A Phase 2b, Randomized, Double-Blinded, Parallel-Group, Placebo-Controlled Study to Evaluate the Efficacy and Safety of Rezpegaldesleukin in Adults with Moderate-to-Severe Atopic Dermatitis

P0559

Jonathan Silverberg[1], Raj Chovatiya[2], David Rosmarin[3], Lisa Beck[4], Melinda Gooderham[5], Andrew Blauvelt[6], Stephen Schleicher[7], Sohail Chaudhry[8], Christie Fanton[8], Danni Yu[8], Yi Liu[8], Jenny Gilbert[8], Karyn Cassello[8], Connie Louie-Tse[8], Wang Waltz[8], Zachary Lee[8], Charleen Jue[8], Mario Marcondes[8], Brian Kotzin[8], Mary Tagliaferri[8], Jonathan Zalevsky[8], Thomas Bieber[9]

[1]Department Of Dermatology, George Washington University School Of Medicine, Washington, DC, United States; [2]Feinberg School Of Medicine, Northwestern University, Chicago, IL, United States; [3]Indiana University School of Medicine, Indianapolis, IN, United States; [4]University Of Rochester Medical Center, Rochester, NY, United States; [5]Department of Medicine, Queen's University, Kingston, ON, Canada; [6]Oregon Medical Research Center, Portland, United States; [7]DermDox Centers for Dermatology, Sugarloaf, PA, United States; [8]Nektar Therapeutics, San Francisco, CA, United States; [9]Christine Kühne Center For Allergy Research And Education, University Of Bonn, Bonn, Germany

BACKGROUND

- Atopic dermatitis (AD) is a chronic, relapsing inflammatory skin disorder.[1-3]
- Approximately 1 in 10 individuals has a lifetime risk of developing AD.[1-5]
- Dysfunction of regulatory T cells (Treg) may play a role in AD immunopathogenensis.[6]
- Targeting the Treg pathway is a novel therapeutic approach for restoring immune homeostasis in patients with moderate-to-severe AD.[7]
- Rezpegaldesleukin (REZPEG: NKTR-358) is a polyethylene glycol (PEG)-conjugated recombinant human interleukin 2 (rhIL-2) with the ability to selectively promote the activation and expansion of Tregs, while having relatively minimal effect on conventional T cells (Tcons).[7]
- In healthy volunteers and patients with SLE, REZPEG treatment resulted a in dose-dependent, selective, and up-to 17-fold increase in CD25^{bright} Tregs over baseline that was sustained for 20–30 days.[7]
- A Phase 1b study of REZPEG for patients with moderate-to-severe AD demonstrated a rapid time to response (2-4 weeks) during induction therapy and a prolonged durability of response (i.e., majority of responders retained that response throughout the 36-week follow-up without additional systemic therapy).[8]
- These results support further development of REZPEG for patients with AD.

Figure 1: Role of Regulatory T Cells in Autoimmune Disease

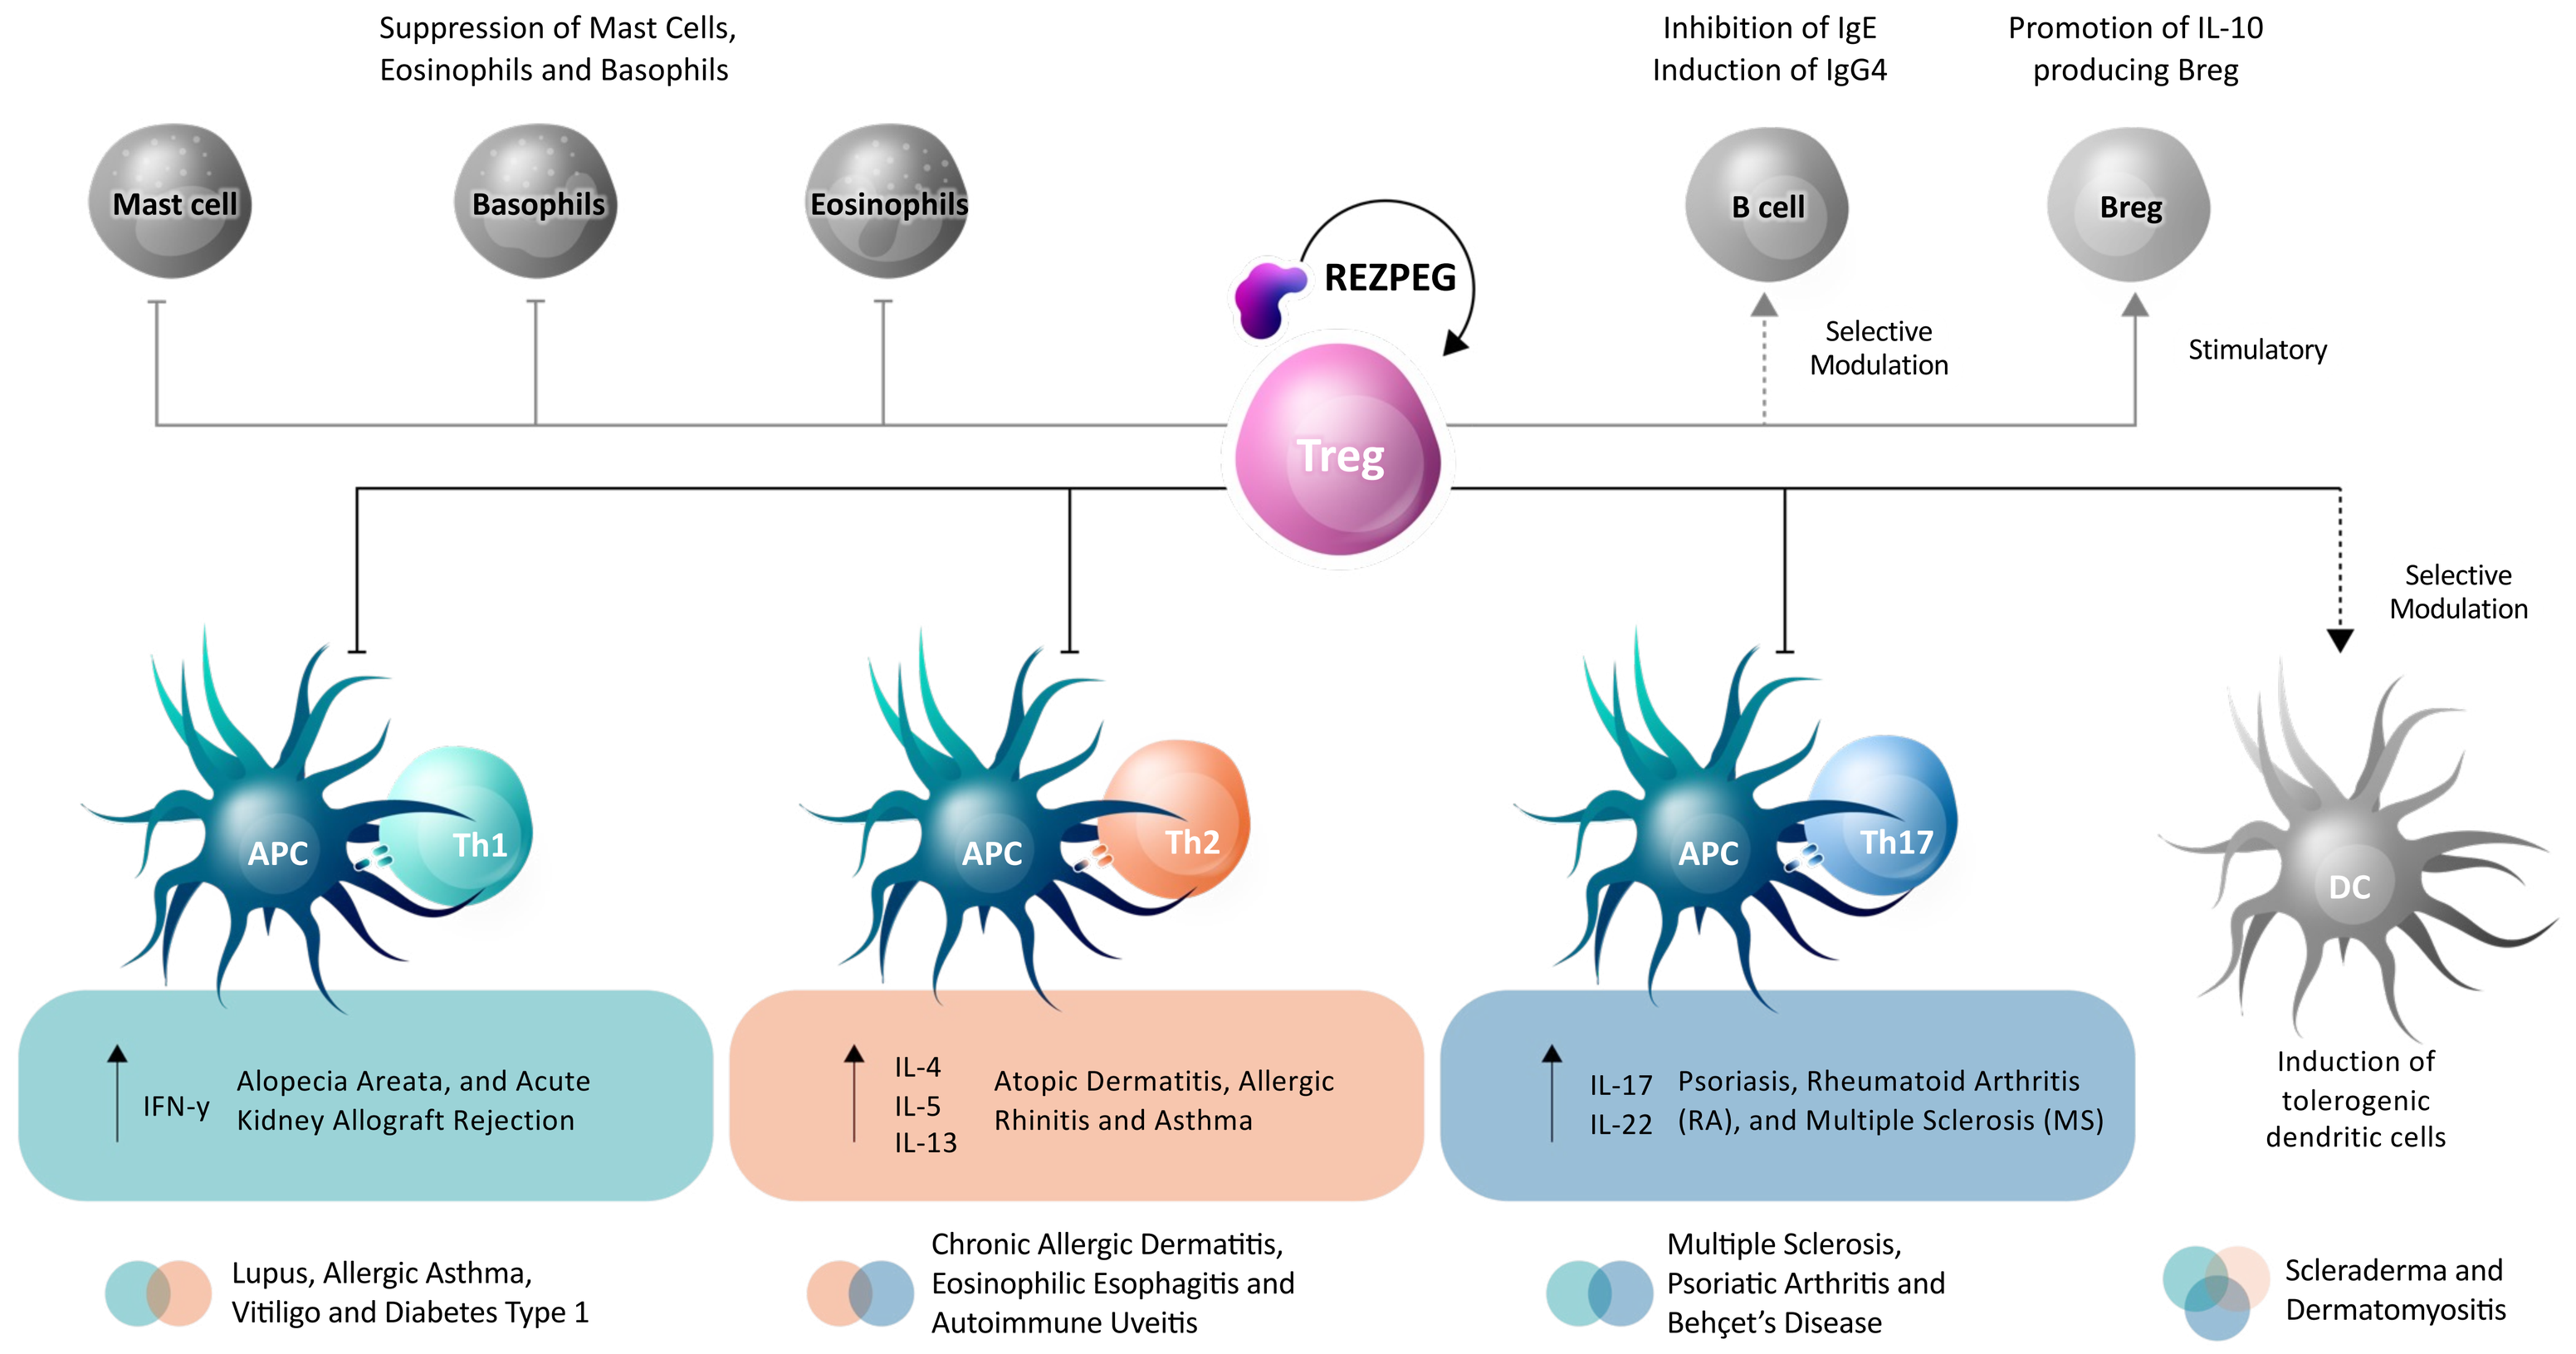
Suppression of Mast Cells, Eosinophils and Basophils
Mast cell
Basophils
Eosinophils
REZPEG
Treg
Inhibition of IgE Induction of IgG4
B cell
Selective Modulation
Promotion of IL-10 producing Breg
Breg
Stimulatory
Selective Modulation
APC
Th1
APC
Th2
APC
Th17
DC
IFN-γ
Alopecia Areata, and Acute Kidney Allograft Rejection
IL-4
IL-5
IL-13
Atopic Dermatitis, Allergic Rhinitis and Asthma
IL-17
IL-22
Psoriasis, Rheumatoid Arthritis (RA), and Multiple Sclerosis (MS)
Induction of tolerogenic dendritic cells
Lupus, Allergic Asthma, Vitiligo and Diabetes Type 1
Chronic Allergic Dermatitis, Eosinophilic Esophagitis and Autoimmune Uveitis
Multiple Sclerosis, Psoriatic Arthritis and Behçet's Disease
Scleraderma and Dermatomyositis

Figure 2: Phase 1b Study of REZPEG in Atopic Dermatitis
Percent Change From Baseline for EASI Score

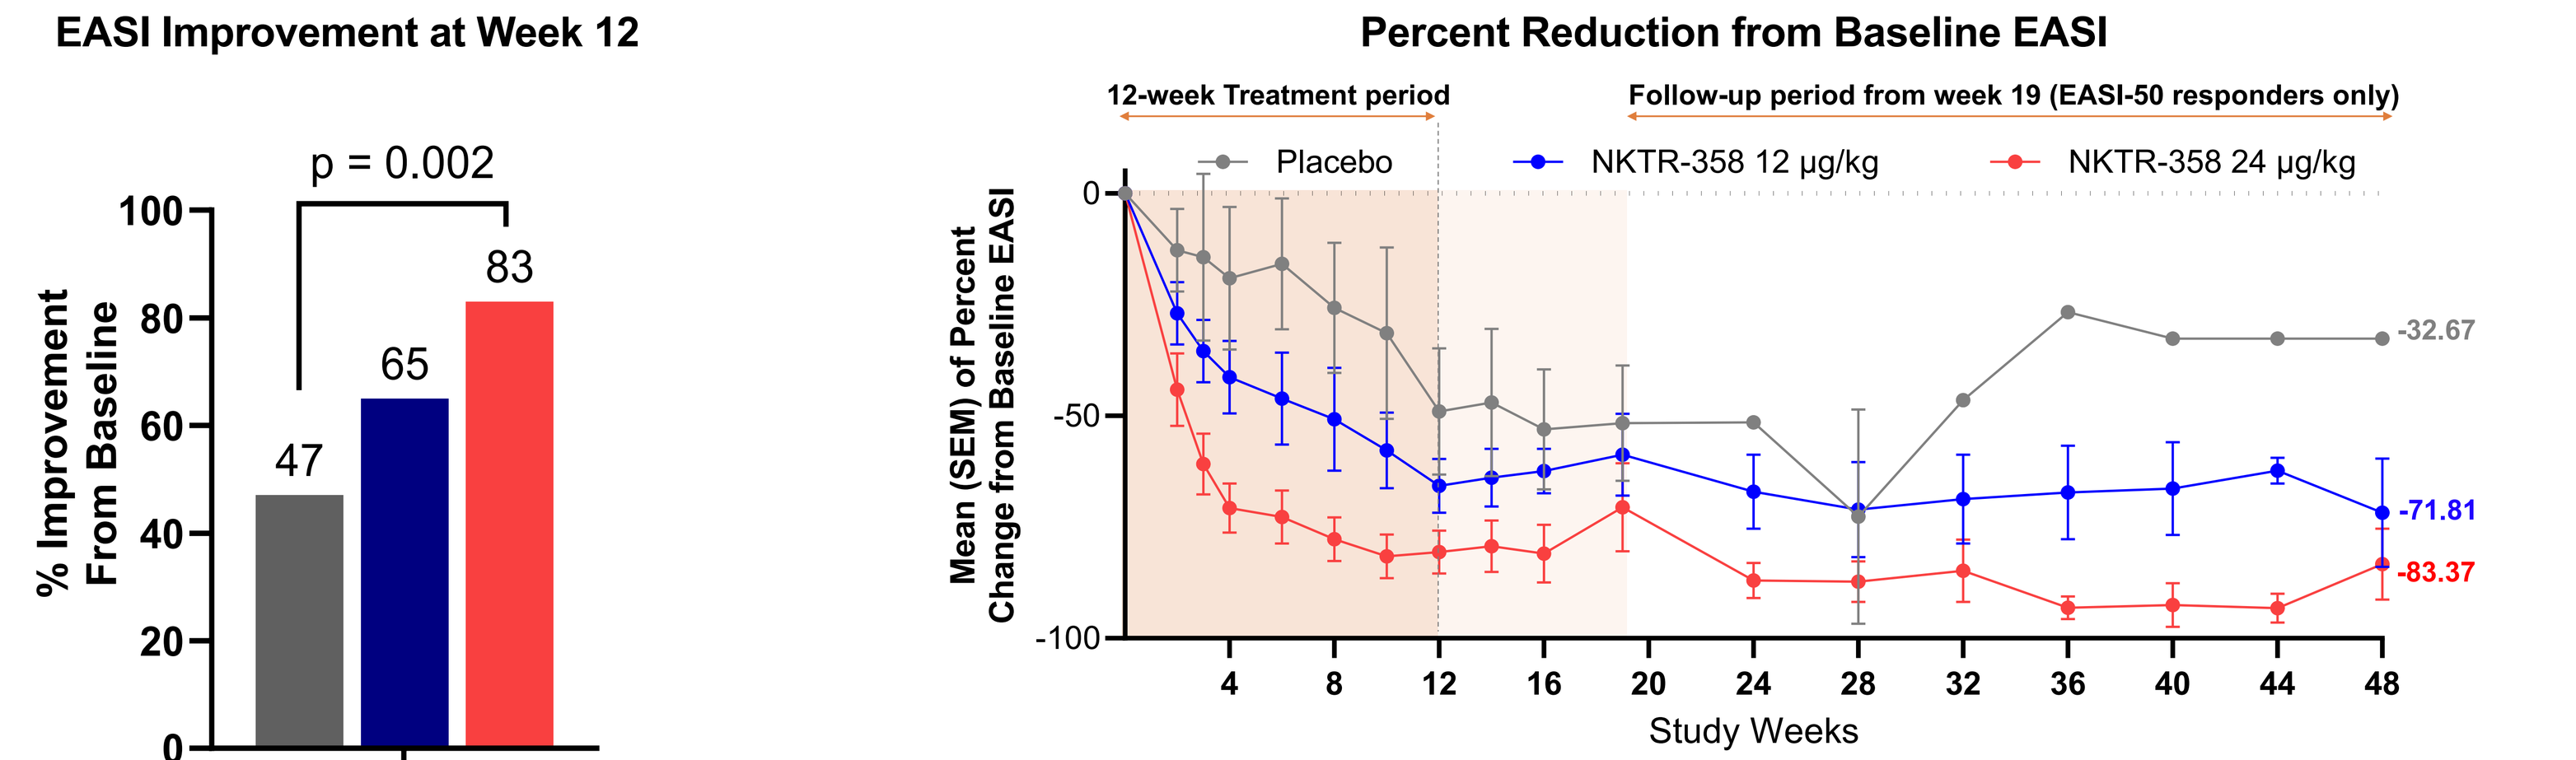
EASI Improvement at Week 12
p = 0.002
% Improvement From Baseline
100
80
60
40
20
0
47
65
83
LS Mean*
Percent Reduction from Baseline EASI
12-week Treatment period
Follow-up period from week 19 (EASI-50 responders only)
Placebo
NKTR-358 12 μg/kg
NKTR-358 24 μg/kg
Mean (SEM) of Percent Change from Baseline EASI
0
-50
-100
4
8
12
16
20
24
28
32
36
40
44
48
Study Weeks
-32.67
-71.81
-83.37

Weeks	0	2	3	4	6	8	10	12	14	16	19	24	28	32	36	40	44	48
PBO, n	10	8	6	8	6	8	5	7	5	5	6	1	2	1	1	1	1	1
REZPEG 12 μg/kg, n	16	16	15	15	14	14	12	12	12	12	12	7	5	5	5	4	3	4
REZPEG 24 μg/kg, n	17	16	16	15	15	14	13	12	13	13	13	8	9	9	7	7	7	7

n = number of participants who were evaluated at each defined timepoint

SEM: Standard error of the mean; continuous endpoint using observed data; *EASI Improvement results are least squares (LS) mean percent change from baseline obtained from Mixed Model for Repeated Measures (MMRM) as specified in the statistical analysis plan (SAP) defined in the protocol (generated by independent statistical audit firm)

Figure 3: Phase 1b Study of REZPEG in Atopic Dermatitis
Proportion of EASI-75 Responders at Week 12 and at Week 48

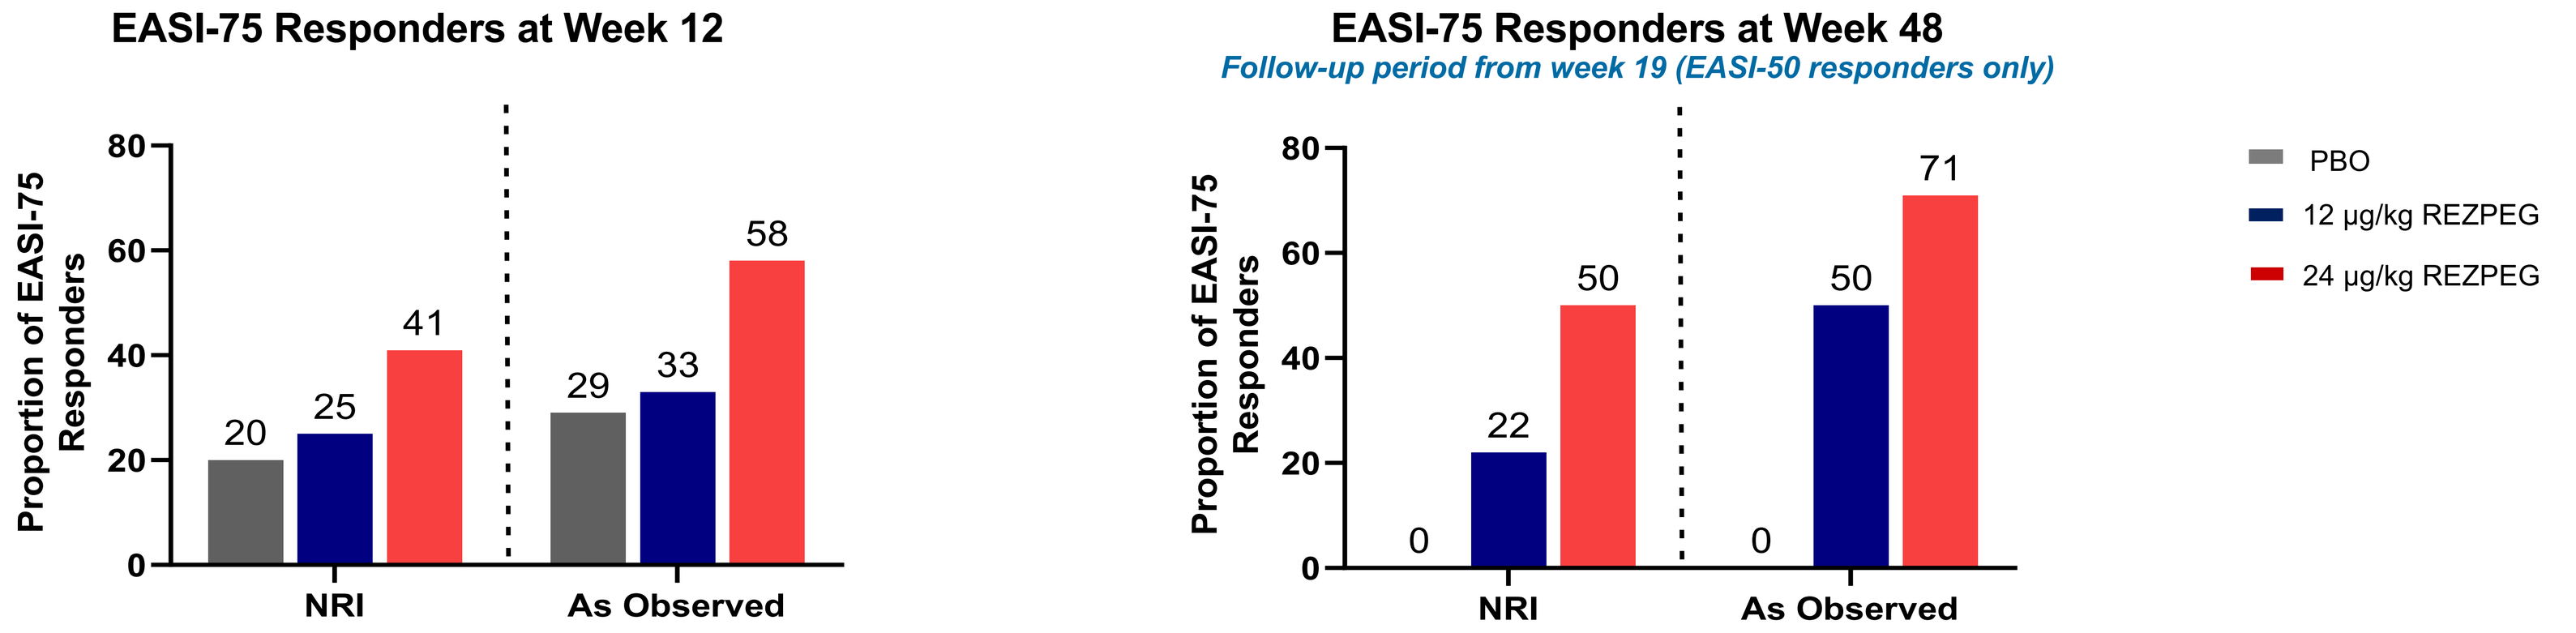
EASI-75 Responders at Week 12
Proportion of EASI-75 Responders
80
60
40
20
0
20
25
41
29
33
58
NRI
As Observed
EASI-75 Responders at Week 48
Follow-up period from week 19 (EASI-50 responders only)
0
22
50
0
50
71
NRI
As Observed
PBO
12 μg/kg REZPEG
24 μg/kg REZPEG

Patients were followed until Week 19, and those with ≥EASI-50 response at Week 19 were followed until Week 48 or until EASI-25 response criteria were no longer met. NRI: non-responder imputation

STUDY DESIGN

Figure 4: Phase 2b Study Design

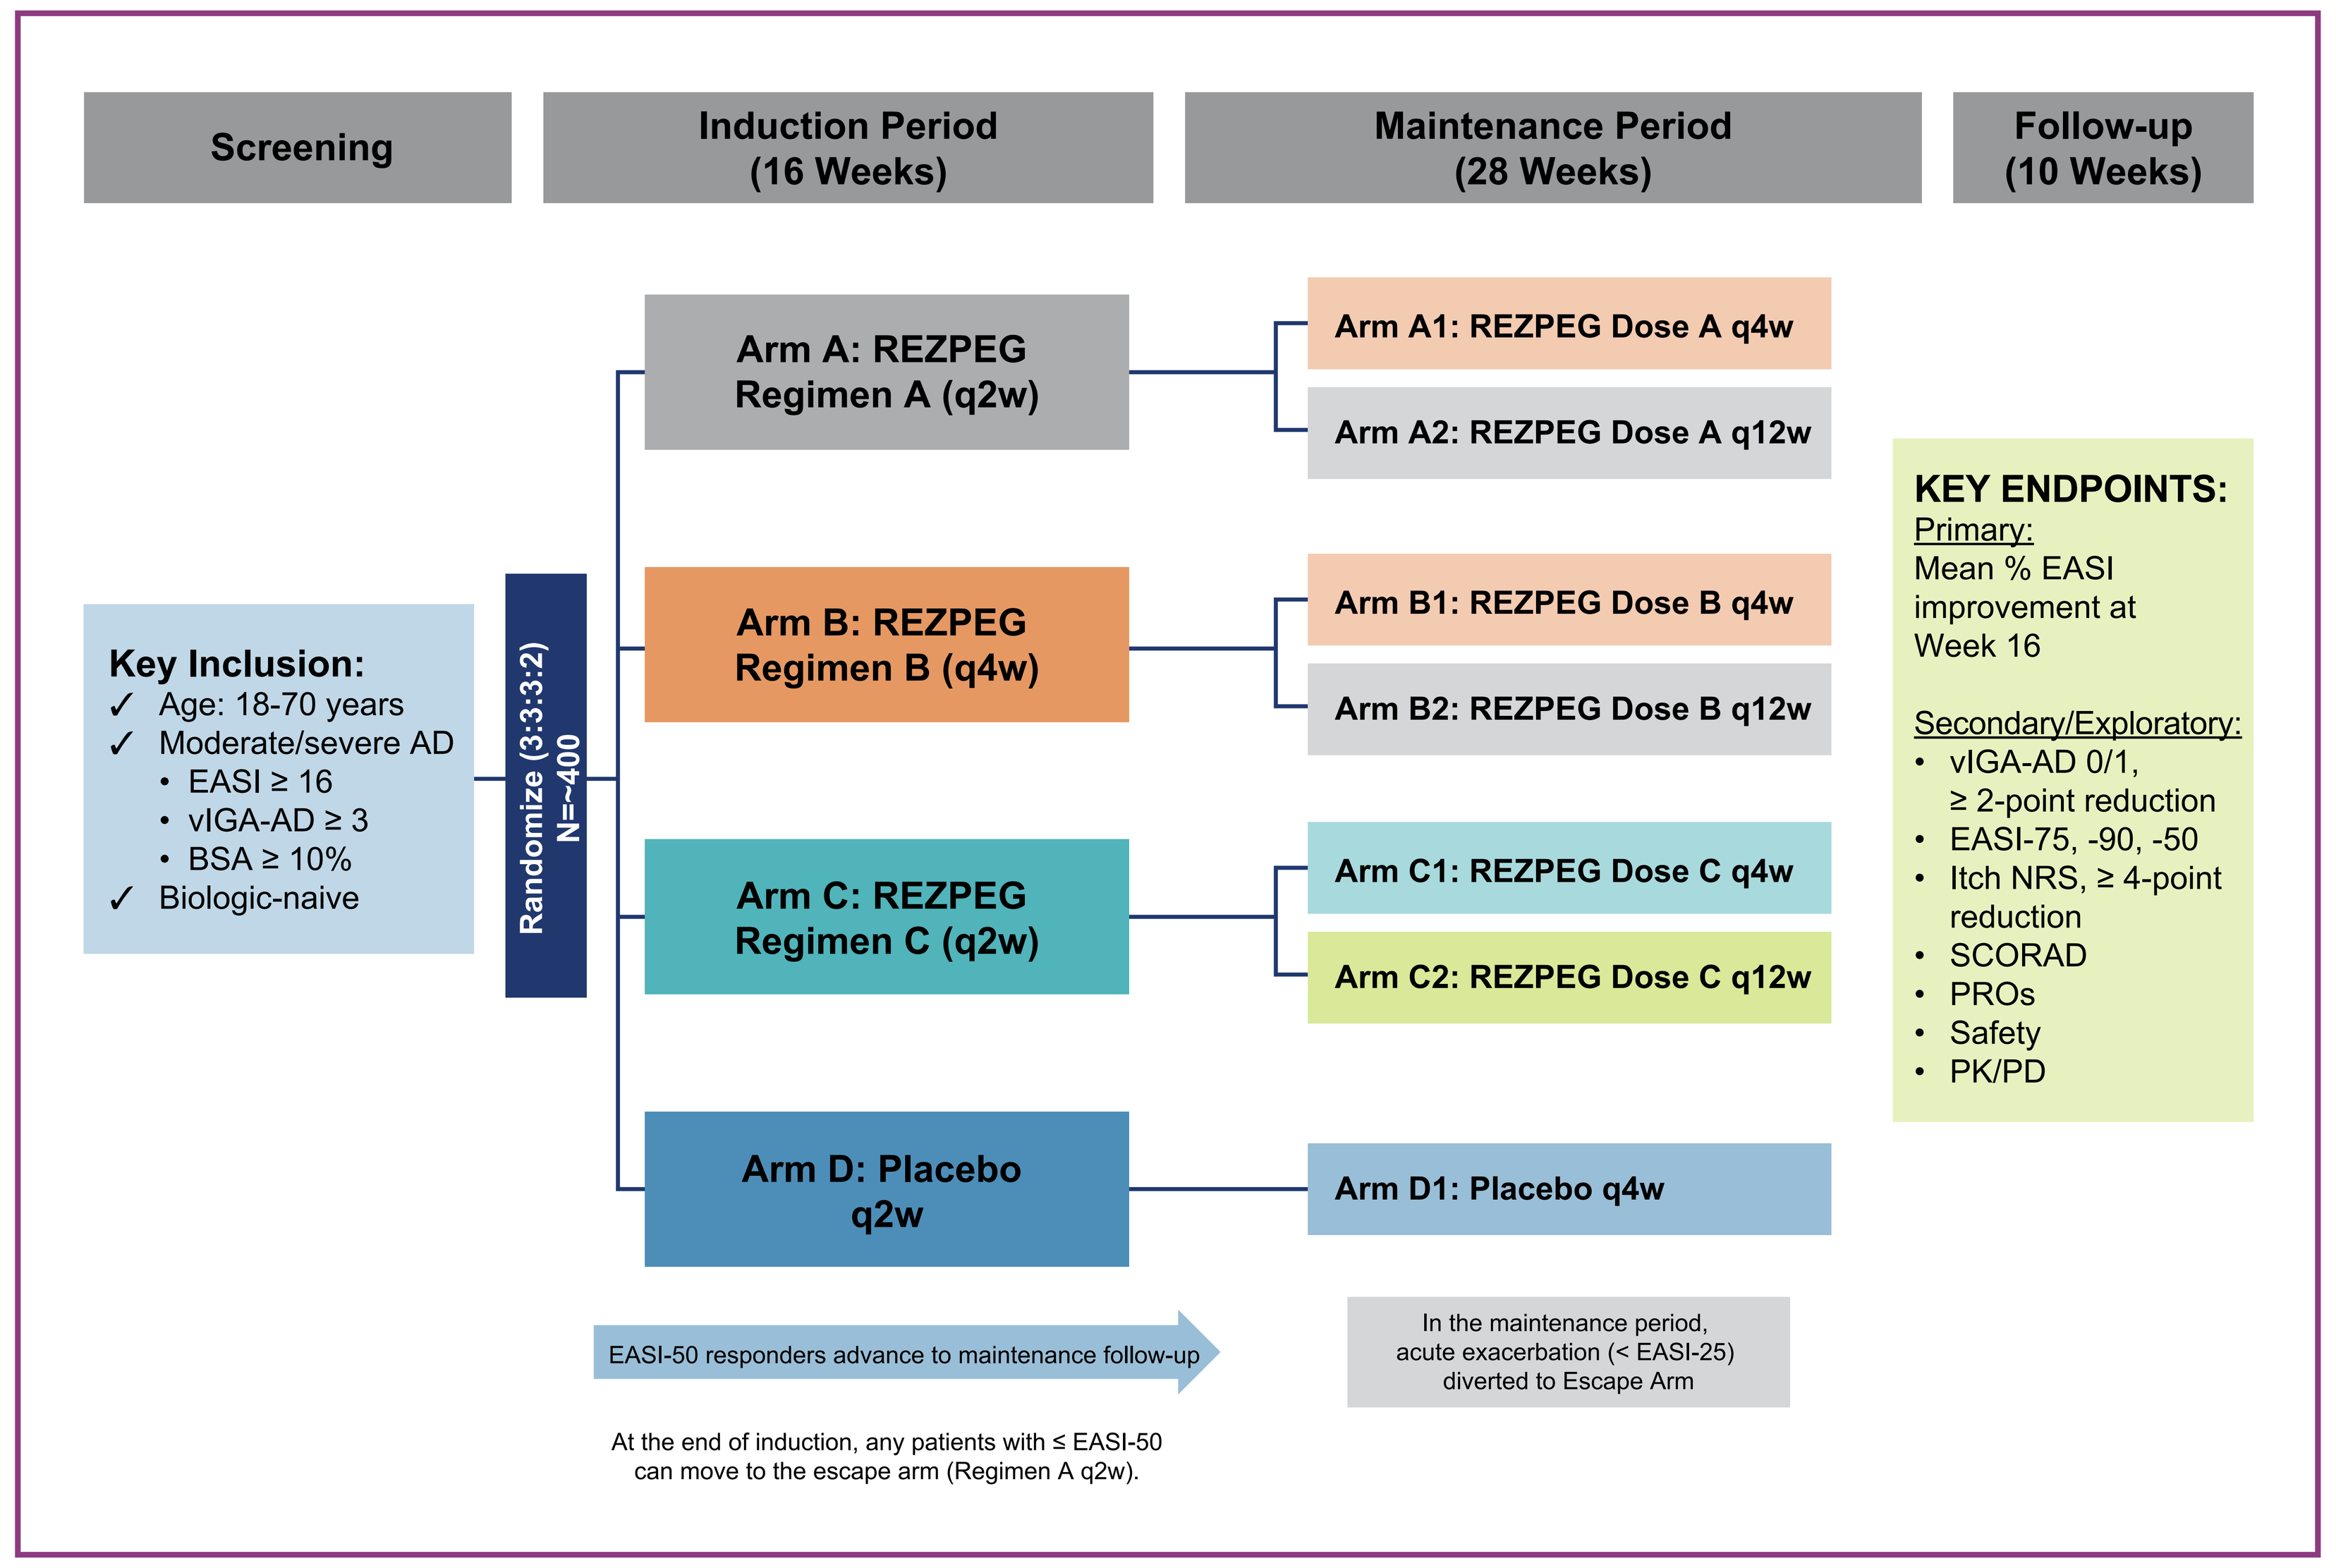
Screening
Induction Period (16 Weeks)
Maintenance Period (28 Weeks)
Follow-up (10 Weeks)
Key Inclusion:
✓ Age: 18-70 years
✓ Moderate/severe AD
• EASI ≥ 16
• vIGA-AD ≥ 3
• BSA ≥ 10%
✓ Biologic-naive
Randomize (3:3:3:2) N=~400
Arm A: REZPEG Regimen A (q2w)
Arm B: REZPEG Regimen B (q4w)
Arm C: REZPEG Regimen C (q2w)
Arm D: Placebo q2w
Arm A1: REZPEG Dose A q4w
Arm A2: REZPEG Dose A q12w
Arm B1: REZPEG Dose B q4w
Arm B2: REZPEG Dose B q12w
Arm C1: REZPEG Dose C q4w
Arm C2: REZPEG Dose C q12w
Arm D1: Placebo q4w
KEY ENDPOINTS:
Primary:
Mean % EASI improvement at Week 16
Secondary/Exploratory:
• vIGA-AD 0/1, ≥ 2-point reduction
• EASI-75, -90, -50
• Itch NRS, ≥ 4-point reduction
• SCORAD
• PROs
• Safety
• PK/PD
EASI-50 responders advance to maintenance follow-up
In the maintenance period, acute exacerbation (< EASI-25) diverted to Escape Arm
At the end of induction, any patients with ≤ EASI-50 can move to the escape arm (Regimen A q2w).

Phase 2b Study for Patients with Atopic Dermatitis

- This trial is a Phase 2b, randomized, double-blinded, placebo-controlled, international, multicenter study of REZPEG vs placebo for biologic-naïve patients with moderate-to-severe AD.
- Patients will be randomly assigned in a 3:3:3:2 ratio to 3 different REZPEG dosing regimens vs. placebo, administered subcutaneously, during the induction period.
- Patients on the REZPEG arms with an at-least EASI-50 response following the 16 week induction period will be re-randomized to a maintenance REZPEG administration every 4 or 12 weeks.
- Re-randomized patients with an acute exacerbation defined as <EASI-25 and patients that do not achieve an EASI-50 at end of induction will be placed in an open-label escape arm and administered REZPEG.

Key inclusion criteria

- ✓ Adult patients aged 18-70 years
- ✓ Chronic AD for at least 1 year and for whom topical treatment was inadequate or inadvisable
- ✓ Moderate-to-severe atopic dermatitis:
 - Eczema Area and Severity Index (EASI) score ≥ 16
 - Investigator's Global Assessment (IGA) AD score ≥ 3
 - Total Body Surface Area (BSA) affected ≥ 10%
- ✓ Systemic biologic and JAK-inhibitor naive

Primary Endpoints

- The primary endpoint for this study is the least-square mean percent reduction in EASI from baseline at end of induction.

Secondary Endpoints

- To evaluate proportion of patients at the end of induction with:
 - IGA 0/1 with at-least 2 point reduction
 - EASI-75, -90, -50
 - SCORAD-75, -50
 - Itch Numerical Rating Scale [NRS] improvement of ≥ 4 points
 - Improvement in % BSA involvement
- Additional endpoints at the end of induction:
 - Safety/tolerability
 - Various patient reported outcomes (PROs)
 - Pharmacokinetics and Pharmacodynamics
- To evaluate the assessed efficacy and safety endpoint at all other timepoints
 - During Induction
 - During Maintenance
 - During Follow-up

STUDY STATUS

Figure 5: Countries Included in the Study

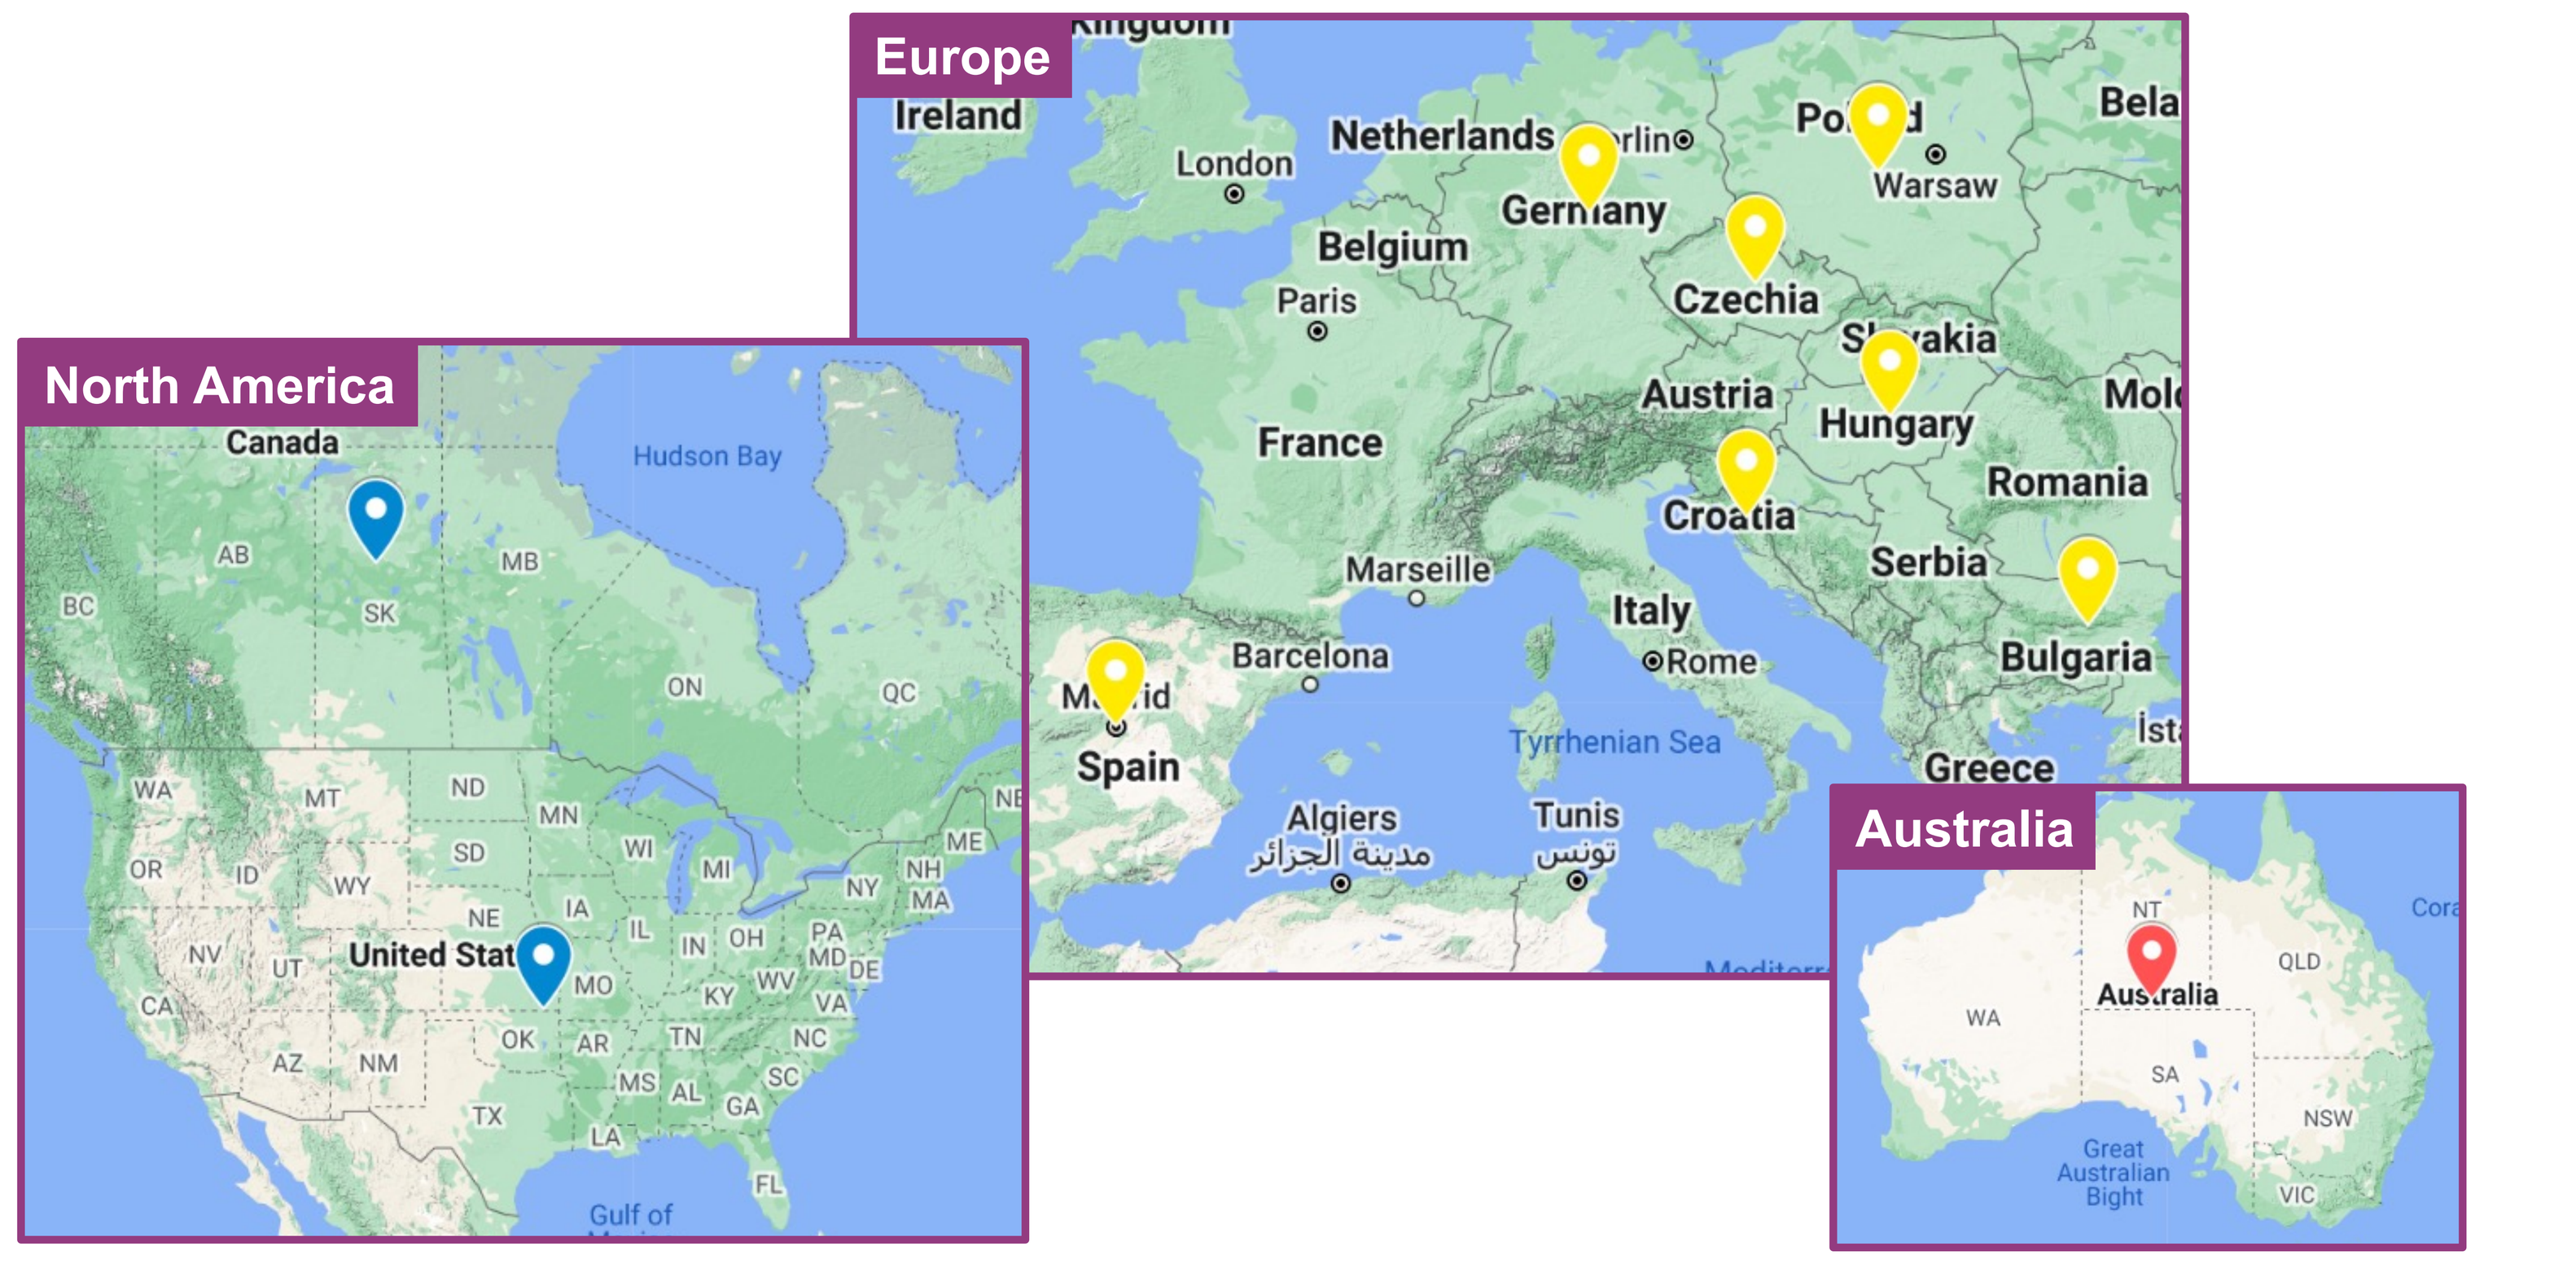
Europe
North America
Australia
Canada
Hudson Bay
United Stat
Ireland
London
Netherlands
Belgium
Paris
Germany
Warsaw
Czechia
Austria
Hungary
Romania
France
Croatia
Serbia
Marseille
Italy
Rome
Barcelona
Spain
Bulgaria
Tyrrhenian Sea
Algiers
Tunis
Greece
WA
NT
QLD
SA
NSW
VIC
Great Australian Bight

- This study is initiating in North America and other parts of the world (**Figure 5**):
 - North America (Canada, United States)
 - Europe (Bulgaria, Croatia, Czech Republic, Germany, Hungary, Poland, Spain)
 - APAC (Australia)
- Please contact the Sponsor (Nektar) with any questions

ACKNOWLEDGMENTS

This study is funded by Nektar Therapeutics, San Francisco, CA.
The study will be approved by the institutional review board of each participating site.

ABBREVIATIONS

AD, Atopic dermatitis; Treg, regulatory T cells; PEG, polyethylene glycol; rhIL-2, recombinant human interleukin 2; Tcons, conventional T cells; Th1, Type 1 T helper cells; Th2, Type 2 T helper cells; Th17, Type 17 T helper cells; RA, rheumatoid arthritis; MS, multiple sclerosis; APC, antigen-presenting cells; EASI, Eczema Area and Severity Index; vIGA-AD, Validated Investigator Global Assessment scale for Atopic Dermatitis; BSA, Body Surface Area; q2w, once every 2 weeks; q4w, once every 4 weeks; q12w, once every 12 weeks; Itch NRS, Itch Numerical Rating Scale; SCORAD, Scoring of Atopic Dermatitis Index; PROs, patient reported outcomes; PK, Pharmacokinetics; PD, Pharmacodynamics; APAC, Asia-Pacific

REFERENCES

1. Silverberg, J. I. & Hanifin, J. M. Adult eczema prevalence and associations with asthma and other health and demographic factors: A US population–based study. *Journal of Allergy and Clinical Immunology* vol. 132 1132–1138 (2013).
2. Abuabara, K. et al. Prevalence of Atopic Eczema Among Patients Seen in Primary Care: Data From The Health Improvement Network. *Ann. Intern. Med*. 170, 354–356 (2019).
3. Silverberg, J. I. Public Health Burden and Epidemiology of Atopic Dermatitis. *Dermatologic Clinics* vol. 35 283–289 (2017).
4. Shaw, T. E., Currie, G. P., Koudelka, C. W. & Simpson, E. L. Eczema Prevalence in the United States: Data from the 2003 National Survey of Children's Health. *Journal of Investigative Dermatology* vol. 131 67–73 (2011).
5. Chiesa Fuxench, Z. C. et al. Atopic Dermatitis in America Study: A Cross-Sectional Study Examining the Prevalence and Disease Burden of Atopic Dermatitis in the US Adult Population. *J. Invest. Dermatol*. 139, 583–590 (2019).
6. Agrawal R, et al. The role of regulatory T cells in atopic dermatitis. *Curr Probl Dermatol*. (2011) 41:112-124.
7. Fanton C, et al. Selective expansion of regulatory T cells by NKTR-358 in healthy volunteers and patients with systemic lupus erythematosus. *J Transl Autoimmun*. (2022) 5:100152.
8. Nektar Therapeutics. (2023, August 7). Nektar Announces Promising New and Corrected Rezpegaldesleukin Efficacy Data Which Were Previously Reported in 2022 and Incorrectly Calculated by Former Collaborator Eli Lilly & Company [Press Release].

Poster presented at the EADV Congress 2023 | Oct 11-14, 2023.
Corresponding author Jonathan Silverberg: jonathanisilverberg@gmail.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without written permission from the authors.