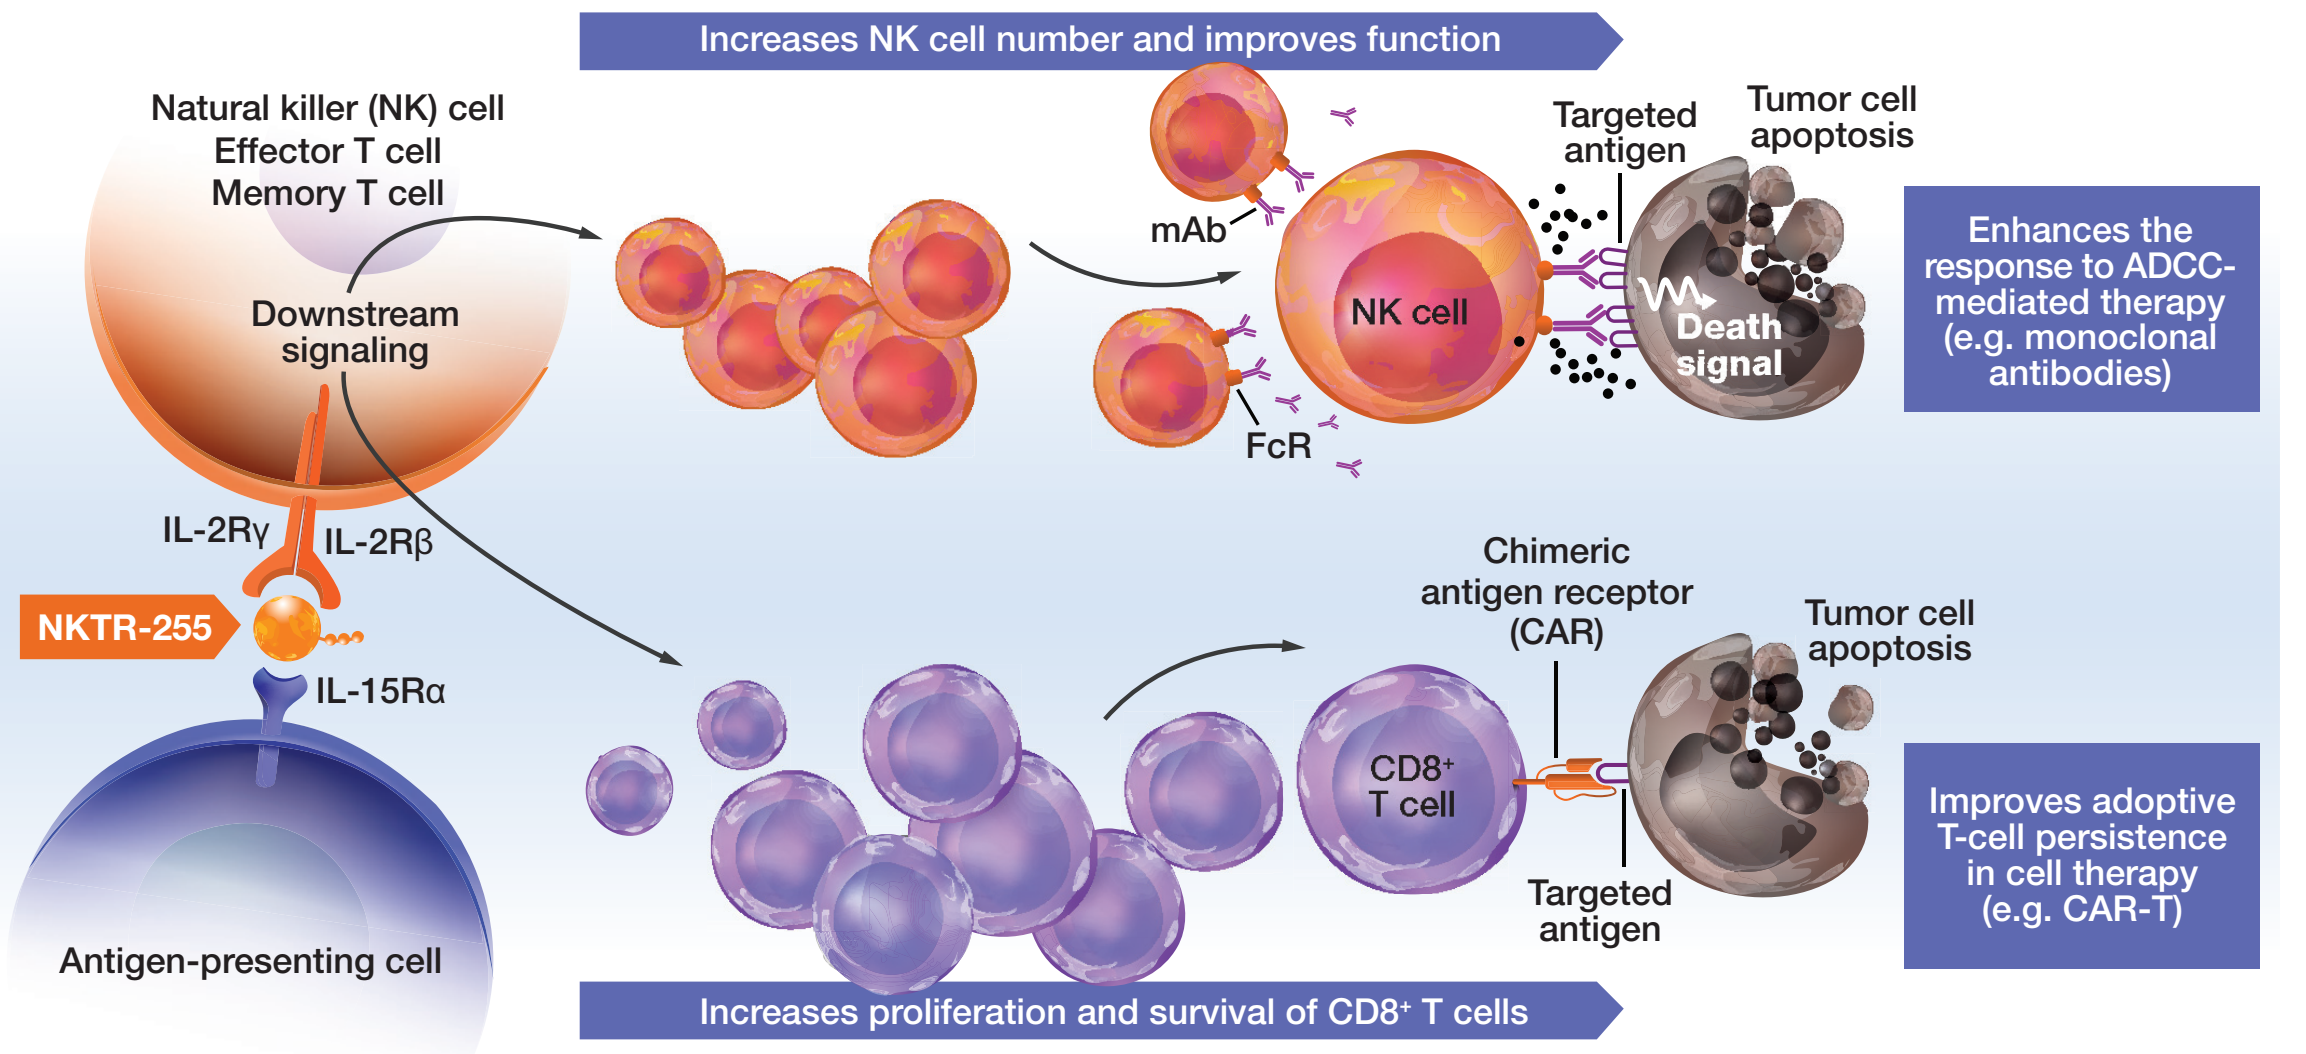
Increases NK cell number and improves function
Natural killer (NK) cell
Effector T cell
Memory T cell
Downstream signaling
mAb
FcR
NK cell
Targeted antigen
Tumor cell apoptosis
Death signal
Enhances the response to ADCC-mediated therapy (e.g. monoclonal antibodies)
IL-2Rγ
IL-2Rβ
NKTR-255
IL-15Rα
Antigen-presenting cell
Chimeric antigen receptor (CAR)
CD8+ T cell
Targeted antigen
Tumor cell apoptosis
Improves adoptive T-cell persistence in cell therapy (e.g. CAR-T)
Increases proliferation and survival of CD8+ T cells

ADCC, antibody-dependent cellular cytotoxicity; CAR-T, chimeric antigen receptor T-cell therapy; CD, cluster of differentiation; FcR, Fc receptor; IL-2R, interleukin-2 receptor; IL-15R, interleukin-15 receptor; mAb, monoclonal antibody; NK, natural killer.

PHASE 1B/2 STUDY DESIGN

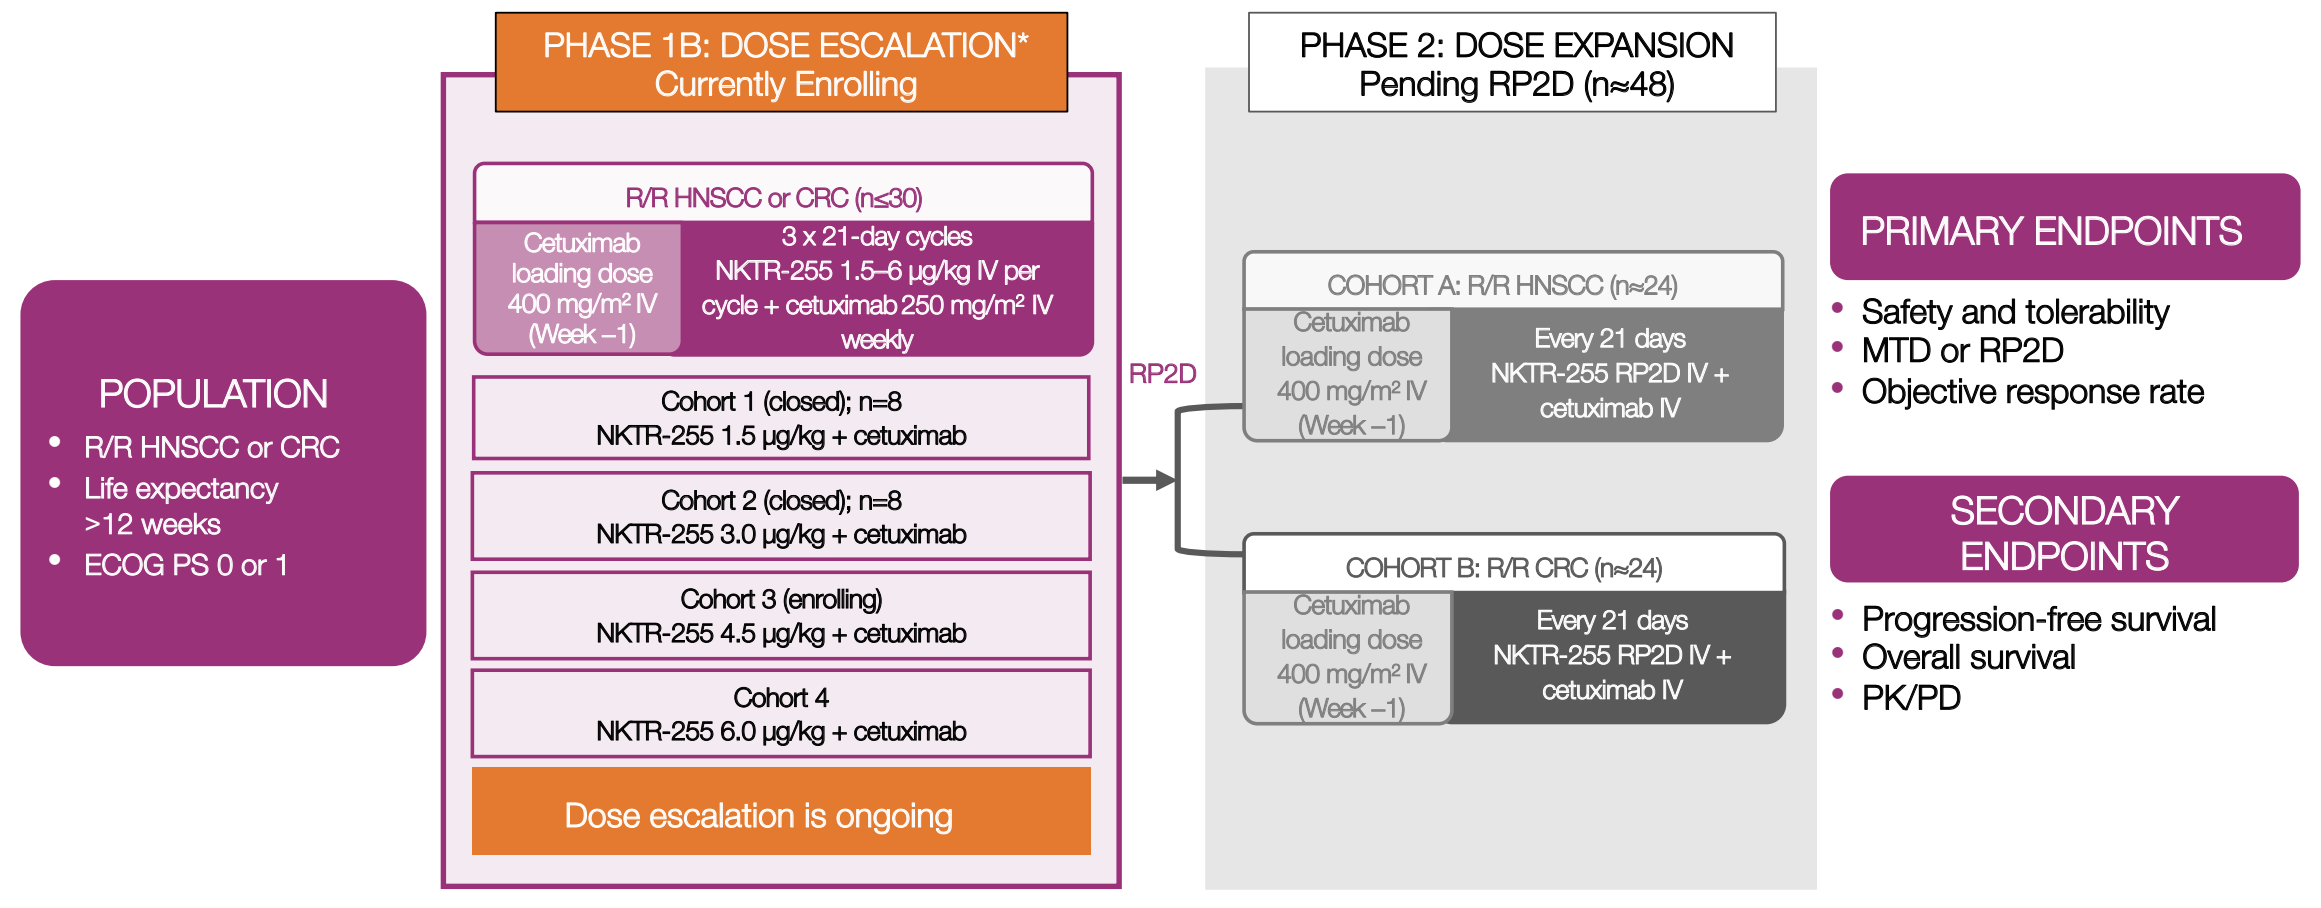
POPULATION
R/R HNSCC or CRC
Life expectancy >12 weeks
ECOG PS 0 or 1
PHASE 1B: DOSE ESCALATION*
Currently Enrolling
R/R HNSCC or CRC (n≤30)
Cetuximab loading dose 400 mg/m² IV (Week –1)
3 x 21-day cycles NKTR-255 1.5–6 μg/kg IV per cycle + cetuximab 250 mg/m² IV weekly
Cohort 1 (closed); n=8
NKTR-255 1.5 μg/kg + cetuximab
Cohort 2 (closed); n=8
NKTR-255 3.0 μg/kg + cetuximab
Cohort 3 (enrolling)
NKTR-255 4.5 μg/kg + cetuximab
Cohort 4
NKTR-255 6.0 μg/kg + cetuximab
Dose escalation is ongoing
RP2D
PHASE 2: DOSE EXPANSION
Pending RP2D (n≈48)
COHORT A: R/R HNSCC (n≈24)
Cetuximab loading dose 400 mg/m² IV (Week –1)
Every 21 days NKTR-255 RP2D IV + cetuximab IV
COHORT B: R/R CRC (n≈24)
Cetuximab loading dose 400 mg/m² IV (Week –1)
Every 21 days NKTR-255 RP2D IV + cetuximab IV
PRIMARY ENDPOINTS
Safety and tolerability
MTD or RP2D
Objective response rate
SECONDARY ENDPOINTS
Progression-free survival
Overall survival
PK/PD

NCT04616196. *Dose-escalation rules: Successive cohorts each receive escalating doses of NKTR-255 every 21 days plus a fixed dose of cetuximab weekly to determine the MTD/RP2D. A two-parameter Bayesian logistic regression model was used to guide each dose level and determine the MTD. The MTD will be declared when at least 6 patients have been evaluated at a dose and the posterior probability of targeted toxicity is at least 60% for that dose. Patients who achieve an optimal response (PR or CR), will be given the option to continue NKTR-255 (every 28 days) as maintenance therapy. CR, complete response; CRC, colorectal cancer; ECOG PS, Eastern Cooperative Oncology Group performance status; HNSCC, head and neck squamous cell carcinoma; IV, intravenous; MTD, maximum tolerated dose; PD, pharmacodynamics; PK, pharmacokinetics; PR, partial response; RP2D, recommended phase 2 dose; R/R, relapsed/refractory.

Study Procedures and Assessments (at October 13, 2021)

Safety and Tolerability	PK and PD	Efficacy
• AEs were assessed by CTCAE v5.0 • The safety evaluable population includes all patients who received ≥1 dose of treatment	• PK with concentration–time profiles (data cutoff: June 8, 2021) • PD (data cutoff: July 30, 2021) – Assessment of CD4+ T cells, CD8+ T cells, and NK Cells – Evaluation of inflammatory cytokines – Evaluation of Tregs	• Objective response: assessed by investigator every 9 weeks (±1 week) per RECIST v1.1 • Per protocol, efficacy-evaluable population defined as patients with ≥1 post-baseline, on-treatment radiographic scan

RESULTS

Heavily Pretreated Population Enrolled in Dose Escalation Phase

Patient Demographics and Disease Characteristics

Patients with HNSCC		N=4
Median age, years		59.5
Sex, n (%)	Female Male	0 4 (100)
ECOG PS, n (%)	0 1	2 (50) 2 (50)
Metastatic disease, n (%)		4 (100)
Median (range) number of prior therapies		4 (1–4)
Previous EGFR-targeted therapy, n (%)		1 (25)
Previous CPI, n (%)		3 (75)

Patients with CRC		N=10
Median age, years		59.5
Sex, n (%)	Female Male	2 (20) 8 (80)
ECOG PS, n (%)	0 1	4 (40) 6 (60)
Mutational status, n (%)	*BRAF/RAS* all wild-type *KRAS* or *NRAS* mutation *RAS* wild-type *BRAF* missing	5 (50) 4 (40) 1 (10)
Median (range) number of prior therapies		3 (2–8)
Previous EGFR-targeted therapy, n (%)		4 (40)
Previous CPI, n (%)		3 (30)

Data cutoff: October 13, 2021. CPI, checkpoint inhibitor; CRC, colorectal cancer; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; HNSCC, head and neck squamous cell carcinoma.

NKTR-255 + Cetuximab was Well Tolerated with no Grade 4/5 TRAEs

Selected TRAEs; n (%)	NKTR-255 dose		Total (N=14)
	1.5 μg/kg (n=7)	3.0 μg/kg (n=7)	
Grade 1 or 2 (≥1 patient)			
Chills	1 (14.3)	1 (14.3)	2 (14.3)
Fatigue	1 (14.3)	2 (28.6)	3 (21.4)
Infusion-related reaction	2 (28.6)	6 (85.7)	8 (57.1)
Nausea	0	2 (28.6)	2 (14.3)
Grade 3 (all)			
Infusion-related reaction/hypoxia*	0	1 (14.3)	1 (7.1)

Data cutoff: October 13, 2021. *This event was considered a NKTR-255-related DLT and resolved within 24 hours. DLT, dose-limiting toxicity; TRAE, treatment-related adverse event.

NKTR-255 Demostrates Extended $T_{½}$ with Minimal Accumulation

Concentration–time profiles

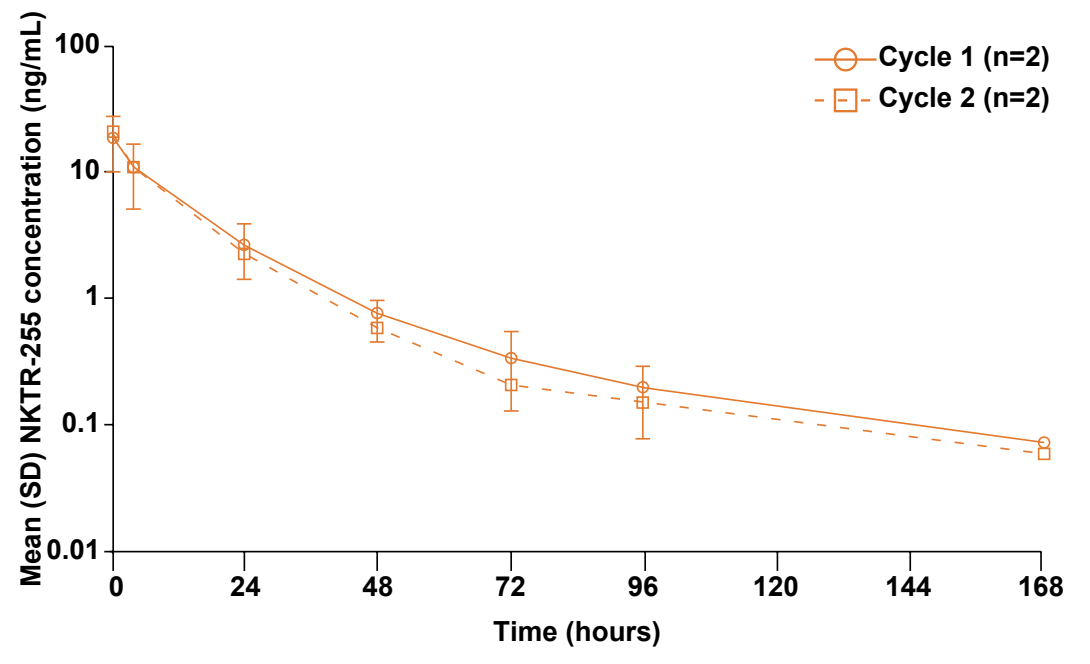
Cycle 1 (n=2)
Cycle 2 (n=2)
Mean (SD) NKTR-255 concentration (ng/mL)
100
10
1
0.1
0.01
0
24
48
72
96
120
144
168
Time (hours)

Preliminary PK analyses

Dose	Cycle	T_{max} (hr)	C_{max} (ng/mL)	AUC_{last} (hr*ng/mL)	$T_{½}$ (hr)	CL (L/hr)
1.5 μg/kg	1 (n=2)	0.58 (0.52, 0.65)	18.4 (8.90)	209 (50.1)	27.8 (33.2)	0.79 (69.9)
	2 (n=2)	0.53 (0.52, 0.53)	21.5 (30.9)	204 (18.4)	20.2 (62.2)	0.70 (41.7)

PK parameters: Mean (coefficient of variance %). All pre-dose NKTR-255 concentrations at Cycle 2+ were below the lower limit of quantification of 0.05 ng/mL.

The mean $T_{½}$ of NKTR-255 (1.5 μg/kg) was 27.8 hours, which is ≈10-fold longer than that reported for rhIL-15,[4] with no or minimal accumulation following repeat dosing.

Data cutoff: June 8, 2021.
A validated method was used to measure plasma concentration of NKTR-255, which was expressed in IL-15 content.
AUC, area under the curve; CL, clearance; C_{max}, peak concentration; IL-15, interleukin-15; PK, pharmacokinetic; $T_{½}$, half-life; T_{max}, time to maximum concentration; SD, standard deviation.

NKTR-255 + Cetuximab Induced Transient Upregulation and Rapid Decline of Inflammatory Cytokines by Day 2–3

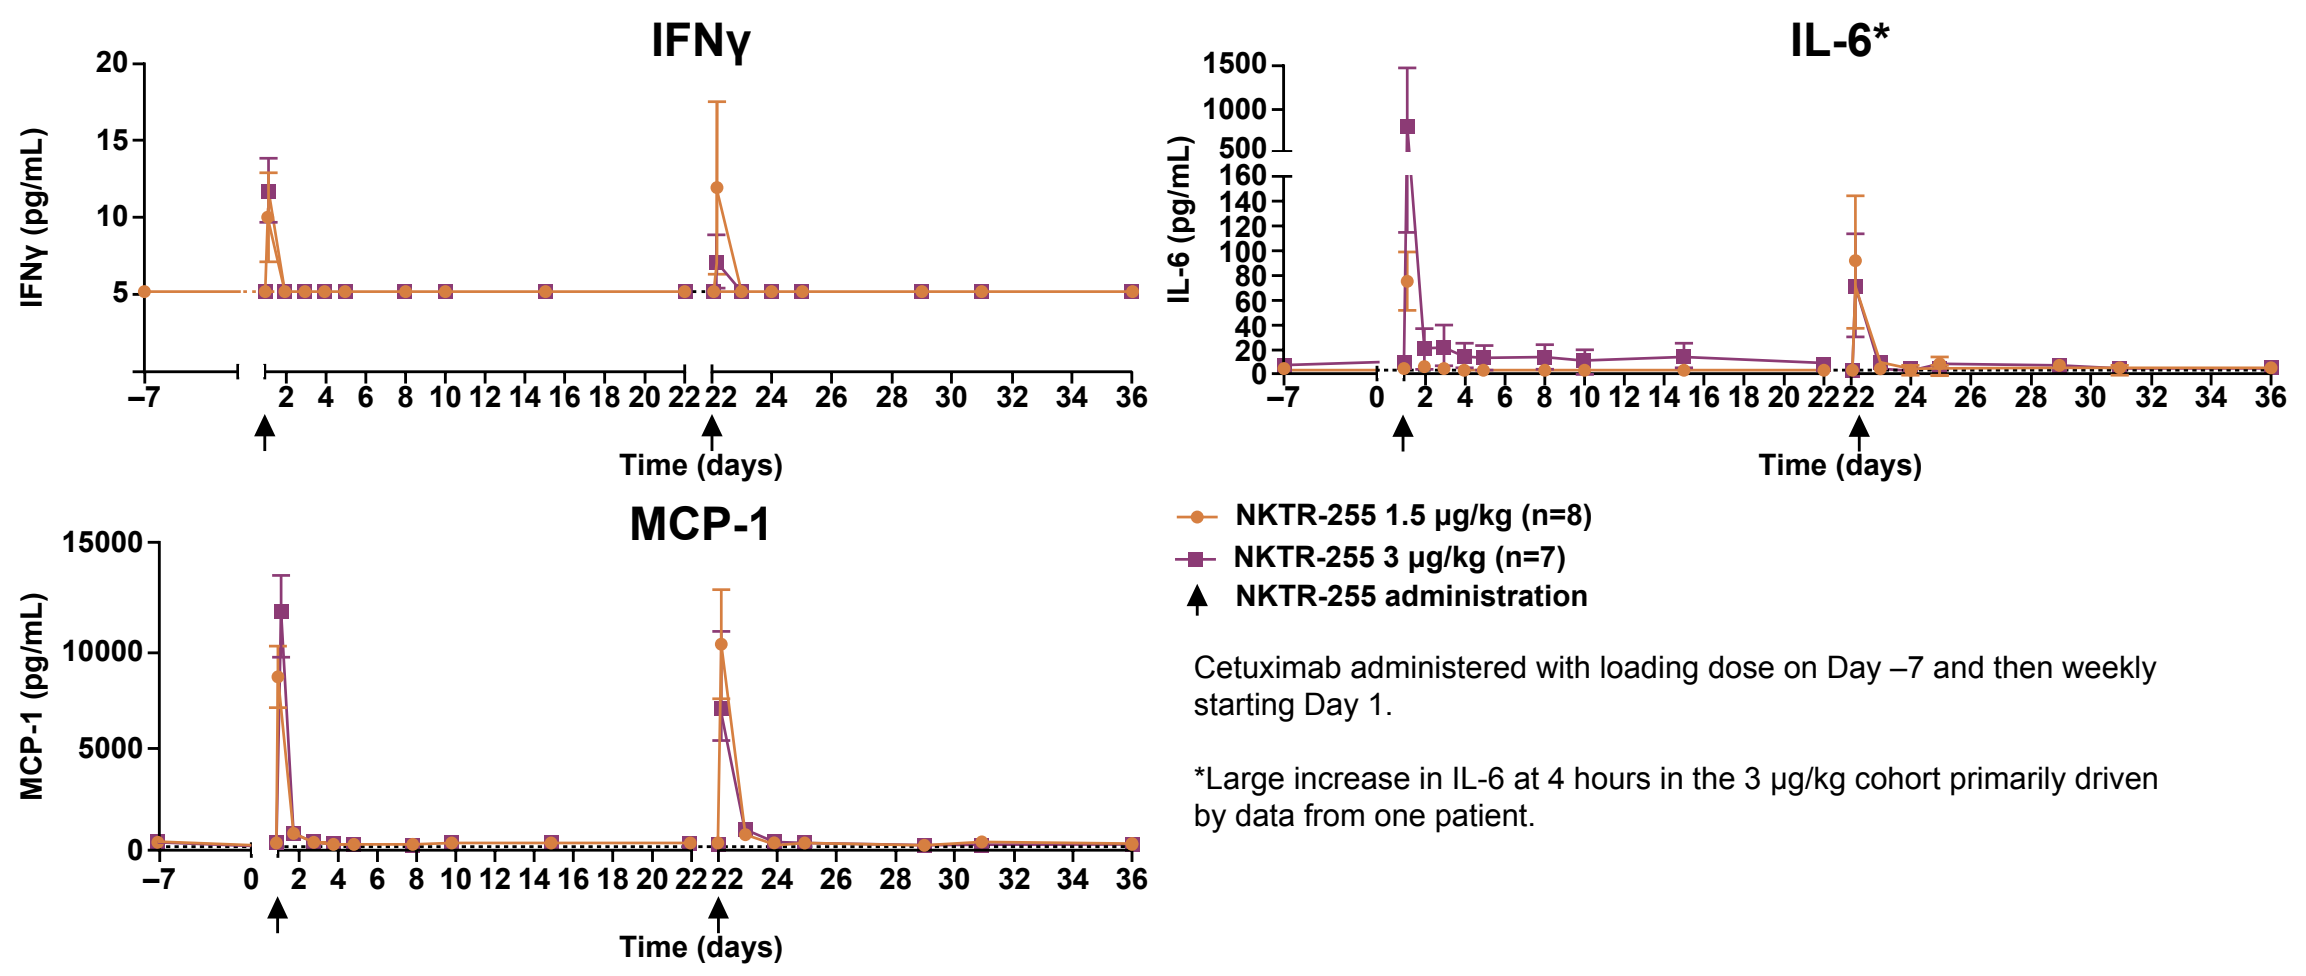
IFNγ
IFNγ (pg/mL)
IL-6*
IL-6 (pg/mL)
MCP-1
MCP-1 (pg/mL)
Time (days)
NKTR-255 1.5 μg/kg (n=8)
NKTR-255 3 μg/kg (n=7)
NKTR-255 administration

Cetuximab administered with loading dose on Day –7 and then weekly starting Day 1.

*Large increase in IL-6 at 4 hours in the 3 μg/kg cohort primarily driven by data from one patient.

IFN, interferon; IL, interleukin; MCP, monocyte chemoattractant protein.

NKTR-255 + Cetuximab Shows Evidence of Preliminary Efficacy at Initial Doses

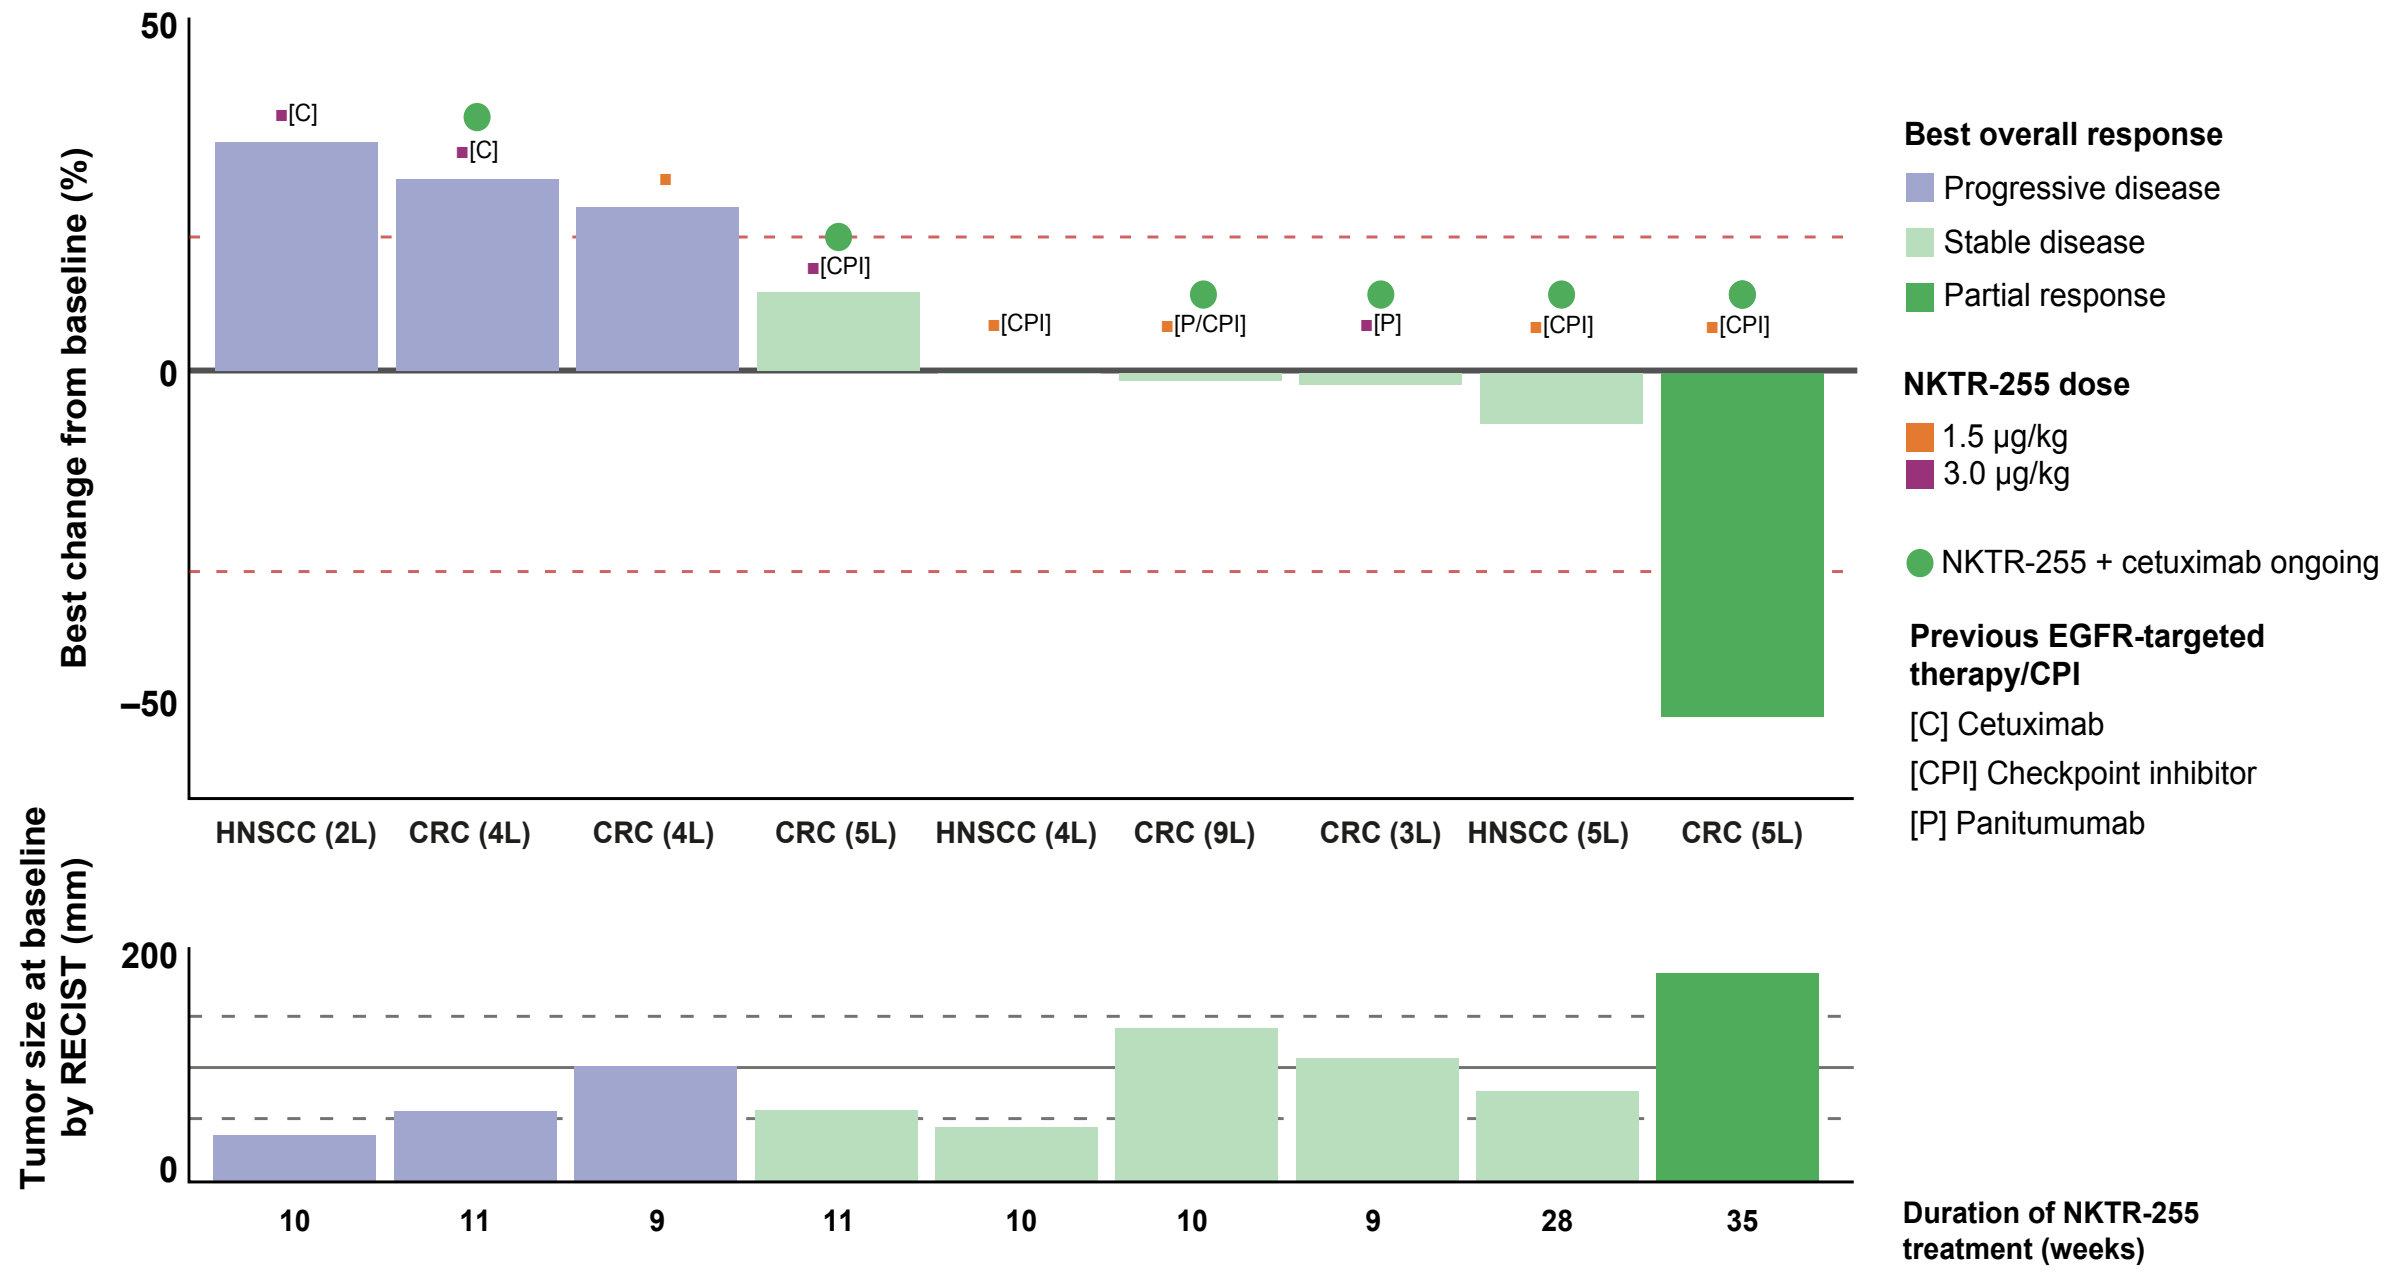
Best change from baseline (%)
50
0
–50
[C]
[C]
[CPI]
[CPI]
[P/CPI]
[P]
[CPI]
[CPI]
HNSCC (2L)
CRC (4L)
CRC (4L)
CRC (5L)
HNSCC (4L)
CRC (9L)
CRC (3L)
HNSCC (5L)
CRC (5L)
Tumor size at baseline by RECIST (mm)
200
0
10
11
9
11
10
10
9
28
35
Duration of NKTR-255 treatment (weeks)
Best overall response
Progressive disease
Stable disease
Partial response
NKTR-255 dose
1.5 μg/kg
3.0 μg/kg
NKTR-255 + cetuximab ongoing
Previous EGFR-targeted therapy/CPI
[C] Cetuximab
[CPI] Checkpoint inhibitor
[P] Panitumumab

One patient experienced a DLT and was not included in the analysis, 4 patients experienced symptoms consistent with progression and discontinued without protocol specified tumor assessment. CPI, checkpoint inhibitor; C, cetuximab; DLT, dose-limiting toxicity; CRC, colorectal cancer; HNSCC, head and neck squamous cell carcinoma; P, panitumumab; RECIST, Response Evaluation Criteria in Solid Tumors.

NKTR-255 + Cetuximab Led to Greater Expansion of NK Cells* in Patients with Clinical Benefit Versus Patients with Progressive Disease, Supporting the MOA of NKTR-255

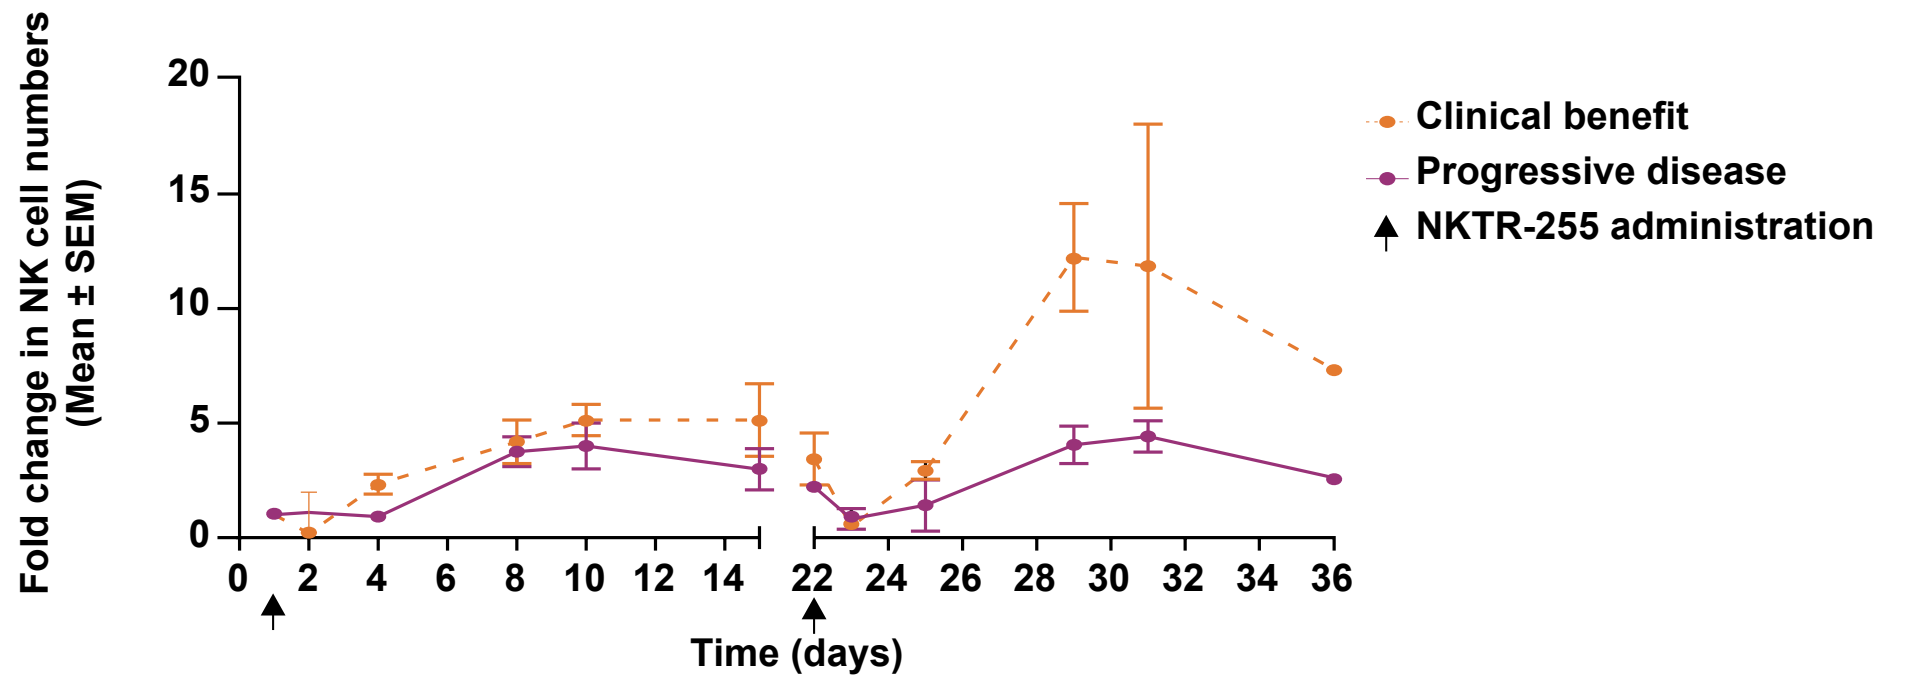
Fold change in NK cell numbers (Mean ± SEM)
Time (days)
Clinical benefit
Progressive disease
NKTR-255 administration

*Data combined for both Cohorts 1 and 2. Cycle 1: clinical benefit n=5, progressive disease n=3. NK, natural killer; SEM, standard error of the mean.

NKTR-255 + Cetuximab Led to Expansion and Increased Proliferative Capacity of NK and CD8+ T Cells

Minimal changes in CD4+ Tregs in blood were observed with NKTR-255 + cetuximab (data not shown)

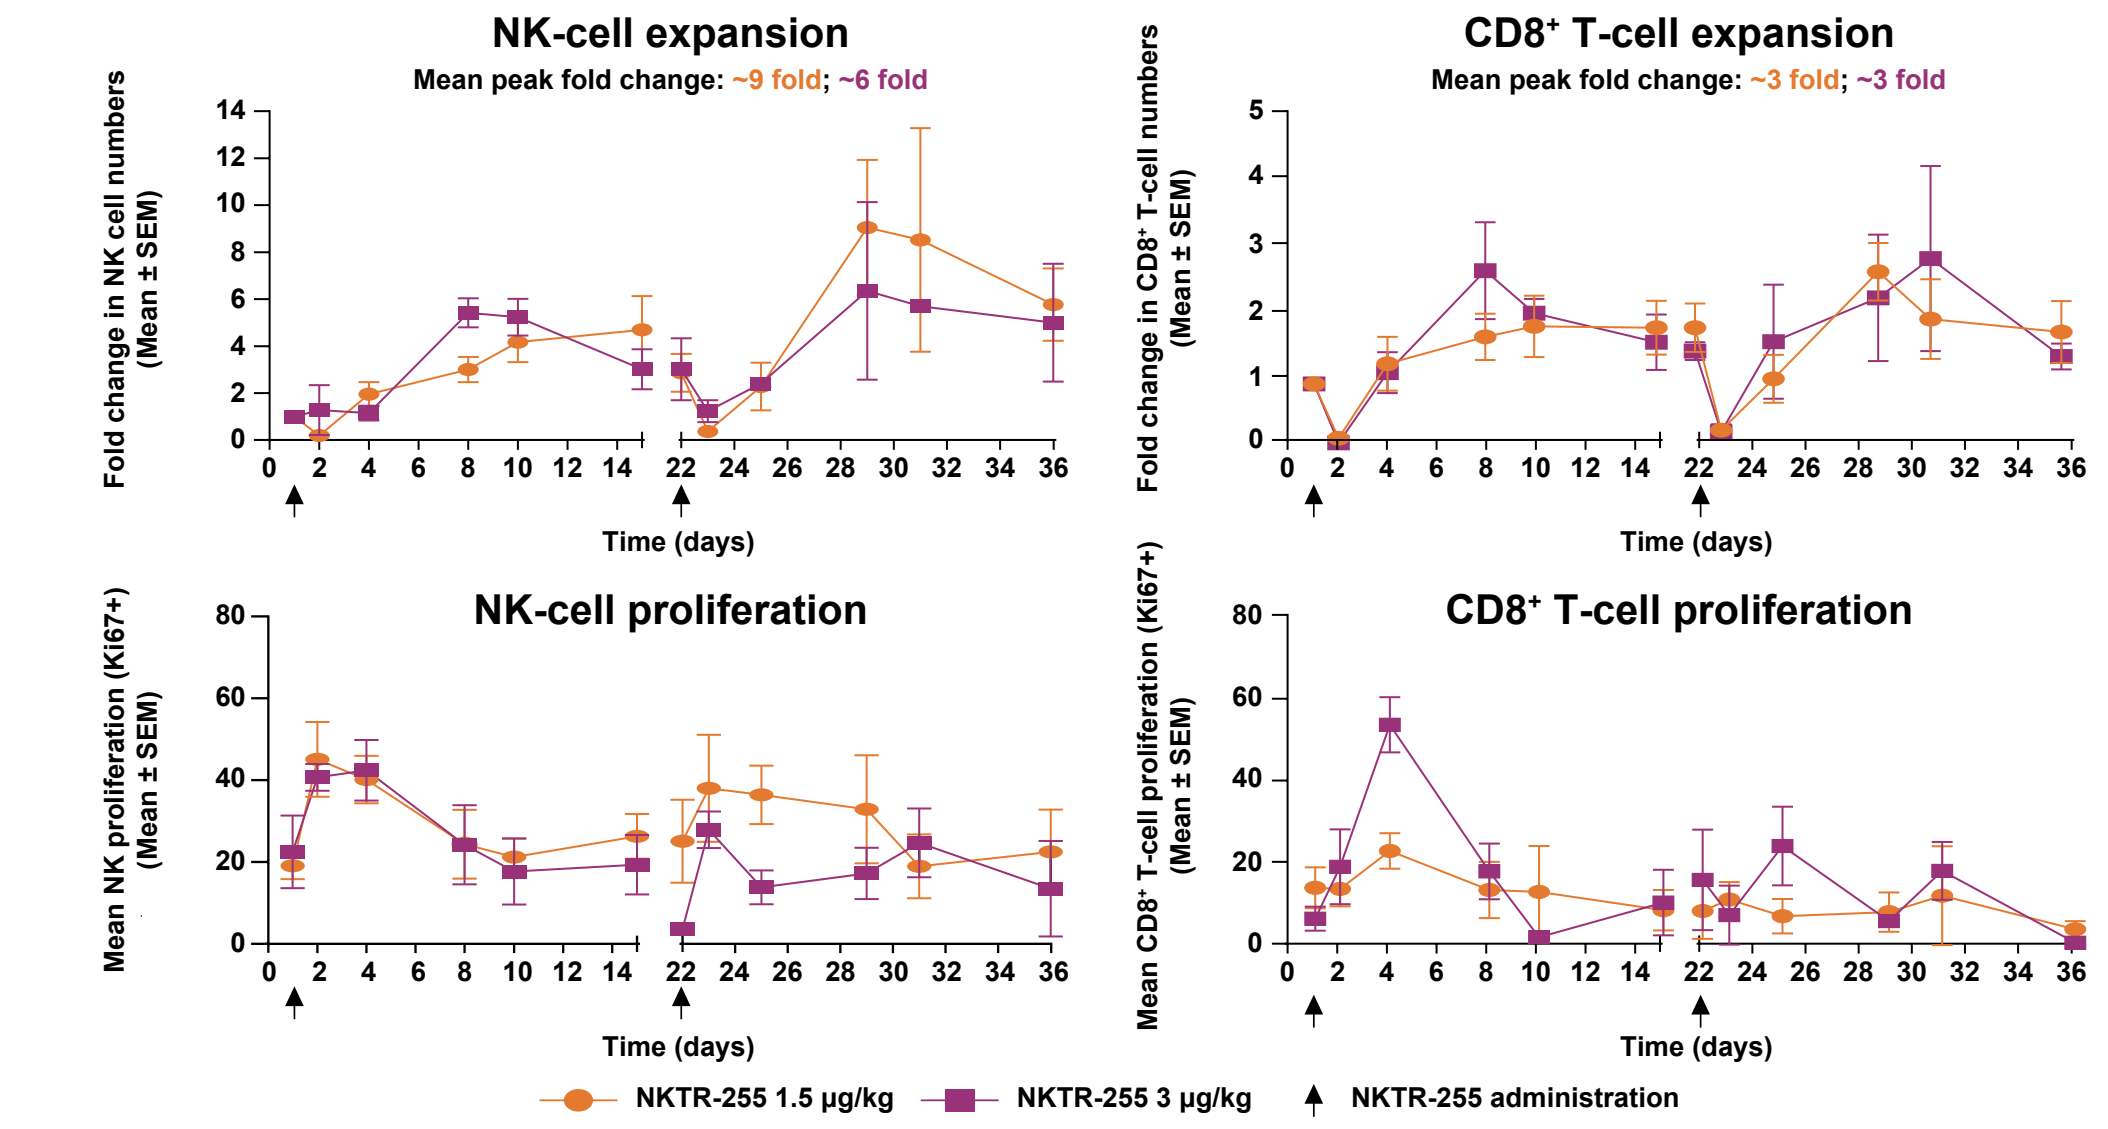
NK-cell expansion
Mean peak fold change: ~9 fold; ~6 fold
Fold change in NK cell numbers (Mean ± SEM)
CD8+ T-cell expansion
Mean peak fold change: ~3 fold; ~3 fold
Fold change in CD8+ T-cell numbers (Mean ± SEM)
NK-cell proliferation
Mean NK proliferation (Ki67+) (Mean ± SEM)
CD8+ T-cell proliferation
Mean CD8+ T-cell proliferation (Ki67+) (Mean ± SEM)
Time (days)
NKTR-255 1.5 μg/kg
NKTR-255 3 μg/kg
NKTR-255 administration

Cetuximab administered with loading dose on Day –7 (not shown), and then weekly starting Day 1. CD, cluster of differentiation; NK, natural killer; SEM, standard error of the mean.

4L CRC Patient Achieved a Confirmed PR (–51.5%) with NKTR-255 + Cetuximab

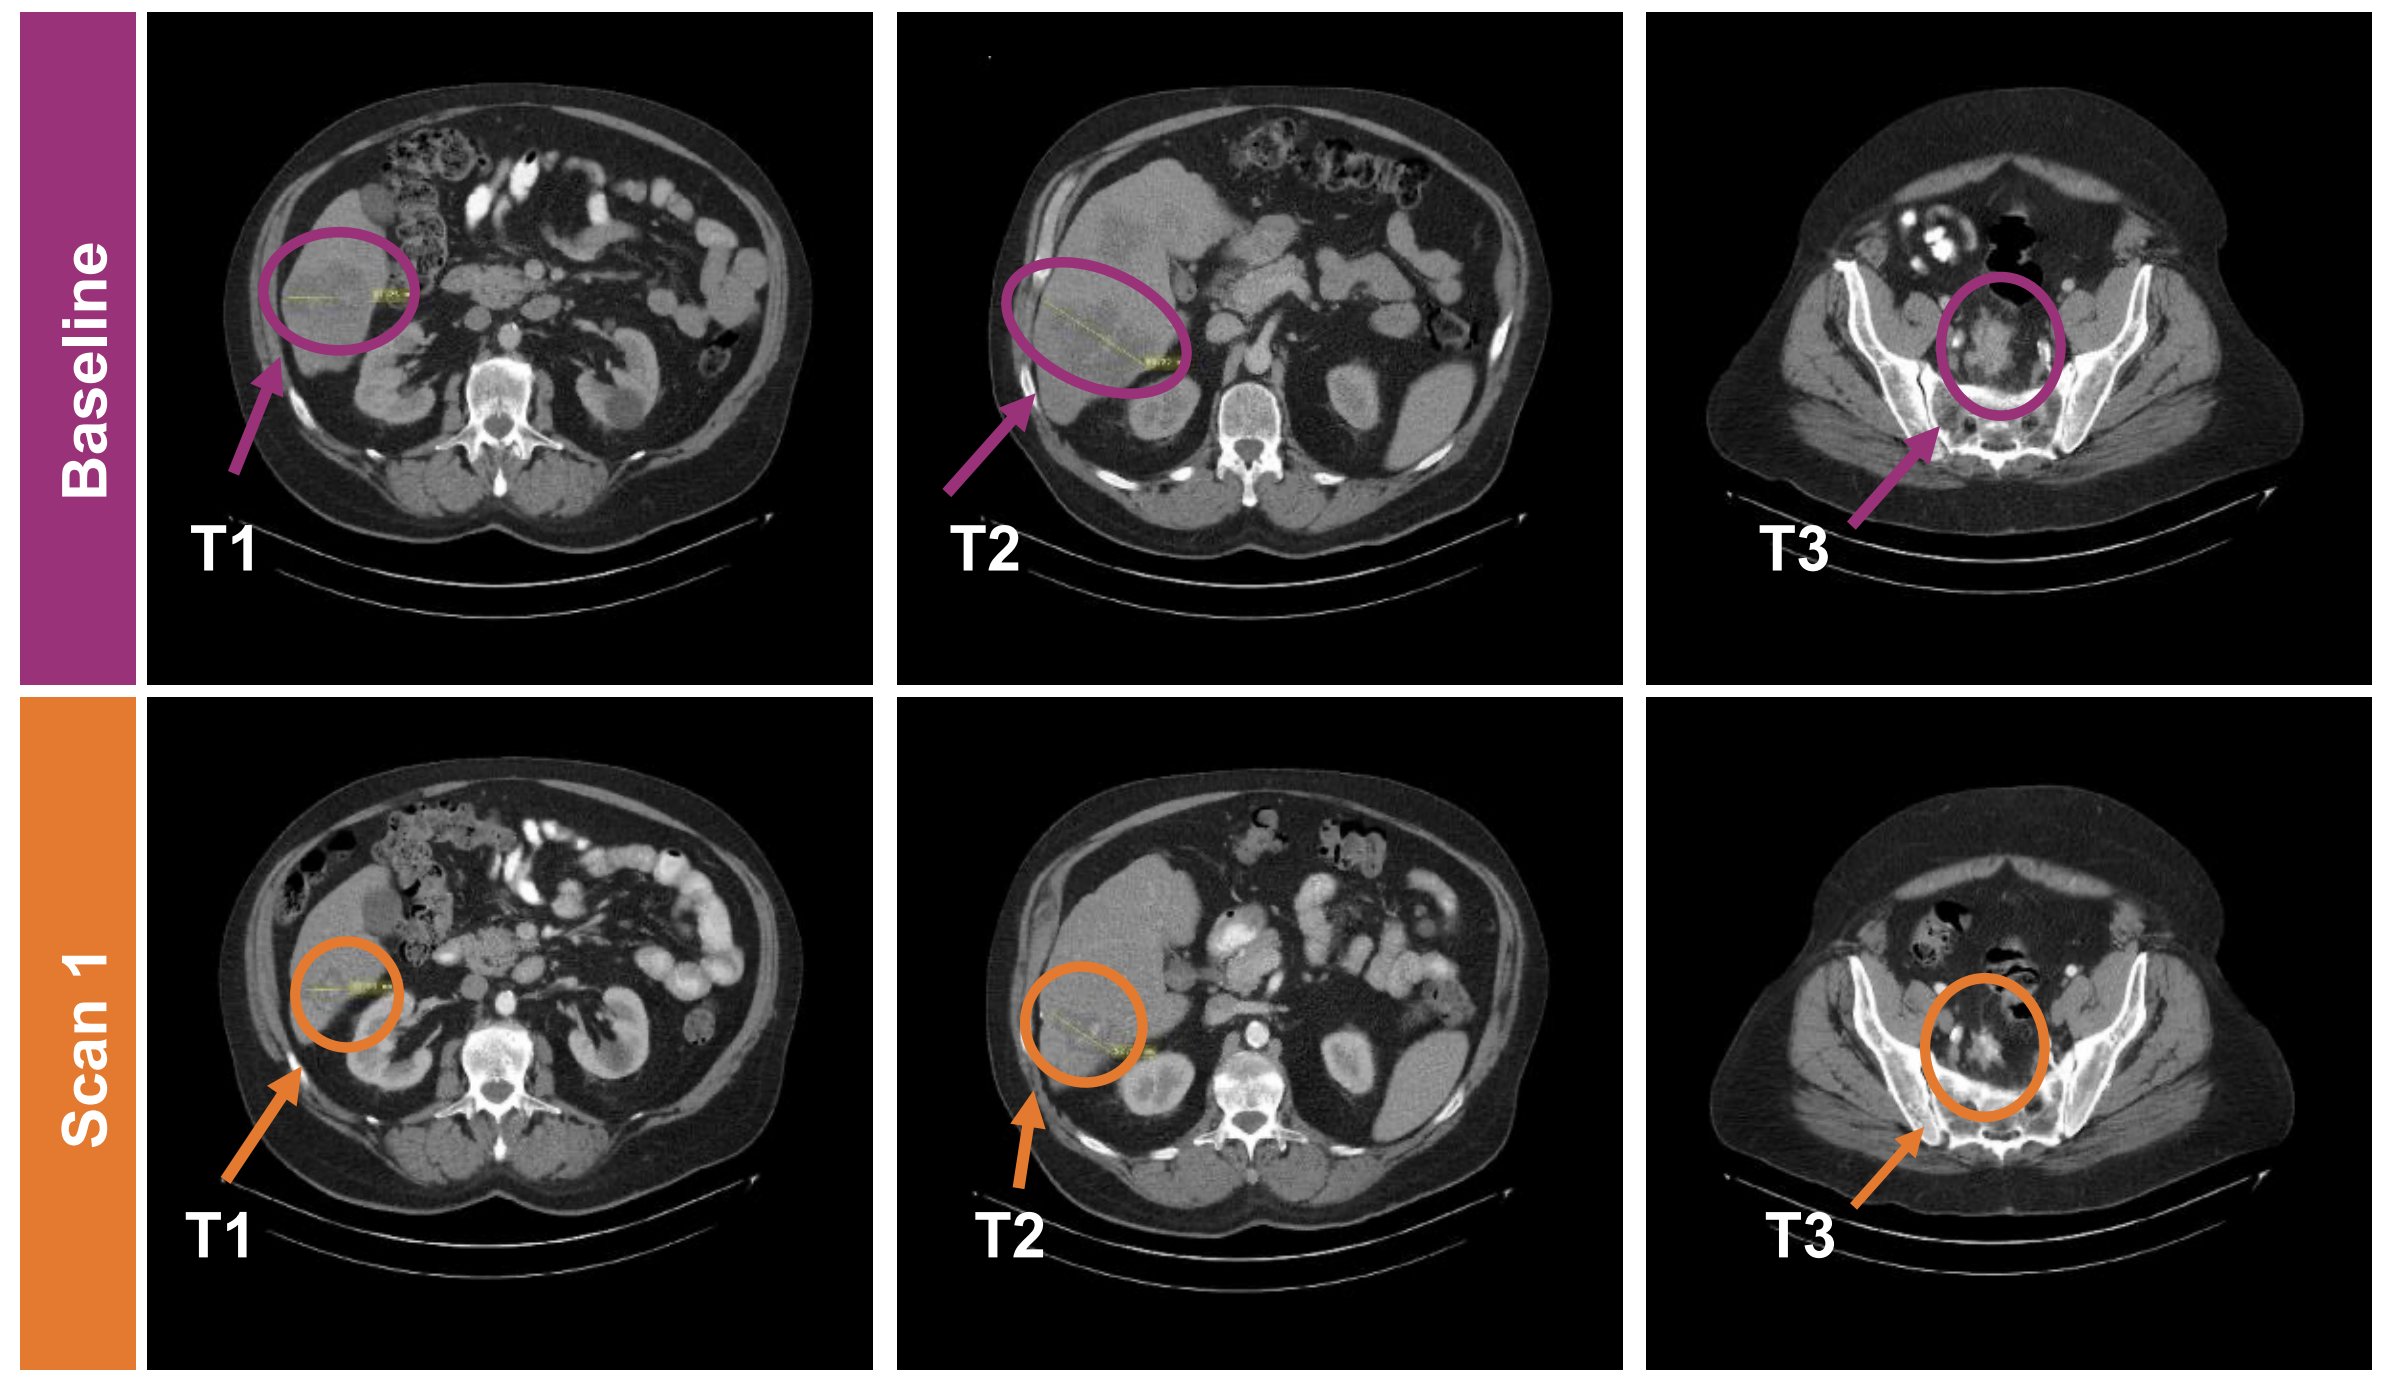
Baseline
Scan 1
T1
T2
T3

	Tumor assessment	Lesion	Baseline	Scan 1 (Week 9)	Scan 2 (Week 18)
Target lesions measurements, mm	T1 – Liver	Right dome liver	**61** x 51	**38** x 34	**22** x 14
	T2 – Liver	Interior right lobe liver	**86** x 69	**52** x 36	**38** x 22
	T3 – Pelvis	Pelvic mass	**47** x 42	**32** x 25	**34** x 23
Non-target lesions	NT01 – Lung	Lung nodule	Present	Present	Present
Sum of diameters, mm (% change from baseline)			**194**	**122 (–37.5)**	**94 (–51.5)**
Overall response (RECIST v1.1)				**PR**	**PR**

Prior treatment (best objective response): 1) FOLFOX + nivolumab + bevacizumab (PR, Mar 19 to Nov 19); 2. Aflibercept + FOLFIRI (SD, Feb 2 to Mar 20); 3. Regorafenib (SD, Mar 20 to Jun 20); 4. Trifluridine/tipiracil (progressive disease, Jul 20 to Oct 20). CRC, colorectal cancer; RECIST, Response Evaluation Criteria in Solid Tumors.

CONCLUSIONS

- **The combination of NKTR-255 + cetuximab was well tolerated at NKTR-255 doses of 1.5 and 3.0 μg/kg Q21D and TRAEs were generally low-grade, transient, and easily managed**
- **Early evidence of on-target biological activity was observed: NKTR-255 led to expansion and proliferation of NK and CD8+ T cells**
- **Early evidence of clinical activity was observed with 1 patient achieving a PR and 5 patients experiencing SD in this heavily pre-treated and highly refractory patient population**
- **The MTD/RP2D has not yet been reached and dose escalation of NKTR-255 + cetuximab in patients with R/R HNSCC and CRC is ongoing**

ACKNOWLEDGMENTS
We would like to thank all patients, their families, and the investigators who are participating in this study. This study is funded by Nektar Therapeutics, San Francisco, CA, USA. All authors contributed to and approved the poster. Medical writing assistance was provided by Suzanne Patel PhD and was funded by Nektar Therapeutics.

DISCLOSURES
M. Altan has served as an advisor to GlaxoSmithKline and Shattuck Labs, and has received institutional funding for research from Adaptimmune Therapeutics, Bristol Myers Squibb, Genentech, GlaxoSmithKline, Jounce Therapeutics, Lilly, Merck, Nektar Therapeutics, Novartis, and Shattuck Labs.

ABBREVIATIONS
ADCC, antibody-dependent cellular cytotoxicity; AE, adverse event; C, cetuximab; CAR-T, chimeric antigen receptor T-cell therapy; CD, cluster of differentiation; CPI, checkpoint inhibitor; CRC, colorectal cancer; CTCAE, Common Terminology Criteria for Adverse Events; CR, complete response; DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; FcR, Fc receptor; HNSCC, head and neck squamous cell carcinoma; IFN, interferon; IL, interleukin; IV, intravenous; mAb, monoclonal antibody; MCP-1, monocyte chemoattractant protein-1; MOA, mechanism of action; MTD, maximum tolerated dose, NK, natural killer; P, panitumumab; PD, pharmacodynamic; PD-1, programmed death-1; PK, pharmacokinetic; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; Q21D, every 21 days; rh, recombinant human; RP2D, recommended phase 2 dose; R/R, relapsed/refractory; SD, stable disease; TRAE, treatment-related AE; Tregs, regulatory T cells; SEM, standard error of the mean.

REFERENCES
1. Miyazaki T, et al. *J Immunother Cancer* 2021;9:e002024; 2. Kivimäe S, et al. *J Immunother Cancer* 2019;7(Suppl. 1):P619; 3. Altan M, et al. European Society for Medical Oncology Annual Meeting, September 16–21, 2021: Poster 1026TIP; 4. Conlon KC, et al. *J Clin Oncol* 2015;33:74–82.

Poster presented at the Society for Immunotherapy of Cancer (SITC) November 10–14, 2021. Corresponding author: Mehmet Altan (maltan@mdanderson.org)