Pharmacokinetic and pharmacodynamic study of NKTR-255, a polymer-conjugated IL-15, in cynomolgus monkey

Takahiro Miyazaki, Peiwen Kuo, Mekhala Maiti, Palakshi Obalapur, Murali Addepalli, Werner Rubas, Paul Sims, Ping Zhang, Mario Marcondes, Loui Madakamutil, Jonathan Zalevsky

Nektar Therapeutics, San Francisco, California

NEKTAR

Introduction

Interleukin-15 (IL-15) is a common γc cytokine that activates and provides survival benefit to memory T and NK cells. IL-15 is predominantly produced by myeloid cells and its receptor is a heterotrimeric receptor consisting of the IL-15 receptor α subunit and IL-2/IL-15 receptor β, γ subunits. Exploiting the therapeutic value of native IL-15 has been challenging due to its unfavorable pharmacokinetic properties and undesirable tolerability profile. NKTR-255 is a polymer-conjugated human IL-15 that retains binding affinity to the α subunit of the IL-15 receptor and exhibits reduced clearance to provide a sustained pharmacodynamics response. Here we investigate the biological effects of NKTR-255 in naïve cynomolgus monkey.

IL-15-mediated signaling through cis- and trans-presentation[1]

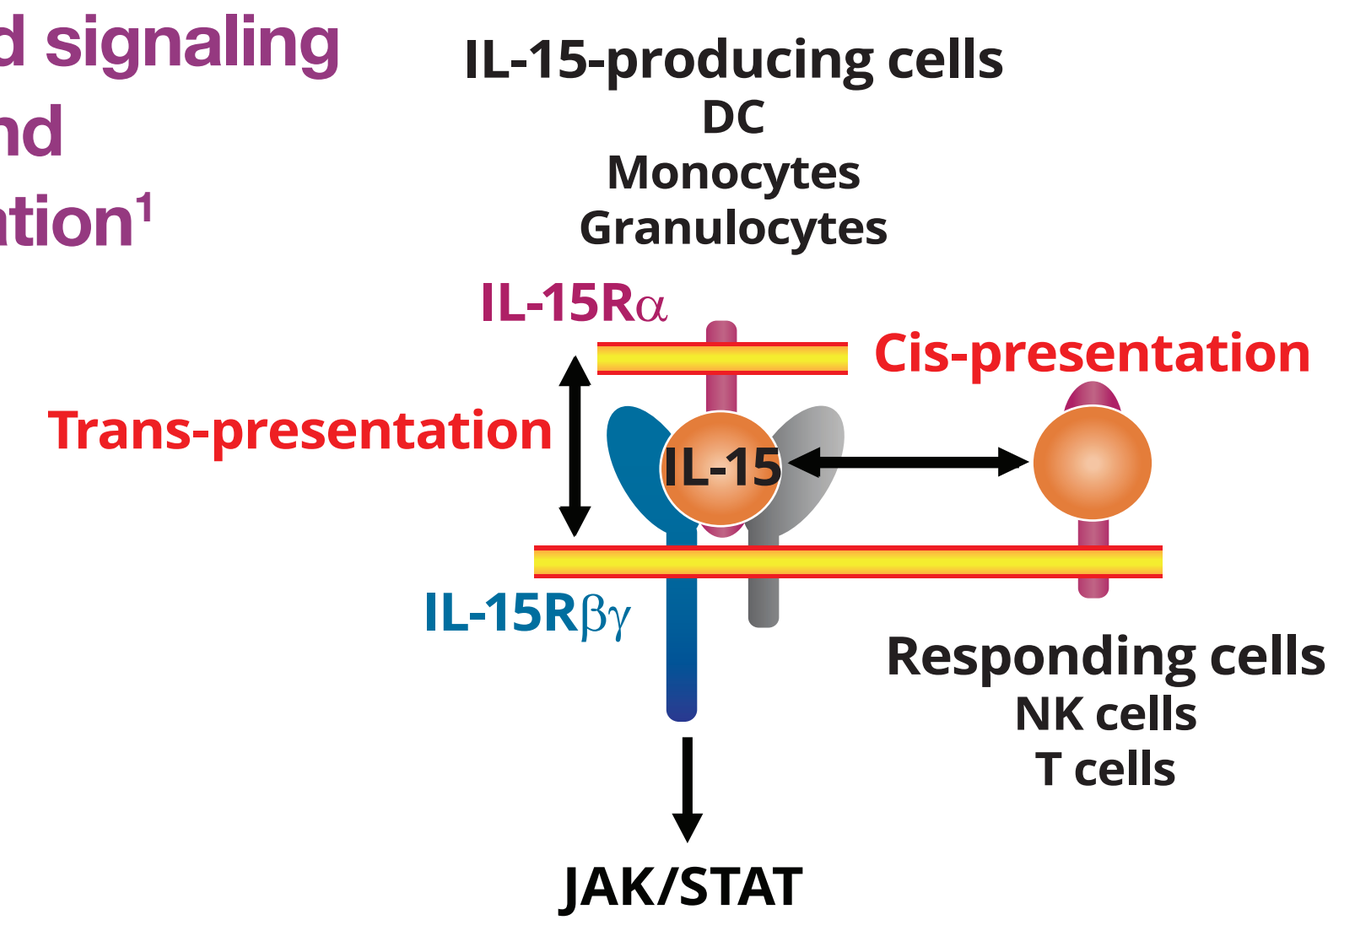
IL-15-producing cells
DC
Monocytes
Granulocytes
IL-15Rα
Cis-presentation
Trans-presentation
IL-15
IL-15Rβγ
Responding cells
NK cells
T cells
JAK/STAT

IL-15 binds the unique IL-15Rα chain and presents to the IL-2/IL-15Rβγ complex on the same (cis) or adjacent cell (trans). Engagement of the IL-2/IL-15R βγ complex can induce JAK-STAT signaling, increasing survival and proliferation. This process is crucial for the proper support of IL-15 biology.[2]

Methods

In vitro assay:

Cynomolgus monkey whole blood was stimulated with the indicated concentration of NKTR-255 or IL-15 for 20 minutes and the percentage of pSTAT5 positive populations in each of NK, CD4 T and CD8 T cells was determined by flow cytometry.

In vivo PK:

Male cynomolgus monkeys received single IV doses of 0.05 mg/kg IL-15 or 0.1 mg/kg NKTR-255. Blood samples were collected to determine the plasma concentrations of NKTR-255 by enzyme-linked immunosorbent assay.

In vivo PD:

Male cynomolgus monkeys received a single IV dose of NKTR-255 at 0.001, 0.01 or 0.1 mg/kg. Blood samples were collected to assess the effects of NKTR-255 on peripheral immune cell populations at multiple time points. Flow cytometry was used to measure STAT5 phosphorylation, Ki-67 expression, Granzyme B expression and frequency of cell populations.

Results

NKTR-255 induced dose-dependent phosphorylation of STAT5 in monkey whole blood (EC50 values NK cells: 6.9 ng/ml, CD8 T cells: 39 ng/ml, CD4 T cells: 53 ng/ml). The half-life and clearance of NKTR-255 were 26x longer and 38x lower, respectively, than of IL-15. NKTR-255 engaged the IL-15 signaling pathway, *in vivo*, demonstrating both robust and sustained STAT5 phosphorylation in lymphocytes. NKTR-255 drove the proliferation of total CD8 T cells and NK cells in a dose-dependent manner, with dramatic and durable increases observed in Ki67 positive population and absolute cell numbers (NK cells: 6.1 fold; CD8 T cells: 7.8 fold from baseline on day 5 at 0.1 mg/kg). These effects were strongly biased towards CD8 T cells and NK cells, with substantially less induction of CD4 T cells. The Ki67 response analyses of the T cell subpopulation revealed a higher response of memory populations than of naïve T cells. Among memory T cells, effector memory T cells showed the highest response over stem cell memory T cells and central memory T cells. NKTR-255 also increased the expression of Granzyme B in both NK and CD8 T cells, concomitant with an enhancement in target cell lysis. Finally, repeated administration of NKTR-255 sustained the Ki67 response of NK and CD8 T cells at levels similar to those observed at the single dose.

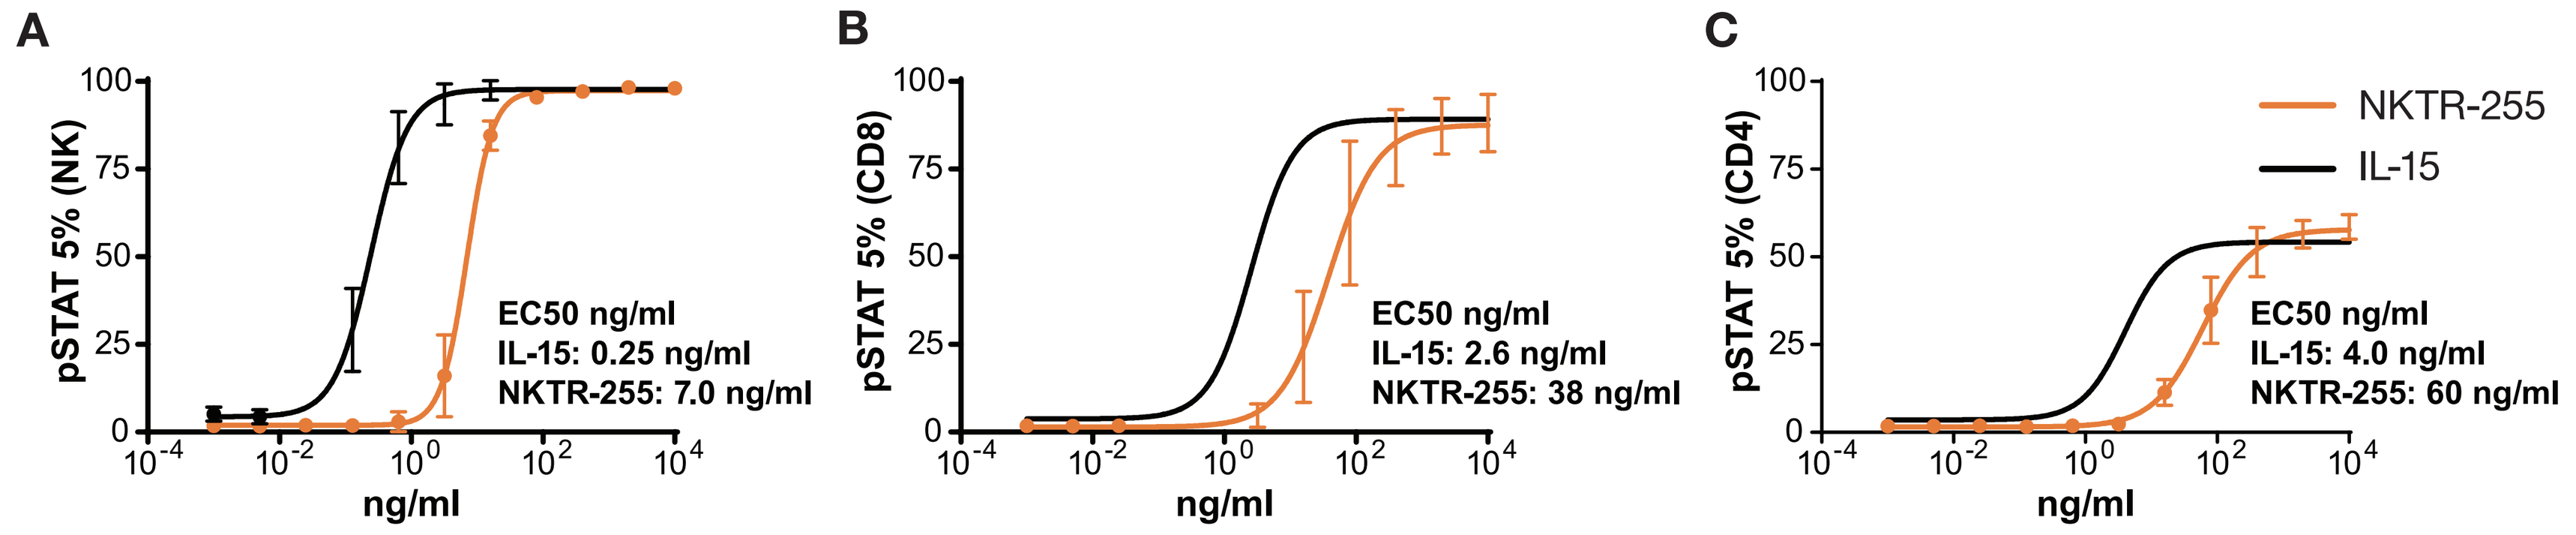
A
B
C
pSTAT 5% (NK)
pSTAT 5% (CD8)
pSTAT 5% (CD4)
ng/ml
EC50 ng/ml
IL-15: 0.25 ng/ml
NKTR-255: 7.0 ng/ml
EC50 ng/ml
IL-15: 2.6 ng/ml
NKTR-255: 38 ng/ml
EC50 ng/ml
IL-15: 4.0 ng/ml
NKTR-255: 60 ng/ml
NKTR-255
IL-15

Figure 1. *In vitro*, NKTR-255 showed a dose-dependent phosphorylation of STAT5 in NK cells, CD8 T cells and CD4 T cells
Monkey whole blood was stimulated with the indicated concentration of NKTR-255 or IL-15 for 20 minutes and the percentage of pSTAT5 positive populations in each NK (A), CD8 T (B) or CD4 T cells (C) was determined by flow cytometry.

Results

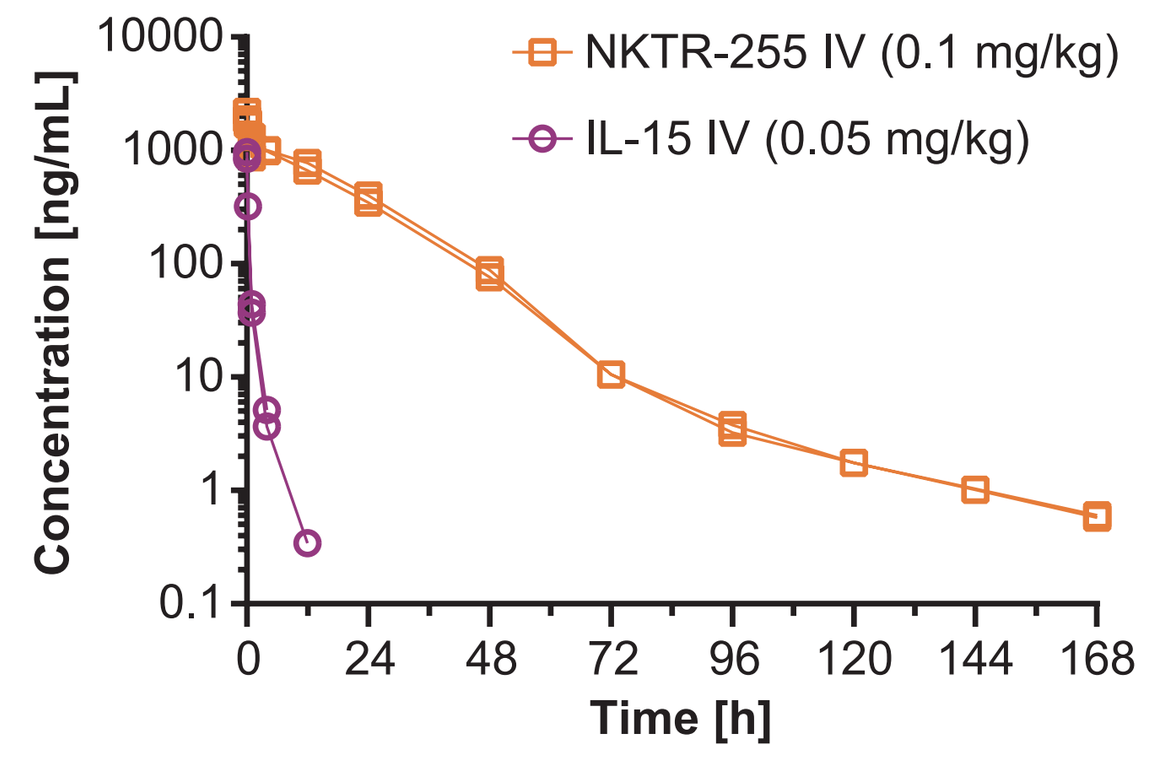
NKTR-255 IV (0.1 mg/kg)
IL-15 IV (0.05 mg/kg)
Concentration [ng/mL]
Time [h]

Test Article	IV Dose [mg/kg]	CL [mL/hr/kg]	$t_{1/2}$ [h]	Vd [mL/kg]
IL-15	0.05	161.4	1.2	112
NKTR -255	0.1	4.26	30.5	72.8

Figure 2. A single dose of NKTR-255 exhibited reduced clearance and longer half-life than IL-15
Monkeys received single IV doses of IL-15 at 0.05 mg/kg or NKTR-255 at 0.1 mg/kg. Blood samples were collected to determine the plasma concentrations of NKTR-255 or IL-15.

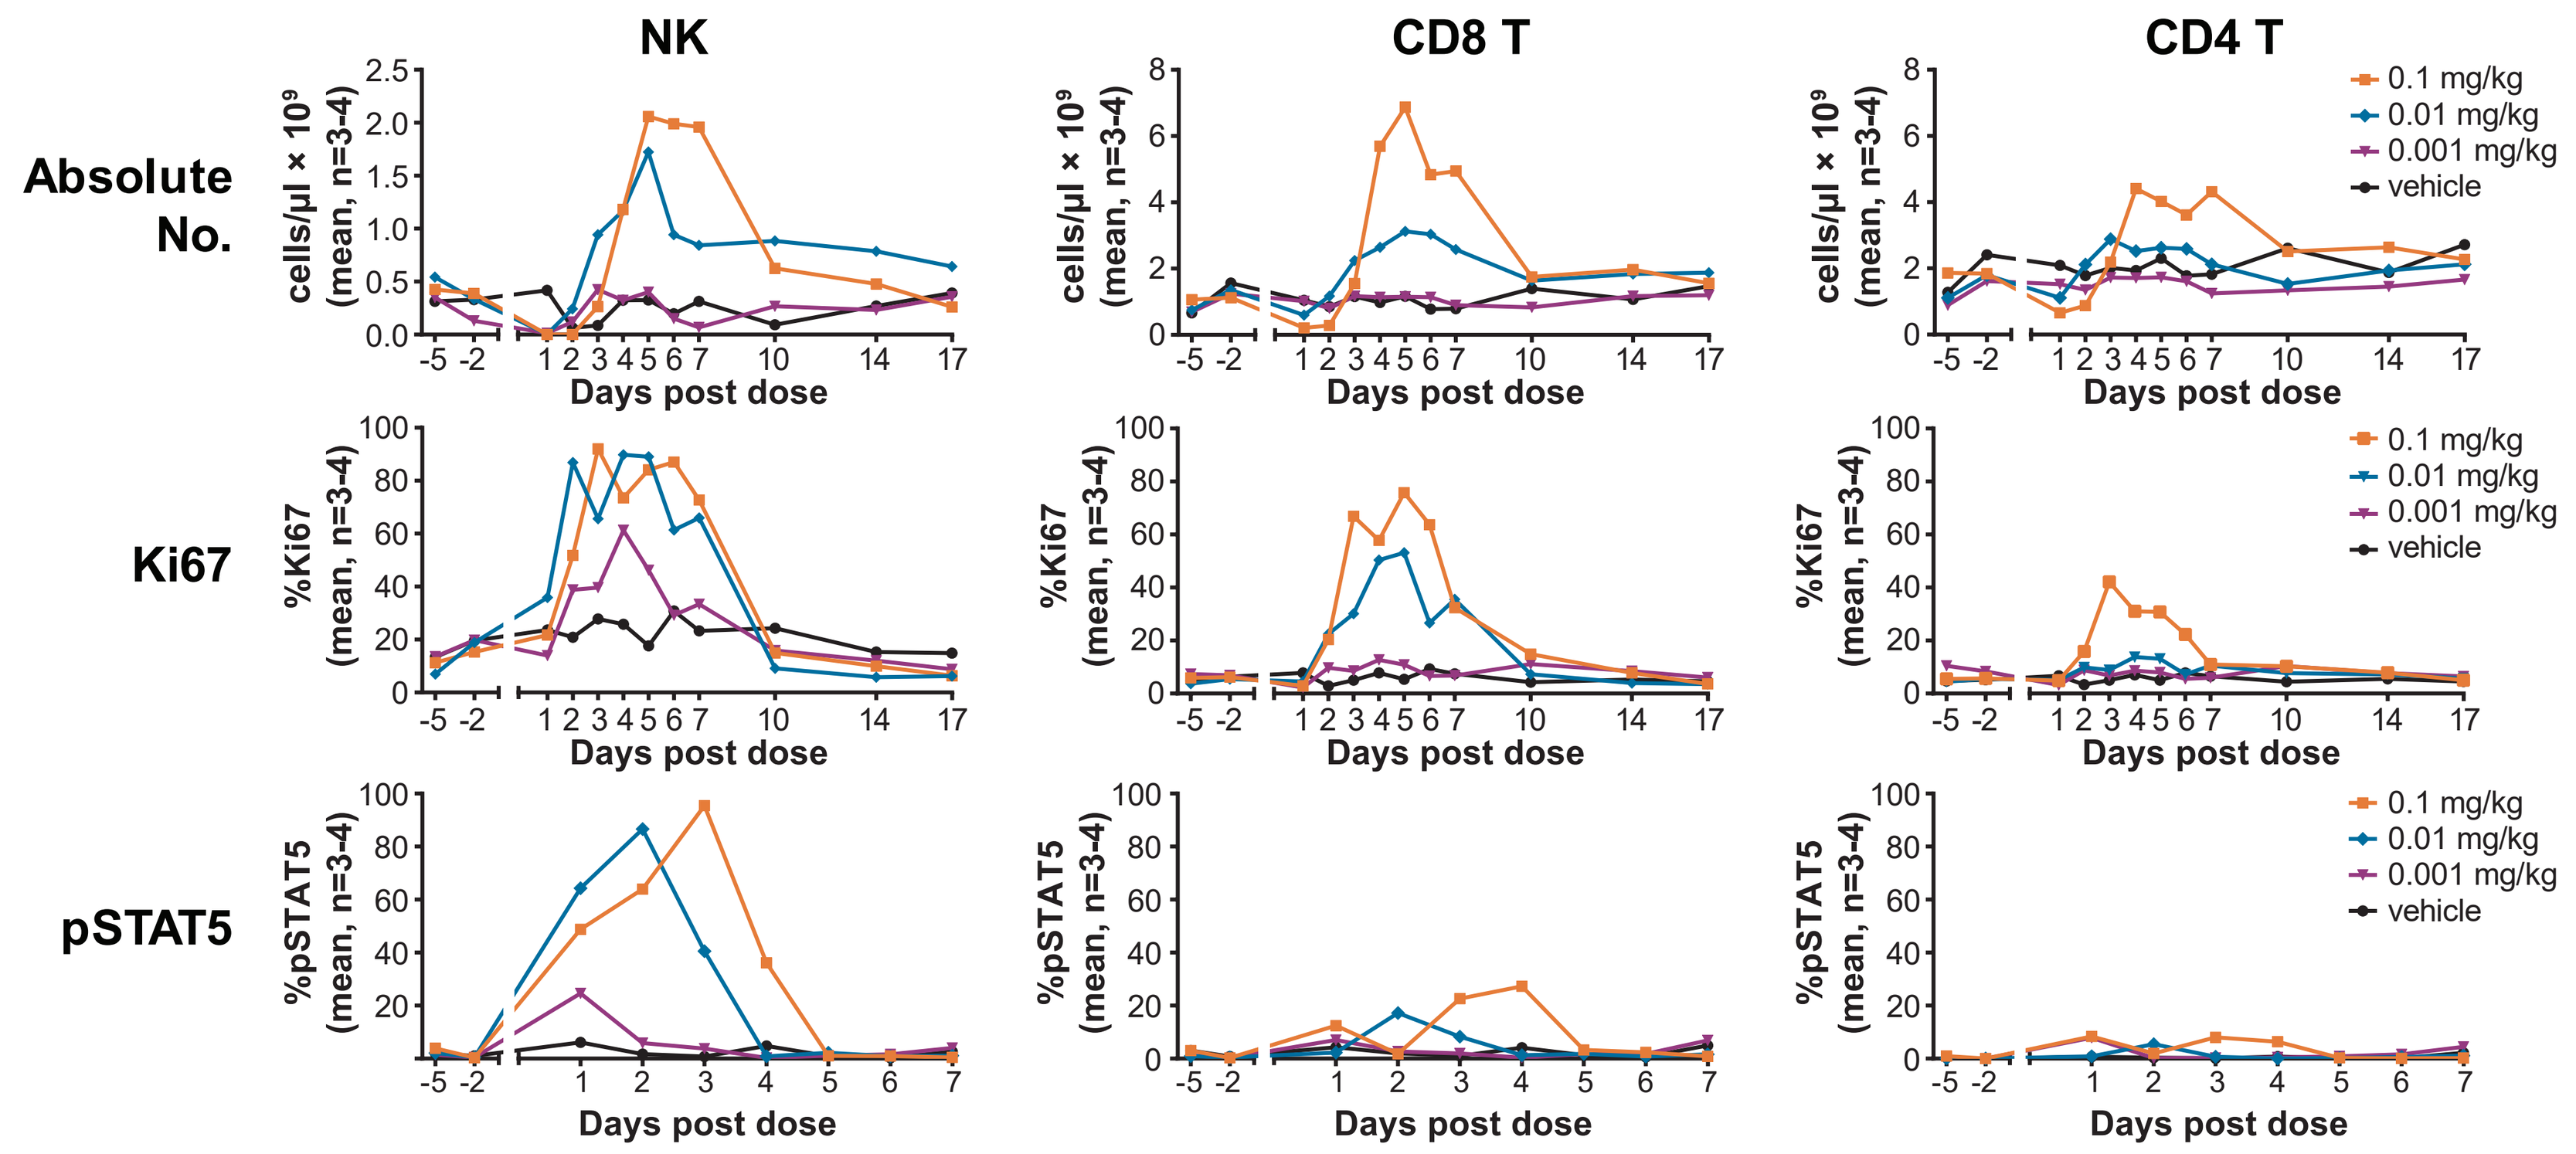
NK
CD8 T
CD4 T
Absolute No.
Ki67
pSTAT5
cells/µl × 10⁹ (mean, n=3-4)
%Ki67 (mean, n=3-4)
%pSTAT5 (mean, n=3-4)
Days post dose
0.1 mg/kg
0.01 mg/kg
0.001 mg/kg
vehicle

Figure 3. A single dose of NKTR-255 provided dominant effect to both NK cells and CD8 T cells, but not CD4 T cells
Monkeys received single IV doses of 0.001, 0.01 or 0.1 mg/kg of NKTR-255. Blood samples were collected to assess pSTAT5, Ki67 positive populations and the absolute number of peripheral NK, CD8 T or CD4 T cells by flow cytometry.

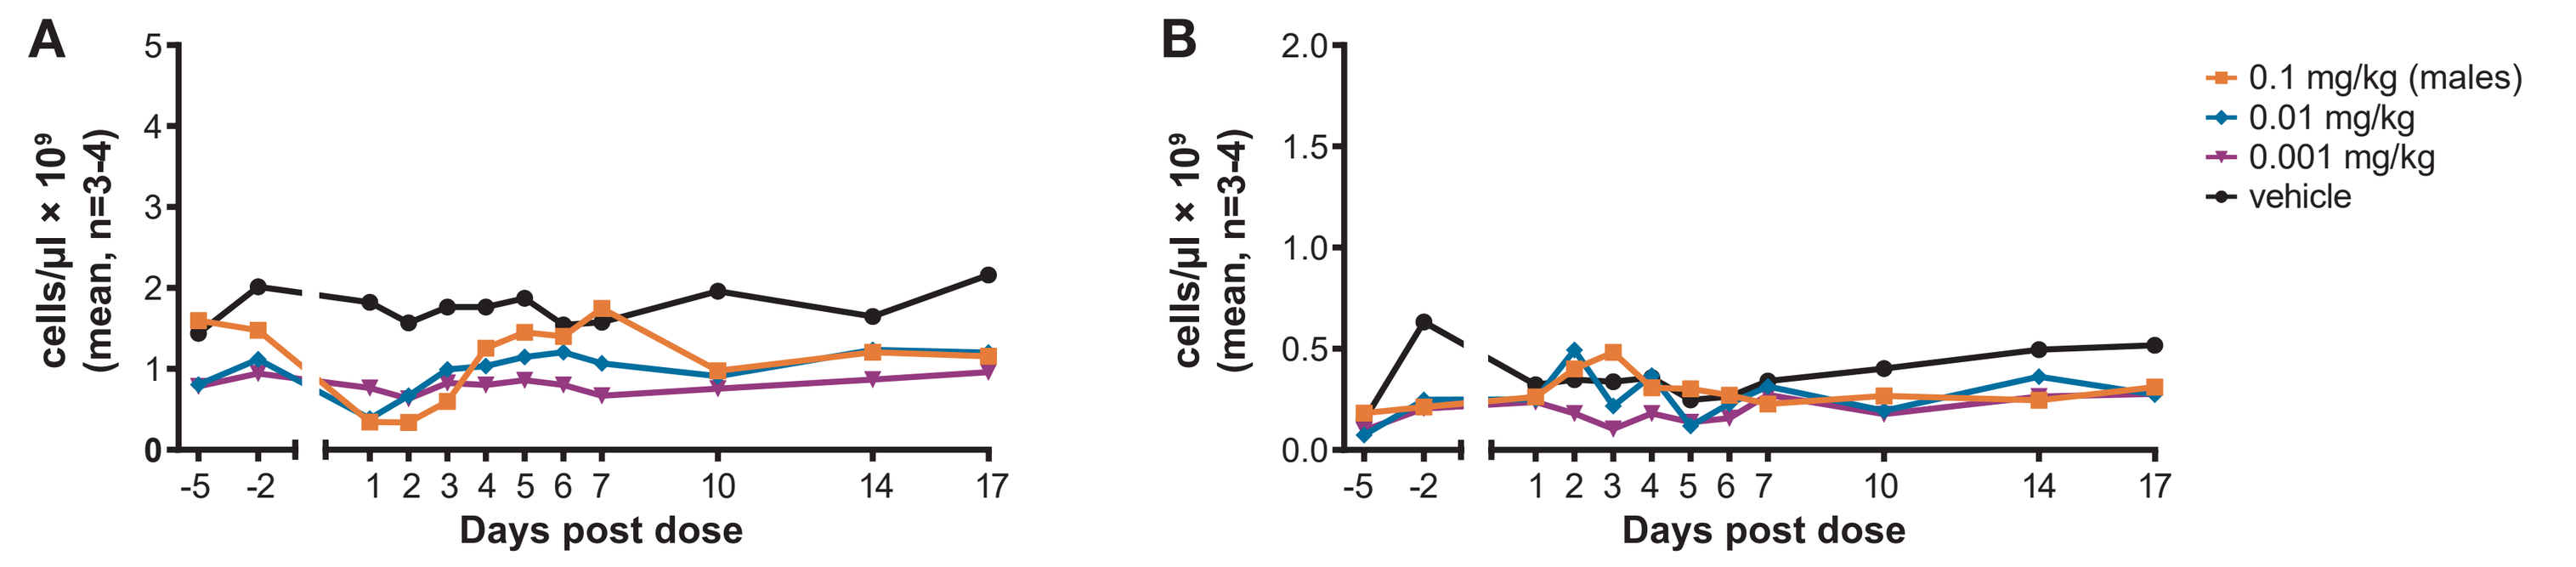
A
B
cells/µl × 10⁹ (mean, n=3-4)
Days post dose
0.1 mg/kg (males)
0.01 mg/kg
0.001 mg/kg
vehicle

Figure 4. A single dose of NKTR-255 did not significantly impact B cells and monocytes
Monkeys received single IV doses of 0.001, 0.01 or 0.1 mg/kg of NKTR-255. Blood samples were collected to assess the absolute number of peripheral B cells (A) or monocytes (B) by flow cytometry.

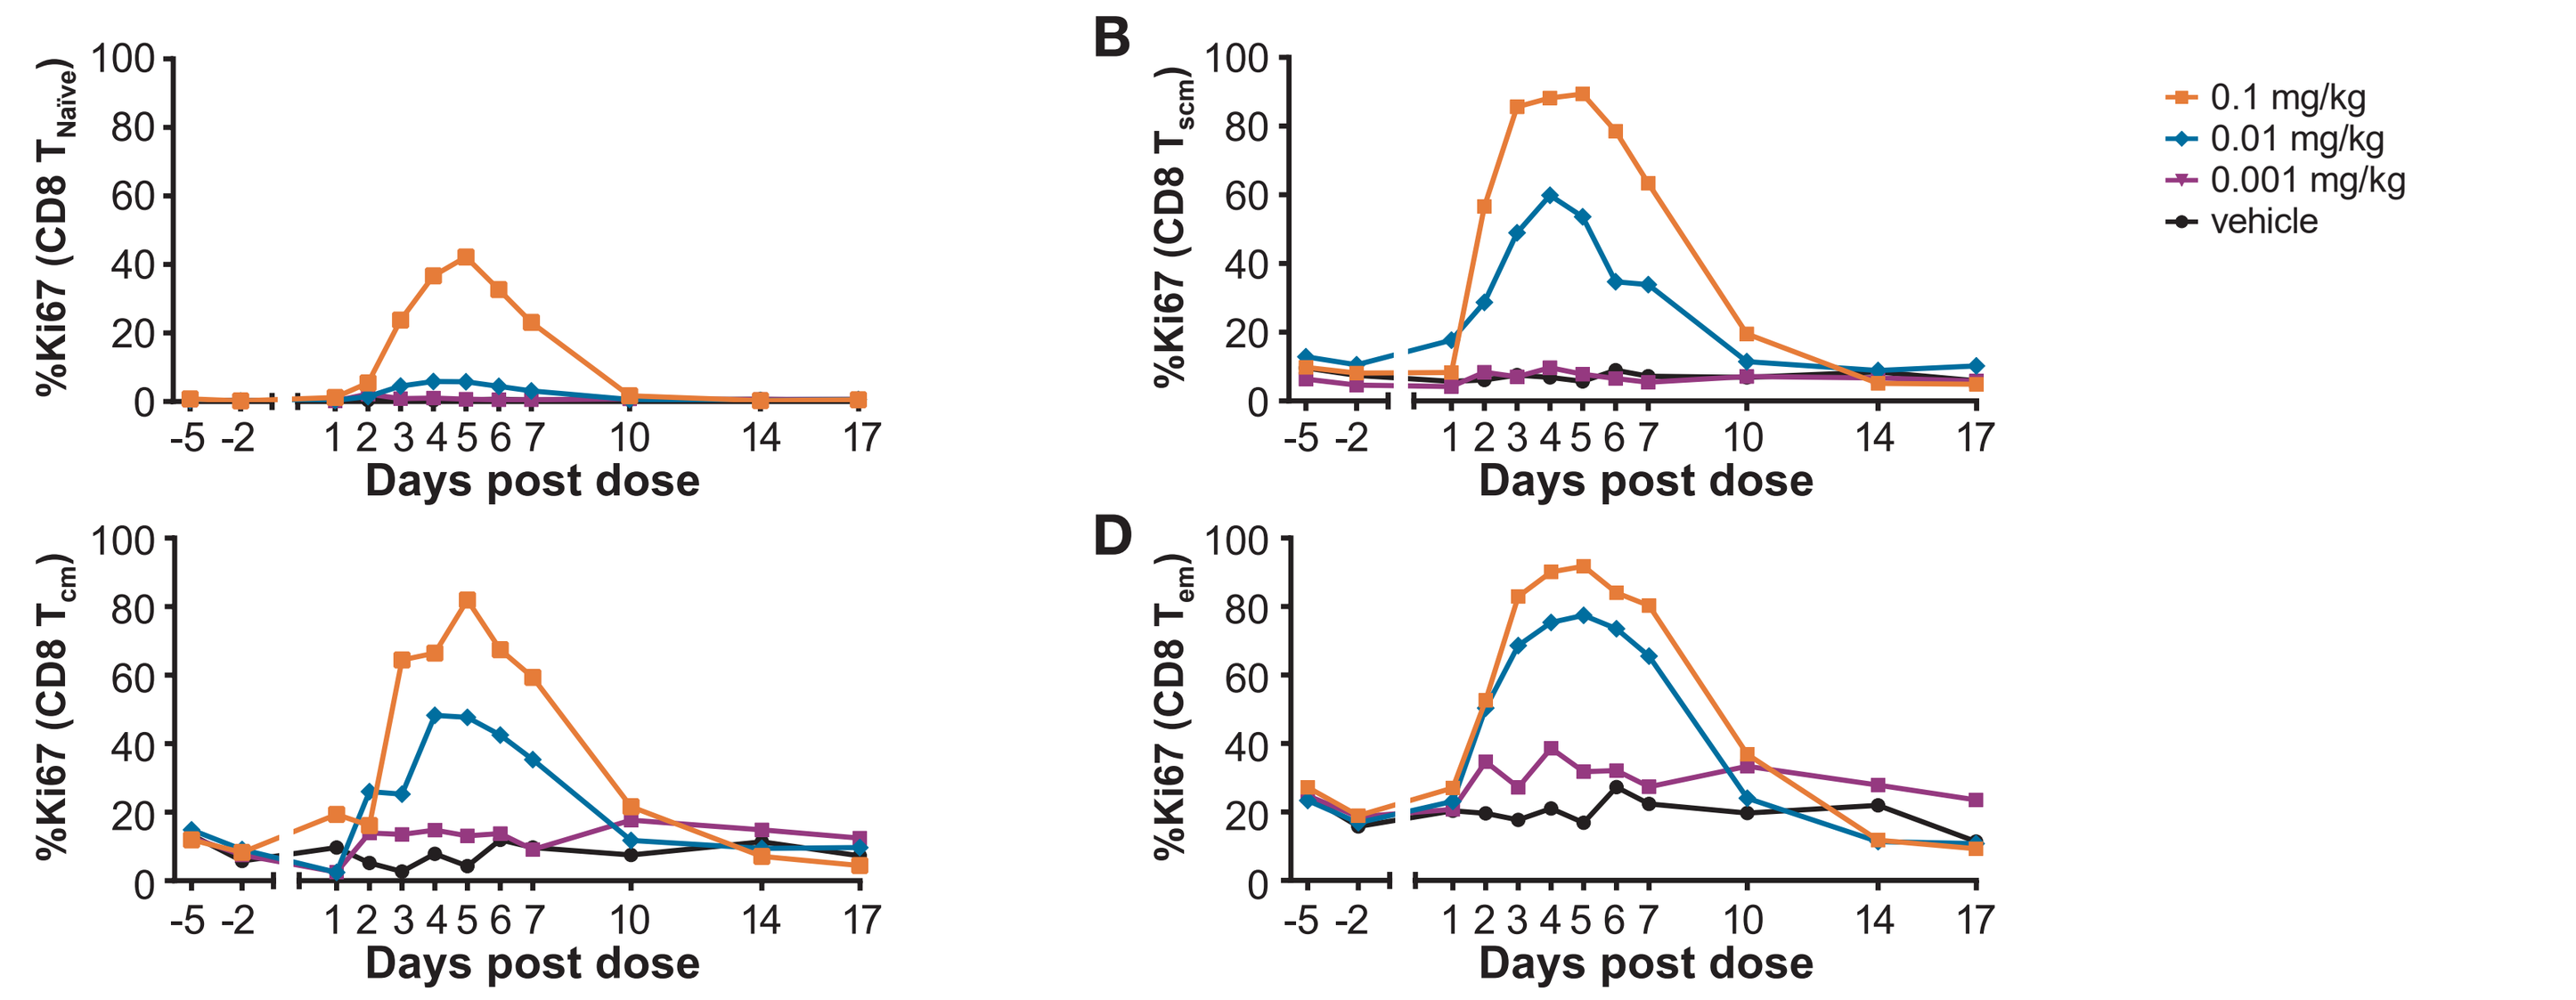
A
B
C
D
%Ki67 (CD8 T Naïve)
%Ki67 (CD8 T scm)
%Ki67 (CD8 T cm)
%Ki67 (CD8 T em)
Days post dose
0.1 mg/kg
0.01 mg/kg
0.001 mg/kg
vehicle

Figure 5. A single dose of NKTR-255 resulted in a higher response of memory CD8 T cell populations than of naïve CD8 T cells
Monkeys received single IV doses of 0.001, 0.01 or 0.1 mg/kg of NKTR-255. Blood samples were collected to assess Ki67 positive populations of naïve CD8 T cells (A), stem cell memory CD8 T cells (B), central memory CD8 T cells (C) or effector memory CD8 T cells (D) by flow cytometry.

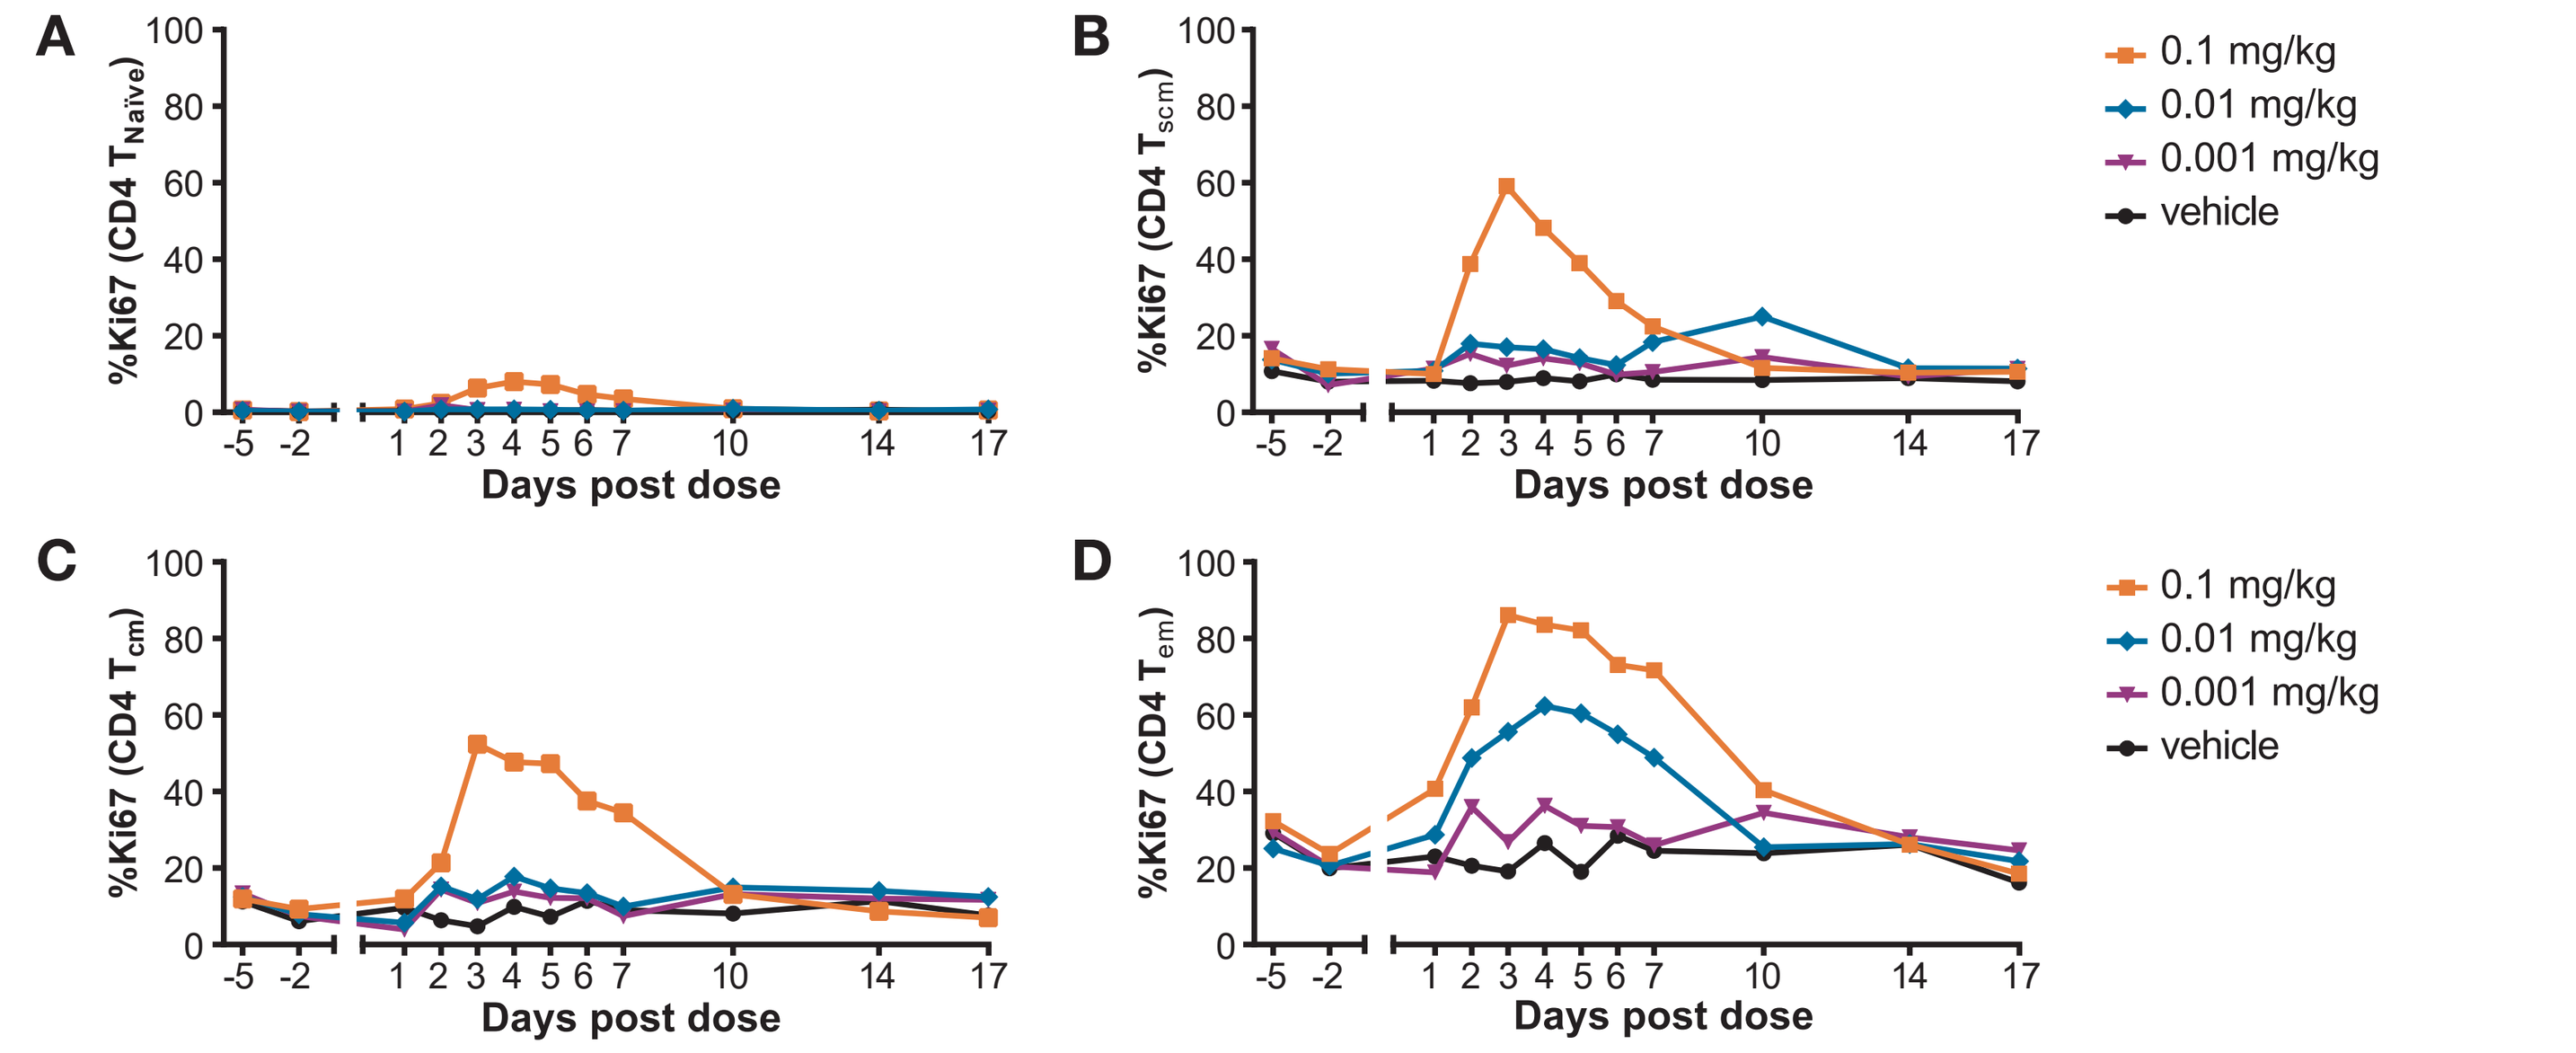
A
B
C
D
%Ki67 (CD4 T Naïve)
%Ki67 (CD4 T scm)
%Ki67 (CD4 T cm)
%Ki67 (CD4 T em)
Days post dose
0.1 mg/kg
0.01 mg/kg
0.001 mg/kg
vehicle

Figure 6. A single dose of NKTR-255 resulted in a higher response of memory CD4 T cell populations than of naïve CD4 T cells
Monkeys received single IV doses of 0.001, 0.01 or 0.1 mg/kg of NKTR-255. Blood samples were collected to assess Ki67 positive populations of naïve CD4 T cells (A), stem cell memory CD4 T cells (B), central memory CD4 T cells (C) or effector memory CD4 T cells (D) by flow cytometry.

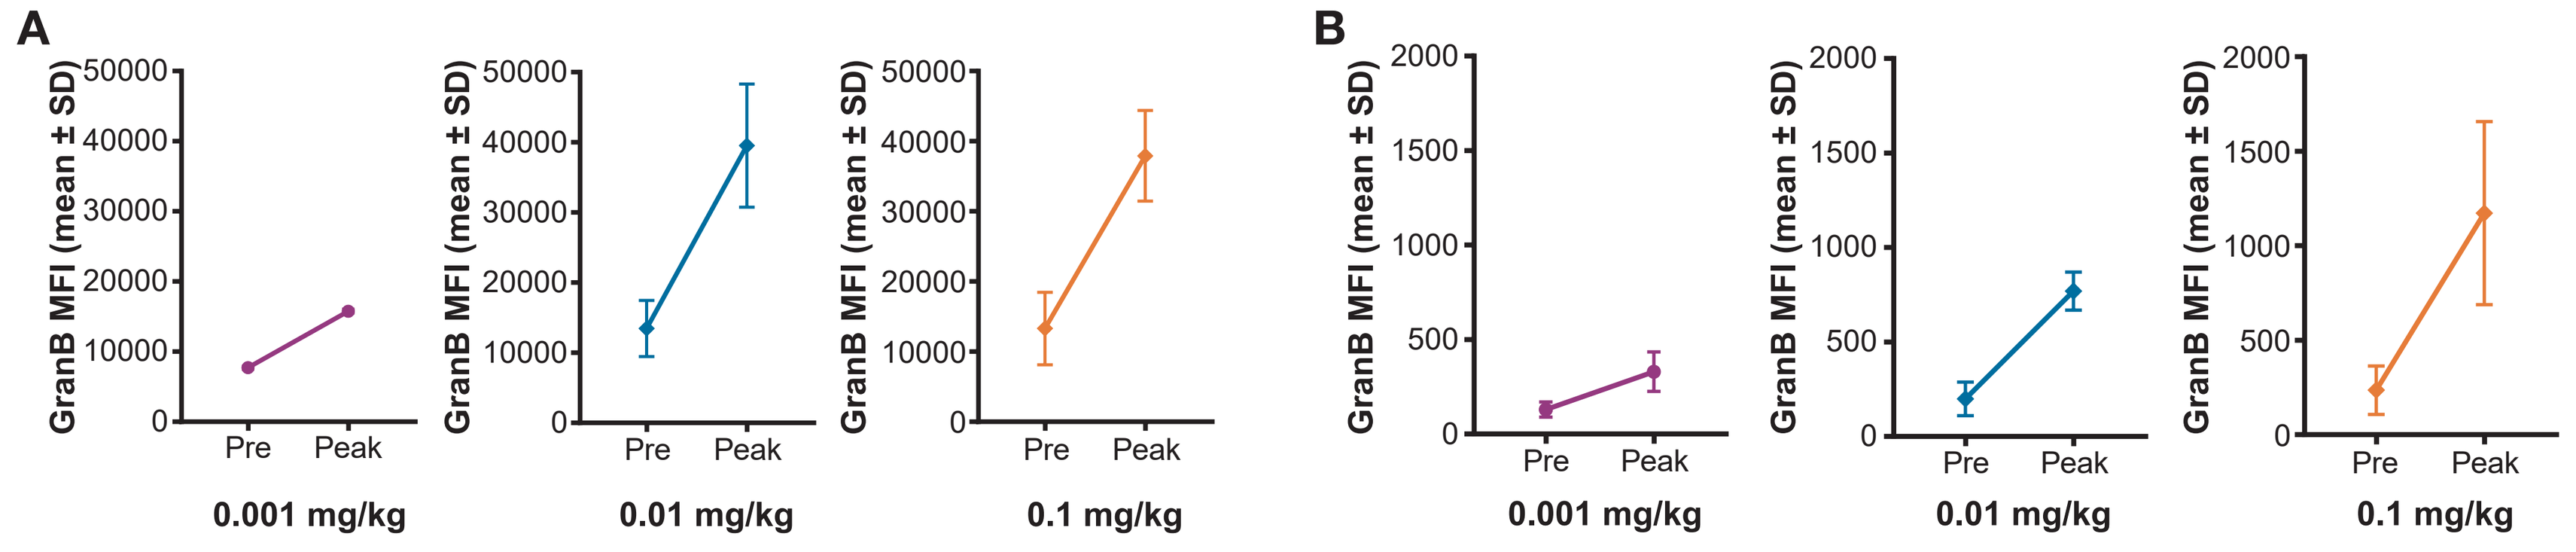
A
B
GranB MFI (mean ± SD)
Pre
Peak
0.001 mg/kg
0.01 mg/kg
0.1 mg/kg

Figure 7. A single dose of NKTR-255 increased Granzyme B on peripheral NK and CD8 T cells
Monkeys received single IV doses of 0.001, 0.01 or 0.1 mg/kg of NKTR-255. Blood samples were collected to assess Granzyme B expression on peripheral NK (A) or CD8 T (B) cells by flow cytometry.

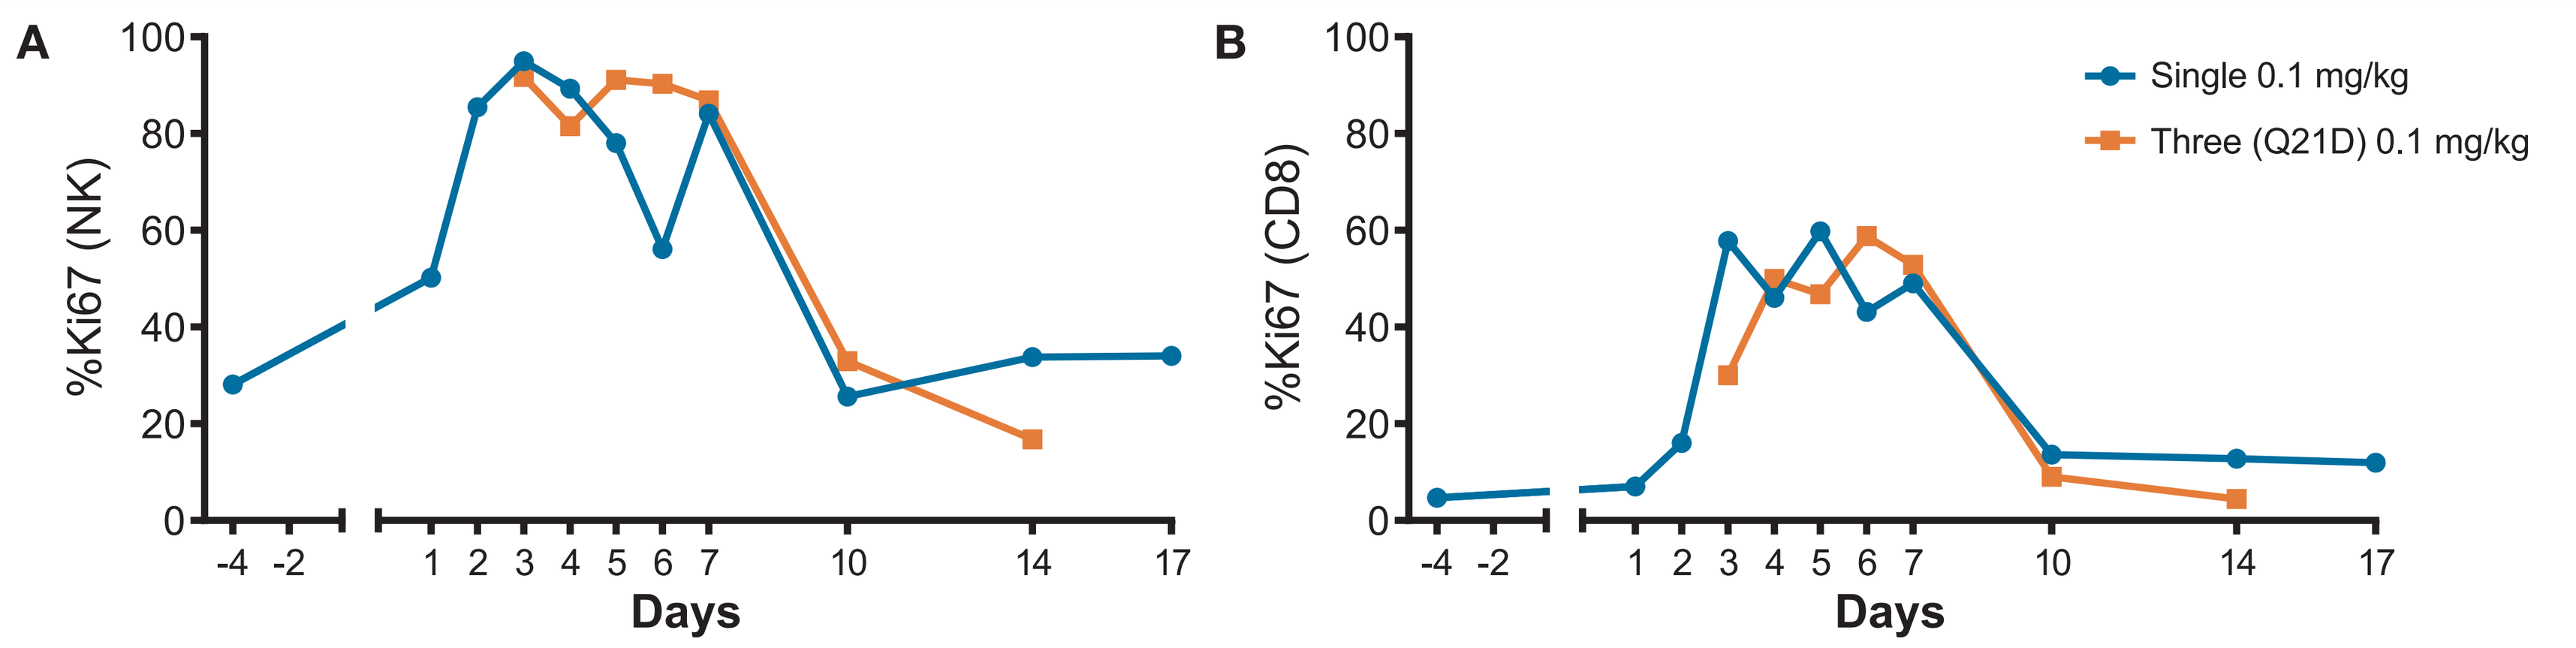
A
B
%Ki67 (NK)
%Ki67 (CD8)
Days
Single 0.1 mg/kg
Three (Q21D) 0.1 mg/kg

Figure 8. Repeat dosing of NKTR-255 did not reduce the magnitude of Ki67 responses in NK and T cells
Monkeys received single or three (Q21D) IV doses of 0.1 mg/kg of NKTR-255. Blood samples were collected after the first (single) or third (Q21Dx3) dose at the indicated time points and assessed for Ki67 positivity in NK cells (A) and CD8 T cells (B) by flow cytometry.

Conclusions

- NKTR-255 engages the JAK/STAT5 pathway with 10-fold less potency compared with IL-15
- A single dose of NKTR-255 exhibited reduced clearance and longer half-life than IL-15
- A single dose of NKTR-255 substantially enhances *in vivo* proliferation and activation of NK and CD8 T cells
- A single dose of NKTR-255 resulted in a higher response of memory populations than of naïve T cells
- A single dose of NKTR-255 increased levels of a cytotoxic enzyme in NK and CD8 T cells
- Repeat dosing of NKTR-255 does not reduce the magnitude of Ki67 responses in NK and T cells

References

1. Marcais et al., Regulation of mouse NK cell development and function by cytokines. *Front. Immunol.* 12 Dec 2013
2. Stonier and Schluns. Trans-presentation: a novel mechanism regulating IL-15 delivery and responses. *Immunol Lett.* 2010.

Presented at ASH Annual Meeting, December 2018