Preclinical efficacy and tolerability of NKTR-255, a polymer conjugated IL-15 for immuno-oncology

Peiwen Kuo, Peter Kirk, Murali Addepalli, Phi Quach, Jane Gunther, Laurie VanderVeen, Ping Zhang, Palakshi Obalapur, Mekhala Maiti, Takahiro Miyazaki, Jonathan Zalevsky

Nektar Therapeutics, San Francisco, CA, United States of America

BACKGROUND

IL-15 is a cytokine that activates T cells and NK cells and has long been recognized for its potential as an immunotherapeutic agent for the treatment of cancer. Exploiting this potential has been challenging due to unfavorable pharmacokinetic properties. NKTR-255 is a polymer-conjugated IL-15 that shows improved plasma exposure while retaining potency and high affinity for IL-15Rα. Here we investigate the pharmacodynamics, preclinical efficacy and tolerability of NKTR-255.

IL-15-mediated signaling through cis- and trans-presentation

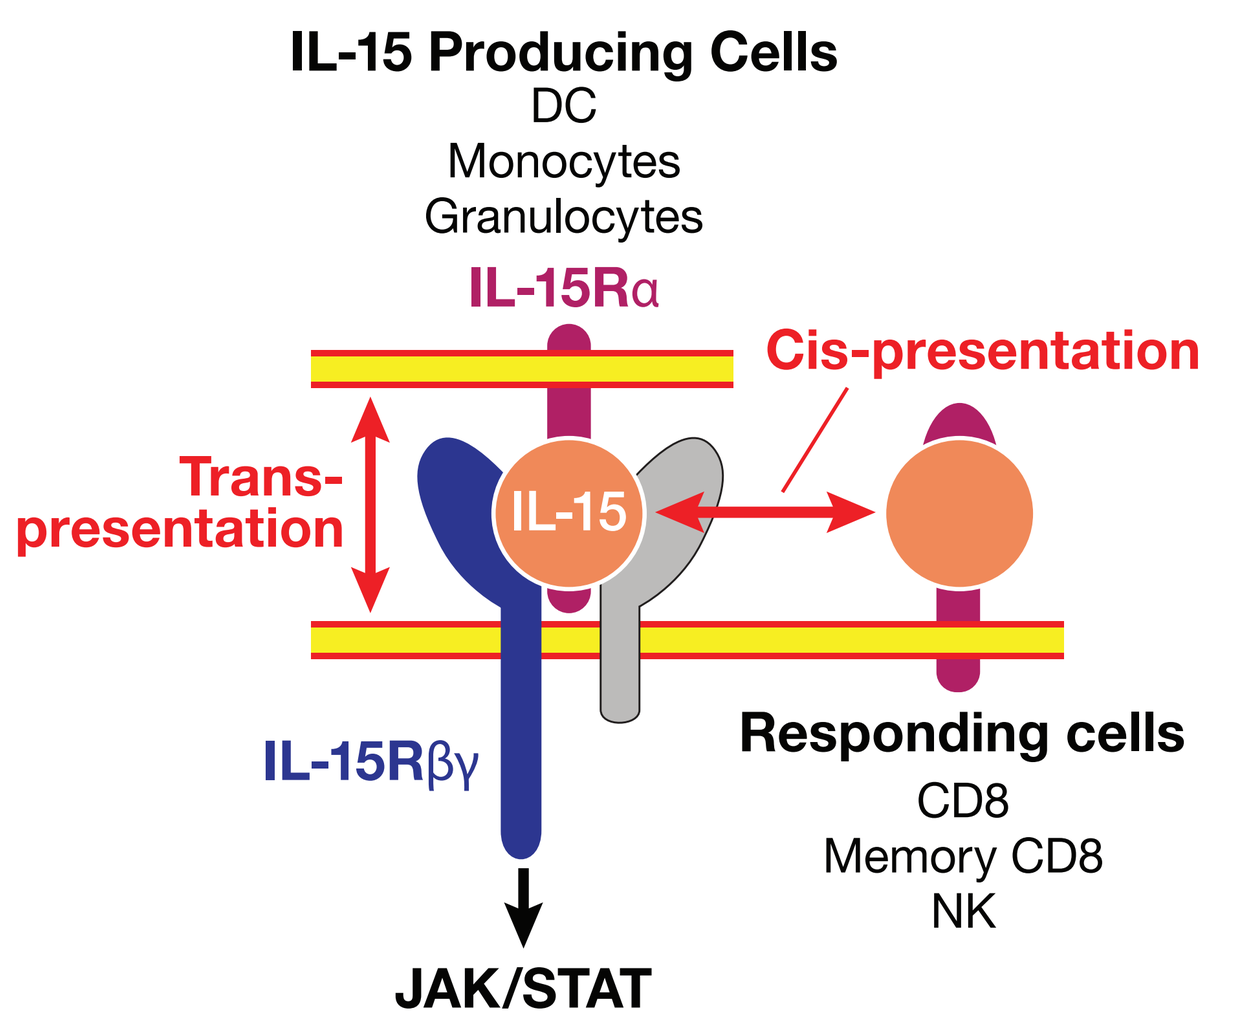
IL-15 Producing Cells
DC
Monocytes
Granulocytes
IL-15Rα
Cis-presentation
Trans-presentation
IL-15
IL-15Rβγ
Responding cells
CD8
Memory CD8
NK
JAK/STAT

Adapted from: Marcais et al. 2013.[1]

A presenting cell synthesizes the unique IL-15Rα chain and IL-15, bound together, and trans-presents IL-15 to the IL-2/IL-15Rβγ receptor complex on the same (cis) or adjacent cell (trans).

Engagement of the IL-2/IL-15Rβγ complex can induce JAK-STAT signaling, increasing survival and proliferation. This process is crucial for the proper support of NK, CD8, and CD8 memory T cells.[2]

METHODS

To assess the pharmacodynamic effects in mice and non-human primates, NKTR-255 was delivered intravenously and whole blood was collected at the indicated time points; flow cytometry was used to measure signaling activity (STAT5 phosphorylation), proliferative status (Ki-67 expression) and absolute frequency of various lymphocyte subpopulations. Pharmacokinetic analysis was conducted following a single intravenous injection of NKTR-255 or IL-15 in naive mice. Analytes were quantified using ELISA-based plasma measurement. Cytotoxicity assays were performed by incubating NK cells purified from spleens of NKTR-255-treated mice with dye-labeled YAC-1 target cells and assessing extent of cell-killing by flow cytometry. Efficacy was studied using the CT-26 lung metastasis model. Briefly, 10^5 CT-26 cells were injected into the tail-vein of mice, NKTR-255 treatment was initiated the following day, lungs were harvested on day 13 and metastases were counted. Some mice were treated with anti-asialo GM1 to deplete NK cells. Cell-based potency was determined by treating human peripheral blood mononuclear cells (PBMC) *in vitro* with a range of concentrations of NKTR-255 or IL-15 and then assessing CD8 and CD56bright NK cell pSTAT5.

RESULTS

NKTR-255 exhibits superior plasma exposure compared to conventional IL-15, inducing rapid and sustained signaling in lymphocytes following intravenous administration in mice and non-human primates. This sustained signal results in proliferation of CD8 T-cells, and a preferential expansion of the CD8 central memory population. In mice, NK cells increase in number and in Granzyme B expression, concomitant with an increase in cytotoxic potential. Notably, the robust induction of CD8 and NK cell proliferation is maintained upon repeat dosing. In a mouse model of tumor metastasis to the lungs, NKTR-255 treatment results in an 85% reduction in the number of metastases. This efficacy is primarily driven by NK cells, as demonstrated in cell-depletion studies. The activity of NKTR-255 translates to higher species, supported by sustained CD8 and NK cell increases in cynomolgus monkey and induction of CD8 and NK pSTAT5 in human PBMC *in vitro*. Toxicology assessments demonstrate that NKTR-255 is well tolerated at efficacious dose levels.

Figure 1. NKTR-255 exhibits prolonged plasma exposure and sustained engagement of the IL-15 signaling pathway.

The half-life of NKTR-255 greatly exceeds that of conventional IL-15 ($t_{1/2}$ = 12 hours vs. < 1 hour, respectively). In a separate experiment, NKTR-255 is a potent driver of JAK/STAT signaling in murine CD8 and NK cells with prolonged phosphorylation seen at both 0.03 and 0.3 mg/kg dose levels.

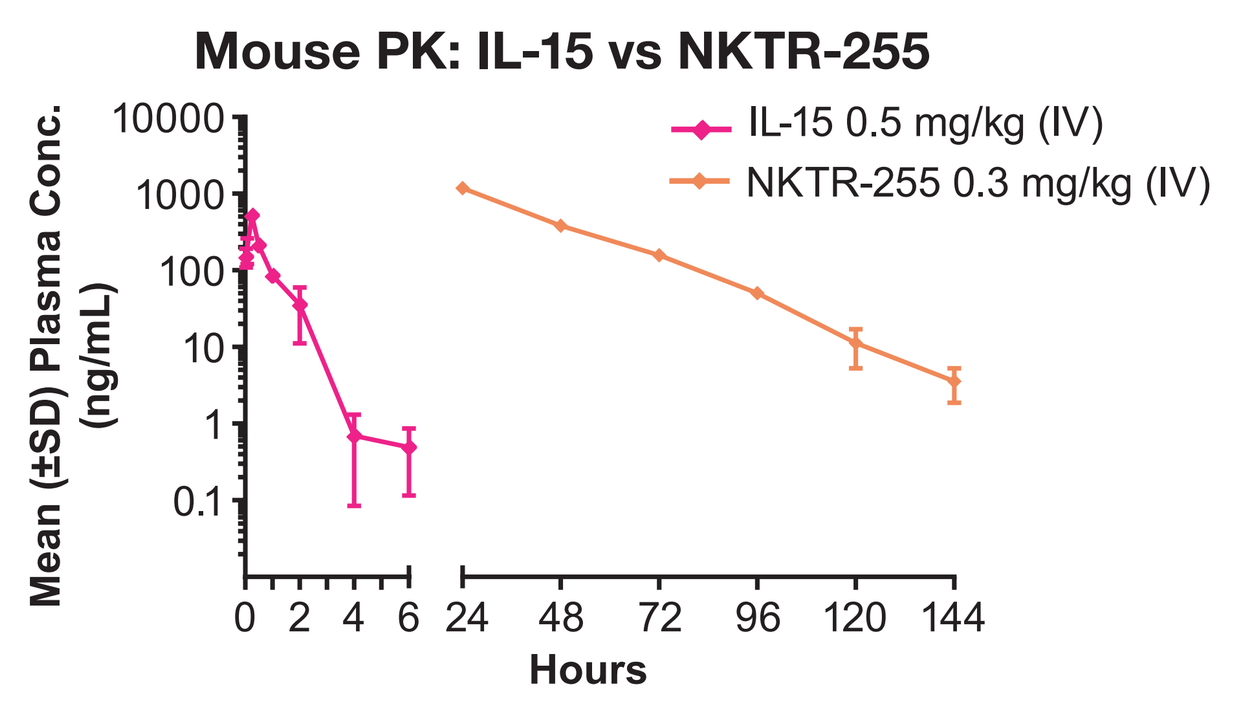
Mouse PK: IL-15 vs NKTR-255
Mean (±SD) Plasma Conc. (ng/mL)
IL-15 0.5 mg/kg (IV)
NKTR-255 0.3 mg/kg (IV)
Hours

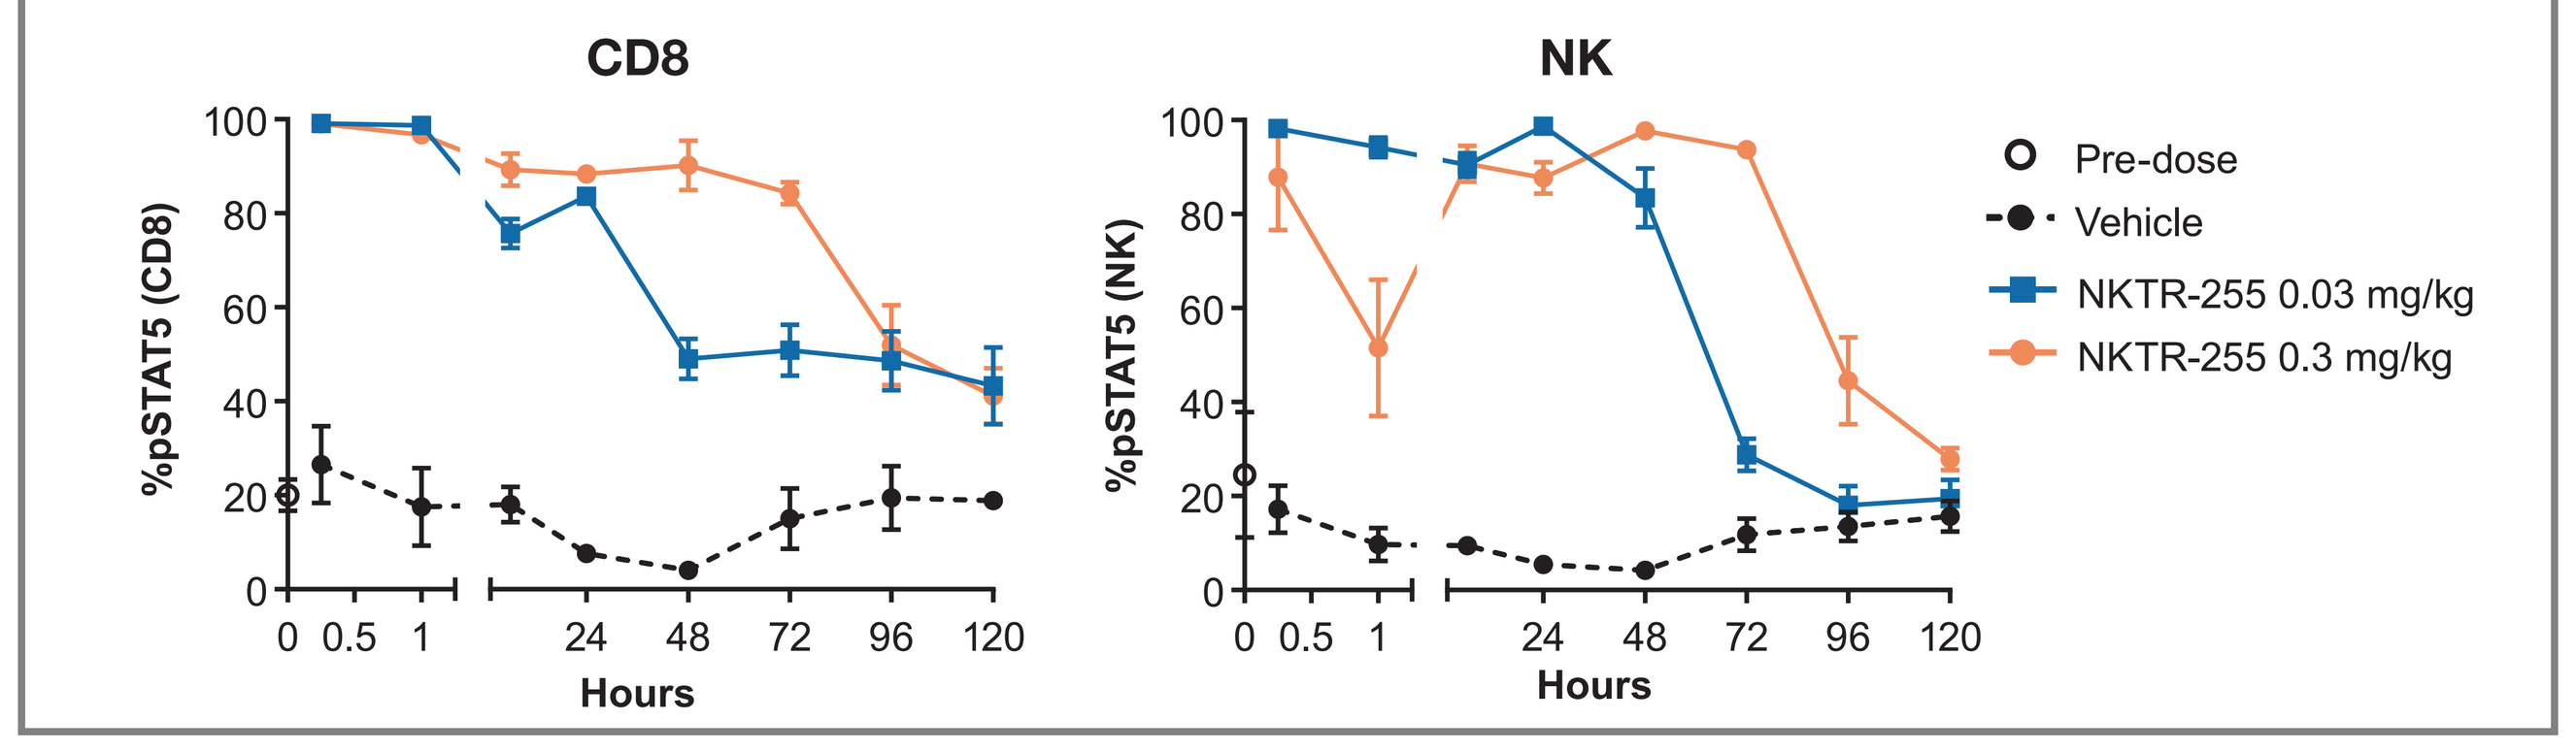
CD8
NK
%pSTAT5 (CD8)
%pSTAT5 (NK)
Hours
Pre-dose
Vehicle
NKTR-255 0.03 mg/kg
NKTR-255 0.3 mg/kg

RESULTS

Figure 2. Single dose NKTR-255 robustly increases CD8 and CD8 memory T cells.

NKTR-255 from 0.01 to 1.5 mg/kg induces a dose-dependent increase in cell numbers (top graphs) and %Ki-67 positivity (bottom graphs) within total CD8, central memory and effector memory T cells in mice. NKTR-255 at a pre-clinical IO efficacious dose (0.3 mg/kg), increases CD8, CD8 Tcm and CD8 Tem by 6.4X, 37.9X and 14.5X, respectively.

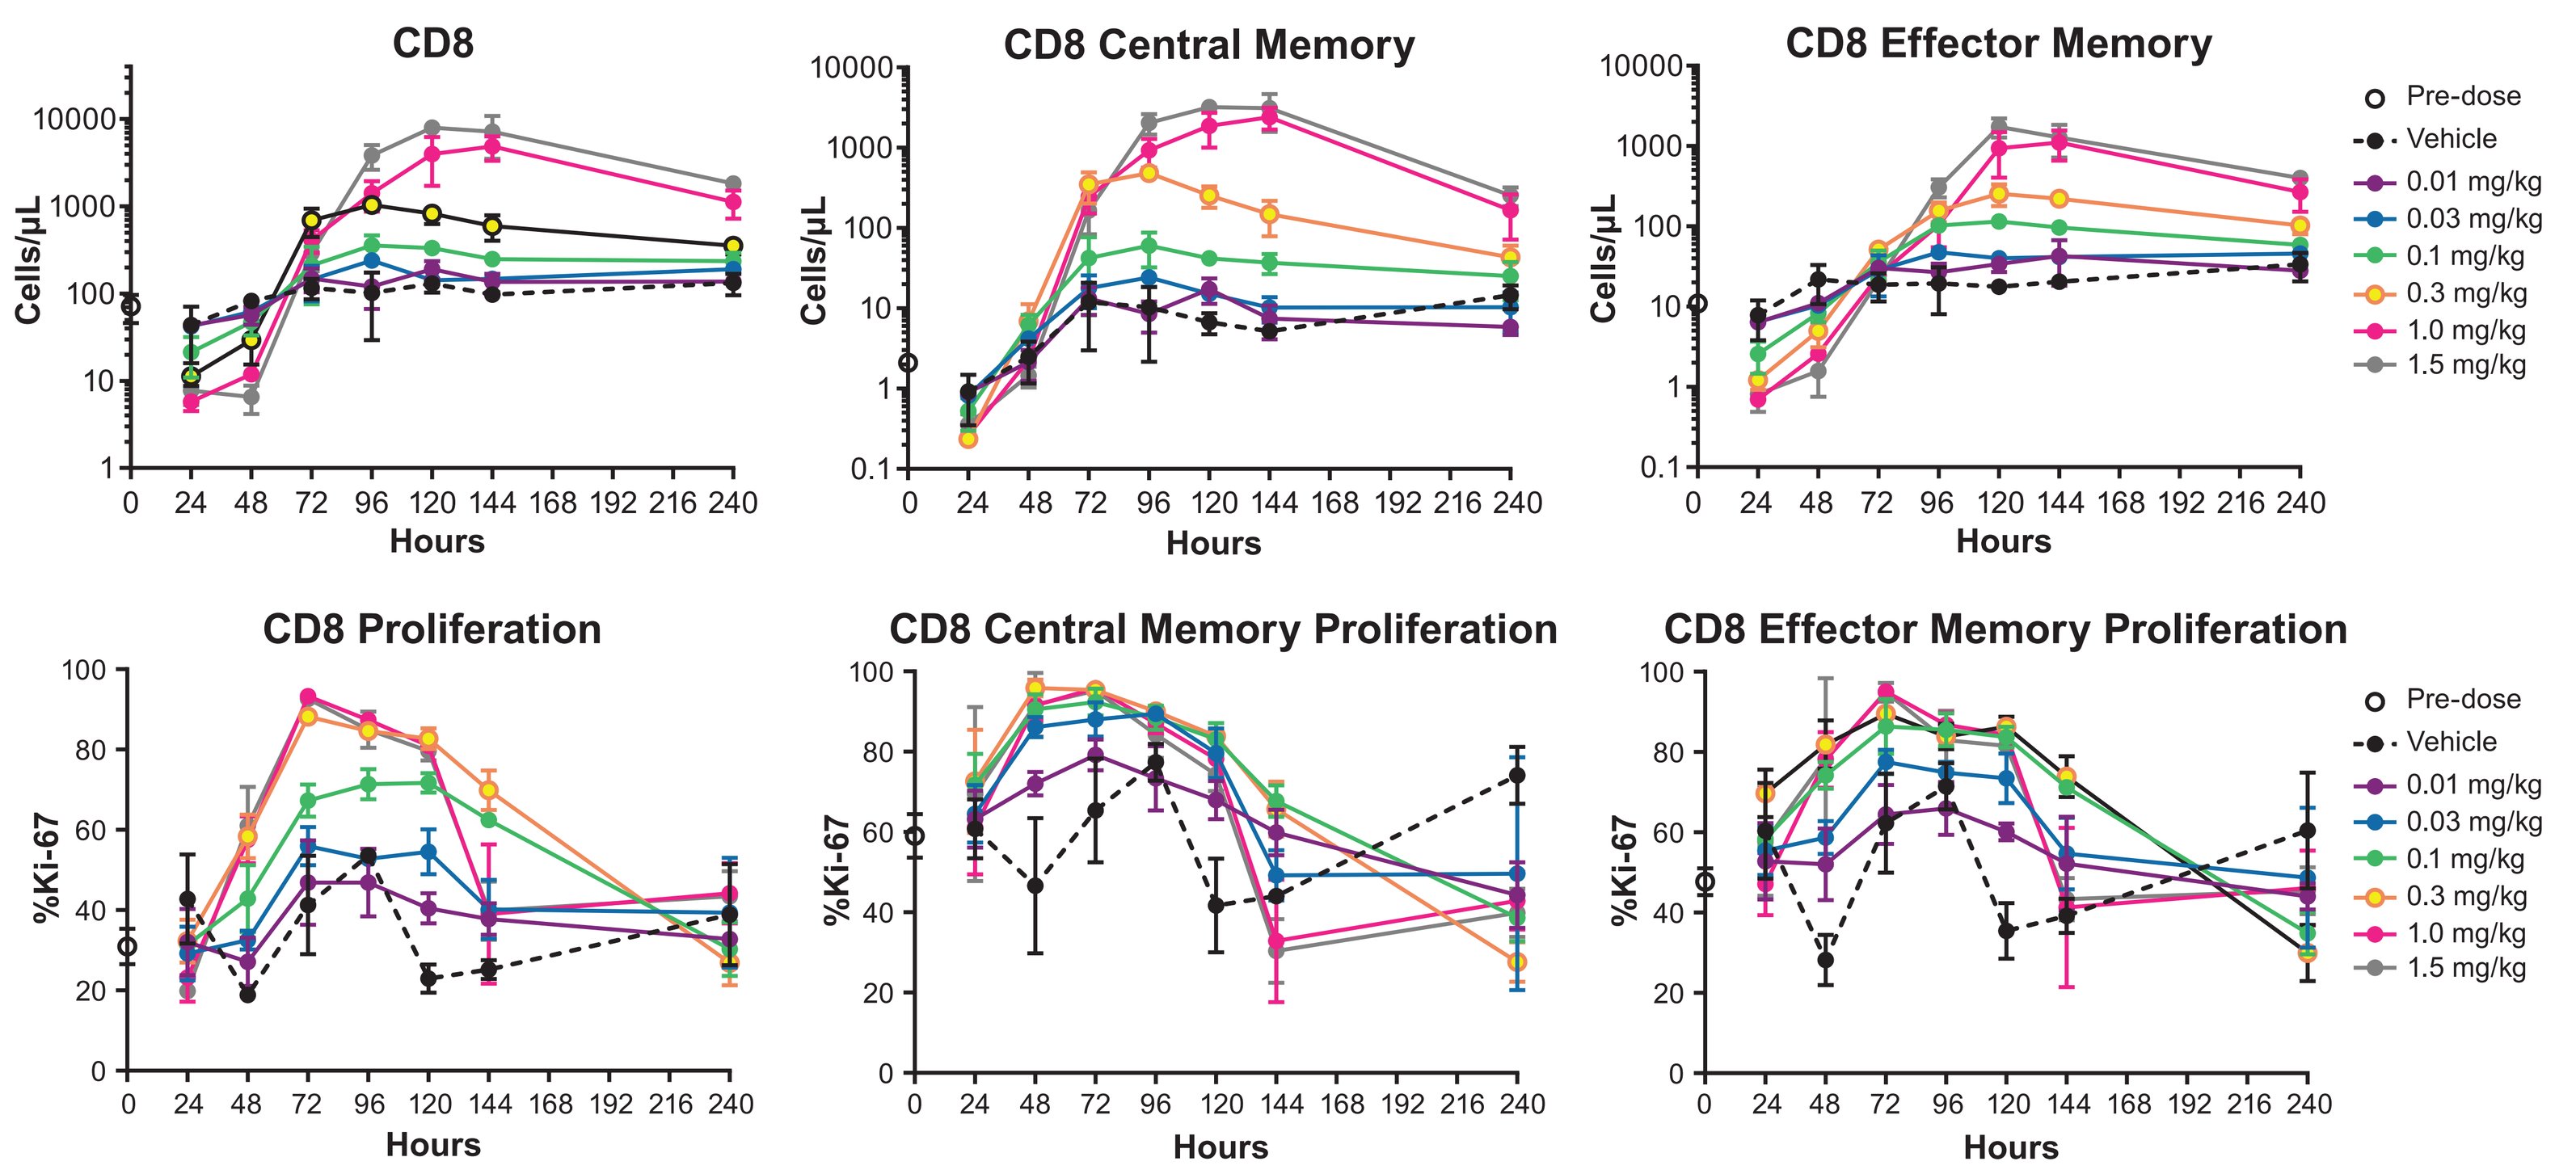
CD8
CD8 Central Memory
CD8 Effector Memory
CD8 Proliferation
CD8 Central Memory Proliferation
CD8 Effector Memory Proliferation
Cells/μL
%Ki-67
Hours
Pre-dose
Vehicle
0.01 mg/kg
0.03 mg/kg
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
1.5 mg/kg

Figure 3. NKTR-255-mediated increase of CD8 and memory CD8 T cells is maintained after repeat dosing.

NKTR-255 induction of murine CD8 and memory CD8 T cells is enhanced when dosed weekly for 3 doses compared to a single dose. Repeat dosing increased total CD8, Tcm and Tem 35.3X, 183X and 73.8X, respectively.

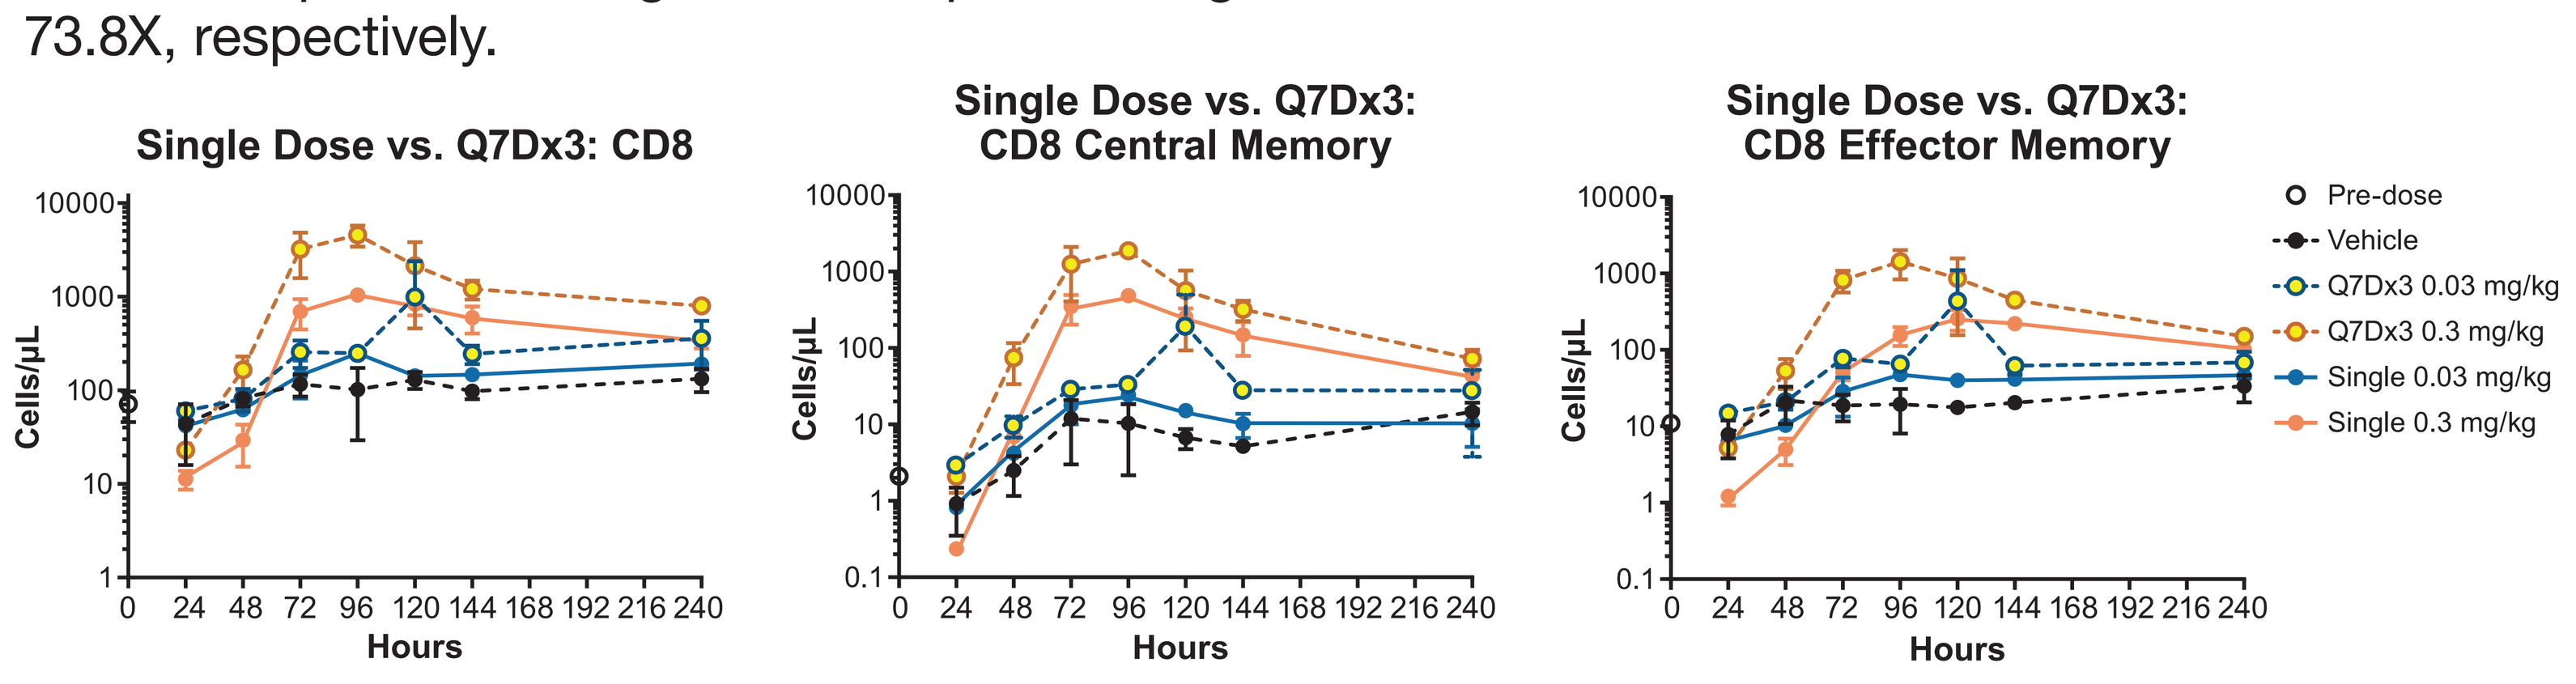
Single Dose vs. Q7Dx3: CD8
Single Dose vs. Q7Dx3: CD8 Central Memory
Single Dose vs. Q7Dx3: CD8 Effector Memory
Cells/μL
Hours
Pre-dose
Vehicle
Q7Dx3 0.03 mg/kg
Q7Dx3 0.3 mg/kg
Single 0.03 mg/kg
Single 0.3 mg/kg

Figure 4. Single and repeat dosing of NKTR-255 increases NK cells.

NK cells increase dramatically in a dose-dependent manner after a single injection of NKTR-255 along with a robust Ki-67 response. Repeat dosing of NKTR-255 generates similar NK cell numbers compared to a single dose in mice.

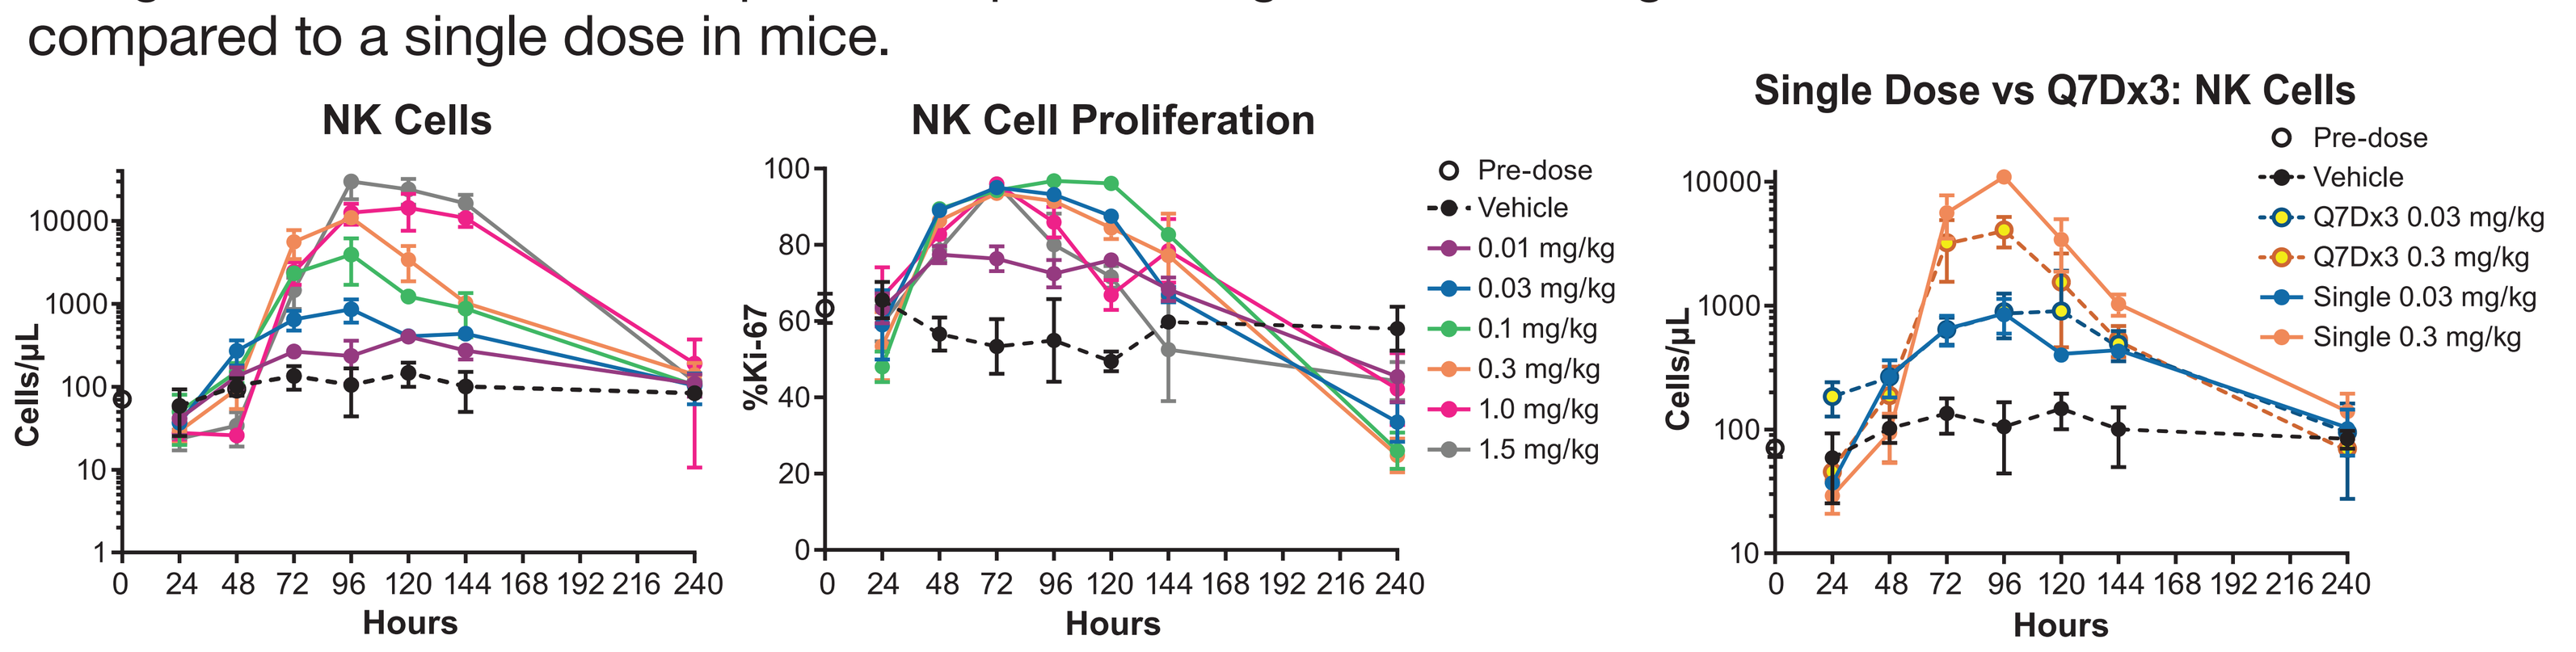
NK Cells
NK Cell Proliferation
Single Dose vs Q7Dx3: NK Cells
Cells/μL
%Ki-67
Hours
Pre-dose
Vehicle
0.01 mg/kg
0.03 mg/kg
0.1 mg/kg
0.3 mg/kg
1.0 mg/kg
1.5 mg/kg
Q7Dx3 0.03 mg/kg
Q7Dx3 0.3 mg/kg
Single 0.03 mg/kg
Single 0.3 mg/kg

Figure 5. NKTR-255 increases NK cell cytotoxicity.

Splenic NK cells isolated from NKTR-255-treated mice exhibit enhanced cytolytic activity against target cells, both in strength and duration, compared to conventional IL-15. Cytotoxicity increased with Granzyme B expression assessed in blood.

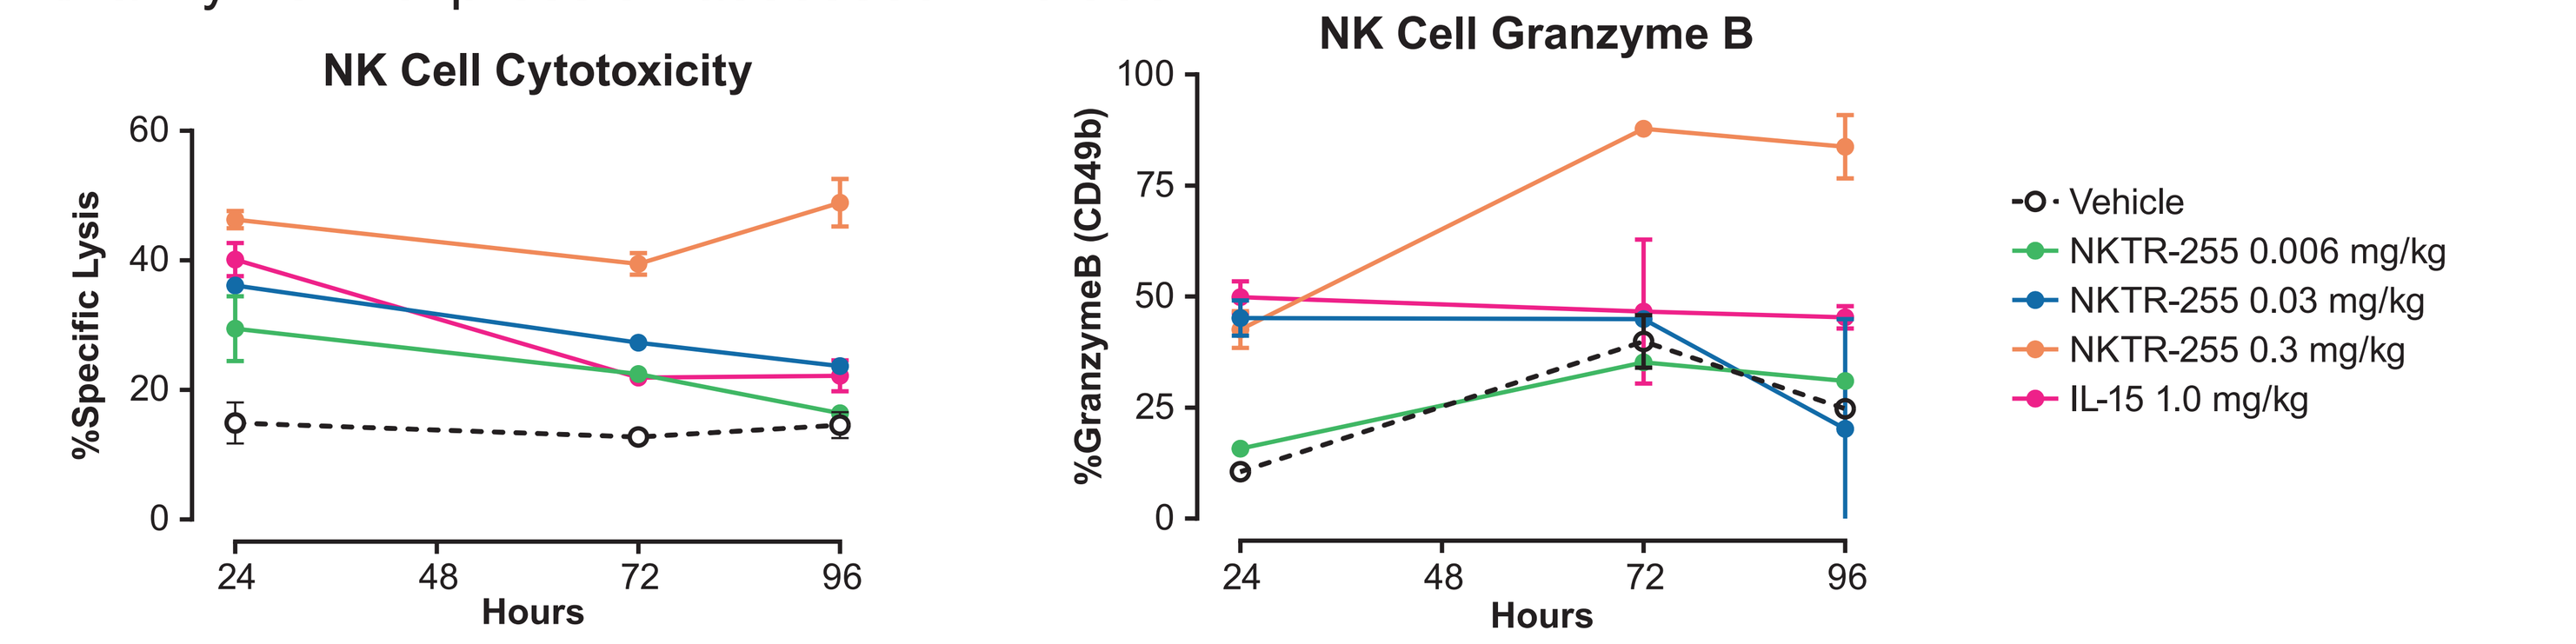
NK Cell Cytotoxicity
NK Cell Granzyme B
%Specific Lysis
%GranzymeB (CD49b)
Hours
Vehicle
NKTR-255 0.006 mg/kg
NKTR-255 0.03 mg/kg
NKTR-255 0.3 mg/kg
IL-15 1.0 mg/kg

Figure 6. NKTR-255 exhibits single agent efficacy and extends survival.

NKTR-255 dosed twice, one week apart, significantly attenuated CT-26 metastasis to the lung and prolonged survival. This anti-metastasis effect was lost in mice depleted of NK cells.

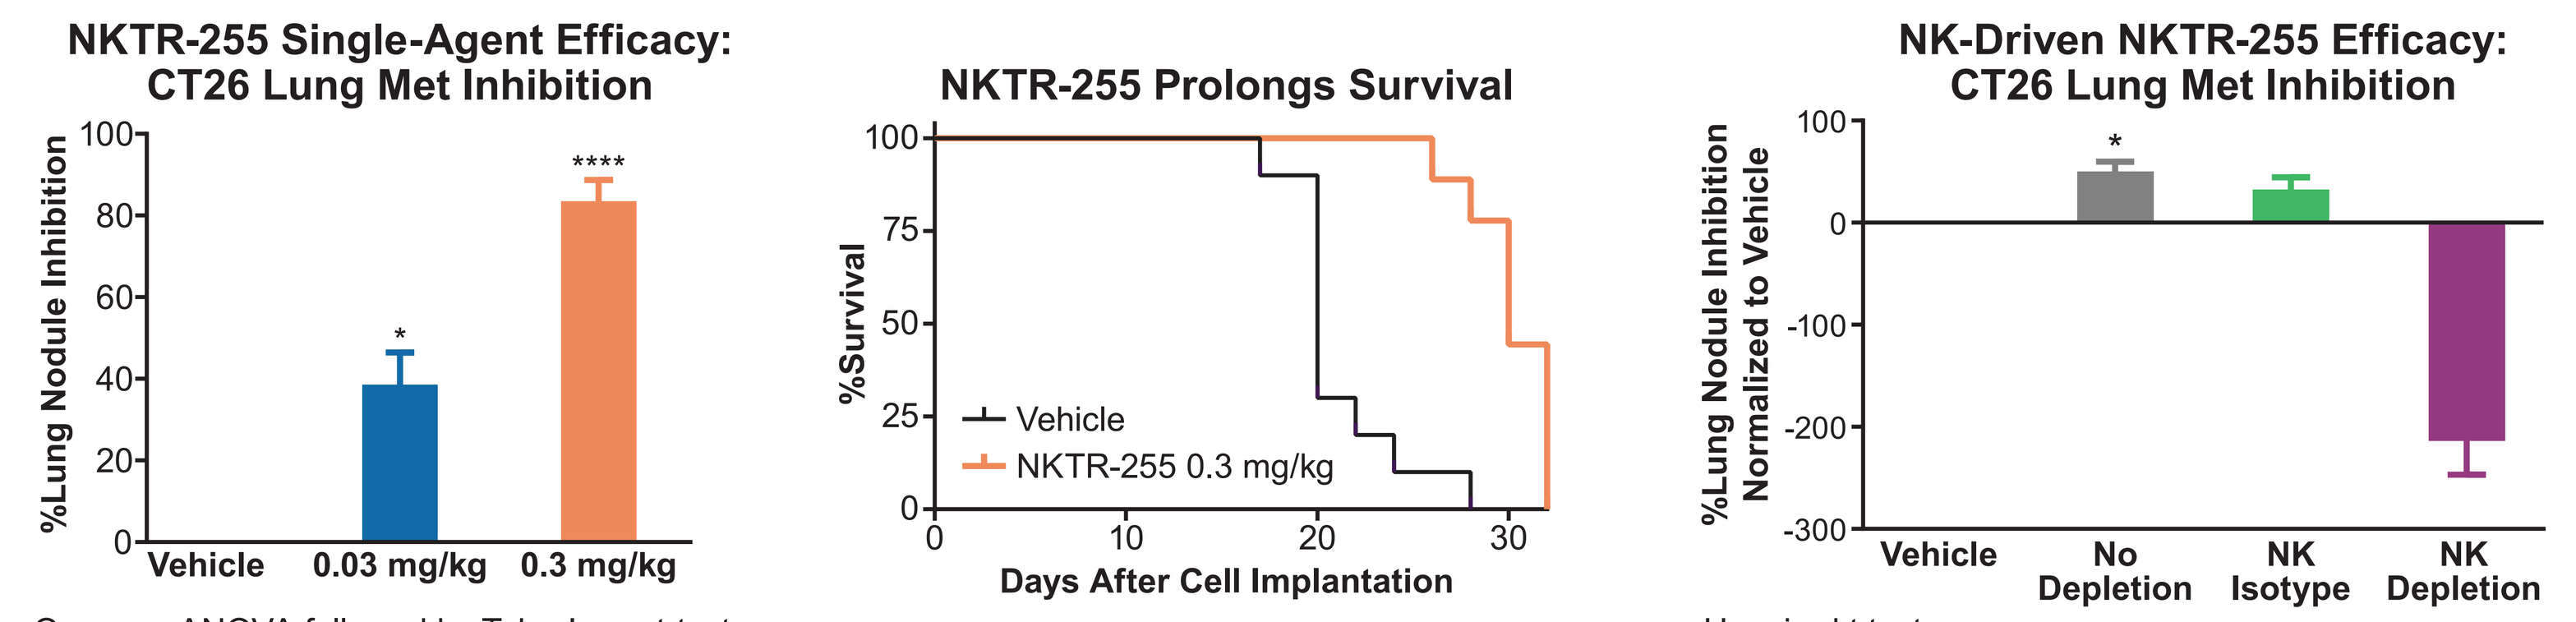
NKTR-255 Single-Agent Efficacy: CT26 Lung Met Inhibition
%Lung Nodule Inhibition
Vehicle
0.03 mg/kg
0.3 mg/kg
NKTR-255 Prolongs Survival
%Survival
Vehicle
NKTR-255 0.3 mg/kg
Days After Cell Implantation
NK-Driven NKTR-255 Efficacy: CT26 Lung Met Inhibition
%Lung Nodule Inhibition Normalized to Vehicle
Vehicle
No Depletion
NK Isotype
NK Depletion

One-way ANOVA followed by Tukey's post-test
*p<0.05; ****p<0.0001 vs. vehicle

Unpaired t test
*p<0.01 vs. vehicle

Figure 7. CD8 and memory subpopulations increase dramatically in cyno after single dose NKTR-255.

Cynomolgus monkeys (cyno) dosed with 0.1 mg/kg of NKTR-255 had a 7-10X increase in CD8 T cells and robust Ki-67 expression. CD8 Tcm and CD8 Tem increased 27-30X and 21-33X, respectively.

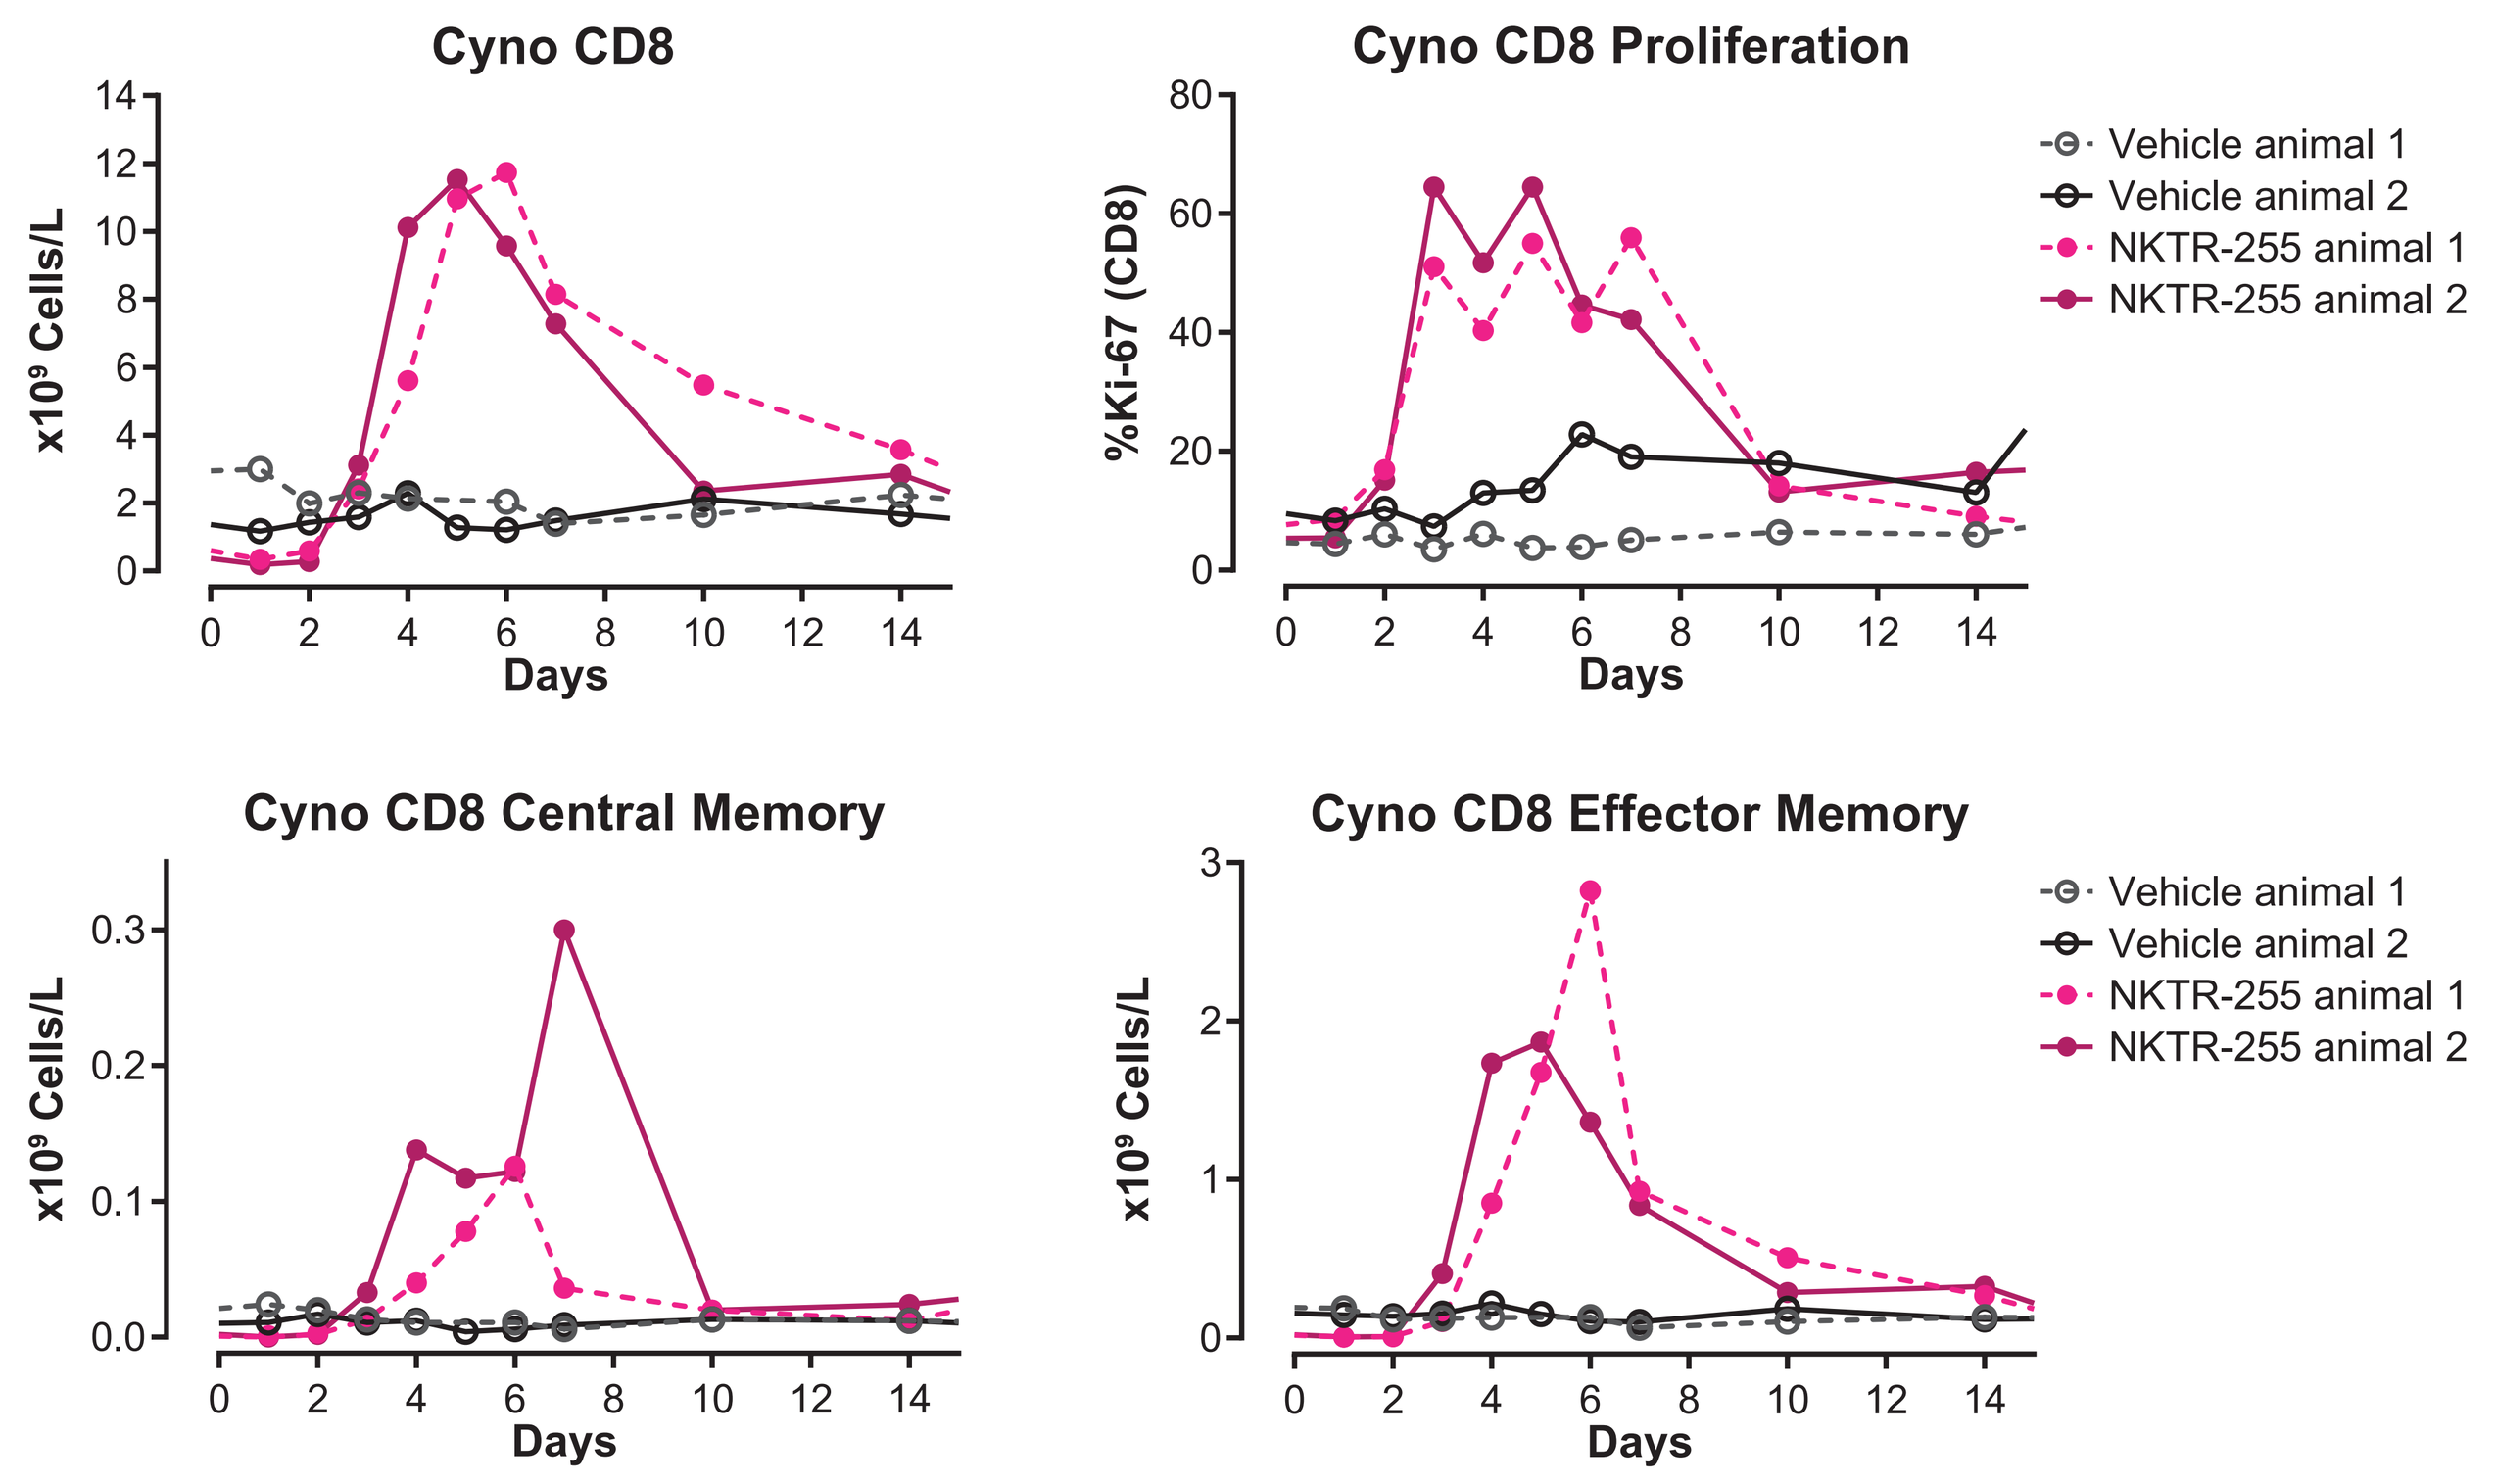
Cyno CD8
Cyno CD8 Proliferation
Cyno CD8 Central Memory
Cyno CD8 Effector Memory
x10⁹ Cells/L
%Ki-67 (CD8)
Days
Vehicle animal 1
Vehicle animal 2
NKTR-255 animal 1
NKTR-255 animal 2

Figure 8. NKTR-255 drives NK cell proliferation in cyno.

NK cells increased 9-10X after NKTR-255 treatment and significant Ki-67 expression was observed.

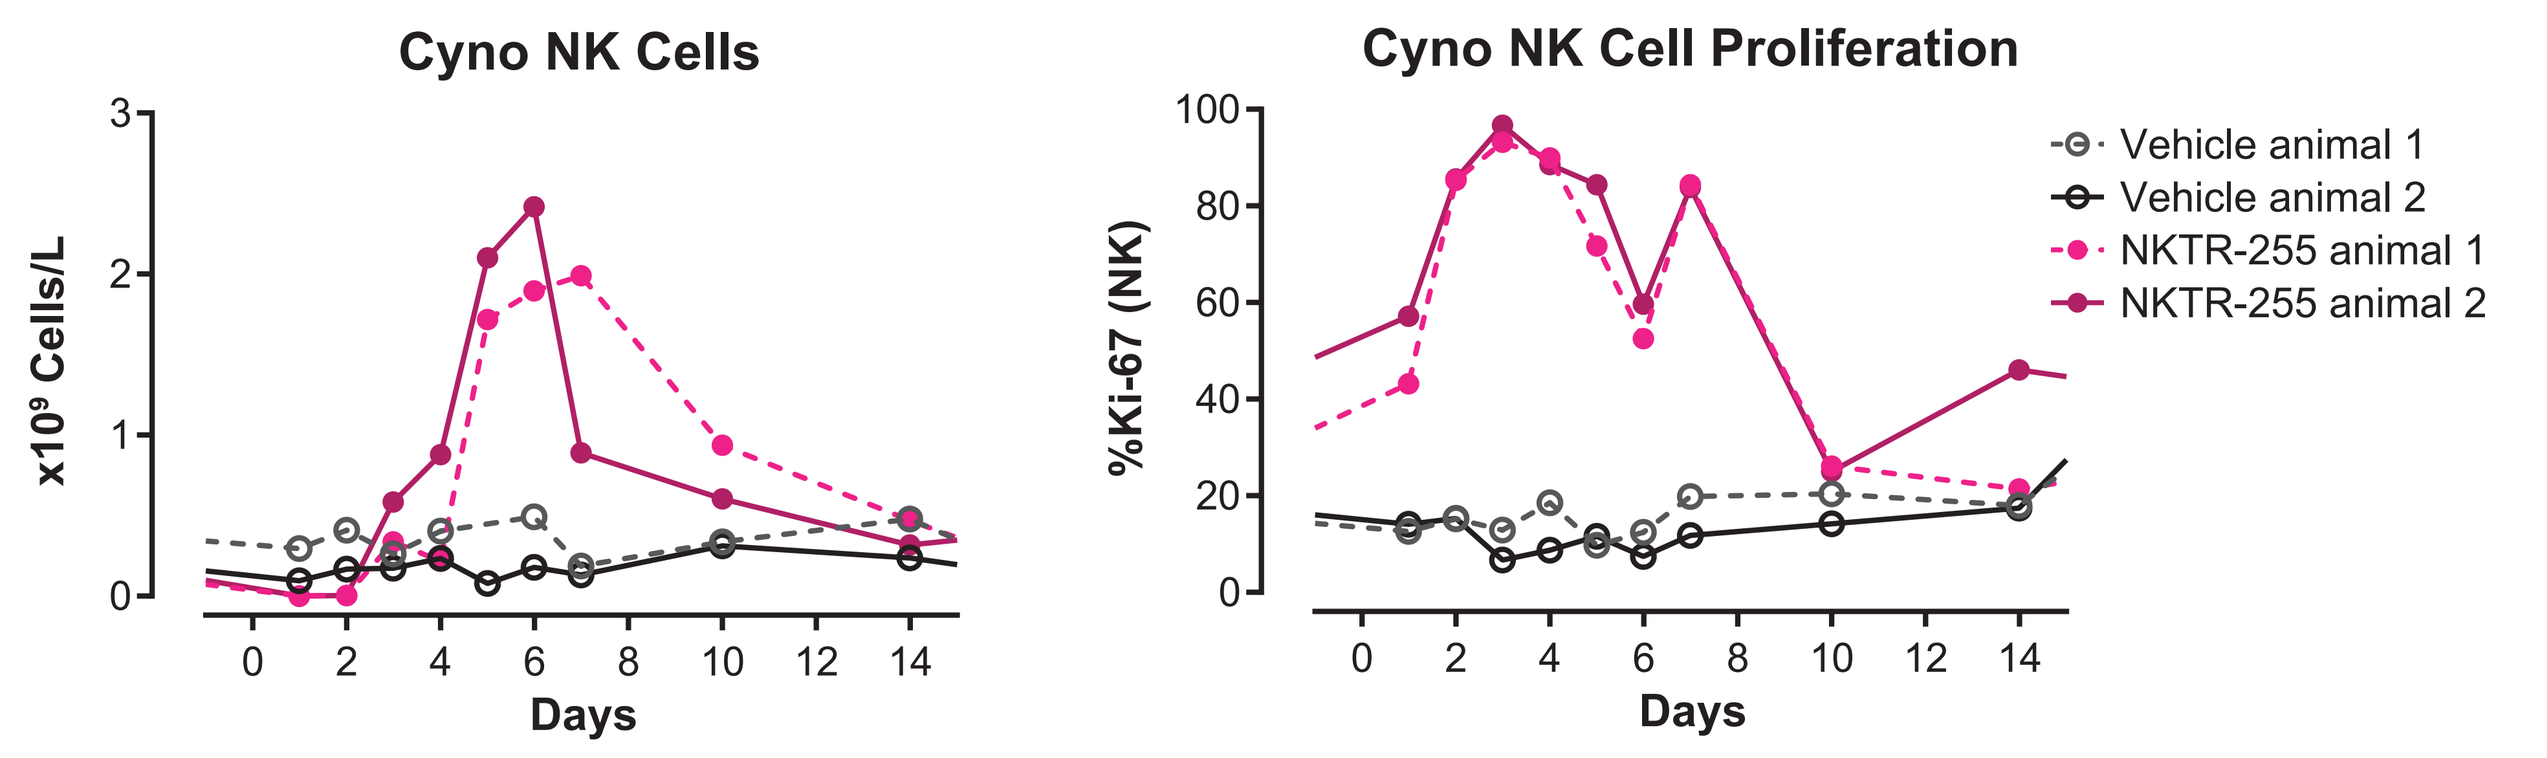
Cyno NK Cells
Cyno NK Cell Proliferation
x10⁹ Cells/L
%Ki-67 (NK)
Days
Vehicle animal 1
Vehicle animal 2
NKTR-255 animal 1
NKTR-255 animal 2

Figure 9. CD8 and NK cells from human PBMC exhibit a robust pSTAT5 response to NKTR-255 treatment.

NKTR-255 is 5.5X and 15X less potent than IL-15 in CD8 and CD56 bright NK cells, respectively. However, NKTR-255 achieves the same maximum response as conventional IL-15.

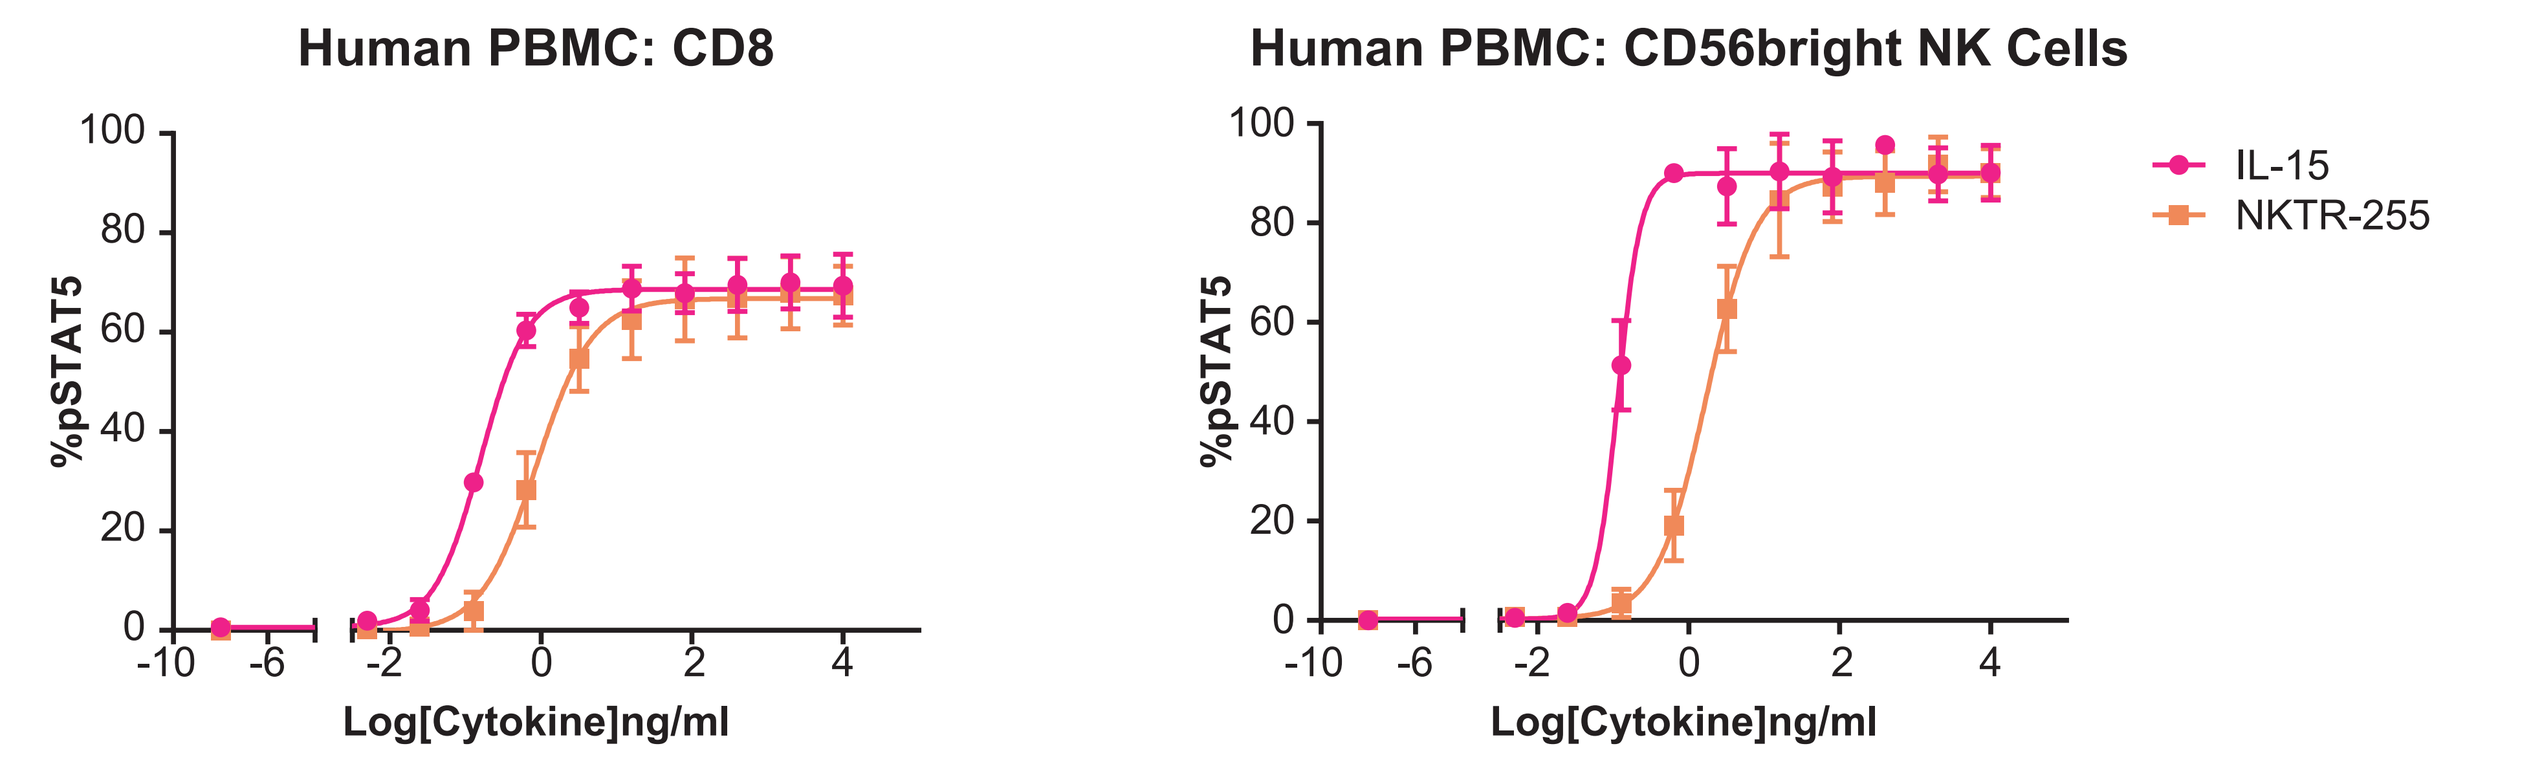
Human PBMC: CD8
Human PBMC: CD56bright NK Cells
%pSTAT5
Log[Cytokine]ng/ml
IL-15
NKTR-255

Compound	EC50 (ng/mL-%pSTAT5	
	CD8	CD56bright
IL-15	0.16	0.11
NKTR-255	0.87	1.7

CONCLUSIONS

- NKTR-255 extends plasma exposure and engages the IL-15 pathway, inducing robust signaling in a sustained manner unlike conventional IL-15
- Substantial proliferation of CD8, CD8 memory and NK cells was observed in rodents and non-human primates
- Repeat dosing did not reduce the magnitude of CD8 and NK cell responses in mice
- NKTR-255 has single-agent efficacy in a murine lung metastasis model, attenuating lung nodule formation and prolonging survival; this effect was NK cell-dependent
- NKTR-255 translates well to the human system, supported by robust CD8 and NK STAT5 phosphorylation in human PBMCs
- NKTR-255 efficacy and tolerability support its further development for the clinic

REFERENCES

1) Marcais et al. Regulation of mouse NK cell development and function by cytokines. *Front. Immunol.* 12 Dec 2013.
2) Stonier and Schluns. Trans-presentation: a novel mechanism regulating IL-15 delivery and responses. *Immunol Lett.* 2010.

Poster #P332: Society for Immunotherapy of Cancer 2017 Annual Meeting