Combining Bempegaldesleukin (CD122-preferential IL-2 pathway agonist) and NKTR-262 (TLR7/8 agonist) pairs local innate activation with systemic CD8+ T cell expansion to enhance anti-tumor immunity

EARLE A. CHILES RESEARCH INSTITUTE

PROVIDENCE Cancer Institute

Annah S. Rolig[1], Daniel Rose[1], Saul Kivimäe[2], Werner Rubas[2], William L. Redmond[1]

[1] Earle A. Chiles Research Institute, Providence Cancer Institute, Portland, OR [2] Nektar Therapeutics, San Francisco, CA

Introduction

- Previously, we demonstrated that radiation therapy (RT) combined with Bempegaldesleukin (BEMPEG;NKTR-214), a first-in-class CD122-preferential IL-2 pathway agonist, enhanced the anti-tumor efficacy of radiation therapy (RT) through a T cell-dependent mechanism, although systemic responses were modest.
- Therefore, we explored alternative approaches to improve systemic tumor-specific immunity.
- Because toll-like receptor (TLR) signaling can induce antigen-presenting cell differentiation and reduce intratumoral immune suppression, we evaluated whether intratumoral NKTR-262, a polymer-modified TLR 7/8 agonist, combined with systemic BEMPEG treatment resulted in improved tumor-specific immunity and survival compared to BEMPEG combined with RT.
- Finally, comparison of BEMPEG combined with either RT or NKTR-262 will reveal mechanisms driving systemic anti-tumor responses.

Hypothesis

We hypothesized that BEMPEG/NKTR-262 therapy would be superior to BEMPEG/RT because TLR7/8 agonists enhance priming and antigen presentation, resulting in potent proinflammatory, anti-tumor immunity.

Treatment groups

- Vehicle control
- RT
- BEMPEG
- NKTR-262
- BEMPEG/RT
- BEMPEG/NKTR-262

Methods

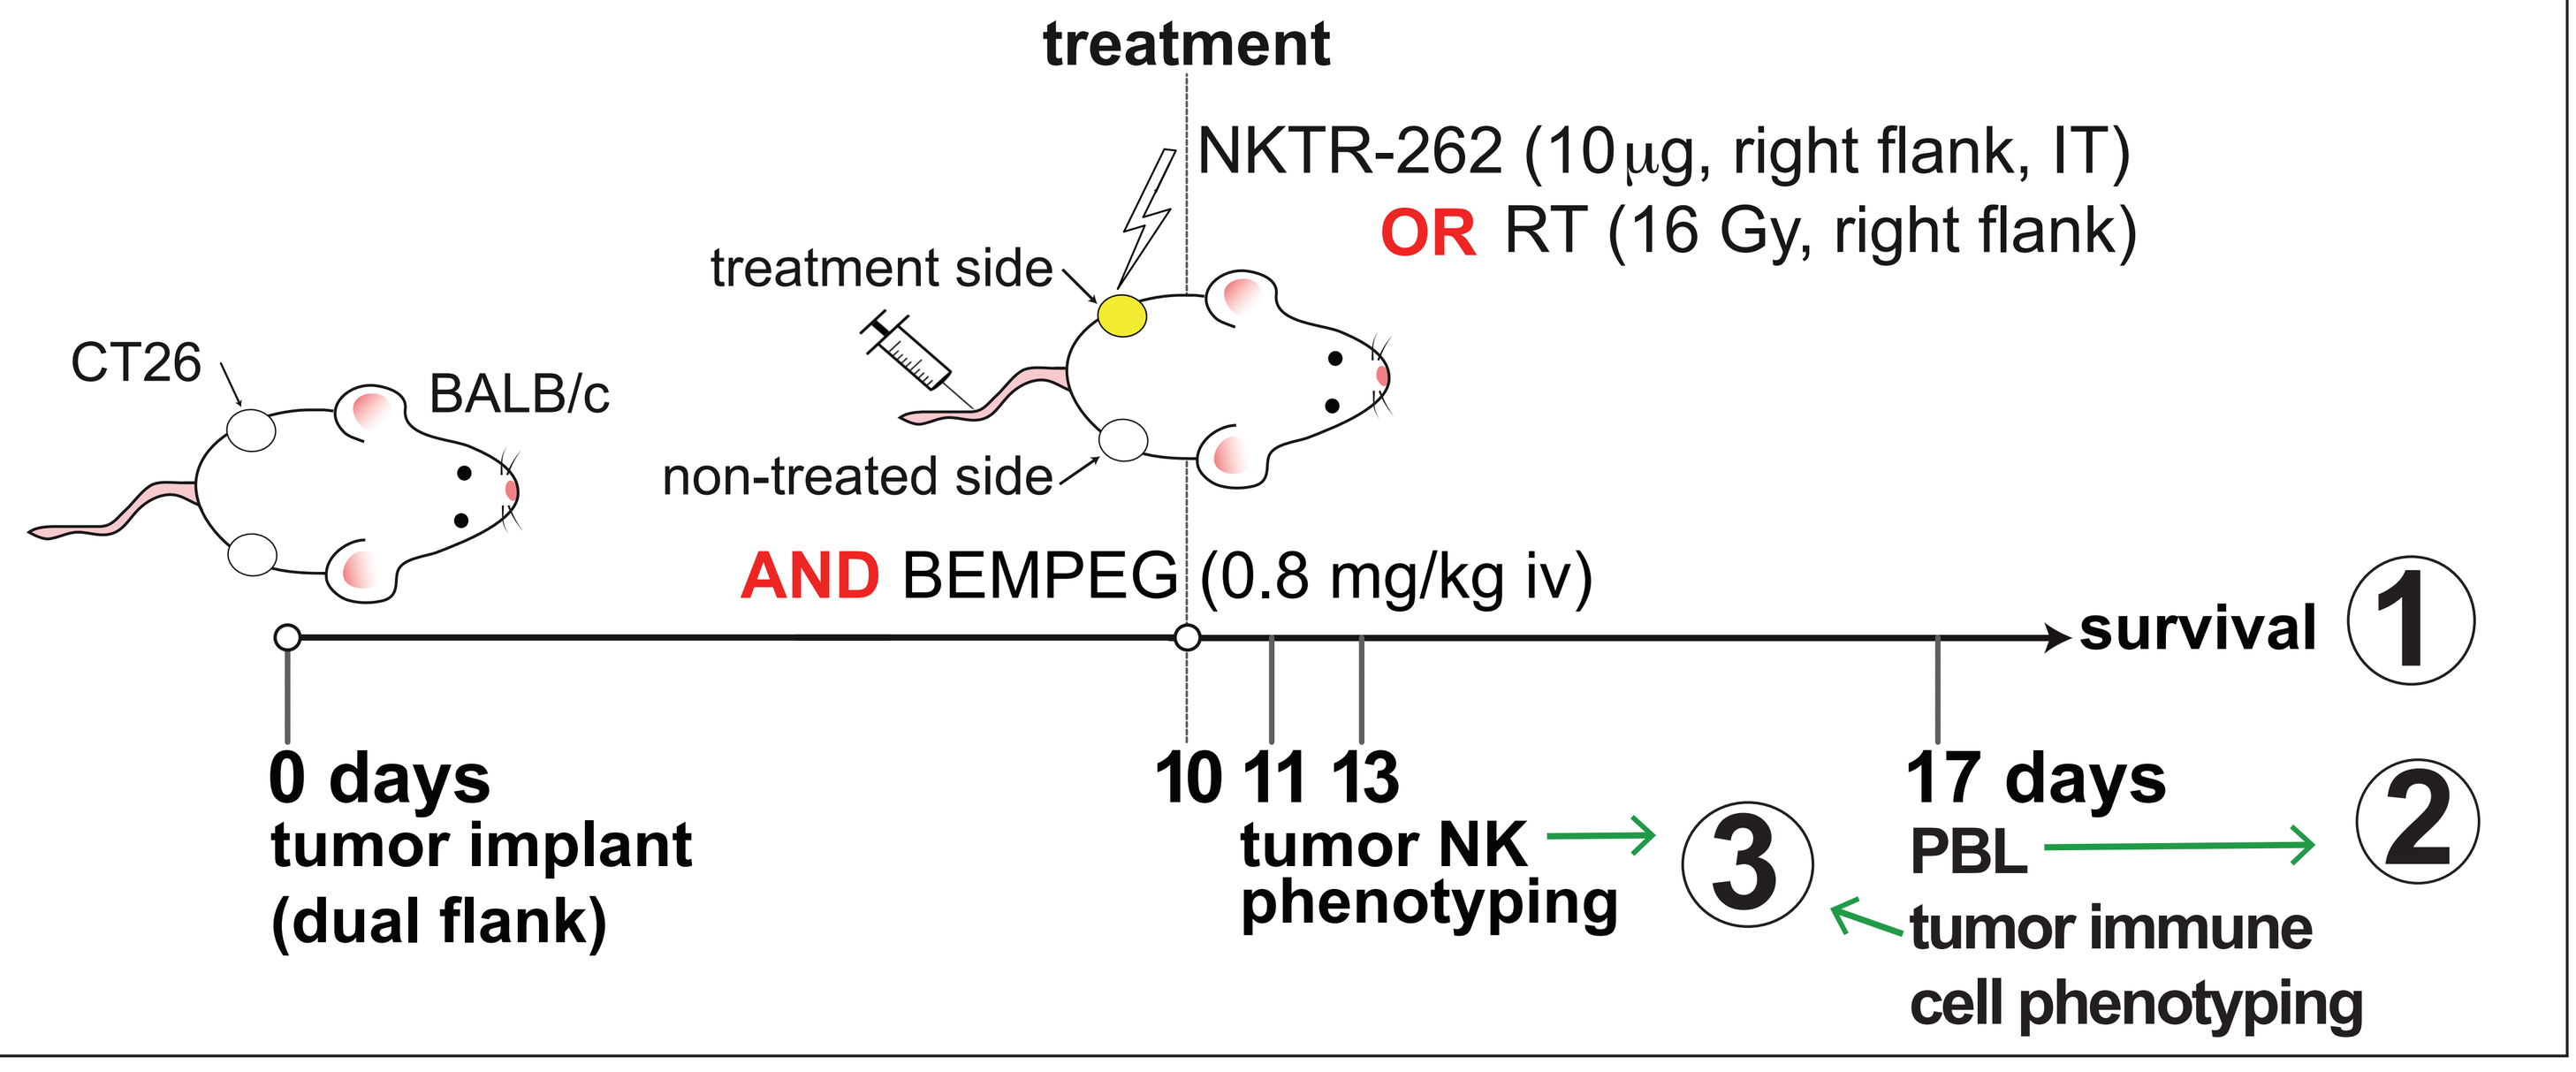
treatment
NKTR-262 (10μg, right flank, IT)
OR RT (16 Gy, right flank)
treatment side
CT26
BALB/c
non-treated side
AND BEMPEG (0.8 mg/kg iv)
survival 1
0 days
tumor implant
(dual flank)
10 11 13
tumor NK phenotyping
3
17 days
PBL
2
tumor immune cell phenotyping

1 BEMPEG/NKTR-262 treatment improves survival over BEMPEG/RT and depends on CD8 T and NK cells

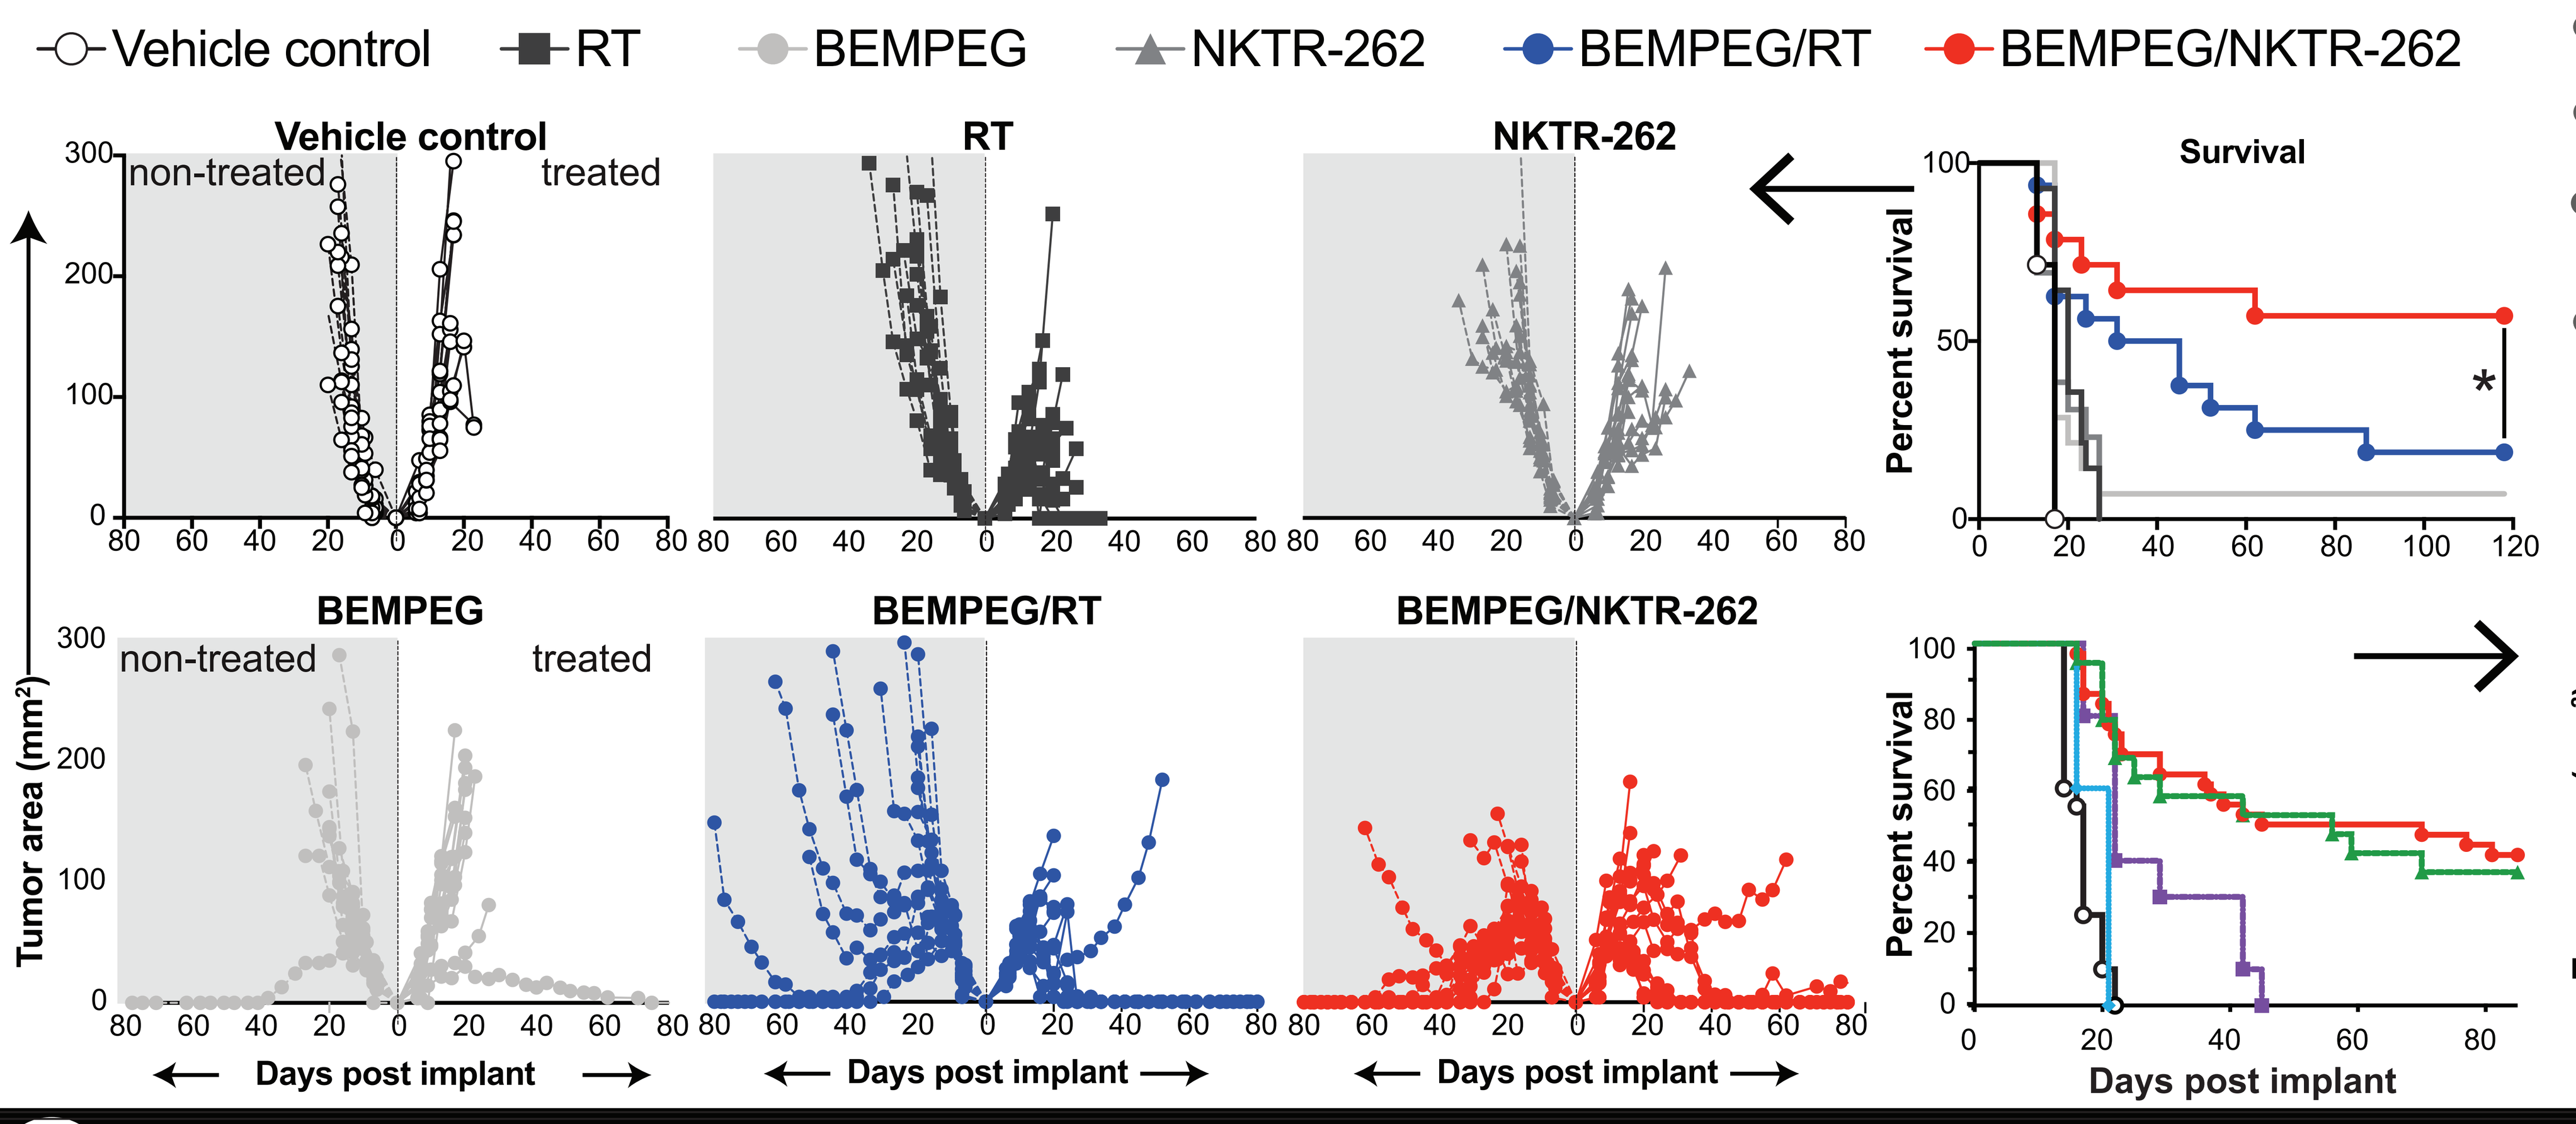
Vehicle control
RT
BEMPEG
NKTR-262
BEMPEG/RT
BEMPEG/NKTR-262
Vehicle control
RT
NKTR-262
BEMPEG
BEMPEG/RT
BEMPEG/NKTR-262
non-treated
treated
Tumor area (mm²)
Days post implant
Survival
Percent survival

- Mice bearing dual flank CT26 tumors were treated as described, and tumor area (mm2) was determined every 2-3 days.
- BEMPEG/NKTR-262 combination treatment significantly reduced tumor growth and increased survival (top center).
- This response depends primarily on CD8 T and NK cells as depletion of CD8 (purple) or NK (light blue), but not CD4 (green) cells, abrogated the effects in comparison to BEMPEG/RT.
- For depletion, animals were treated using either 20 μl anti-asialo-GM1 or 200 μg anti-CD8 (clone 53-6.7) delivered once intraperitoneally (ip) 1 day before treatment (D9), or 200 μg anti-CD4 (clone GK1.5) given 1x/week (ip) for 6 weeks. N>10 from two independent experiments. * p<0.05, log-rank test.

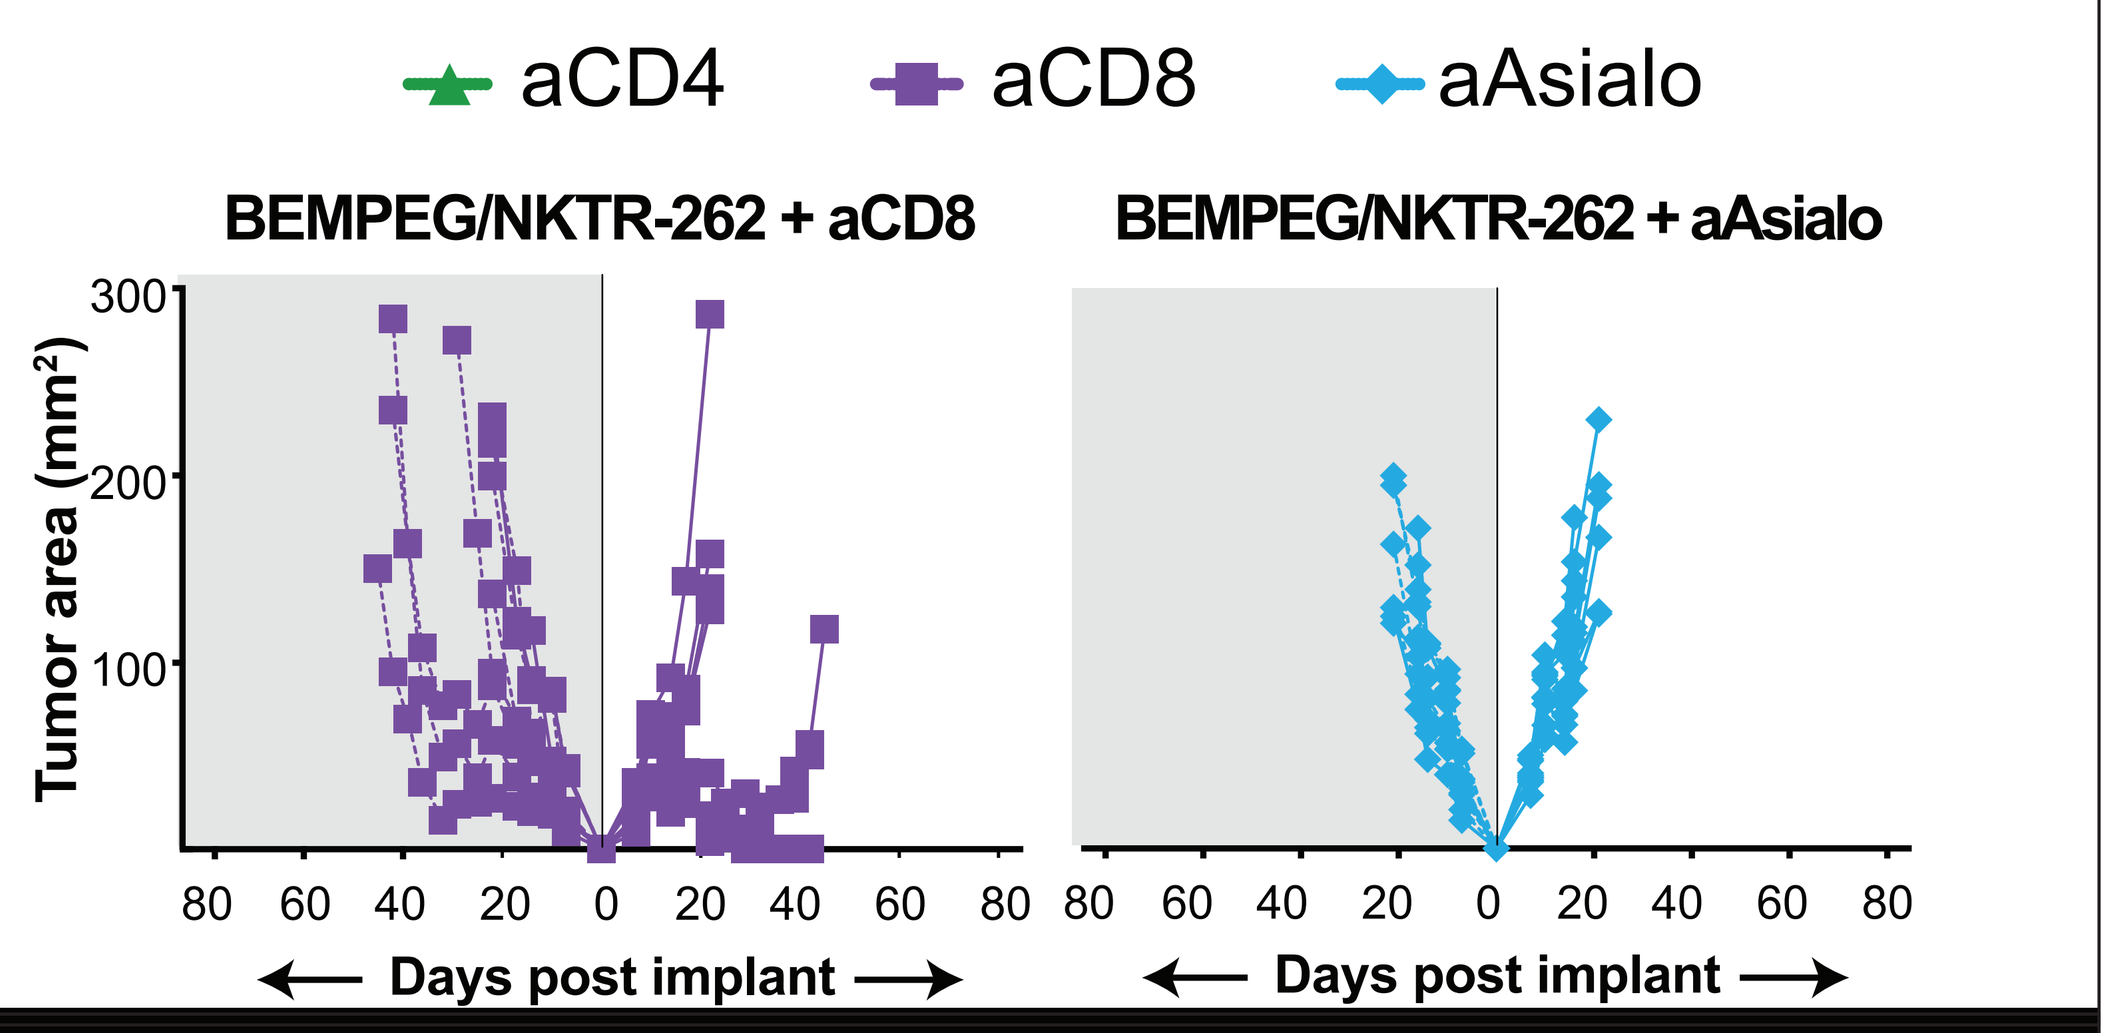
aCD4
aCD8
aAsialo
BEMPEG/NKTR-262 + aCD8
BEMPEG/NKTR-262 + aAsialo
Tumor area (mm²)
Days post implant

2 Peripheral blood immune cell phenotyping

BEMPEG/NKTR-262 induces significantly greater expansion of CD8 T cells in the blood than BEMPEG/RT

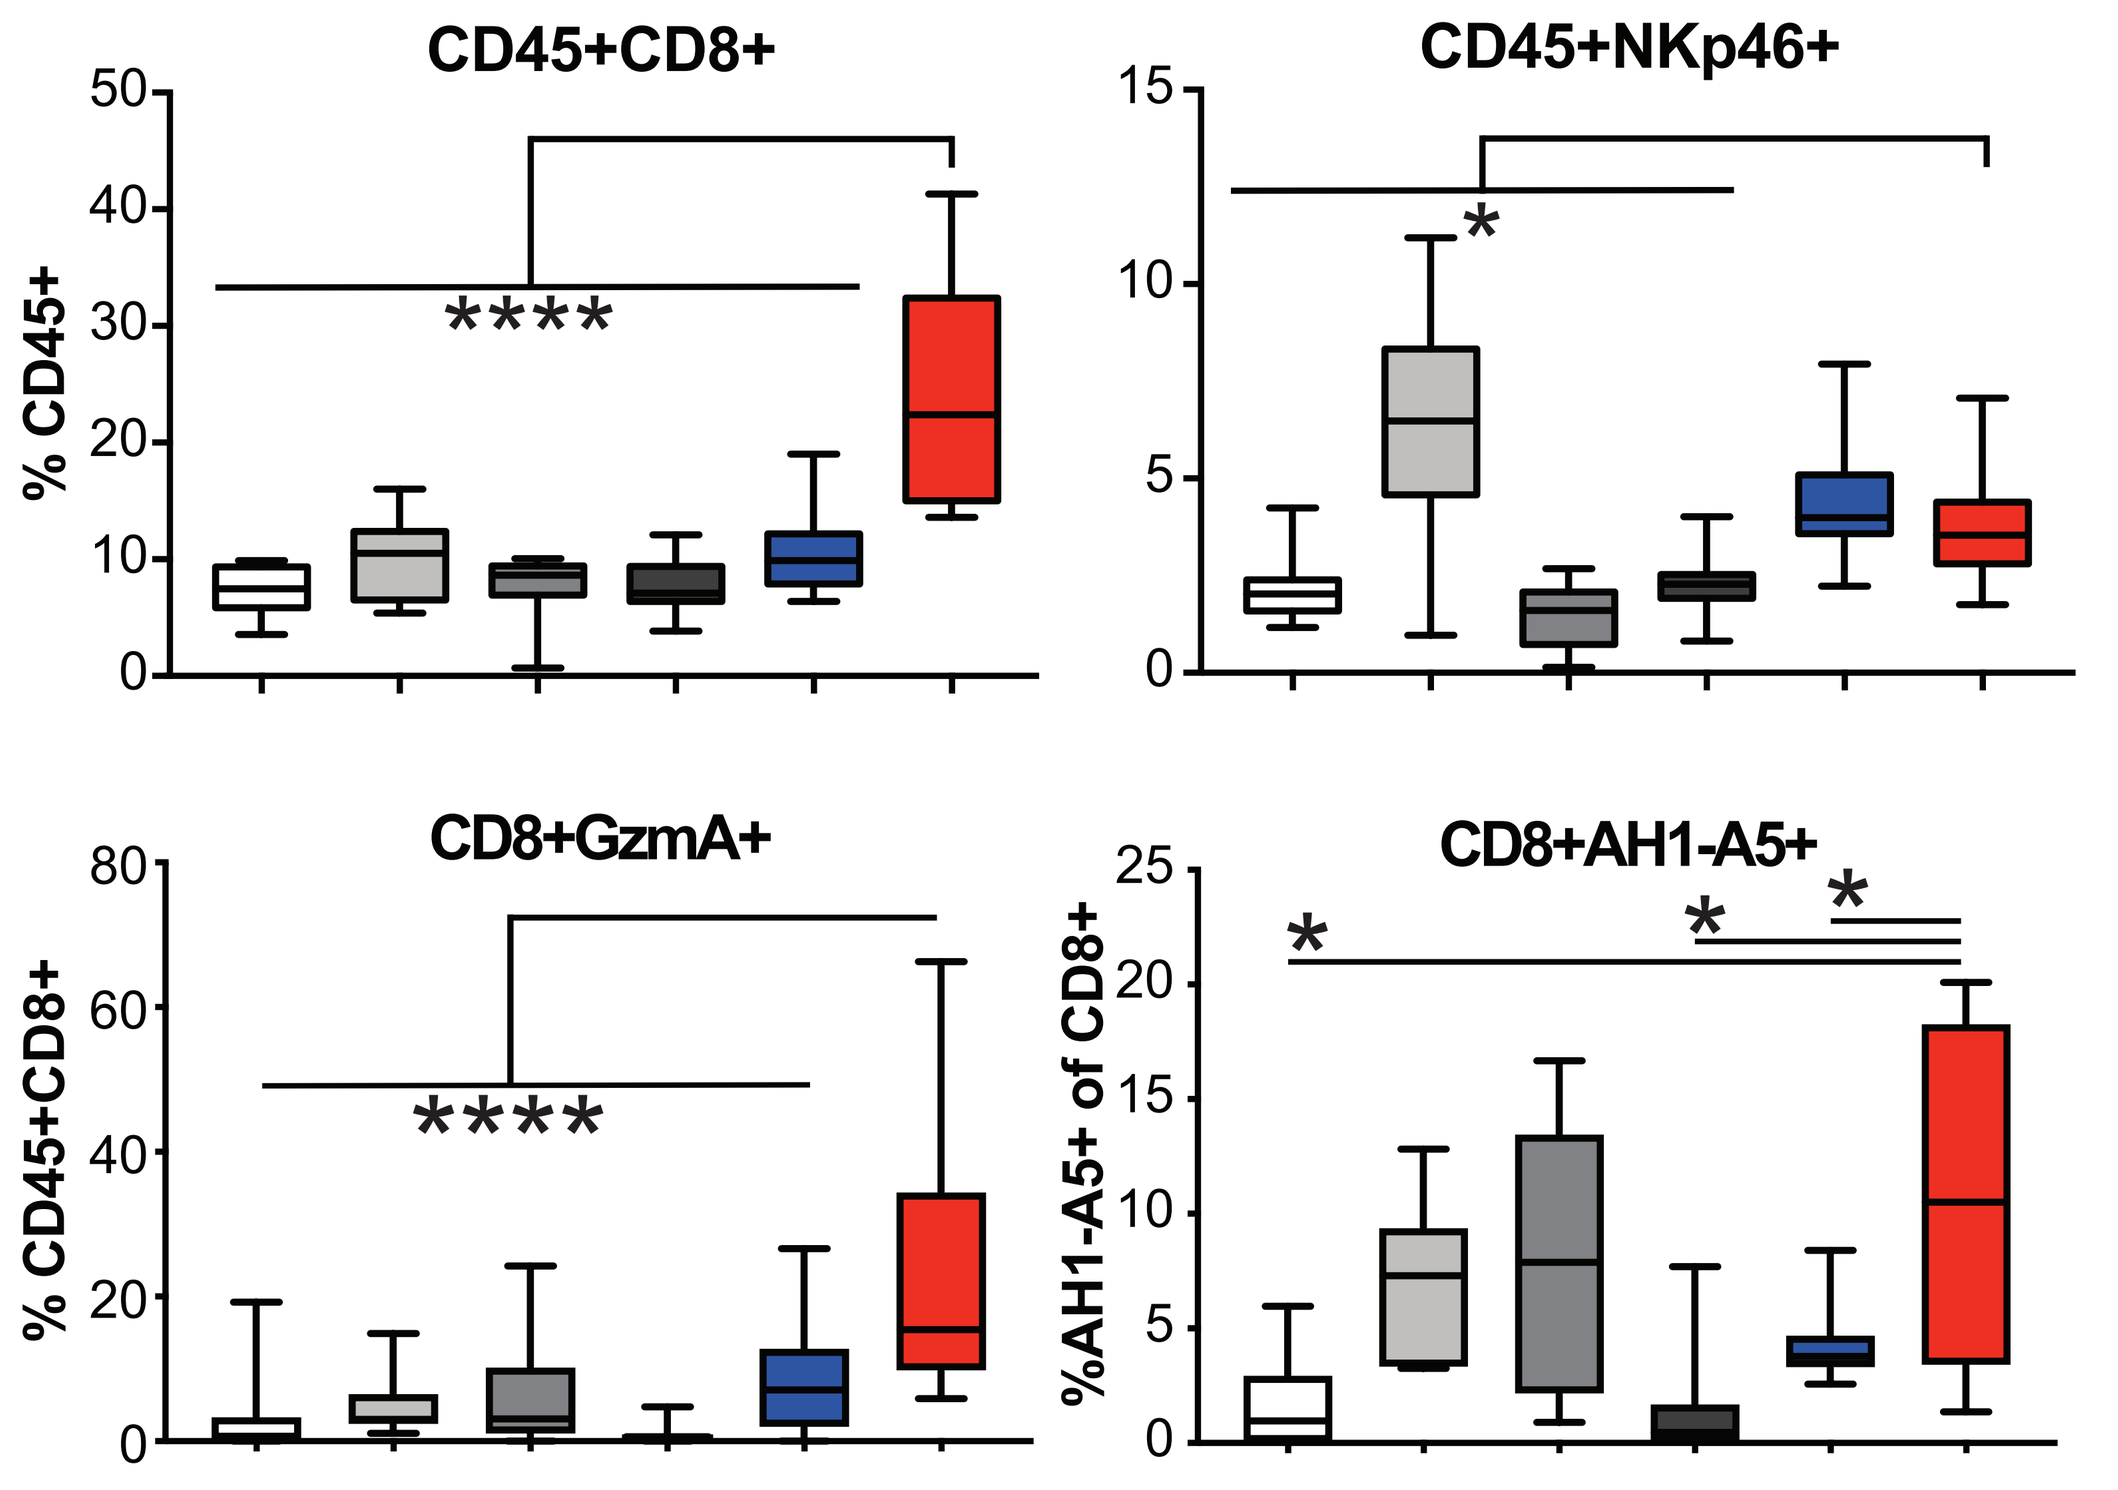
Vehicle
BEMPEG
NKTR-262
RT
BEMPEG/RT
BEMPEG/NKTR-262
CD45+CD8+
CD45+NKp46+
CD8+GzmA+
CD8+AH1-A5+
% CD45+
% CD45+CD8+
%AH1-A5+ of CD8+

- Blood drawn 7 days post- treatment; flow cytometry used to montior changes in immune cell phenotypes.
- BEMPEG/NKTR-262 in combination significantly enhanced CD8 T cell responses over BEMPEG/RT: increased % of total CD8 T cells, % of granzyme+ , and % of tumor specific (AH1-A5+) CD8 T cells.
- BEMPEG/NKTR-262 treatment induced AH1-A5+ and AH1-A5- CD8 T cells with increased granzyme A levels over BEMPEG/RT, suggesting increased priming and epitope spreading. N=7-14 from 2 experiments. One-way ANOVA; *p<0.05, **p<0.01, ****p<0.0001.

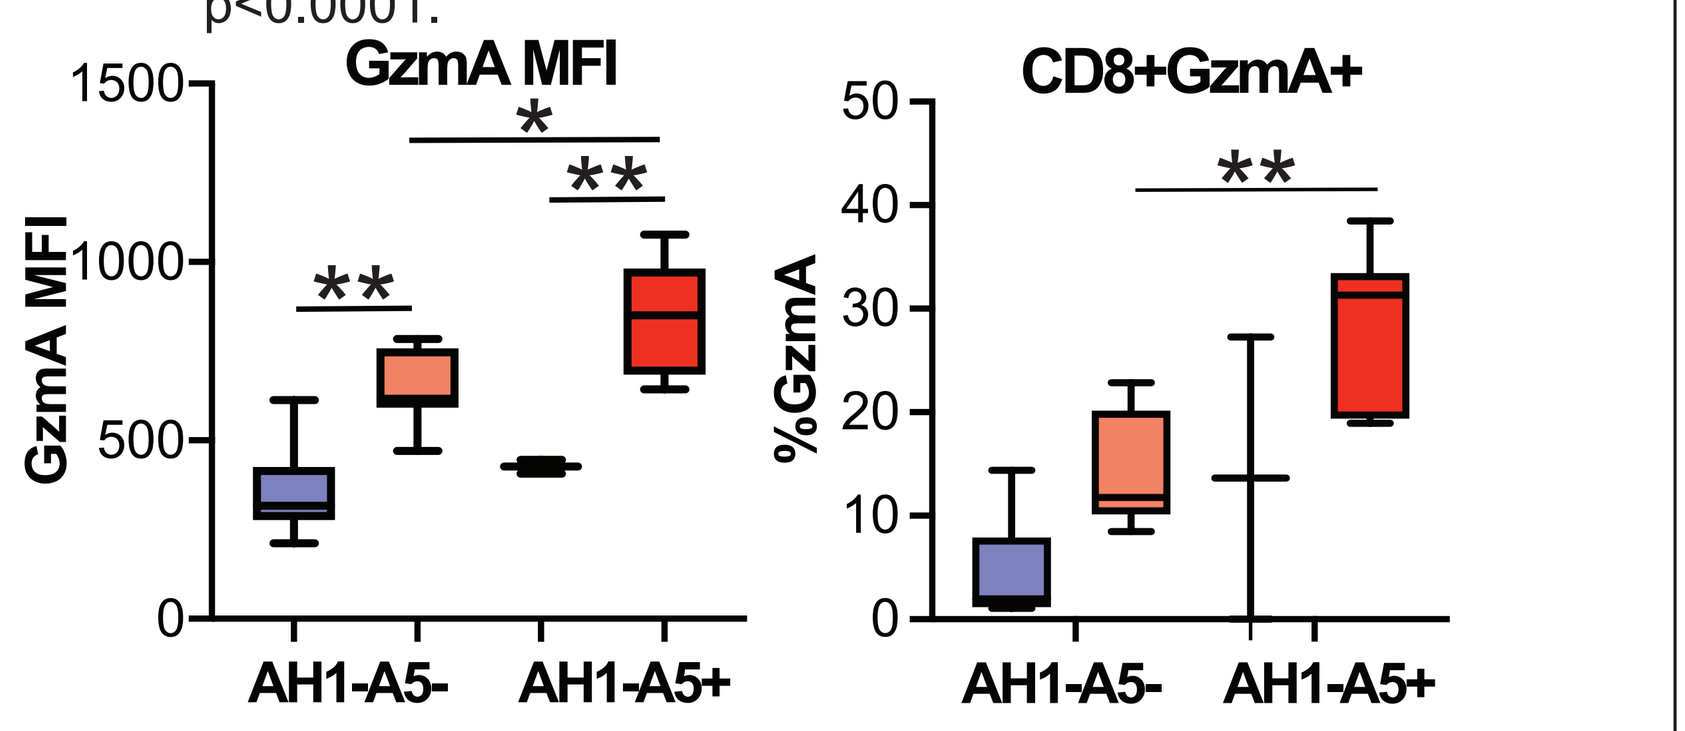
GzmA MFI
CD8+GzmA+
%GzmA
AH1-A5-
AH1-A5+

NK cells are required for CD8 T cell expansion after BEMPEG/NKTR-262 combination therapy

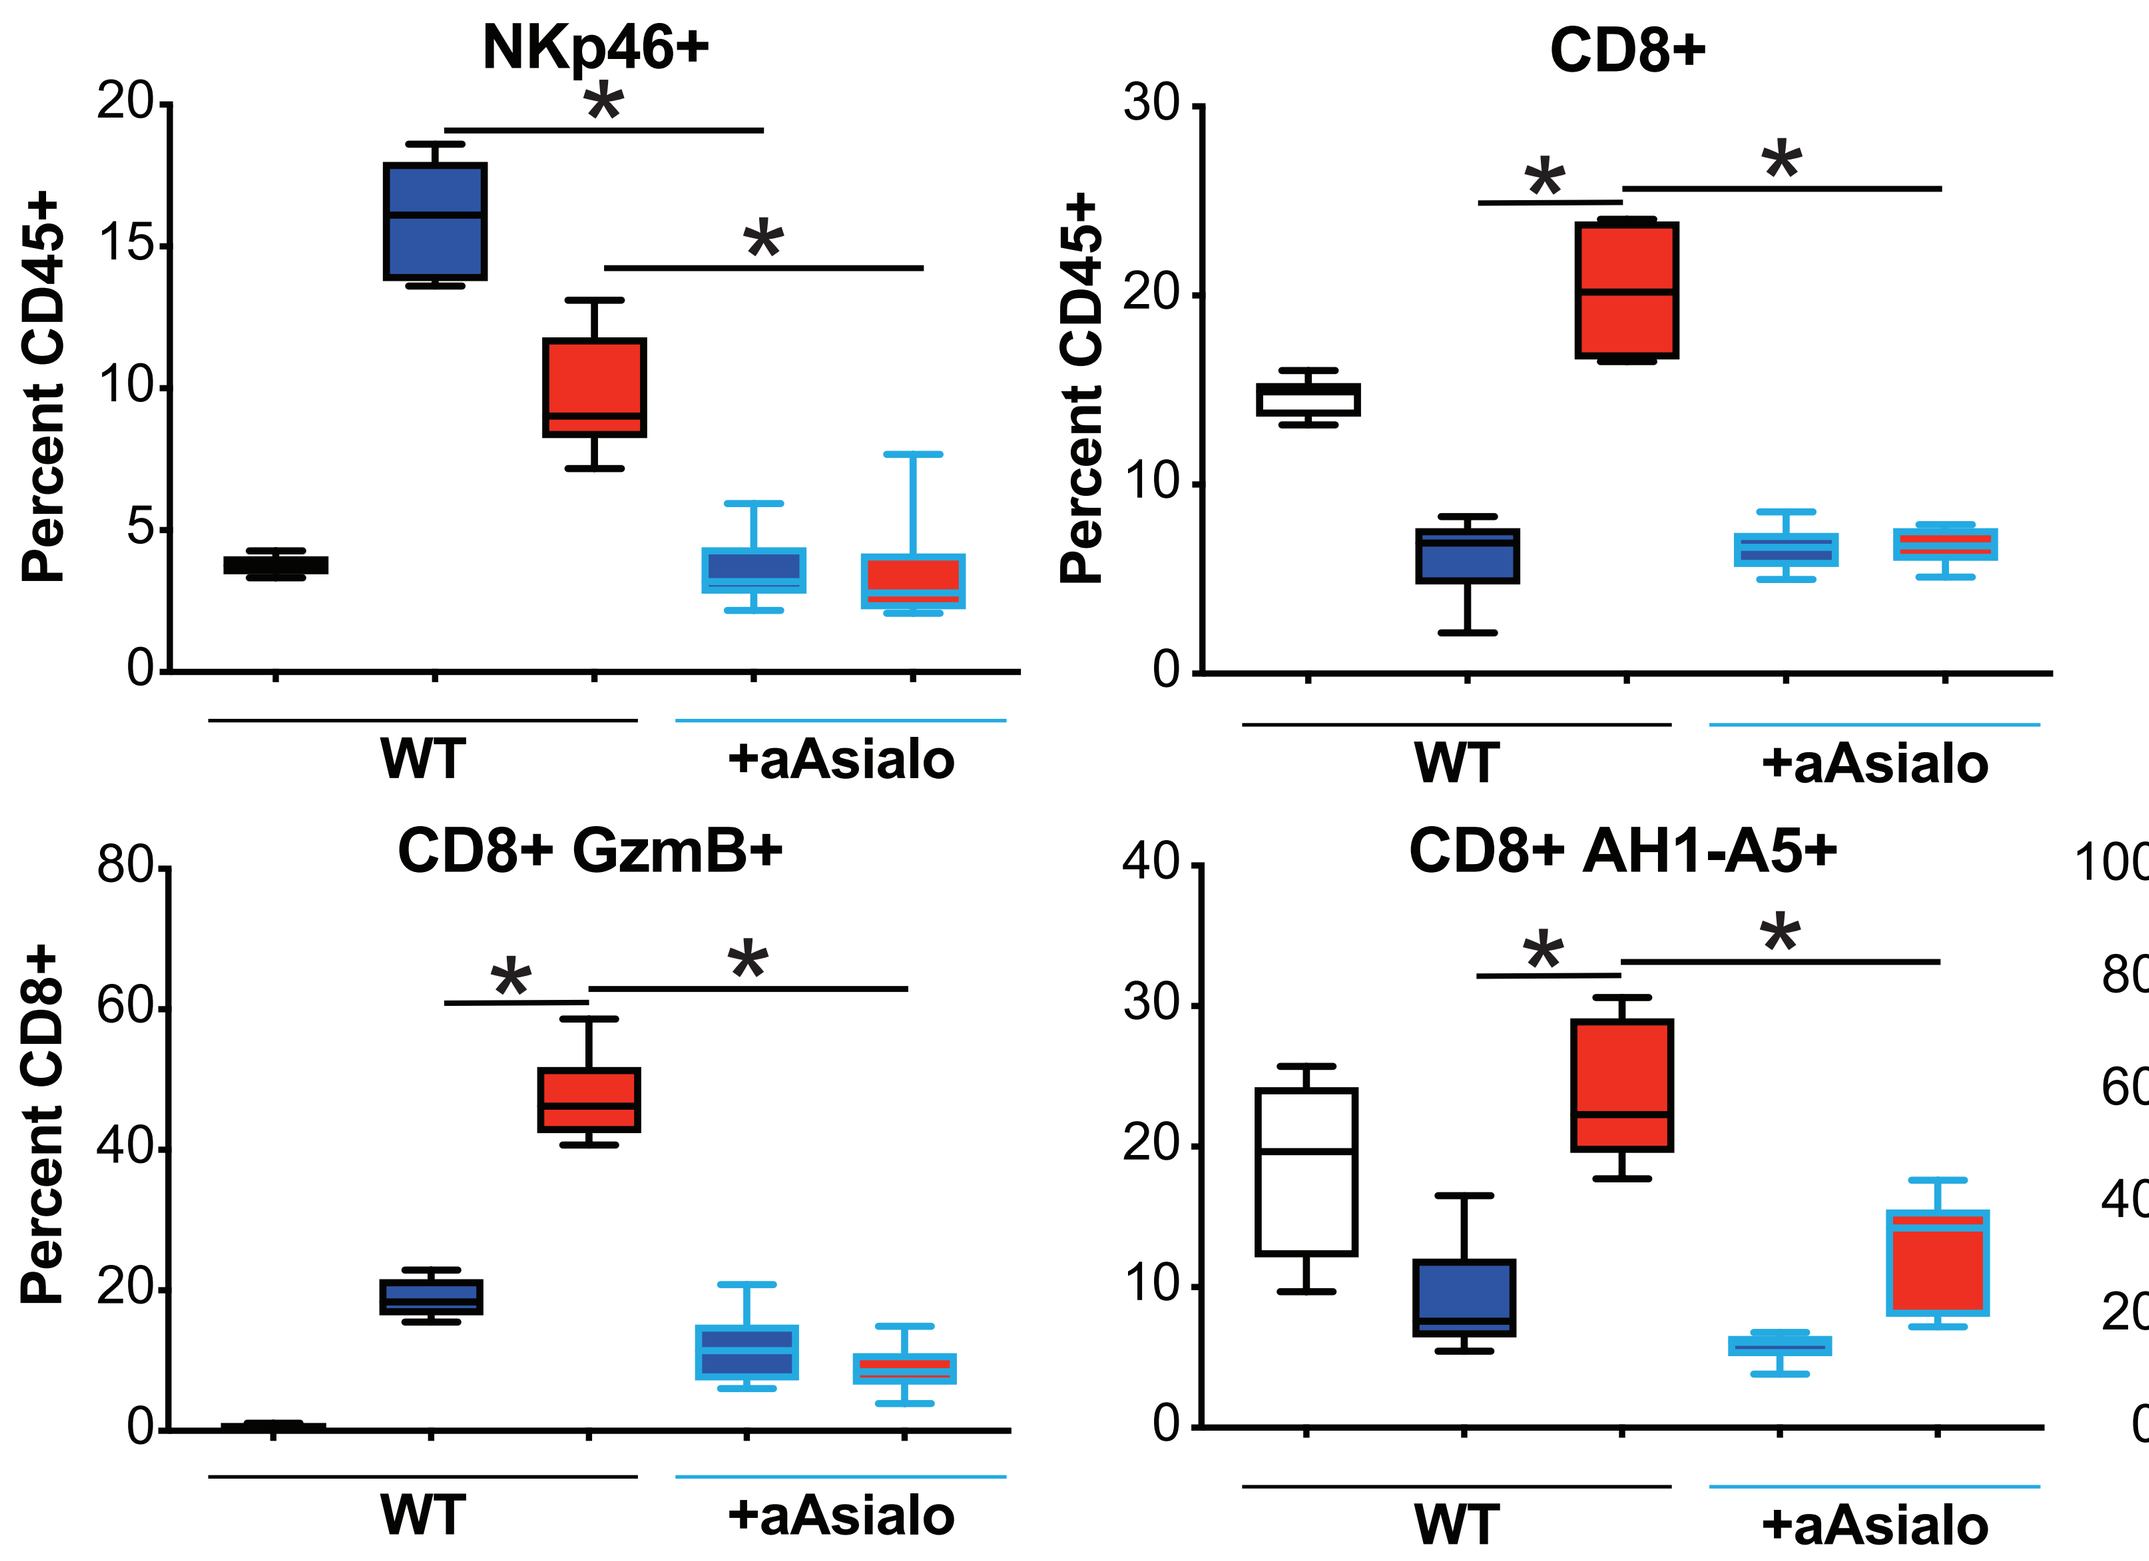
NKp46+
CD8+
CD8+ GzmB+
CD8+ AH1-A5+
Percent CD45+
Percent CD8+
WT
+aAsialo

- Blood drawn 4 days post-treatment; aAsialo delivered 1 day prior to therapy. Flow cytometry used to monitor changes to immune cell phenotypes. N=5, one experiment. ANOVA, *p<0.05.
- Loss of NK cell expansion (light blue outline) prevented expansion of CD8 T cells in the periphery. The CD8 T cells that were present had reduced granzyme B, AH1-A5+ (tumor specific), and Ki-67 (proliferation).
- Changes due to reduction of NK cells were more significant in the BEMPEG/NKTR-262 combination therapy than they were in BEMPEG/RT combination therapy.

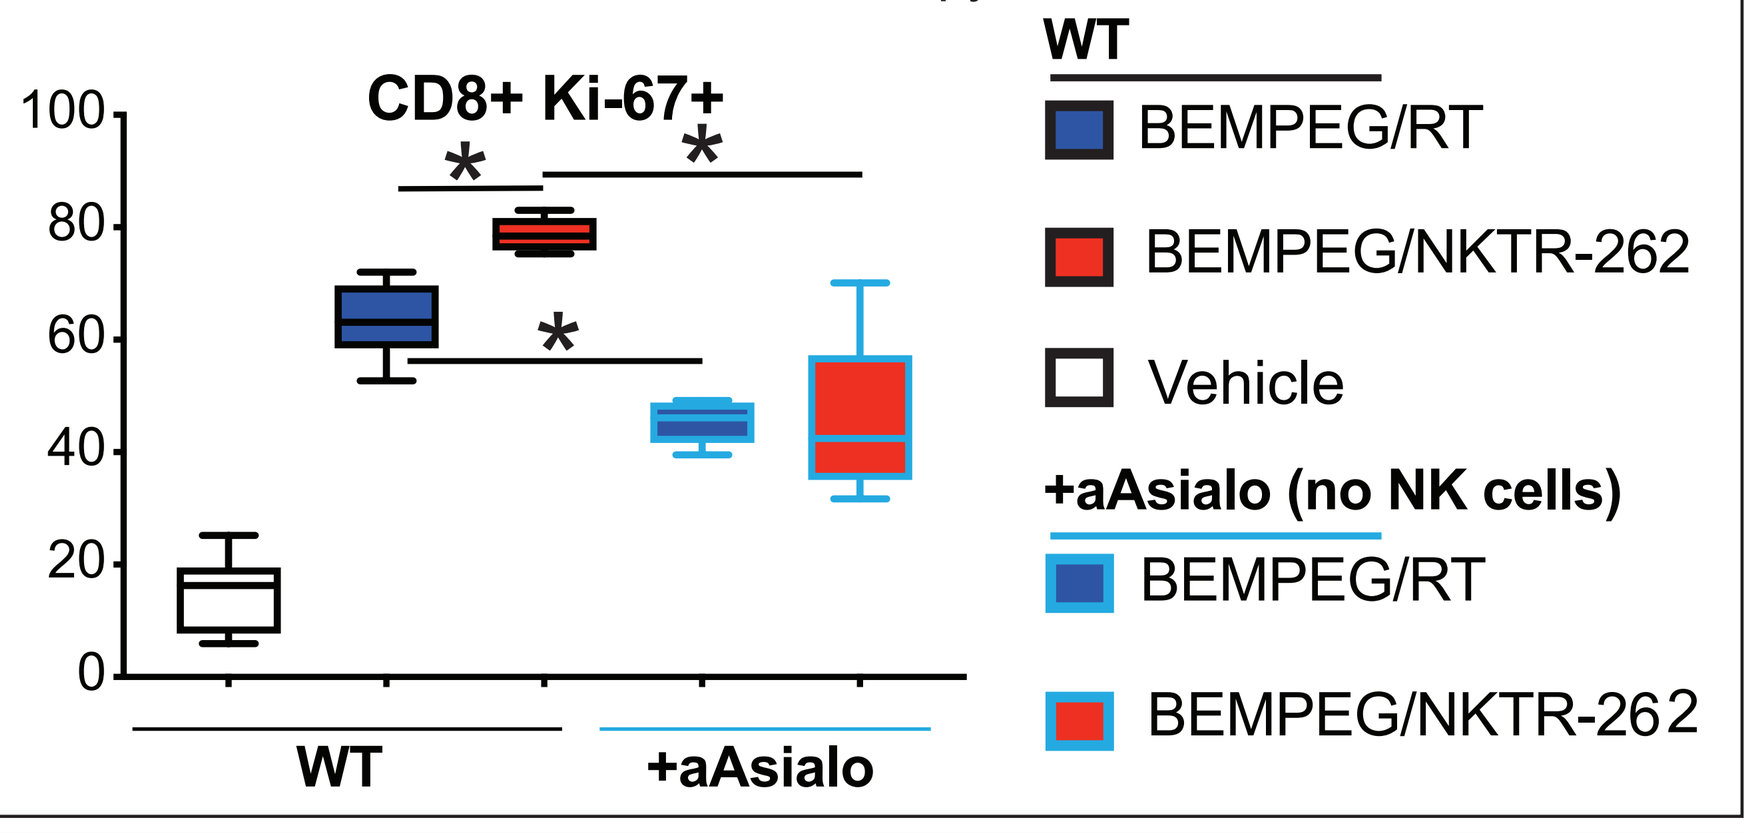
CD8+ Ki-67+
WT
+aAsialo
WT
BEMPEG/RT
BEMPEG/NKTR-262
Vehicle
+aAsialo (no NK cells)
BEMPEG/RT
BEMPEG/NKTR-262

3 Tumor infiltrating immune cell phenotyping

BEMPEG/NKTR-262 induces less exhausted and more active CD8 T cell responses

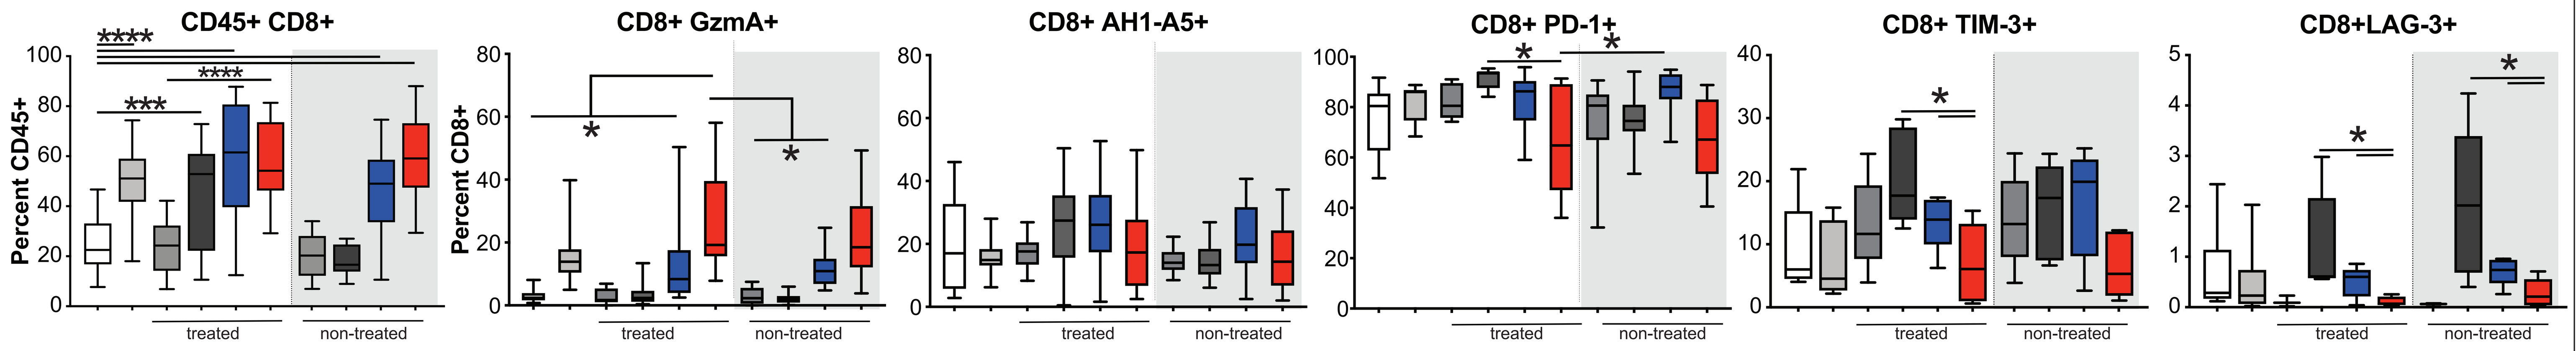
Vehicle
BEMPEG
NKTR-262
RT
BEMPEG/RT
BEMPEG/NKTR-262
CD45+ CD8+
CD8+ GzmA+
CD8+ AH1-A5+
CD8+ PD-1+
CD8+ TIM-3+
CD8+LAG-3+
Percent CD45+
Percent CD8+
treated
non-treated

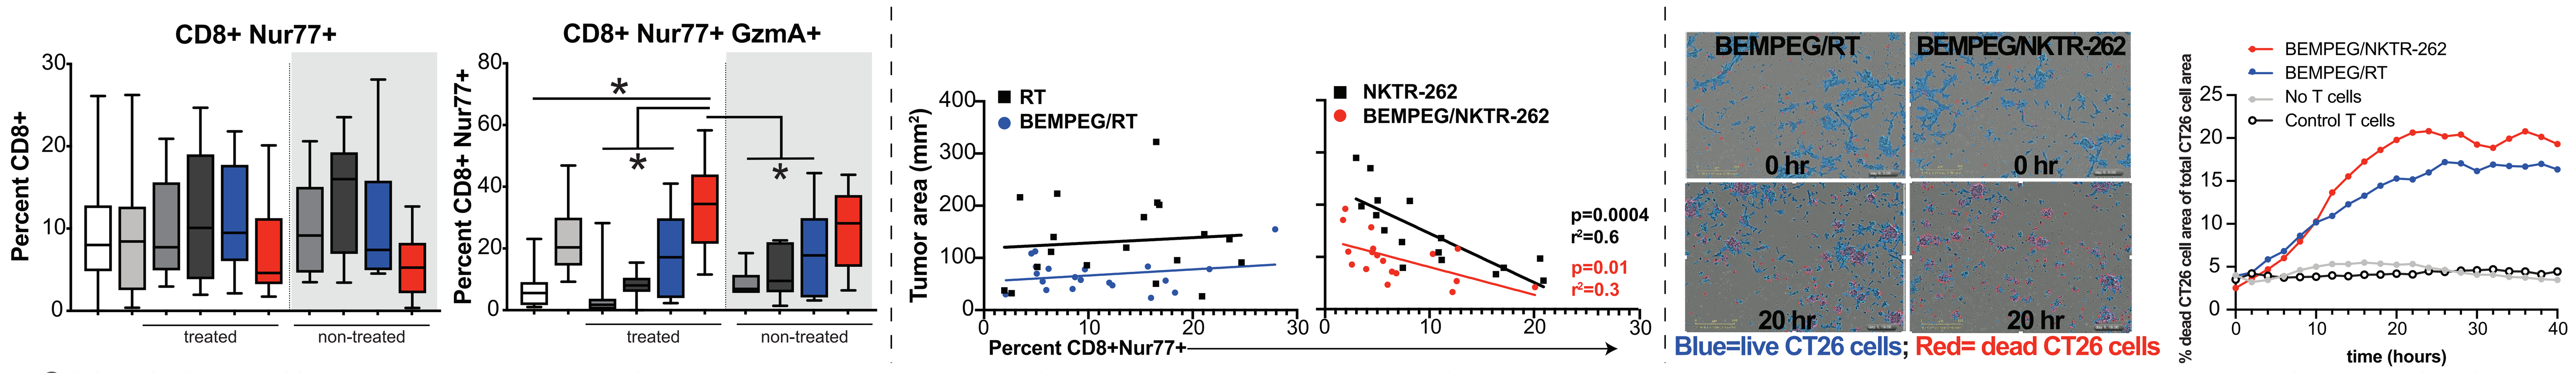
CD8+ Nur77+
CD8+ Nur77+ GzmA+
Percent CD8+
Percent CD8+ Nur77+
treated
non-treated
RT
BEMPEG/RT
NKTR-262
BEMPEG/NKTR-262
Tumor area (mm²)
Percent CD8+Nur77+
p=0.0004
r²=0.6
p=0.01
r²=0.3
BEMPEG/RT
BEMPEG/NKTR-262
0 hr
20 hr
Blue=live CT26 cells; Red= dead CT26 cells
No T cells
Control T cells
% dead CT26 cell area of total CT26 cell area
time (hours)

- (TOP ROW) BEMPEG/NKTR-262 treatment resulted in CD8 T cells with enhanced expression of gramzyme A and reduced expression of exhaustion markers PD-1, Tim3, and Lag3. N>8 from 2 independent experiments. One-way ANOVA; *p<0.05, ***p<0.001, ****p<0.0001.
- (BOTTOM ROW) Nur77-GFP+ mice bearing dual flank CT26 tumors treated with BEMPEG/NKTR-262 had a similar fraction of CD8+ Nur77+ cells, but the Nur77+ cells expressed increased GzmA levels (left). The Nur77+ cells induced by BEMPEG/NKTR-262 are functional anti-tumor cells, which is represented by a significant negative relationship between those cells and tumor size in the BEMPEG/NKTR-262 treated mice. N>8 from two independent expt (middle). Incucyte assay with CD8 T cells isolated from tumors treated with either BEMPEG/NKTR-262 or BEMPEG/RT demonstrated CD8 T cells from BEMPEG/NKTR-262 tumors are superior at killing CT26 cells grown in culture (right, from 1 experiment).

NK cells increase CD8+ T cell tumor infiltrate, 7 dpt

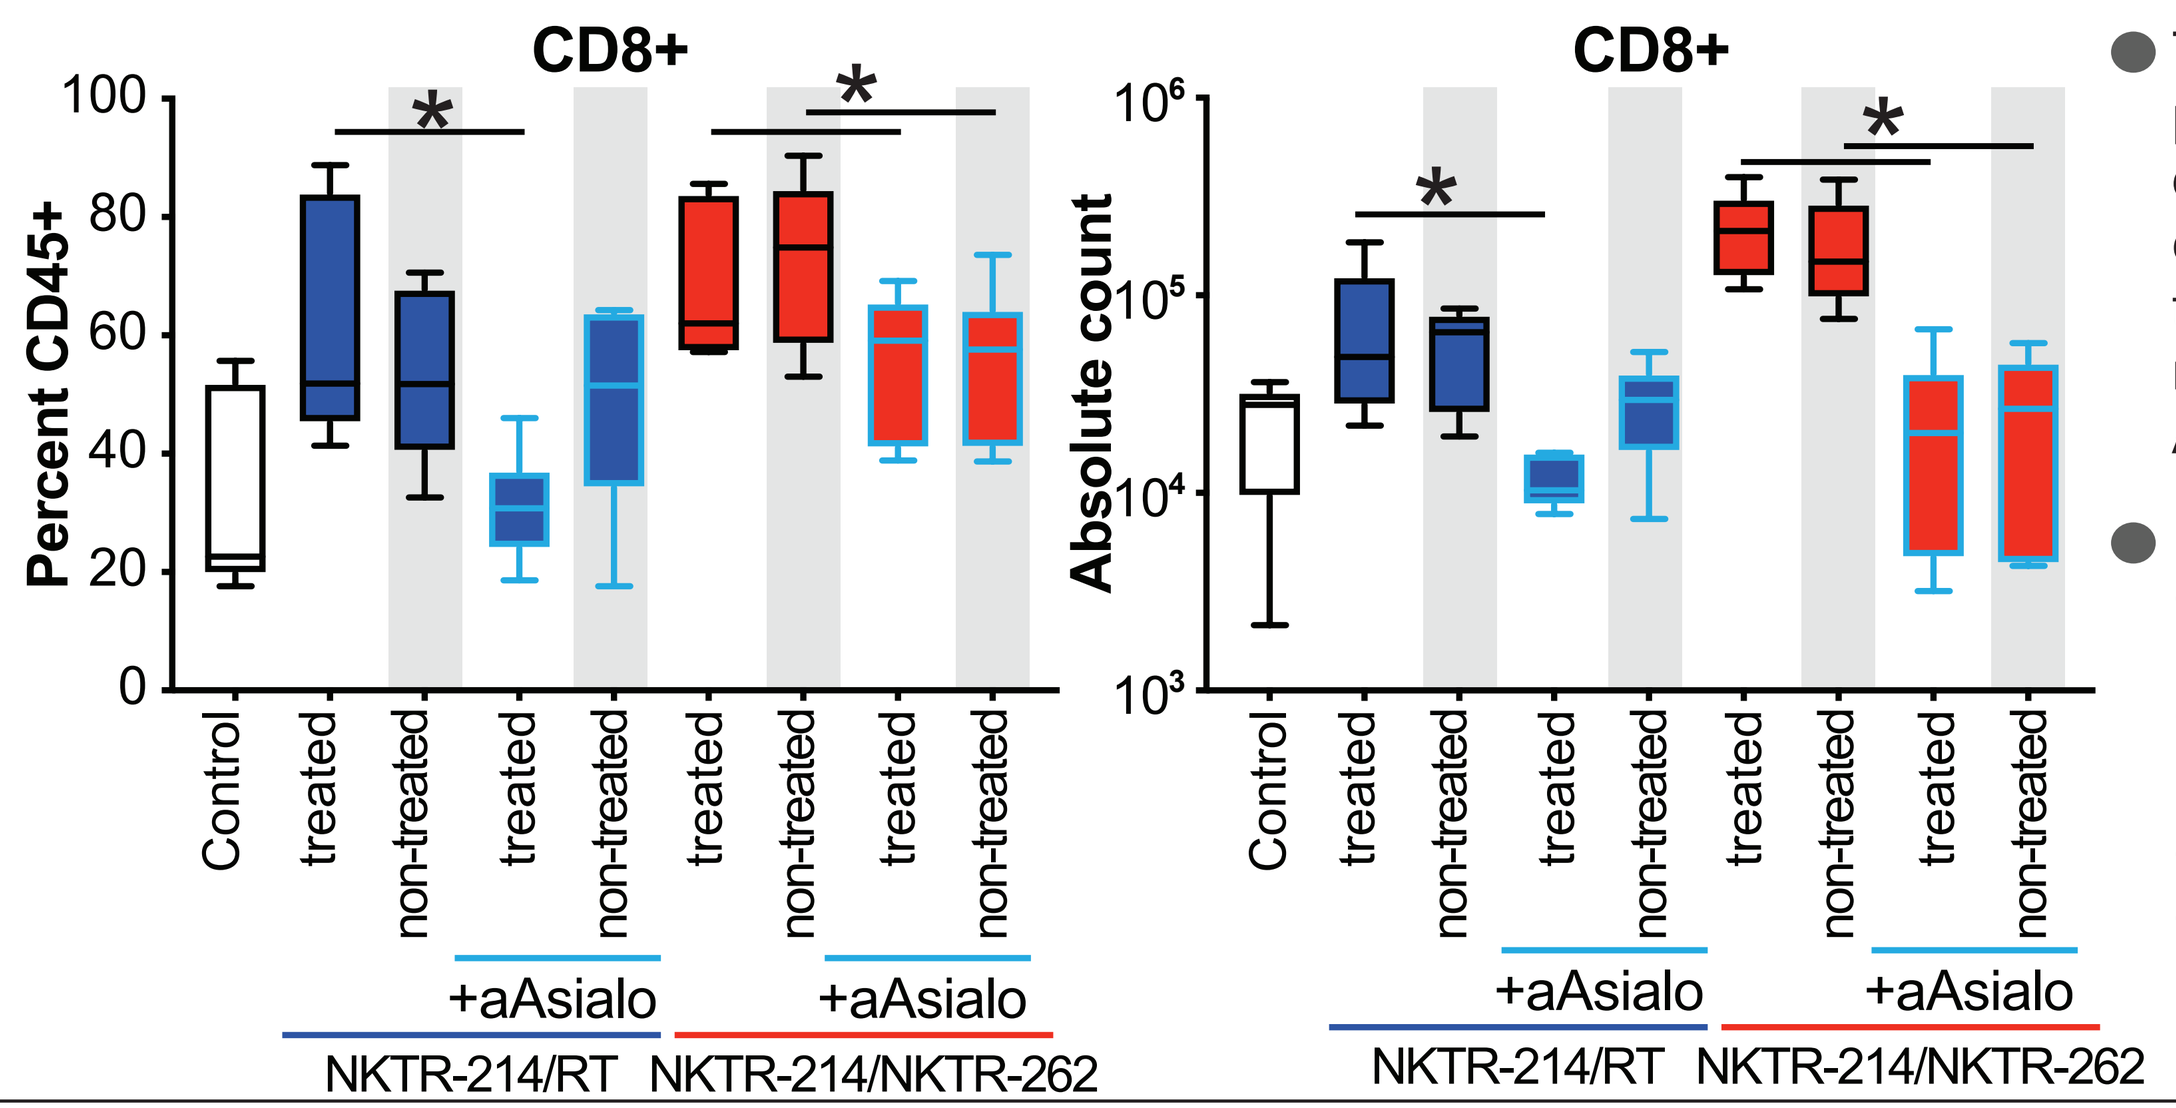
CD8+
Percent CD45+
Absolute count
Control
treated
non-treated
+aAsialo
NKTR-214/RT
NKTR-214/NKTR-262

- Tumors harvested 7 days post-treatment. aAsialo antibody delivered 1 day prior to therapy. Flow cytometry used to monitor changes to immune cell phenotypes. N=5 mice from 1 experiment. One way ANOVA, *p<0.05.
- Loss of NK cell expansion reduces the % (left) and absolute number (right) of CD8 T cells in the tumor. The effect is greater in the BEMPEG/NKTR-262 treated tumors than in the BEMPEG/RT treated tumors.

NKTR-262 increases high effector NK cells, 1 dpt

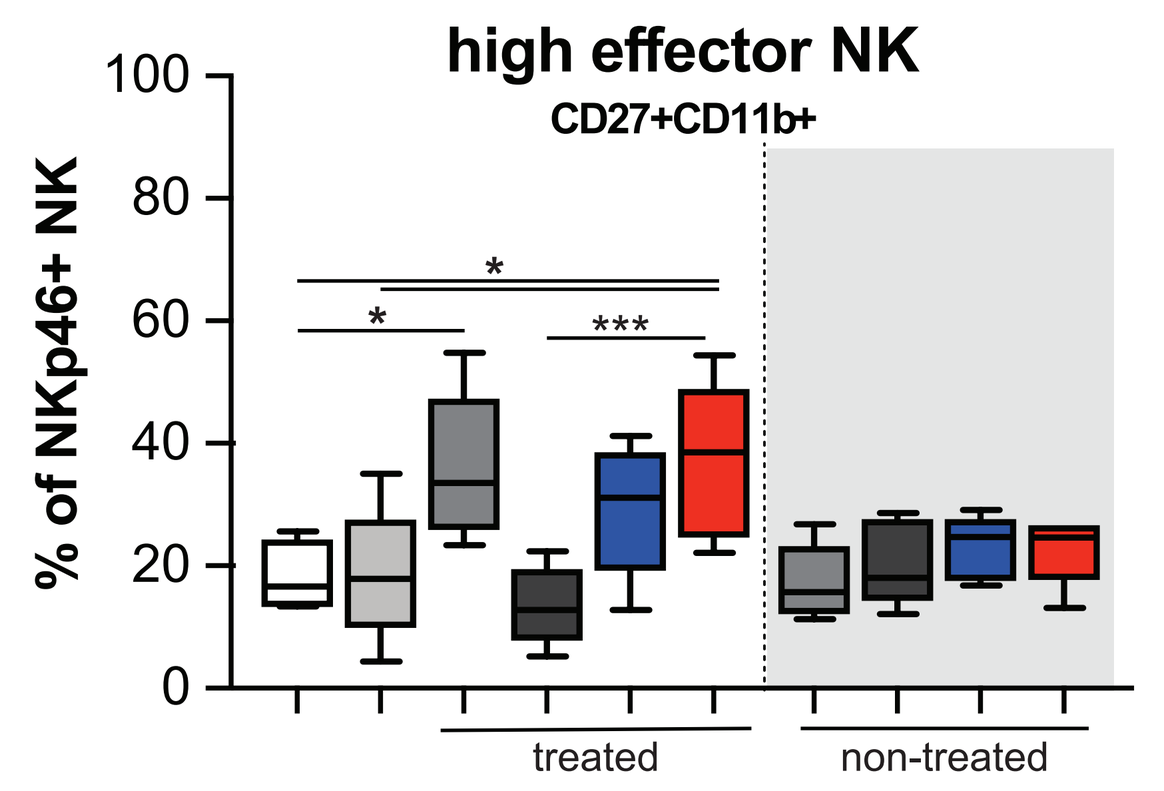
high effector NK
CD27+CD11b+
% of NKp46+ NK
treated
non-treated

Vehicle, BEMPEG, NKTR-262, RT, BEMPEG/RT, BEMPEG/NKTR-262

- Tumors harvested 1 (shown) and 3 days (not shown) post-treatment. aAsialo antibody delivered 1 day prior to therapy. Flow cytometry used to monitor changes to immune cell phenotypes. N=5 mice from 1 experiment. One way ANOVA; *p<0.05, ***p<0.001.
- NKTR-262 induces an increase in the percent of high effector NK cells (CD11b+CD27+) in the tumor early post treatment. The overall number of total NK cells is not significantly different across groups (data not shown).

Conclusions

- BEMPEG/NKTR-262 treatment produces a higher fraction of activated tumor antigen-specific cytotoxic CD8 T cells systemically, correlating with superior anti-tumor efficacy relative to BEMPEG combined with RT.
- Efficacy of BEMPEG/NKTR-262 combination therapy depends on CD8 T cells and NK cells.
- BEMPEG/NKTR-262 combination therapy induces intratumoral CD8 T cells that have increased activity as deomonstrated by increased granzyme expression and increased tumor killing, and reduced conversion to an exhausted phenotype (PD-1+, TIM-3+, LAG-3+)
- Loss of NK cells reduces CD8 T cell percentages and function in the peripheral blood and in the tumor, suggesting a connection between early NK cell function and anti-tumor adaptive immune responses.

Acknowledgements

Nektar Therapeutics

EACRI Flow Core and Cancer Research Animal Division (CRAD)

NEKTAR