NKTR-255, a polymer-conjugated IL-15 receptor agonist, enhances efficacy of therapeutic monoclonal antibodies with ADCC activity in solid tumor models

Saul Kivimäe, Takahiro Miyazaki, Rhoneil Pena, Marlene Hennessy, Wildaliz Nieves, Andrew Moffett, Phi Quach, Mekhala Maiti, Lawrence Chinn, Loui Madakamutil and Jonathan Zalevsky

Nektar Therapeutics, San Francisco, CA

NEKTAR

Poster # P619

Introduction

- IL-15 is a critical cytokine for NK cell survival and functional activation. Therapeutic application of native IL-15 has been challenging due to unfavorable pharmacokinetic properties and tolerability.
- NKTR-255 is a polymer conjugated human IL-15 that retains binding affinity to the IL-15 receptor alpha subunit and exhibits reduced clearance to provide a sustained pharmacodynamic response including proliferation and functional activation of NK cells.[1]
- NKTR-255 has potential to increase efficacy of tumor targeting antibody therapies with ADCC mechanism of action by expanding NK cells with increased cytotoxicity and enhanced surface expression of low affinity Fc receptor CD16.
- We have previously demonstrated ADCC enhancement by NKTR-255 with daratumumab and rituximab in B cell lymphoma xenograft tumor models[1]. Here we extend this therapeutic concept into solid tumor setting showing NKTR-255 dependent enhancement of ADCC in vitro with colorectal and head and neck squamous cell carcinoma (HNSCC) tumor cell lines and improvement of cetuximab and trastuzumab efficacy in vivo correlating with proliferation and functional activation of tumor resident NK cells in multiple xenograft solid tumor models in mice.

Antibody-dependent cellular cytotoxicity (ADCC) by NK cells

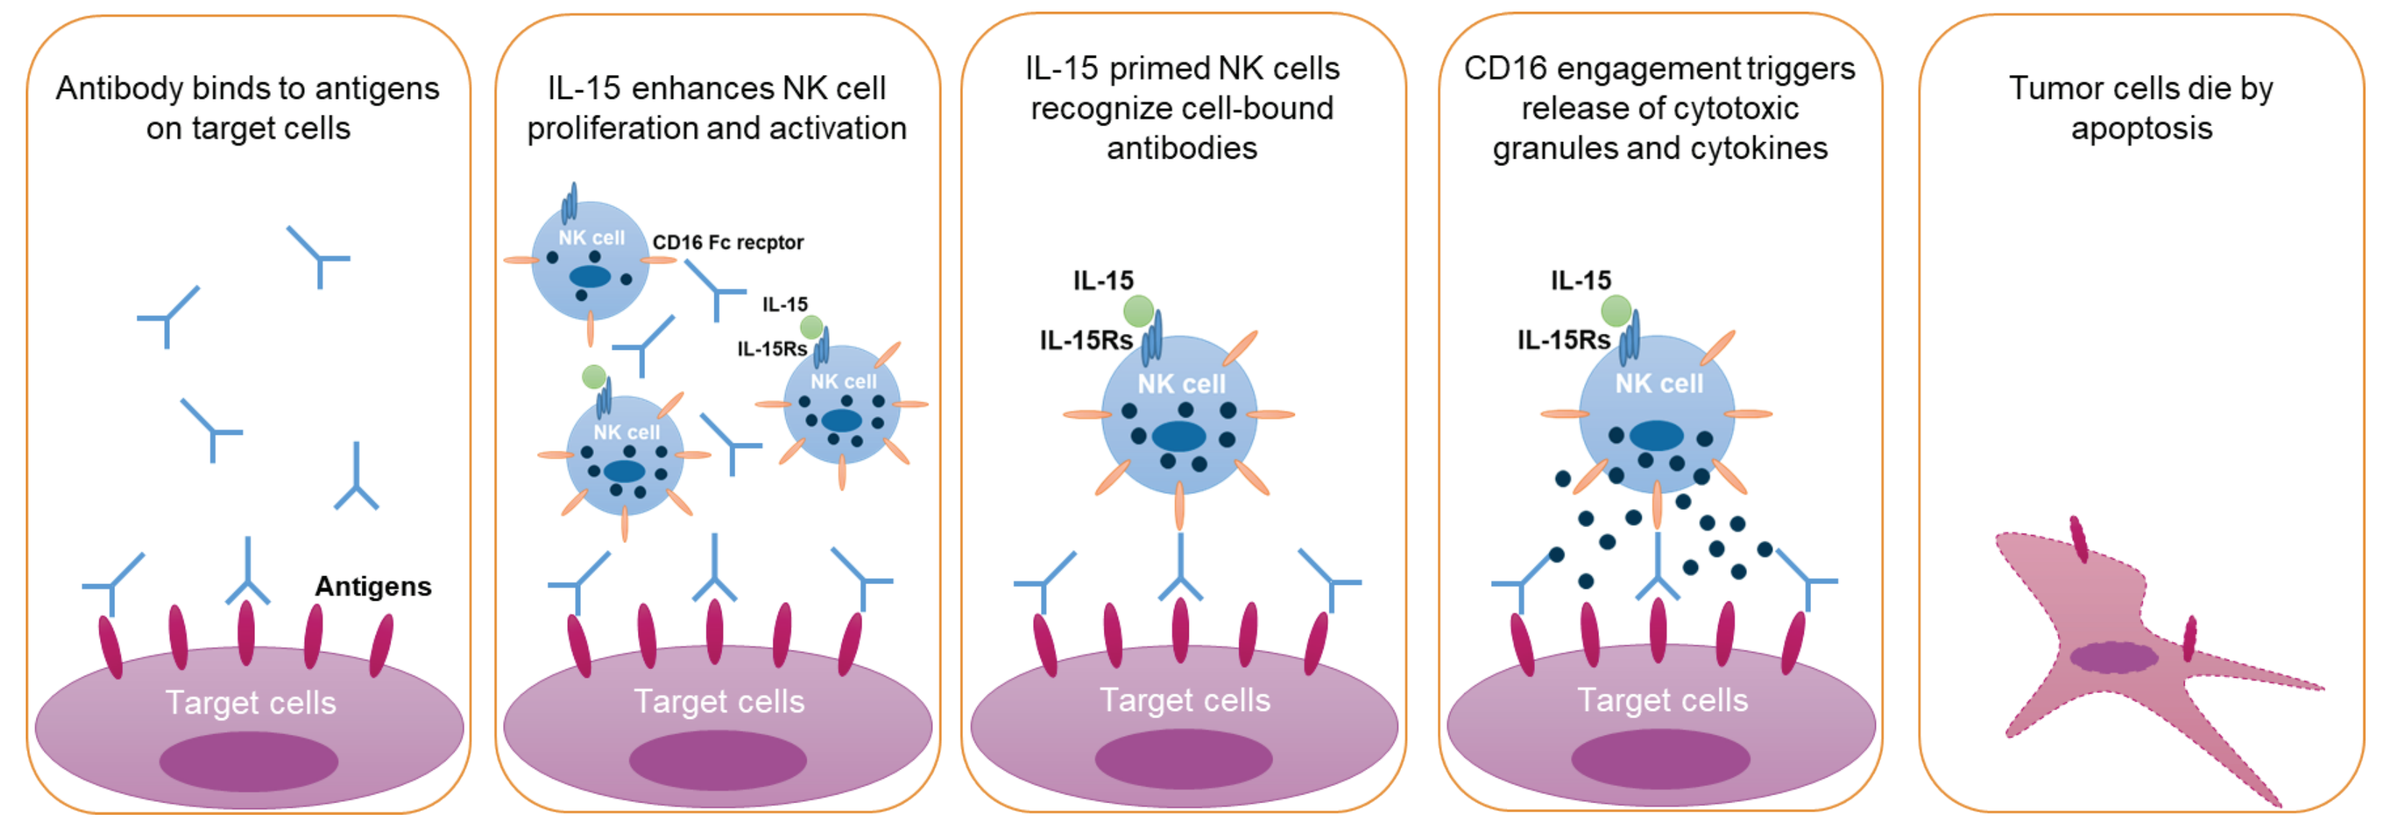
Antibody binds to antigens on target cells
IL-15 enhances NK cell proliferation and activation
IL-15 primed NK cells recognize cell-bound antibodies
CD16 engagement triggers release of cytotoxic granules and cytokines
Tumor cells die by apoptosis
Antigens
Target cells
NK cell
CD16 Fc receptor
IL-15
IL-15Rs

ADCC is a crucial mechanism in tumor depletion by tumor-targeted antibodies. CD16 Fc receptors on NK cells recognize tumor cell-bound antibodies and the CD16 engagement triggers release of cytotoxic granules and cytokines to kill tumor cells. IL-15 enhances NK-mediated ADCC.[2]

Results

Synergistic activation and degranulation enhancement in human NK cells by NKTR-255 and CD16 Fc receptor crosslinking

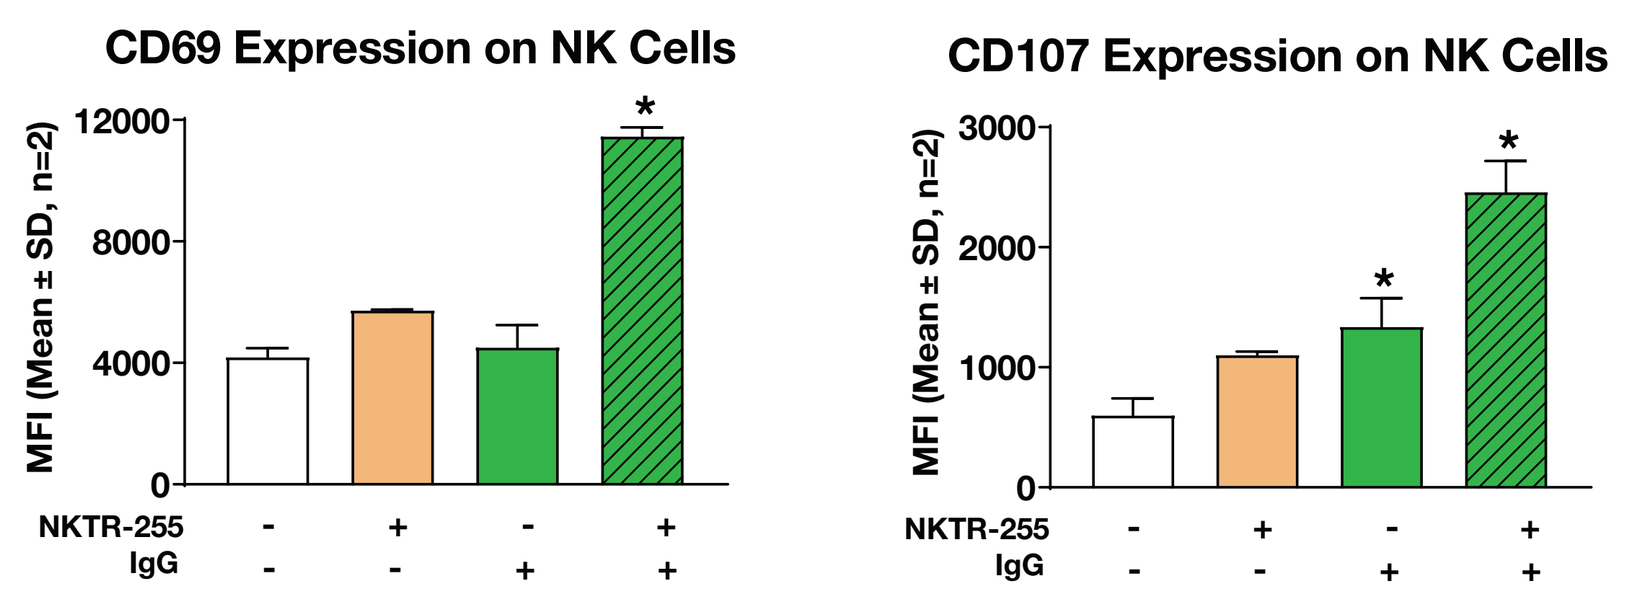
CD69 Expression on NK Cells
CD107 Expression on NK Cells
MFI (Mean ± SD, n=2)
NKTR-255
IgG

Human PBMCs were cultured overnight with 100 ng/ml NKTR-255 on non-coated or 0.1 mg/ml hIgG coated plates. CD69 and CD107 expression on NK cells was measured by flow cytometry (*, p<0.05 relative to no treatment, one-way ANOVA; SD – standard deviation).

NKTR-255 enhances cetuximab mediated NK cells ADCC against HNSCC and colorectal cancer cell lines in vitro

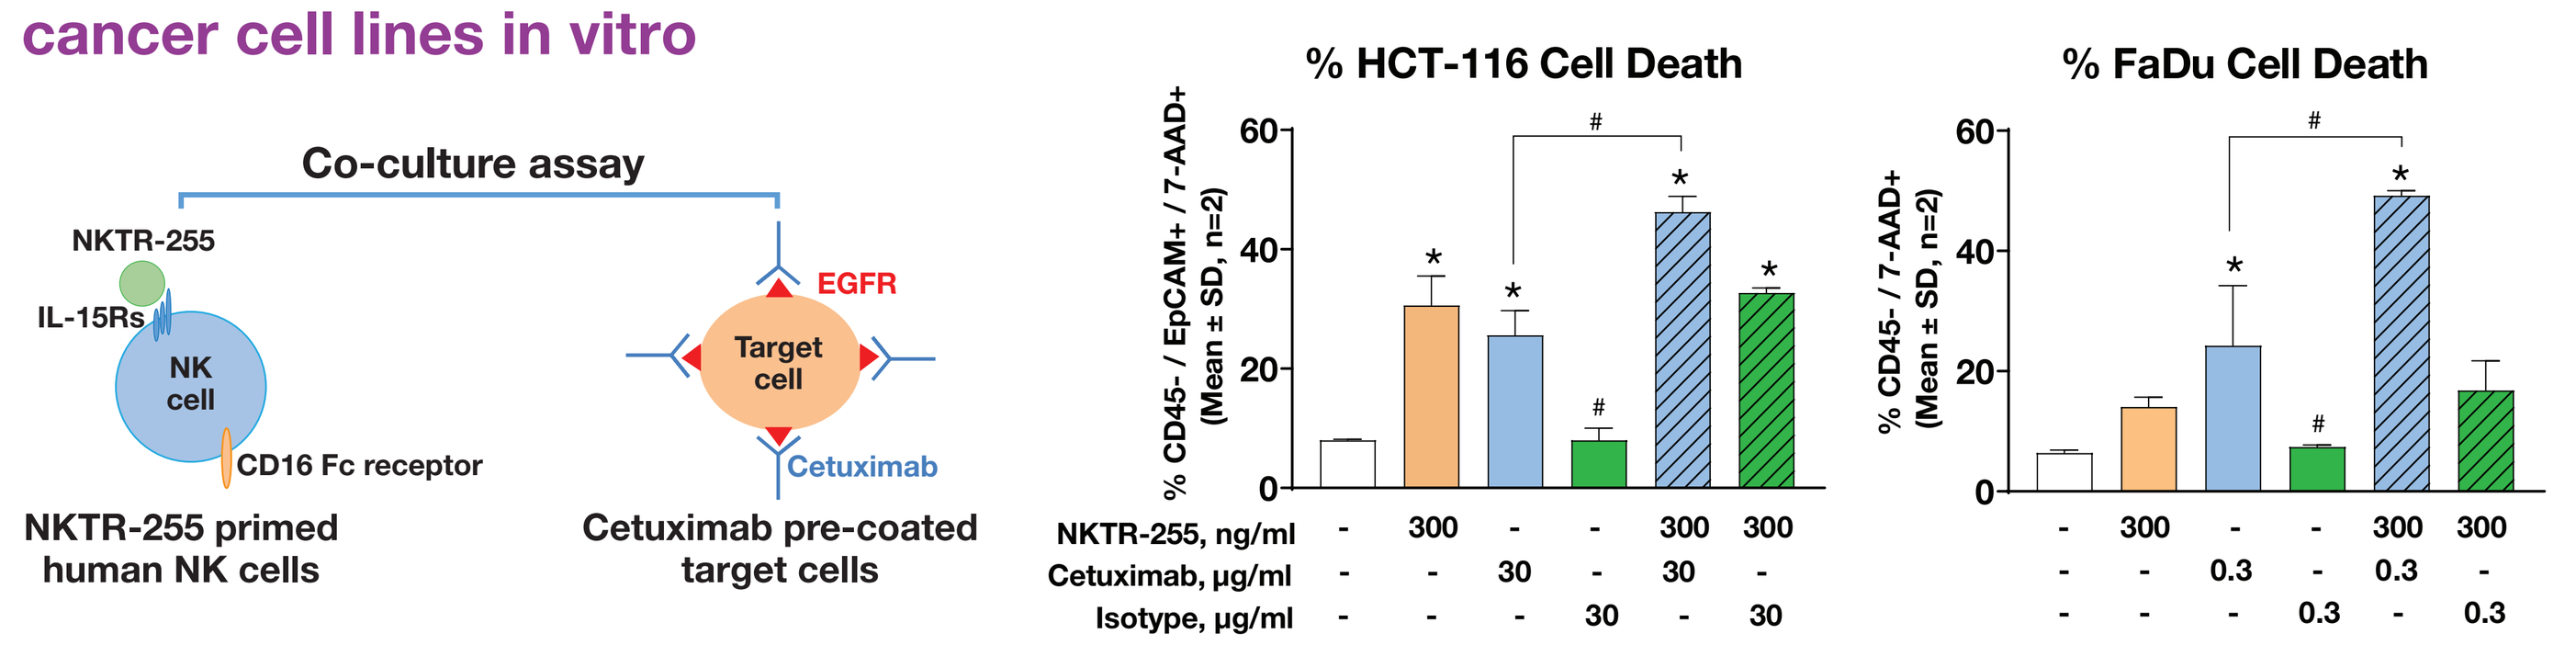
Co-culture assay
NKTR-255
IL-15Rs
NK cell
CD16 Fc receptor
EGFR
Target cell
Cetuximab
NKTR-255 primed human NK cells
Cetuximab pre-coated target cells
% HCT-116 Cell Death
% FaDu Cell Death
% CD45- / EpCAM+ / 7-AAD+ (Mean ± SD, n=2)
% CD45- / 7-AAD+ (Mean ± SD, n=2)
NKTR-255, ng/ml
Cetuximab, µg/ml
Isotype, µg/ml

Purified human NK cells were cultured overnight with or without NKTR-255. Activated NK cells (effector cells) were co-cultured with cetuximab pre-coated FaDu (HNSCC) or HCT-116 (colorectal cancer) cells (target cells) at indicated antibody concentrations at effector:target ratio of 10 for 3 hrs at 37°C. NK cells killing activity was evaluated by detecting 7-AAD stained target cells (*, p<0.05 relative to no treatment, #, p<0.05 relative to cetuximab single agent, one-way ANOVA; SD – standard deviation).

Results

NKTR-255 combination treatment with cetuximab induces NK cell expansion and increased functional activation in blood and tumor tissue in human HNSCC and colorectal cancer xenograft tumor models in SCID mice in vivo

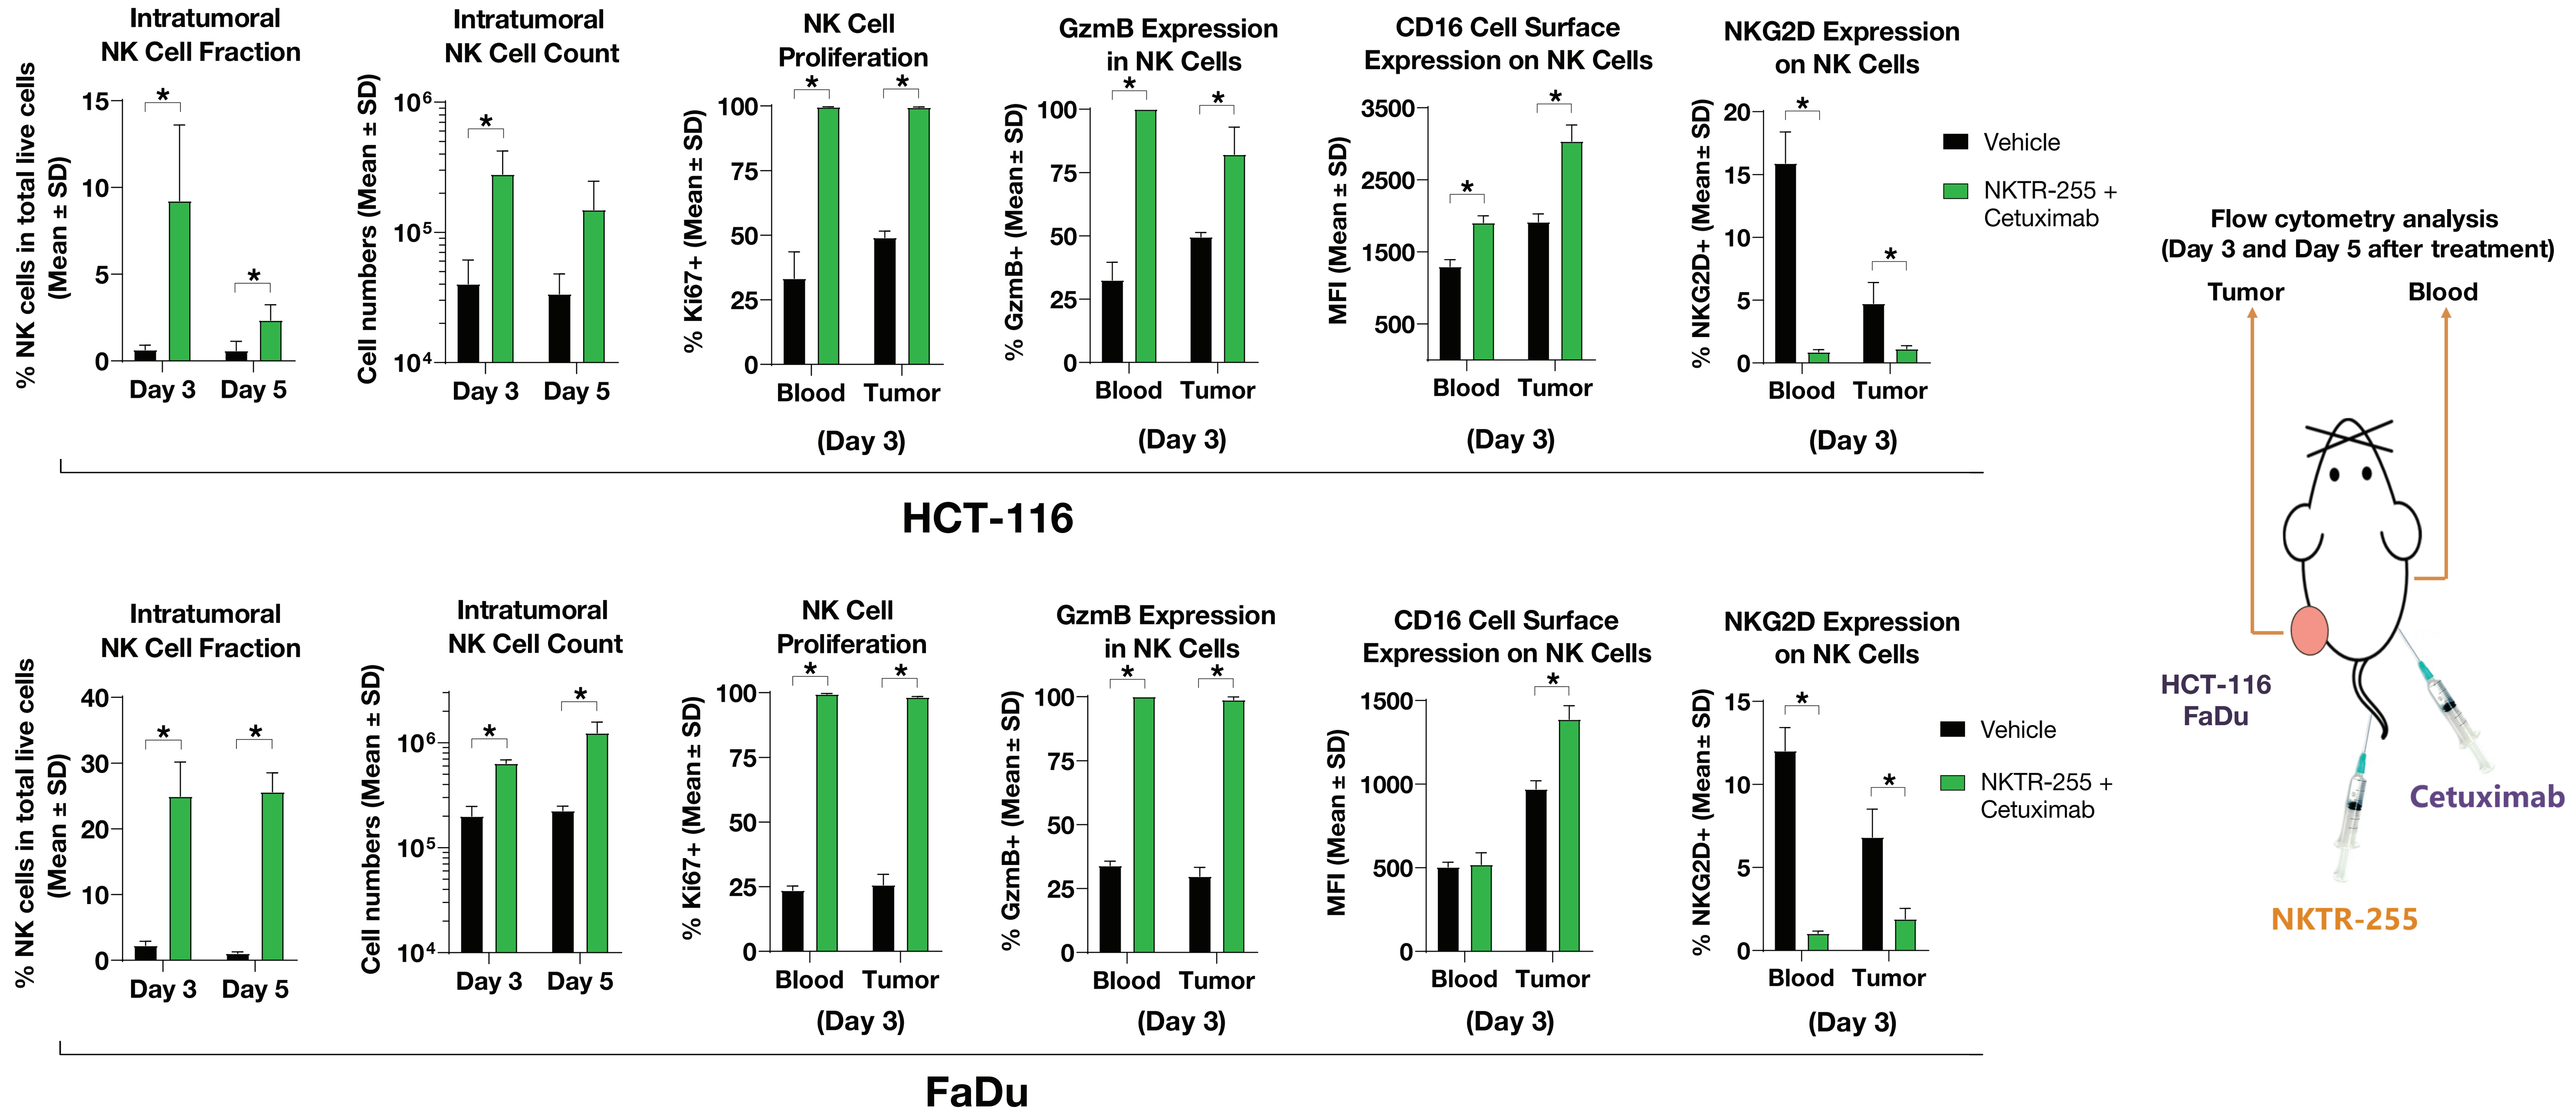
Intratumoral NK Cell Fraction
Intratumoral NK Cell Count
NK Cell Proliferation
GzmB Expression in NK Cells
CD16 Cell Surface Expression on NK Cells
NKG2D Expression on NK Cells
% NK cells in total live cells (Mean ± SD)
Cell numbers (Mean ± SD)
% Ki67+ (Mean± SD)
% GzmB+ (Mean± SD)
MFI (Mean ± SD)
% NKG2D+ (Mean± SD)
Day 3
Day 5
Blood
Tumor
(Day 3)
Vehicle
NKTR-255 + Cetuximab
HCT-116
FaDu
Flow cytometry analysis
(Day 3 and Day 5 after treatment)
Tumor
Blood
HCT-116
FaDu
Cetuximab
NKTR-255

Balb/c SCID mice (n=4/group) bearing subcutaneous HCT-116 or FaDu tumors were treated with NKTR-255 (0.3 mg/kg, IV) and cetuximab (20 mg/kg, IP) 7 days after tumor inoculation at mean tumor volume of ~150mm³. NK cells in blood and in tumors were analyzed by flow cytometry on Day 3 and Day 5 after treatment. (*, p<0.05 relative to vehicle treatment, multiple t-test; SD – standard deviation).

NKTR-255 combination treatment with cetuximab produces tumor growth inhibition activity and induces tumor growth delay in cetuximab resistant human colorectal cancer xenograft tumor models

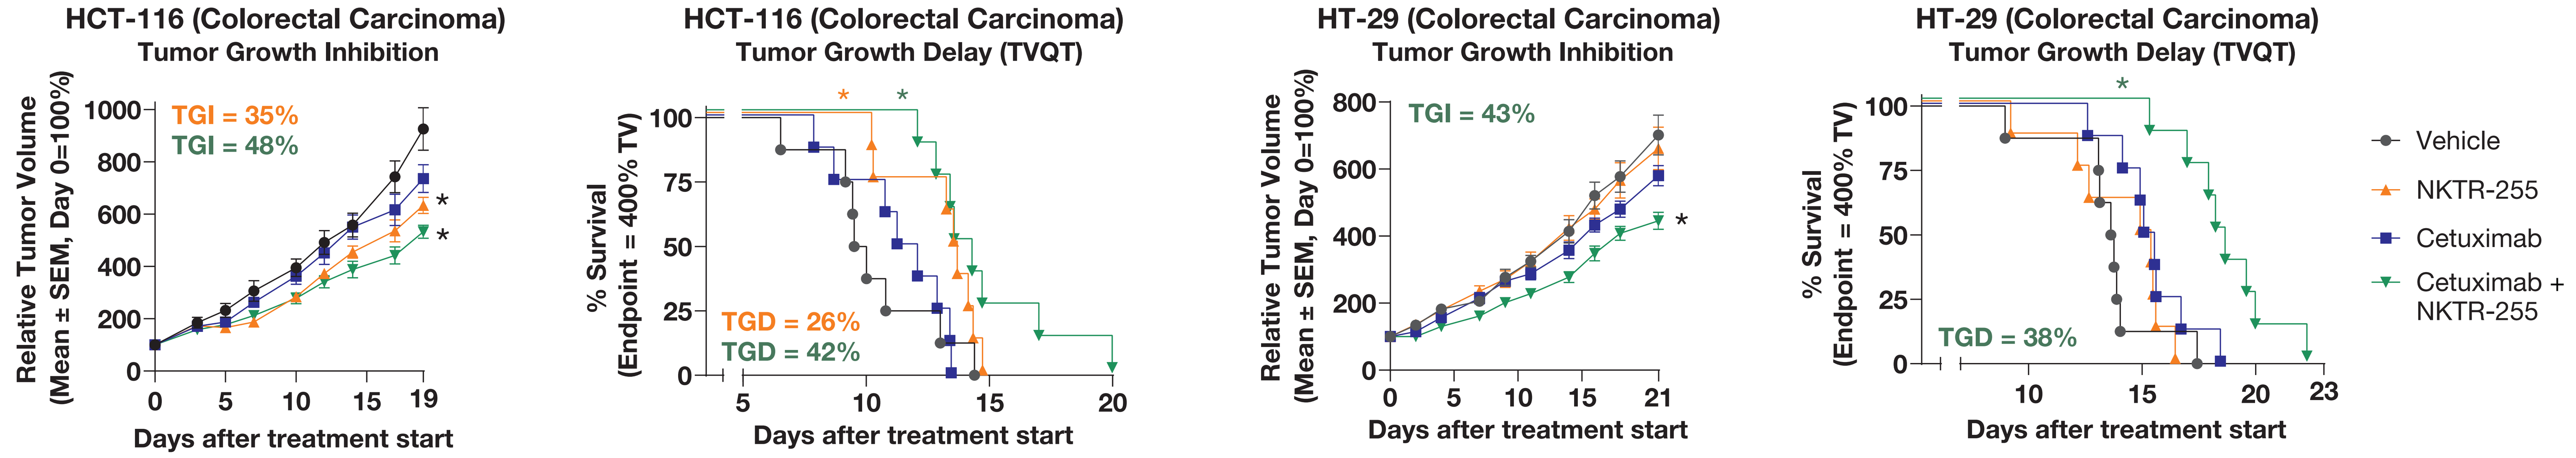
HCT-116 (Colorectal Carcinoma)
Tumor Growth Inhibition
TGI = 35%
TGI = 48%
HCT-116 (Colorectal Carcinoma)
Tumor Growth Delay (TVQT)
TGD = 26%
TGD = 42%
HT-29 (Colorectal Carcinoma)
Tumor Growth Inhibition
TGI = 43%
HT-29 (Colorectal Carcinoma)
Tumor Growth Delay (TVQT)
TGD = 38%
Relative Tumor Volume (Mean ± SEM, Day 0=100%)
% Survival (Endpoint = 400% TV)
Days after treatment start
Vehicle
NKTR-255
Cetuximab
Cetuximab + NKTR-255

Balb/c SCID mice (n=8/group) bearing subcutaneous HT-29 or HCT-116 xenograft tumors were treated with NKTR-255 (0.3 mg/kg, IV, q7dx3) and with cetuximab (40 mg/kg, IP, BIWx3) starting treatment at mean tumor volume of ~150mm³. Relative tumor volumes normalized to starting tumor size were graphed to quantify tumor growth inhibition (TGI) by treatments. Tumor growth delay (TGD) in drug treated animals was evaluated relative to vehicle treatment by measuring tumor volume quadrupling time (TVQT) indicating time to reaching 400% tumor volume (TV) growth from baseline as predefined survival endpoint (*, p<0.05 relative to vehicle treatment, one-way ANOVA; SEM – standard error of the mean).

NKTR-255 enhances trastuzumab tumor growth inhibition activity in human tumor xenograft models in mice

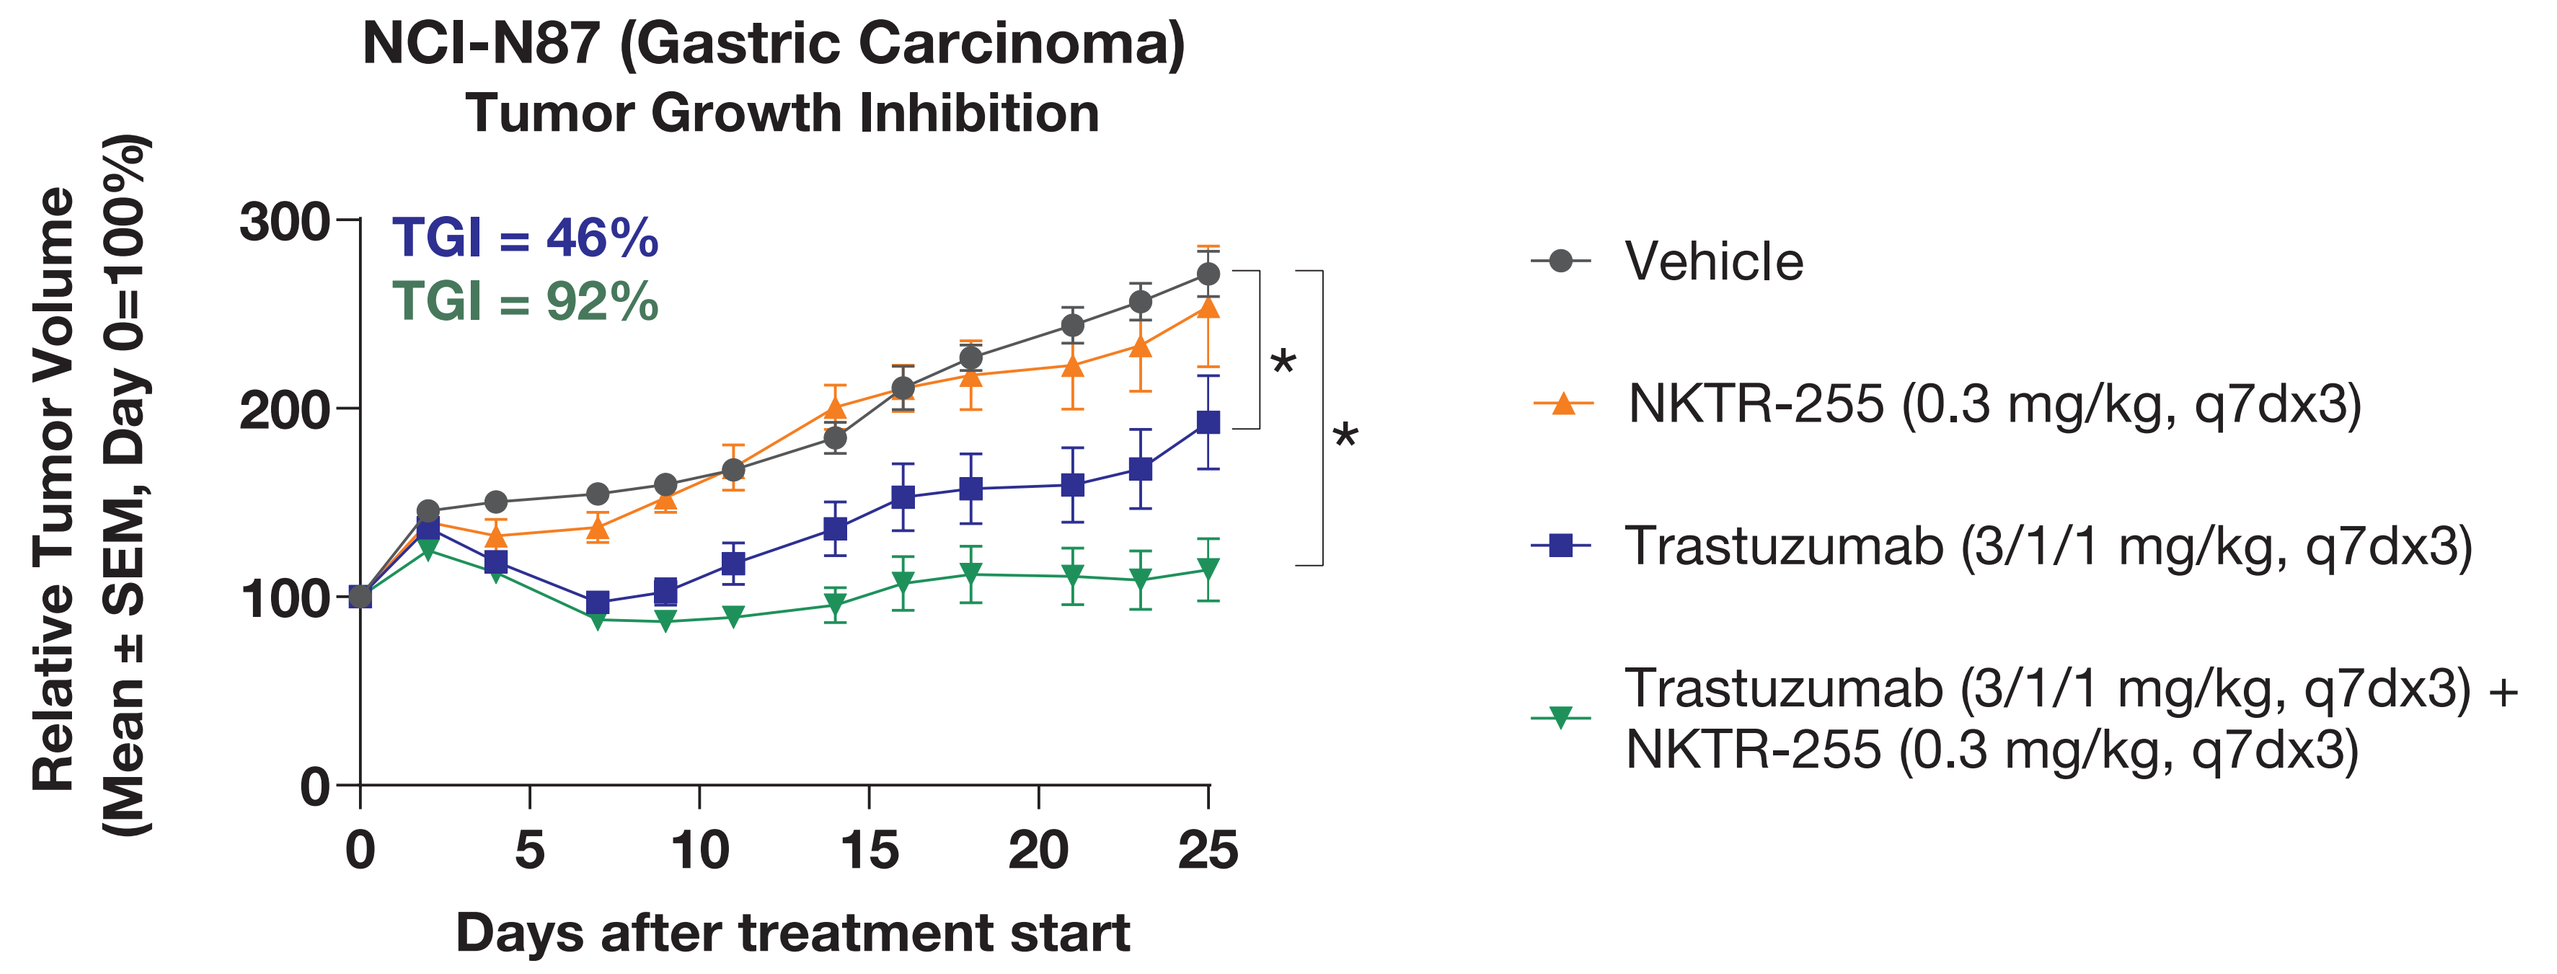
NCI-N87 (Gastric Carcinoma)
Tumor Growth Inhibition
TGI = 46%
TGI = 92%
Relative Tumor Volume (Mean ± SEM, Day 0=100%)
Days after treatment start
Vehicle
NKTR-255 (0.3 mg/kg, q7dx3)
Trastuzumab (3/1/1 mg/kg, q7dx3)
Trastuzumab (3/1/1 mg/kg, q7dx3) + NKTR-255 (0.3 mg/kg, q7dx3)

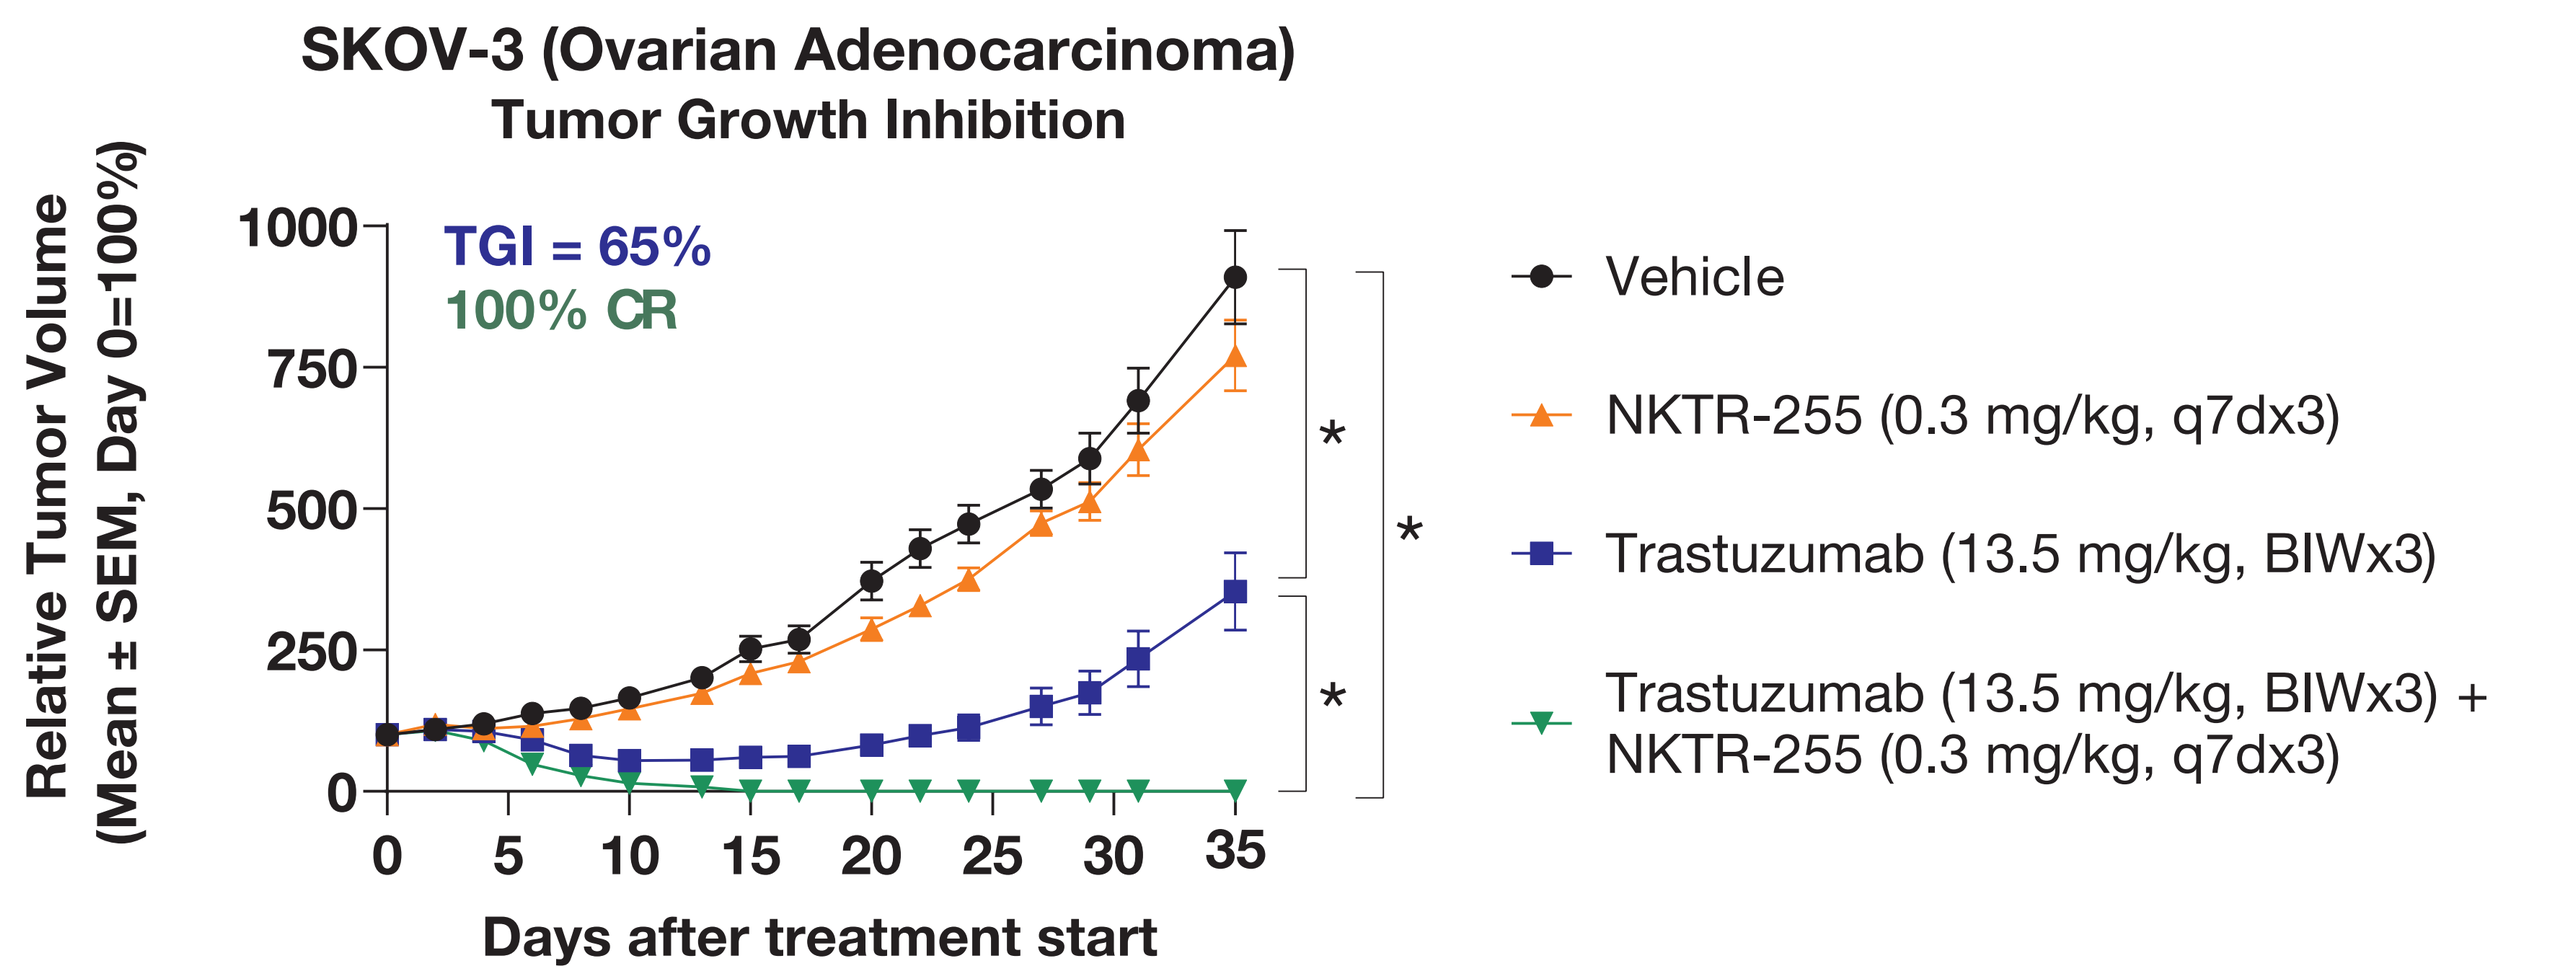
SKOV-3 (Ovarian Adenocarcinoma)
Tumor Growth Inhibition
TGI = 65%
100% CR
Relative Tumor Volume (Mean ± SEM, Day 0=100%)
Days after treatment start
Vehicle
NKTR-255 (0.3 mg/kg, q7dx3)
Trastuzumab (13.5 mg/kg, BIWx3)
Trastuzumab (13.5 mg/kg, BIWx3) + NKTR-255 (0.3 mg/kg, q7dx3)

Balb/c nude mice (n=10/group) bearing subcutaneous NCI-N87 or SKOV-3 xenograft tumors were treated IV with NKTR-255 and trastuzumab at indicated doses starting treatment at mean tumor volume of ~150mm³. Relative tumor volumes normalized to starting tumor size were graphed to quantify tumor growth inhibition (TGI) by treatments (*, p<0.05 relative to vehicle, one-way ANOVA; SEM – standard error of the mean).

Conclusions

- NKTR-255/IL-15R and mAb Fc/CD16 signals in combination synergistically activate NK cells indicating ADCC therapeutic mechanism enhancement potential for NKTR-255.
- NKTR-255 enhances cetuximab-mediated killing of colorectal and HNSCC tumor cells in vitro.
- NKTR-255 induces proliferation and cytotoxic activation of NK cells in blood and in the tumor environment of colorectal and HNSCC xenograft solid tumor models in vivo.
- NKTR-255 in combination with cetuximab induces tumor growth inhibition in cetuximab single agent resistant. solid tumor xenograft models.
- NKTR-255 enhances efficacy of trastuzumab in solid tumor xenograft models.
- NKTR-255 has a potential to be applied with certain cancer therapies to enhance ADCC dependent therapeutic activity of tumor-targeting monoclonal antibodies in solid tumors.

References

1. Miyazaki et al. NKTR-255, a polymer-conjugated IL-15 enhances anti-tumor NK cell responses and synergizes with monoclonal antibodies to provide long-term survival in human lymphoma model. AACR. 2019, Poster 3265.
2. Wei et al. NK cell-mediated antibody-dependent cellular cytotoxicity in cancer immunotherapy. *Front Immunol.* 2015; 27.

Presented at Society for Immunotherapy of Cancer 2019 Annual Meeting