Poster No. 1603/11

NKTR-255 Engages the IL-15 Pathway Driving CD8 T Cell Survival and CD8 Memory T Cell Proliferation

Peiwen Kuo, Mekhala Maiti, Phi Quach, Murali Addepalli, Arunasree Lanka, Poornachandra Mathamsetti, Christie Fanton, Ping Zhang, Peter Kirk, Takahiro Miyazaki, Jonathan Zalevsky

Nektar Therapeutics, San Francisco, California

NEKTAR

Introduction

- IL-15-based therapy has great potential to augment immune responses
 - IL-15 plays important roles in both the innate and adaptive immune system
 - IL-15, a pleiotropic cytokine, is a key regulator of homeostasis and survival of CD8 and CD8 memory T cells
 - Targeting the IL-15 pathway is a promising therapeutic approach to induce long-term T cell activation and durable memory responses
- Rapid plasma clearance of conventional IL-15 poses a significant limitation in its development as an immuno-oncology therapeutic. We therefore engineered a polymer-conjugated IL-15 designed to deliver sustained signaling, which overcomes C_{max}-driven cytotoxicity of naked IL-15[1]

NKTR-255-Mediated Signaling Through Trans-Presentation[2]

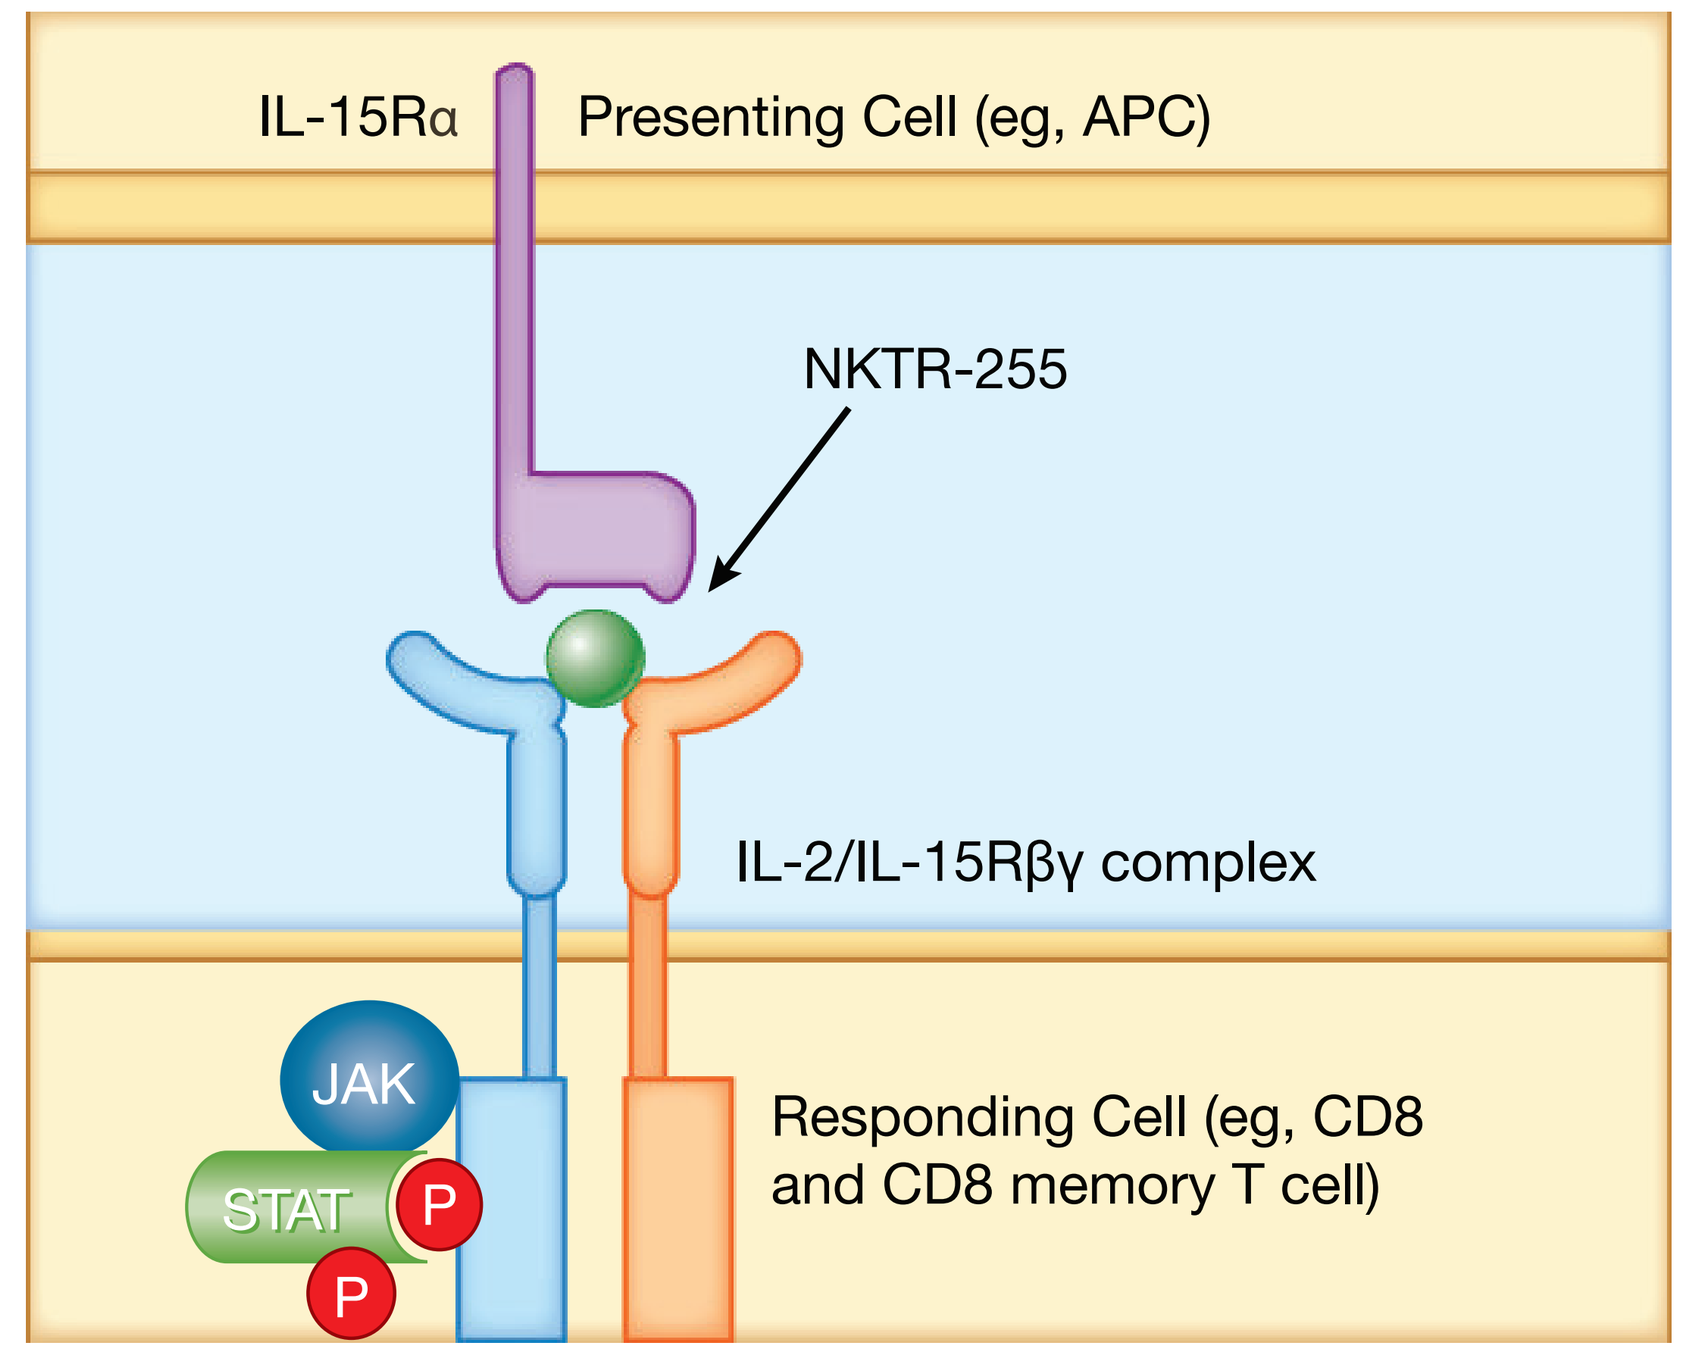
IL-15Rα
Presenting Cell (eg, APC)
NKTR-255
IL-2/IL-15Rβγ complex
JAK
STAT
P
P
Responding Cell (eg, CD8 and CD8 memory T cell)

A unique IL-15 receptor alpha (IL-15Rα) chain and IL-15 cytokine are synthesized in the presenting cell, bound together in the endoplasmic reticulum, and transported to the cell surface. The IL-15/IL-15Rα complex is presented to the IL-2/IL-15Rβγ-receptor complex on the adjacent responding cell (*trans*-presentation). Engagement of the IL-2/IL-15Rβγ can induce JAK-STAT signaling, increasing CD8 and CD8 memory T cells survival and proliferation.[3] NKTR-255 is designed to optimally engage the IL-15 receptor complex and provide durable pathway activation.

IL-2/IL-15Rβγ, receptor subunit shared by IL-2 and IL-15; APC, antigen-presenting cell; JAK, Janus Kinase; STAT, signal transducer and activator of transcription; P, phosphorylation.

- NKTR-255, a polymer-engineered IL-15, is highly active and provides long cytokine exposure to improve CD8 T cell memory
 - Plasma and tumor pharmacokinetics reveal improved exposure compared to IL-15
 - Provides sustained engagement of the IL-15 pathway by signaling through the private IL-15Rα and shared IL-2/IL-15Rβγ complex in vivo

NKTR-255 Induces Sustained Signaling in CD8 T Cells

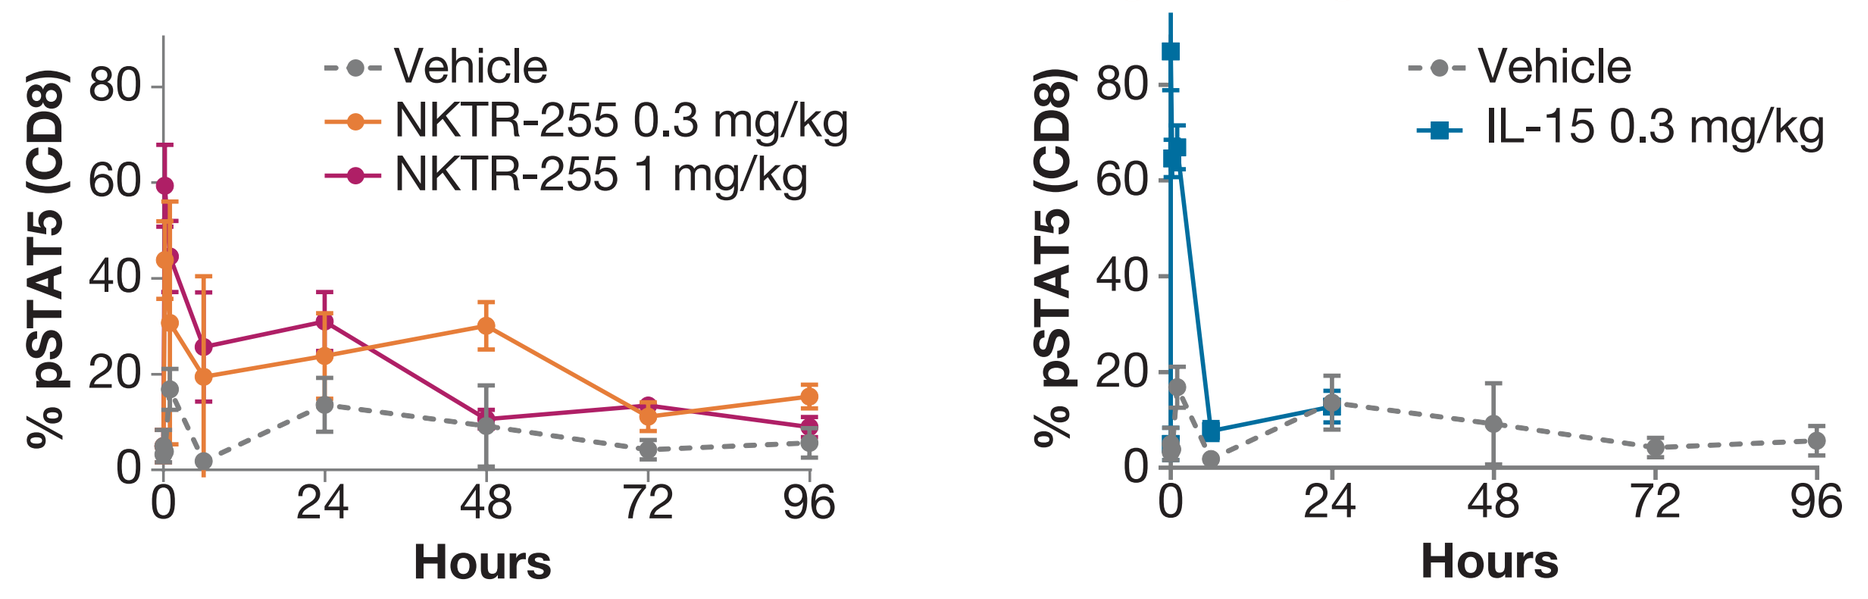
% pSTAT5 (CD8)
Vehicle
NKTR-255 0.3 mg/kg
NKTR-255 1 mg/kg
IL-15 0.3 mg/kg
Hours

STAT5 phosphorylation persists within CD8 T cells after NKTR-255 treatment (left), whereas parent IL-15 action is short-lived (right).

Methods

- Immunophenotyping of CD8 T cell subpopulations was performed in naïve and tumor-bearing Balb/c mice treated with NKTR-255. Cell surface staining of CD3, CD8, CD44, CD62L and Sca-1 was conducted to identify effector (Tem: CD44hi, CD62Llo), central (Tcm: CD44hi, CD62Lhi) and stem (Tscm: CD44hi, CD62Lhi, Sca-1+) memory T cells. Intracellular staining of Ki-67 and Bcl-2 were also analyzed by flow cytometry.
- The CT-26 metastasis model involved injection of 1x10^5 tumor cells through the tail vein of Balb/c mice on day 0. NKTR-255 at 0.03, 0.1, 0.3, 1 or 3mg/kg and IL-15 at 0.3mg/kg were dosed on days 1, 5, and 10. Lung nodule quantification and immunophenotyping were conducted three days after the last dose.
- Cynomolgus monkeys received a single injection of NKTR-255 at various doses and were subject to immunophenotyping of CD8 subpopulations, including effector memory cells (CD45A- CD197-) and central memory cells (CD45RA-CD197+).
- Human whole blood and peripheral blood mononuclear cells (PBMCs) were stimulated with IL-15 (0.0001-1000 ng/ml) or NKTR-255 (0.001-10,000 ng/ml). pSTAT5 response in CD8 T cells was monitored by flow cytometry at various time points.

Results

Figure 1. Single Dose NKTR-255 Increases CD8 Memory T Cells

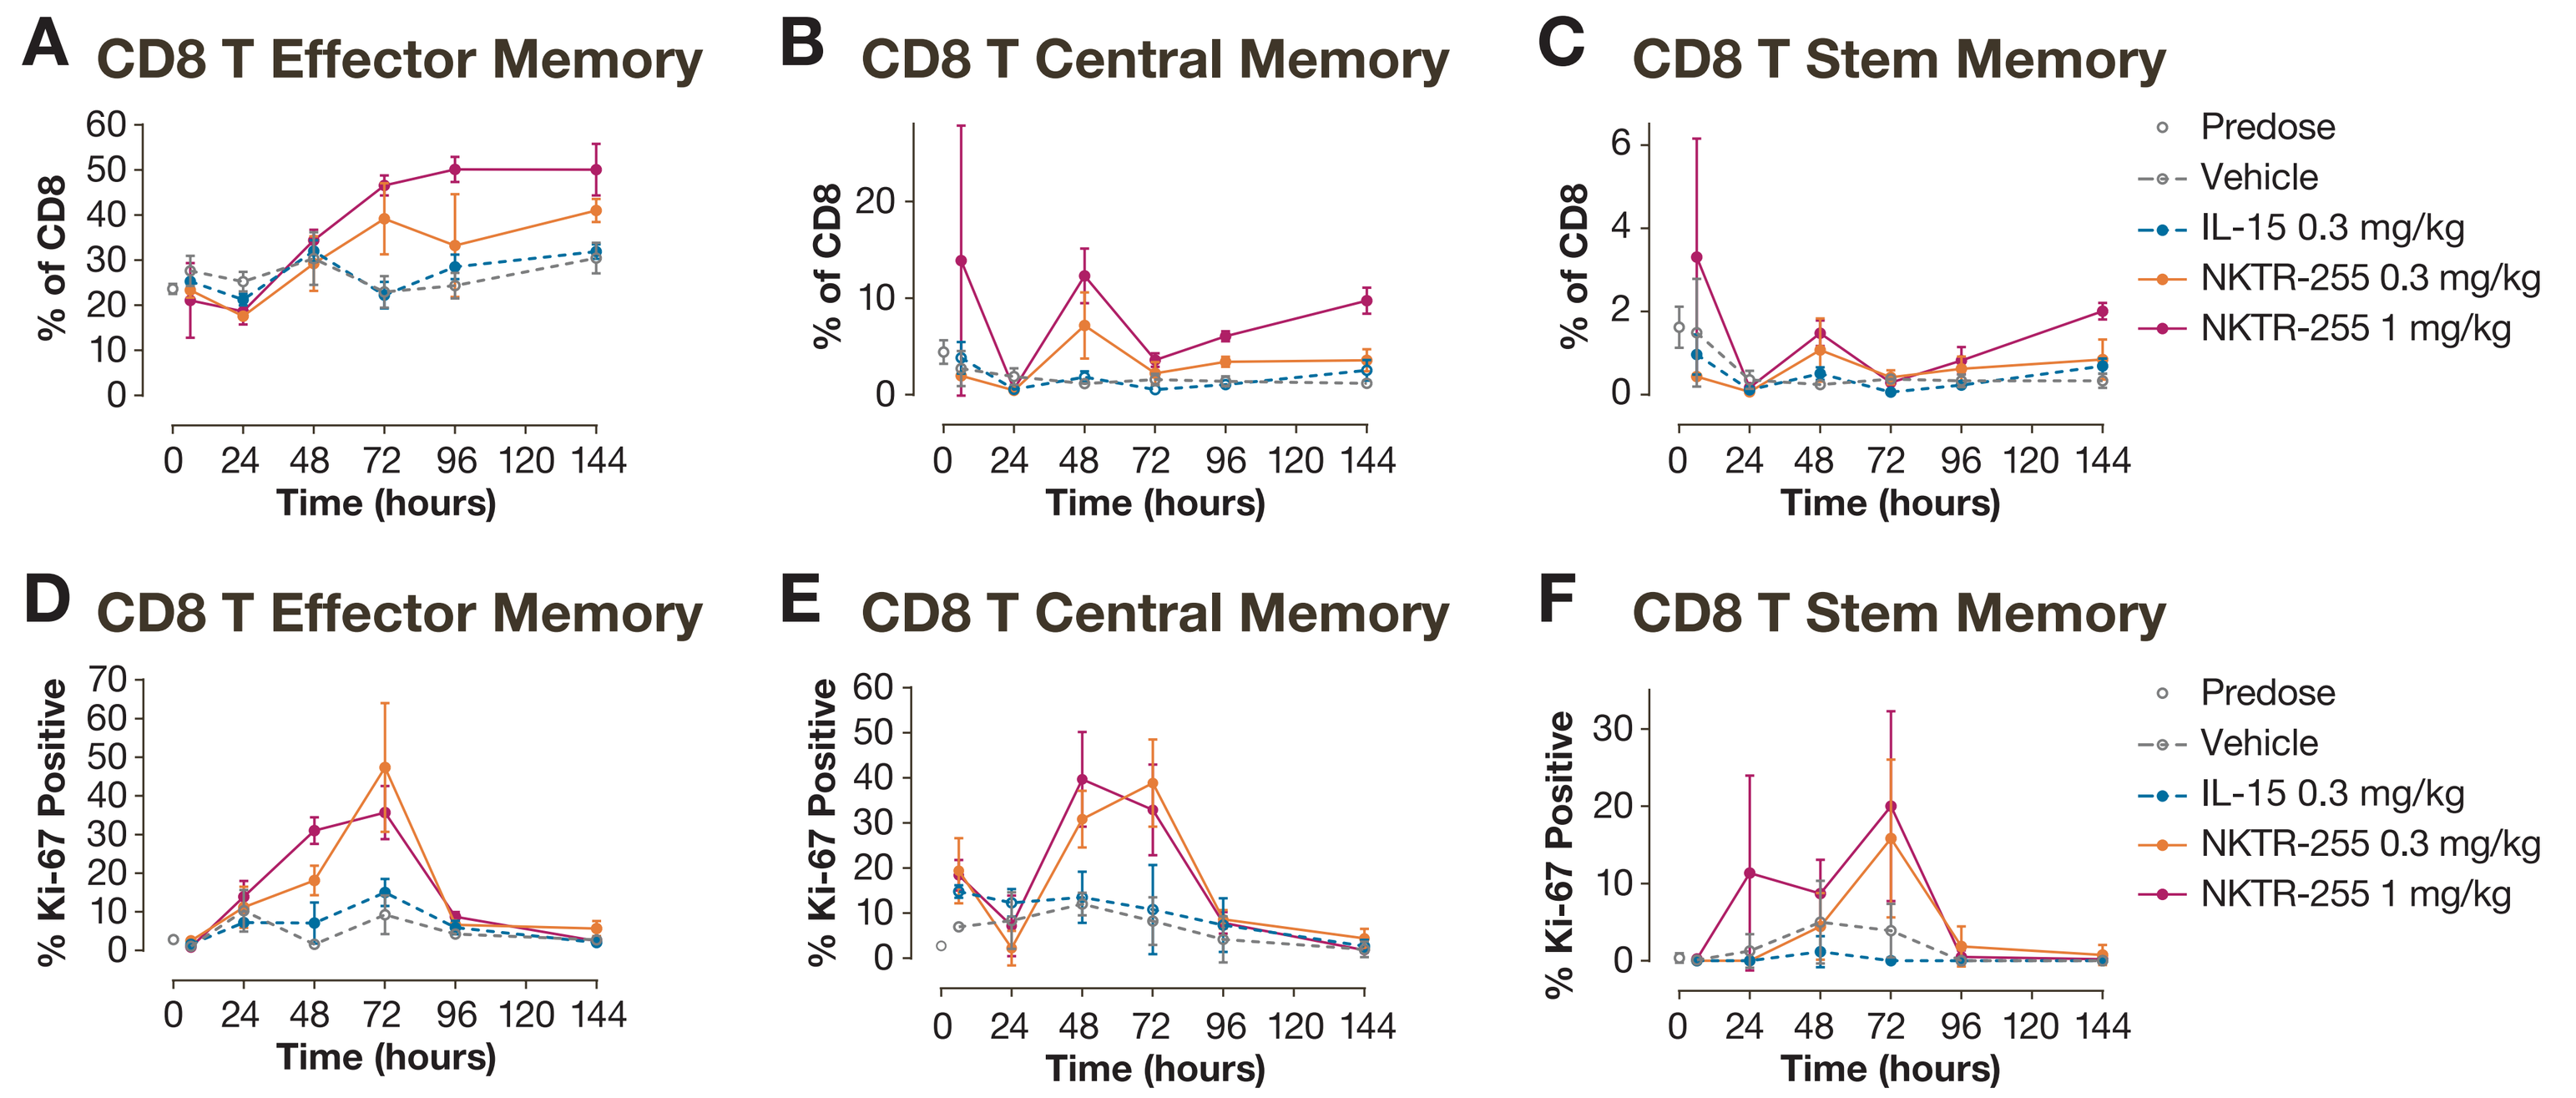
A CD8 T Effector Memory
B CD8 T Central Memory
C CD8 T Stem Memory
% of CD8
D CD8 T Effector Memory
E CD8 T Central Memory
F CD8 T Stem Memory
% Ki-67 Positive
Time (hours)
Predose
Vehicle
IL-15 0.3 mg/kg
NKTR-255 0.3 mg/kg
NKTR-255 1 mg/kg

Tem, T effector memory; Tcm, T central memory; Tscm, T stem memory.

(A-C) In naïve mice, single dose NKTR-255 (0.3-1 mg/kg) increased the number of CD8 Tem, Tcm, and Tscm in a dose-dependent manner. At the 0.3 mg/kg tumor efficacy dose, maximum fold increases of 1.8, 4.6, and 3.2 in CD8 Tem, Tcm, and Tscm subpopulations, respectively, compared to vehicle were observed. NKTR-255 administered at 1 mg/kg induced CD8 Tem, Tcm, and Tscm maximum fold increases of 2.1, 7.9, and 6.1, respectively, compared to vehicle. The expansion of the memory subpopulations persisted for at least 144 hours after a single dose. A single administration of conventional IL-15 did not affect CD8 memory T cells.

(D-F) NKTR-255 boosts CD8 memory T cell proliferation. Ki-67 positive CD8 Tem, Tcm, and Tscm increased 5.1, 4.7, and 4.1 fold, respectively, compared to vehicle at 0.3 mg/kg. In CD8 Tem, Tcm, and Tscm Ki-67 positivity increased 3.9, 3.3, and 5.1 fold, respectively, compared to vehicle at 1 mg/kg. Proliferation peaked between 48 to 72 hours after treatment. Single-dose IL-15 did not increase proliferation among the memory subpopulations.

Figure 2. NKTR-255 Single Agent Efficacy in a CT-26 Lung Metastasis Model

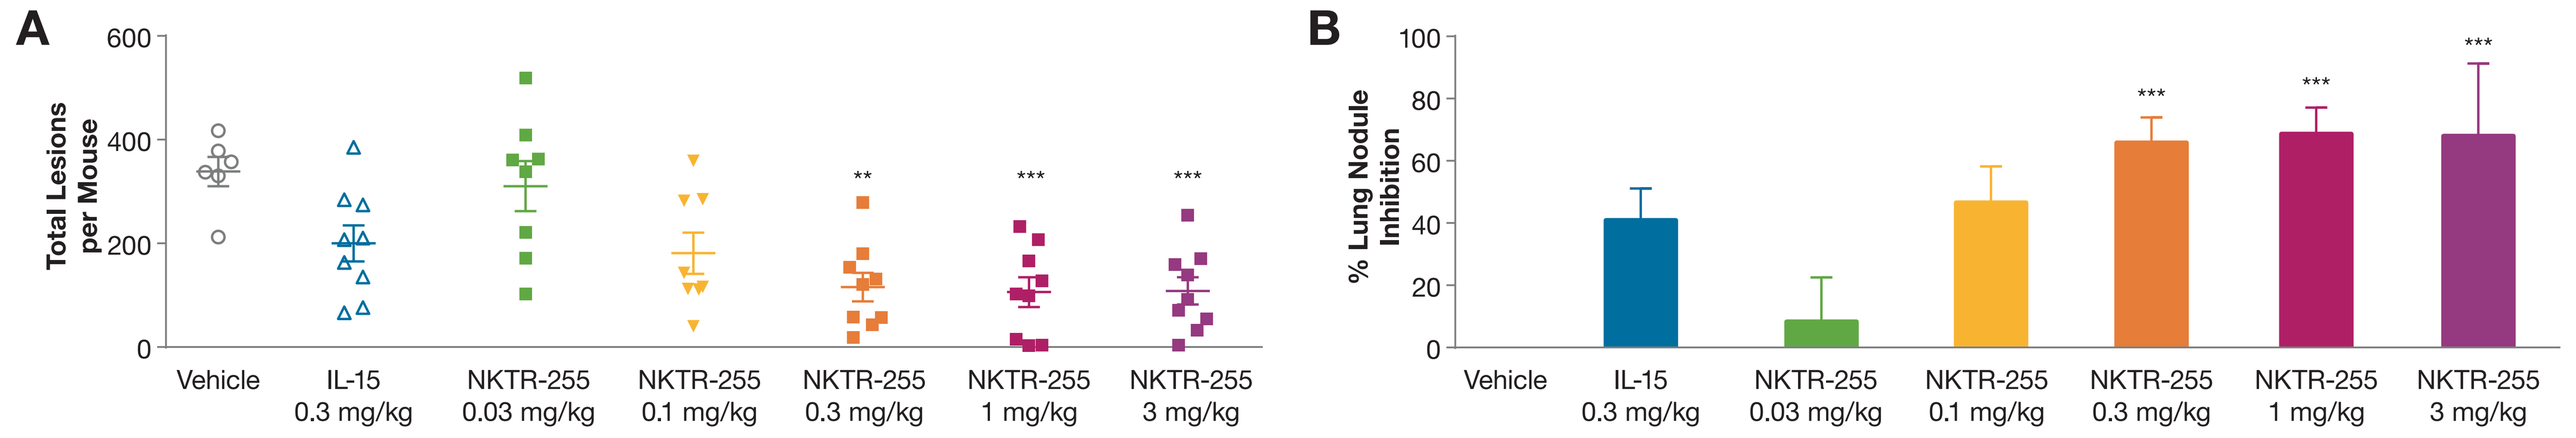
A
Total Lesions per Mouse
B
% Lung Nodule Inhibition
Vehicle
IL-15 0.3 mg/kg
NKTR-255 0.03 mg/kg
NKTR-255 0.1 mg/kg
NKTR-255 0.3 mg/kg
NKTR-255 1 mg/kg
NKTR-255 3 mg/kg

p<0.01, * p<0.001 One-way ANOVA followed by Tukey's post-test versus vehicle.

(A) Dose response of NKTR-255 in a CT-26 lung metastasis model demonstrates significant reduction of lung nodules at 0.3 to 3 mg/kg dose levels.

(B) NKTR-255 is approximately three times more potent than parent IL-15 as an inhibitor of lung nodule formation.

Figure 3. NKTR-255 Increases CD8 T Cell Proliferation and Survival in Tumor-Bearing Mice

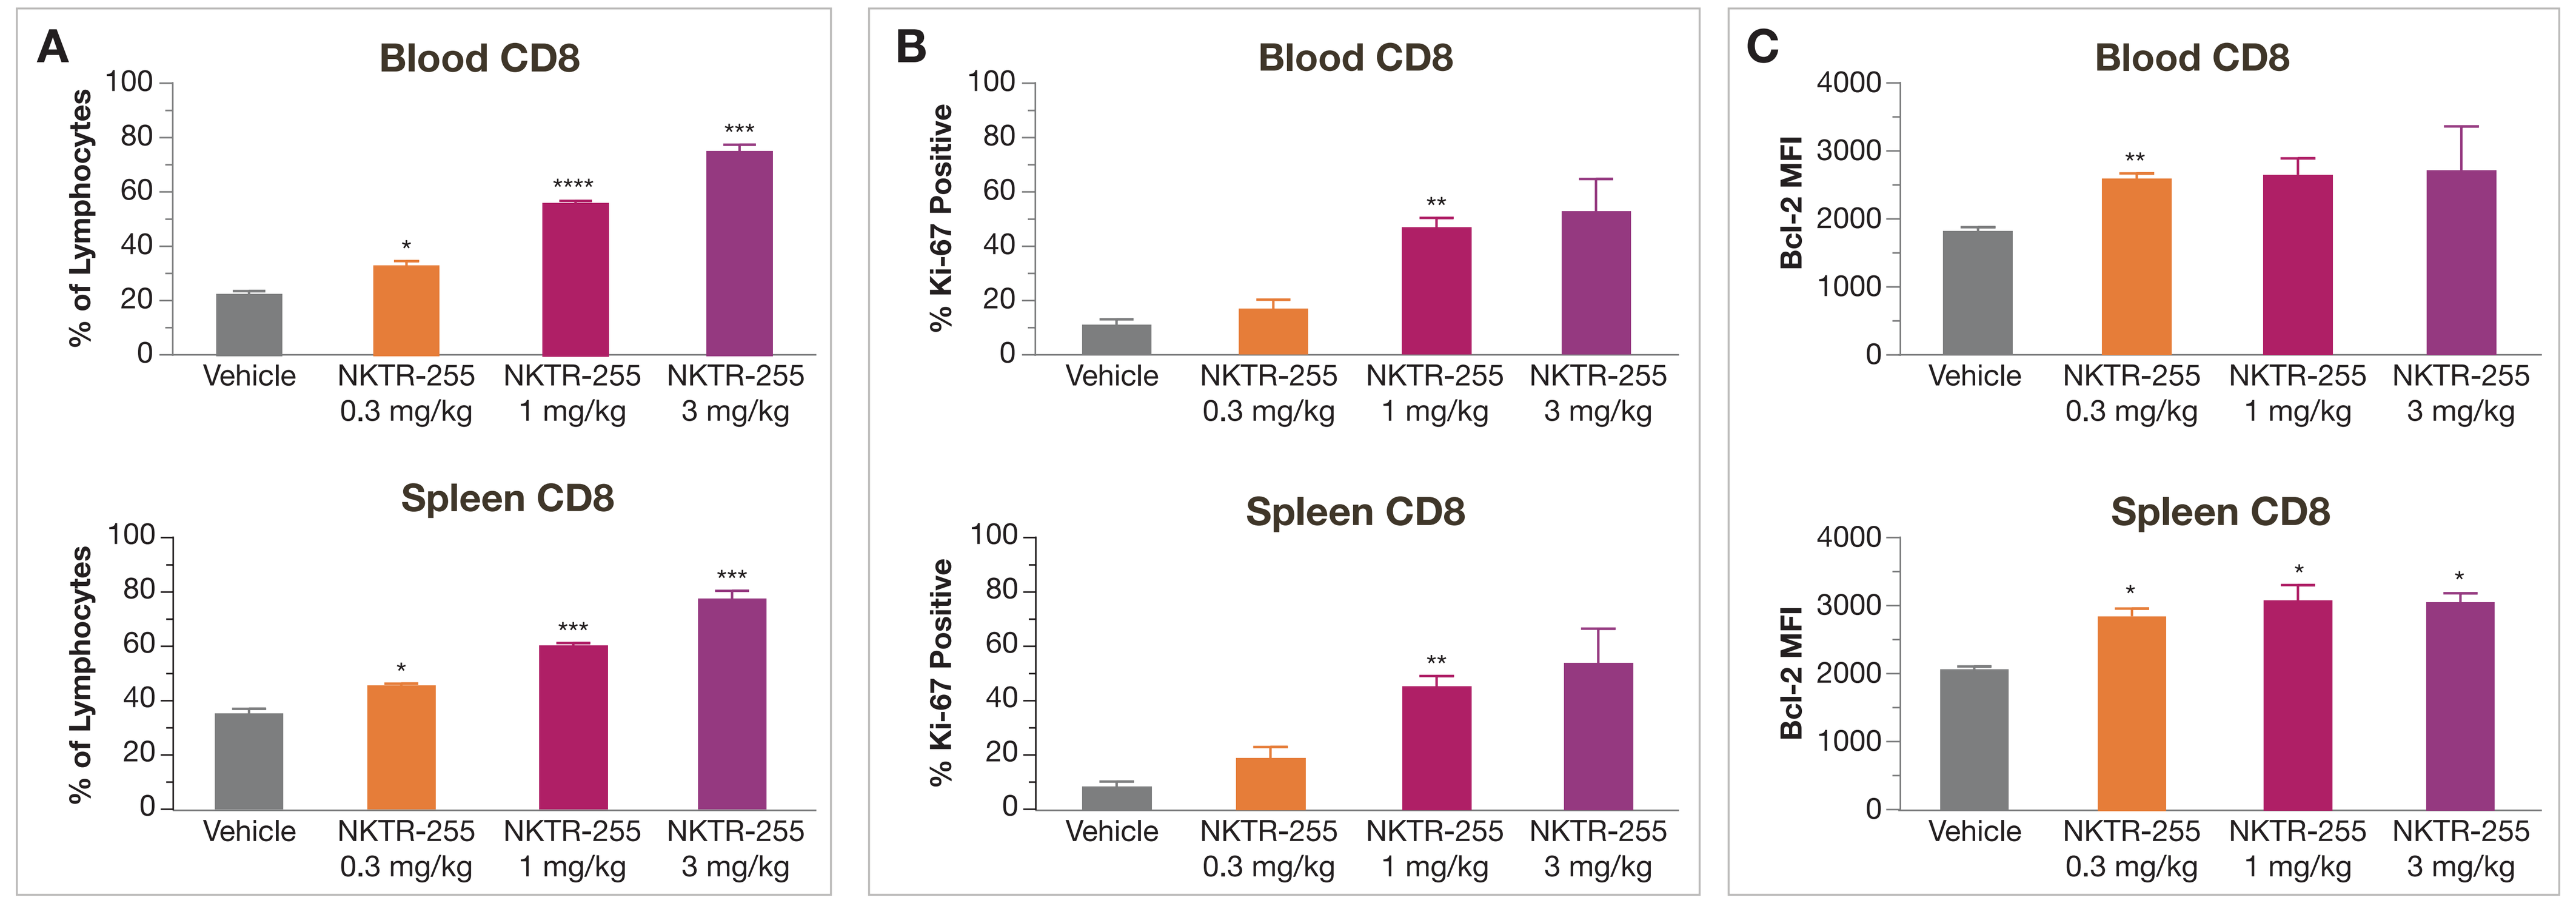
A
B
C
Blood CD8
Spleen CD8
% of Lymphocytes
% Ki-67 Positive
Bcl-2 MFI
Vehicle
NKTR-255 0.3 mg/kg
NKTR-255 1 mg/kg
NKTR-255 3 mg/kg

MFI, mean fluorescence index.

*p<0.05, **p<0.01, ***p<0.001, ***p<0.0001 vs vehicle.

(A) In blood, NKTR-255 administered at 0.3, 1, and 3 mg/kg induced a dose-dependent increase of CD8 T cells 1.5, 2.5, and 3.3-fold versus vehicle, respectively. Similar observations were made in spleen with increases of 1.3, 1.7, and 2.2-fold at 0.3, 1, and 3 mg/kg dose levels versus vehicle.

(B) Ki-67 immunophenotyping revealed significant dose-dependent increases in CD8 T cell proliferation in blood with 1.7, 4.6, and 5.3-fold changes and spleen with 2.5, 5.7, and 6.9-fold changes at the same low, mid, and high dose levels compared to vehicle.

(C) Finally, NKTR-255 treatment increased pro-survival Bcl-2 MFI in CD8 as much as 1.5-fold in both blood and spleen.

Figure 4. NKTR-255 Effect on CD8 and Memory CD8 is Translatable to Monkey and Human

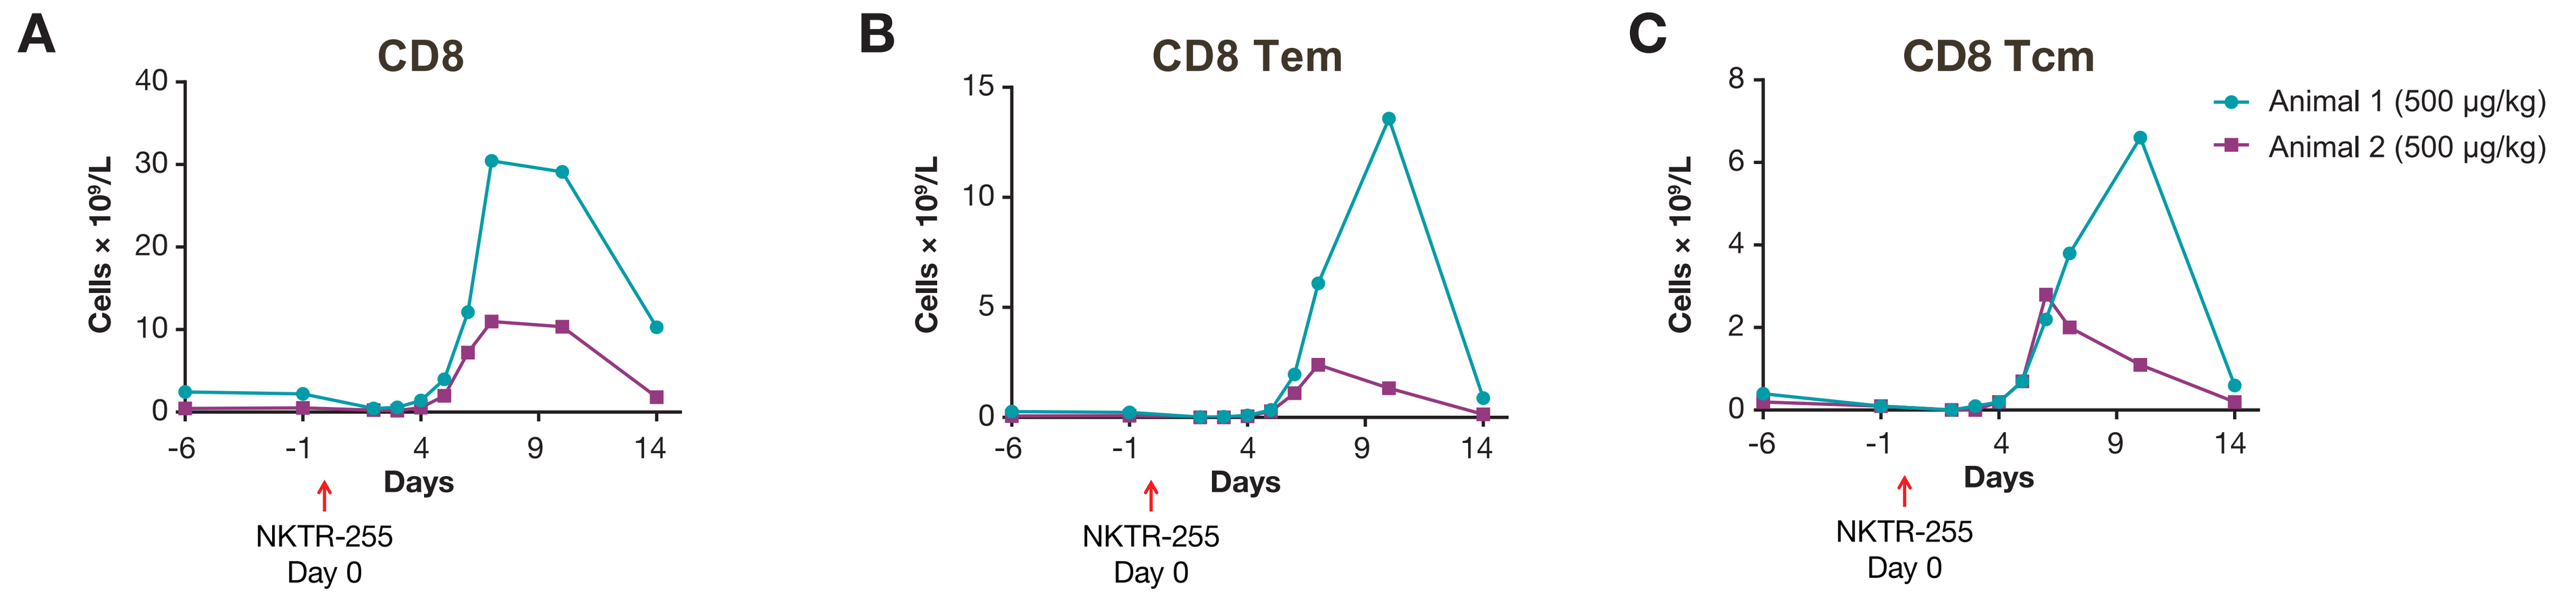
A CD8
B CD8 Tem
C CD8 Tcm
Cells × 10⁹/L
Days
NKTR-255 Day 0
Animal 1 (500 µg/kg)
Animal 2 (500 µg/kg)

(A-C) In cynomolgus monkey, single dose NKTR-255 at 500 µg/kg increased CD8, CD8 Tem, and CD8 Tcm.

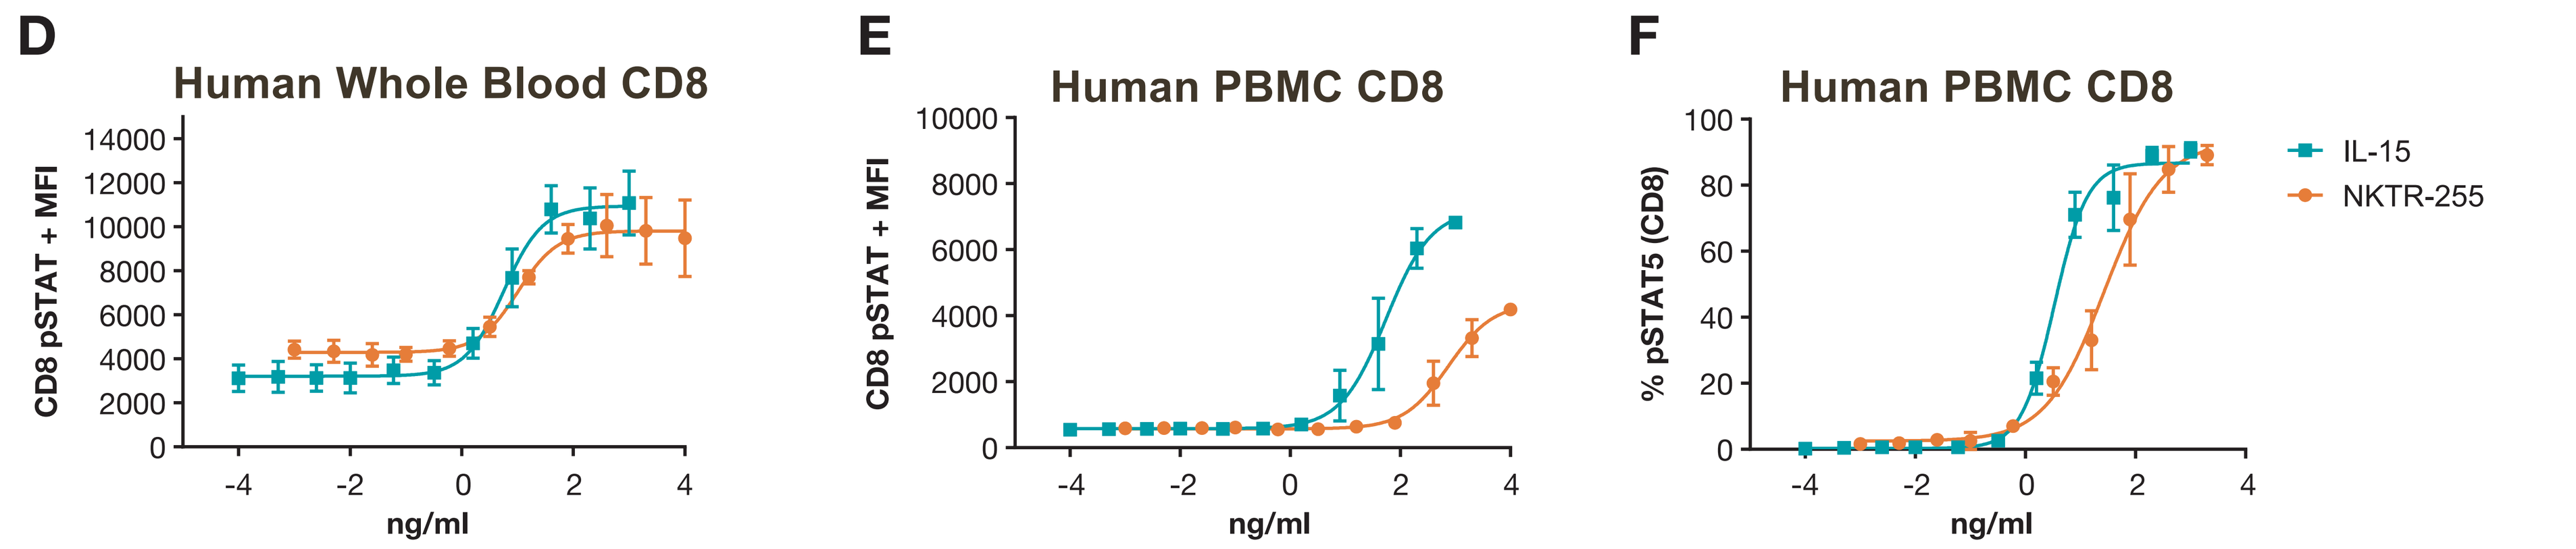
D Human Whole Blood CD8
E Human PBMC CD8
F Human PBMC CD8
CD8 pSTAT + MFI
% pSTAT5 (CD8)
ng/ml
IL-15
NKTR-255

Compound	EC50 (ng/ml)
IL-15	5.6
NKTR-255	10.19

Compound	EC50 (ng/ml)
IL-15	55.07
NKTR-255	751.4

Compound	EC50 (ng/ml)
IL-15	4.9
NKTR-255	27.03

(D-F) In human whole blood, in vitro, CD8 T cells showed dose-dependent induction of pSTAT5+ MFI after a 10-minute treatment with IL-15 or NKTR-255 (n=3). Similar to whole blood, CD8 T cells from human PBMCs (n=3) showed dose-dependent induction of pSTAT5 in both positive MFI and percent positivity after a 10-minute treatment with IL-15 or NKTR-255.

Conclusions

- Single dose NKTR-255 results in sustained IL-15-mediated activity not achievable with conventional IL-15
- NKTR-255 targets CD8 and CD8 memory T cells and robustly drives their proliferation in a prolonged manner
- CD8 Tem, Tcm, and Tscm populations all respond to NKTR-255, proliferating at least fourfold more than vehicle at 0.3 mg/kg
- NKTR-255 has single agent efficacy in the CT-26 lung metastatic model, demonstrating significant lung nodule inhibition
- In tumor-bearing mice, NKTR-255 increases blood and splenic CD8 proliferation and pro-survival factor, Bcl-2
- Single dose NKTR-255 increases CD8 and CD8 memory T cells in cynomolgus monkeys
- NKTR-255 increases CD8 STAT5 phosphorylation in human PBMCs and whole blood in vitro
- NKTR-255 has great potential as an immunotherapeutic with robust stimulation of CD8 and CD8 memory T cells

References

1. Conlon et al. *J Clin Oncol.* 2015;33:74-82.
2. Ikemizu et al. *Nat Immunol.* 2012;13:1141-2.
3. Stonier and Schluns. *Immunol Lett.* 2010;127:85-92.

Presented April 3, 2017 at AACR in Washington, DC