Baseline Tumor Immune Signatures Associated with Response to Bempegaldesleukin (NKTR-214) and Nivolumab

Abstract 2623

Mike Hurwitz[1], Daniel Cho[2], Arjun Balar[2], Brendan Curti[3], Arlene Siefker-Radtke[4], Mario Sznol[1], Harriet Kluger[1], Chantale Bernatchez[4], Christie Fanton[5], Ernesto Iacucci[5], Yi Liu[5], Tuan Nguyen[5], Willem Overwijk[5], Jonathan Zalevsky[5], Mary Tagliaferri[5], Ute Hoch[5], Adi Diab[4]

[1]Yale Cancer Center, New Haven, CT; [2]NYU Langone Health, New York, NY; [3]Providence Cancer Institute, Portland, OR; [4]The University of Texas MD Anderson Cancer Center, Houston, TX; [5]Nektar Therapeutics, San Francisco, CA

BACKGROUND

Bempegaldesleukin Preferential Signaling Through the IL-2 Receptor Pathway

- Bempegaldesleukin (BEMPEG; NKTR-214) is a CD122-preferential IL-2 pathway agonist that has been shown to increase tumor-infiltrating lymphocytes, T cell clonality and PD-1 expression[1,2]
- BEMPEG combined with checkpoint inhibitor nivolumab (NIVO) has been shown to convert baseline tumors from PD-L1 negative (<1%) to PD-L1 positive (≥1%)[3-5]
- Low levels of baseline tumor-infiltrating lymphocytes (TILs)[6-8] and T cell–inflammation[9] is predictive of a poor response to checkpoint inhibitors (CPIs)

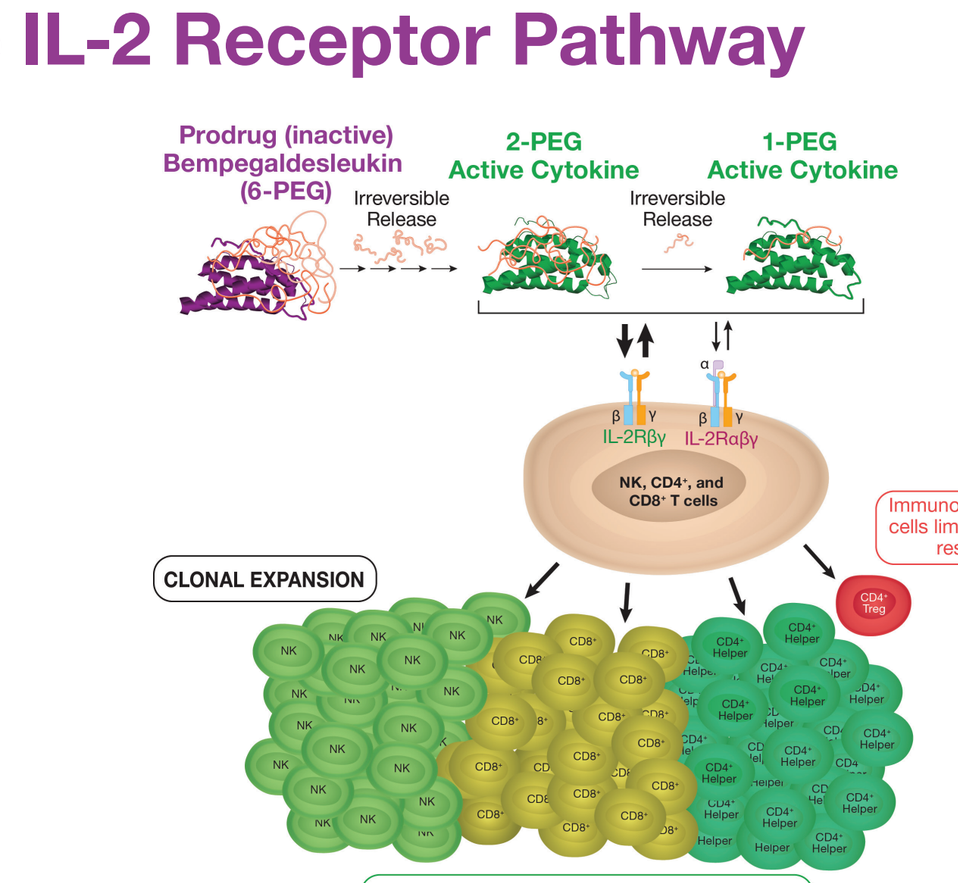
Prodrug (inactive) Bempegaldesleukin (6-PEG)
Irreversible Release
2-PEG Active Cytokine
Irreversible Release
1-PEG Active Cytokine
IL-2Rβγ
IL-2Rαβγ
NK, CD4+, and CD8+ T cells
Immunosuppressive cells limit anti-tumor response
CLONAL EXPANSION
Stimulates Immune Response to Kill Tumor Cells

Rapid Activation of the Immune System was Observed with BEMPEG and NIVO

Increase in Lymphocytes with Every Treatment Cycle*

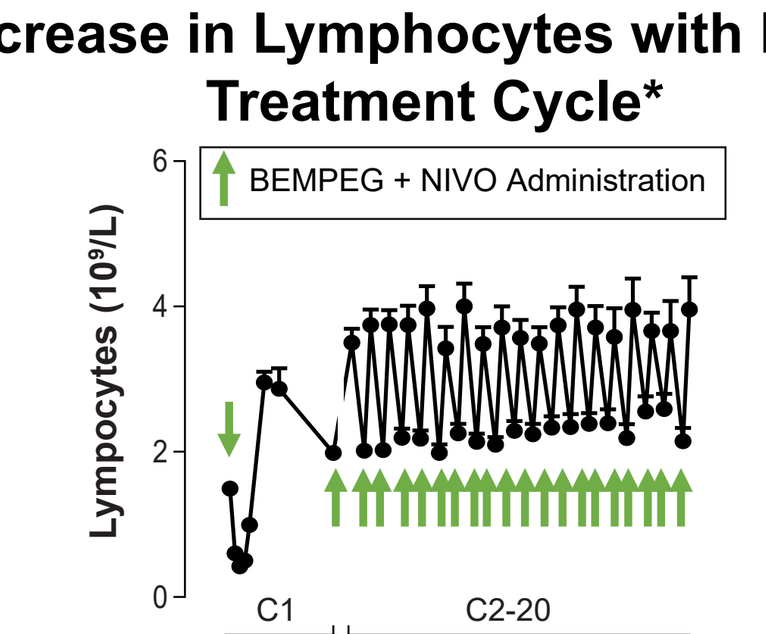
BEMPEG + NIVO Administration
Lymphocytes (10^9/L)
C1
C2-20
Time (Days)

Lymphocyte effects of the BEMPEG + NIVO combination are driven by BEMPEG, as a similar pattern is observed with monotherapy[2]

*Lymphocyte levels were obtained from standard hematology analysis. All efficacy evaluable melanoma (n=38) and mUC (n=27) in the BEMPEG plus NIVO combination enrolled in PIVOT-02 (n=65, Mean+SD) were included in the analyses.

On-Treatment Increase in TIL and PD-L1

Change in CD8 Infiltrate in MEL[3,^]

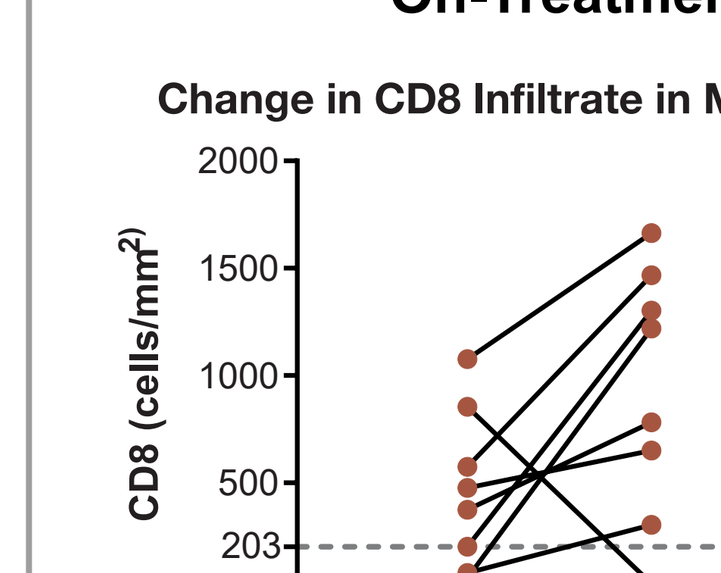
CD8 (cells/mm²)
Baseline
Week 3

PD-L1 Conversion in UC[4,#]

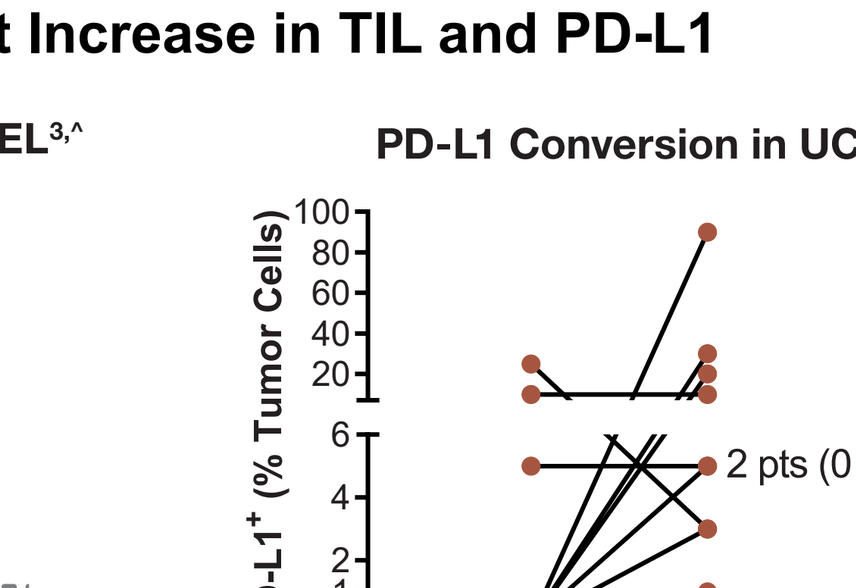
PD-L1+ (% Tumor Cells)
2 pts (0→5)
3 pts (0→0)
Baseline
Week 3

^IHC for CD8 was obtained by standard methods. All patients with first-line melanoma (1L MEL) with matched Baseline and Week 3 biopsy (n=8) were included in the analysis.

#All patients with 1L urothelial carcinoma (UC) with matched Baseline and Week 3 biopsy (n=13) at time of data cut were included and assessed for PD-L1 expression (28-8 PharmDx).

PIVOT-02 Study Schema

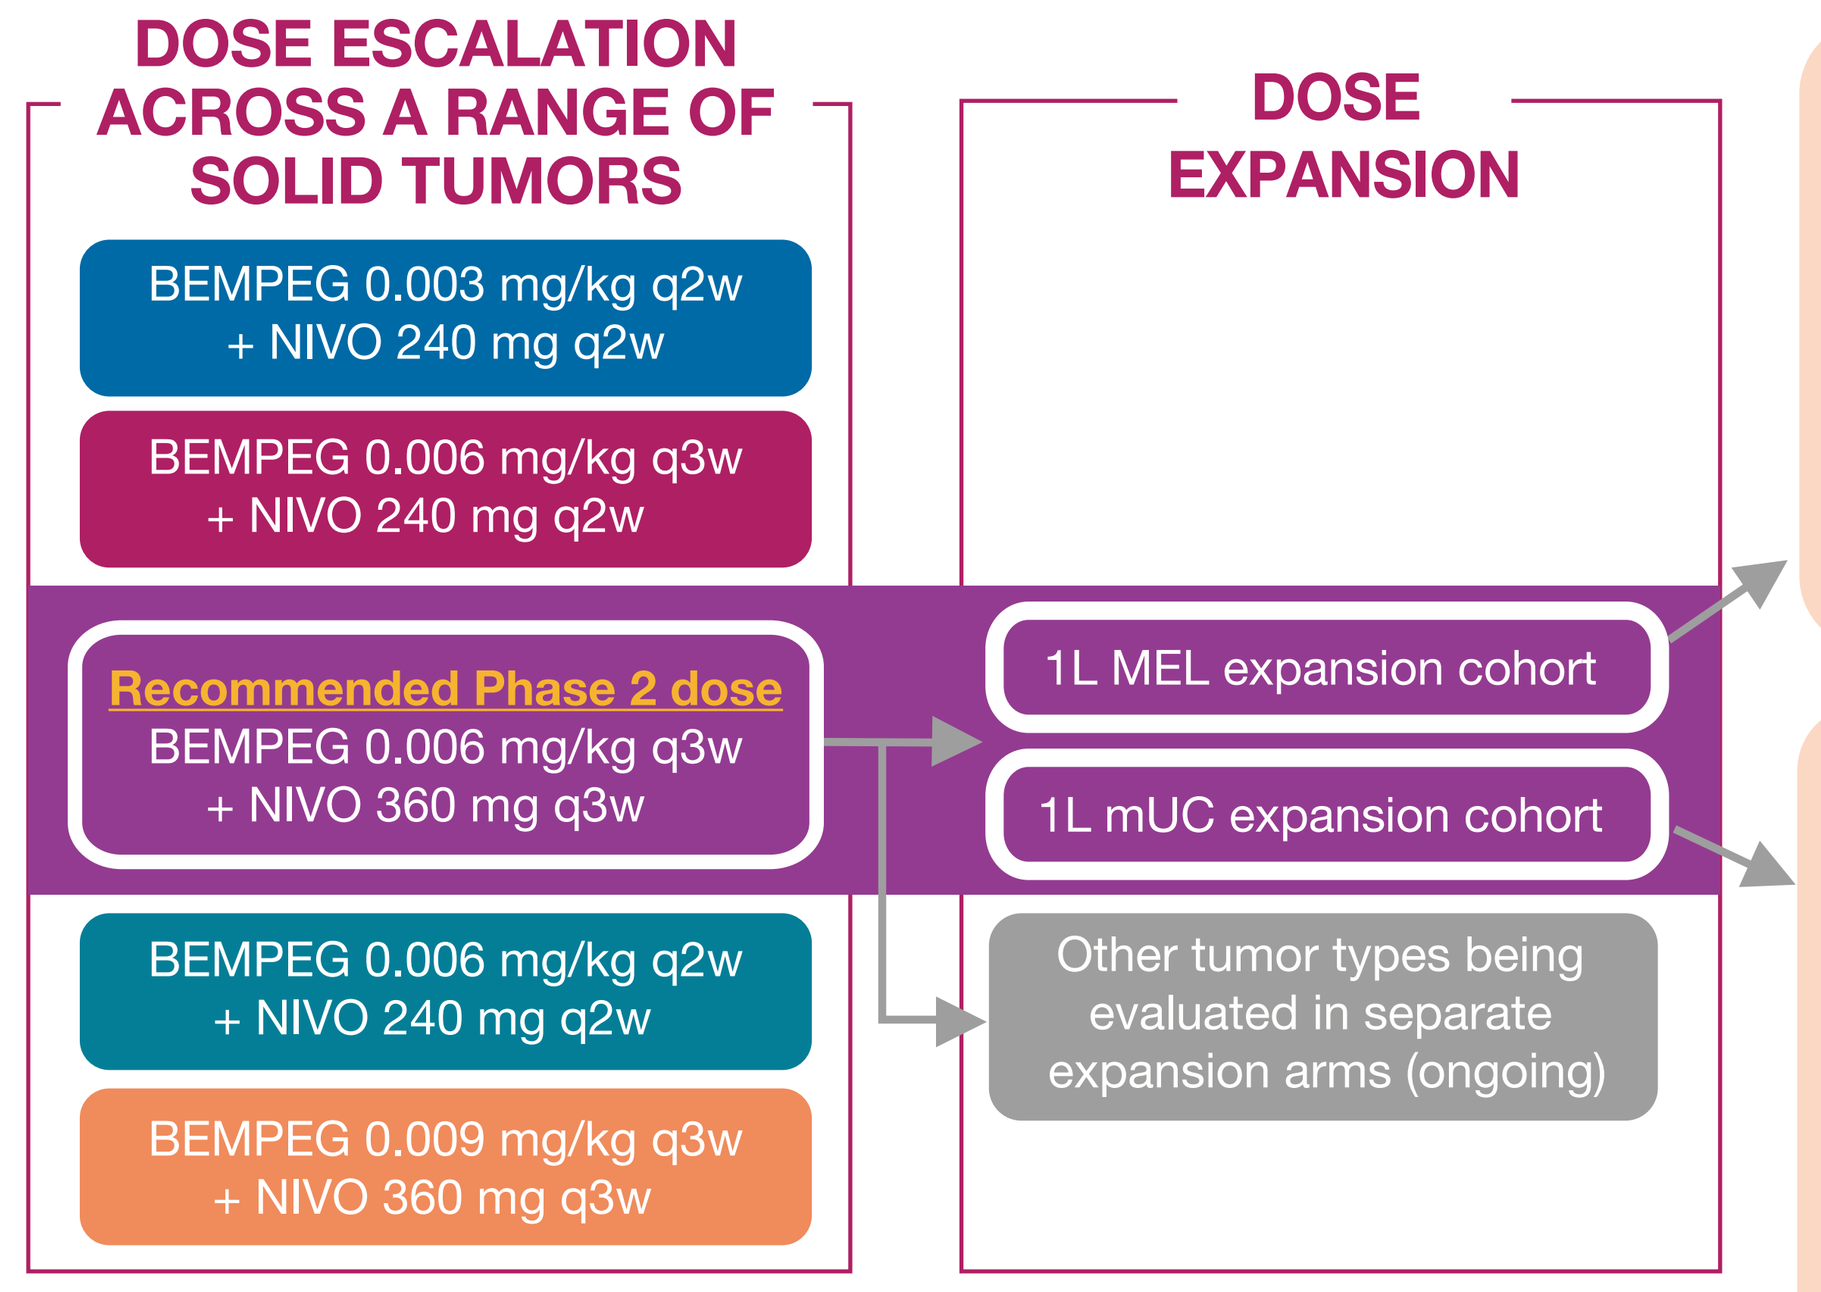
DOSE ESCALATION ACROSS A RANGE OF SOLID TUMORS
BEMPEG 0.003 mg/kg q2w + NIVO 240 mg q2w
BEMPEG 0.006 mg/kg q3w + NIVO 240 mg q2w
Recommended Phase 2 dose
BEMPEG 0.006 mg/kg q3w + NIVO 360 mg q3w
BEMPEG 0.006 mg/kg q2w + NIVO 240 mg q2w
BEMPEG 0.009 mg/kg q3w + NIVO 360 mg q3w
DOSE EXPANSION
1L MEL expansion cohort
1L mUC expansion cohort
Other tumor types being evaluated in separate expansion arms (ongoing)

Key MEL Inclusion Criteria

- 1L Metastatic Melanoma (with known BRAF status)
- Measurable disease per RECIST v1.1
- ECOG 0-1
- 41 MEL patients enrolled and received at least one dose of BEMPEG + NIVO
- As of March 29, 2019, 38 patients were efficacy evaluable defined as patients with ≥1 post-baseline scan (3 patients discontinued prior to first scan due to an unrelated TEAE [n=1] and patient decision [n=2])

Key mUC Inclusion Criteria

- Unresectable locally advanced or metastatic disease
- Cisplatin-ineligible
- Cisplatin-eligible who refused SOC
- ECOG 0-1
- 41 patients with mUC enrolled and received at least one dose of BEMPEG + NIVO
- As of December 3, 2018, 27 patients were efficacy evaluable defined as ≥1 post-baseline scan (26/27 stage IV mUC): (1 patient was excluded for non-eligibility (no target lesion), and 3 patients discontinued prior to first scan [1 due to patient decision, 1 due to clinical progression; 1 due to death from disease], 10 patients pending first scan in database)

Baseline tumors and gene expression were assessed by IHC (28-8 PharmDx) and Nanostring PanCancer Panel

ECOG PS: Eastern Cooperative Oncology Group performance score; MEL: melanoma; mUC: metastatic urothelial carcinoma; RECIST: response evaluation criteria in solid tumors; SOC: standard of care

- PIVOT-02 is a multicenter, Phase 1/2 study evaluating BEMPEG plus NIVO and includes a cohort of patients with metastatic melanoma, and patients with locally advanced or metastatic urothelial carcinoma (mUC) who are cisplatin-ineligible or cisplatin-eligible who have refused standard of care
- PIVOT-02 recently reported preliminary clinical and safety data for melanoma[3] and mUC[4]
 - BEMPEG plus NIVO in mUC was well tolerated and demonstrated promising clinical benefit
 - ORR of 48% in efficacy-evaluable population; in cisplatin-ineligible, 44%; ORR in refused SOC, 55%
 - Therapy demonstrated deep responses with CR rate of 19% (median 78% tumor shrinkage among responders)
 - No relapses observed among responders
 - Responses were observed regardless of baseline PD-L1 expression; ORR in PD-L1 positive patients was 50% and ORR in PD-L1 negative was 45%
- **Here, we report the baseline tumor immune signatures associated with response to BEMPEG plus NIVO in 1L Stage IV melanoma (data cut-off, March 29, 2019) and 1L mUC (data cut-off, December 3, 2018) cohorts, as well as updated response data for melanoma**

RESULTS

Stage IV 1L Melanoma Cohort at RP2D: Best Overall Response by Independent Radiology

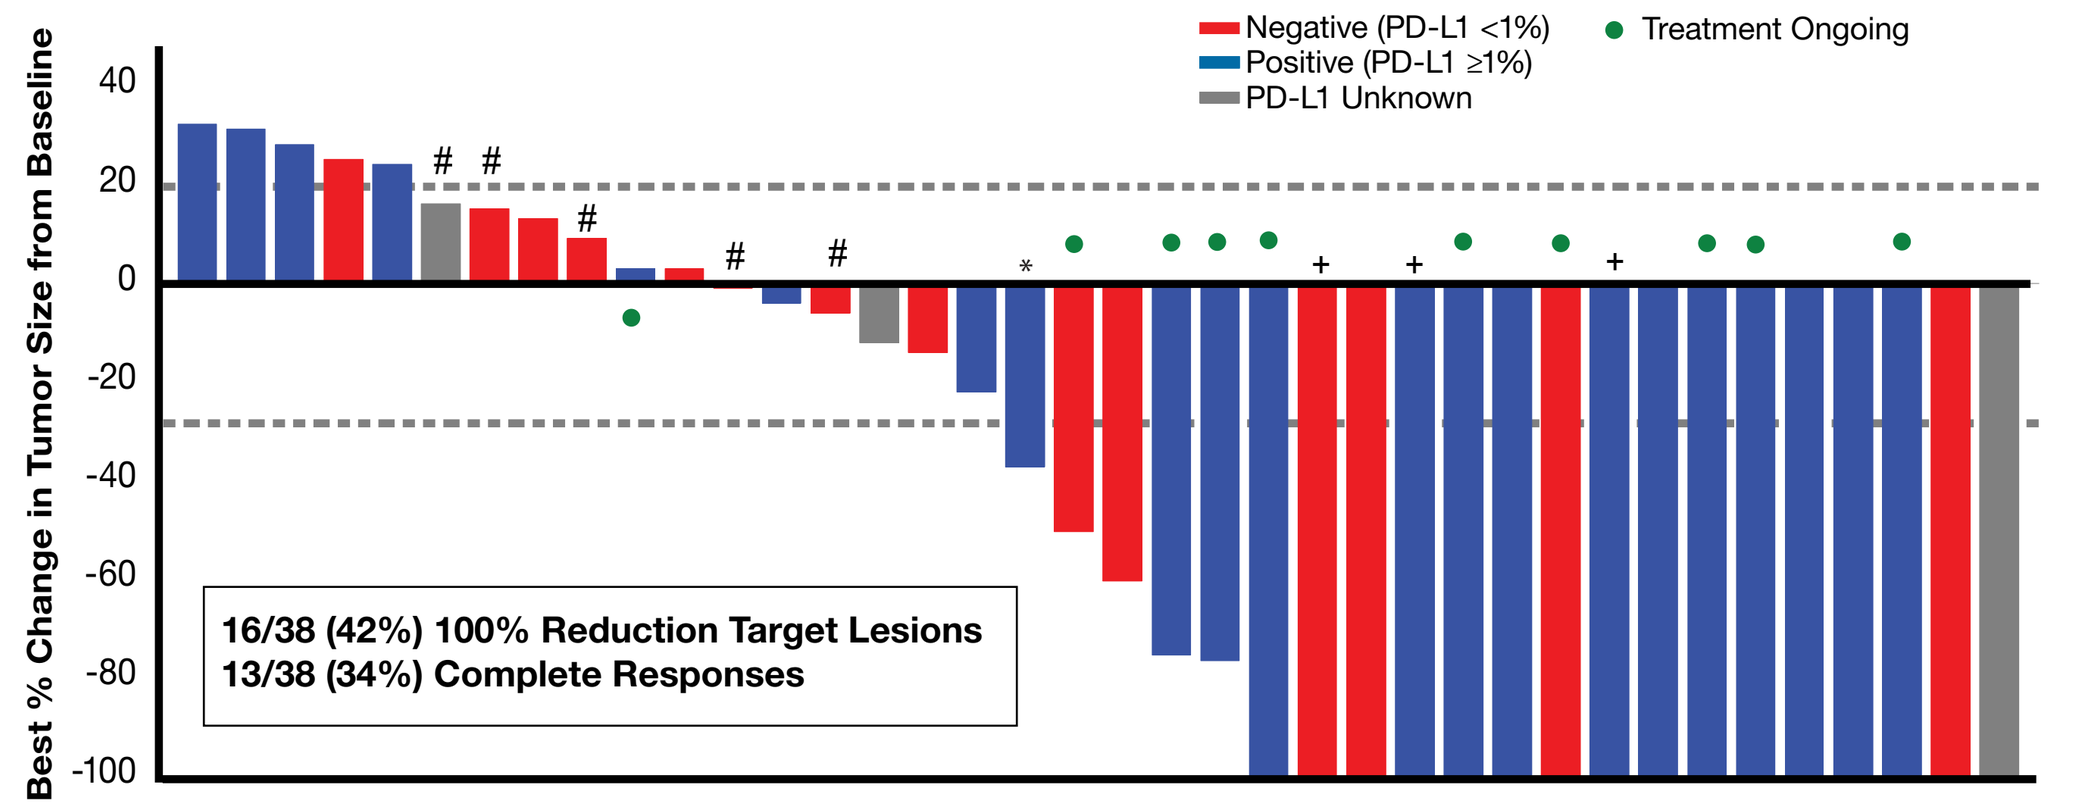
Negative (PD-L1 <1%)
Positive (PD-L1 ≥1%)
PD-L1 Unknown
Treatment Ongoing
Best % Change in Tumor Size from Baseline
16/38 (42%) 100% Reduction Target Lesions
13/38 (34%) Complete Responses

1L Melanoma (n=38 Efficacy Evaluable**)	Overall Response Rate
Confirmed ORR (CR+PR)	20 (53%)
CR	13 (34%)
DCR (CR+PR+SD)	28 (74%)
PD-L1 negative (n=14)	6 (43%)
PD-L1 positive (n=21)	13 (62%)
PD-L1 unknown (n=3)	1 (33%)
LDH > ULN (n=11)	5 (45%)
Liver metastases (n=10)	5 (50%)

#Best overall response is PD due to non-target lesion progression or presence of new lesion. *Best overall response is SD. +Best overall response is PR. CR for target lesion(s). Non-target lesion(s) still present.

**Efficacy evaluable population includes patients who have measurable disease (per RECIST 1.1) at baseline and also have at least one post-baseline tumor assessment.

ITT = 41: 3 patients are excluded because they are not response evaluable:1 patient discontinued treatment after 1 dose due to unrelated adverse event (MI); 1 patient discontinued treatment after 1 dose due to patient decision; 1 patient discontinued treatment after 3 doses due to patient decision.

Stage IV 1L Melanoma Cohort at RP2D: CR Rate 34% (13/38) with 12.7 Months Median Follow-up by Independent Radiology

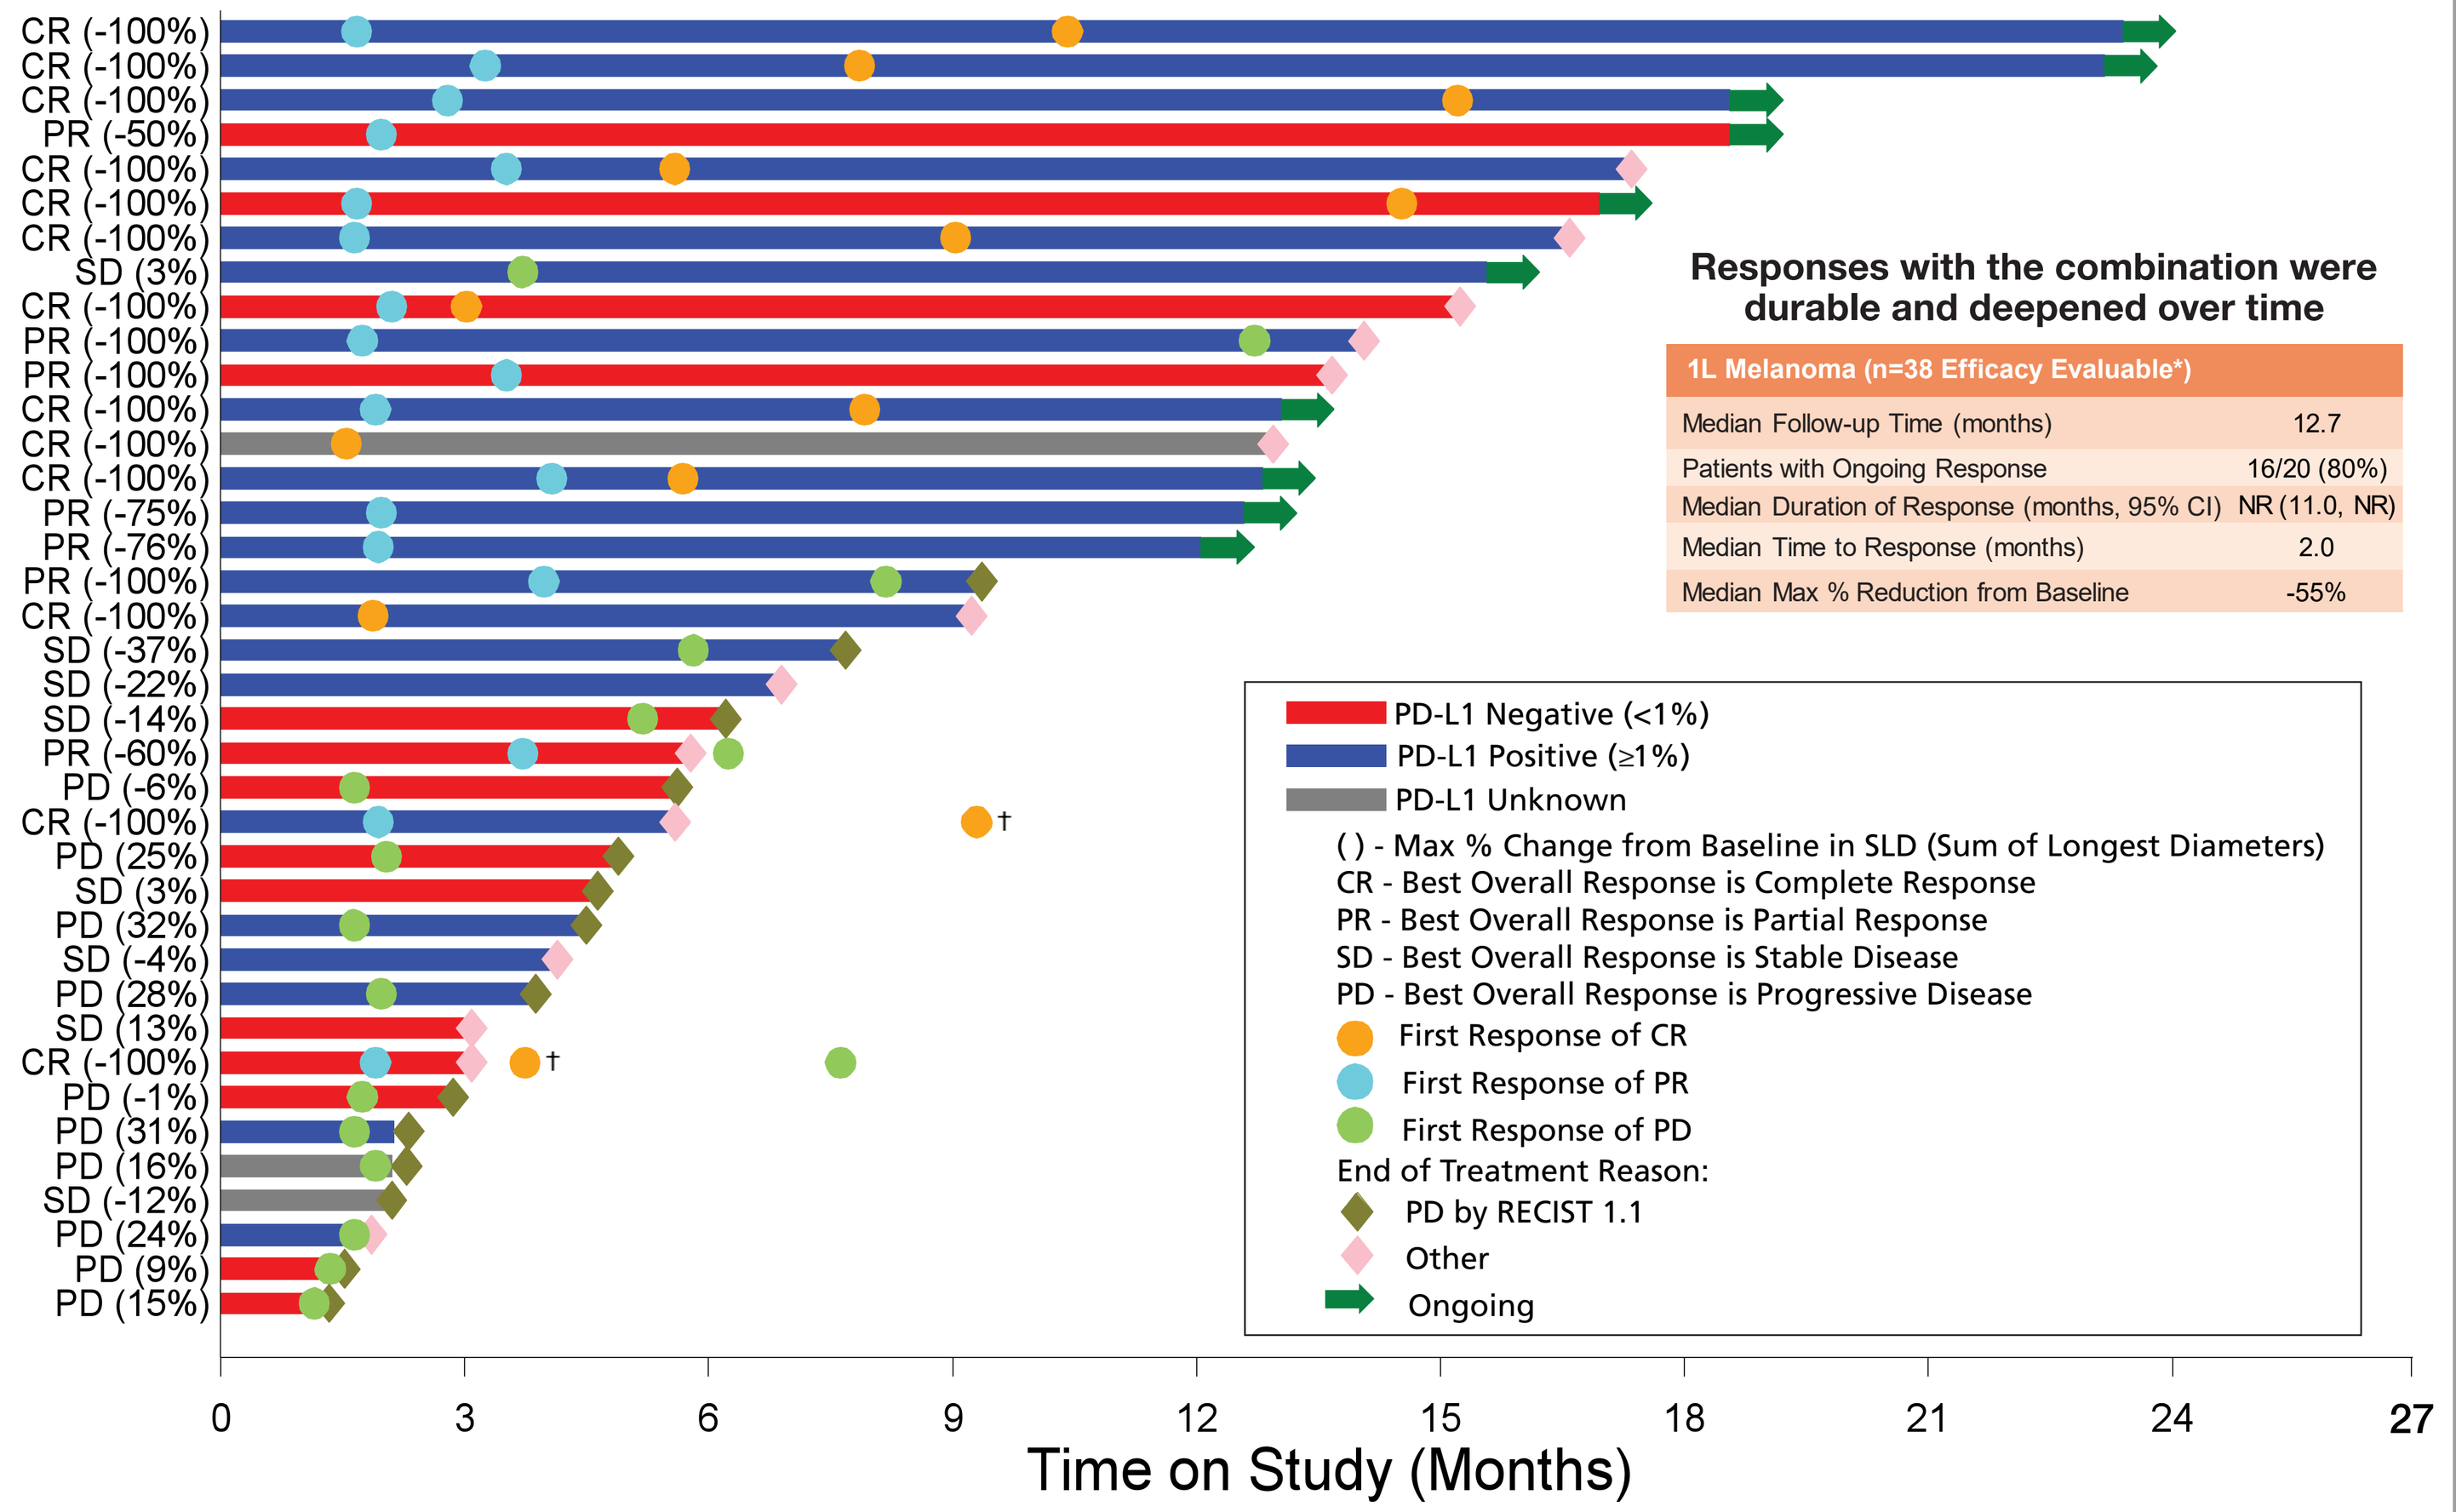
CR (-100%)
CR (-100%)
CR (-100%)
PR (-50%)
CR (-100%)
CR (-100%)
CR (-100%)
SD (3%)
CR (-100%)
PR (-100%)
PR (-100%)
CR (-100%)
CR (-100%)
CR (-100%)
PR (-75%)
PR (-76%)
PR (-100%)
CR (-100%)
SD (-37%)
SD (-22%)
SD (-14%)
PR (-60%)
PD (-6%)
CR (-100%)
PD (25%)
SD (3%)
PD (32%)
SD (-4%)
PD (28%)
SD (13%)
CR (-100%)
PD (-1%)
PD (31%)
PD (16%)
SD (-12%)
PD (24%)
PD (9%)
PD (15%)
PD-L1 Negative (<1%)
PD-L1 Positive (≥1%)
PD-L1 Unknown
() - Max % Change from Baseline in SLD (Sum of Longest Diameters)
CR - Best Overall Response is Complete Response
PR - Best Overall Response is Partial Response
SD - Best Overall Response is Stable Disease
PD - Best Overall Response is Progressive Disease
First Response of CR
First Response of PR
First Response of PD
End of Treatment Reason:
PD by RECIST 1.1
Other
Ongoing
Time on Study (Months)

Responses with the combination were durable and deepened over time

1L Melanoma (n=38 Efficacy Evaluable*)	
Median Follow-up Time (months)	12.7
Patients with Ongoing Response	16/20 (80%)
Median Duration of Response (months, 95% CI)	NR (11.0, NR)
Median Time to Response (months)	2.0
Median Max % Reduction from Baseline	-55%

*Efficacy evaluable population includes patients who have measurable disease (per RECIST 1.1) at baseline and also have at least one post-baseline tumor assessment. ITT = 41: 3 patients are excluded because they are not response evaluable: 1 patient discontinued treatment after 1 dose due to unrelated adverse event (MI); 1 patient discontinued treatment after 1 dose due to patient decision; 1 patient discontinued treatment after 3 doses due to patient decision. †CRs noted beyond discontinuation of treatment were confirmed for patients who had no intervening therapy.

Stage IV IO-Naïve 1L Melanoma Treatment-Related Adverse Events (AEs) at RP2D

Preferred Term[1]	Total (N=41)[a]
All Treatment-Related Grade 3-4	**6 (14.6%)[b]**
Atrial fibrillation* (Grade 3)	2 (4.9%)
Hyperglycemia (Grade 4)	1 (2.4%)
Acute kidney injury, Blood creatinine increased, Dyspnea, Hypernatremia, Hypoxia (all Grade 3)	1 each (2.4%)
Treatment-Related Grade 1-2 in (>30% listed below)	**39 (95.1%)**
Flu-like symptoms**	33 (80.5%)
Rash***	29 (70.7%)
Fatigue	27 (65.9%)
Pruritus	20 (48.8%)
Nausea	17 (41.5%)
Arthralgia	15 (36.6%)
Myalgia	13 (31.7%)
Any imAE (Grade ≥3)	**2 (4.9%)**
Patients who discontinued due to a TRAE (blood creatinine increased, cerebrovascular accident, hyperglycemia, pharyngitis)	**4 (9.8%)**

The combination of BEMPEG plus NIVO is well tolerated, and treatment-related adverse events (TRAEs) are similar to what was previously reported at SITC 2018[3]

[a]N = 41, safety population defined as patients with ≥ 1 dose of study treatment. [1] Patients are only counted once under each preferred term using highest grade.

[b]3 patients with previously reported Gr3s were re-categorized by investigator and these changes are reflected in the March 29 data cut.

*1 patient with previous history of atrial fibrillation since 2015; 1 patient experienced atrial fibrillation 1 month after last dose of study drug.**Flu-like symptoms included the following MedDRA PTs: Chills, Influenza, Influenza-like Illness, Pyrexia. ***Rash included the following MedDRA PTs: Erythema, Rash, Rash erythematous, Rash generalized, Rash macular, Rash maculo-papular, Rash maculovesicular, Rash papular, Rash pruritic, Rash pustular, Rash vesicular, Exfoliative rash.

Baseline Biomarkers and Scoring Methods

Baseline biomarker, scoring system, and number of patients with evaluable biomarkers in efficacy evaluable 1L MEL and 1L UC patients

Baseline Biomarker^	Experimental Method	Unit	Scoring System	1L MEL (N=38)* N (%)	1L UC (N=27)* N (%)
IFNg+	Gene Expression**	– log10 (p-value)	< or ≥ Median	21 (55%)	Not Available
PD-L1+ (28-8 PharmDx, Tumor)	IHC	%	<1% (neg) or ≥1% (pos)	35 (92%)	23 (85%)
PD-L1 CPS (28-8)	IHC	%	<10% (neg) or ≥10% (pos)	Not Done	23 (85%)
CD3+ TIL	IHC	%	< or ≥ Median	28 (74%)	22 (81%)
CD8+TIL	IHC	Cells/mm²	< or ≥ Median	29 (76%)	24 (89%)
PD1+CD8+TIL	IHC	%	< or ≥ Median	25 (66%)	21 (78%)
Ki67+CD8+TIL	IHC	%	< or ≥ Median	27 (71%)	23 (85%)
CD68+Cells	IHC	%	< or ≥ Median	28 (74%)	22 (81%)
PD-L1+CD68+Cells	IHC	%	< or ≥ Median	28 (74%)	21 (78%)
PD1+Cells	IHC	%	< or ≥ Median	26 (68%)	22 (81%)

*Efficacy evaluable population. **Gene expression (Nanostring PanCancer Panel) data were used to obtain an IFNg score. The IFNg score is based on the statistical significance of the ranking of the genes found in the signature (CD3D, IDO1,CCL5, CD2, CXCL13, IL2RG, HLA-E, CXCR6, LAG3, CXCL10, STAT1, GZMB, CXCL9, IFNg and PRF1) compared to a uniform distribution.[10] ^Additional baseline biomarkers: total Ki67+ cells, PD-L1+CD3+TIL, and total FoxP3 + cells were measured but showed negligible ORR difference, data not shown.

Response rates, baseline demographics and prognostic factors in unselected biomarker populations are similar compared to efficacy evaluable population

In 1L Melanoma, Univariate Analyses Show Enrichment Based Upon Multiple Biomarkers, Most Strongly Baseline IFNg and TIL Scores

Baseline Biomarker	Median Cutoff	< Median[a] # CR+PR/ Total	< Median[a] ORR[b] (%)	≥ Median # CR+PR/ Total	≥ Median ORR (%)
IFNg	1.2	2/10	20	9/11	81.8
PD-L1	1	6/14	42.9	13/21	61.9
CD3+TIL	18.9	4/14	28.6	11/14	78.6
CD8+TIL	203	4/14	28.6	12/15	80.0
PD1+CD8+TIL	4.7	4/12	33.3	10/13	76.9
Ki67+CD8+TIL	31.5	6/13	46.2	9/14	64.3
CD68+Cells	7.0	6/14	42.9	9/14	64.3
PD-L1+CD68+Cells	29.1	6/14	42.9	9/14	64.3
PD1+Cells	3.2	5/13	38.5	9/13	69.2

Difference in ORR (95% CI)[c]

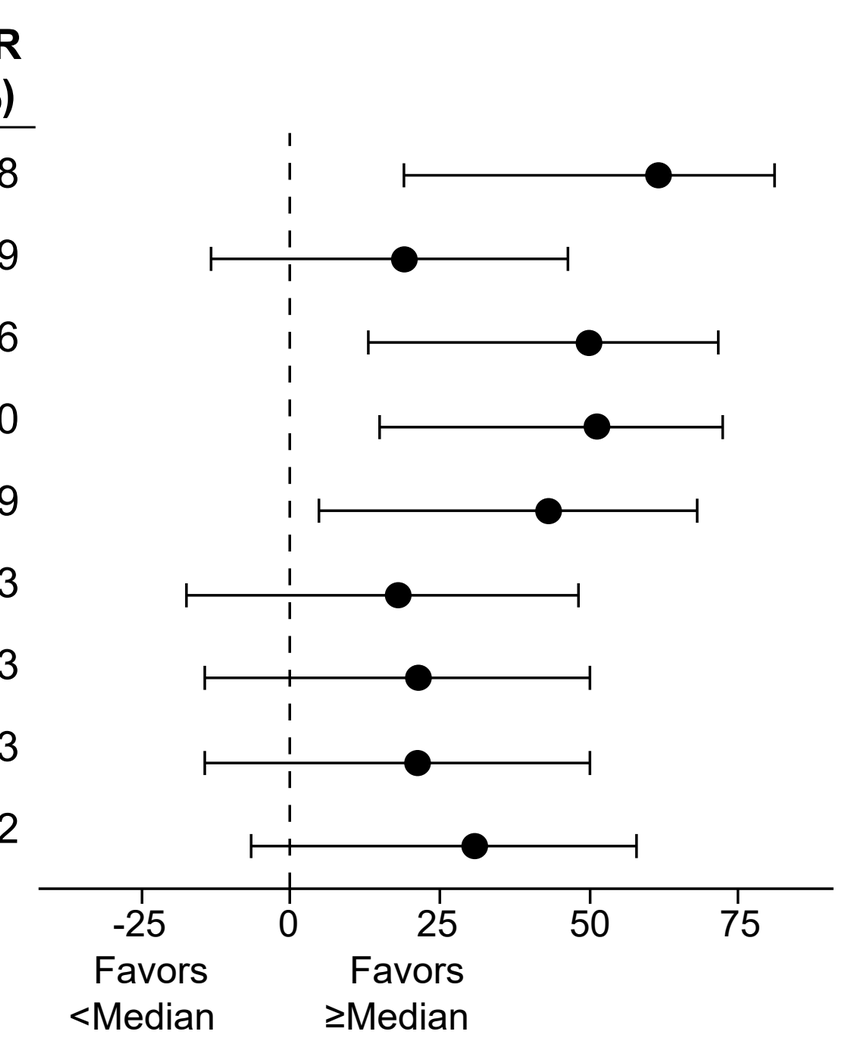
-25
0
25
50
75
Favors <Median
Favors ≥Median

[a]Except for PD-L1, which scored by negative vs positive

[b]Best response (RECIST 1.1) by independent central review

[c]95% confidence interval for risk difference is based on Wilson method

In mUC, Unlike Single-Agent CPIs, Favorable ORRs Observed Regardless of Baseline PD-L1 Status

Baseline Biomarker	Median Cutoff	< Median[a] # CR+PR/ Total	< Median[a] ORR[b] (%)	≥ Median # CR+PR/ Total	≥ Median ORR (%)
PD-L1	1	5/11	45.5	6/12	50.0
PD-L1 CPS	10	6/14	42.9	5/9	55.6
CD3+TIL	15.5	5/11	45.5	5/11	45.5
CD8+TIL	70.5	5/12	41.7	6/12	50.0
PD1+CD8+TIL	0	4/11	36.4	5/10	50.0
Ki67+CD8+TIL	23.5	5/11	45.5	6/12	50.0
CD68+Cells	7.5	4/11	36.4	6/11	54.5
PD-L1+CD68+Cells	1.2	3/10	30.0	7/11	63.6
PD1+Cells	0.7	5/11	45.5	5/11	45.5

Difference in ORR (95% CI)[c]

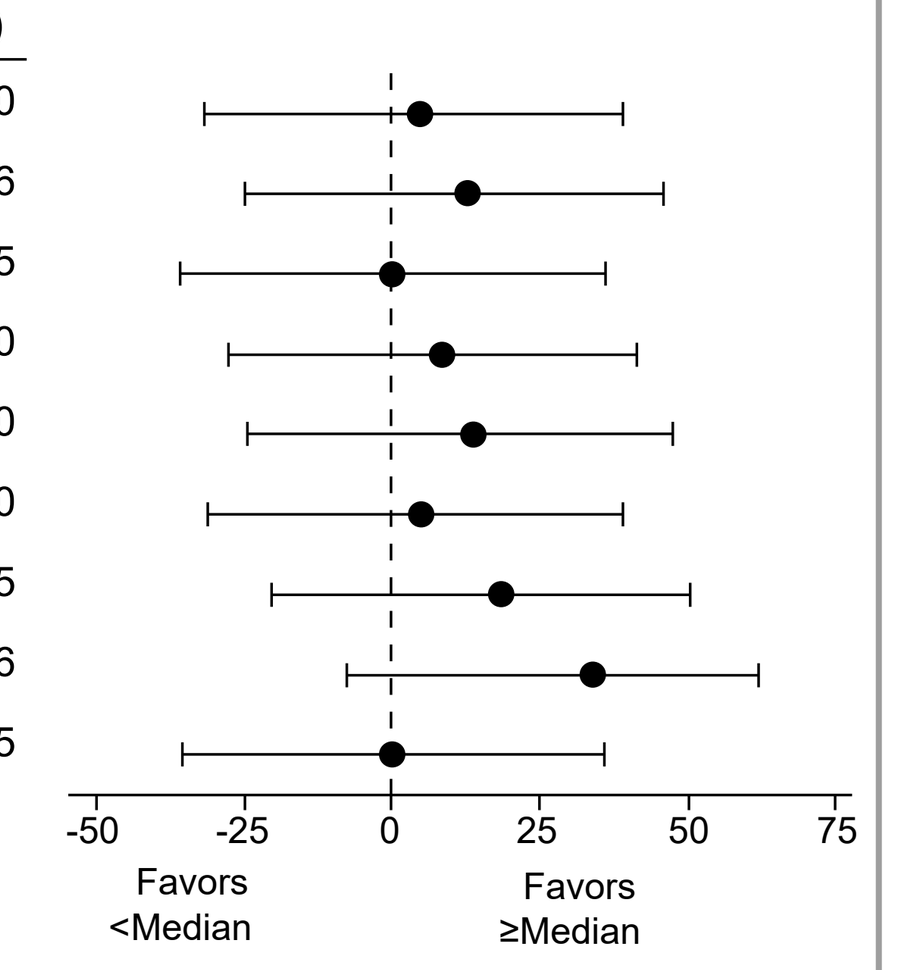
-50
-25
0
25
50
75
Favors <Median
Favors ≥Median

Response rate maintained in PD-L1 neg and CD8-TIL low (50%, 4/8) or CD3-TIL low (43%, 3/7)

[a]Except for PD-L1, which scored by negative vs positive

[b]Best response (RECIST 1.1) by independent central review

[c]95% confidence interval for risk difference is based on Wilson method

In 1L Melanoma, Paired Analyses Show Encouraging Response Rate in Patients with Favorable and Unfavorable Tumor Microenvironment (TME)*

CD8-TIL vs %PD-L1+

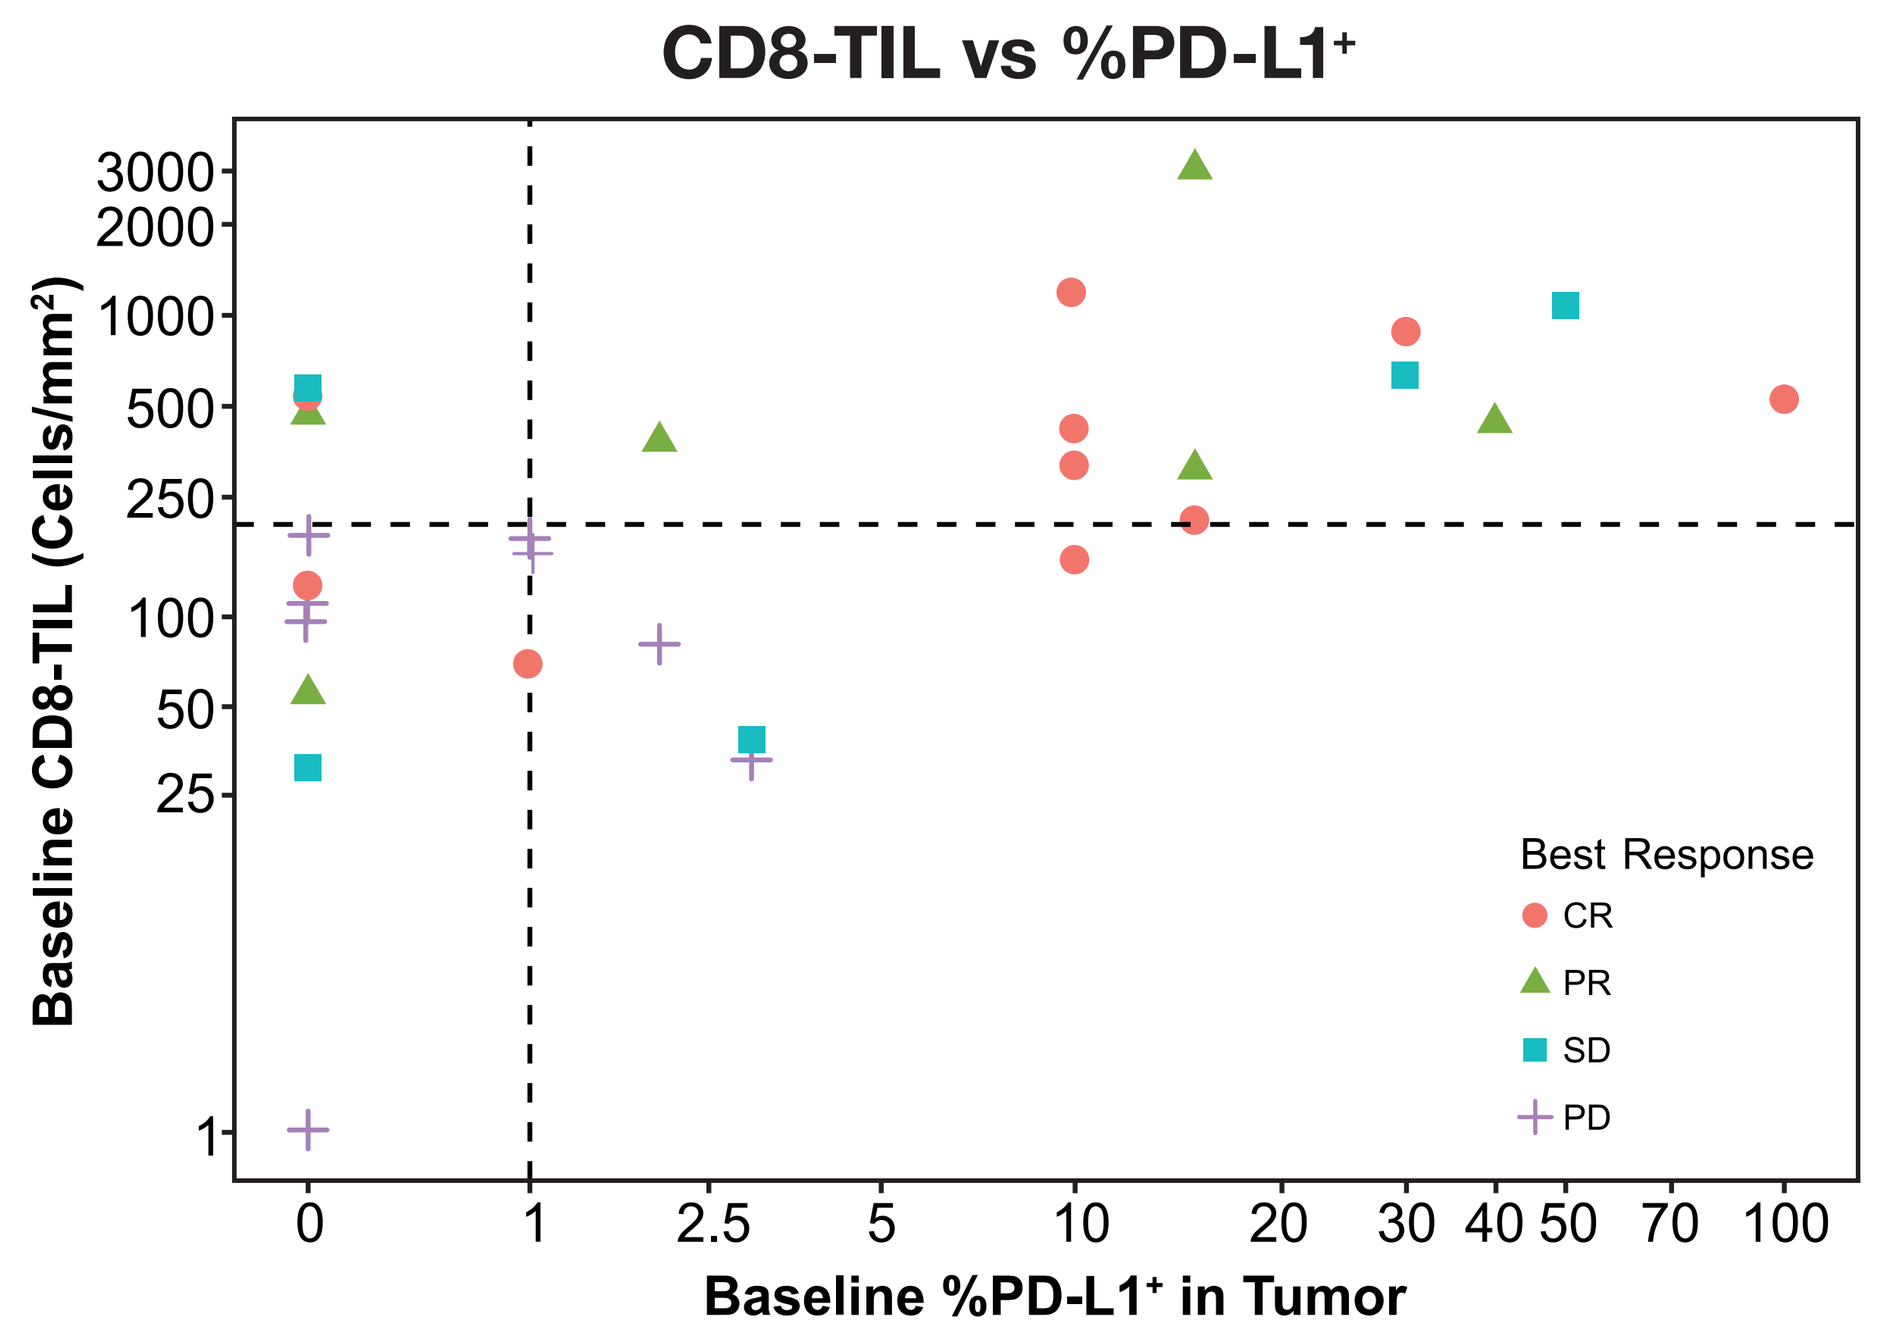
Baseline CD8-TIL (Cells/mm²)
Baseline %PD-L1+ in Tumor
Best Response
CR
PR
SD
PD

	PD-L1 Neg	PD-L1 Pos
CD8+TIL ≥Median	67% (2/3)	**83% (10/12)**
CD8+TIL <Median	**29% (2/7)**	29% (2/7)

CD8-TIL vs IFNg

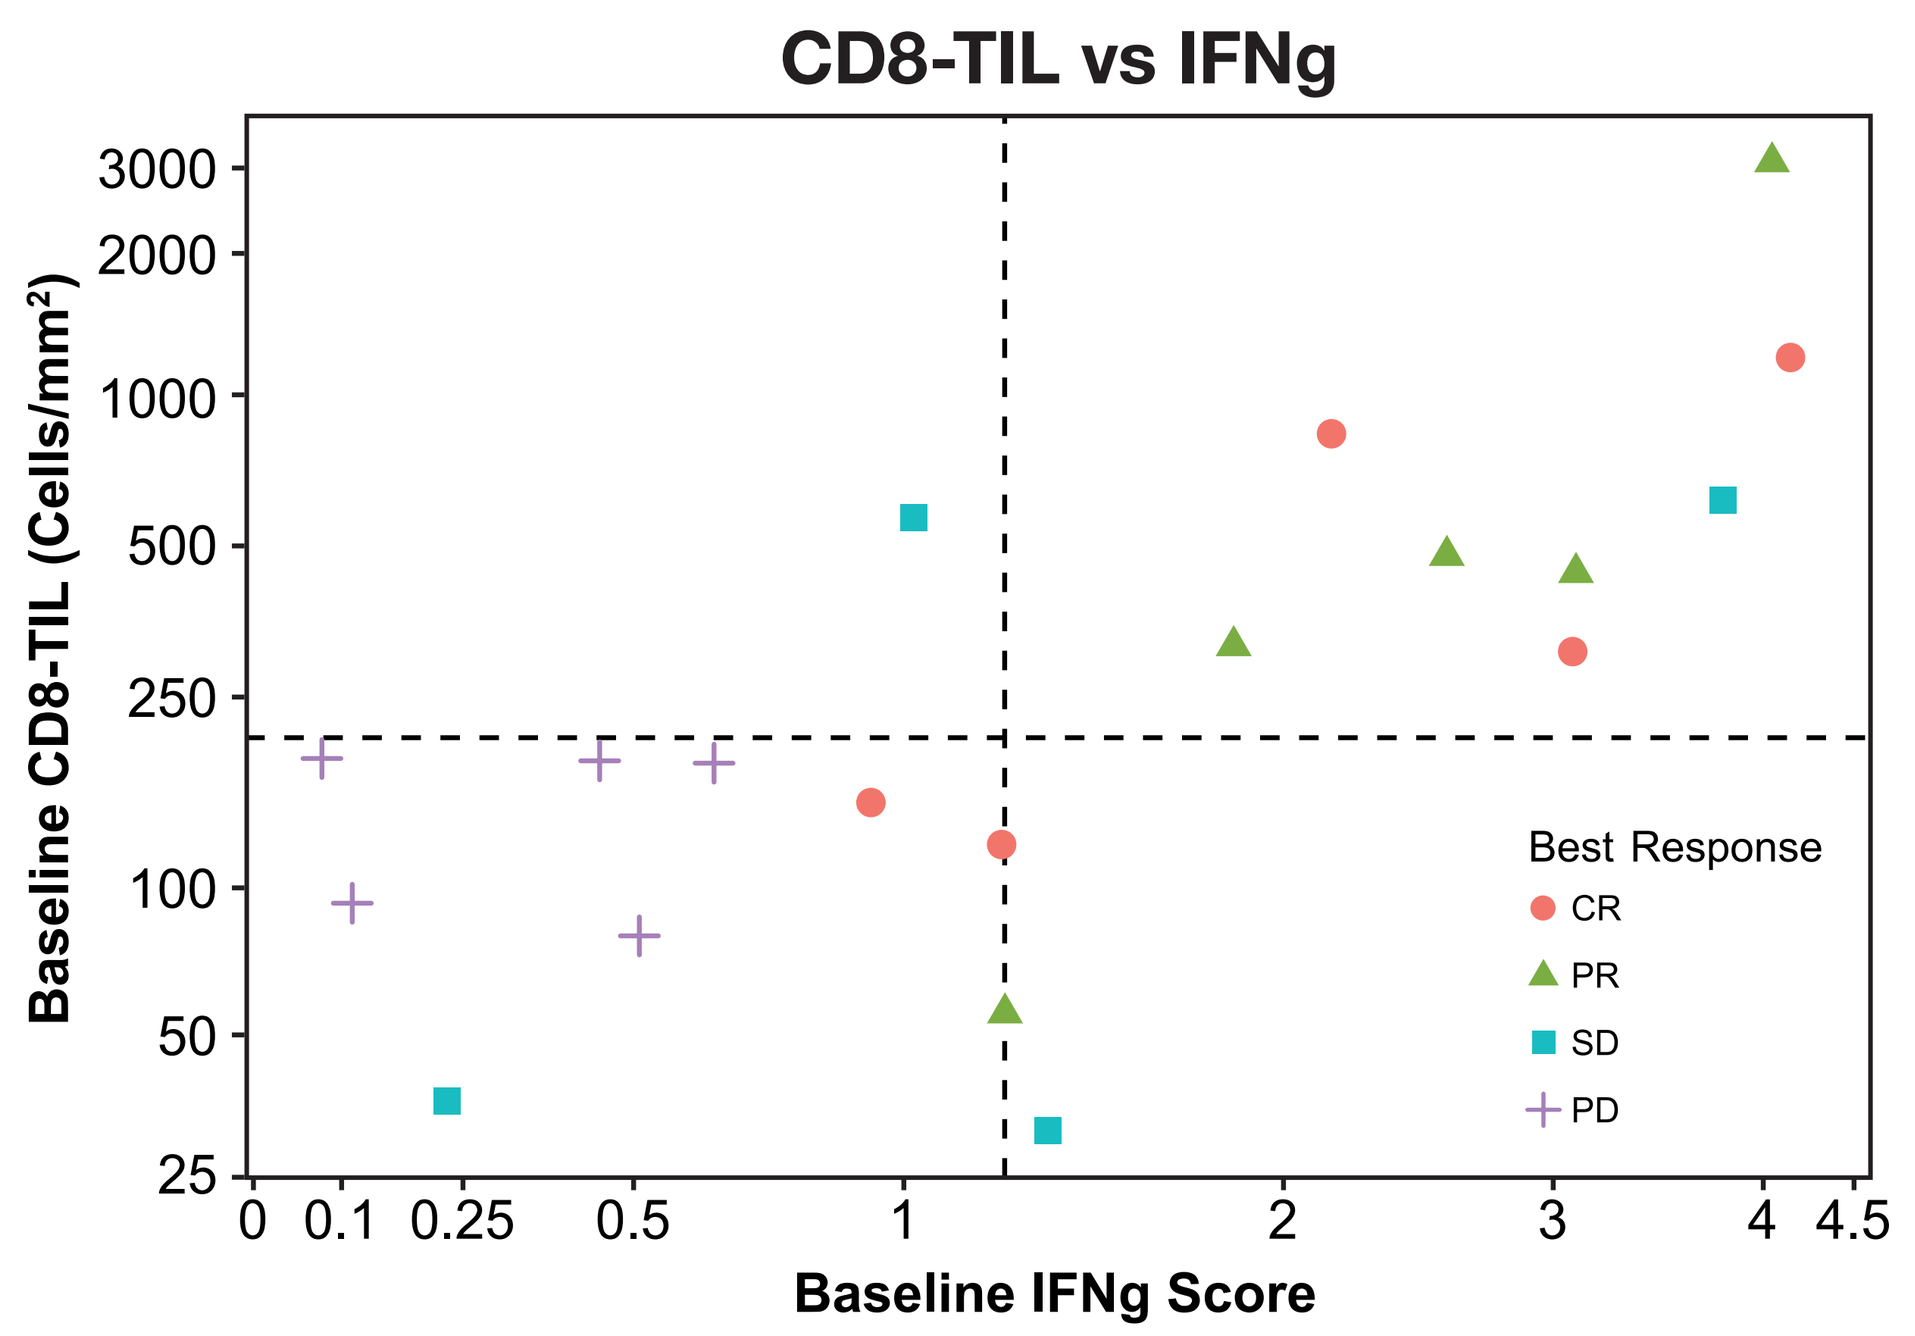
Baseline CD8-TIL (Cells/mm²)
Baseline IFNg Score
Best Response
CR
PR
SD
PD

	IFNg <Median	IFNg ≥Median
CD8+TIL ≥Median	0% (0/1)	**88% (7/8)**
CD8+TIL <Median	**25% (2/8)**	50% (1/2)

IFNg vs %PD-L1+

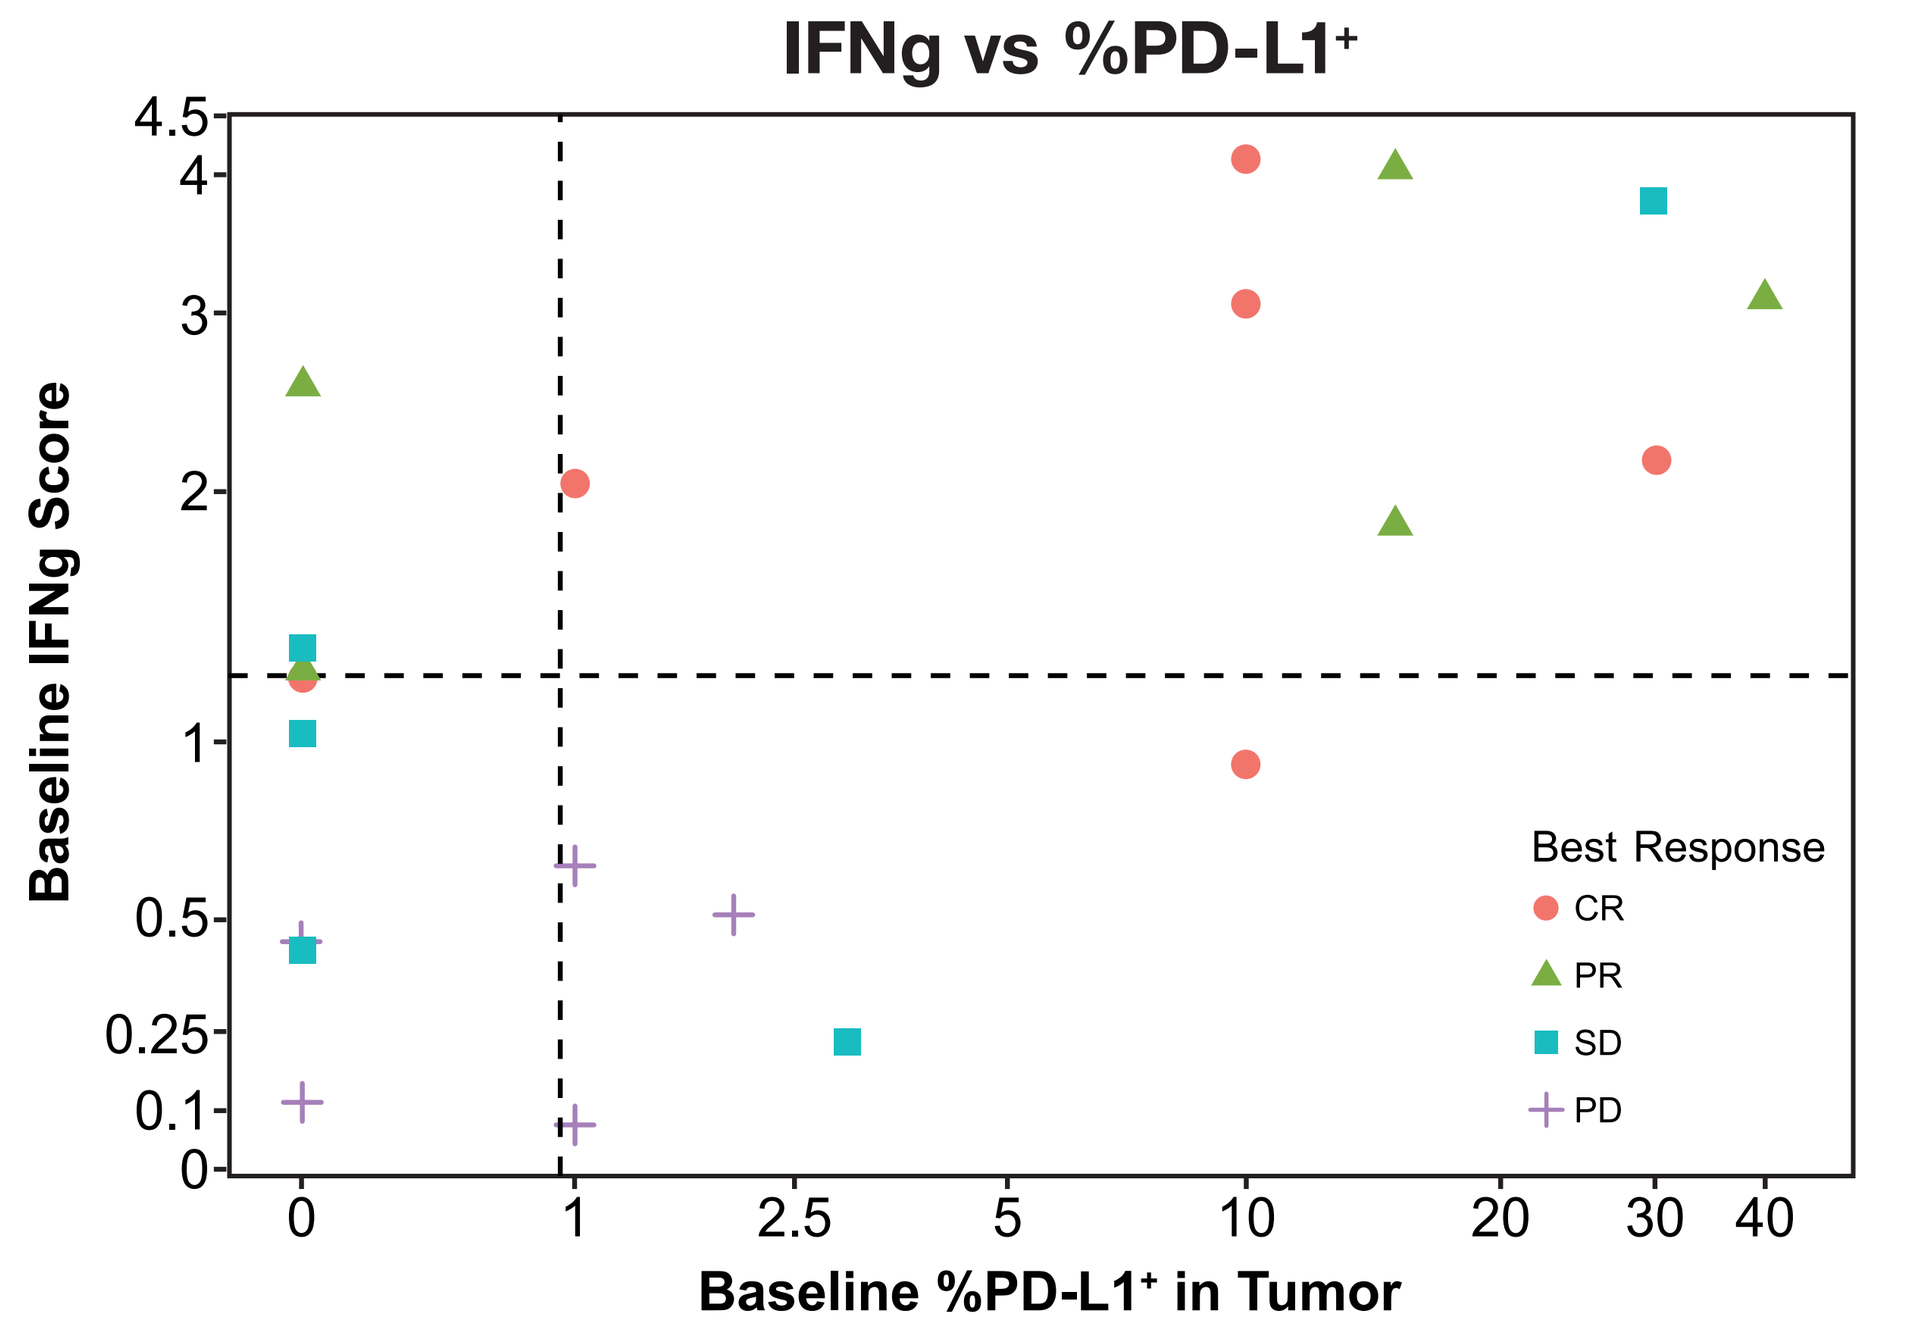
Baseline IFNg Score
Baseline %PD-L1+ in Tumor
Best Response
CR
PR
SD
PD

	PD-L1 Neg	PD-L1 Pos
IFNg ≥Median	67% (2/3)	**88% (7/8)**
IFNg <Median	**20% (1/5)**	20% (1/5)

*Unfavorable TME is defined as low/low by TILs/PD-L1, IFNg/TILs, and IFNg/PD-L1[8-10]

2x2 tables are based on median cutoffs of CD8-TIL and IFNg (≥ vs. <), and PD-L1 (≥1% vs. <1%)

Median: 203 cells/mm² (CD8+TIL); 1.2 (IFNg)

Spearman correlation on scale from 0-1 was 0.51 (CD8-TIL and PD-L1), 0.68 (IFNg and CD8-TIL), 0.55 (IFNg and PD-L1)

Dotted line marks the median cutoff (CD8-TIL and IFNg) or negative/positive status (PD-L1)

CONCLUSIONS

- Exploratory biomarker analyses of PIVOT-02 baseline tumor biopsies identified immune signatures that enrich for response in patients with 1L MEL and not 1L mUC
- Notable response rates were observed regardless of PD-L1 expression or unfavorable tumor microenvironments
- BEMPEG in combination with NIVO showed anti-tumor activity in the efficacy-evaluable patients
 - 1L Melanoma ORR 53%, CR 34%, DCR 74%
 - 1L mUC ORR 48%, CR 19%, DCR 70%
- The value of baseline biomarkers as predictive factors of response to BEMPEG, in combination with NIVO, is being further explored in PIVOT-02 Clinical trial (NCT02983045), PIVOT IO 001 - Phase 3 1L Melanoma (NCT03635983) and PIVOT 10 - Phase 2 1L Urothelial Cancer (NCT03785925)

REFERENCES

1. Charych D, et al. *PLoS One.* 2017;12: e0179431.
2. Bentebibel SE, et al. *Cancer Discov.* May 8 2019 DOI: 10.1158/2159-8290.CD-18-1495.
3. Diab A, et al. Oral presentation at SITC; November 7-11, 2018; Washington, D.C., USA. Abstract #O4.
4. Siefker-Radtke, et al. Poster presentation at ASCO GU; February 13-16, 2019, San Francisco, CA, USA. Abstract #388.
5. Opdivo (nivolumab) [package insert]. Princeton, NJ: Bristol-Myers Squibb; 2019.
6. Daud AI, et al. *J Clin Oncol.* 2016;34:4102-09.
7. Daud AI, et al. *J Clin Invest.* 2016;126:3447-52.
8. Tumeh PC, et al. *Nature.* 2014;515:568-71.
9. Ayers, https://doi.org/10.1172/JCI91190.
10. Subramanian A, et al. *Proc Natl Acad Sci USA.* 2005;102(43):15545–50.

ACKNOWLEDGEMENTS

The investigators would like to acknowledge the patients and clinical teams for their participation in this study; Dako for collaborative development of the PD-L1 IHC 28-8 pharmDx assay; Bristol-Myers Squibb (Princeton, NJ). This study was sponsored by Nektar Therapeutics. All authors contributed to and approved the presentation. Writing and editorial assistance was provided by Phillips Gilmore Oncology Communications and funded by Nektar Therapeutics.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster

Presented at ASCO, Chicago, IL, on June 1, 2019. Poster Session: Developmental Immunotherapy and Tumor Immunobiology. For questions or permission to reprint any portion of this poster, contact michael.hurwitz@yale.edu